Poster P1174

Tedizolid Activity against a Global Collection of Gram-Positive Bacterial Isolates Causing Bone and Joint Infections (2017–2019)

Cecilia G. Carvalhaes[1], Helio S. Sader[1], Jennifer M. Streit[1], Robert K. Flamm[1], Rodrigo E. Mendes[1]

[1] JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Despite the advances in current health care, bone and joint infections (BJI) remain difficult-to-treat infections that frequently involve prolonged, systemic antibiotic use.
- Tedizolid was approved in the United States in 2014 and Europe in 2015 to treat adults with acute bacterial skin and skin structure infections (ABSSSIs) and is currently under investigation as a treatment option for BJI in adults and children.
- Tedizolid belongs to the oxazolidinone class with activity against Gram-positive pathogens due to the inhibition of the early steps of bacterial protein synthesis.
- This study assessed the *in vitro* activity of tedizolid and comparator agents against a contemporary global collection of Gram-positive isolates causing BJI.

Materials and Methods

Organism collection

- A total of 523 Gram-positive cocci were collected from patients with BJI as part of the Surveillance of Tedizolid Activity and Resistance (STAR) program for 2017–2019.
- The isolates were recovered from a global network of 68 medical centres, including 23 sites in the United States (224 isolates; 42.8%), 28 in Europe (200 isolates; 38.2%), 7 in Latin America (52 isolates; 9.9%), and 10 in the Asia-Pacific region (47 isolates; 9.0%; Figure 1).
- Bacterial isolates were determined to be significant by local criteria as the probable cause of BJI and only 1 isolate per patient infection episode was included.
- Bacterial identification was confirmed by matrix-assisted laser desorption ionization–time of flight mass spectrometry (Bruker Daltonics, Massachusetts, USA) following the manufacturer's instructions.

Antimicrobial susceptibility testing

- Broth microdilution tests were performed according to the Clinical and Laboratory Standards Institute (CLSI) guidelines to determine the susceptibility of tedizolid and numerous comparator agents.
 - Broth microdilution used 96-well reference panels manufactured by JMI Laboratories (North Liberty, Iowa, USA) containing cation-adjusted Mueller-Hinton broth (CAMHB) as testing media.
 - CAMHB supplemented with 2.5-5% lysed horse blood was used for streptococci.
 - CAMHB supplemented with 50 mg/L calcium (Ca^{2+}) was used for testing daptomycin.
- Quality control (QC) strains included *Staphylococcus aureus* ATCC 29213, *Enterococcus faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619.
- EUCAST (2019) breakpoint criteria for tedizolid and comparator agents were utilized.

Results

- *S. aureus* (379, 72.5%) was the most common Gram-positive pathogen recovered from BJI followed by β-haemolytic streptococci (BHS; 81; 15.5%), coagulase-negative staphylococci (CoNS; 25; 4.8%), *E. faecalis* (20; 3.8%), and Viridans group streptococci (VGS; 11; 2.1%) (Figure 2).
- Tedizolid was active against *S. aureus* isolates ($MIC_{50/90}$, 0.25/0.25 mg/L) and retained *in vitro* activity against methicillin-resistant *S. aureus* subset (MRSA; $MIC_{50/90}$, 0.12/0.25 mg/L).
 - Tedizolid, linezolid, vancomycin, and daptomycin inhibited all *S. aureus* isolates at the respective susceptible breakpoints (Table 1).
 - MRSA rates varied among regions (17.2%-35.1%); however, based on MIC_{90}, tedizolid showed consistent potencies (MIC_{90}, 0.25-0.5 mg/L), regardless of geographic region (Table 2).
- Among the entire collection, only 1 CoNS isolate (*Staphylococcus cohnii*) was resistant to linezolid (MIC, 8 mg/L) and tedizolid (MIC, >1 mg/L).
- Tedizolid ($MIC_{50/90}$, 0.25/0.5 mg/L) was also active against *E. faecalis* isolates, as well as daptomycin ($MIC_{50/90}$, 1/1 mg/L), linezolid ($MIC_{50/90}$, 1/2 mg/L), and vancomycin ($MIC_{50/90}$, 1/2 mg/L; Table 1).
- All BHS isolates were inhibited by tedizolid at ≤0.5 mg/L ($MIC_{50/90}$, 0.25/0.25 mg/L; 100% susceptible).
 - Penicillin, linezolid, vancomycin, and daptomycin were also active (100% susceptible) against all BHS (Figure 3).
- Tedizolid and vancomycin inhibited all VGS isolates at the respective susceptible breakpoints (Table 1).
 - No differences on tedizolid activity against BHS or VGS were observed among geographic regions.

Conclusions

- Tedizolid demonstrated potent *in vitro* activity against this worldwide collection of contemporary Gram-positive isolates causing BJI.
- Tedizolid showed consistent *in vitro* activity, regardless of geographic area.
- High susceptibility rates were observed by tedizolid and comparator agents against the most frequent organisms and organism groups, including MRSA.
- These findings support the clinical development of tedizolid for treating BJI infections caused by Gram-positive pathogens.

Figure 1 Number of isolates and medical centres by geographic region

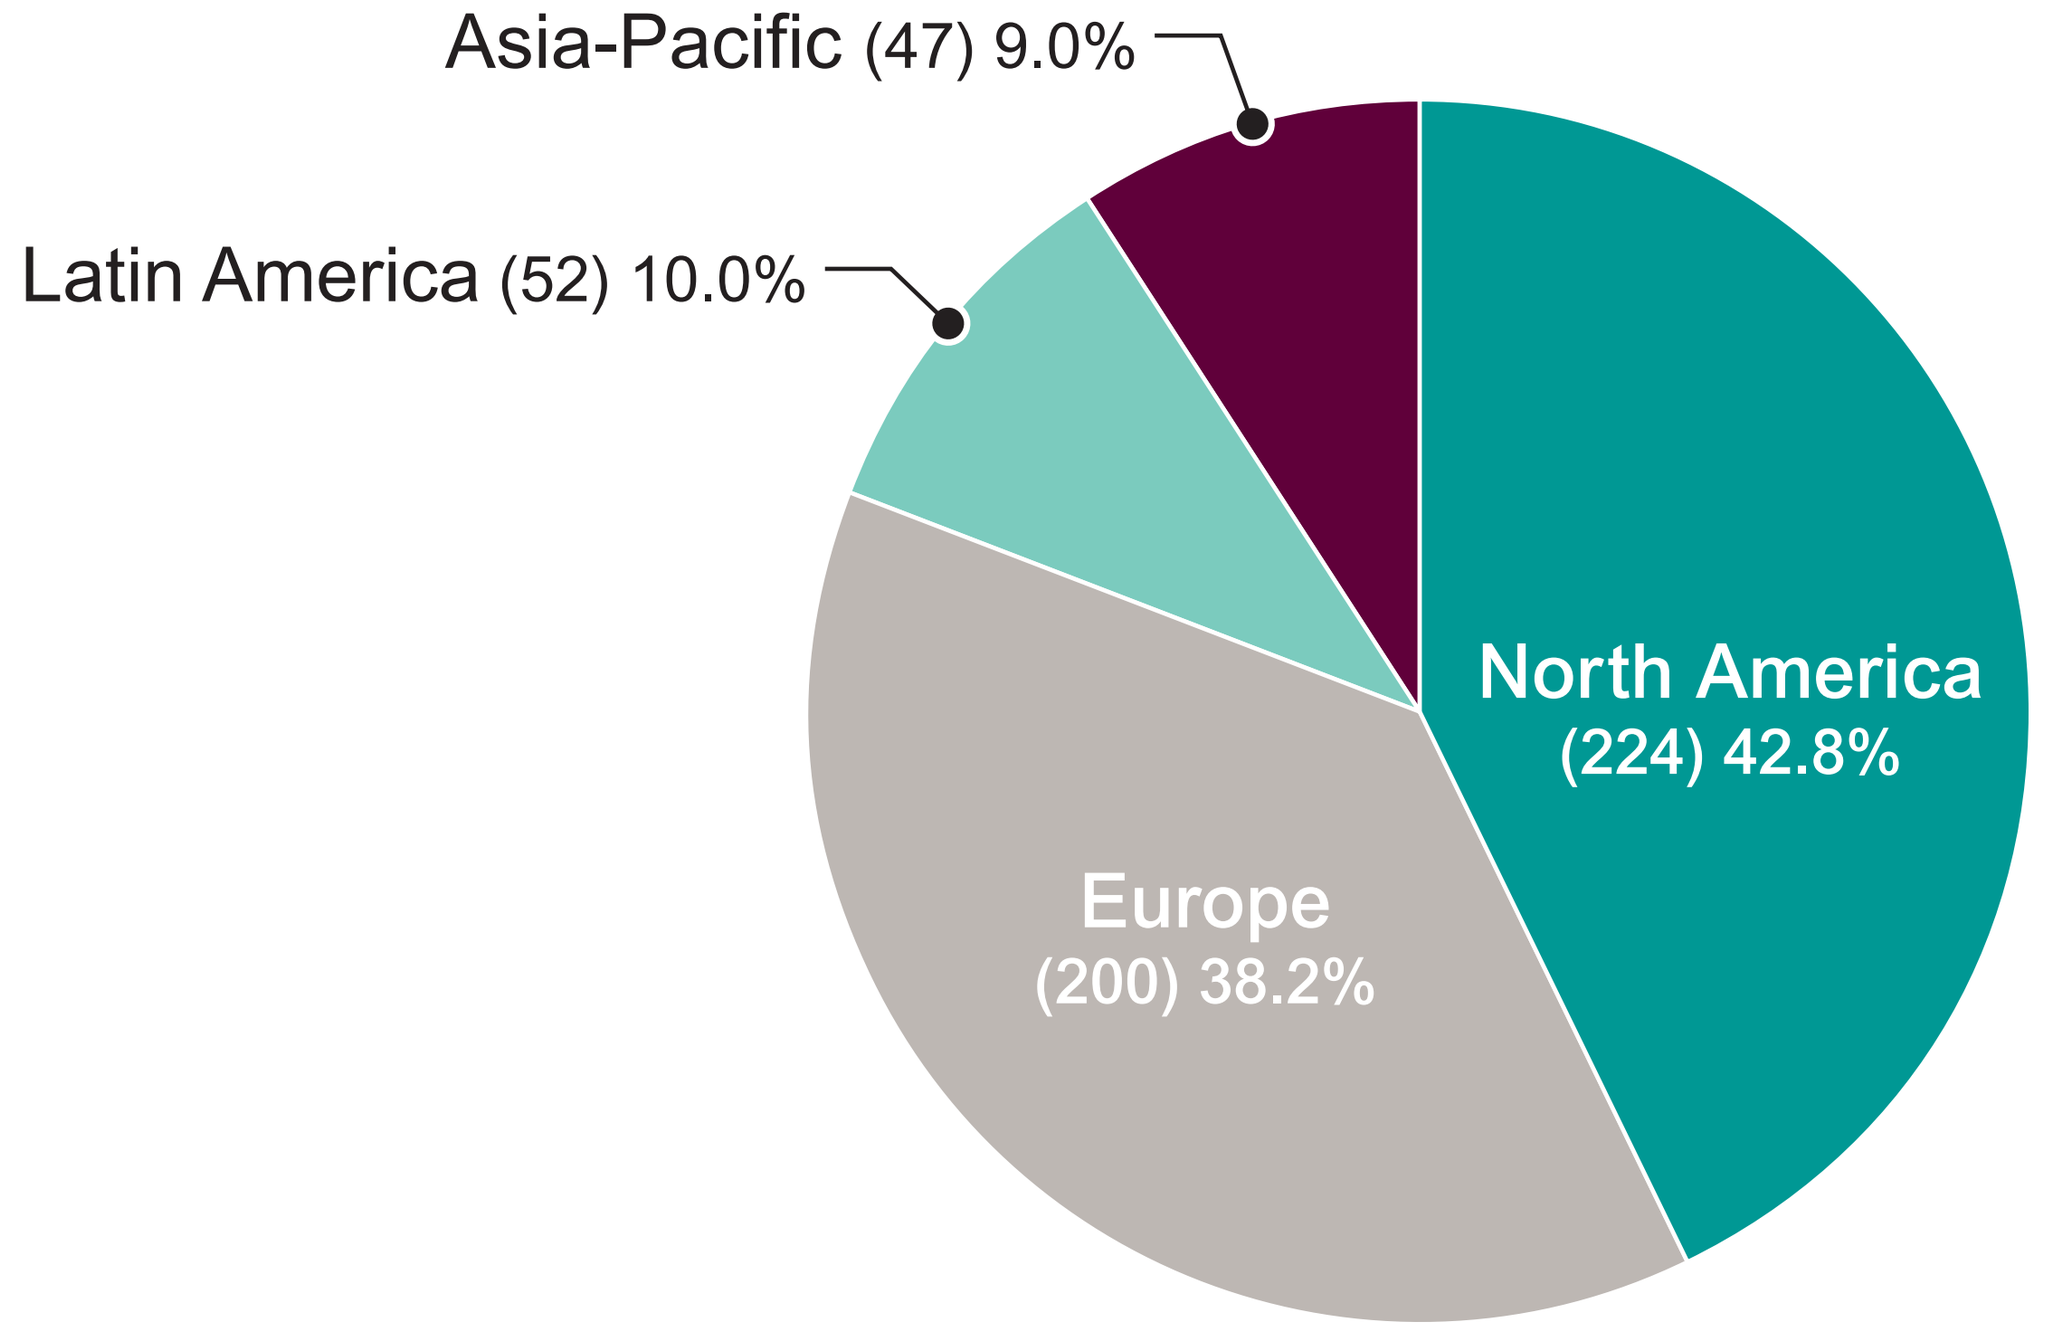

Asia-Pacific (47) 9.0%
Latin America (52) 10.0%
North America
(224) 42.8%
Europe
(200) 38.2%

Figure 2 Gram-positive isolates recovered from patients with BJI worldwide (2017–2019)

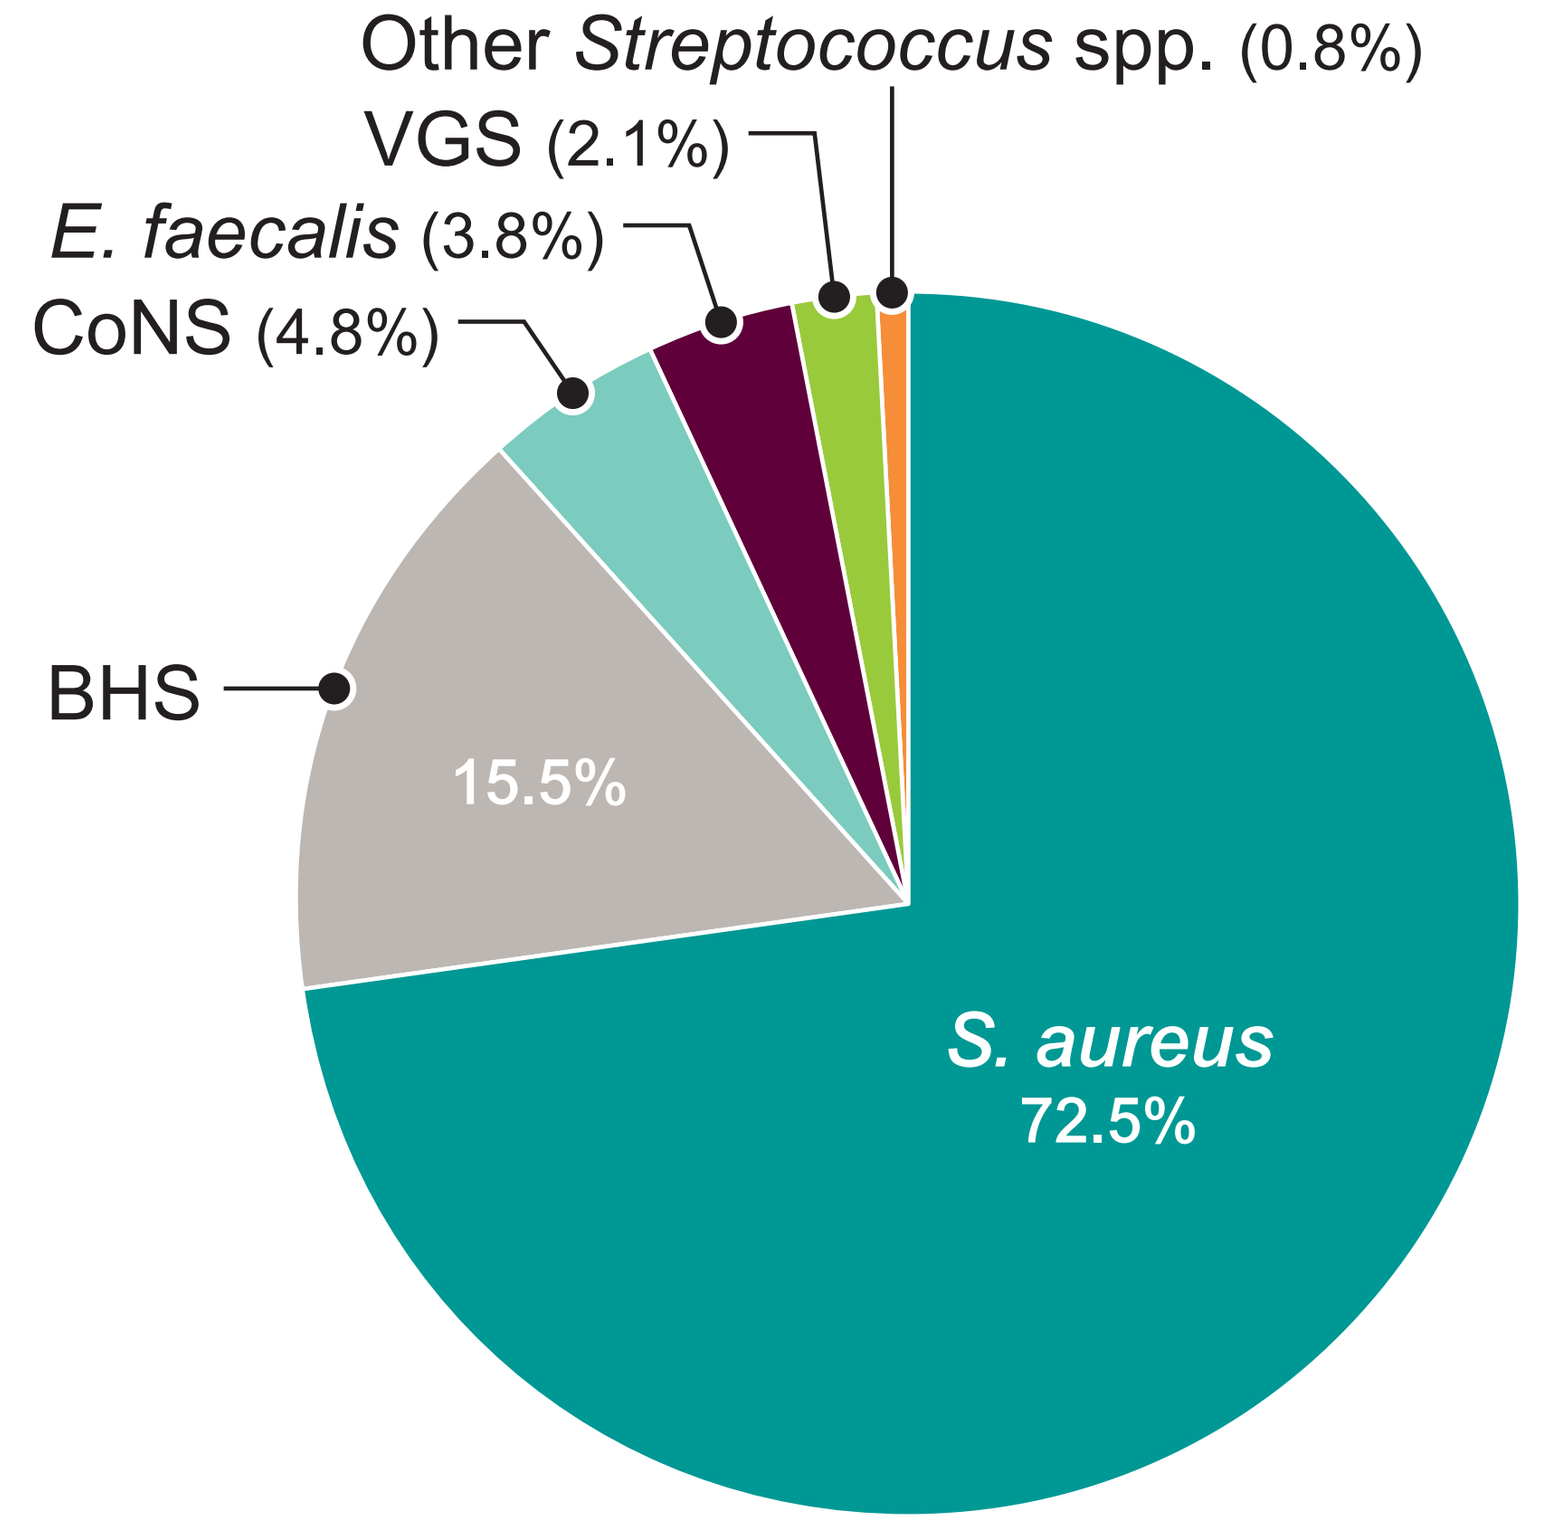

Other *Streptococcus* spp. (0.8%)
VGS (2.1%)
E. faecalis (3.8%)
CoNS (4.8%)
BHS
15.5%
S. aureus
72.5%

Figure 3 Tedizolid and comparators susceptibility rates against Gram-positive pathogens causing BJI worldwide (2017–2019)

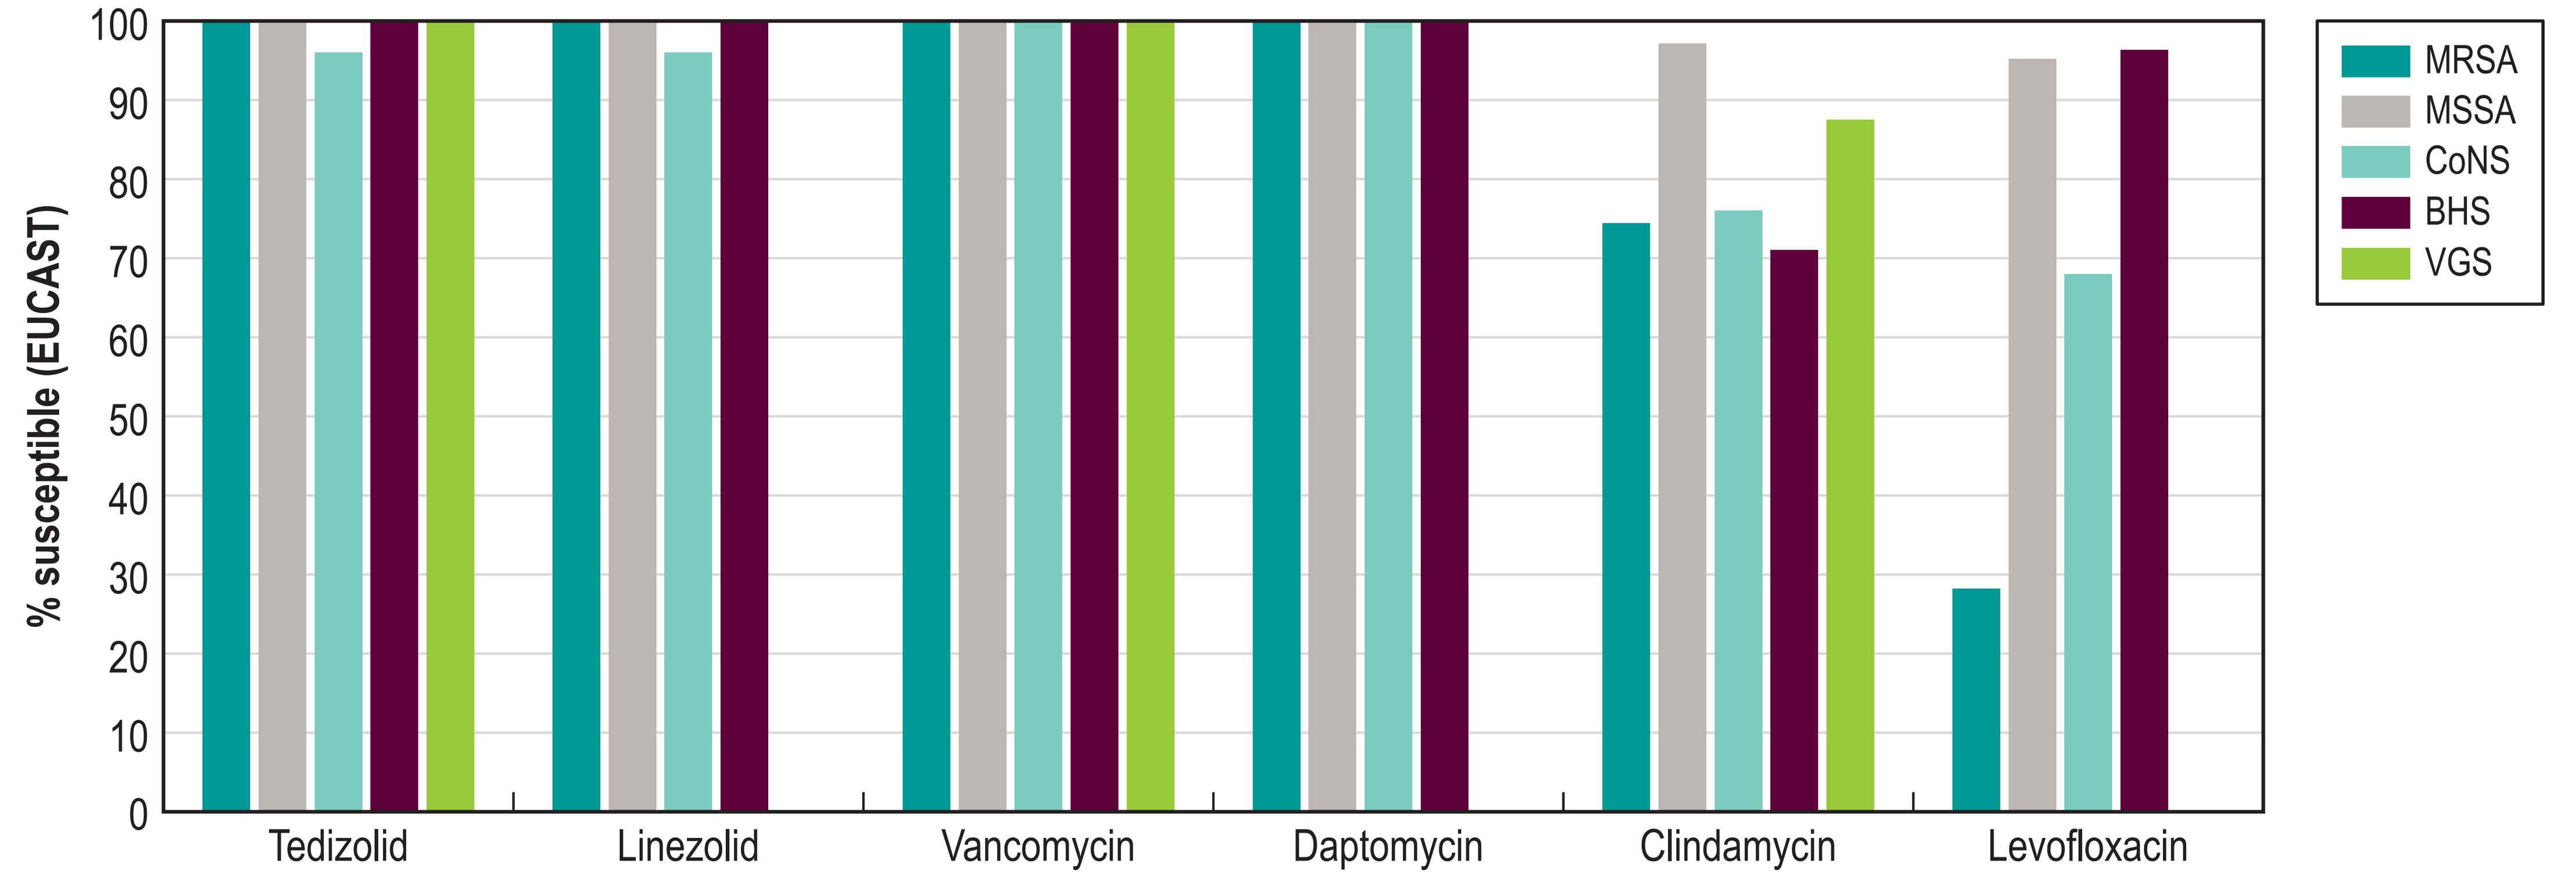

% susceptible (EUCAST)
0
10
20
30
40
50
60
70
80
90
100
Tedizolid
Linezolid
Vancomycin
Daptomycin
Clindamycin
Levofloxacin
MRSA
MSSA
CoNS
BHS
VGS

Breakpoint not available for some agent/organism combinations

Table 1 Activity of tedizolid and comparator agents against Gram-positive pathogens causing BJI worldwide (2017–2019)

Organism (no. tested)	MIC_{50}/MIC_{90} in mg/L (%S, EUCAST)						
Phenotype	Tedizolid	Linezolid	Vancomycin	Daptomycin	Clindamycin	Levofloxacin	Penicillin
MSSA (276)	0.25/0.25 (100)	1/2 (100)	1/1 (100)	0.25/0.25 (100)	0.06/0.06 (97.1)	0.25/0.25 (95.2)	ND
MRSA (103)	0.12/0.25 (100)	1/2 (100)	1/1 (100)	0.25/0.5 (100)	0.06/>2 (74.4)	>4/>4 (28.2)	ND
CoNS (25)	0.12/0.25 (96)	1/1 (96)	1/2 (100)	0.25/0.5 (100)	0.06/>2 (76.0)	0.25/>4 (68.0)	ND
E. faecalis (20)	0.25/0.5 (—)	1/2 (100)	1/2 (100)	1/1 (—)	ND	1/>4 (81.2)[b]	ND
BHS (81)	0.25/0.25 (100)	1/2 (100)	0.5/0.5 (100)	0.12/0.25 (100)	≤0.25/>2 (71.0)	0.5/1 (96.3)	0.03/0.06 (100)
VGS (11)	0.25/0.25 (100)[a]	1/2 (—)	0.5/1 (100)	0.25/0.5 (—)	≤0.25/— (87.5)	1/2 (—)	0.03/0.25 (90.9)

S, susceptible; MRSA, methicillin-resistant *S. aureus*; BHS = β-haemolytic streptococci; VGS = viridans group streptococci; ND, not determined; "—" breakpoint not available.
[a] Tedizolid breakpoint for *S. anginosus* group used for VGS
[b] Levofloxacin breakpoint for uncomplicated urinary tract infection only

Table 2 Tedizolid activity against MRSA isolates causing BJI and MRSA rate per geographic region

Geographic region	MRSA rate (%)	Tedizolid $MIC_{50/90}$ (mg/L)
North America	34.6	0.12 / 0.25
Europe	17.2	0.12 / 0.25
Latin America	27.3	0.25 / 0.5
Asia Pacific	35.1	0.12 / 0.25
All regions	27.2	0.12 / 0.25

Acknowledgements

Funding for this research was provided by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA.

References

Carvalhaes CG, Sader HS, Flamm RK, Mendes RE. Tedizolid in vitro activity against Gram-positive clinical isolates causing bone and joint infections in hospitals in the USA and Europe (2014-17). J Antimicrob Chemother. 2019;74(7):1928–1933.

Locke JB, Zurenko GE, Shaw KJ, Bartizal K. Tedizolid for the management of human infections: in vitro characteristics. Clin Infect Dis. 2014;58(Suppl 1):S35–42.

Osmon DR, Berbari EF, Berendt AR *et al.* Diagnosis and management of prosthetic joint infection: clinical practice guidelines by the Infectious Diseases Society of America. *Clin Infect Dis* 2013; 56: e1-e25.

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.

EUCAST (2019). Breakpoint tables for interpretation of MICs and zone diameters. Version 9.0, January 2019. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_9.0_Breakpoint_Tables.pdf

European public assessment report (EPAR) for Sivextro. Available at https://www.ema.europa.eu/en/medicines/human/EPAR/sivextro

Sinvextro (tedizolid phosphate) package insert. Revised August 2017. Document available online at: https://www.merck.com/product/usa/pi_circulars/s/sivextro/sivextro_pi.pdf

Contact Information
Cecilia Carvalhaes, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia-carvalhaes@jmilabs.com

This poster was originally intended for presentation at the 30th ECCMID (Paris, France; April 18–21, 2020). This meeting was canceled due to the COVID-19 pandemic. The corresponding accepted abstract can be found in the 30th ECCMID abstract book (Abstract 2359).

Presented at ECCMID 2020, April 18–21, 2020, Paris, France

Copyright © 2020 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.