1823

Presented at IDWeek 2016
October 26-30, 2016
New Orleans, LA, USA

Activity of Ceftolozane/Tazobactam and Comparators Tested Against Carbapenem-Nonsusceptible *Pseudomonas aeruginosa* Isolates

Shortridge D; Castanheira M; Duncan LR; Sader HS; Flamm RK

JMI Laboratories, North Liberty, Iowa, USA

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Ste A
North Liberty, IA 52317, USA
319-665-3370
dee-shortridge@jmilabs.com

INTRODUCTION

- Ceftolozane, a novel oxyimino-aminothiazolyl cephalosporin, exerts potent activity against Enterobacteriaceae (similar to other oxyimino-aminothiazolyl cephalosporins) and has demonstrated greater activity than ceftazidime against *Pseudomonas aeruginosa*
 - Ceftolozane maintains its stability against many *P. aeruginosa* resistance mechanisms, including AmpC hyperproduction and efflux mechanisms, and is little affected by porin deficiency
 - However, as with other oxyimino-aminothiazolyl cephalosporins, ceftolozane's activity is compromised in bacteria producing extended-spectrum β-lactamases (ESBLs), stably derepressed AmpC β-lactamases, and carbapenemases
- Tazobactam, a penicillanic acid sulfone, is a well-established β-lactamase inhibitor that extends the spectrum of β-lactam agents
- Ceftolozane/tazobactam is a novel antibacterial combination with activity against *P. aeruginosa* (including drug-resistant strains) and against other common Gram-negative pathogens, among them many ESBL-producing Enterobacteriaceae
- Although carbapenems are some of the most effective agents for treating Gram-negative infections (including those caused by *P. aeruginosa*), carbapenem resistance in *P. aeruginosa* limits the available treatment options; these pathogens are commonly resistant to other β-lactam agents as well
 - Empiric and targeted therapies to treat infections with these organisms are becoming increasingly limited
- In the present study, we compared the activity of ceftolozane/tazobactam on *P. aeruginosa* isolates nonsusceptible to ≥1 carbapenem (doripenem, imipenem, meropenem) between 2012 and 2015 from both the United States and Europe

MATERIALS AND METHODS

Sampling sites and organisms

- *P. aeruginosa* isolates were consecutively collected by the Program to Assess Ceftolozane/Tazobactam Susceptibility (PACTS) between 2012 and 2015 from 74 medical centers throughout Europe and from all 9 US census divisions
- *P. aeruginosa* isolates were selected if they were nonsusceptible to ≥1 carbapenem (doripenem, imipenem, meropenem) according to Clinical and Laboratory Standards Institute (CLSI) 2016 breakpoints
- The isolates were collected from patients in 17 European countries (including Turkey and Israel) and the United States (21 states)
- All organisms were isolated from documented infections, and only 1 isolate per patient infection episode was included in the surveillance collection
- The distribution of infection type for the carbapenem-nonsusceptible *P. aeruginosa* isolates is shown in **Figure 1**. Most (53%) isolates were from hospital patients with pneumonia

Figure 1. Infection type for 2507 carbapenem-nonsusceptible *P. aeruginosa* isolates (2012-2015).

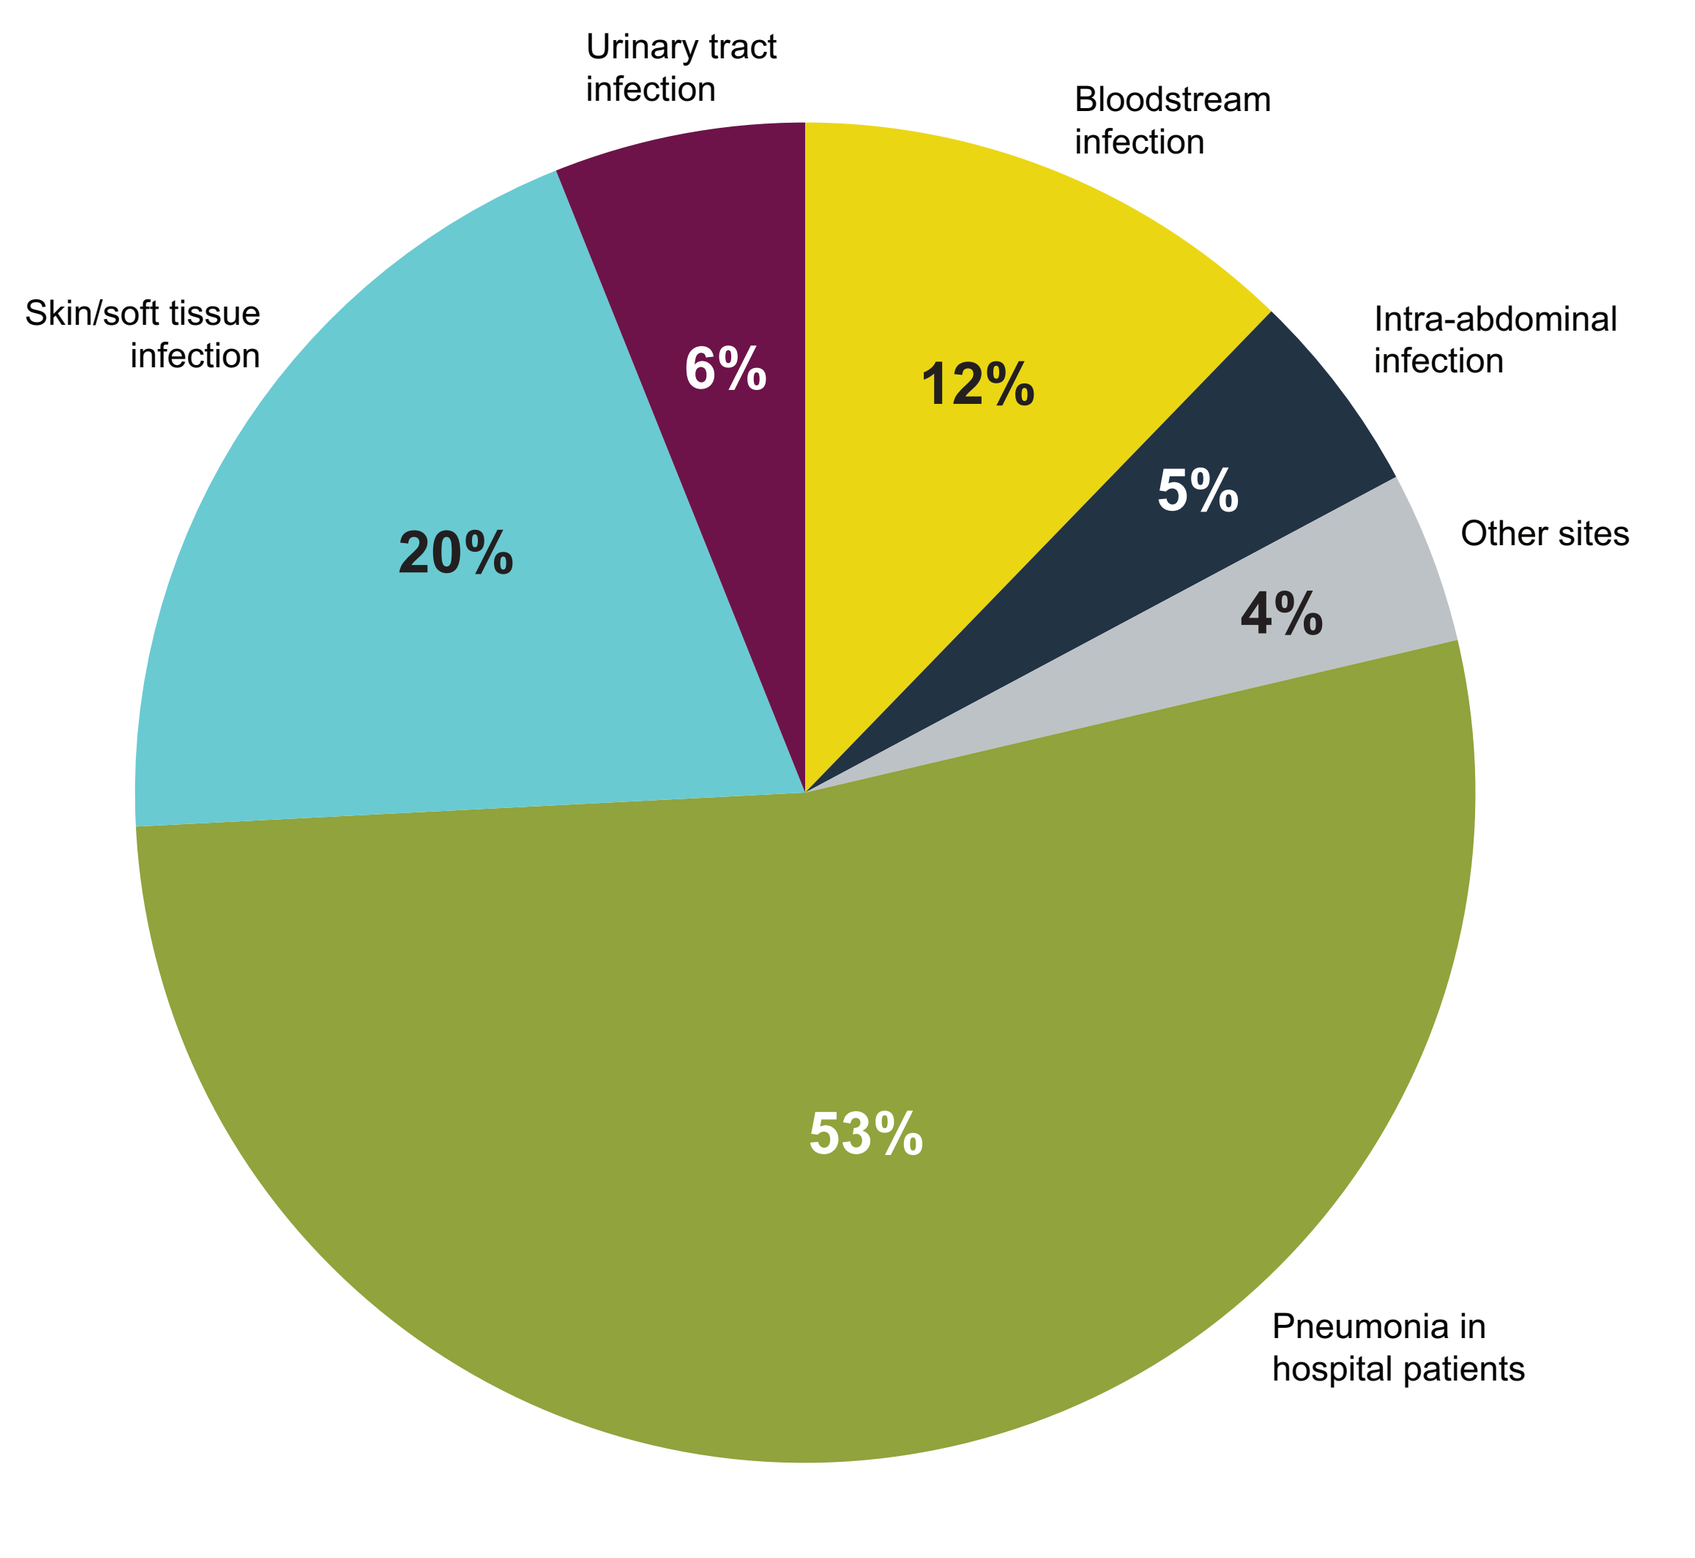
Urinary tract infection
Bloodstream infection
Intra-abdominal infection
Other sites
Skin/soft tissue infection
Pneumonia in hospital patients
6%
12%
5%
4%
20%
53%

Antimicrobial susceptibility testing

- Minimum inhibitory concentration (MIC) values were determined using the reference CLSI broth microdilution method (M07-A10)
 - Quality control (QC) ranges and interpretive criteria for comparator compounds used the CLSI M100-S26 guidelines
 - QC strains included *Escherichia coli* ATCC 25922 and NCTC 13353, *Klebsiella pneumoniae* ATCC 700603, and *P. aeruginosa* ATCC 27853
 - All QC results were within published ranges

RESULTS

- Among 8423 *P. aeruginosa* isolates, 2507 (29.8%) were nonsusceptible using the CLSI criteria of ≥1 μg/mL for the carbapenems meropenem, imipenem, and doripenem
- Carbapenem-nonsusceptible *P. aeruginosa* displayed low susceptibility rates for various agents tested (**Table 1**)
 - Colistin ($MIC_{50/90}$, 1/2 μg/mL; 98.8/99.7% susceptible per CLSI/EUCAST criteria) was the most active agent, followed by ceftolozane/tazobactam and amikacin
- Ceftolozane/tazobactam ($MIC_{50/90}$, 1/>32 μg/mL) inhibited 77.9% of the carbapenem-nonsusceptible isolates at current susceptible CLSI/EUCAST breakpoints
 - Amikacin was active against 78.8% of the isolates using the CLSI interpretive criteria and 68.4% of the isolates applying EUCAST breakpoints
 - Cefepime, ceftazidime, and piperacillin/tazobactam were active against 52.5%, 50.3%, and 42.0% of the isolates, respectively, using CLSI or EUCAST breakpoints

Table 1. Activity of ceftolozane/tazobactam and comparators versus carbapenem-nonsusceptible *P. aeruginosa*

	All N = 2507				United States n = 894				Europe n = 1613			
Antimicrobial Agent	MIC_{50}	MIC_{90}	%S CLSI	%S EUCAST	MIC_{50}	MIC_{90}	%S CLSI	%S EUCAST	MIC_{50}	MIC_{90}	%S CLSI	%S EUCAST
Ceftolozane/tazobactam	1	>32	77.9	77.9	1	4	90.0	90.0	2	>32	71.2	71.2
Amikacin	4	>32	78.8	68.4	4	16	91.7	82.3	8	>32	71.7	60.8
Cefepime	8	>16	52.5	52.5	8	>16	60.4	60.4	16	>16	48.2	48.2
Ceftazidime	8	>32	50.3	50.3	8	>32	61.6	61.6	16	>32	44.0	44.0
Colistin	1	2	98.8	99.7	1	2	98.8	99.7	1	2	98.9	99.7
Meropenem	8	>8	18.8	18.8	8	>8	21.7	21.7	8	>8	17.2	17.2
Levofloxacin	>4	>4	37.0	26.1	4	>4	39.0	27.3	>4	>4	35.8	25.4
Piperacillin/tazobactam	32	>64	42.0	42.0	16	>64	51.5	51.5	32	>64	36.7	36.7

CLSI = Clinical and Laboratory Standards Institute; EUCAST = European Committee on Antimicrobial Susceptibility Testing; MIC_{50} = minimum inhibitory concentration to inhibit growth of 50% of isolates; MIC_{90} = minimum inhibitory concentration to inhibit growth of 90% of isolates; S = susceptible.

- Ceftolozane/tazobactam inhibited 90% of the carbapenem-nonsusceptible *P. aeruginosa* from the United States and 71.2% of the isolates from Europe at current susceptible CLSI/EUCAST breakpoints
 - For carbapenem-nonsusceptible *P. aeruginosa* in this study, ceftolozane/tazobactam inhibited 70% (1271/1815), 77% (1804/2343), and 73% (1490/2035) of the doripenem-, imipenem-, and meropenem-nonsusceptible isolates, respectively
- Overall, except for colistin, isolates from Europe displayed lower susceptibility rates for comparator agents than isolates from the United States (**Table 1**)
- **Tables 2** and **3** show the activity of ceftolozane/tazobactam and comparator agents versus carbapenem-nonsusceptible *P. aeruginosa* isolates from the United States and Europe, respectively
- **Figure 2** shows the ceftolozane/tazobactam susceptibility rates for carbapenem-nonsusceptible *P. aeruginosa* from the 9 US census divisions. Differences in activity between them range from 78.5% susceptible in the Pacific division to 98.6% susceptible in the West North Central division
- **Figure 3** shows the ceftolozane/tazobactam susceptibility rates for carbapenem-nonsusceptible *P. aeruginosa* from the 17 participating European countries (including Turkey and Israel). Differences in activity between the countries range from 0.0% susceptible in Belarus to 97.9% susceptible in Ireland

Table 2. Activity of ceftolozane/tazobactam and comparators tested against carbapenem-nonsusceptible *P. aeruginosa* isolates from the United States (PACTS, 2012-2015)

	$MIC_{50/90}$, μg/mL							
Division (no. isolates)	Ceftolozane/ tazobactam	Amikacin	Cefepime	Ceftazidime	Colistin	Levofloxacin	Meropenem	Piperacillin/ tazobactam
New England (102)	1/4	4/16	8/>16	4/>32	1/2	>4/>4	4/>8	8/>64
Mid-Atlantic (138)	1/8	4/16	8/>16	8/>32	1/2	4/>4	8/>8	32/>64
East North Central (127)	1/4	4/8	8/>16	8/>32	1/2	4/>4	8/>8	16/>64
West North Central (72)	1/4	4/8	8/>16	4/>32	1/2	>4/>4	4/>8	16/>64
South Atlantic (128)	1/8	4/32	8/>16	4/>32	1/2	4/>4	8/>8	16/>64
East South Central (76)	1/4	4/16	8/>16	4/32	1/2	>4/>4	8/>8	16/>64
West South Central (111)	1/4	4/32	8/>16	8/>32	1/2	>4/>4	8/>8	32/>64
Mountain (47)	1/2	4/8	8/16	4/>32	1/2	4/>4	8/>8	16/>64
Pacific (93)	1/16	8/>32	8/>16	8/>32	1/2	>4/>4	8/>8	32/>64

MIC_{50} = minimum inhibitory concentration to inhibit growth of 50% of isolates; MIC_{90} = minimum inhibitory concentration to inhibit growth of 90% of isolates; PACTS = Program to Assess Ceftolozane/Tazobactam Susceptibility.

Table 3. Activity of ceftolozane/tazobactam and comparators tested against carbapenem-nonsusceptible *P. aeruginosa* isolates from Europe (PACTS, 2012-2015)

	$MIC_{50/90}$, μg/mL							
Country (no. isolates)	Ceftolozane/ tazobactam	Amikacin	Cefepime	Ceftazidime	Colistin	Levofloxacin	Meropenem	Piperacillin/ tazobactam
Belarus (30)	>32/>32	>32/>32	>16/>16	32/>32	≤0.5/1	>4/>4	>32/>32†	64/>64
Belgium (63)	2/>32	8/>32	16/>16	16/>32	1/2	>4/>4	>8/>8	32/>64
Czech Republic (25)	1/>32	4/>32	8/>16	16/>32	1/2	>4/>4	8/>8	32/>64
France (119)	1/4	4/32	8/>16	8/>32	2/2	4/>4	4/8	32/>64
Germany (144)	1/8	4/>32	8/>16	8/>32	1/2	4/>4	8/>8	32/>64
Greece (51)	8/>32	8/>32	16/>16	16/>32	2/2	>4/>4	>8/>8	32/>64
Ireland (48)	1/4	4/32	8/16	4/>32	1/2	2/>4	4/>8	16/>64
Israel (71)	1/4	4/16	8/>16	8/>32	1/2	4/>4	4/>8	16/>64
Italy (148)	2/>32	8/>32	16/>16	16/>32	1/2	>4/>4	8/>8	32/>64
Poland (202)	2/>32	16/>32	16/>16	16/>32	1/2	>4/>4	8/>8	64/>64
Portugal (111)	16/32	16/16	16/>16	>32/>32	1/2	>4/>4	>8/>8	>64/>64
Russia (179)	8/>32	>32/>32	16/>16	32/>32	1/2	>4/>4	>8/>8	32/>64
Spain (144)	2/16	4/16	8/>16	16/>32	1/2	>4/>4	8/>8	64/>64
Sweden (29)	1/4	4/8	8/16	4/>32	1/2	1/>4	4/>8	16/>64
Turkey (177)	1/16	4/32	8/>16	8/>32	1/2	2/>4	8/>8	32/>64
Ukraine (28)	32/>32	>32/>32	16/>16	32/>32	1/2	>4/>4	>8/>8	64/>64
United Kingdom (44)	1/2	2/4	4/16	4/32	1/2	1/>4	8/>8	16/64

MIC_{50} = minimum inhibitory concentration to inhibit growth of 50% of isolates; MIC_{90} = minimum inhibitory concentration to inhibit growth of 90% of isolates; PACTS = Program to Assess Ceftolozane/Tazobactam Susceptibility.

†Test range for meropenem changed in 2015.

Figure 2. Comparison of ceftolozane/tazobactam susceptibility rates† for carbapenem-nonsusceptible *P. aeruginosa* in US census divisions (2012-2015).

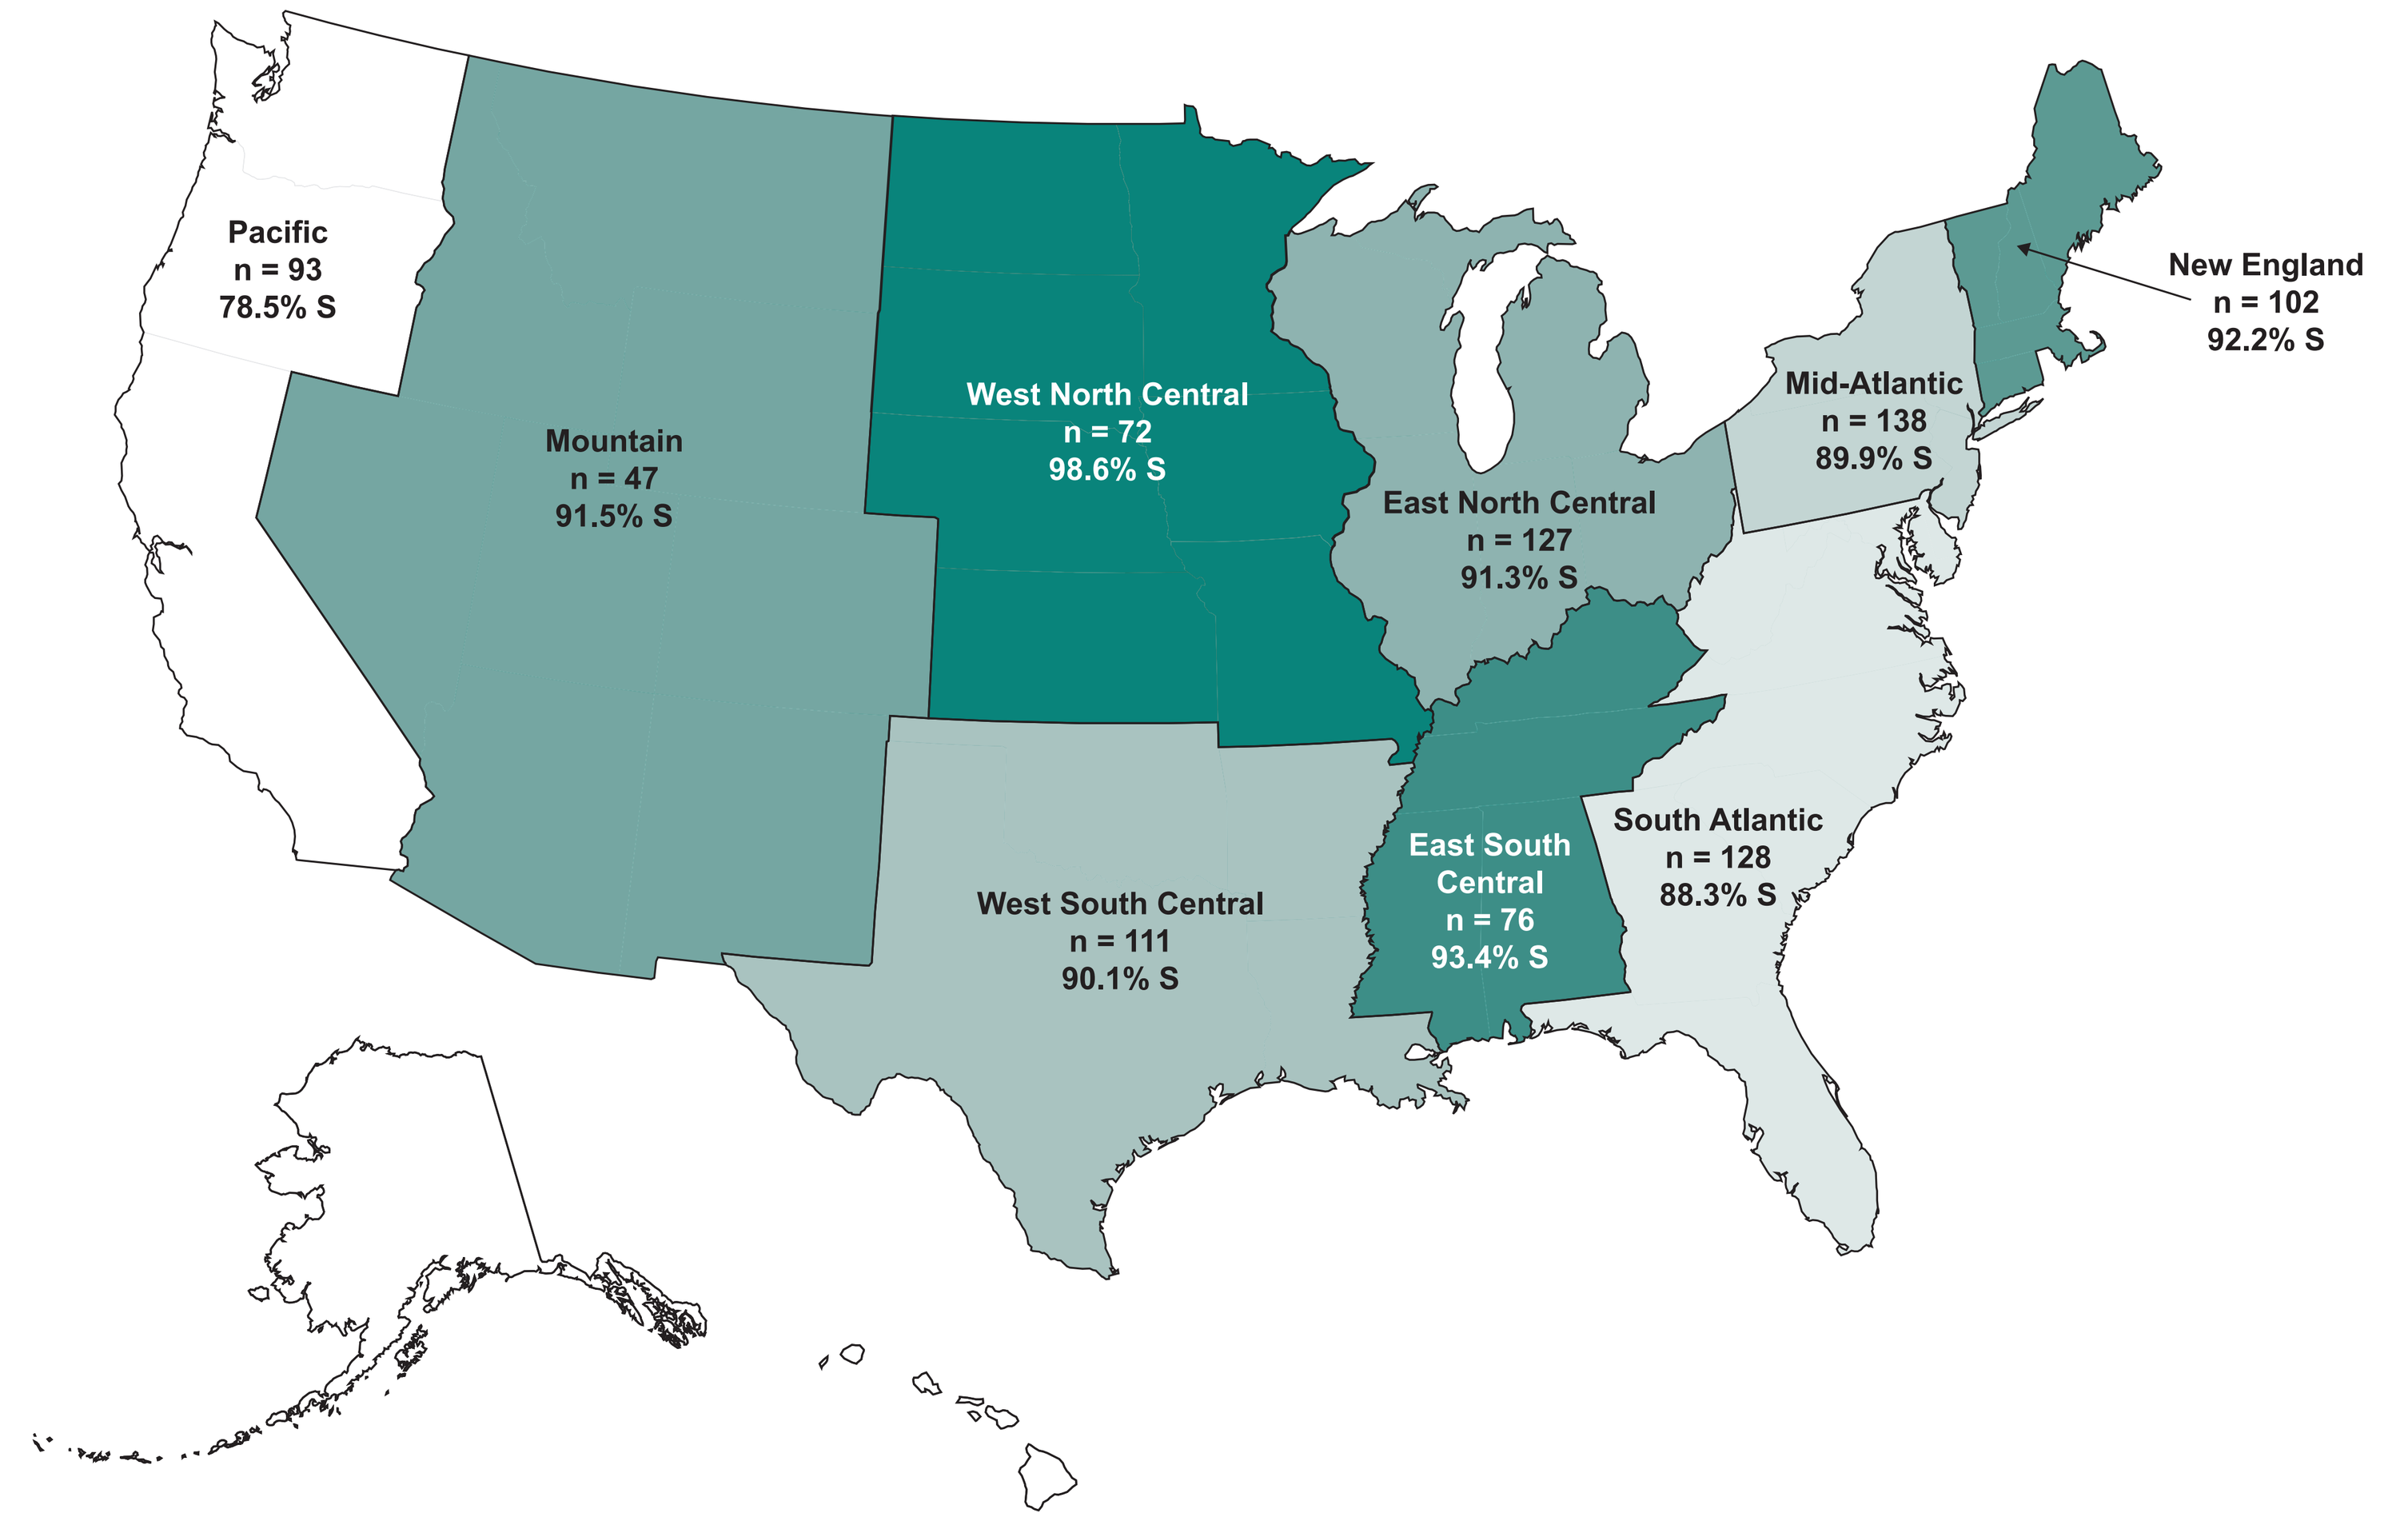
Pacific
n = 93
78.5% S
Mountain
n = 47
91.5% S
West North Central
n = 72
98.6% S
East North Central
n = 127
91.3% S
Mid-Atlantic
n = 138
89.9% S
New England
n = 102
92.2% S
West South Central
n = 111
90.1% S
East South Central
n = 76
93.4% S
South Atlantic
n = 128
88.3% S

CLSI = Clinical and Laboratory Standards Institute; EUCAST = European Committee on Antimicrobial Susceptibility Testing; S = susceptibility.
Green coloration denotes susceptibility rates: the darker the color, the greater the susceptibility of *P. aeruginosa* to ceftolozane/tazobactam in that division.
†Based on 2016 CLSI/EUCAST.

Figure 3. Comparison of ceftolozane/tazobactam susceptibility rates† for carbapenem-nonsusceptible *P. aeruginosa* in European countries (2012-2015).

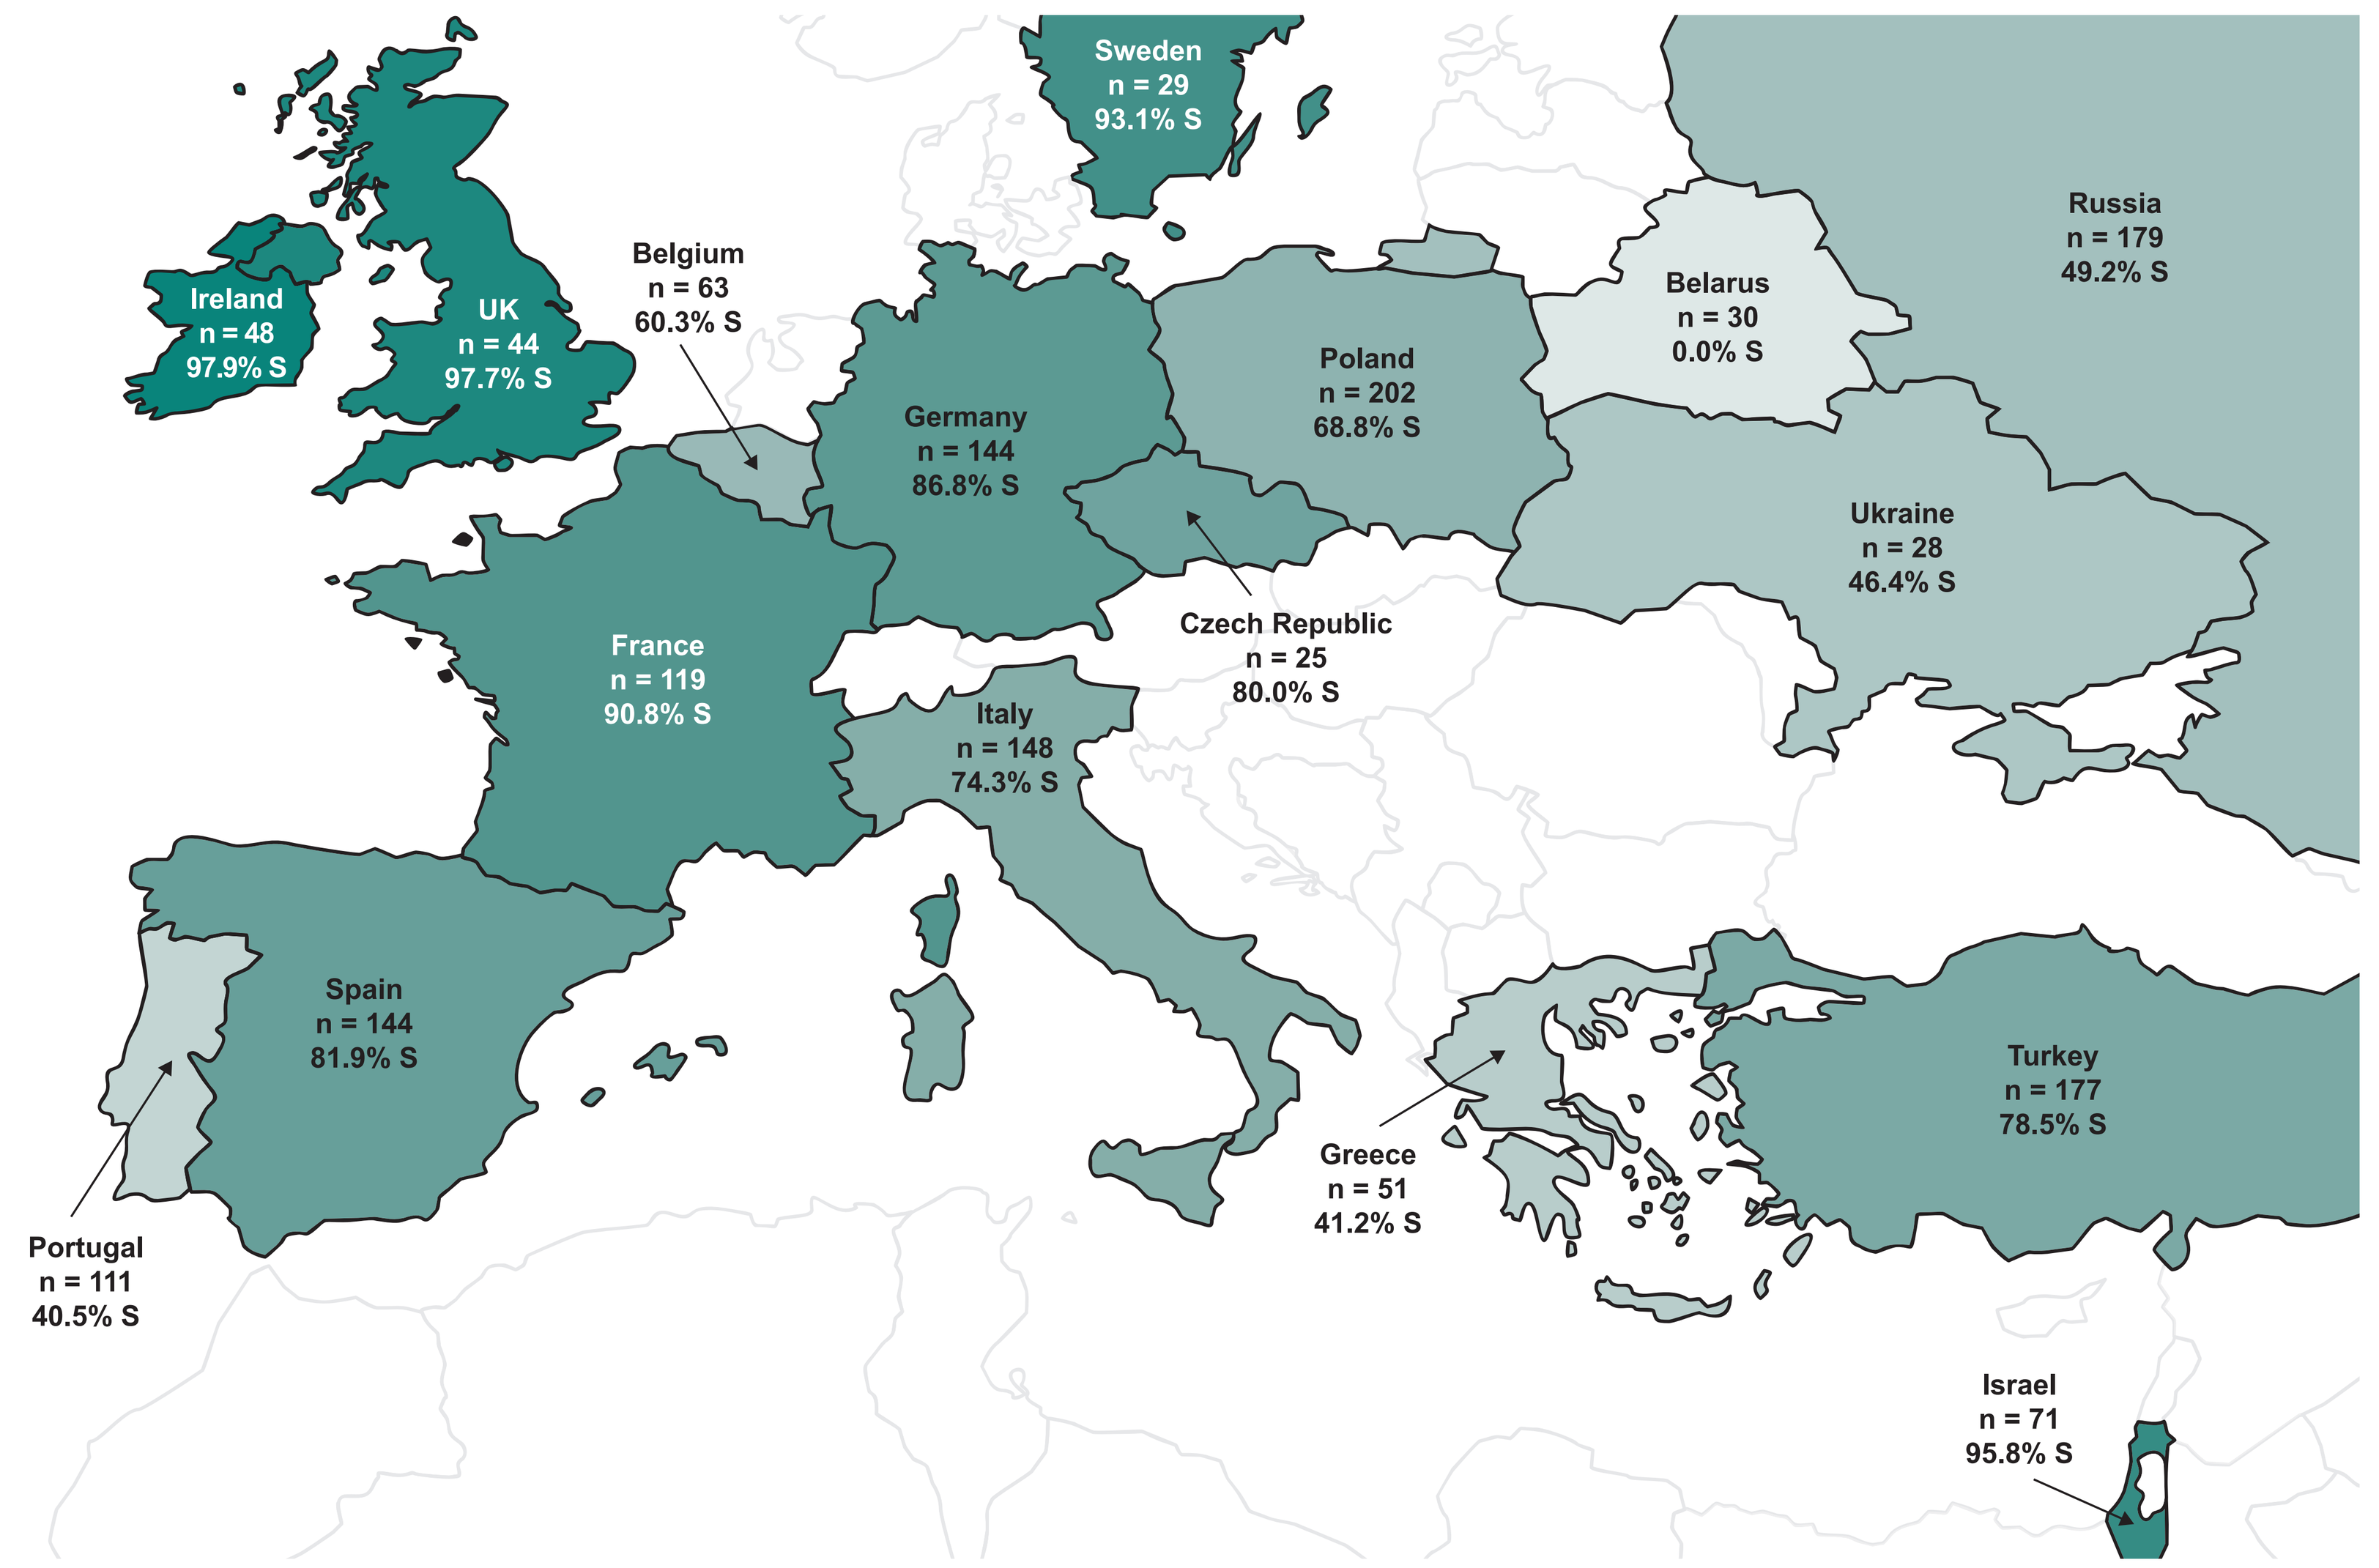
Sweden
n = 29
93.1% S
Ireland
n = 48
97.9% S
UK
n = 44
97.7% S
Belgium
n = 63
60.3% S
Belarus
n = 30
0.0% S
Russia
n = 179
49.2% S
Poland
n = 202
68.8% S
Germany
n = 144
86.8% S
Ukraine
n = 28
46.4% S
France
n = 119
90.8% S
Czech Republic
n = 25
80.0% S
Italy
n = 148
74.3% S
Spain
n = 144
81.9% S
Turkey
n = 177
78.5% S
Greece
n = 51
41.2% S
Portugal
n = 111
40.5% S
Israel
n = 71
95.8% S

CLSI = Clinical and Laboratory Standards Institute; EUCAST = European Committee on Antimicrobial Susceptibility Testing; S = susceptibility.
Green coloration denotes susceptibility rates: the darker the color, the greater the susceptibility of *P. aeruginosa* to ceftolozane/tazobactam in that country.
†Based on 2016 CLSI/EUCAST.

CONCLUSIONS

- *P. aeruginosa* is a leading cause of nosocomial infections. High mortality rates and increasing levels of carbapenem resistance have been observed worldwide
- Ceftolozane/tazobactam retained activity against 77.9% of the carbapenem-nonsusceptible *P. aeruginosa* overall (US and Europe) and 90% of the isolates from the United States
- Ceftolozane/tazobactam was active against >70% of the carbapenem-nonsusceptible *P. aeruginosa* from Europe (including Turkey and Israel), where metallo-β-lactamase–producing isolates are more commonly described. However, there was a broad range of susceptibility within Europe (including Turkey and Israel)
- Ceftolozane/tazobactam may represent a valuable treatment option for Gram-negative infections, including those caused by carbapenem-nonsusceptible *P. aeruginosa*

REFERENCES

Clinical and Laboratory Standards Institute. M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard tenth edition. Wayne, PA: CLSI; 2015.

Clinical and Laboratory Standards Institute. M100-S26. Performance standards for antimicrobial susceptibility testing: 26th informational supplement. Wayne, PA: CLSI; 2016.

Craig WA, Andes DR. In vivo activities of ceftolozane, a new cephalosporin, with and without tazobactam against *Pseudomonas aeruginosa* and Enterobacteriaceae, including strains with extended-spectrum beta-lactamases, in the thighs of neutropenic mice. *Antimicrob Agents Chemother*. 2013;57:1577-1582.

European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, 2016. http://www.eucast.org/clinical_breakpoints/. Accessed September 13, 2016.

Farrell DJ, Flamm RK, Sader HS, Jones RN. Antimicrobial activity of ceftolozane-tazobactam tested against Enterobacteriaceae and *Pseudomonas aeruginosa* with various resistance patterns isolated in U.S. hospitals (2011-2012). *Antimicrob Agents Chemother*. 2013;57:6305-6310.

Sader HS, Farrell DJ, Castanheira M, Flamm RK, Jones RN. Antimicrobial activity of ceftolozane/tazobactam tested against *Pseudomonas aeruginosa* and Enterobacteriaceae with various resistance patterns isolated in European hospitals (2011-12). *J Antimicrob Chemother*. 2014;69:2713-2722.

Skalweit MJ. Profile of ceftolozane/tazobactam and its potential in the treatment of complicated intra-abdominal infections. *Drug Des Devel Ther*. 2015;9:2919-2925.

ACKNOWLEDGMENTS

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ, USA. Editorial assistance was provided by Sally Mitchell, PhD (ApotheCom, Yardley, PA, USA). This assistance was funded by Merck & Co., Inc., Kenilworth, NJ, USA.

http://tinyurl.com/h8o7eyk

Copyright © 2016, Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc. All rights reserved.