Poster #P1883

Activity of Tedizolid and Comparators against *Enterococcus* spp. that Include Multidrug-Resistant Clinical Isolates from European and US Medical Centres (2016–2018)

Cecilia G. Carvalhaes[1], Helio S. Sader[1], Jennifer M. Streit[1], Robert K. Flamm[1], Rodrigo E. Mendes[1]

[1]JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Tedizolid is an oxazolidinone-class antimicrobial with potent *in vitro* activity against gram-positive organisms, including vancomycin-resistant enterococci (VRE)
- Tedizolid is approved by the European Medicines Agency and the United States Food and Drug Administration for the treatment of skin and skin structure infections
- Tedizolid is currently under investigation for serious infections, such as pneumonia and bone and joint infections
- *Enterococcus* spp. pathogens are the second most common cause of gram-positive hospital-associated infections with significant mortality rates
- VRE isolates, mainly *Enterococcus faecium*, commonly exhibit a multidrug-resistance phenotype, causing infections with limited treatment options
- This study assessed the *in vitro* activity of tedizolid and comparators against a contemporary collection of enterococci, including VRE and isolates exhibiting high-level aminoglycoside resistance (HLAR)

Materials and Methods

Organism collection

- A total of 3,953 enterococci were collected from US (1,829 isolates) and European (and adjacent) medical centres (2,124 isolates) during 2016–2018
- The isolates were recovered from 73 medical centres, including 33 sites from the United States and 40 sites from Europe and surrounding countries; Figure 1 lists total isolates per participating country
- The most frequent pathogens were *E. faecalis* (*n* = 2,525; 63.9%), followed by *E. faecium* (*n* = 1,300; 32.9%), and other *Enterococcus* spp. (*n* = 128; 3.2%), including *E. avium* (*n* = 39; 1,0%), 37 *E. gallinarum* (*n* = 37; 0.9%), *E. casseliflavus* (*n* = 26; 0.7%), *E. raffinosus* (*n* = 10; 0.3%), *E. hirae* (*n* = 9; 0.2%) and *E. durans* (*n* = 7; 0.2%)
 - *Enterococcus* spp. distribution in Europe and the United States are shown in Figure 1
- Enterococcal isolates determined to be significant by local criteria as the probable cause of bloodstream infection (BSI); pneumonia in hospitalized patients (PIHP); intra-abdominal infection (IAI); skin and skin structure infection (SSTI); and urinary tract infection (UTI) are shown in Figure 2
- Only 1 isolate per patient infection episode was included
- Bacterial identification was confirmed by matrix-assisted laser desorption ionization–time of flight mass spectrometry (Bruker Daltonics, Massachusetts, USA) following the manufacturer's instructions

Antimicrobial susceptibility testing

- Susceptibility (S) testing was performed by broth microdilution according to European Committee of Antimicrobial Susceptibility Testing (EUCAST) methods using 96-well panels manufactured by JMI Laboratories (North Liberty, Iowa, USA)
- Quality assurance was performed by concurrently testing EUCAST quality control (QC) reference strains (*Staphylococcus aureus* ATCC 29213 and *E. faecalis* ATCC 29212)
 - All QC results were within published acceptable ranges
- Susceptibility determinations and HLAR screenings were based on EUCAST breakpoint criteria (2019)

Results

- Overall, tedizolid ($MIC_{50/90}$, 0.25/0.25 mg/L) inhibited all but 6 enterococci at MIC of ≤0.5 mg/L (Table 1)
- Isolates with VRE and/or HLAR phenotypes showed similar tedizolid MIC_{50} (0.25 mg/L) and MIC_{90} results (0.25-0.5 mg/L), regardless of enterococcal species (Table 1)
- Although VRE isolates were more frequently observed in the US hospitals than in European hospitals (66.8% vs.18.3% of *E. faecium* and 3.3% vs. 0.7% of *E. faecalis*, respectively), tedizolid MIC_{50} and MIC_{90} results remained at 0.25 mg/L and 0.25/0.5 mg/L, respectively, against enterococci from US and European hospitals (Figure 1)
- *E. faecalis* showed high S rates (≥98.0%S) to linezolid, vancomycin, and ampicillin, and these agents, except vancomycin, remained active against the combined VRE and HLAR subset (1.6% of all *E. faecalis;* Table 1)
- Tedizolid ($MIC_{50/90}$, 0.25/0.25 mg/L) exhibited MIC results 4- to 8-fold lower than those of linezolid ($MIC_{50/90}$, 1/2 mg/L; 99.7%S) when tested against *E. faecium* (Table 1)
- A total of 8.7% of *E. faecium* isolates were highly resistant (VRE and HLAR), and similar tedizolid MIC results were obtained against the VRE, HLAR, and VRE/HLAR subsets ($MIC_{50/90}$, 0.25/0.25 mg/L; Table 1)
- Five linezolid non-S isolates (MIC, >4 mg/L) were observed, and 4 of those isolates were inhibited by tedizolid at ≤1 mg/L
- Two VRE isolates that showed elevated daptomycin MIC results (MIC >4 mg/L) were inhibited by tedizolid at ≤0.5 mg/L

Table 1 Activity of tedizolid and comparators against *Enterococcus* spp. isolates by resistant phenotype

	$MIC_{50/90}$ (%S, EUCAST[a])				
Organism (no. tested) Phenotype	Tedizolid	Linezolid	Daptomycin	Ampicillin	Vancomycin
All (3,953)	0.25/0.25 (-)	1/2 (99.9)	1/2 (-)	1/>16 (71.1)	1/>16 (85.4)
E. faecalis (2,525)	0.25/0.25 (-)	1/2 (>99.9)	0.5/1 (-)	1/1 (100)	1/2 (98.1)
HLAR (754)	0.25/0.25 (-)	1/2 (99.9)	0.5/1 (-)	1/2 (100)	1/1 (94.7)
VRE (48)	0.25/0.25 (-)	1/2 (100)	0.5/1 (-)	1/2 (100)	-
HLAR and VRE (40)	0.25/0.25 (-)	1/2 (100)	0.5/1 (-)	1/2 (100)	-
E. faecium (1,300)	0.25/0.25 (-)	1/2 (99.7)	1/2 (-)	>16/>16 (12.9)	1/>16 (60.6)
HLAR (345)	0.25/0.25 (-)	1/2 (99.4)	1/2 (-)	>16/>16 (3.2)	1/>16 (67.2)
VRE (512)	0.25/0.25 (-)	1/2 (99.8)	1/2 (-)	>16/>16 (1.0)	-
HLAR and VRE (113)	0.25/0.25 (-)	1/2 (100)	1/2 (-)	>16/>16 (0.0)	-
Other *Enterococcus* spp. (128)	0.25/0.5 (-)	1/2 (100)	0.5/1 (-)	≤0.5/2 (92.2)	1/8 (86.7)
Vancomycin non-S (17)	0.25/0.5 (-)	1/2 (100)	1/4 (-)	1/2 (100)	-

HLAR, high-level aminoglycoside resistance; VRE, vancomycin-resistant *Enterococcus;* S, susceptible; -, VRE selected isolates.

[a] Susceptibility rate based on EUCAST v.9.0 (2019) criteria.

Figure 2 Distribution of enterococci isolates by infection type

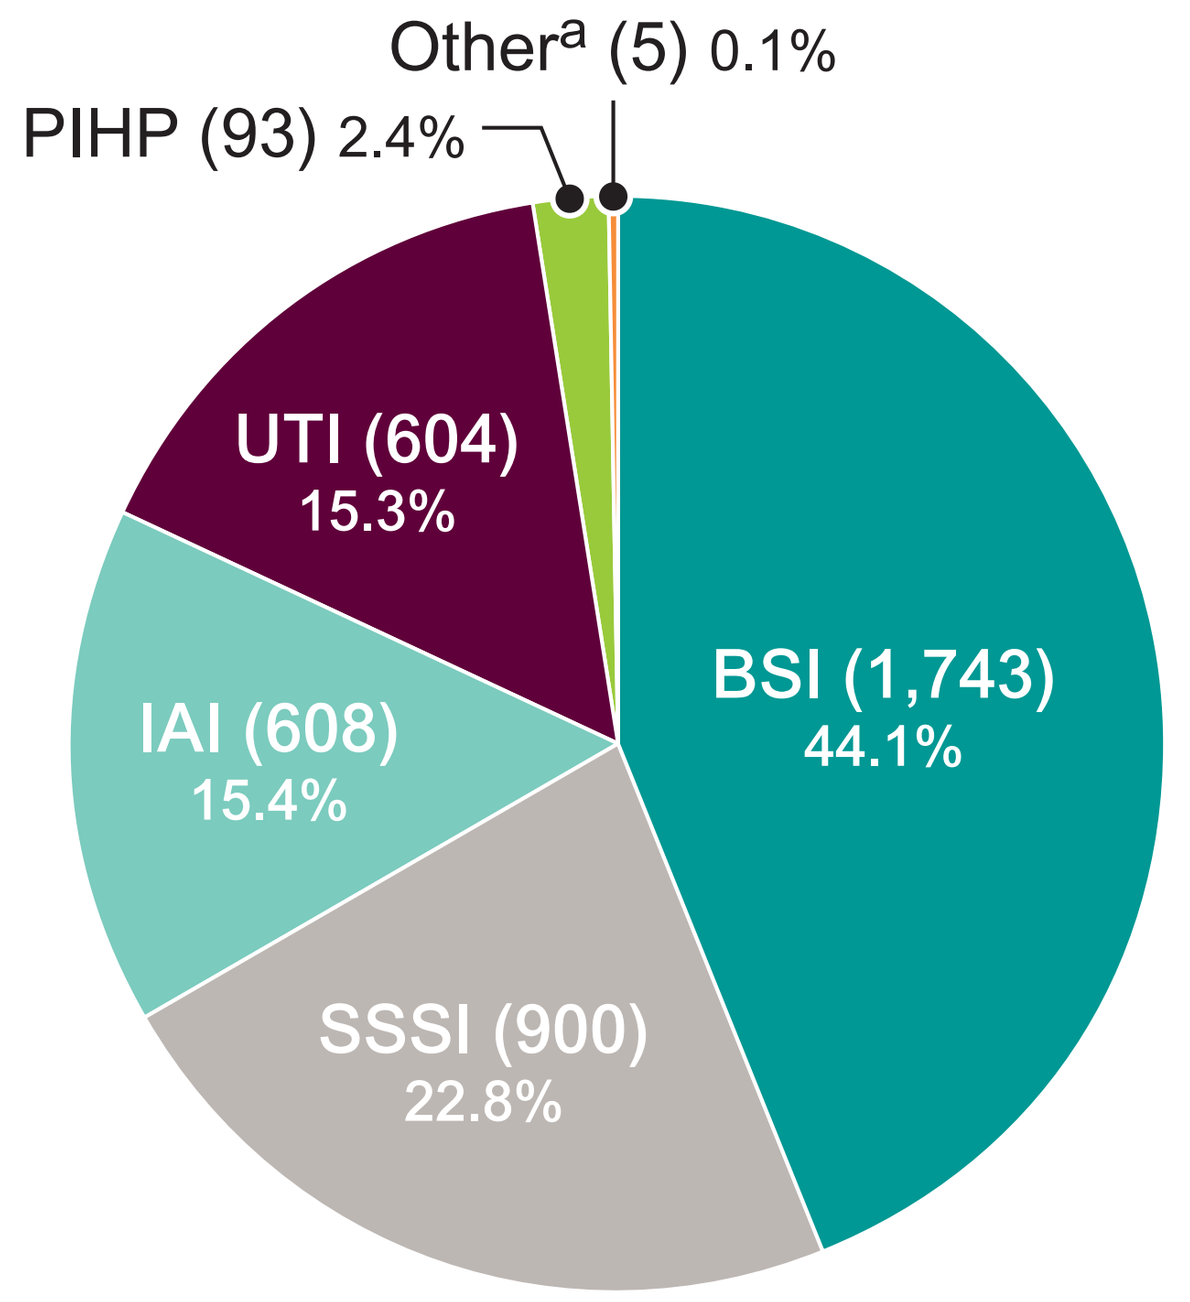

Other[a] (5) 0.1%
PIHP (93) 2.4%
UTI (604)
15.3%
IAI (608)
15.4%
SSSI (900)
22.8%
BSI (1,743)
44.1%

[a] Other sites include genital tract (2) and unspecified body site (3).

BSI, bloodstream infection; SSSI, skin and skin structure infection; IAI, intra-abdominal infection; UTI, urinary tract infection; PIHP, pneumonia in hospitalized patient.

Figure 1 Tedizolid activity and VRE and HLAR rates among enterococci recovered from the United States and Europe

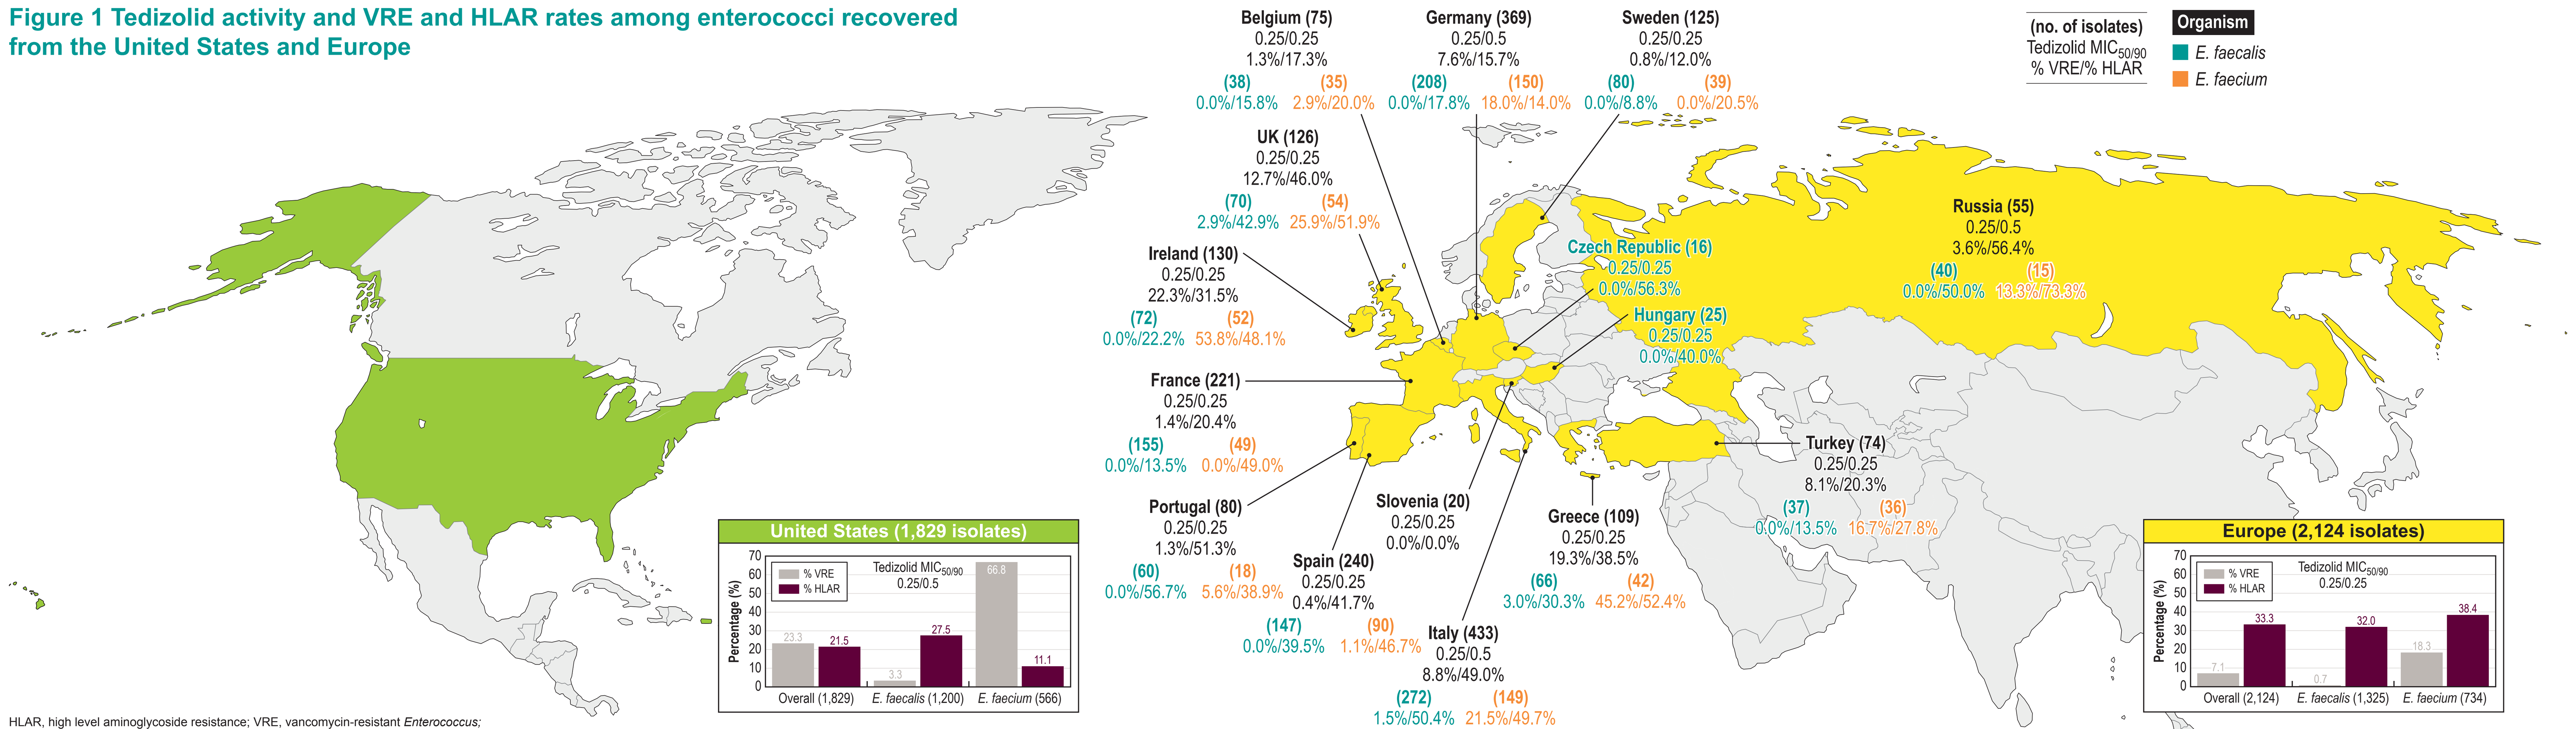

(no. of isolates)
Tedizolid MIC50/90
% VRE/% HLAR
Organism
E. faecalis
E. faecium
Belgium (75)
0.25/0.25
1.3%/17.3%
(38) 0.0%/15.8%
(35) 2.9%/20.0%
Germany (369)
0.25/0.5
7.6%/15.7%
(208) 0.0%/17.8%
(150) 18.0%/14.0%
Sweden (125)
0.25/0.25
0.8%/12.0%
(80) 0.0%/8.8%
(39) 0.0%/20.5%
UK (126)
0.25/0.25
12.7%/46.0%
(70) 2.9%/42.9%
(54) 25.9%/51.9%
Ireland (130)
0.25/0.25
22.3%/31.5%
(72) 0.0%/22.2%
(52) 53.8%/48.1%
France (221)
0.25/0.25
1.4%/20.4%
(155) 0.0%/13.5%
(49) 0.0%/49.0%
Portugal (80)
0.25/0.25
1.3%/51.3%
(60) 0.0%/56.7%
(18) 5.6%/38.9%
Spain (240)
0.25/0.25
0.4%/41.7%
(147) 0.0%/39.5%
(90) 1.1%/46.7%
Slovenia (20)
0.25/0.25
0.0%/0.0%
Italy (433)
0.25/0.5
8.8%/49.0%
(272) 1.5%/50.4%
(149) 21.5%/49.7%
Greece (109)
0.25/0.25
19.3%/38.5%
(66) 3.0%/30.3%
(42) 45.2%/52.4%
Czech Republic (16)
0.25/0.25
0.0%/56.3%
Hungary (25)
0.25/0.25
0.0%/40.0%
Russia (55)
0.25/0.5
3.6%/56.4%
(40) 0.0%/50.0%
(15) 13.3%/73.3%
Turkey (74)
0.25/0.25
8.1%/20.3%
(37) 0.0%/13.5%
(36) 16.7%/27.8%
United States (1,829 isolates)
% VRE
% HLAR
Tedizolid MIC50/90
0.25/0.5
Percentage (%)
23.3
21.5
3.3
27.5
66.8
11.1
Overall (1,829)
E. faecalis (1,200)
E. faecium (566)
Europe (2,124 isolates)
% VRE
% HLAR
Tedizolid MIC50/90
0.25/0.25
Percentage (%)
7.1
33.3
0.7
32.0
18.3
38.4
Overall (2,124)
E. faecalis (1,325)
E. faecium (734)

HLAR, high level aminoglycoside resistance; VRE, vancomycin-resistant *Enterococcus;*

(-) no isolate was detected.

Note: Data for Belarus, Israel, Poland, and Romania are not shown/graphed due to a limited number of isolates (*n*<10) recovered.

Figure 3. MIC distribution of oxazolidinones against (A.) *E. faecalis* (*n* = 2,525) and (B.) *E. faecium* (*n* = 1,300) isolates

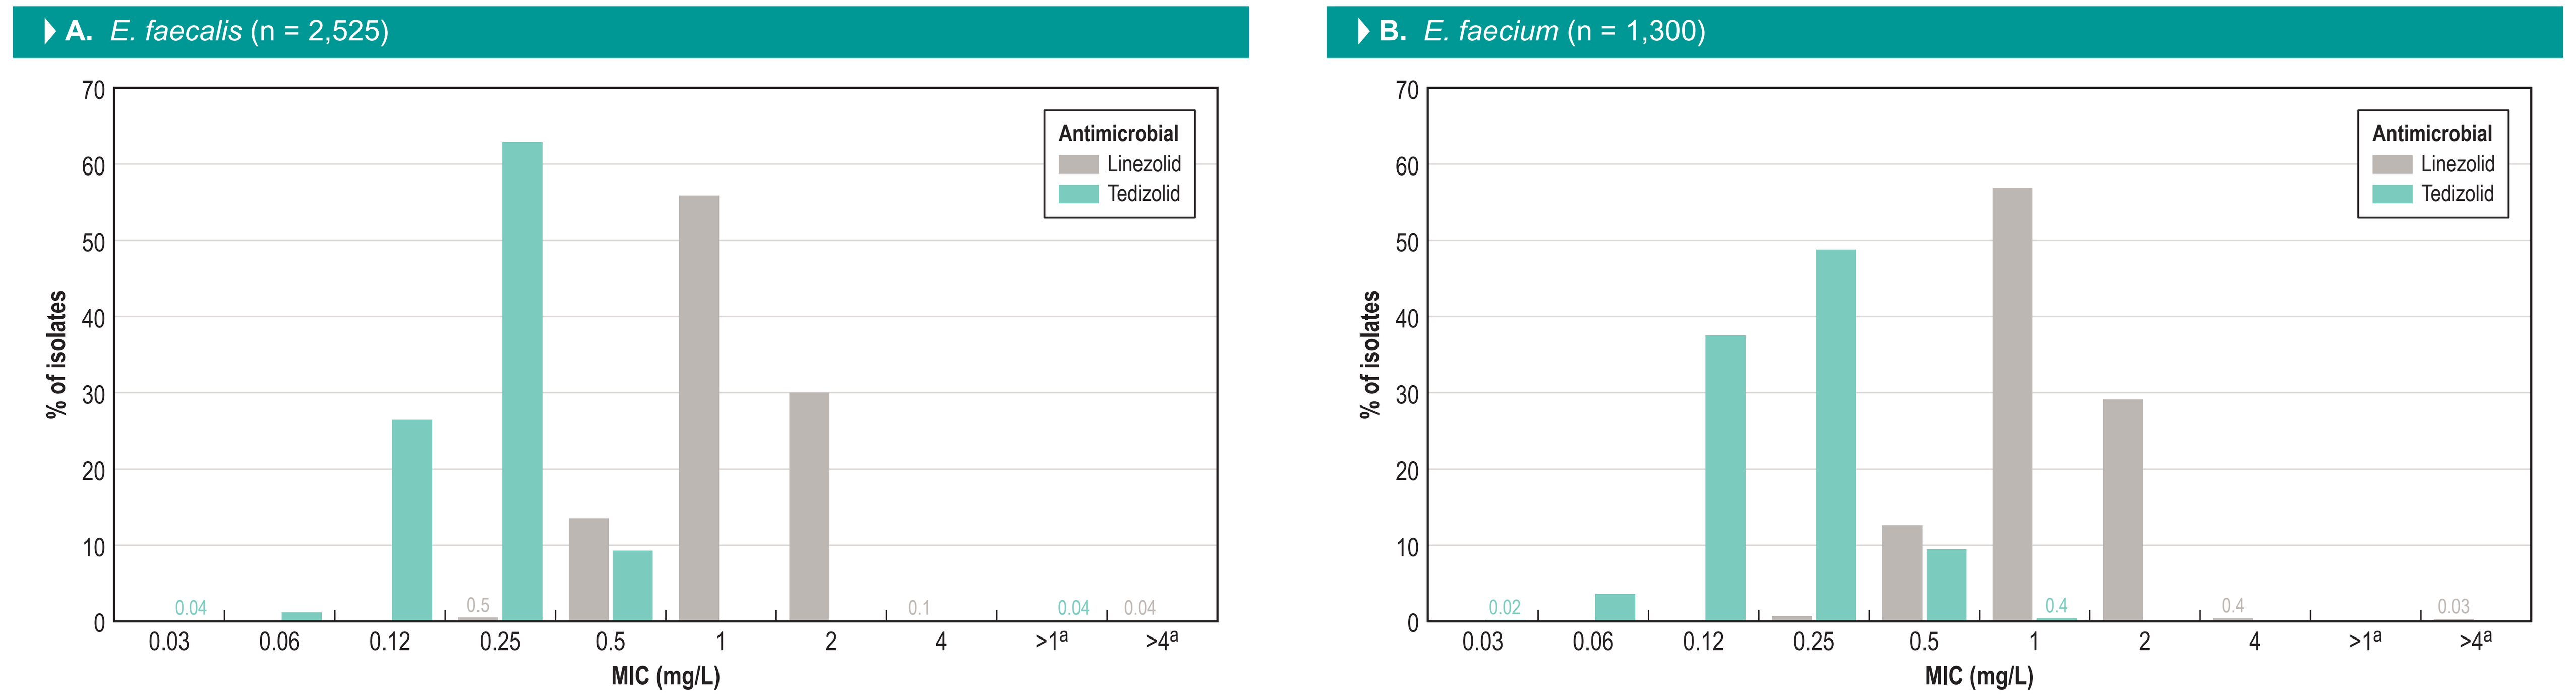

A. E. faecalis (n = 2,525)
B. E. faecium (n = 1,300)
Antimicrobial
Linezolid
Tedizolid
% of isolates
MIC (mg/L)
0.03
0.06
0.12
0.25
0.5
1
2
4
>1a
>4a
0.04
0.5
0.1
0.04
0.04
0.02
0.4
0.4
0.03

[a] Greater than the highest concentration tested for linezolid (>4 mg/L) and tedizolid (>1 mg/L)

Conclusions

- Tedizolid showed good activity ($MIC_{50/90}$, 0.25/0.25 mg/L) against this 2016-2018 US and European collection of *Enterococcus* spp. isolates responsible for BSI, PIHP, IAI, SSSI, and UTI
- Overall, tedizolid remained potent against this challenge set of enterococcal clinical isolates, regardless of resistance phenotype (HLAR or VRE)
- Tedizolid was 4- to 8-fold more active than linezolid against both *E. faecalis* and *E. faecium* isolates, regardless of resistance phenotype
- Tedizolid may be considered for treating serious enterococcal infections, granted clinical approval

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ USA

The database used for this poster was amended to include the entire 2018 collection.

References

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.

EUCAST (2019). Breakpoint tables for interpretation of MIC's and zone diameters. Version 9.0, January 2019. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_9.0_Breakpoint_Tables.pdf. Accessed January 2019.

Locke JB, Zurenko GE, Shaw KJ, Bartizal K. Tedizolid for the management of human infections: in vitro characteristics. Clin Infect Dis. 2014;58(Suppl 1):S35–42

Sinvextro (tedizolid phosphate) package insert. Revised August 2017. Document available online at: https://www.merck.com/product/usa/pi_circulars/s/sivextro/sivextro_pi.pdf

European public assessment report (EPAR) for Sivextro. Available at https://www.ema.europa.eu/en/medicines/human/EPAR/sivextro

Contact Information

Cecilia G. Carvalhaes, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia-carvalhaes@jmilabs.com

https://bit.ly/2Gq6S7p

Presented at the ECCMID 2019, Amsterdam, the Netherlands, April 13–16, 2019

Copyright © 2019 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.