Poster Number: 333

Tedizolid Activity against Gram-Positive Bacterial Isolates Causing Bone and Joint Infections in the United States (2015–2019)

Cecilia G. Carvalhaes, Helio S. Sader, Jennifer M. Streit, Mariana Castanheira, Rodrigo E. Mendes
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Prolonged systemic antibiotic courses are frequently used to manage difficult-to-treat bone and joint infections (BJI).
- New strategies are needed to overcome the treatment challenges posed by BJI infections caused by methicillin-resistant *Staphylococcus aureus* (MRSA) and other resistant pathogens.
- Tedizolid is an oxazolidinone-class antimicrobial that inhibits protein synthesis and exhibits activity against staphylococci, streptococci, and enterococci, including MRSA and vancomycin-resistant *Enterococcus* spp. (VRE).
- Tedizolid was approved by the European Medicines Agency, the United States Food and Drug Administration, and other regulatory agencies for the treatment of acute bacterial skin and skin structure infections (ABSSSI), and has been considered as a therapy candidate for BJI in adults and children.
- This study assessed the *in vitro* activity of tedizolid and comparator agents against a contemporary collection of Gram-positive isolates causing BJI in the US.

Materials and Methods

- A total of 493 Gram-positive isolates were collected from patients with BJI between 2015 and 2019 as part of the Surveillance of Tedizolid Activity and Resistance (STAR) Program.
- The isolates were recovered from 30 medical centers in the US, including all 9 Census regions.
- Pathogens mostly included *Staphylococcus aureus* (n=310; 62.9%), β-hemolytic streptococci (BHS; n=79; 16.0%), coagulase-negative staphylococci (CoNS; n=52; 10.5%), and *Enterococcus faecalis* (n=37; 7.5%). Pathogen distribution is shown in Figure 1.
- Only Gram-positive isolates (1 per patient per infection episode) determined to be clinically significant by local criteria as the probable cause of infection were included.
- Bacterial identification was performed by the participating centers and confirmed by the monitoring laboratory (JMI Laboratories, North Liberty, IA) using matrix-assisted laser desorption ionization–time of flight mass spectrometry (MALDI-TOF; Bruker Daltonics, Billerica, MA) following the manufacturer's instructions.
- Susceptibility testing was performed by broth microdilution according to Clinical and Laboratory Standards Institute M07 (2018) guidelines.
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories and contained cation-adjusted Mueller-Hinton broth, with 2.5–5% lysed horse blood added for streptococci.
- Quality assurance was performed by concurrently testing CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213, *E. faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619).
- Breakpoint criteria for MIC interpretations were those criteria from the CLSI M100 (2020) and EUCAST (2020) documents.

Results

- Tedizolid (MIC$_{50/90}$, 0.12/0.25 mg/L) inhibited all *S. aureus* at the CLSI breakpoint (≤0.5 mg/L; 100% susceptible; Table 1; Figure 2).
 - Tedizolid, linezolid, vancomycin, and daptomycin had 100% susceptibility rates against *S. aureus* isolates (Table 1).
 - Ceftaroline and clindamycin inhibited 98.7% and 89.4% of *S. aureus* at the current CLSI breakpoint, respectively.
- MRSA was observed in 35.8% of *S. aureus* causing BJI. Tedizolid activity against MRSA isolates (MIC$_{50/90}$, 0.12/0.25 mg/L) was equivalent to tedizolid activity against methicillin-susceptible *S. aureus* isolates (MIC$_{50/90}$, 0.12/0.25 mg/L; Table 2).
 - MRSA isolates remain susceptible to tedizolid, linezolid, daptomycin, and vancomycin (100% susceptible).
 - Ceftaroline and trimethoprim/sulfamethoxazole inhibited 96.4% and 94.6% of MRSA isolates at the susceptible breakpoints, respectively.
 - Clindamycin (76.6%) and erythromycin (18.9%) showed limited activity against MRSA.
- All CoNS isolates were inhibited by tedizolid at ≤0.5 mg/L (susceptible breakpoint for *S. aureus*).
 - High methicillin-resistance rate was observed among CoNS (71.2%).
 - Tedizolid, linezolid, vancomycin, and daptomycin displayed 100% susceptibility against this CoNS set (Table 1).
- Tedizolid was active against all BHS (100% susceptible; Table 1).
 - Tedizolid tested against *S. pyogenes* (n=24) and *S. agalactiae* (n=44) displayed MIC$_{50}$ and MIC$_{90}$ values of 0.12 and 0.25 mg/L, respectively.
 - Against *S. dysgalactiae* (n=11), tedizolid had MIC$_{50/90}$ of 0.25/0.25 mg/L.
 - Penicillin, linezolid, vancomycin, and daptomycin also were active against BHS (100% susceptible).
- Tedizolid (MIC$_{50/90}$, 0.25/0.25 mg/L; 100% susceptible) was 4- to 8-fold more potent than linezolid (MIC$_{50/90}$, 1/1 mg/L) and vancomycin (MIC$_{50/90}$, 1/2 mg/L) against *E. faecalis* (Table 1).
- Gram-positive isolates resistant to oxazolidinone were not observed in this collection.

Figure 1 Distribution of Gram-positive isolates causing BJI in US medical centers (2015–2019)

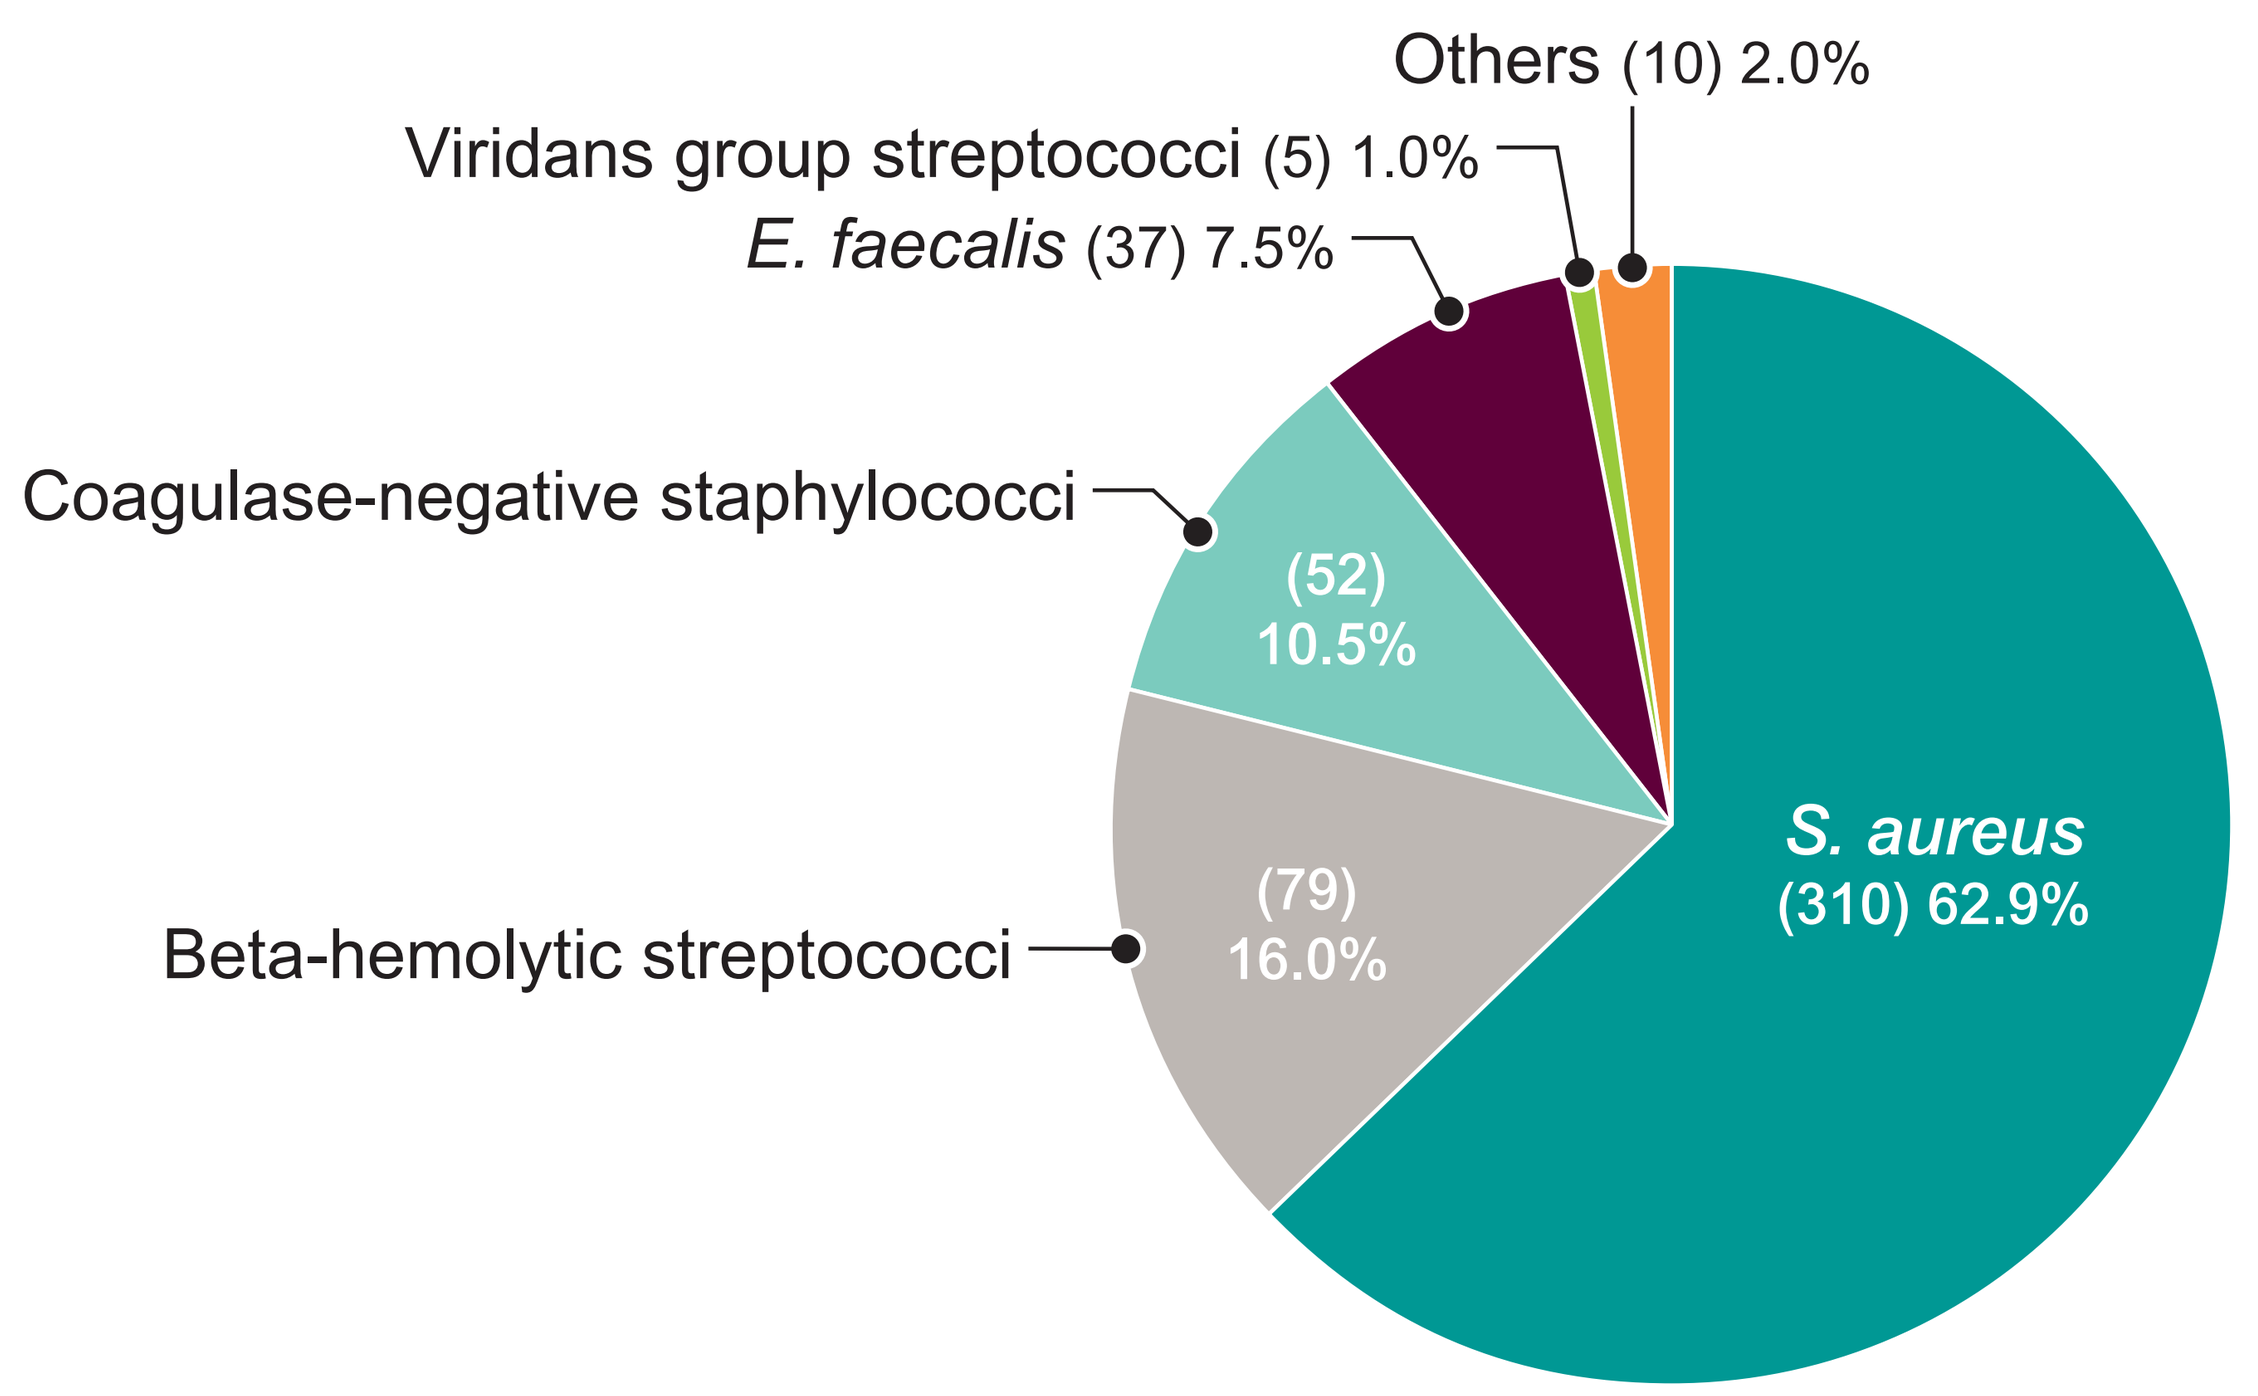
Others (10) 2.0%
Viridans group streptococci (5) 1.0%
E. faecalis (37) 7.5%
Coagulase-negative staphylococci
(52) 10.5%
Beta-hemolytic streptococci
(79) 16.0%
S. aureus (310) 62.9%

Table 1 Activity of tedizolid and comparators against Gram-positive cocci causing BJI in US medical centers (2015–2019)

Organism[a] (no. tested) Phenotype	MIC$_{50}$ / MIC$_{90}$ (% susceptible)[b]											
	Tedizolid		Linezolid		Ceftaroline		Daptomycin		Clindamycin		Vancomycin	
S. aureus (310)	0.12 / 0.25	(100)	1 / 2	(100)	0.25 / 1	(98.7)	0.25 / 0.5	(100)	≤0.25 / >2	(89.4)	0.5 / 1	(100)
CoNS (52)	0.12 / 0.12	(100)[c]	0.5 / 1	(100)	0.25 / 0.5	(94.2)[e]	0.25 / 0.5	(100)	≤0.25 / >2	(69.2)	1 / 2	(100)
BHS (79)	0.12 / 0.25	(100)[d]	1 / 1	(100)	≤0.008 / 0.015	(100)	0.12 / 0.25	(100)	≤0.25 / >2	(65.8)	0.5 / 0.5	(100)
S. pyogenes (24)	0.12 / 0.25	(100)	1 / 2	(100)	≤0.008 / 0.015	(100)	≤0.06 / 0.12	(100)	≤0.25 / ≤0.25	(91.7)	0.25 / 0.5	(100)
S. agalactiae (44)	0.12 / 0.25	(100)	1 / 1	(100)	0.015 / 0.015	(100)	0.25 / 0.5	(100)	>2 / >2	(43.2)	0.5 / 0.5	(100)
S. dysgalactiae (11)	0.25 / 0.25	(100)[d]	1 / 2	(100)	≤0.008 / ≤0.008	(100)	≤0.06 / ≤0.06	(100)	≤0.25 / ≤0.25	(100)	0.25 / 0.25	(100)
E. faecalis (37)	0.25 / 0.25	(100)	1 / 1	(100)	2 / 8	—	1 / 1	(100)	—		1 / 2	(94.6)

[a] BHS, β-hemolytic streptococci; CoNS, coagulase-negative staphylococci.
[b] CLSI (2020), according to breakpoint availability.
[c] Tedizolid breakpoint for *S. aureus* used for CoNS for comparison purposes.
[d] Tedizolid breakpoint for *S. pyogenes* and *S. agalactiae* used for *S. dysgalactiae*.
[e] Ceftaroline breakpoint for *S. aureus* used for CoNS for comparison purposes.

Figure 2 MIC distribution of oxazolidinones against the main BJI pathogens, *S. aureus* and beta-hemolytic streptococci

A. *S. aureus* (n = 310)

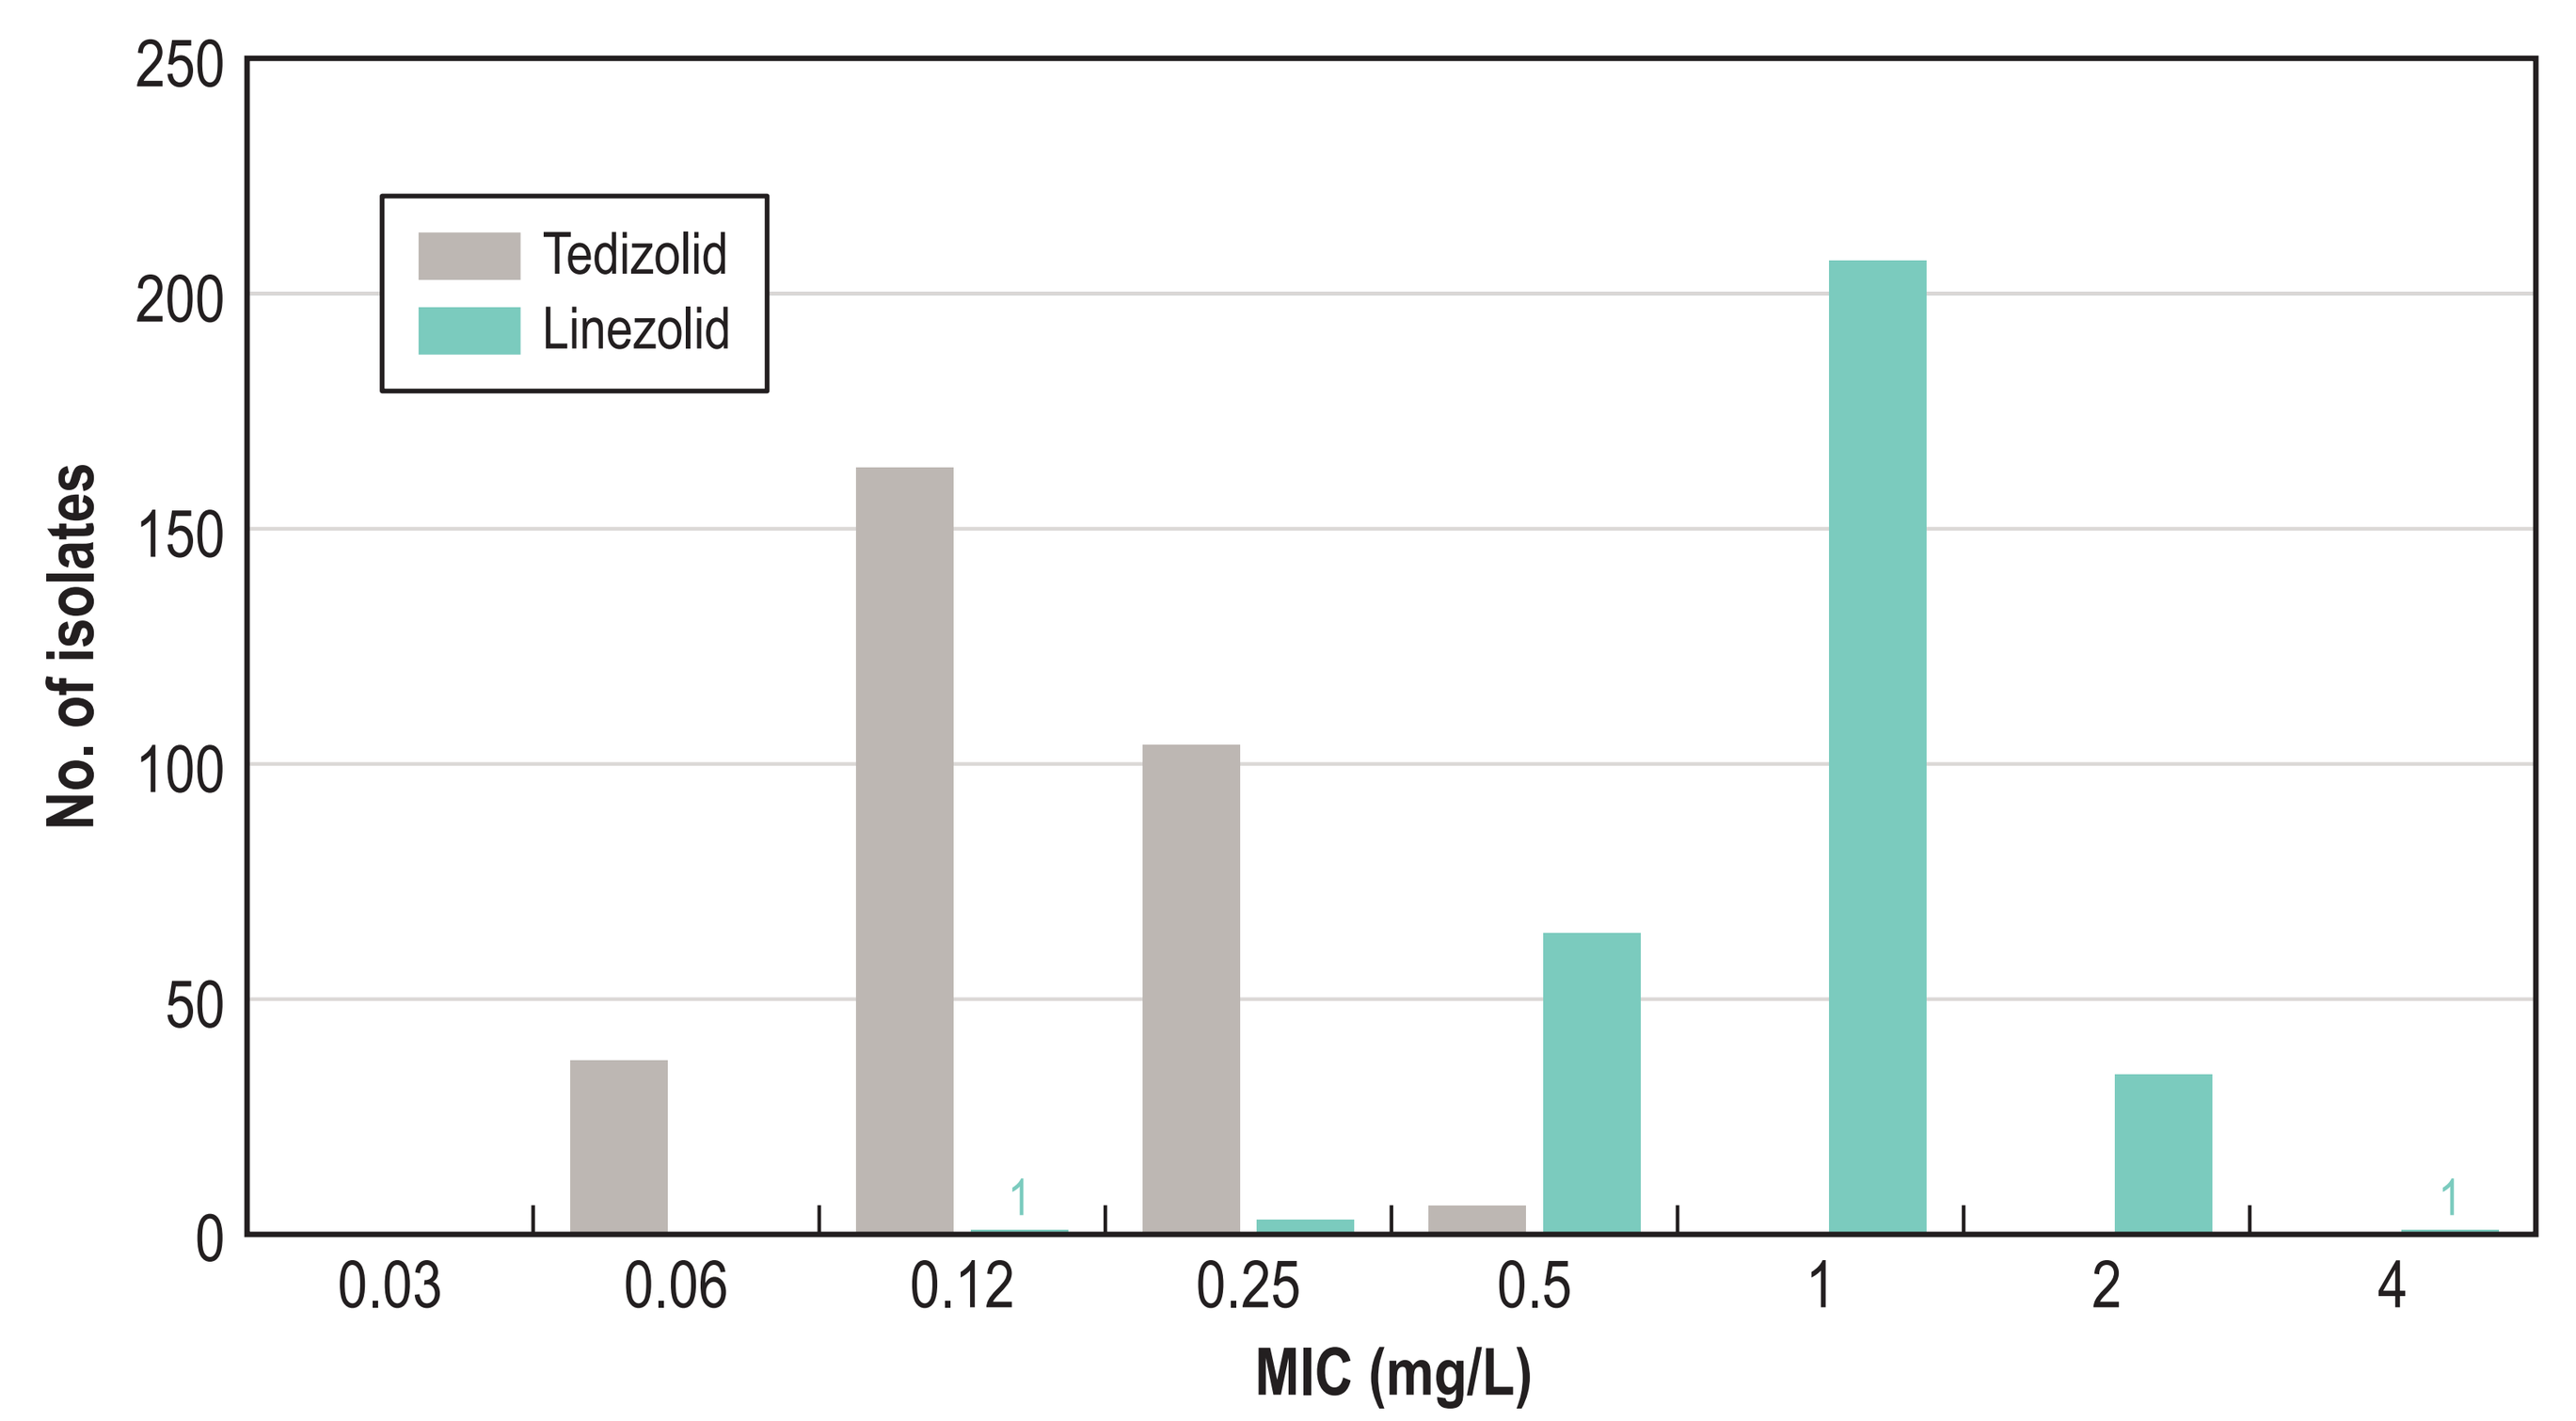
250
200
150
100
50
0
No. of isolates
Tedizolid
Linezolid
1
1
0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (mg/L)

B. Beta-hemolytic streptococci (n = 79)

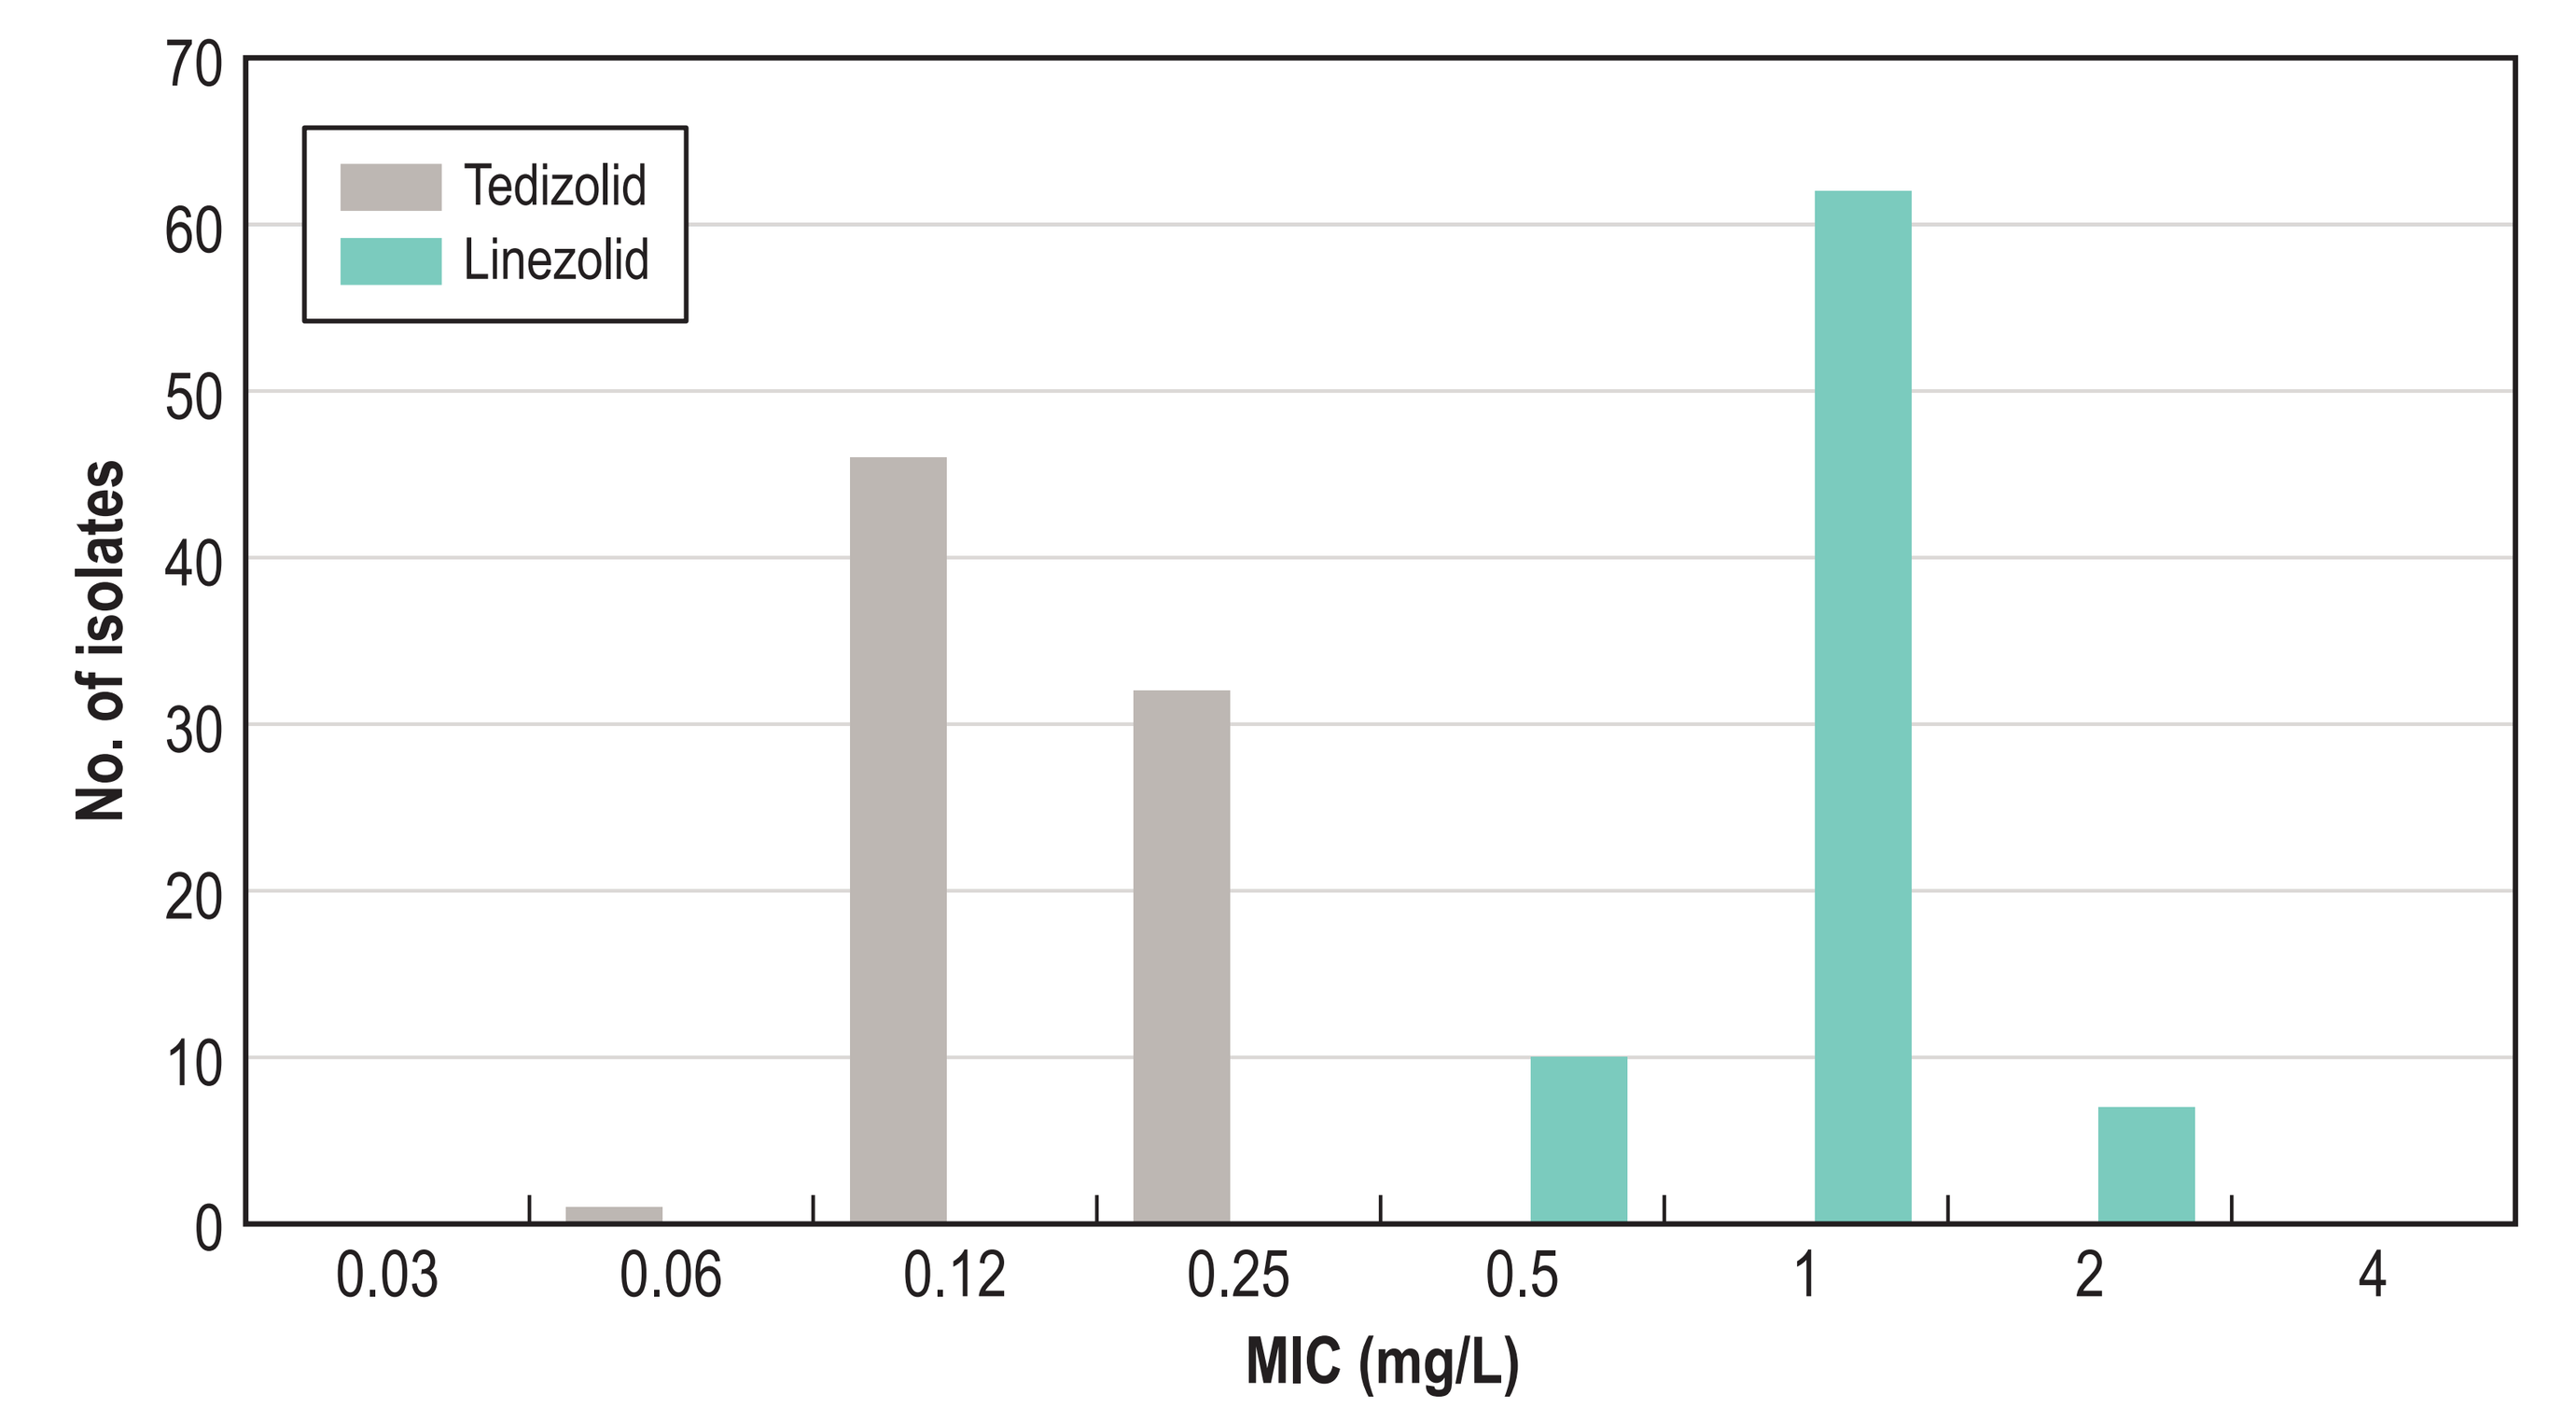
70
60
50
40
30
20
10
0
No. of isolates
Tedizolid
Linezolid
0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (mg/L)

Organisms included: *Streptococcus agalactiae* (44), *S. dysgalactiae* (11), *S. pyogenes* (24)

Conclusions

- *S. aureus* and BHS were responsible for >75% of Gram-positive isolates causing BJI in patients across US hospitals during a 5-year period.
- Tedizolid demonstrated potent *in vitro* activity against this collection of contemporary Gram-positive BJI isolates.
- Tedizolid and comparator agents showed high susceptibility rates against the most frequent organisms and organism groups, including MRSA.
- These findings support the clinical development of tedizolid as an additional option for treating BJI caused by Gram-positive pathogens.

Table 2 Activity of tedizolid and comparators against *S. aureus* isolates causing BJI in US medical centers (2015–2019)

Antimicrobial agent (n)	mg/L		CLSI[a]		EUCAST[a]	
	MIC$_{50}$	MIC$_{90}$	%S	%R	%S	%R
MSSA (199)						
Tedizolid	0.12	0.25	100.0	0.0	100.0	0.0
Linezolid	1	2	100.0	0.0	100.0	0.0
Ceftaroline	0.25	0.25	100.0	0.0	100.0[b]	0.0
Clindamycin	≤0.25	≤0.25	96.5	3.5	95.5	3.5
Daptomycin	0.25	0.5	100.0		100.0	0.0
Erythromycin	0.25	>8	68.8	21.6	69.3	26.6
TMP-SMT	≤0.5	≤0.5	100.0	0.0	100.0	0.0
Vancomycin	0.5	1	100.0	0.0	100.0	0.0
MRSA (111)						
Tedizolid	0.12	0.25	100.0	0.0	100.0	0.0
Linezolid	1	1	100.0	0.0	100.0	0.0
Ceftaroline	0.5	1	96.4	0.0	96.4[b]	0.0
Clindamycin	≤0.25	>2	76.6	23.4	76.6	23.4
Daptomycin	0.25	0.5	100.0		100.0	0.0
Erythromycin	>8	>8	18.9	81.1	18.9	81.1
TMP-SMT	≤0.5	≤0.5	94.6	5.4	94.6	5.4
Vancomycin	1	1	100.0	0.0	100.0	0.0

[a] Criteria as published by CLSI 2020 and EUCAST 2020.
[b] Using other than pneumonia breakpoints.
TMP-SMT, trimethoprim-sulfamethoxazole

Acknowledgements

Funding for this research was provided by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA. Merck was involved in the design and decision to present these results, and JMI Laboratories received compensation for preparing the poster. Merck did not contribute to decisions in the collection, analysis, or interpretation of the data.

References

Carvalhaes CG, Sader HS, Flamm RK, Mendes RE. Tedizolid *in vitro* activity against Gram-positive clinical isolates causing bone and joint infections in hospitals in the USA and Europe (2014–17). J Antimicrob Chemother. 2019;74(7):1928–1933.

Clinical and Laboratory Standards Institute (2020). M100Ed30. Performance standards for antimicrobial susceptibility testing: 30th informational supplement. Wayne, PA: CLSI.

EUCAST (2020). Breakpoint tables for interpretation of MICs and zone diameters. Version 10.0, July 2020. Available at https://eucast.org/clinical_breakpoints/. Accessed July 2020.

Locke JB, Zurenko GE, Shaw KJ, Bartizal K. Tedizolid for the management of human infections: *in vitro* characteristics. Clin Infect Dis. 2014;58(Suppl 1):S35–42.

Sivextro (tedizolid phosphate) package insert. Revised August 2017. Document available online at https://www.merck.com/product/usa/pi_circulars/s/sivextro/sivextro_pi.pdf.

Contact Information:
Cecilia Carvalhaes, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia-carvalhaes@jmilabs.com

https://bit.ly/2E5TCoS

Presented at IDWeek 2020

Copyright © 2020 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.