ASM Microbe 2019
Poster Friday - AAR-542

Update on the *In Vitro* Activity of Dalbavancin against Indicated Species (*Staphylococcus aureus, Enterococcus faecalis*, β-Hemolytic Streptococci, and *Streptococcus anginosus* Group) Collected from United States Hospitals in 2017–2018

Helio S. Sader, Jennifer M. Streit, S.J. Ryan Arends, Rodrigo E. Mendes, Robert K. Flamm

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit www.allergancongressposters.com/363765

Charges may apply.
No personal information is stored.

INTRODUCTION

- Dalbavancin belongs to the lipoglycopeptide class of antimicrobial agents that act by interrupting bacterial cell wall synthesis, resulting in bacterial death
- Dalbavancin allows for very convenient parenteral administration, which can be a single dose of 1500 mg or a dose of 1000 mg followed by 500 mg a week later for treating acute bacterial skin and skin structure infections (ABSSSIs)
- Dalbavancin was approved in the United States (2014) and Europe (2015) to treat adults with ABSSSI caused by
 - Methicillin-resistant (MRSA) and -susceptible *Staphylococcus aureus* (MSSA)
 - *Streptococcus pyogenes*
 - *Streptococcus agalactiae*
 - *Streptococcus dysgalactiae*
 - *Streptococcus anginosus* group
 - Vancomycin-susceptible *Enterococcus faecalis*
- This study updates dalbavancin activity against contemporary isolates of these species/groups

MATERIALS AND METHODS

Bacterial isolates

- A total of 12,138 unique isolates of the indicated species were consecutively collected from 70 United States (US) medical centers in 2017–2018
- Isolates were determined to be clinically significant based on local guidelines and were submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA)
- Participating laboratories initially identified isolates and JMI confirmed bacterial identifications by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany)

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility (S) by broth microdilution following guidelines in the CLSI M07 document (2018)
- The dalbavancin breakpoints approved by the US Food and Drug Administration (FDA) and CLSI for indicated species were applied (i.e., ≤0.25 mg/L), and breakpoint criteria for comparator agents were those from CLSI (2019)
- Quality assurance was performed by concurrently testing CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213, *E. faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619)

RESULTS

- Dalbavancin $MIC_{50/90}$ values were 0.03/0.06 mg/L against *S. aureus* (100.0%S), including MRSA isolates (Table 1 and Figure 1)
- Dalbavancin was 16- to 32-fold more active than vancomycin ($MIC_{50/90}$, 1/1 mg/L) and 4- to 8-fold more active than daptomycin ($MIC_{50/90}$, 0.25/0.25 mg/L) against MRSA (Table 1)
- Susceptibility rates for ceftaroline and trimethoprim-sulfamethoxazole (TMP-SMX) were 94.0% and 96.9%, respectively, among MRSA (Table 1)
- All (100.0%) vancomycin-susceptible *E. faecalis* isolates were dalbavancin-susceptible, with the highest dalbavancin MIC value of 0.25 mg/L (1,007/1,008 [99.9%] inhibited at ≤0.12 mg/L; Table 1 and Figure 1)
- Dalbavancin MIC values for vancomycin-susceptible *E. faecalis* isolates were 32-fold and 16-fold lower than those of vancomycin ($MIC_{50/90}$, 1/2 mg/L) and daptomycin ($MIC_{50/90}$, 0.5/1 mg/L), respectively (Table 1)
- Dalbavancin was highly active against β-hemolytic streptococci ($MIC_{50/90}$, 0.008/0.015 mg/L), with MIC_{90} values 16- to 32-fold lower than daptomycin (MIC_{90}, 0.25 mg/L) and vancomycin (MIC_{90}, 0.5 mg/L), respectively (Table 1)
- β-hemolytic streptococci susceptibility rates for tetracycline, clindamycin, and levofloxacin were 55.6%, 82.4%, and 99.2%, respectively (Table 1)
- All *S. anginosus* group isolates were inhibited at ≤0.004 mg/L of dalbavancin (Table 1 and Figure 1)
- *S. anginosus* group susceptibility rates for penicillin, clindamycin, and tetracycline were 100.0%, 83.6%, and 60.0%, respectively (Table 1)

Figure 1. Dalbavancin MIC distributions when tested against clinical isolates of indicated species collected from US medical centers (2017–2018)

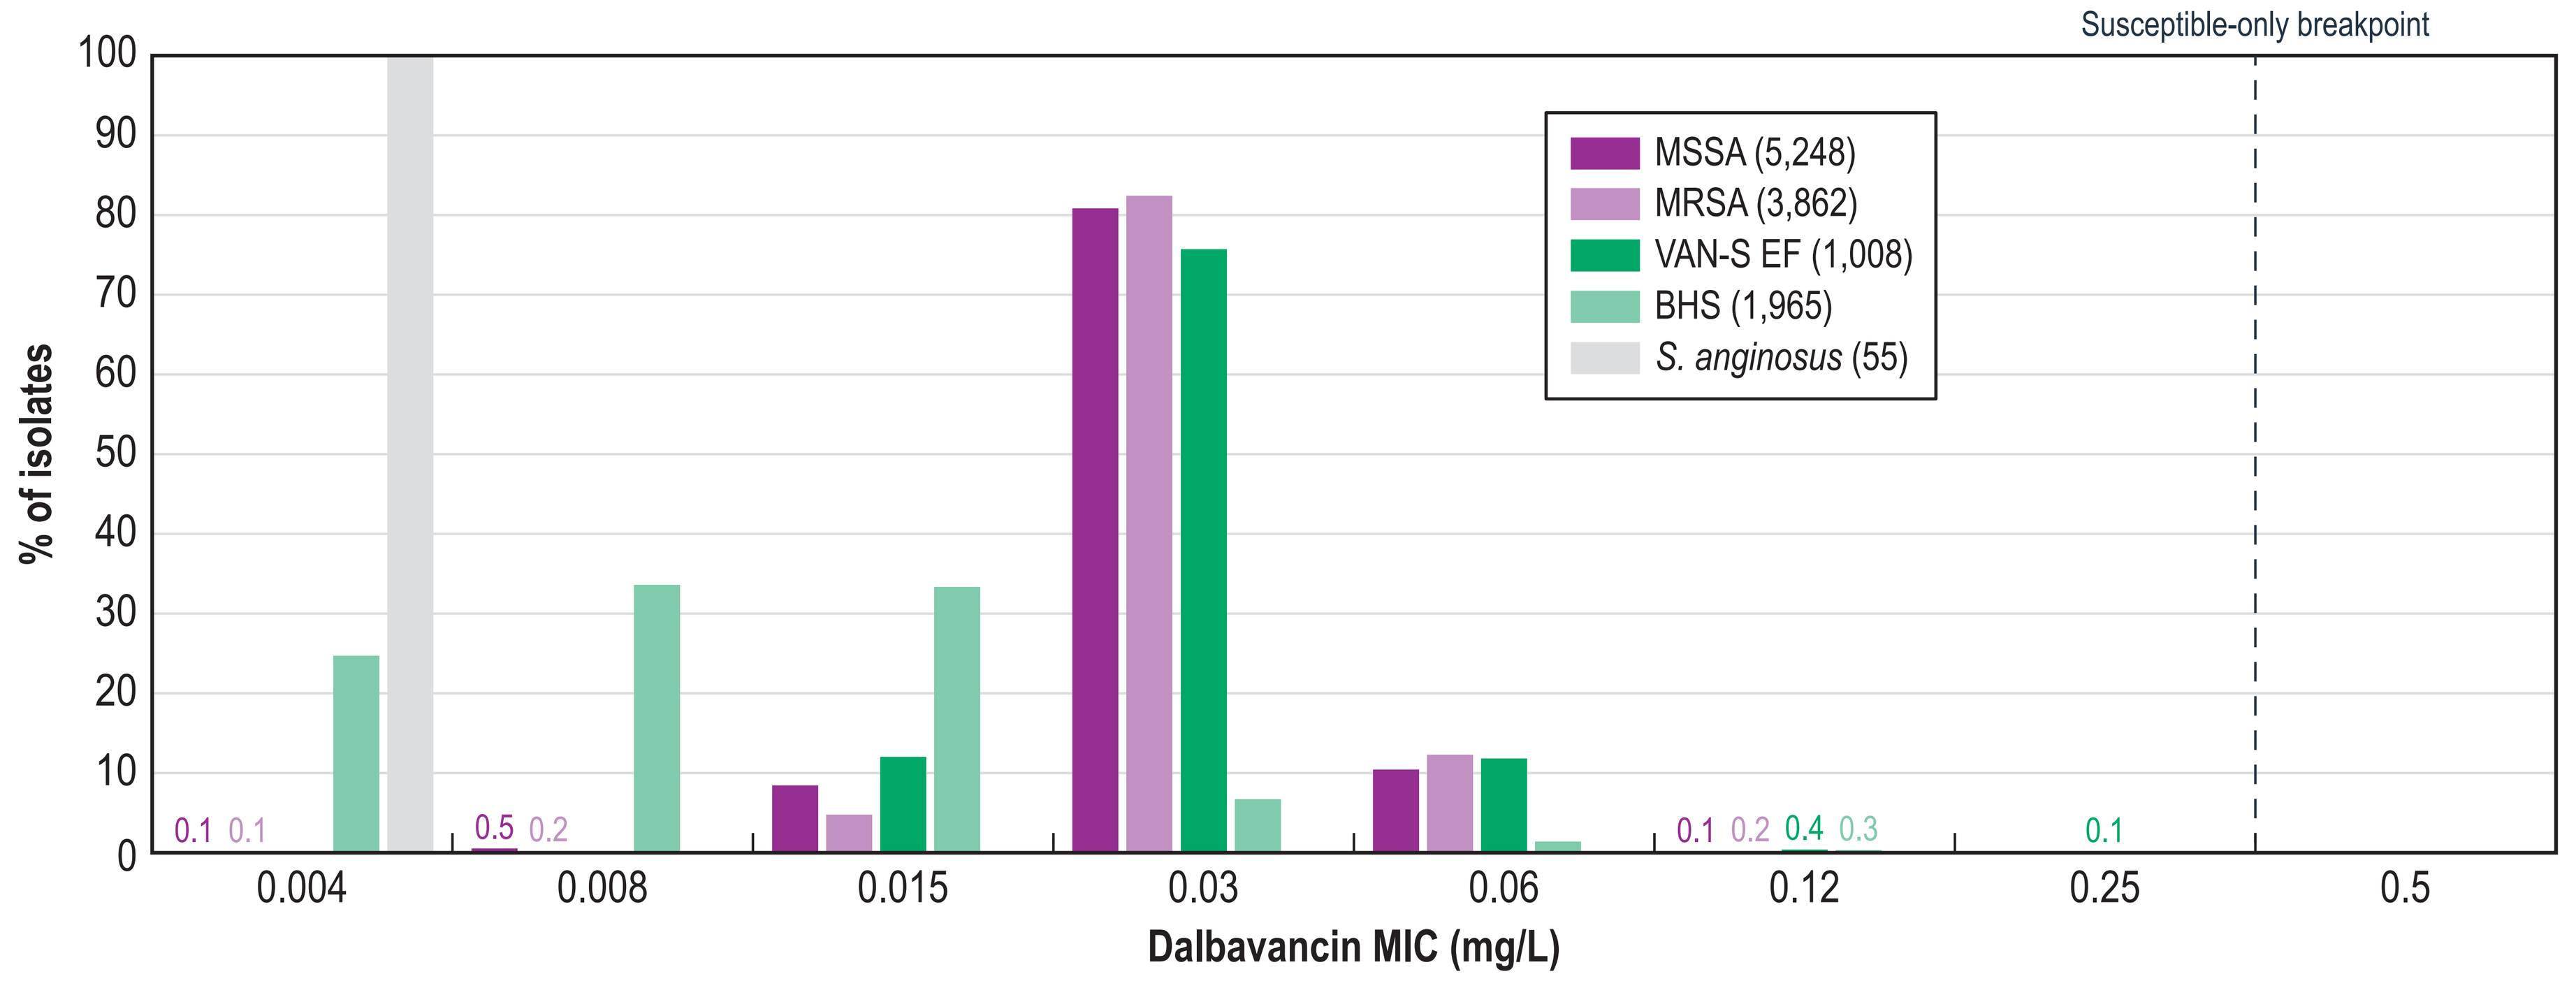

Susceptible-only breakpoint
% of isolates
100
90
80
70
60
50
40
30
20
10
0
MSSA (5,248)
MRSA (3,862)
VAN-S EF (1,008)
BHS (1,965)
S. anginosus (55)
0.1 0.1
0.5 0.2
0.1 0.2 0.4 0.3
0.1
0.004
0.008
0.015
0.03
0.06
0.12
0.25
0.5
Dalbavancin MIC (mg/L)

MSSA, methicillin-susceptible *Staphylococcus aureus*; MRSA, methicillin-resistant *S. aureus*; VAN-S EF, vancomycin-susceptible *Enterococcus faecalis*; BHS, β-hemolytic streptococci.

Table 1 Activity of dalbavancin and comparator antimicrobial agents against clinical isolates of indicated species collected from US medical centers (2017–2018)

Antimicrobial agent / organism (no. tested)	MIC_{50}	MIC_{90}	CLSI[a]	
			%S	%R
Methicillin-susceptible *S. aureus* (5,248)				
Dalbavancin	0.03	0.06	100.0	
Telavancin	0.06	0.06	100.0	
Teicoplanin	0.5	0.5	100.0	0.0
Vancomycin	1	1	100.0	0.0
Daptomycin	0.25	0.25	100.0	
Linezolid	1	2	100.0	0.0
Ceftaroline	0.25	0.25	100.0	0.0
Oxacillin	0.5	1	100.0	0.0
Clindamycin	0.06	0.12	95.6	4.1
Erythromycin	0.25	>8	65.7	28.1
Levofloxacin	0.25	4	89.6	10.3
Tetracycline	≤0.5	≤0.5	95.4	3.4
TMP-SMX	≤0.5	≤0.5	99.4	0.6
Methicillin-resistant *S. aureus* (3,862)				
Dalbavancin	0.03	0.06	100.0	
Telavancin	0.06	0.06	100.0	
Teicoplanin	0.5	0.5	100.0	0.0
Vancomycin	1	1	100.0	0.0
Daptomycin	0.25	0.25	>99.9	
Linezolid	1	2	>99.9	<0.1
Ceftaroline	1	1	94.0	<0.1
Oxacillin	>2	>2	0.0	100.0
Clindamycin	0.06	>2	73.8	25.8
Erythromycin	>8	>8	13.8	83.3
Levofloxacin	4	>4	33.7	65.9
Tetracycline	≤0.5	1	92.5	6.5
TMP-SMX	≤0.5	≤0.5	96.9	3.1
Vancomycin-susceptible *E. faecalis* (1,008)				
Dalbavancin	0.03	0.06	100.0	
Telavancin	0.12	0.25	99.9	
Teicoplanin	0.5	0.5	100.0	0.0
Vancomycin	1	2	100.0	0.0
Daptomycin	0.5	1	97.1	0.0
Linezolid	1	2	99.9	0.0
Ampicillin	1	1	100.0	0.0
Levofloxacin	1	>4	80.4	19.3
β-hemolytic streptococci (1,965)				
Dalbavancin	0.008	0.015	100.0	
Vancomycin	0.5	0.5	100.0	
Daptomycin	≤0.06	0.25	100.0	
Linezolid	1	2	100.0	
Ceftaroline	≤0.008	0.015	100.0	
Ceftriaxone	0.03	0.06	100.0	
Penicillin	0.015	0.06	100.0	
Clindamycin	≤0.25	>2	82.4	16.6
Erythromycin	0.06	>16	65.9	33.1
Levofloxacin	0.5	1	99.2	0.7
Tetracycline	0.5	>4	55.6	43.8
TMP-SMX	≤0.12	0.25		
S. anginosus group (55)				
Dalbavancin	≤0.004	≤0.004	100.0	
Vancomycin	0.5	1	100.0	
Daptomycin	0.25	0.25	100.0	
Linezolid	1	2	100.0	
Ceftaroline	0.03	0.03		
Ceftriaxone	0.25	0.25	100.0	0.0
Penicillin	0.03	0.06	100.0	0.0
Clindamycin	≤0.25	>2	83.6	16.4
Erythromycin	≤0.015	>16	74.5	25.5
Levofloxacin	0.5	1	100.0	0.0
Tetracycline	1	>4	60.0	38.2
TMP-SMX	≤0.12	≤0.12		

TMP-SMX, trimethoprim-sulfamethoxazole
[a] Criteria as published by CLSI (2019).

CONCLUSIONS

- Dalbavancin retained activity against 100.0% of isolates from indicated species collected in US hospitals in 2017–2018 (n=12,138)
- Dalbavancin MIC_{50} values ranged from ≤0.004 mg/L to 0.03 mg/L and MIC_{90} values ranged from ≤0.004 mg/L to 0.06 mg/L for these isolates, with 99.99% of isolates inhibited at ≤0.12 mg/L of dalbavancin
- In summary, dalbavancin remains highly active against clinical isolates of indicated species causing infections in US medical centers

ACKNOWLEDGMENTS

The authors would like to thank all participants of the International Dalbavancin Evaluation of Activity (IDEA) for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.
3. Dalvance® (2018). Available at https://www.allergan.com/assets/pdf/dalvance_pi.
4. Pfaller MA, Mendes RE, Duncan LR, et al. (2018). Activity of dalbavancin and comparator agents against Gram-positive cocci from clinical infections in the USA and Europe 2015–16. *J Antimicrob Chemother in press*.

Allergan

ASM Microbe, June 20–24, 2019, San Francisco, California