IDWEEK 2020 | Poster #1590

Activity of meropenem-vaborbactam and single-agent comparators against *Enterobacterales* isolates, including KPC-producing isolates, from European patients hospitalized with pneumonia including ventilator-associated pneumonia (2014–2019)

Dee Shortridge, Lalitagauri Deshpande, Leonard Duncan, Jennifer Streit, Mariana Castanheira

JMI Laboratories, North Liberty, IA, USA

Introduction

- Carbapenem-resistant *Enterobacterales* (CRE) isolates are a growing global concern.
- Among carbapenemases detected in *Enterobacterales* species, *Klebsiella pneumoniae* carbapenemases (KPCs) have disseminated worldwide and are endemic in many hospitals in various countries.
- *K. pneumoniae* containing KPC are often multidrug resistant and have limited treatment options.
- Colistin and tigecycline are often used as treatments for infections caused by KPC-producing isolates.
 - Both compounds have limitations that concern clinicians, such as toxicity or low plasma concentrations. In addition, tigecycline is not indicated by EMA to treat pneumonia.
 - Resistance to colistin, and less often to tigecycline, have been reported among KPC-producing *K. pneumoniae* (Sader, Castanheira et al. 2015).
- Vaborbactam is a cyclic boronic acid β-lactamase inhibitor that has activity against Ambler class A, including KPC, and C enzymes.
 - Vaborbactam combined with meropenem enhanced the activity of this carbapenem against KPC-producing isolates in comparison to meropenem tested alone.
 - Vaborbactam does not inhibit class B metallo-β-lactamases (MBL) or class D carbapenemases (Lomovskaya, Sun et al. 2017).
- Meropenem-vaborbactam (MVB) recently was approved in Europe for the treatment of complicated urinary tract infections (cUTIs), including acute pyelonephritis, complicated intra-abdominal infections (cIAI), hospital-acquired bacterial pneumonia (HABP), ventilator-associated bacterial pneumonia (VABP), and bacteremia that occurs with any of the infections listed above.
- We evaluated the activity of MVB and single-agent comparators against 6,846 *Enterobacterales* isolates from patients hospitalized with pneumonia (PHP) in European hospitals from 2014–2019.

Materials and Methods

- 6,846 *Enterobacterales* PHP clinical isolates collected in 40 European hospitals across 20 countries were susceptibility tested using reference broth microdilution methods (CLSI 2018).
 - EUCAST and CLSI interpretive criteria were used (CLSI 2020, EUCAST 2020)
- *Enterobacterales* isolates also were screened for the extended spectrum beta-lactamase (ESBL) phenotype (CLSI 2020). Isolates that were ESBL screen-positive isolates included both carbapenem-susceptible and carbapenem-nonsusceptible isolates; a subset of carbapenem-susceptible ESBL- phenotype isolates (ESBL non-CR) were also analyzed.
- Carbapenem-resistant isolates were subjected to whole genome sequencing and analyzed for the presence of carbapenemases (Castanheira, Huband et al. 2017).
- The susceptibilities of isolates from intensive care unit (ICU) and non-ICU patients were compared.
 - 3,218 isolates were from patients in the ICU and 2,627 were non-ICU. ICU vs. non-ICU designations were made by the submitting hospital.
 - A subset of 1,890 isolates from ICU patients with VABP were included.
 - The VABP designation was made by the submitting site. Only isolates from patients with VABP in ICU were included in the analysis.
 - A total of 1,001 isolates did not have an ICU or non-ICU designation from the submitting site. These 1,001 isolates were not included in the ICU/non-ICU analysis but were included in the total.

Results

- The top 5 species isolated from PHP caused by *Enterobacterales* are shown in Figure 1.
 - The most common *Enterobacterales* species isolated from patients hospitalized with pneumonia (PHP) were *Klebsiella pneumoniae* (n=1,877) and *Escherichia coli* (n=1,646).
- The susceptibilities of MVB and comparators tested against *Enterobacterales* for ICU and non-ICU patients, ICU patients with VABP and resistance groups are shown in Table 1.
- Overall, MVB had the highest percent susceptibility of the drugs tested, with 97.7/98.0% using CLSI or EUCAST breakpoints (CLSI, ≤4/8/≥16; EUCAST, ≤8/-/≥16 mg/L). MVB MIC distributions for all isolates and subgroups from patients in the ICU, ICU with VABP, and non-ICU are shown in Figure 2.
 - Amikacin was the most active comparator against all *Enterobacterales* (96.2/94.2% susceptible CLSI/EUCAST).
- For *Enterobacterales* isolates from ICU patients, MVB susceptibility was 96.6/97.1%. For non-ICU isolates, MVB susceptibility was 98.2/98.5% (CLSI/EUCAST) as shown in Table 1.
 - The susceptibilities of comparators for ICU vs non-ICU isolates were similar, except for levofloxacin (76.0/76.0%S ICU and 70.3/70.3%S non-ICU, CLSI/EUCAST).
 - Susceptibilities were similar between ICU PHP and VABP subsets.
- Overall, 20% of isolates had an ESBL-phenotype, 76.4% of these were carbapenem-susceptible.
 - Against isolates with an ESBL-phenotype, MVB inhibited 85.8/87.5% (CLSI/EUCAST). All ESBL-phenotype isolates that were non-CRE were susceptible to meropenem-vaborbactam.
- 5.3% of all isolates were CRE.
 - Meropenem-vaborbactam nonsusceptible CRE isolates contained Class B (NDM, VIM) or Class D (OXA-48-like group) carbapenamases. These isolates were also resistant to other comparators including amikacin, gentamicin, and levofloxacin.
- MVB inhibited 100% of CRE KPC-producing isolates as shown in Figure 3.
 - Colistin was the most active comparator against KPC-producing isolates (81.3% susceptible, EUCAST).
- Figure 4 shows the distribution of KPC, Class B MBL, and Class D OXA-48-like producing isolates in different European Countries. Striking differences were present in the carbapenemase distribution.
 - Russia and Turkey, had primarily OXA-48-producing isolates.
 - Belarus had primarily MBL-producing isolates, and Greece had both MBL and KPC-producing isolates.
 - Western European countries had primarily KPC-producing isolates.
- There were 75 KPC-producing isolates from 7 countries.
 - KPC-producing isolates were mainly *K. pneumoniae* (n=71), which included 58 bla_{KPC-3}, 16 bla_{KPC-2}, and 1 bla_{KPC-12}. Four *E. coli* had bla_{KPC-3}
 - Italy had the highest number of KPC-producing isolates at 42 (56%).
 - 29 KPC-producing isolates were from ICU patients, 36 from non-ICU.

Table 1 Susceptibility of meropenem-vaborbactam and comparators against *Enterobacterales* isolates from patients hospitalized with pneumonia

Organisms and organism groups (n)	% susceptible using CLSI/ EUCAST[a] breakpoints					
	Meropenem-vaborbactam	Meropenem	Amikacin	Gentamicin	Levofloxacin	Colistin[b]
Enterobacterales (6,846)	97.7/98.0	94.6/95.1	96.2/94.2	86.0/85.3	75.0/75.0	-/76.8
ESBL-phenotype (1,368)	89.5/90.6	75.7/77.4	83.0/75.1	49.9/48.8	22.6/22.6	-/88.3
ESBL nonCR[c] (1045)	100.0/100.0	98.6/100.0	94.0/87.0	53.4/52.4	28.4/28.4	-/92.5
CRE (362)	56.6/63.0	3.6/7.7	51.9/41.4	42.5/39.8	8.9/8.9	-/74.2
KPC-producing (75)	100.0/100.0	0.0/0.0	56.0/48.0	61.3/60.0	6.7/6.7	-/81.3
MBL or OXA-48[d] (157)	21.0/29.9	6.4/12.1	40.8/32.5	27.4/26.1	8.3/8.3	-/85.3
ICU isolates (3,218)	96.6/97.1	93.4/94.2	95.0/93.0	85.2/84.6	76.0/76.0	-/76.4
ESBL-phenotype (705)	85.8/87.5	72.6/75.3	79.1/71.2	47.1/45.7	22.8/22.8	-/86.1
ESBL nonCR (519)	100.0/100.0	97.7/100.0	92.1/85.0	49.9/48.7	29.4/29.4	-/90.5
CRE (206)	47.6/55.3	3.9/9.2	47.1/37.9	42.2/39.8	9.3/9.3	-/73.3
KPC-producing (29)	100.0/100.0	0.0/0.0	48.3/37.9	79.3/75.9	6.9/6.9	-/62.1
MBL or OXA-48 (113)	19.5/30.1	6.2/10.6	37.2/29.2	28.3/26.5	7.1/7.1	-/89.4
ICU-VAP isolates (1,890)	95.9/96.7	92.1/93.2	93.7/91.5	83.3/82.6	74.3/74.3	-/76.8
ESBL-phenotype (455)	84.2/86.8	69.9/73.8	75.6/67.3	46.2/44.6	20.7/20.7	-/86.5
ESBL nonCR (324)	100.0/100.0	96.6/100.0	89.2/82.4	49.1/47.8	27.5/27.5	-/90.4
CRE (144)	45.8/56.2	4.9/11.1	45.8/34.7	41.7/38.9	7.7/7.7	-/77.1
KPC-producing (11)	100.0/100.0	0.0/0.0	27.3/18.2	81.8/81.8	0.0/0.0	-/72.7
MBL or OXA-48 (89)	21.3/34.8	6.7/11.2	39.3/30.3	31.5/29.2	8.0/8.0	-/92.1
non-ICU isolates (2,627)	98.2/98.5	94.2/94.5	96.4/94.0	84.5/83.7	70.3/70.3	-/78.2
ESBL-phenotype (530)	92.3/92.8	74.3/75.1	84.9/76.6	49.8/48.9	19.2/19.2	-/89.4
ESBL nonCR (396)	100.0/100.0	99.2/100.0	94.9/88.1	53.5/52.8	24.7/24.7	-/93.7
CRE (153)	69.9/74.5	3.3/5.9	59.5/47.1	43.8/40.5	8.5/8.5	-/75.7
KPC-producing (46)	100.0/100.0	0.0/0.0	60.9/54.3	50.0/50.0	6.5/6.5	-/93.5
MBL or OXA-48 (41)	26.8/31.7	7.3/17.1	53.7/43.9	26.8/26.8	12.2/12.2	-/75.0

[a] CLSI M100 30[th] Ed., EUCAST v10.0 (2020)
[b] No CLSI susceptible category for colistin
[c] CR carbapenem resistant
[d] Metallo-β-lactamase (MBL) included NDM (n=50) or VIM (n=14); OXA-48 included OXA-48-like carbapenemases.

Figure 1 Five most common *Enterobacterales* species isolated from pneumonia

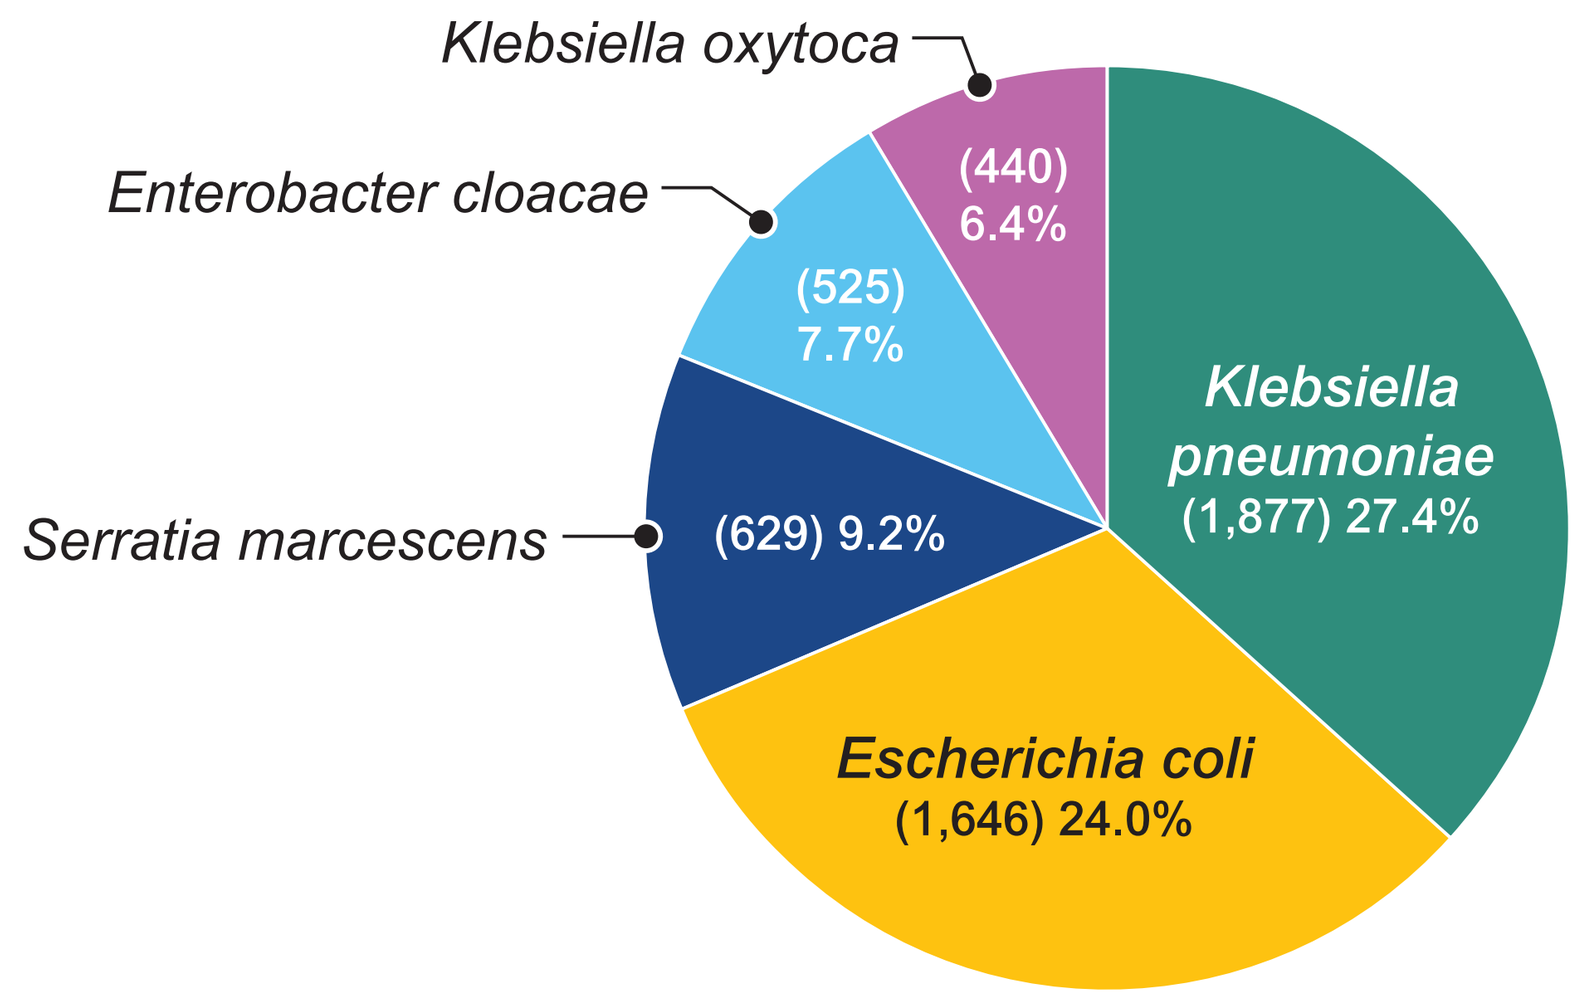

Klebsiella oxytoca
Enterobacter cloacae
Serratia marcescens
(440) 6.4%
(525) 7.7%
(629) 9.2%
Klebsiella pneumoniae (1,877) 27.4%
Escherichia coli (1,646) 24.0%

Figure 2 Meropenem-vaborbactam MIC (mg/L) distribution of *Enterobacterales* isolates from PHP* for ICU patients, ICU patients with VAP, non-ICU patients, and patients for which ICU status was not available

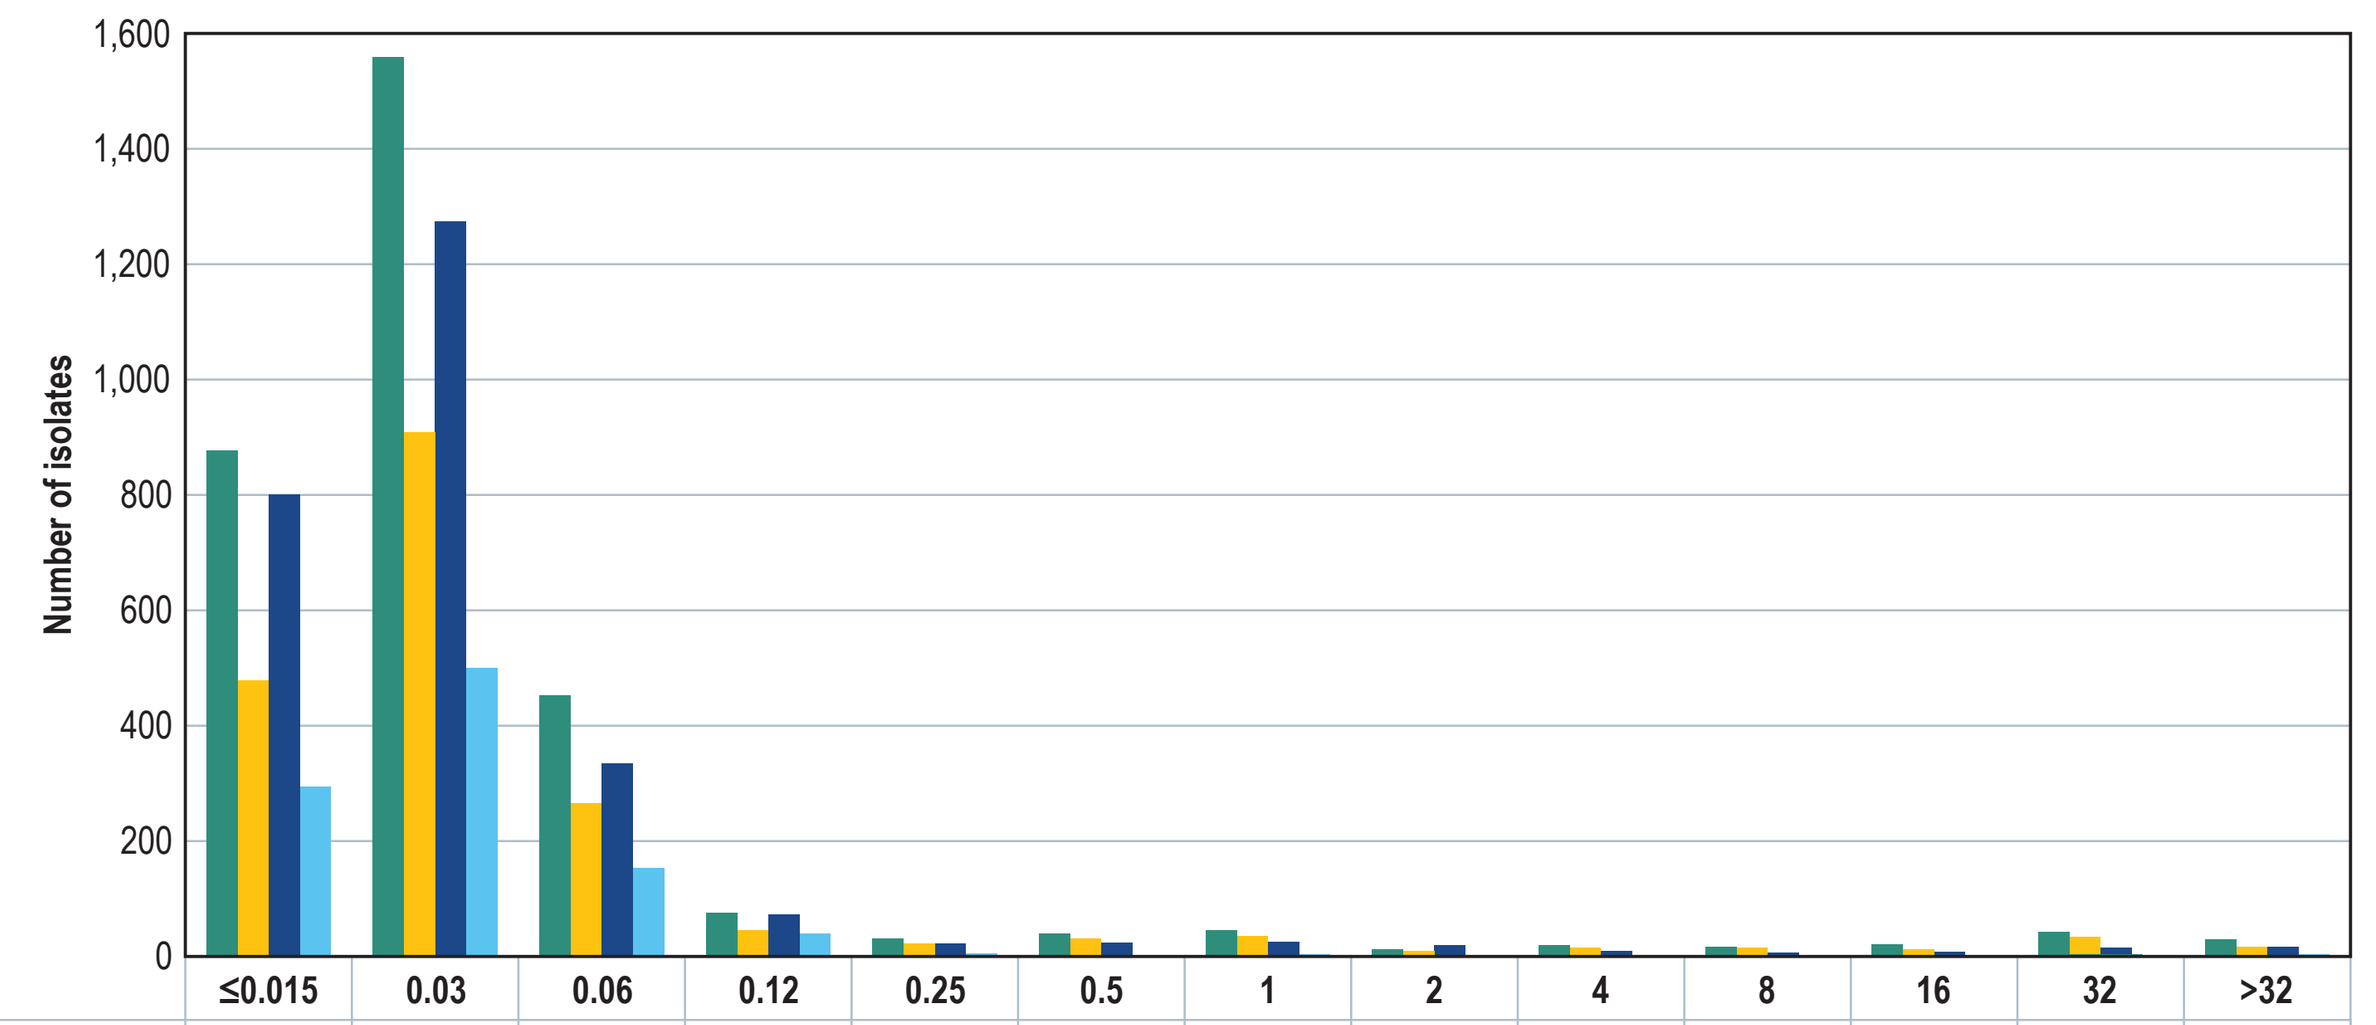

Number of isolates
1,600
1,400
1,200
1,000
800
600
400
200
0

	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32
ICU	877	1,559	452	76	31	39	45	12	19	16	21	42	29
ICU-VAP	479	908	265	46	23	31	35	10	15	15	12	34	17
non-ICU	800	1,274	335	73	22	24	25	19	9	7	8	15	16
No info	294	500	153	40	5	1	4	1					3

*PHP, patients hospitalized with pneumonia; ICU. intensive care unit; ICU-VAP, ICU patients with ventilator-associated pneumonia; non-ICU, in hospital service other than ICU; no info, ICU status was not indicated by submitting laboratory

Figure 3 Activity of meropenem and meropenem-vaborbactam against KPC-producing isolates

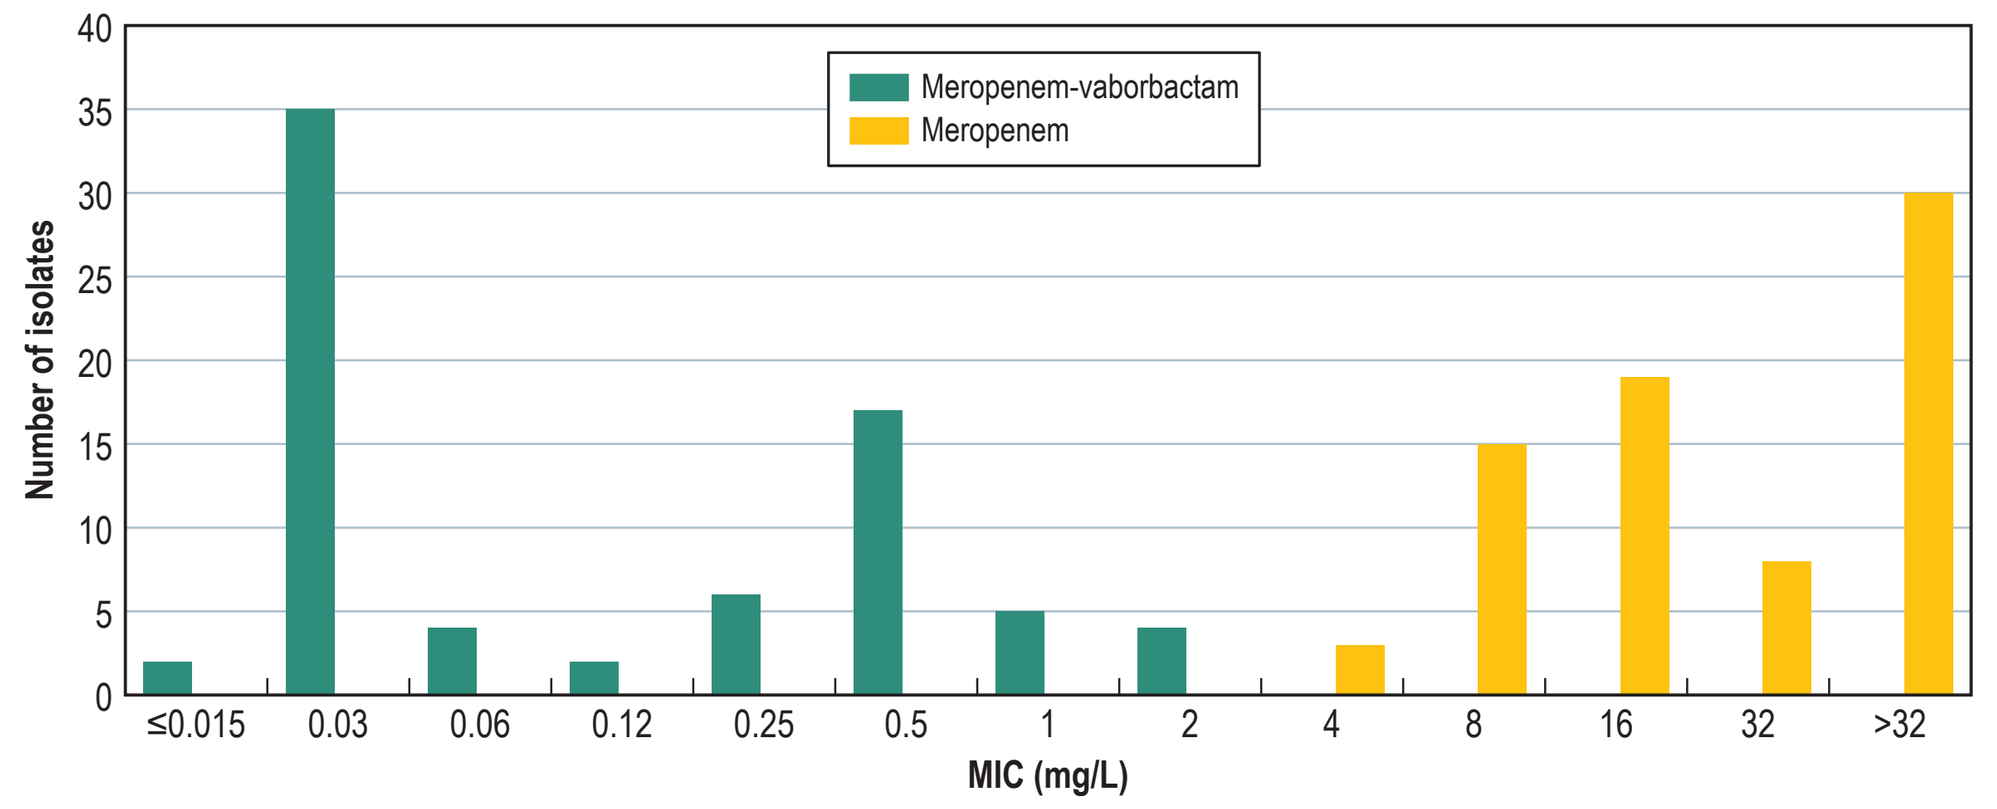

Meropenem-vaborbactam
Meropenem
Number of isolates
40
35
30
25
20
15
10
5
0
≤0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 >32
MIC (mg/L)

KPC, carbapenemase

Figure 4 European countries with KPC, metallo-β-lactamases or OXA-48-like carbapenemases

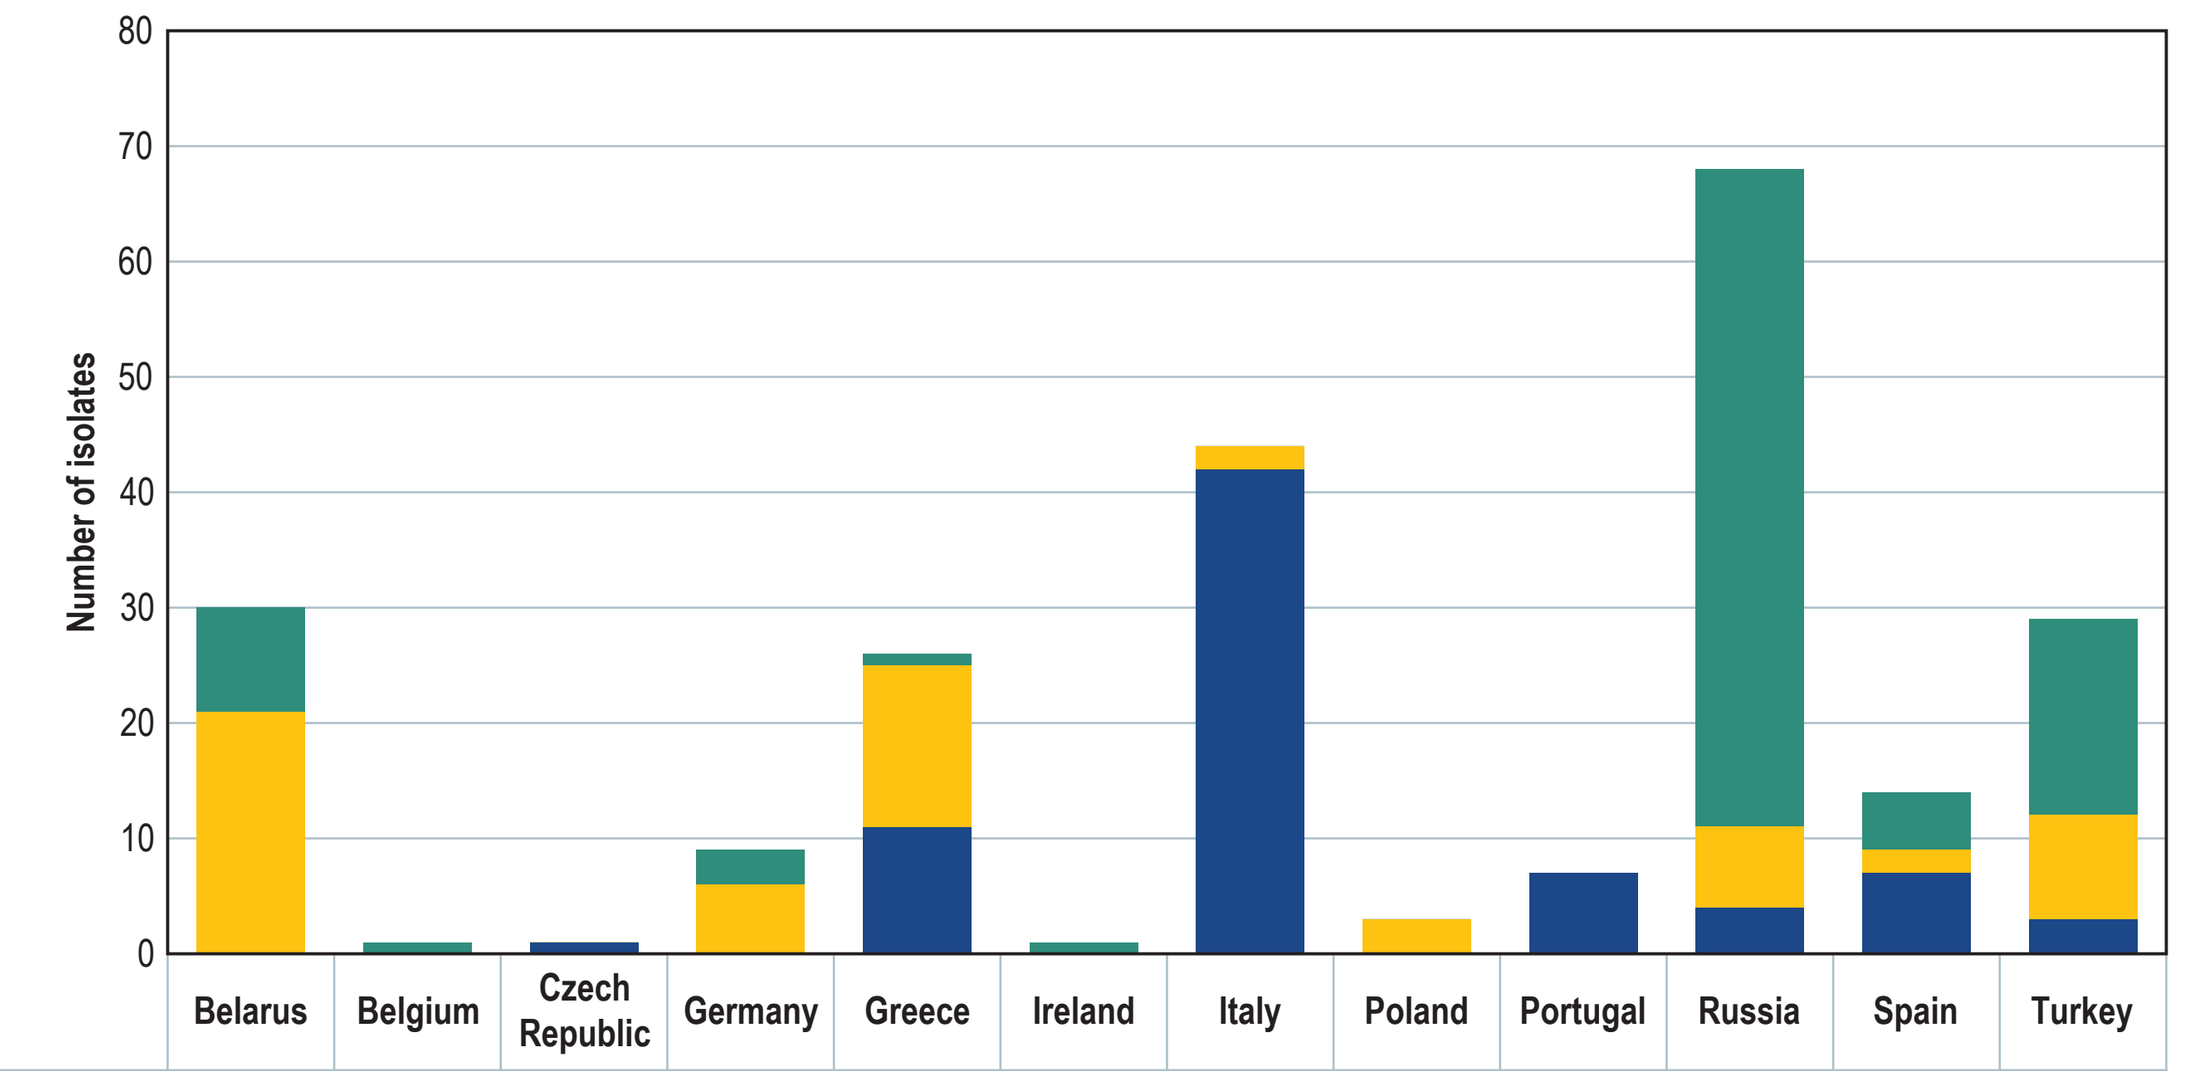

Number of isolates
80
70
60
50
40
30
20
10
0

	Belarus	Belgium	Czech Republic	Germany	Greece	Ireland	Italy	Poland	Portugal	Russia	Spain	Turkey
OXA-48-like	9	1	0	3	1	1	0	0	0	57	5	17
MBL	21	0	0	6	14	0	2	3	0	7	2	9
KPC	0	0	1	0	11	0	42	0	7	4	7	3

OXA-48-like carbapenemases
MBL- NDM (n=50) or VIM (n=14) metallo-β-lactamases;
KPC- *Klebsiella pneumoniae* carbapenemase, KPC 2, 3 and 12.

Conclusions

- Meropenem-vaborbactam had the highest susceptibility (97.7/98.2%) of tested agents against pathogens isolated from patients hospitalized with pneumonia, including VABP.
 - Isolates from ICU and non-ICU patients had similar susceptibilities for all antimicrobials except levofloxacin, which had a lower susceptibility rate in non-ICU isolates.
- The overall rate of CRE isolates was 5.3%, and the type of carbapenemase varied by country, with more KPC-producing isolates in Western European, and more OXA-48 and MBL-producing isolates in Eastern European countries.
- Meropenem-vaborbactam was very active against *Enterobacterales* isolates producing KPC enzymes inhibiting 100% of the studied isolates.
- These data suggest that meropenem-vaborbactam is a useful treatment option for both ICU and non-ICU patients hospitalized with pneumonia, including VABP, even when pneumonia is caused by carbapenem-resistant strains, in regions with a high prevalence of KPC.

Acknowledgements

Funding for this study was provided by A. MENARINI Industrie Farmaceutiche Riunite S.R.I., Florence, Italy.

References

Castanheira, M., M. D. Huband, R. E. Mendes and R. K. Flamm (2017). “Meropenem-Vaborbactam Tested against Contemporary Gram-Negative Isolates Collected Worldwide during 2014, Including Carbapenem-Resistant, KPC-Producing, Multidrug-Resistant, and Extensively Drug-Resistant Enterobacteriaceae.” *Antimicrob Agents Chemother* 61(9).

CLSI (2018). M07 Eleventh edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically. Wayne, PA, Clinical and Laboratory Standards Institute.

CLSI (2020). M100 30th edition. Performance Standards for Antimicrobial Susceptibility Testing. Wayne, PA, Clinical and Laboratory Standards Institute.

EUCAST (2020) “The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters” 2020.

Lomovskaya, O., D. Sun, D. Rubio-Aparicio, K. Nelson, R. Tsivkovski, D. C. Griffith and M. N. Dudley (2017). “Vaborbactam: Spectrum of Beta-Lactamase Inhibition and Impact of Resistance Mechanisms on Activity in Enterobacteriaceae.” *Antimicrob Agents Chemother* 61(11).

Sader, H. S., M. Castanheira, R. K. Flamm, R. E. Mendes, D. J. Farrell and R. N. Jones (2015). “Tigecycline activity tested against carbapenem-resistant Enterobacteriaceae from 18 European nations: results from the SENTRY surveillance program (2010–2013).” *Diagn Microbiol Infect Dis* 83(2): 183–186.

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

MENARINI group