IDWEEK 2020 | Poster Number: 1463

Activity of Delafloxacin against Multi-Drug-Resistant Fastidious Respiratory Pathogens from European Medical Centers (2014–2019)

Dee Shortridge, Jennifer M. Streit, Michael D. Huband, Robert K. Flamm

JMI Laboratories, North Liberty, IA, USA

Introduction

- Delafloxacin is an anionic fluoroquinolone approved in the United States and Europe for the treatment of acute bacterial skin and skin structure infections.
- Delafloxacin recently was approved in the United States for treatment of community-acquired bacterial pneumonia (CABP).
- This study examines the *in vitro* susceptibilities of delafloxacin and comparator agents against recent European isolates of *Streptococcus pneumoniae*, *Haemophilus influenzae*, *H. parainfluenzae*, and *Moraxella catarrhalis*.
- This study also analyzed the delafloxacin susceptibility of isolates resistant to current first-line therapies: Erythromycin-resistant *S. pneumoniae*, multi-drug resistant *S. pneumoniae*, and β-lactamase positive *H. influenzae*.

Materials and Methods

- A total of 2,835 *S. pneumoniae*, 1,484 *H. influenzae*, 959 *M. catarrhalis*, and 20 *H. parainfluenzae* isolates were collected from community-acquired respiratory tract infections (CA-RTI) from European hospitals between 2014 and 2019. Sites included only 1 isolate/patient/infection episode.
- JMI Laboratories confirmed isolate identifications using biochemical and/or molecular methods as needed. Susceptibility testing was performed by broth microdilution methodology according to CLSI (CLSI M07, 2018).
- Where applicable, CLSI/ FDA and EUCAST v. 10.0 (2020) breakpoints were used.
 - Delafloxacin has FDA but not EUCAST breakpoints for CABP organisms *S. pneumoniae* and *Haemophilus* spp. No breakpoints have been determined for *M. catarrhalis*.
 - In EUCAST v. 10.0 (2020), breakpoints for *S. pneumoniae* with levofloxacin and *H. influenzae* with amoxicillin-clavulanate, isolates that previously were susceptible are now categorized as "susceptible-increased exposure, intermediate."
- Multidrug-resistant (MDR) *S. pneumoniae* isolates were categorized as being nonsusceptible (NS) to amoxicillin-clavulanate, erythromycin (ERY), and tetracycline using EUCAST criteria.
- The nitrocefin test (Cefinase, BBL) was used to determine the presence of β-lactamase (BL) for *Haemophilus* species and *M. catarrhalis*.

Results

- The delafloxacin MIC distributions of the 4 main species, *H. influenzae, H. parainfluenzae, M. catarrhalis* and *S. pneumoniae*, are shown in Figure 1.
 - *S. pneumoniae* (n=2,835) was the organism most frequently isolated from CA-RTI, followed by *H. influenzae* (n=1,484).
 - *M. catarrhalis* had 959 isolates, *H. parainfluenzae* had only 20 isolates.
- Delafloxacin was the most active agent against *S. pneumoniae*; it had the lowest $MIC_{50/90}$ values of 0.015/0.03 mg/L (Table 1). Delafloxacin activities were similar when tested against *S. pneumoniae* isolates with resistant phenotypes.
 - MDR isolates had delafloxacin $MIC_{50/90}$ values of 0.015/0.03 mg/L.
- The percent susceptibility of all *S. pneumoniae* and MDR isolates for delafloxacin and comparators are shown in Table 1.
 - Delafloxacin susceptibility, based on FDA susceptible breakpoints for *S. pneumoniae* (≤0.03 mg/L), was 95.0%. No EUCAST breakpoints for delafloxacin are available for *S. pneumoniae* or *Haemophilus spp.*
 - The drugs with the lowest percent susceptibility for *S. pneumoniae*, using EUCAST breakpoints, were penicillin with 71.8% and erythromycin with 76.8%.
 - Using EUCAST criteria, 253 isolates were MDR, which is 8.9% of all isolates.
- The percentage of MDR *S. pneumoniae* isolates by country is shown in Figure 2.
 - The country with the largest number of MDR *S. pneumoniae* was Turkey (n=55, 29.7%).
- Delafloxacin was the most active fluoroquinolone against *H. influenzae* (Table 2).
 - 18.1% of *H. influenzae*, 5.0% of *H. parainfluenzae*, and 86.9% of *M. catarrhalis* were β-lactamase positive. β-lactamase presence did not affect fluoroquinolone MIC values.
- The percent susceptibility of *H. influenzae* and comparators is shown in Table 2.
 - Delafloxacin susceptibility against *H. influenzae* (≤0.004 mg/L) was 97.0%.
 - For *H. influenzae*, only 61.1% were susceptible to trimethoprim-sulfamethoxazole, and the ampicillin resistance was 24.7%.
 - For the β-lactamase negative *H. influenzae*, 8.1% were ampicillin-R using EUCAST breakpoints (data not shown).

Figure 1 Delafloxacin MIC distribution for respiratory tract pathogens

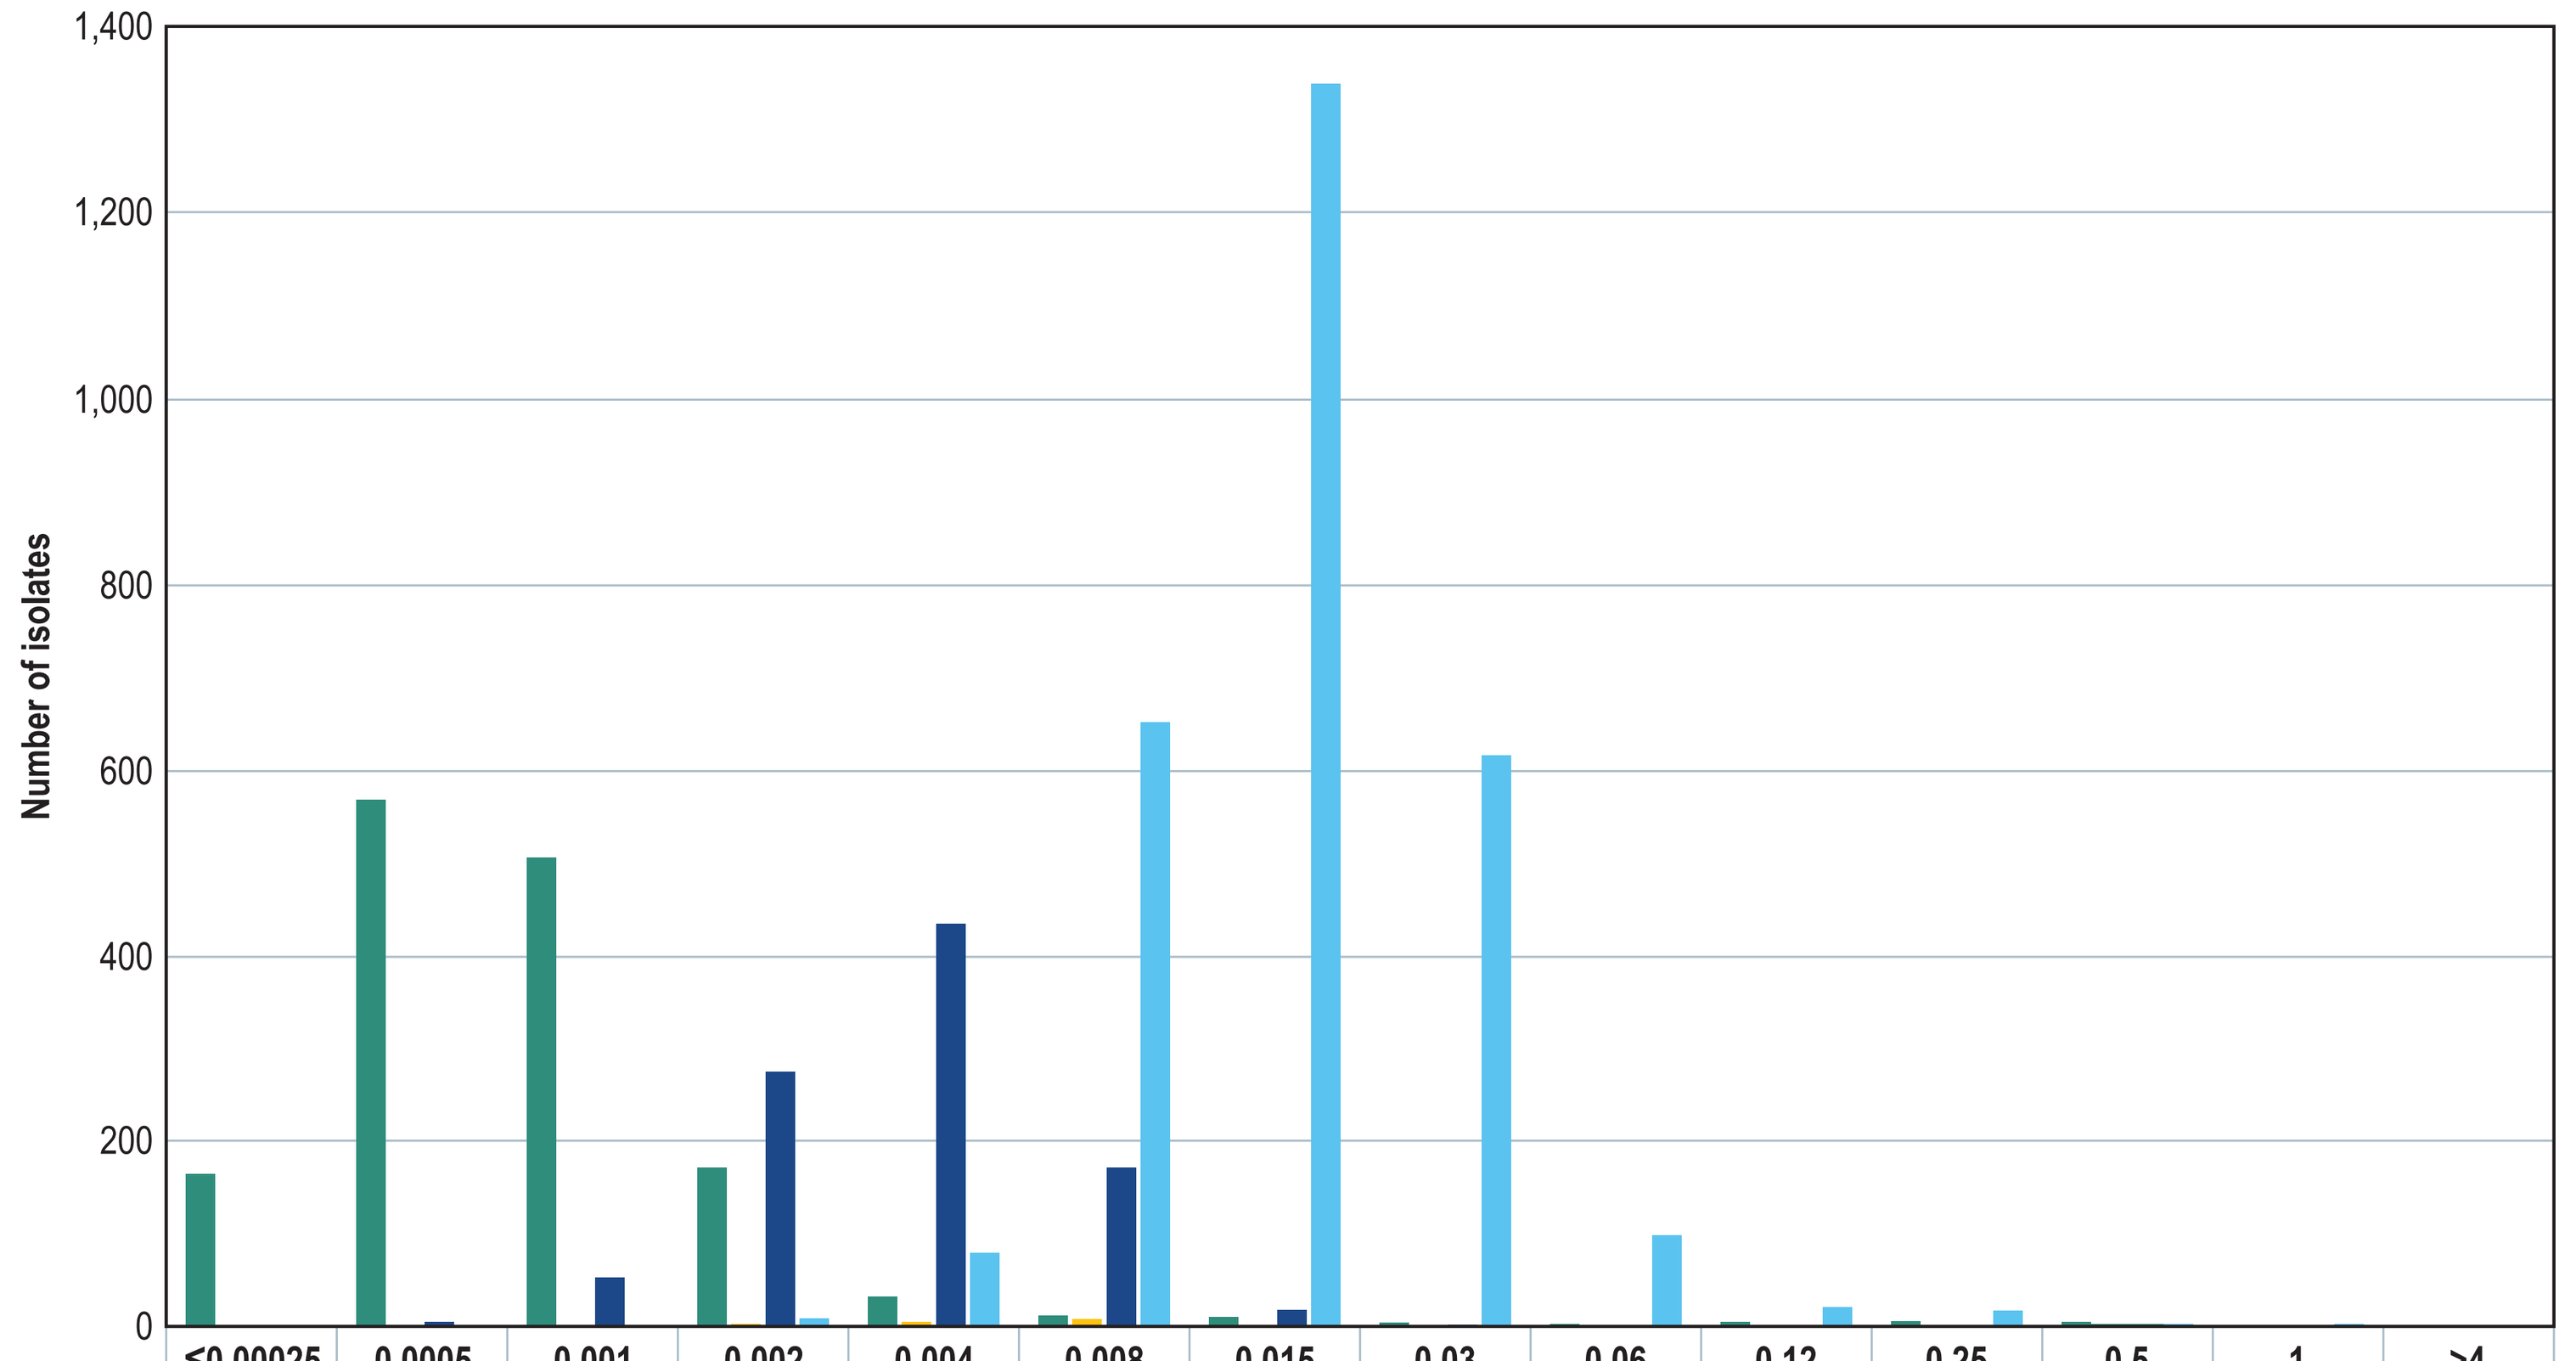
1,400
1,200
1,000
800
600
400
200
0
Number of isolates

	≤0.00025	0.0005	0.001	0.002	0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	>4
Haemophilus influenzae	164	567	505	171	32	12	10	4	3	5	6	5		
Haemophilus parainfluenzae		1		3	5	8		1		1		1		
Moraxella catarrhalis		5	53	274	434	171	18	2	1	1				
Streptococcus pneumoniae				9	79	651	1,338	615	98	21	17	3	3	1

Figure 2 Percent of multi-drug resistance *S. pneumoniae* by country

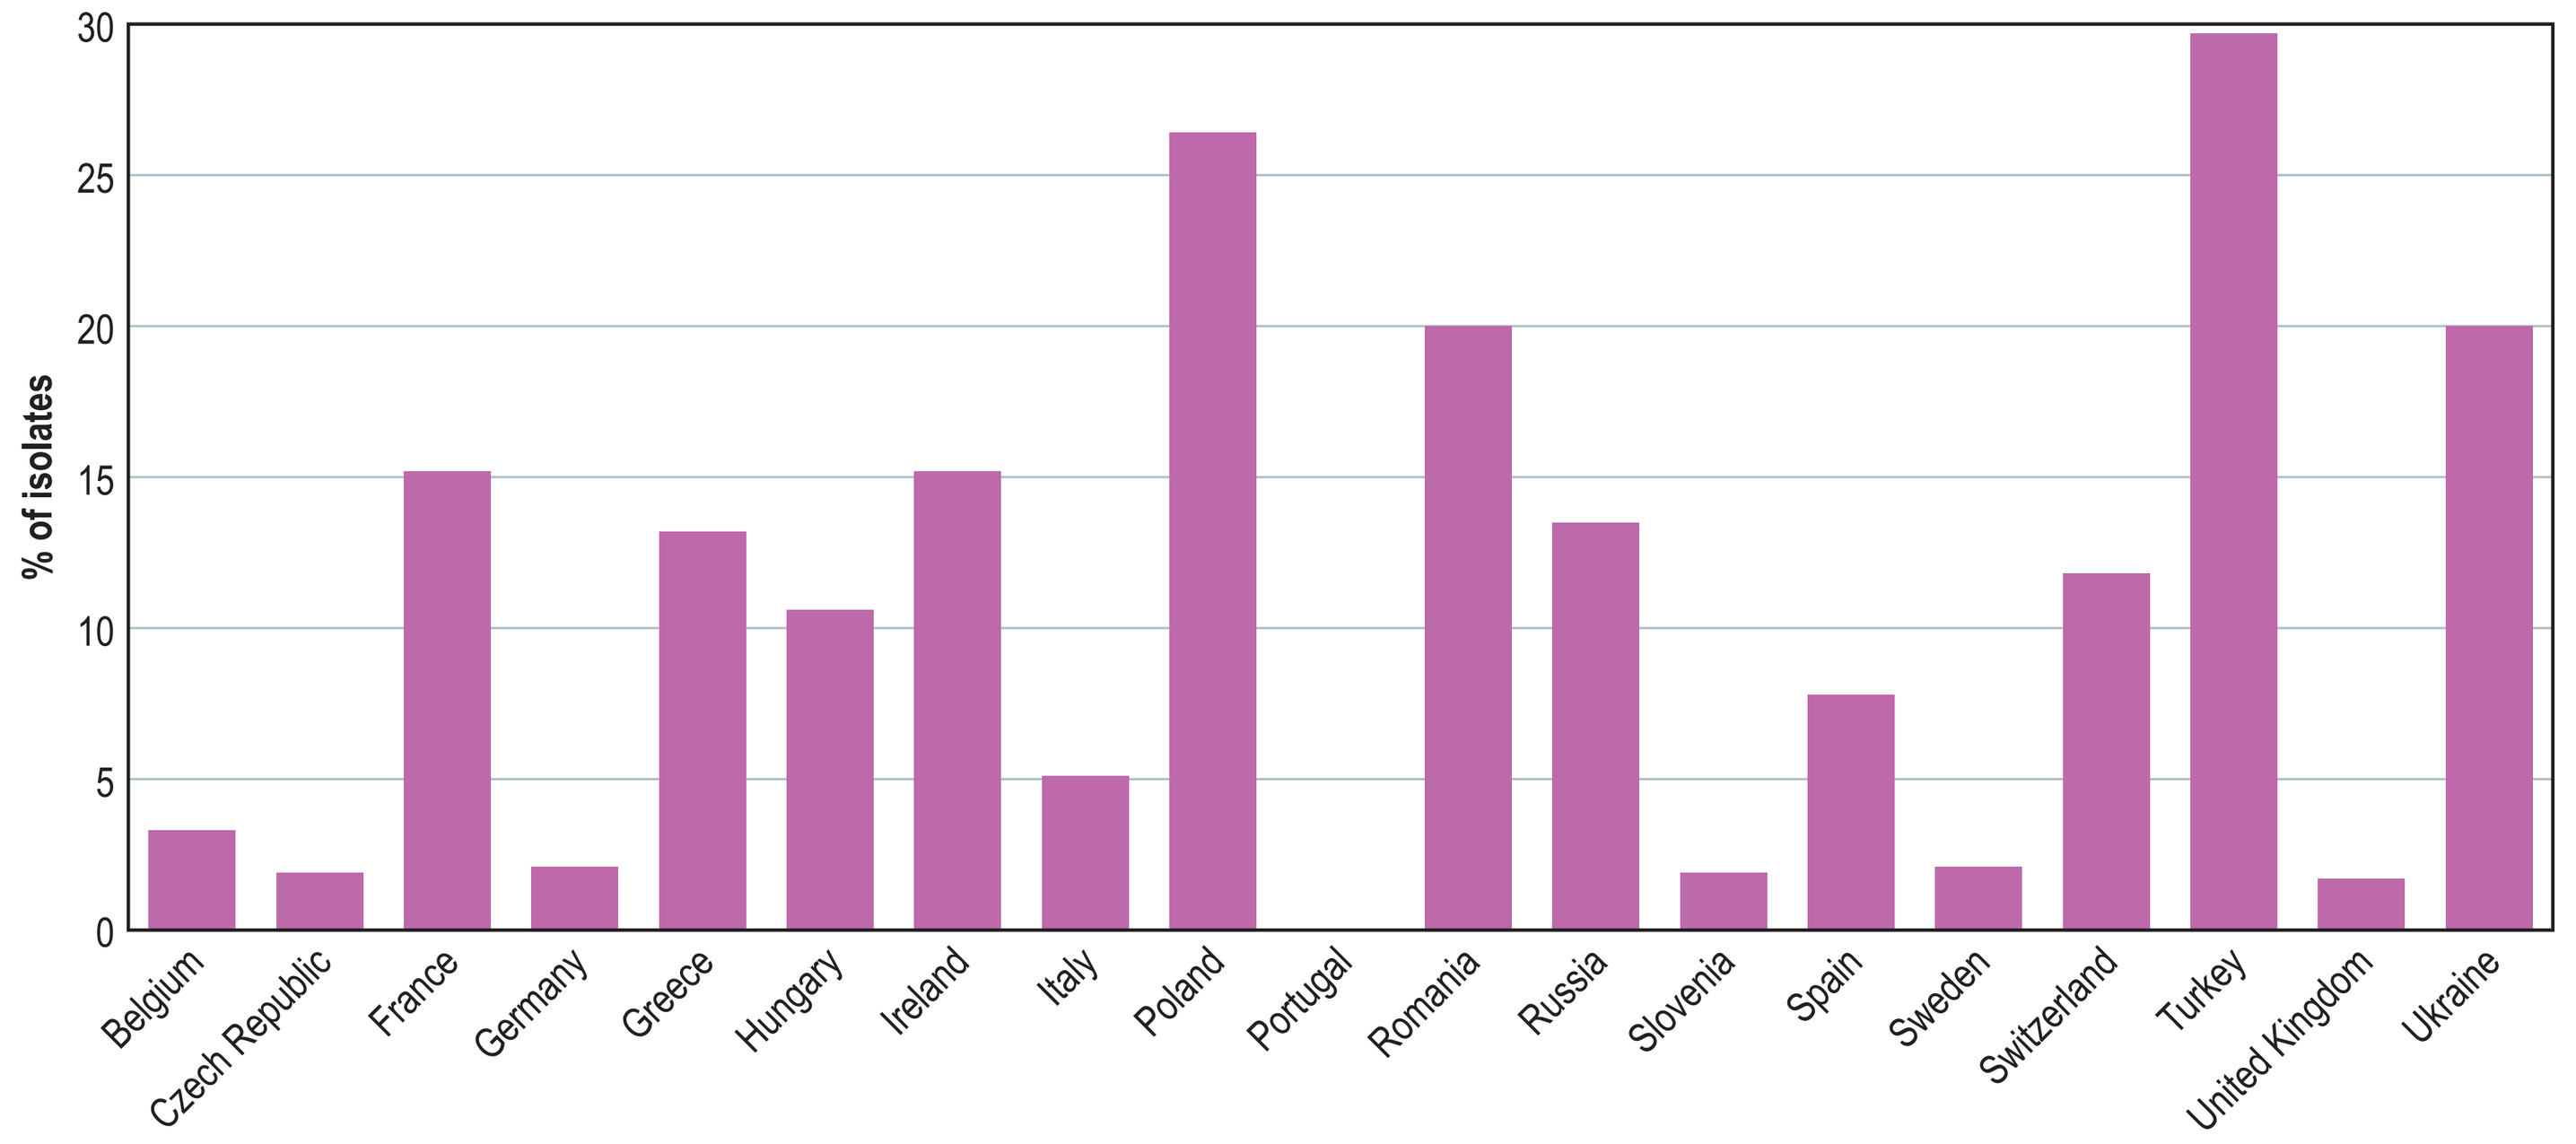
30
25
20
15
10
5
0
% of isolates
Belgium
Czech Republic
France
Germany
Greece
Hungary
Ireland
Italy
Poland
Portugal
Romania
Russia
Slovenia
Spain
Sweden
Switzerland
Turkey
United Kingdom
Ukraine

Table 1 Antimicrobial activity of delafloxacin and comparator agents tested against *S. pneumoniae*

Antimicrobial agent/ organism/organism group	No. of isolates	mg/L			CLSI[a]			EUCAST[a]		
		MIC_{50}	MIC_{90}	MIC range	%S	%I	%R	%S	%I	%R
Streptococcus pneumoniae										
Delafloxacin	2,835	0.015	0.03	≤0.004 to >4	95.0[b,c]					
Levofloxacin	2,835	1	2	0.25 to >4	97.9	0.4	1.7	[d]	97.9	2.1
Moxifloxacin	2,715	≤0.12	0.25	≤0.12 to >4	98.5	0.7	0.8	98.5		1.5
Amoxicillin-clavulanic acid	2,834	≤1	2	≤1 to >4	93.7[e]	2.6	3.7	83.2[f]	4.1	12.7
Ceftaroline	2,833	≤0.015	0.12	≤0.015 to >1	99.9			99.8		0.2
Ceftriaxone	2,834	≤0.06	1	≤0.06 to >2	86.9[g]	9.4	3.7	86.9	12.4	0.7
					96.3[e]	3.0	0.7			
Clindamycin	2,835	≤0.25	>1	≤0.25 to >1	83.4	0.4	16.2	83.8		16.2
Erythromycin	2,833	≤0.12	>2	≤0.12 to >2	76.8	0.2	23	76.8	0.2	23
Meropenem	2,715	≤0.015	0.5	≤0.015 to >1	85.7	8.8	5.5	85.7[g]		14.3
								100.0[e]		0
Penicillin	2,835	≤0.06	2	≤0.06 to >4	71.8[f]	15.2	13	71.8[g]		28.2
					71.8[h]		28.2	71.8[e]	23	5.1
					94.9[i]	4.6	0.5			
Tetracycline	2,835	≤0.5	>4	≤0.5 to >4	78.8	0.4	20.8	78.8	0.4	20.8
Trimethoprim-sulfamethoxazole	2,835	≤0.5	>4	≤0.5 to >4	73.4	8.7	17.8	78.8	3.4	17.8
Multi-drug resistant *S. pneumoniae*										
Delafloxacin	253	0.015	0.03	≤0.004 to 1	90.9[b,c]					
Levofloxacin	253	1	2	0.5 to >4	94.5	1.2	4.3	[d]	94.5	5.5
Moxifloxacin	240	≤0.12	0.25	≤0.12 to >4	96.2	1.2	2.5	96.2		3.8
Amoxicillin-clavulanic acid	253	2	>4	1 to >4	60.9[e]	17.8	21.3	0.0[f]	23.7	76.3
Ceftaroline	252	0.12	0.25	≤0.008 to 1	99.6			98.4		1.6
Ceftriaxone	253	1	2	0.12 to >2	17.4[g]	51	31.6	17.4	76.7	5.9
					68.4[e]	25.7	5.9			
Clindamycin	253	>1	>1	≤0.25 to >1	19.8	0.8	79.4	20.6		79.4
Erythromycin	253	>2	>2	0.5 to >2	0.0	1.2	98.8	0.0	1.2	98.8
Meropenem	240	0.5	1	≤0.008 to >1	10.8	54.2	35	10.8[g]		89.2
								100.0[e]		0
Penicillin	253	2	4	0.5 to >4	0.0[f]	16.6	83.4	0.0[g]		100
					0.0[h]		100.0	0.0[e]	64.4	35.6
					64.4[i]	30.8	4.7			
Tetracycline	253	>4	>4	2 to >4	0.0	1.2	98.8	0.0	1.2	98.8
Trimethoprim-sulfamethoxazole	253	4	>4	≤0.5 to >4	27.7	16.6	55.7	38.3	5.9	55.7

[a] Criteria as published by CLSI (2020) and EUCAST (2020).
[b] Using FDA CABP breakpoints.
[c] FDA breakpoints published 2019-OCT-29.
[d] An arbitrary susceptible breakpoint of ≤0.001 mg/L and/or >50 mm has been published by EUCAST indicating that susceptible should not be reported for this organism-agent combination and intermediate should be interpreted as susceptible increased exposure (EUCAST 2020).
[e] Using non-meningitis breakpoints.
[f] Using oral breakpoints.
[g] Using meningitis breakpoints.
[h] Using parenteral, meningitis breakpoints.
[i] Using parenteral, non-meningitis breakpoints.

Table 2 Antimicrobial activity of delafloxacin and comparator agents tested against *H. influenzae*

Antimicrobial agent/ organism/ organism group	No. of isolates	mg/L			CLSI[a]			EUCAST[a]		
		MIC_{50}	MIC_{90}	MIC range	%S	%I	%R	%S	%I	%R
H. influenzae										
Delafloxacin	1,484	≤0.001	0.002	≤0.001 to >0.25	97.0[b,c]					
Levofloxacin	1,484	≤0.015	0.03	≤0.015 to >2	98.9			97.6		2.4
Ciprofloxacin	1,484	0.015	0.015	≤0.008 to >1	98.8			97.8		2.2
Moxifloxacin	1,400	0.03	0.03	≤0.008 to >1	98.7			98.0		2.0
Amoxicillin-clavulanic acid	1,484	0.5	2	≤0.12 to >8	99.7		0.3	[d,e]	94.6	5.4
								94.6[f]		5.4
Ampicillin	1,484	0.5	>8	≤0.12 to >8	75.3	6.4	18.3	75.3		24.7
Azithromycin	1,484	1	1	≤0.12 to >8	99.1			99.1[g]		
Ceftaroline	1,483	0.008	0.03	≤0.004 to 0.5	100.0			98.3		1.7
Ceftriaxone	1,484	≤0.015	≤0.015	≤0.015 to 0.25	100.0			99.9		0.1
Clarithromycin	1,484	8	16	≤0.12 to >16	89.0	9.7	1.3	100.0[g]		
Meropenem	1,484	0.06	0.12	≤0.008 to 0.5	100.0			99.5[h]		0.5
								100.0[i]		0.0
Tetracycline	1,484	0.5	1	0.12 to >8	99.2	0.0	0.8	98.8	0.4	0.8
Trimethoprim-sulfamethoxazole	1,483	0.12	>4	≤0.06 to >4	61.1	5.9	33	61.1	2.8	36.1
H. influenzae- BLP										
Delafloxacin	268	≤0.001	0.002	≤0.001 to 0.03	99.6[b,c]					
Levofloxacin	268	≤0.015	0.03	≤0.015 to 1	100.0			99.3		0.7
Ciprofloxacin	268	0.015	0.015	≤0.008 to 1	100.0			99.3		0.7
Moxifloxacin	254	0.03	0.03	0.008 to 1	100.0			99.6		0.4
Amoxicillin-clavulanic acid	268	1	2	0.25 to 8	99.6		0.4	[d,e]	94.8	5.2
								94.8[f]		5.2
Ampicillin	268	>8	>8	1 to >8	0.4	1.5	98.1	0.4		99.6
Azithromycin	268	0.5	1	≤0.12 to >8	98.5			98.5[g]		
Ceftaroline	268	0.015	0.03	≤0.004 to 0.5	100.0			95.1		4.9
Ceftriaxone	268	≤0.015	≤0.015	≤0.015 to 0.12	100.0			100.0		0.0
Clarithromycin	268	8	8	0.25 to >16	91.0	7.5	1.5	100.0[g]		
Meropenem	268	0.06	0.12	≤0.008 to 0.5	100.0			99.6[h]		0.4
								100.0[i]		0.0
Tetracycline	268	0.5	0.5	0.25 to >8	97.4	0.0	2.6	96.3	1.1	2.6
Trimethoprim-sulfamethoxazole	268	0.12	>4	≤0.06 to >4	57.1	2.6	40.3	57.1	0.7	42.2

[a] Criteria as published by CLSI (2020) and EUCAST (2020).
[b] Using CABP breakpoints.
[c] FDA breakpoints published 2019-OCT-29.
[d] Using oral breakpoints.
[e] An arbitrary susceptible breakpoint of ≤0.001 mg/L and/or >50 mm has been published by EUCAST indicating that susceptible should not be reported for this organism-agent combination and intermediate should be interpreted as susceptible increased exposure (EUCAST 2020).
[f] Using parenteral breakpoints.
[g] Percentage of wild type based on ECV. EUCAST version 10.0 (2020).
[h] Using meningitis breakpoints.
[i] Using non-meningitis breakpoints.
[j] FDA breakpoints published 2017-DEC-13.

Conclusions

- Delafloxacin demonstrated potent *in vitro* antibacterial activity against *S. pneumoniae, H. influenzae, H. parainfluenzae*, and *M. catarrhalis*.
- Delafloxacin was active against the MDR *S. pneumoniae* that were non-susceptible to the agents commonly used as treatments for CABP: amoxicillin-clavulanate, tetracycline, and erythromycin.
- Resistance to fluoroquinolones was rare for all CA-RTI isolates, with ≥94% susceptiblity overall.
- These data support the utility of delafloxacin in the treatment of CABP, including infections caused by antibiotic-resistant strains of *S. pneumoniae* and *H. influenzae*.

Acknowledgements

Funding for this research was provided by A. MENARINI Industrie Farmaceutiche Riunite S.R.I., Florence, Italy.

References

CLSI (2018). M07 11th edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically. Wayne, PA, Clinical and Laboratory Standards Institute.

CLSI (2020). M100 30th edition. Performance Standards for Antimicrobial Susceptibility Testing. Wayne, PA, Clinical and Laboratory Standards Institute.

EUCAST (2020) "The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters."

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

MENARINI group