ASM Microbe 2018

Friday - 601

In Vitro Activity of a Novel Extended-Spectrum β-Lactamase Inhibitor, AAI101, in Combination with Cefepime, against *Enterobacteriaceae* Isolates Collected during 2016

MD HUBAND[1], PR RHOMBERG[1], KA FEDLER[1], RK FLAMM[1], P KNECHTLE[2], S SHAPIRO[2]

[1] JMI Laboratories, Inc., North Liberty, Iowa, USA; [2] Allecra Therapeutics SAS, F-68300 Saint-Louis, France

Contact Information:
Michael D. Huband
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

To obtain a PDF of this poster:

- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/ASM-Microbe-2018-cefepime.pdf

Charges may apply.
No personal information is stored.

Introduction

- Resistance development to existing β-lactam therapies due to the spread of extended-spectrum β-lactamases (ESBLs) among *Enterobacteriaceae* has created the need for new treatment options
- AAI101 is a novel extended-spectrum β-lactamase inhibitor (Figure 1) highly active against ESBLs and a broad array of other β-lactamases
- The combination of cefepime/AAI101 recently has completed a Phase 2 clinical trial in complicated urinary tract infections and Phase 3 is planned to be initiated in 2018
- Cefepime/AAI101 was granted Qualified Infectious Disease Product and Fast Track designations by the United States Food and Drug Administration
- A collection of 572 recent *Enterobacteriaceae* clinical isolates were tested for susceptibility to cefepime/AAI101, cefepime, meropenem, and piperacillin-tazobactam
- The isolates were obtained mostly from year 2016, primarily collected from North America and Europe, and included challenge strains

Figure 1. Chemical structure of AAI101

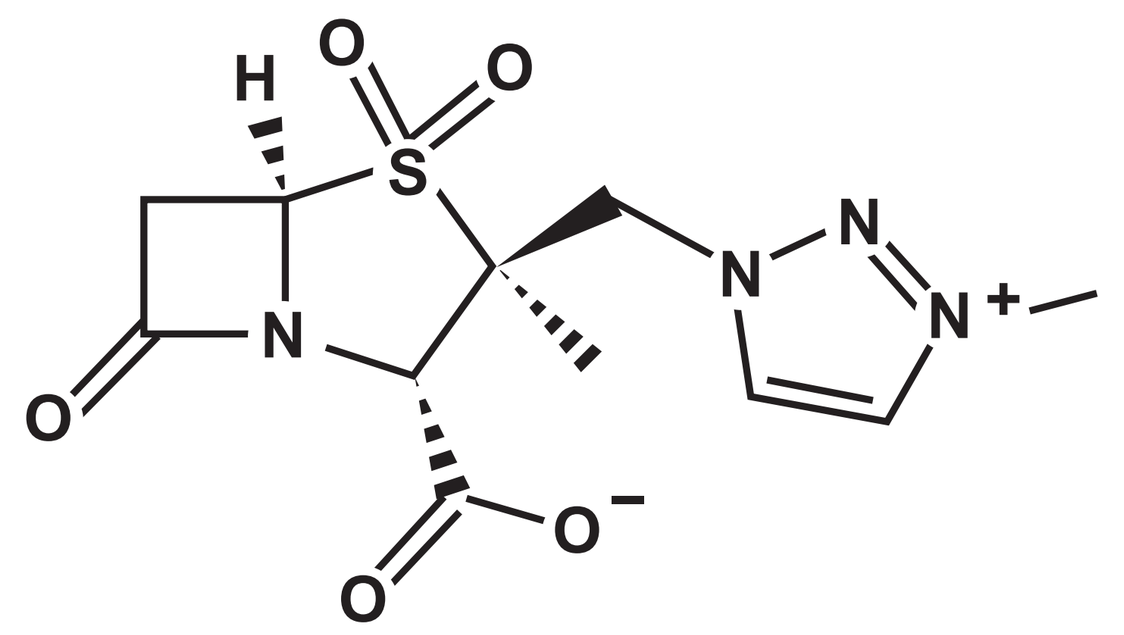

Materials and Methods

- A collection of 572 *Enterobacteriaceae* was obtained from US (74.0%), European (25.0%), and Latin American (1.0%) medical centers, with 91.1% of organisms isolated during 2016
- The panel included 16.8% (96/572) cefepime-nonsusceptible and 5.4% (31/572) carbapenem-resistant isolates with diverse resistance mechanisms
- Organisms were identified by matrix-assisted laser desorption ionization time-of-flight mass spectrometry or next-generation sequencing
- Susceptibility testing was performed using CLSI reference broth microdilution methodology; CLSI interpretive criteria were applied
- Isolates were tested for susceptibility to cefepime/AAI101, cefepime, meropenem, and piperacillin-tazobactam
- Fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL, in combination with cefepime, were chosen for broth microdilution susceptibility testing based on prior microbiological and pharmacokinetic/pharmacodynamic studies
- For comparative purposes, CLSI breakpoint interpretive criteria for cefepime were applied to cefepime/AAI101 combinations

Results

- Against the complete collection of 572 *Enterobacteriaceae*, which included cefepime-nonsusceptible and carbapenem-resistant isolates, AAI101 restored the activity of cefepime to the susceptible interpretive category against 95.1%-97.0% of the isolates with an MIC_{90} value of 0.5 µg/mL (fixed AAI101 concentrations of 4 µg/mL or 8 µg/mL; Table 1 and Figure 2)
 - MIC_{90} values for cefepime-only and piperacillin-tazobactam were in the resistant interpretive category with values of 16 µg/mL and 128 µg/mL, respectively
 - Cefepime/AAI101 activity was comparable to that of meropenem: 97.0% of isolates were inhibited at or below the cefepime susceptible breakpoint of 2 µg/mL, and 94.2% of the isolates inhibited at or below the meropenem susceptible breakpoint of 1 µg/mL, respectively
- Against cefepime-resistant, carbapenem-susceptible isolates of *Enterobacteriaceae* (n=40), AAI101 restored the activity of cefepime to the susceptible interpretive category against 97.5% of the isolates with an MIC_{90} value of 1 µg/mL (fixed AAI101 concentration of 8 µg/mL; Table 2 and Figure 3)
 - MIC_{90} values for piperacillin-tazobactam and meropenem were >128 µg/mL and 0.5 µg/mL, respectively (Table 2 and Figure 4)
- Against carbapenem-resistant isolates of *Enterobacteriaceae* (n=31), cefepime/AAI101 (fixed AAI101 concentration of 8 µg/mL), piperacillin-tazobactam, and meropenem showed 51.6%, 3.2% and 0% susceptible isolates
- Against *K. pneumoniae* (n=135) and *E. coli* (n=121), cefepime-only MIC_{90} values were in the resistant category (MIC_{90} values >128 µg/mL and 32 µg/mL, respectively). AAI101 at fixed concentrations of 4 µg/mL or 8 µg/mL restored cefepime MIC_{90} values to the susceptible-dose-dependent and susceptible interpretive categories (Table 2)
 - Piperacillin-tazobactam MIC_{90} values (>128 µg/mL and 64 µg/mL, respectively) were in the resistant category for *K. pneumoniae* and intermediate interpretive category for *E. coli*
 - Meropenem MIC_{90} value (>8 µg/mL) was in the resistant category for *K. pneumoniae* with 80% susceptible isolates

Table 1 Cumulative percent inhibition results for cefepime/AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against 572 recent *Enterobacteriaceae* isolates

Organism group (no. tested)/antimicrobials	No. of isolates at MIC (µg/mL; cumulative %)																	MIC_{50}	MIC_{90}
	≤0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>[a]		
Enterobacteriaceae (572)[b]																			
Cefepime/AAI101 (fixed AAI101 concentration of 4 µg/mL)	0 0.0	6 1.0	67 12.8	216 50.5	133 73.8	58 83.9	21 87.6	**21** **91.3**[c]	17 94.2	5 95.1	6 96.1	8 97.6	7 98.8	4 99.5	0 99.5	1 99.7	2 100.0	0.03	0.5
Cefepime/AAI101 (fixed AAI101 concentration of 8 µg/mL)	0 0.0	1 0.2	72 12.8	203 48.3	161 76.4	51 85.3	19 88.6	**24** **92.8**	16 95.6	8 97.0	4 97.7	4 98.4	3 99.0	4 99.7	0 99.7	2 100.0		0.06	0.5
Cefepime	0 0.0	2 0.3	62 11.1	180 42.7	125 64.5	35 70.6	23 74.6	17 77.6	15 80.2	16 83.0	6 84.1	22 87.9	**19** **91.3**	19 94.6	5 95.5	4 96.2	22 100.0	0.06	16
Meropenem	0 0.0	6 1.0	202 36.4	178 67.5	110 86.7	**27** **91.4**	6 92.5	5 93.3	5 94.2	2 94.6	2 94.9	6 96.0					23 100.0	0.03	0.12
Piperacillin-tazobactam					0 0.0	2 0.3	39 7.2	39 14.0	85 28.8	185 61.2	79 75.0	29 80.1	27 84.8	8 86.2	16 89.0	**15** **91.6**	48 100.0	2	128

[a] MIC value is greater than the highest concentration tested.
[b] Comprised of 21 *Citrobacter freundii* spp. complex, 21 *Citrobacter koseri*, 28 *Enterobacter aerogenes*, 77 *Enterobacter cloacae* spp. complex, 121 *Escherichia coli*, 27 *Klebsiella oxytoca*, 135 *Klebsiella pneumoniae*, 27 *Morganella morganii*, 27 *Proteus mirabilis*, 21 *Proteus vulgaris* group, 21 *Providencia rettgeri*, 21 *Providencia stuartii*, and 25 *Serratia marcescens* isolates.
[c] MIC_{90} value is in bold.

Figure 2 Cumulative percent inhibition results for cefepime/AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against 572 recent *Enterobacteriaceae* isolates

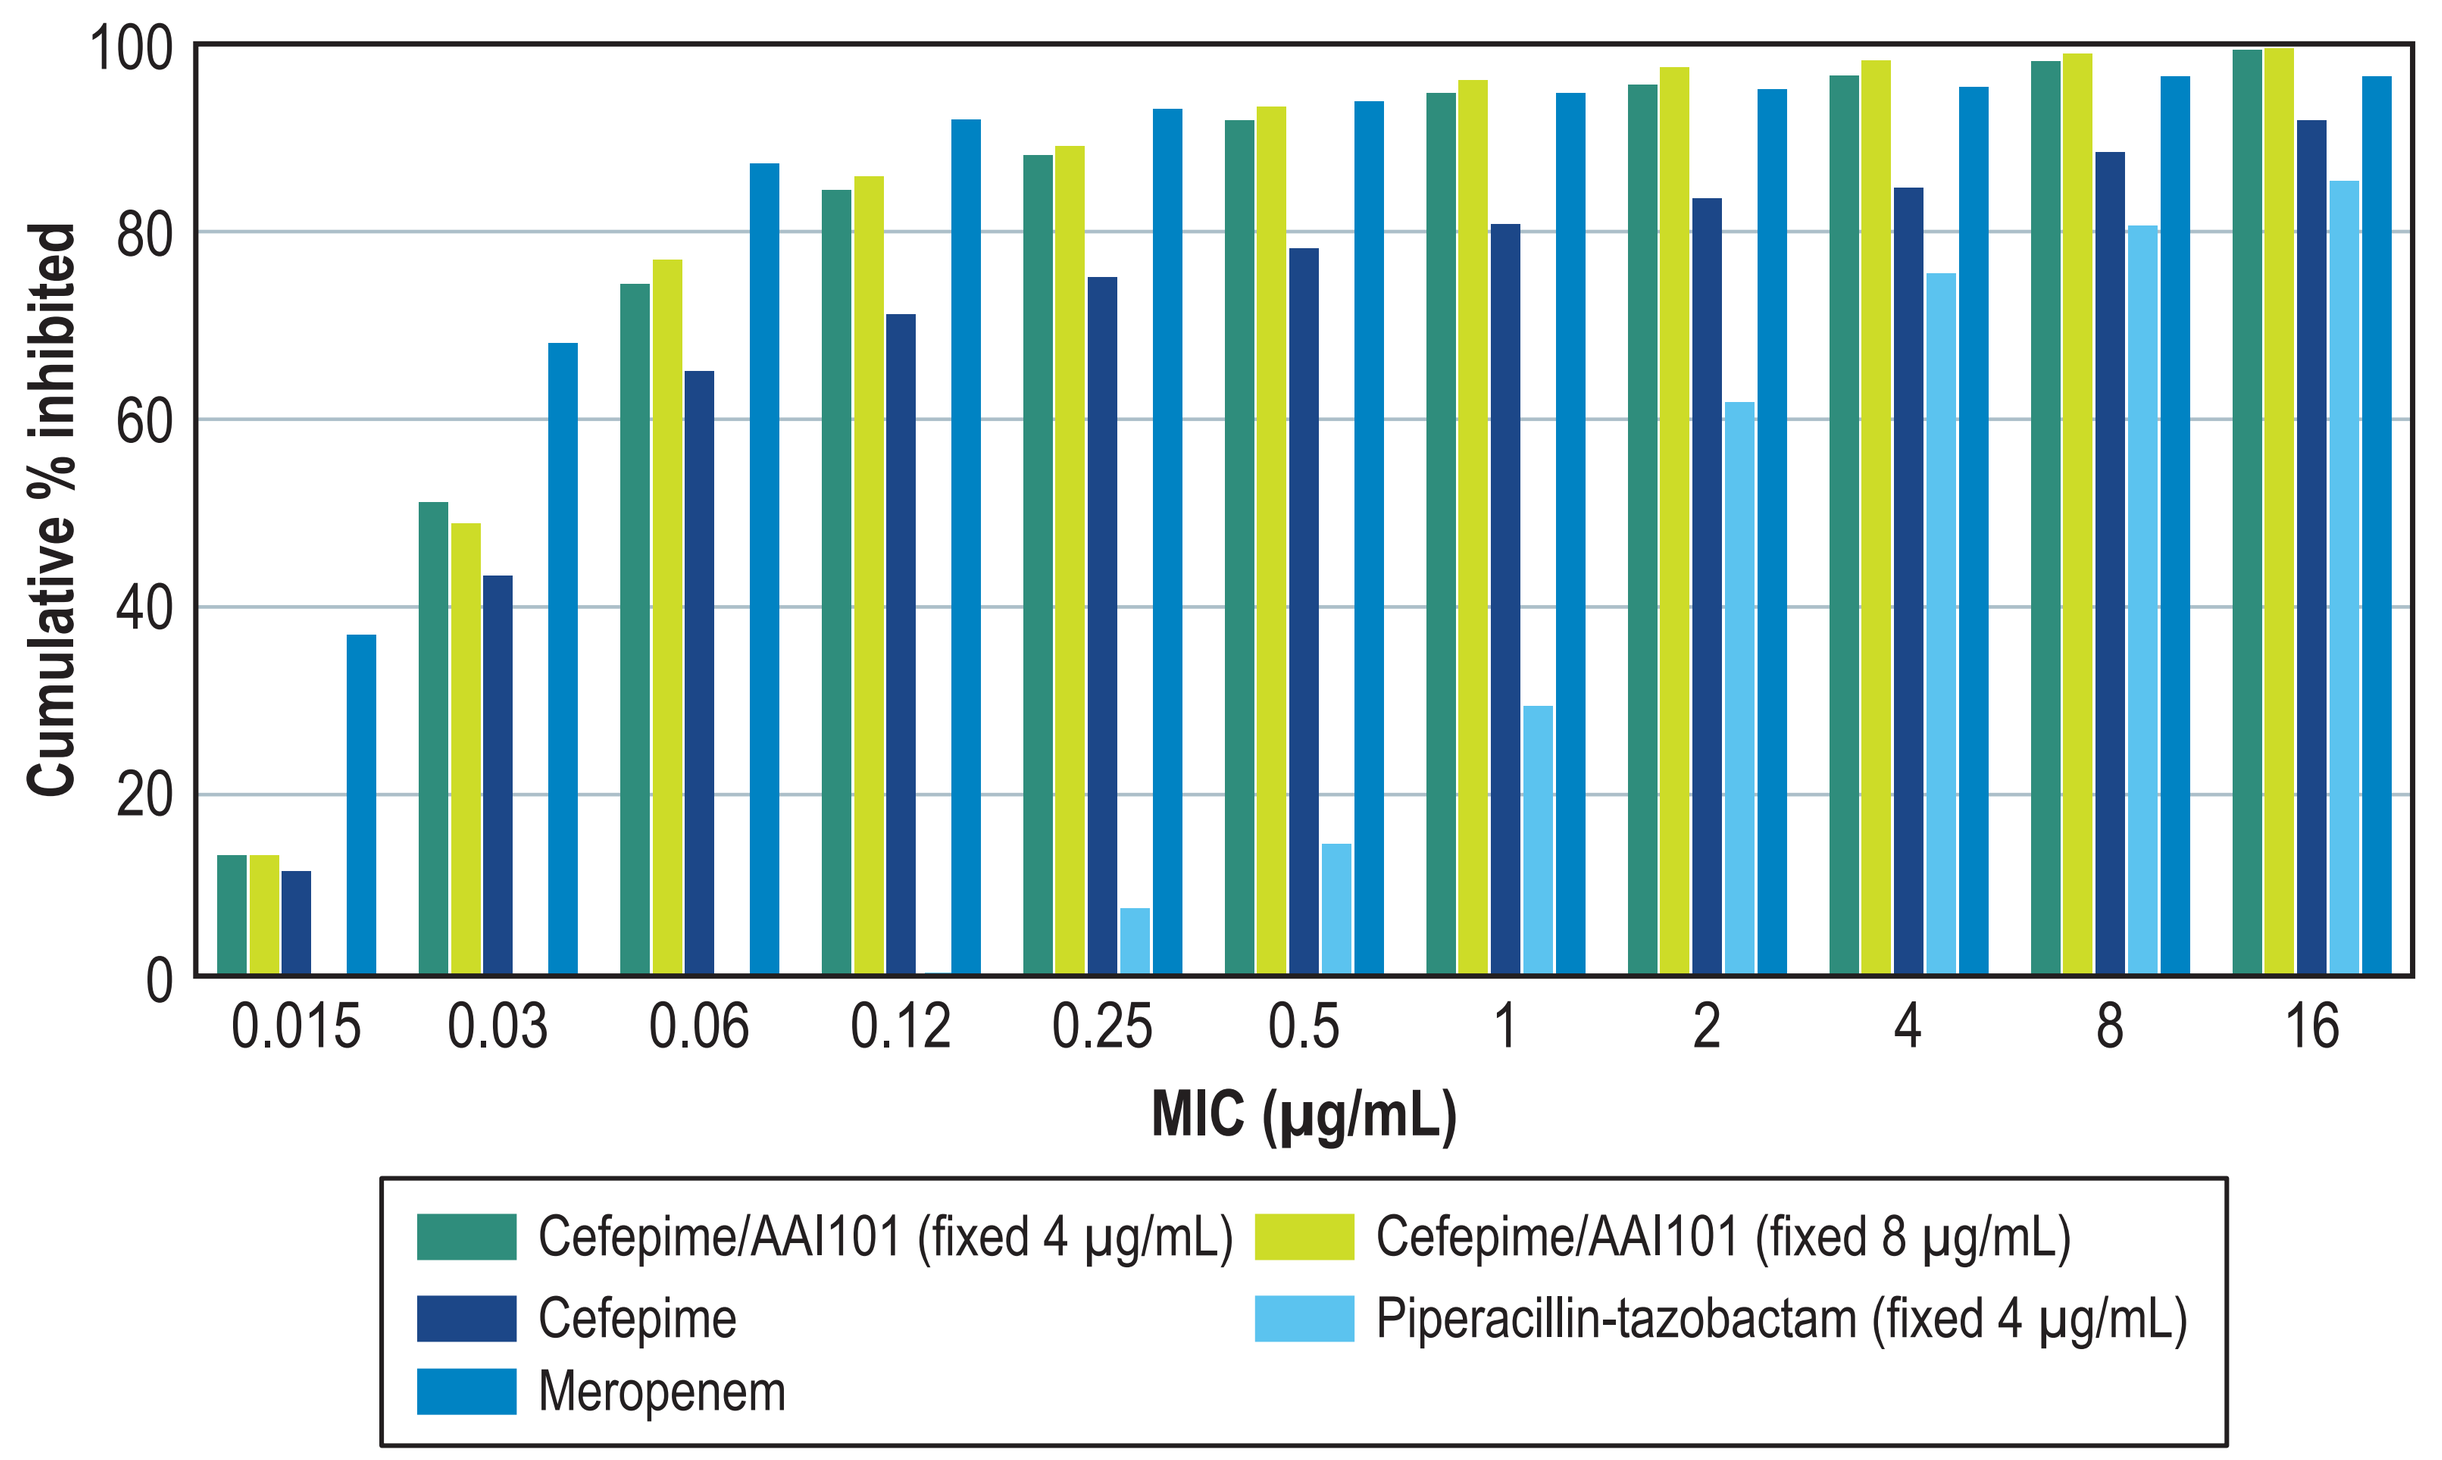

Cumulative % inhibited
MIC (µg/mL)
0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16
Cefepime/AAI101 (fixed 4 µg/mL)
Cefepime/AAI101 (fixed 8 µg/mL)
Cefepime
Piperacillin-tazobactam (fixed 4 µg/mL)
Meropenem

Figure 3 Cumulative percent inhibition results for cefepime/AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against 40 cefepime-resistant, carbapenem-susceptible *Enterobacteriaceae* isolates

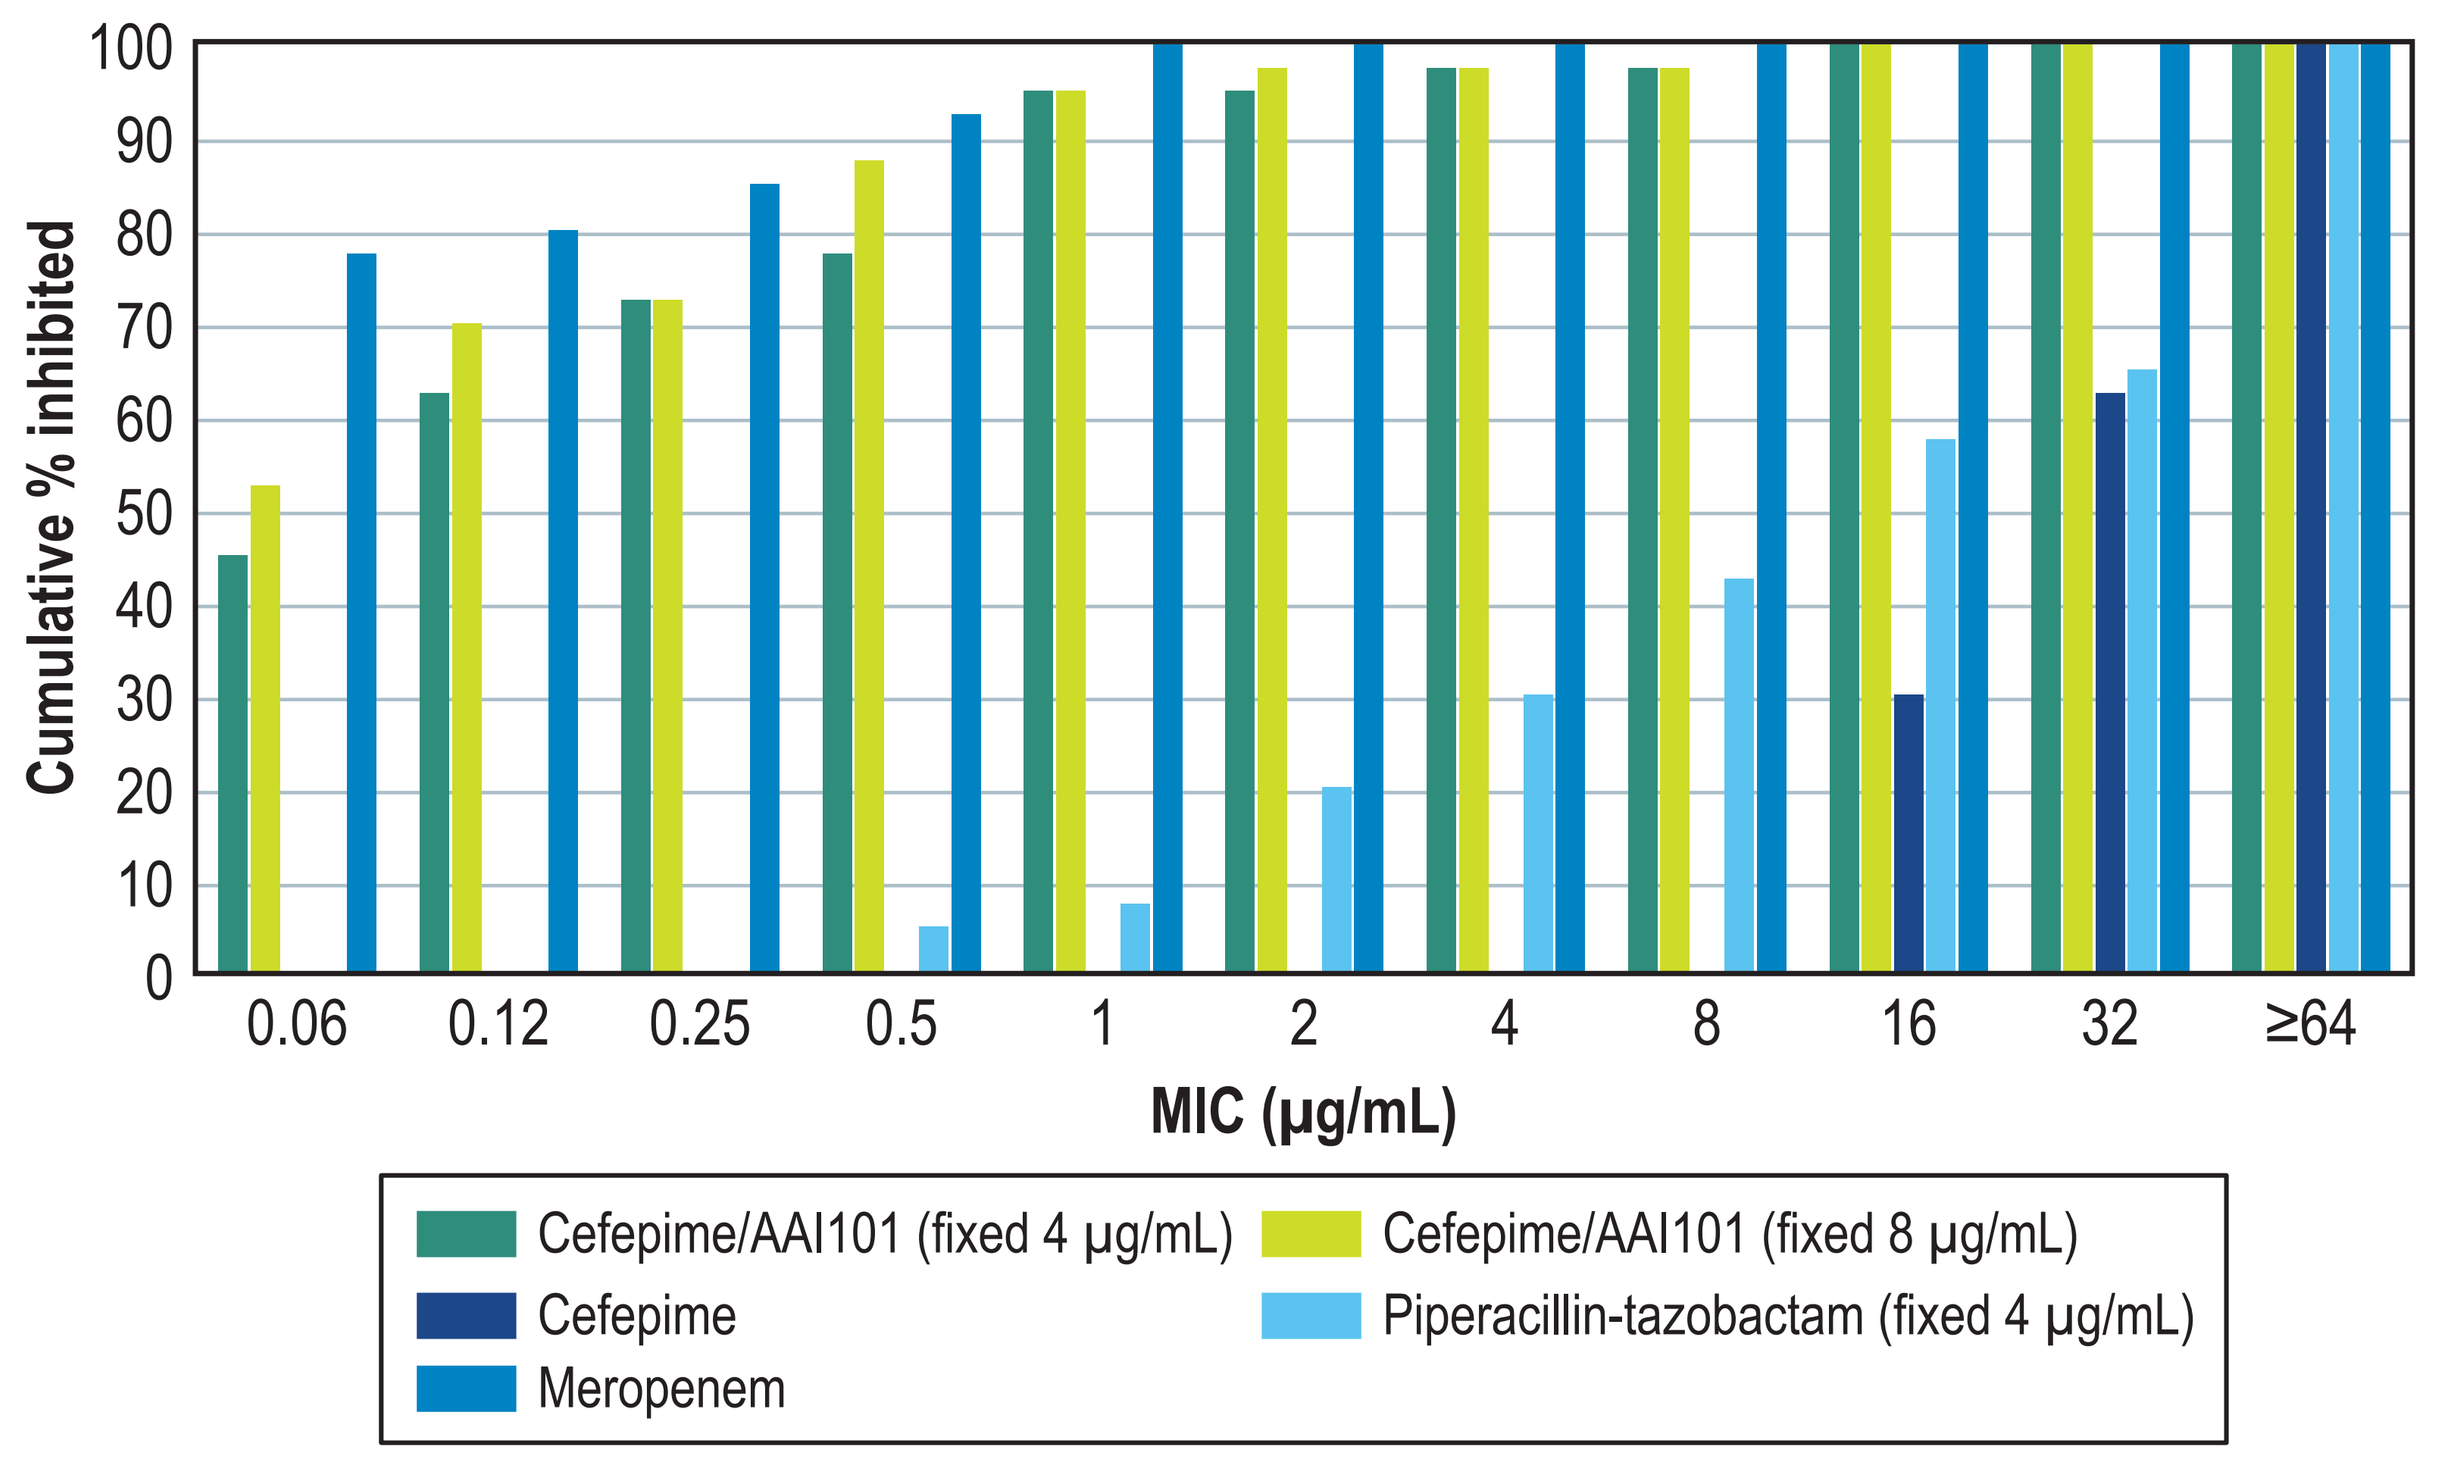

Cumulative % inhibited
MIC (µg/mL)
0.06 0.12 0.25 0.5 1 2 4 8 16 32 ≥64
Cefepime/AAI101 (fixed 4 µg/mL)
Cefepime/AAI101 (fixed 8 µg/mL)
Cefepime
Piperacillin-tazobactam (fixed 4 µg/mL)
Meropenem

Figure 4 Cumulative percent inhibition results for cefepime/AAI101 (fixed AAI101 concentrations of 4 µg/mL and 8 µg/mL) and comparators against 31 carbapenem-resistant *Enterobacteriaceae* isolates

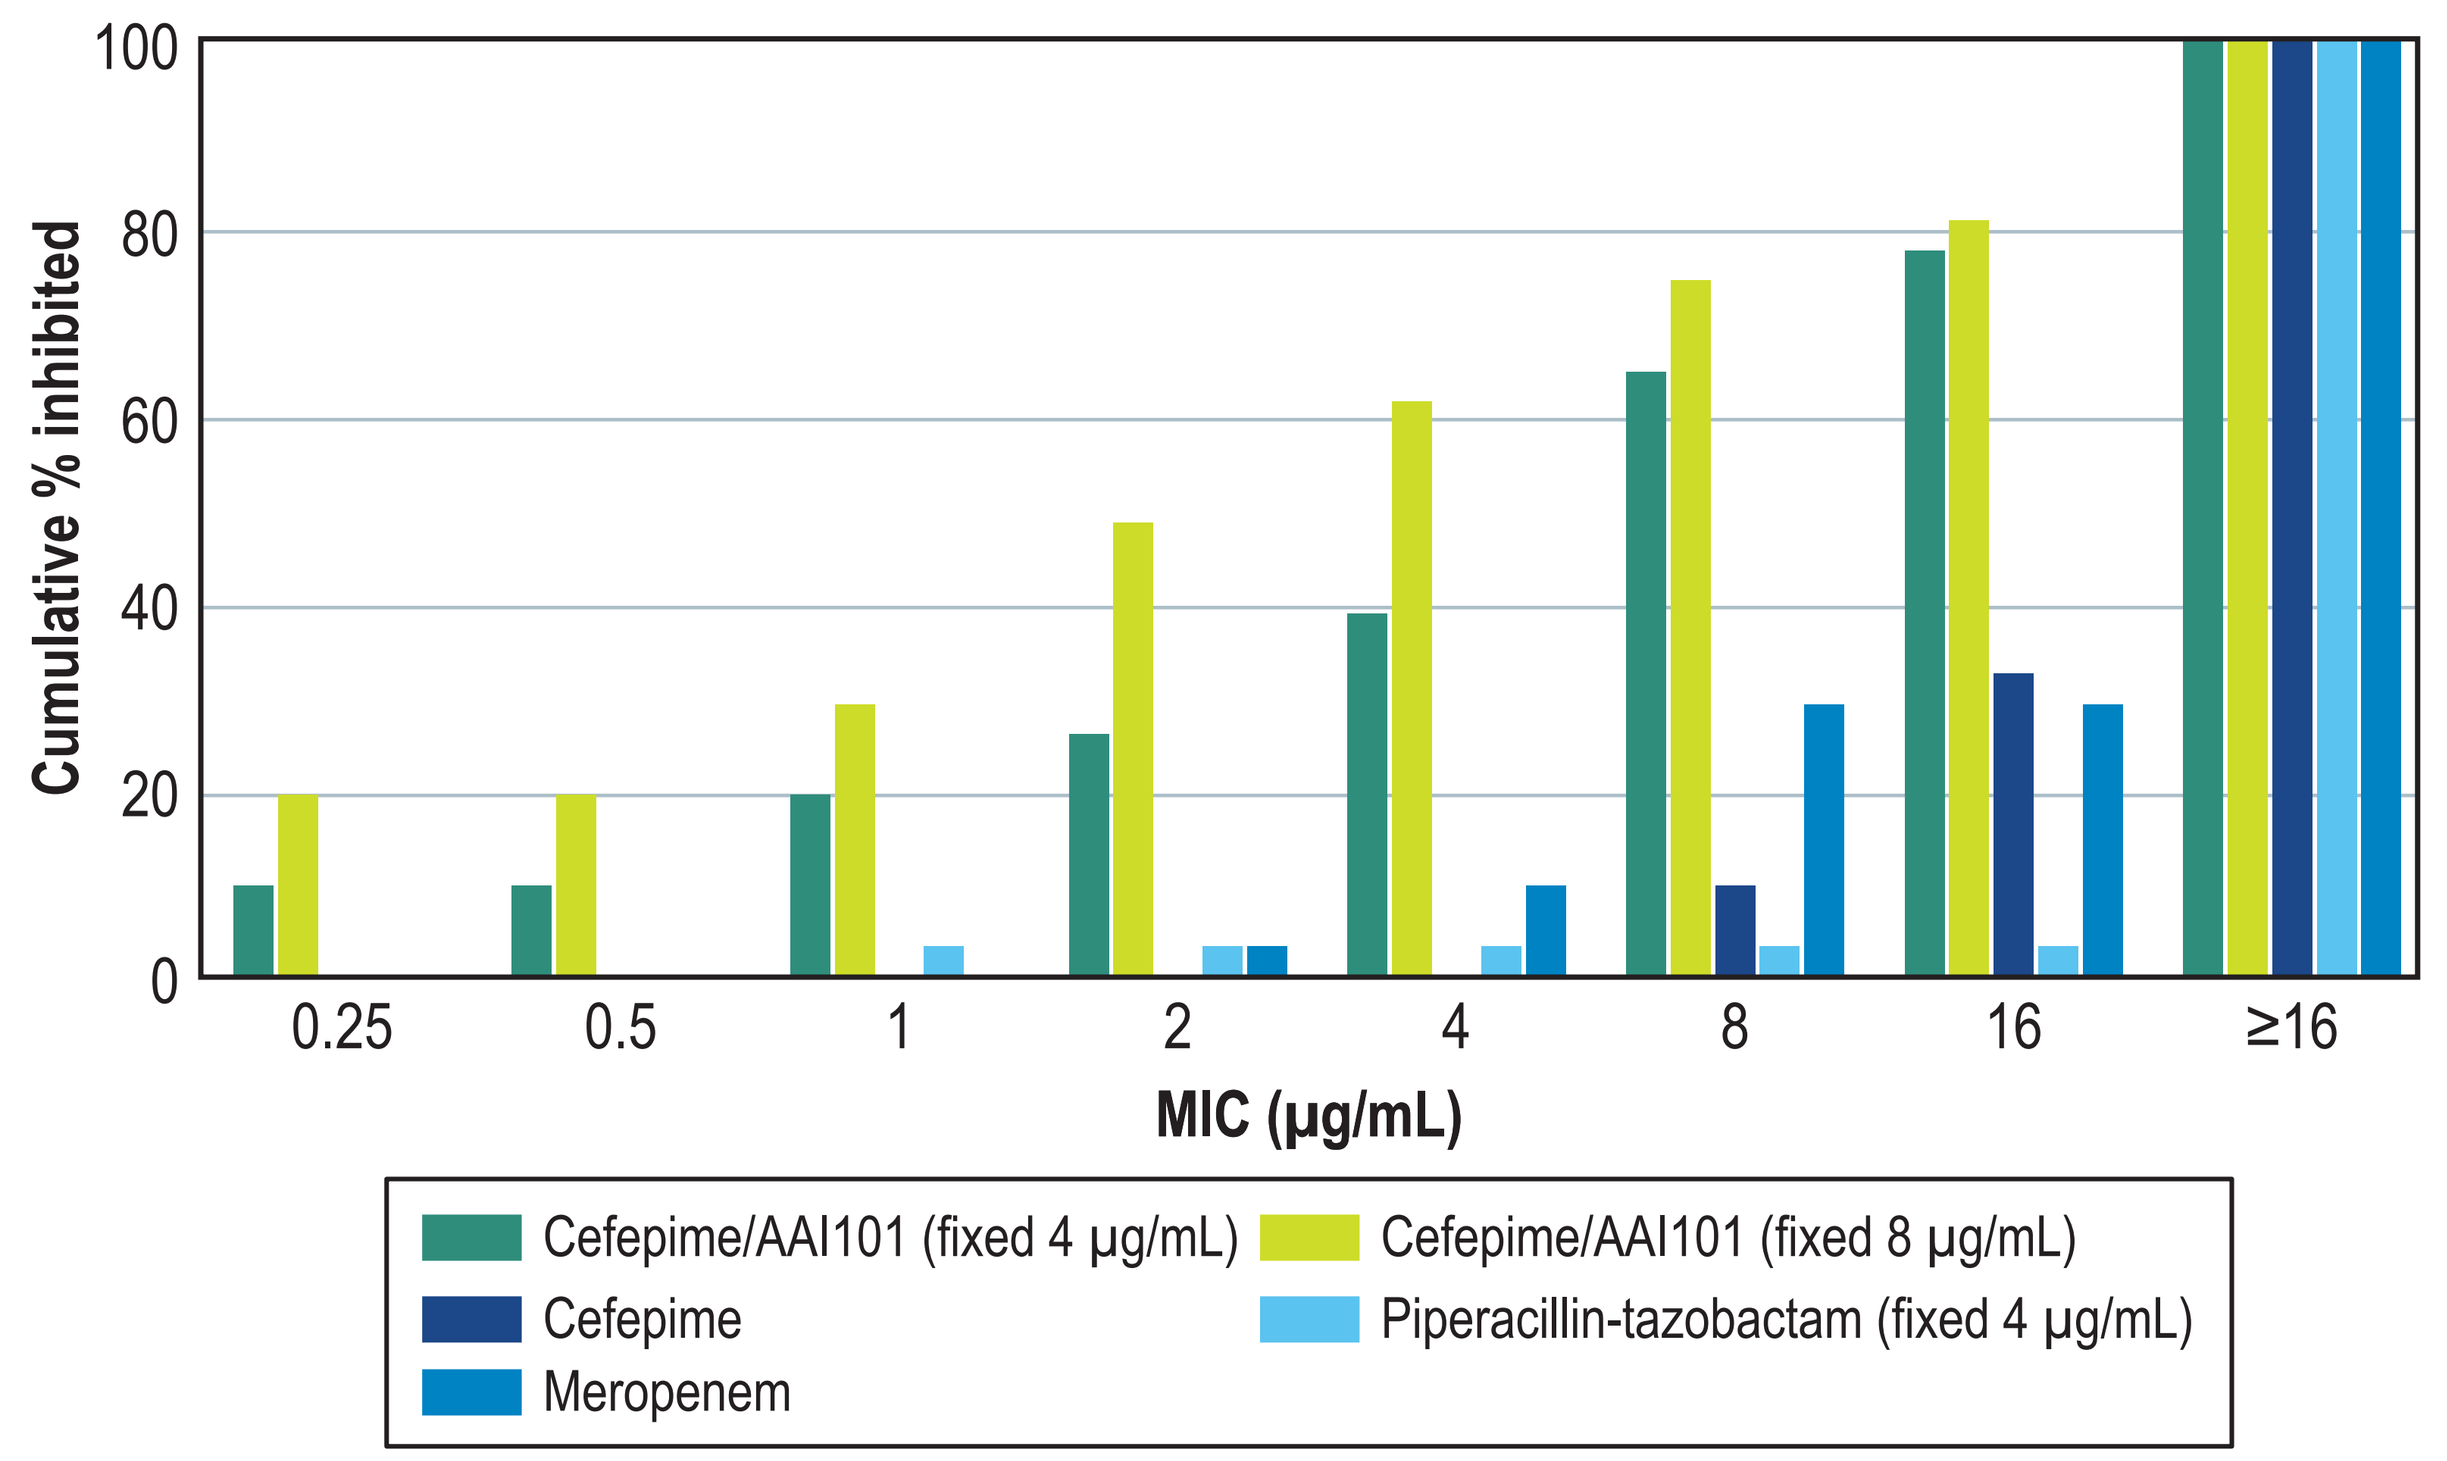

Cumulative % inhibited
MIC (µg/mL)
0.25 0.5 1 2 4 8 16 ≥16
Cefepime/AAI101 (fixed 4 µg/mL)
Cefepime/AAI101 (fixed 8 µg/mL)
Cefepime
Piperacillin-tazobactam (fixed 4 µg/mL)
Meropenem

- Against *Enterobacter* spp. cefepime-only MIC_{90} values were in the susceptible interpretive category for *E. aerogenes* and in the susceptible-dose-dependent category for *E. cloacae*. AAI101 reduced cefepime MIC_{90} values by ≥2 log_2 dilutions and restored cefepime MIC_{90} values to the susceptible interpretive category for both species (Table 2)
 - Piperacillin-tazobactam MIC_{90} values were in the intermediate interpretive category for *E. aerogenes* and resistant interpretive category for *E. cloacae* spp. complex isolates
 - Meropenem MIC_{90} values were in the susceptible interpretive category for both species
- Against *Citrobacter* spp., *K. oxytoca*, *M. morganii*, *Proteus* spp., *Providencia* spp., and *S. marcescens*, MIC_{90} values for cefepime-only, cefepime/AAI101, piperacillin-tazobactam, and meropenem were in the susceptible categories (Table 2)
 - AAI101 reduced cefepime MIC_{90} values by 4 log_2 dilutions for *Proteus* spp. Reductions of 1 to 2 log_2 dilutions by AAI101 were observed for *K. oxytoca*, *Providencia* spp., and *S. marcescens*

Table 2 Antimicrobial activity of cefepime/AAI101 and comparators against 572 recent *Enterobacteriaceae* isolates

Organism/group (n)	$MIC_{50/90}$ (µg/mL)/% susceptible (CLSI)				
	Cefepime	Cefepime/ AAI101 (fixed 4 µg/mL)[a]	Cefepime/ AAI101 (fixed 8 µg/mL)[a]	Piperacillin-tazobactam	Meropenem
Enterobacteriaceae (572)[b]	0.06 / 16 83.0%	0.03 / 0.5 95.1%	0.06 / 0.5 97.0%	2 / 128 84.8%	0.03 / 0.12 94.2%
Enterobacteriaceae cefepime-resistant, carbapenem-susceptible (40)[c]	32 / >128 0.0%	0.12 / 1 95.0%	0.06 / 1 97.5%	16 / >128 57.5%	0.03 / 0.5 100.0%
Enterobacteriaceae carbapenem-resistant (31)[d]	32 / >128 3.2%	8 / 32 25.8%	2 / 32 51.6%	>128 / >128 3.2%	>8 / >8 0.0%
Citrobacter spp. (42)[e]	0.03 / 0.25 97.6%	0.03 / 0.25 100.0%	0.03 / 0.25 100.0%	2 / 16 90.5%	0.015 / 0.03 100.0%
E. aerogenes (28)	0.03 / 0.5 96.4%	0.03 / 0.12 100.0%	0.03 / 0.12 100.0%	2 / 64 82.1%	0.03 / 0.06 100.0%
E. cloacae (77)	0.06 / 4 88.3%	0.06 / 0.5 98.7%	0.06 / 0.5 100%	2 / 128 77.9%	0.03 / 0.12 100.0%
E. coli (121)	0.06 / 32 72.7%	0.03 / 0.12 98.3%	0.03 / 0.12 98.3%	2 / 64 88.4%	0.015 / 0.06 95.9%
K. pneumoniae (135)	0.06 / >128 65.2%	0.06 / 8 83.0%	0.06 / 2 90.4%	4 / >128 66.7%	0.03 / >8 80.0%
K. oxytoca (27)	0.03 / 0.12 100.0%	0.03 / 0.06 100.0%	0.03 / 0.06 100.0%	2 / 8 96.3%	0.015 / 0.03 100.0%
M. morganii (27)	0.03 / 0.06 100.0%	0.03 / 0.06 100.0%	0.03 / 0.06 100.0%	0.5 / 1 100.0%	0.06 / 0.12 100.0%
Proteus spp. (48)[f]	0.06 / 2 91.7%	0.06 / 0.12 97.9%	0.06 / 0.12 97.9%	0.5 / 1 100.0%	0.06 / 0.12 97.9%
Providencia spp. (42)[g]	0.03 / 0.5 97.6%	0.03 / 0.25 97.6%	0.03 / 0.12 97.6%	1 / 8 97.6%	0.06 / 0.06 100.0%
S. marcescens (25)	0.06 / 1 92.0%	0.06 / 0.5 96.0%	0.06 / 0.5 96.0%	2 / 8 96.0%	0.06 / 0.12 96.0%

[a] Cefepime breakpoint interpretive criteria were applied to cefepime/AAI101 combinations for comparison purposes only.
[b] Comprised of 21 *Citrobacter freundii* species complex, 21 *C. koseri*, 28 *Enterobacter aerogenes*, 77 *E. cloacae* species complex, 121 *Escherichia coli*, 27 *Klebsiella oxytoca*, 135 *K. pneumoniae*, 27 *Morganella morganii*, 27 *Proteus mirabilis*, 21 *P. vulgaris* group, 21 *Providencia rettgeri*, 21 *P. stuartii*, and 25 *Serratia marcescens* isolates. The cefepime- and carbapenem-resistant isolates represent subsets of the 572 *Enterobacteriaceae* surveyed.
[c] Comprised of 1 *E. aerogenes*, 6 *E. cloacae* spp. complex, 16 *E. coli*, 14 *K. pneumoniae*, 2 *P. mirabilis*, and 1 *P. stuartii* isolates.
[d] Comprised of 4 *E. coli*, 26 *K. pneumoniae*, and 1 *P. mirabilis* isolates.
[e] Comprised of 21 *C. koseri* and 21 *C. freundii* species complex isolates.
[f] Comprised of 27 *P. mirabilis* and 21 *P. vulgaris* group isolates.
[g] Comprised of 21 *P. rettgeri* and 21 *P. stuartii* isolates.

Conclusions

- AAI101, a novel β-lactamase inhibitor with potent activity against ESBLs and a variety of other β-lactamases, restored the activity of cefepime against a panel of recent *Enterobacteriaceae* collected primarily in 2016 from US and European medical centers
- Activity of AAI101, when combined with cefepime, was most prominent against *K. pneumoniae* and *E. coli*, followed by *E. cloacae* and *Proteus* spp.
- Cefepime/AAI101 was as effective as meropenem and outperformed cefepime and piperacillin-tazobactam against this collection of isolates
- Against *Citrobacter* spp., *K. oxytoca*, *M. morganii*, *Providencia* spp., and *S. marcescens*, cefepime-only demonstrated potent activity
- Cefepime/AAI101 warrants further clinical investigation to evaluate its potential as a therapeutic option for difficult-to-treat gram-negative pathogens
- Cefepime/AAI101 may be a carbapenem-sparing option for settings with increasing resistance to piperacillin-tazobactam

Acknowledgements

This study and poster presentation were funded by a grant from Allecra Therapeutics SAS (Saint-Louis, France).

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07-A11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - eleventh edition*. Wayne, PA: CLSI.