Antimicrobial Susceptibility of *Pseudomonas aeruginosa* to Ceftazidime-Avibactam, Ceftolozane-Tazobactam, Piperacillin-Tazobactam, and Meropenem Stratified by United States Census Divisions: Results from the 2017 INFORM Program

ASM Microbe 2018
Sunday - 388

HS Sader, RK Flamm, M Castanheira

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- *Pseudomonas aeruginosa* represents a major cause of nosocomial infections worldwide, including sepsis, hospital-acquired pneumonia, ventilator-associated pneumonia (VAP), skin and skin structure infections (SSSIs), and urinary tract infections (UTIs)
- The increasing prevalence of multidrug-resistant (MDR) and extensively drug-resistant (XDR) strains of *P. aeruginosa* is a cause of great concern as it makes the selection of appropriate empirical and definitive antimicrobial treatments very difficult
- Antipseudomonal β-lactam agents play an important role in empirically treating *P. aeruginosa* infections
- Resistance mechanisms against these antimicrobial agents include production of intrinsic or acquired β-lactamases, hyperexpression of efflux systems, and the decrease or loss of the outer membrane protein OprD
- Ceftazidime-avibactam and ceftolozane-tazobactam are the most recently US FDA-approved cephalosporin/β-lactamase inhibitor combinations for treating infections caused by gram-negative bacilli, including *P. aeruginosa*
- We evaluated the *in vitro* activity of ceftazidime-avibactam, ceftolozane-tazobactam, and many comparator agents against a large collection of recent clinical PSA isolates from United States medical centers

MATERIALS AND METHODS

Organism collection

- A total of 1,909 *P. aeruginosa* isolates (1 per patient episode) were consecutively collected from 70 US medical centers (35 states from all 9 census divisions) in 2017 as part of the International Network for Optimal Resistance Monitoring (INFORM) program
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of an infection were included in this investigation, and the results were stratified by US census division
- Species identification was confirmed when necessary by MALDI-TOF MS using the Bruker Daltonics MALDI Biotyper (Billerica, Massachusetts, USA) by following manufacturer instructions
- Isolates were categorized as MDR, XDR, or pan drug-resistant (PDR) according to criteria published by Magiorakos *et al.* (2012) that state MDR is nonsusceptible (NS) to ≥1 agent in ≥3 antimicrobial classes, XDR is NS to ≥1 agent in all but ≤2 antimicrobial classes, and PDR is NS to all antimicrobial classes tested

Antimicrobial susceptibility testing

- All isolates were tested for susceptibility using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI)
- Ceftazidime-avibactam and ceftolozane-tazobactam were tested with the β-lactamase inhibitor at a fixed concentration of 4 μg/mL
- Susceptibility interpretations for comparator agents were those found in CLSI document M100 and/or the US FDA package insert
- Quality control (QC) was performed using *Escherichia coli* ATCC 25922 and 35218, *Klebsiella pneumoniae* ATCC 700603 and BAA 1705, and *P. aeruginosa* ATCC 27853

RESULTS

- Ceftazidime-avibactam ($MIC_{50/90}$, 2/8 μg/mL) and ceftolozane-tazobactam ($MIC_{50/90}$, 0.5/2 μg/mL) were the most active compounds after colistin with national susceptibility rates of 96.9% and 97.5%, respectively (Table 1)
- Colistin was active against 99.9% of *P. aeruginosa* isolates overall (Table 1)
- National susceptibility rates for piperacillin-tazobactam ($MIC_{50/90}$, 4/128 μg/mL) and meropenem ($MIC_{50/90}$, 0.5/16 μg/mL) were 77.5% and 76.0%, respectively; 94.8% and 93.1% of isolates were susceptible (S) to amikacin ($MIC_{50/90}$, 4/16 μg/mL) and tobramycin ($MIC_{50/90}$, 0.5/2 μg/mL), respectively (Table 1)
- Ceftazidime-avibactam and ceftolozane-tazobactam were the most active compounds after colistin in all census divisions and had susceptibility rates of 93.3%–99.4% and 92.9%–99.6%, respectively (Table 2)
- Susceptibility rates for piperacillin-tazobactam, meropenem, and tobramycin across census divisions were 70.0%–85.4%, 65.0%–84.2%, and 89.8%–96.8%, respectively (Table 2)
- MDR rates varied from 18.6% (East South Central) to 32.7% (West South Central), and XDR rates varied from 6.8% (South Atlantic) to 19.9% (West South Central; Figure 1)
- Overall, ceftazidime-avibactam and ceftolozane-tazobactam were active against 88.6% and 91.4% of MDR and 80.2% and 83.8% of XDR isolates, respectively (Figure 2)

Table 1 Antimicrobial susceptibility of 1,909 *Pseudomonas aeruginosa* clinical isolates from US medical centers (INFORM program, 2017)

Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %I	CLSI[a] %R
Ceftazidime-avibactam	2	8	96.9		3.1
Ceftolozane-tazobactam	0.5	2	97.5	1.2	1.3
Piperacillin-tazobactam	4	128	77.5	10.3	12.2
Ceftazidime	2	32	82.5	4.3	13.2
Cefepime	4	16	82.4	11.1	6.5
Meropenem	0.5	16	76.0	7.0	17.0
Doripenem	0.5	>8	77.4	6.0	16.6
Imipenem	1	>8	75.7	4.2	20.1
Ciprofloxacin	0.25	>4	77.9	5.9	16.2
Levofloxacin	1	16	72.1	9.0	18.9
Gentamicin	2	8	81.7	9.6	8.7
Amikacin	4	16	94.8	2.5	2.7
Tobramycin	0.5	2	93.1	1.5	5.3
Colistin	0.5	1	99.9		0.1

[a] Criteria as published by CLSI 2018.

Table 2 Antimicrobial susceptibility of 1,909 *Pseudomonas aeruginosa* clinical isolates stratified by US census division (INFORM program, 2017)

Antimicrobial agent	% susceptible by US census division (no. of isolates): New England (157)	Middle Atlantic (334)	East North Central (342)	West North Central (240)	South Atlantic (219)	East South Central (156)	West South Central (156)	Mountain (80)	Pacific (225)
Ceftazidime-avibactam	98.7	96.1	98.0	97.9	97.7	99.4	95.5	95.0	93.3
Ceftolozane-tazobactam	97.5	97.6	98.0	99.6	98.6	98.7	98.1	96.2	92.9
Piperacillin-tazobactam	83.4	71.0	79.5	85.4	78.5	81.4	73.7	70.0	73.3
Ceftazidime	89.2	76.3	86.0	88.3	85.8	89.1	75.3	72.5	76.0
Cefepime	82.8	79.3	86.3	83.3	85.8	87.2	76.3	78.8	78.7
Meropenem	82.2	65.0	78.9	84.2	81.7	82.7	67.3	76.2	70.7
Ciprofloxacin	70.1	77.8	80.1	78.3	80.8	80.8	76.3	77.5	76.0
Levofloxacin	64.3	70.6	76.0	75.0	72.1	75.0	67.3	73.8	71.1
Amikacin	94.9	96.1	95.3	93.8	96.8	94.9	94.9	93.8	91.6
Tobramycin	89.8	93.7	93.3	95.0	96.8	92.9	90.4	90.0	92.0
Colistin	100.0	100.0	99.7	100.0	100.0	100.0	100.0	100.0	100.0

Figure 1 Frequency of multidrug-resistant (MDR) and extensively drug-resistant (XDR) *P. aeruginosa* clinical isolates stratified by US census division

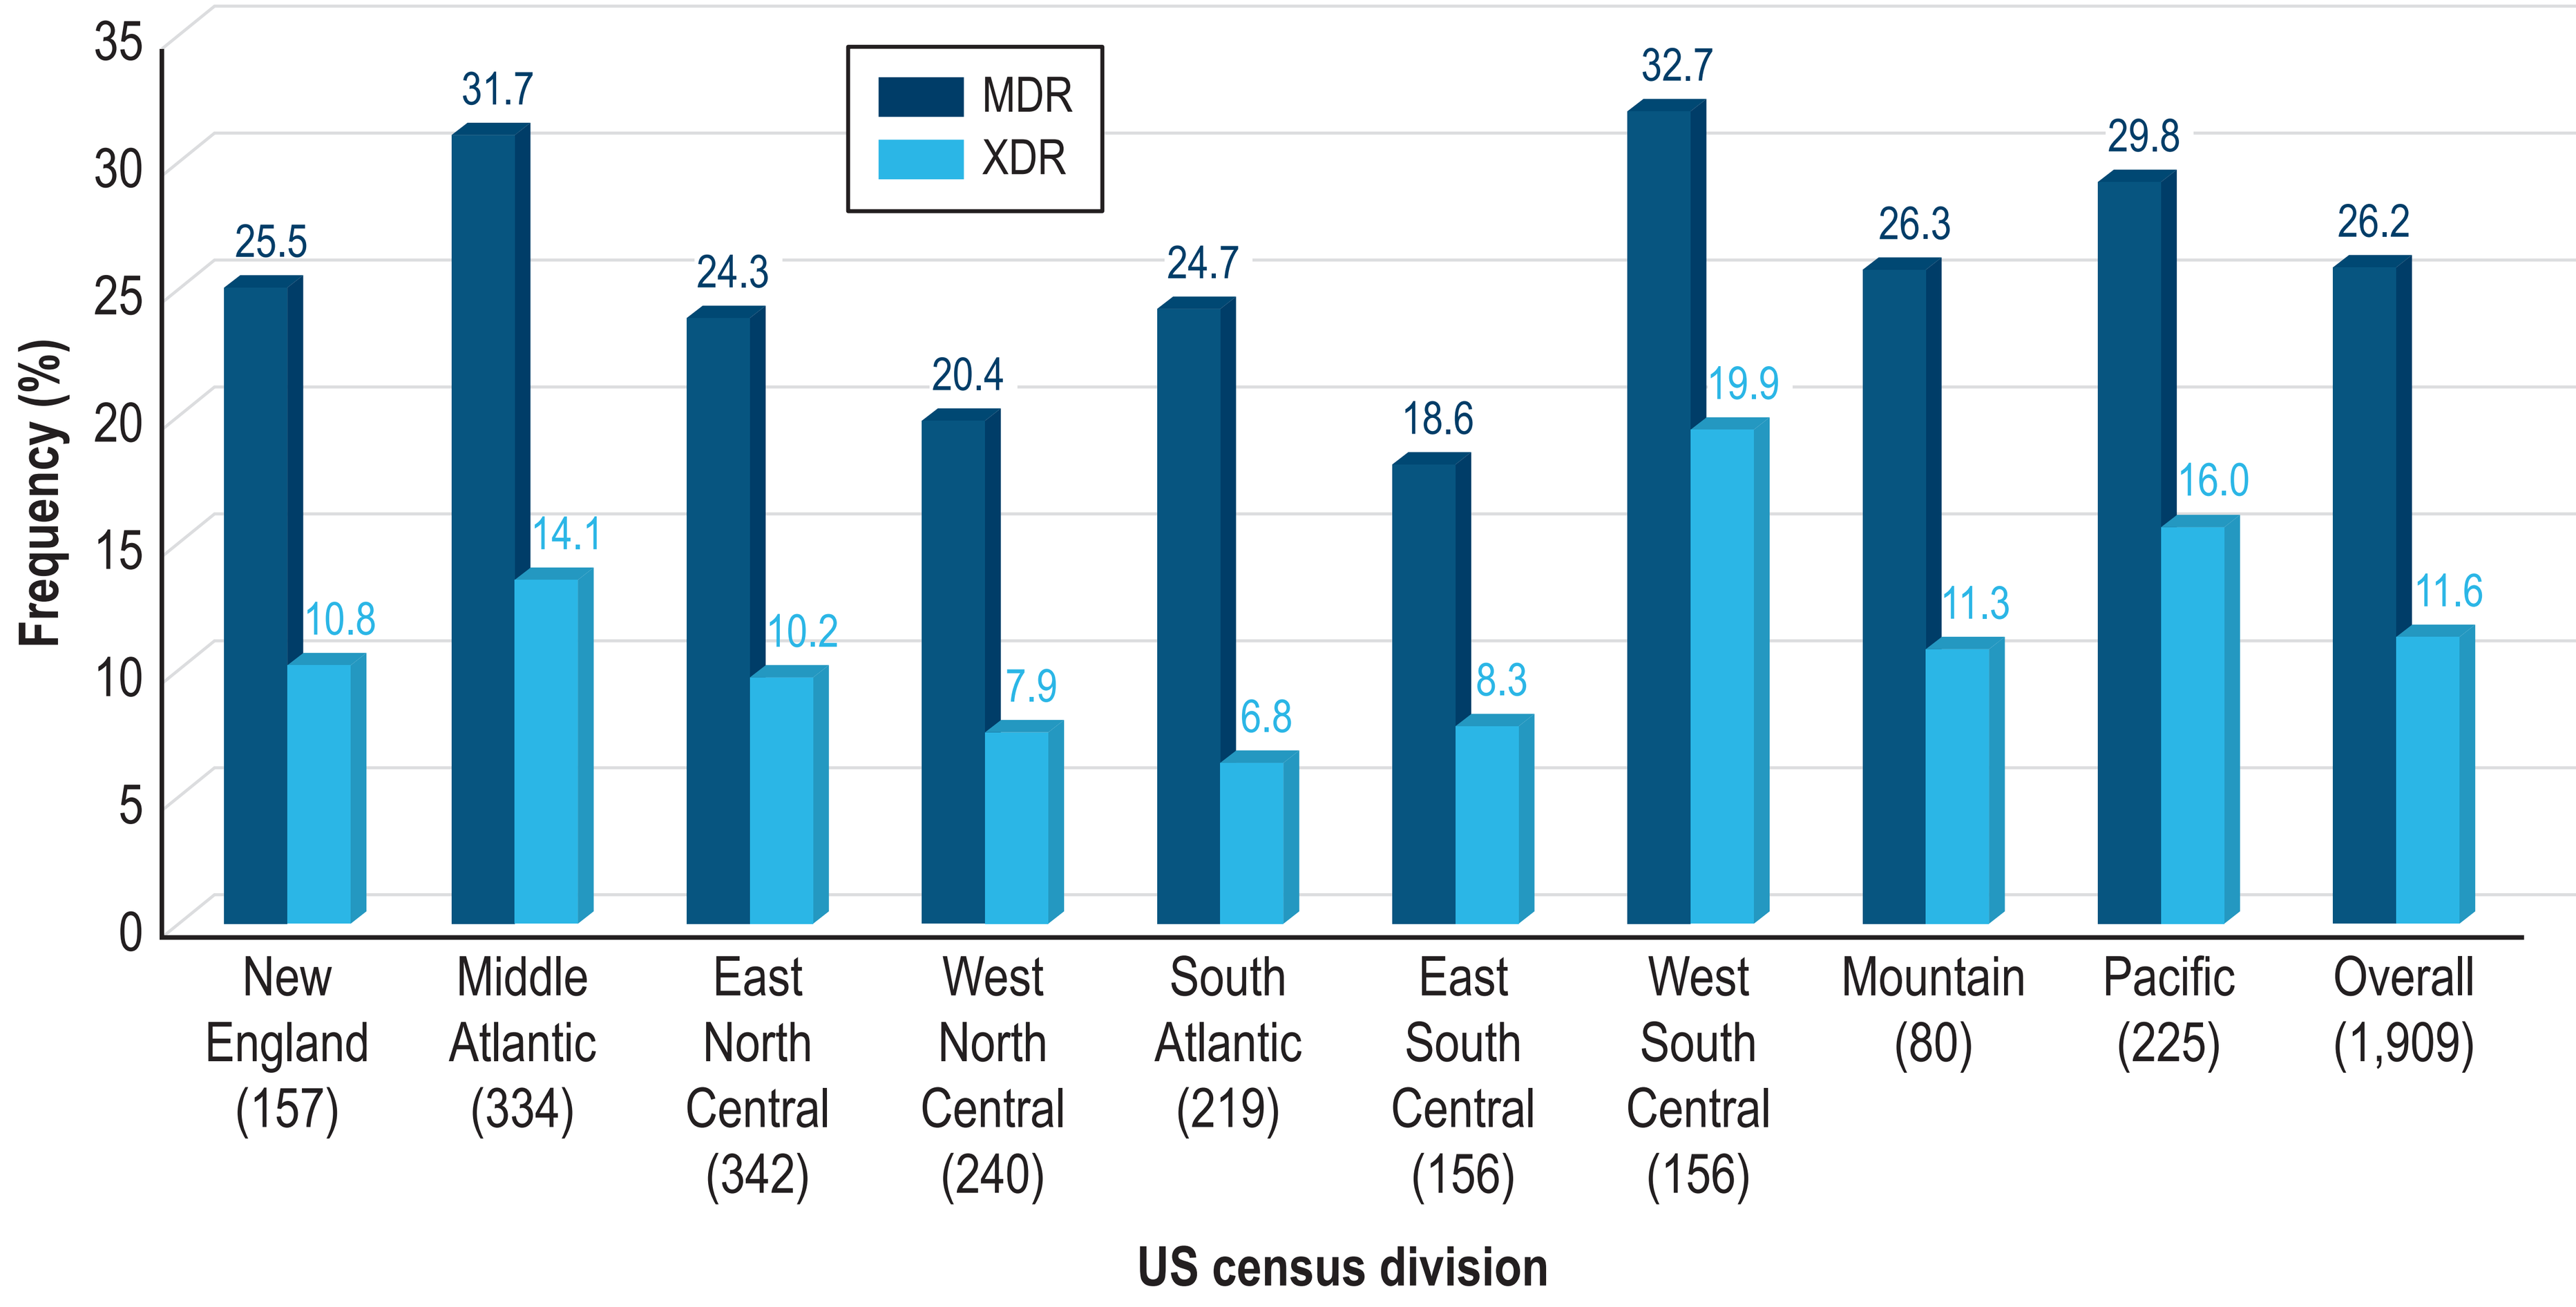
MDR
XDR
Frequency (%)
35
30
25
20
15
10
5
0
25.5
10.8
31.7
14.1
24.3
10.2
20.4
7.9
24.7
6.8
18.6
8.3
32.7
19.9
26.3
11.3
29.8
16.0
26.2
11.6
New England (157)
Middle Atlantic (334)
East North Central (342)
West North Central (240)
South Atlantic (219)
East South Central (156)
West South Central (156)
Mountain (80)
Pacific (225)
Overall (1,909)
US census division

Figure 2 Antimicrobial susceptibility of multidrug-resistant (MDR) and extensively drug-resistant (XDR) *P. aeruginosa* clinical isolates

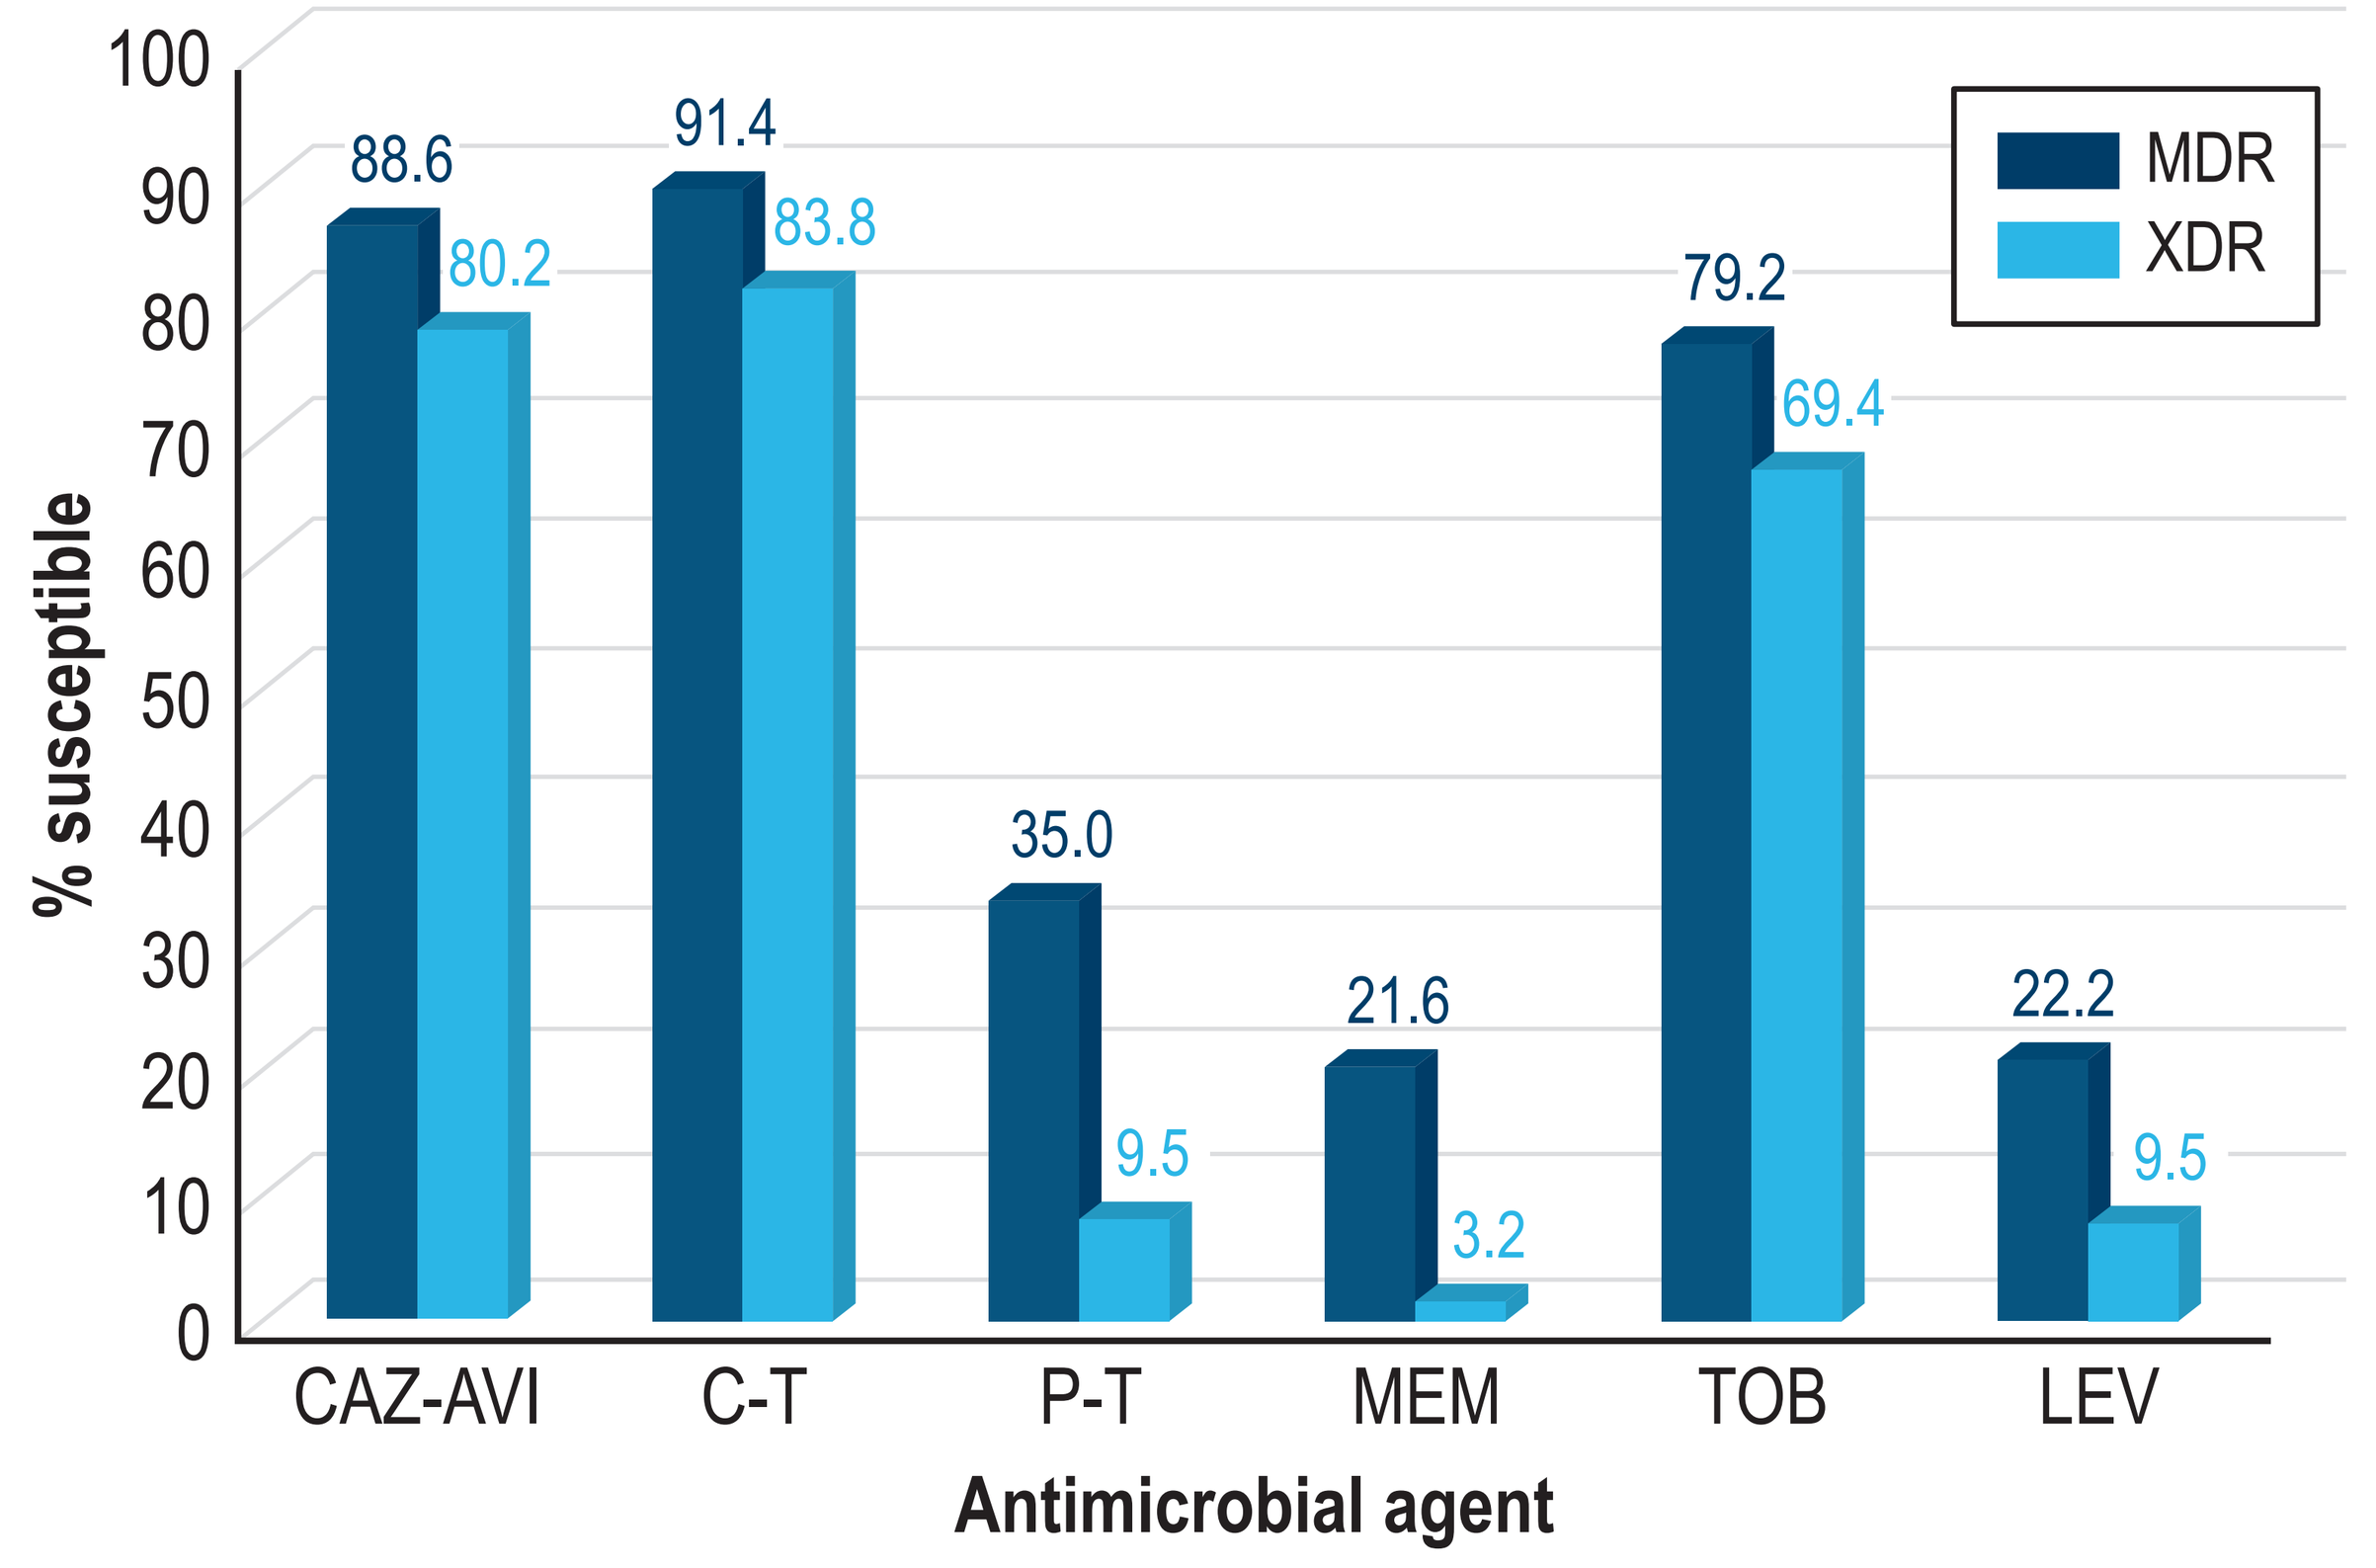
MDR
XDR
% susceptible
100
90
80
70
60
50
40
30
20
10
0
88.6
80.2
91.4
83.8
35.0
9.5
21.6
3.2
79.2
69.4
22.2
9.5
CAZ-AVI
C-T
P-T
MEM
TOB
LEV
Antimicrobial agent

Abbreviations: CAZ-AVI, ceftazidime-avibactam; C-T, ceftolozane-tazobactam; P-T, piperacillin-tazobactam; MEM, meropenem; TOB, tobramycin; and LEV, levofloxacin.

CONCLUSIONS

- Ceftazidime-avibactam and ceftolozane-tazobactam exhibited similar coverage against *P. aeruginosa* regardless of US census division or resistance phenotype
- Ceftazidime-avibactam and ceftolozane-tazobactam demonstrated potent *in vitro* activity against *P. aeruginosa*, including MDR and XDR isolates
- Variations in the susceptibility patterns of *P. aeruginosa* isolates across census divisions were documented for many antimicrobials

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the INFORM program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com /assets/pdf/avycaz_pi. Accessed January 2017.
2. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition.* Wayne, PA: CLSI.
4. Falcone M, Paterson D (2016). Spotlight on ceftazidime/avibactam: a new option for MDR Gram-negative infections. *J Antimicrob Chemother* 71: 2713–2722.
5. Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268–281.
6. Sader HS, Huband MD, Castanheira M, et al. (2017). *Pseudomonas aeruginosa* antimicrobial susceptibility results from four years (2012–2015) of the International Network for Optimal Resistance Monitoring program in the United States. *Antimicrob Agents Chemother* 61: e02252.

Visit www.allergancongressposters .com/634679

Allergan

ASM-Microbe 2018, June 7–11, 2018 in Atlanta, Georgia