Sunday-392

Antimicrobial Activity of Ceftolozane-Tazobactam Tested against Contemporary (2015–2017) Gram-Negative Isolates from Intensive Care Unit Patients with Pneumonia in US Medical Centers

Dee Shortridge, Michael A. Pfaller, Leonard R. Duncan, Jennifer M. Streit, Robert K. Flamm
JMI Laboratories, North Liberty, Iowa, USA

INTRODUCTION

- Ceftolozane-tazobactam is a combination of an antipseudomonal cephalosporin and a β-lactamase inhibitor
 - Ceftolozane-tazobactam has activity against isolates with the most common β-lactam resistance mechanisms employed by *Pseudomonas aeruginosa,* including AmpC production, up-regulated efflux pumps, and porin reductions
 - Ceftolozane-tazobactam also has activity against most extended-spectrum beta-lactamase (ESBL)-producing *Enterobacteriaceae* (new taxonomy: Enterobacterales)
- Ceftolozane-tazobactam has been approved in >50 countries for treatment of adults with complicated urinary tract infections, including acute pyelonephritis, and complicated intra-abdominal infections in combination with metronidazole
 - Clinical trials in hospital-acquired bacterial pneumonia/ventilator-associated bacterial pneumonia are in progress (clinicaltrials.gov Identifier: NCT02070757)
- The Program to Assess Ceftolozane-Tazobactam Susceptibility (PACTS) monitors susceptibility of ceftolozane-tazobactam among gram-negative isolates worldwide
- In the current study, gram-negative isolates were collected from pneumonia in hospitalized patients in the intensive care unit (ICU) in the US from 2015–2017

MATERIALS AND METHODS

- A total of 1,951 gram-negative isolates, including 1,113 *Enterobacteriaceae* and 571 *P. aeruginosa*, were collected from pneumonia in hospitalized patients during 2015-2017 from 29 US hospital ICUs and tested for C-T susceptibility by CLSI broth microdilution at JMI Laboratories
 - Only 1 isolate per patient per infection episode was included
- Other antibiotics tested were amikacin, cefepime, ceftazidime, colistin, levofloxacin, meropenem, and piperacillin-tazobactam
 - Ceftolozane-tazobactam and piperacillin-tazobactam were tested with a fixed 4 mg/L concentration of tazobactam
 - CLSI (2018) and EUCAST (2018) breakpoints were applied; EUCAST breakpoints were used for colistin against *Enterobacteriaceae*
- Resistant phenotypes included *Enterobacteriaceae* resistant to doripenem, imipenem, or meropenem (carbapenem-resistant *Enterobacteriaceae*, CRE) and ESBL-screen-postive phenoytpe isolates, not resistant to carbapenems (ESBL, non-CRE)
 - ESBL and CRE phenotypes were based on CLSI M100 criteria.
- For *P. aeruginosa*, the phenotypes analyzed were ceftazidime-NS, meropenem-NS, cefepime-NS, piperacillin-tazobactam-NS, and NS to all 4 β-lactams (BL-NS)

RESULTS

- The top 3 species isolated from ICU patients hospitalized with pneumonia were *P. aeruginosa* (n=571), *Klebsiella pneumoniae* (n=296), and *Escherichia coli* (n=216)
- The susceptibilities of ceftolozane-tazobactam and comparators for the top enteric pathogens are shown in Tables 1 and 2
- Ceftolozane-tazobactam MIC distributions are shown in Table 3

Figure 1 Susceptibility of ceftolozane-tazobactam and β-lactam comparators against all *P. aeruginosa* isolates and resistance phenotypes

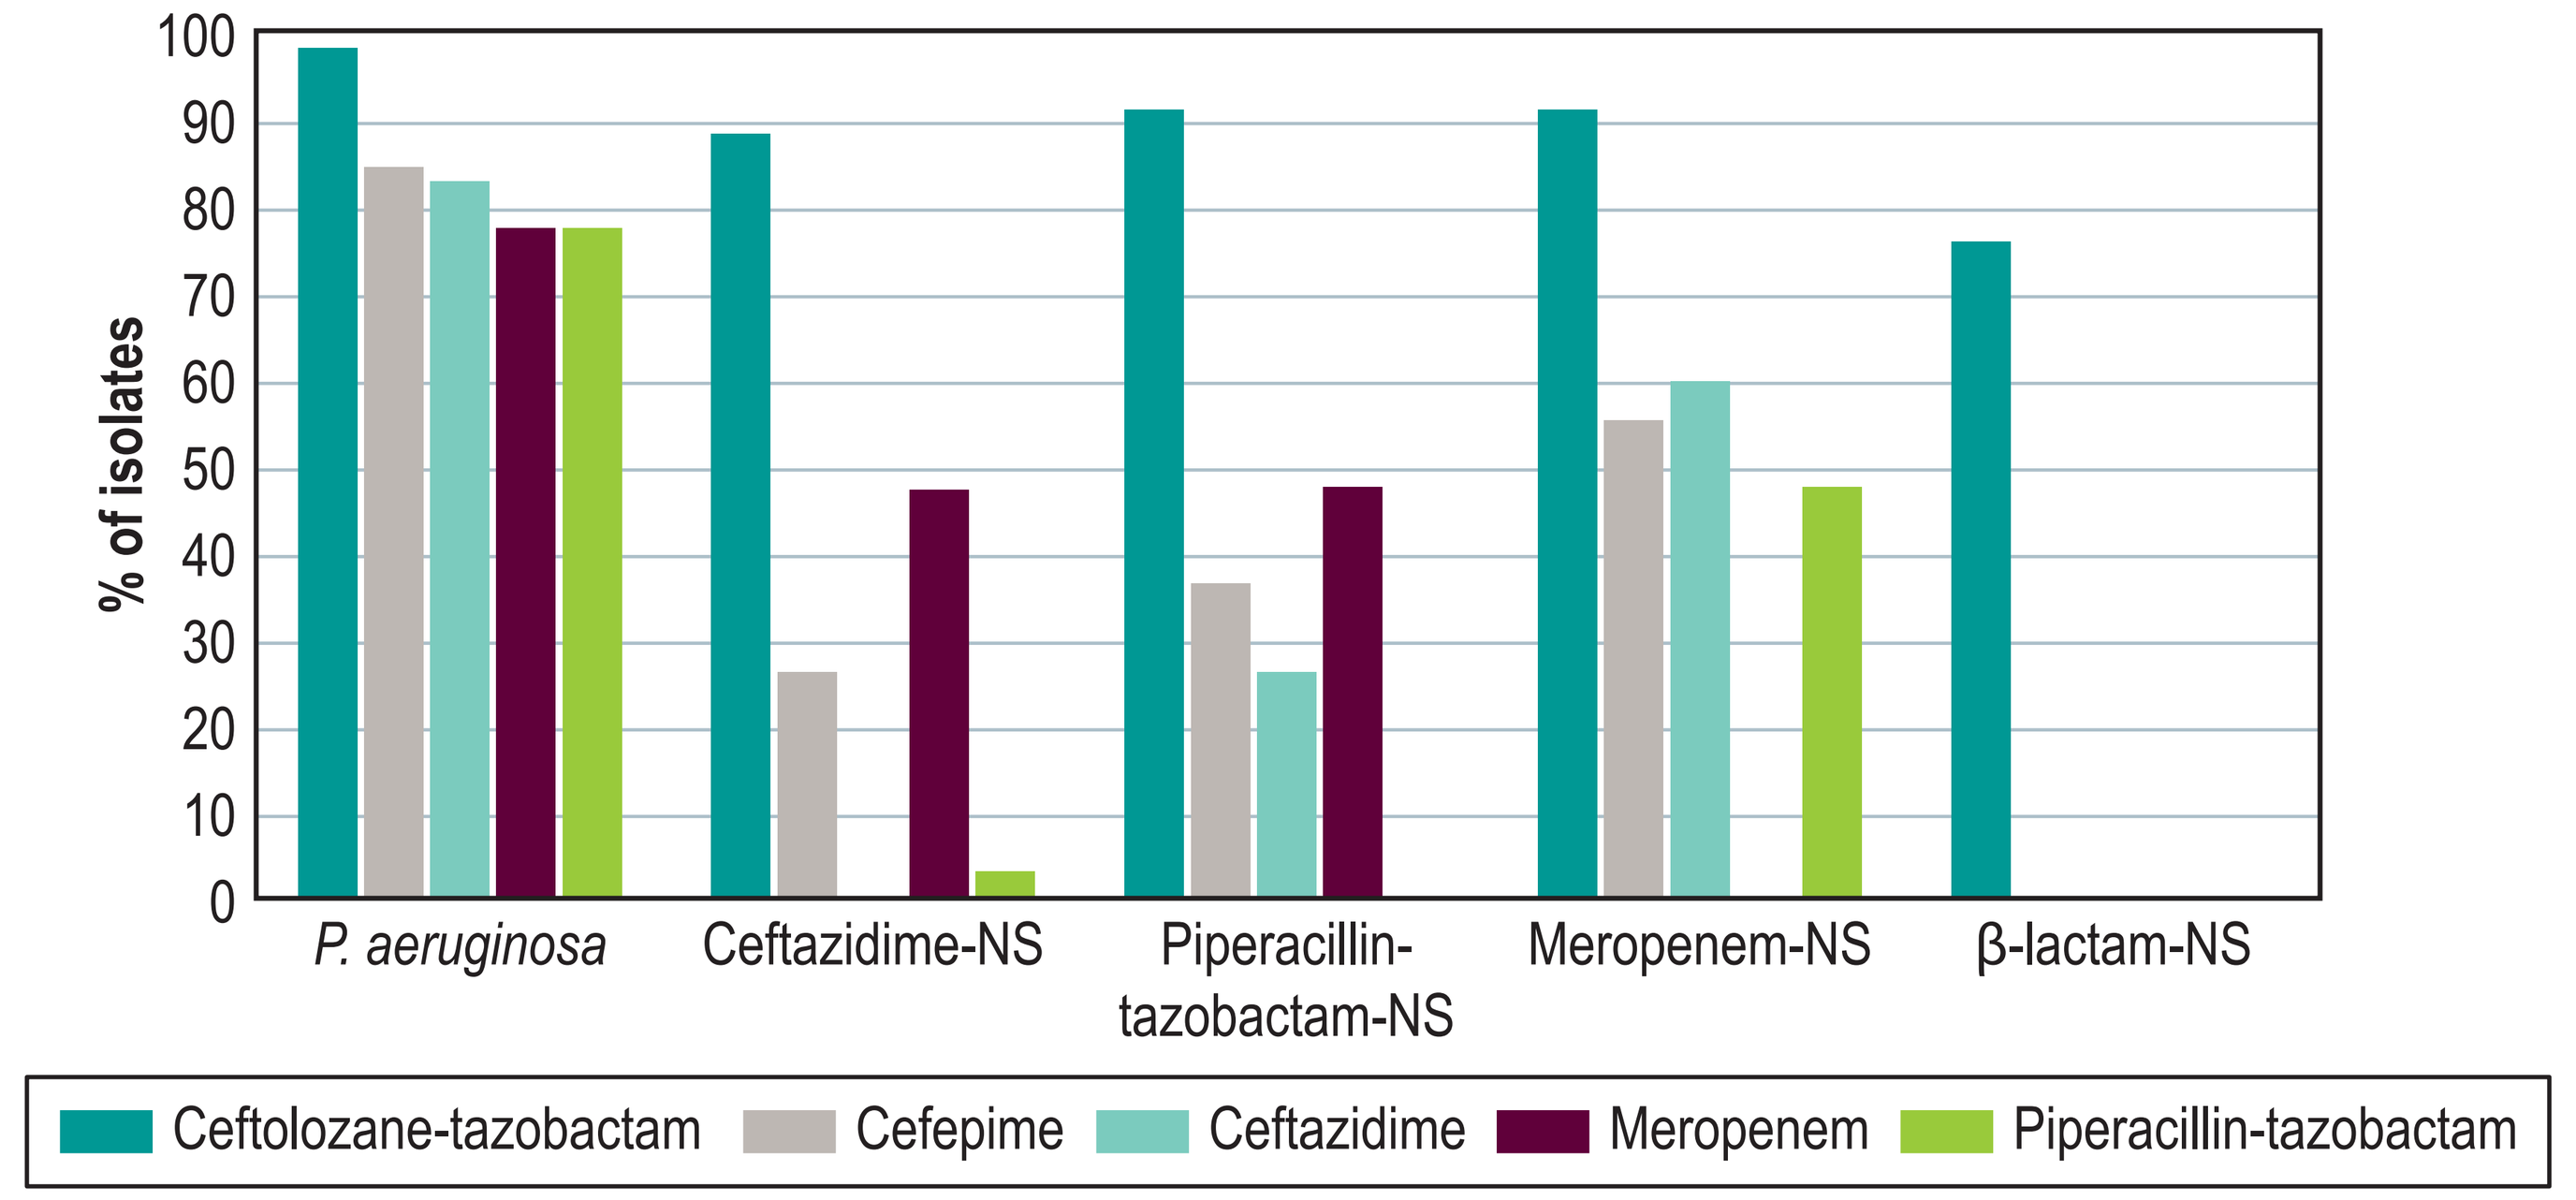
100
90
80
70
60
50
40
30
20
10
0
% of isolates
P. aeruginosa
Ceftazidime-NS
Piperacillin-tazobactam-NS
Meropenem-NS
β-lactam-NS
Ceftolozane-tazobactam
Cefepime
Ceftazidime
Meropenem
Piperacillin-tazobactam

Table 1 Susceptibilities of ceftolozane-tazobactam and comparators for *E. coli* and *K. pneumoniae* isolated from US ICU patients hospitalized with pneumonia

		% susceptible[a]							
	n	Ceftolozane-tazobactam	Cefepime	Ceftazidime	Meropenem	Piperacillin-tazobactam	Levofloxacin	Amikacin	Colistin
E. coli	216	95.8	78.2	79.2	99.1	90.7	61.9	99.1	100.0
Ceftazidime-NS	45	80.0	11.1	0.0	95.6	80.0	17.8	97.8	100.0
Piperacillin-tazobactam-NS	20	65.0	55.0	55.0	90.0	0.0	35.0	95.0	100.0
K. pneumoniae	296	93.6	89.2	85.8	95.3	87.8	92.5	99.3	98.3
Ceftazidime-NS	42	54.8	23.8	0.0	66.7	42.9	50.0	95.2	95.2
Piperacillin-tazobactam-NS	36	47.2	44.4	33.3	63.9	0.0	61.1	94.4	94.4

[a] CLSI (2018), EUCAST (2018) for *E. coli* and *K. pneumoniae* versus colistin

- For *Enterobacteriaceae,* amikacin (99.5% susceptible) and meropenem (97.0% susceptible) were the most active agents followed by ceftolozane-tazobactam (91.9% susceptible) (Table 2)
 - Ceftolozane-tazobactam was not active against 2 CRE *E. coli* and 14 CRE *K. pneumoniae* isolates (Table 3)
- For *P. aeruginosa*, ceftolozane-tazobactam (97.9%) and colistin (99.6%) were the most active agents, followed by amikacin (96.5%), as shown in Table 2 and Figure 2
 - Other β-lactams had <85% susceptibility
 - Levofloxacin had 76.0% susceptibility

Table 2 Ceftolozane-tazobactam and comparator susceptibilities when tested against isolates from US ICU patients hospitalized with pneumonia

Antimicrobial agent	MIC_{50}	MIC_{90}	Range	CLSI[a] %S	CLSI[a] %I	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %I	EUCAST[a] %R
Enterobacteriaceae (n=1,113)									
Ceftolozane-tazobactam	0.25	2	0.03 to >32	91.9	2.2	5.8	89.7		10.3
Amikacin	2	4	≤0.25 to 32	99.5	0.5	0	98.7	0.8	0.5
Cefepime	≤0.12	8	≤0.12 to >16	88.8	2.4	8.8 [b]	86.9	3.1	10.1
Ceftazidime	0.25	32	0.03 to >32	83.5	1.3	15.2	80.8	2.7	16.5
Colistin	0.12	>8	≤0.06 to >8				79.4		20.6
Levofloxacin	0.06	>4	≤0.03 to >4	87.2	1.3	11.5	83	2.7	14.3
Meropenem	0.03	0.06	≤0.015 to >32	97	0.7	2.2	97.8	1.3	1
Piperacillin-tazobactam	2	64	≤0.5 to >64	87.4	5.1	7.5	82.4	5	12.6
E. coli (n=216)									
Ceftolozane-tazobactam	0.25	0.5	0.06 to >32	95.8	1.4	2.8	95.4		4.6
Amikacin	2	4	0.5 to 32	99.1	0.9	0	96.8	2.3	0.9
Cefepime	≤0.12	>16	≤0.12 to >16	78.2	2.3	19.4 [b]	77.8	1.9	20.4
Ceftazidime	0.25	32	0.06 to >32	79.2	1.9	19	75.5	3.7	20.8
Colistin	0.12	0.25	≤0.06 to 1				100		0
Levofloxacin	0.25	>4	≤0.03 to >4	61.9	0.9	37.2	60.5	1.4	38.1
Meropenem	≤0.015	0.03	≤0.015 to 8	99.1	0	0.9	99.1	0.9	0
Piperacillin-tazobactam	2	16	≤0.5 to >64	90.7	0.9	8.3	87	3.7	9.3
K. pneumoniae (n=296)									
Ceftolozane-tazobactam	0.25	1	0.06 to >32	93.6	1	5.4	92.2		7.8
Amikacin	1	2	≤0.25 to 32	99.3	0.7	0	98.6	0.7	0.7
Cefepime	≤0.12	8	≤0.12 to >16	89.2	1	9.8 [b]	87.2	2	10.8
Ceftazidime	0.25	32	0.03 to >32	85.8	1	13.2	82.8	3	14.2
Colistin	0.12	0.25	≤0.06 to >8				98.3		1.7
Levofloxacin	0.06	1	≤0.03 to >4	92.5	1	6.4	86.1	4.4	9.5
Meropenem	0.03	0.03	≤0.015 to >32	95.3	1.4	3.4	96.6	1.4	2
Piperacillin-tazobactam	4	32	0.12 to >64	87.8	4.7	7.4	79.1	8.8	12.2
P. aeruginosa (n=571)									
Ceftolozane-tazobactam	0.5	2	0.12 to >32	97.9	0.4	1.8	97.9		2.1
Amikacin	4	8	≤0.25 to >32	96.5	1.6	1.9	91.6	4.9	3.5
Cefepime	4	16	≤0.5 to >16	84.2	11.2	4.6	84.2		15.8
Ceftazidime	2	32	≤0.25 to >32	82.5	5.6	11.9	82.5		17.5
Colistin	1	1	≤0.5 to 4	99.6		0.4	99.6		0.4
Levofloxacin	0.5	>4	≤0.12 to >4	76.0	7.2	16.8	65.8		34.2
Meropenem	0.5	8	≤0.015 to >32	77.1	7.9	15.1	77.1	13.3	9.6
Piperacillin-tazobactam	4	>64	≤0.5 to >64	77.1	12.4	10.5	77.1		22.9

[a] Criteria as published by CLSI (2018) and EUCAST (2018)
[b] Intermediate interpreted as susceptible-dose dependent

Organisms include: *Citrobacter amalonaticus/farmeri* (1), *C. freundii* species complex (13), *C. koseri* (23), *Enterobacter aerogenes* (98), *E. asburiae* (2), *E. cloacae* (77), *E. cloacae* species complex (82), *E. kobei* (1), *E. taylorae* (1), *Escherichia coli* (216), *Hafnia alvei* (3), *Klebsiella oxytoca* (96), *K. pneumoniae* (296), *K. variicola* (2), *Morganella morganii* (6), *Pantoea agglomerans* (2), *P. dispersa* (1), *Proteus mirabilis* (31), *P. vulgaris* (1), *P. vulgaris* group (2), *Providencia rettgeri* (5), *P. stuartii* (8), *Rahnella aquatilis* (1), *Raoultella ornithinolytica* (4), *R. planticola* (3), *Serratia liquefaciens* (4), *S. marcescens* (132), unspeciated *Pantoea* (1), unspeciated *Raoultella* (1)

Table 3 Antimicrobial activity of ceftolozane-tazobactam tested against the main organisms and organism groups of isolates

Organism/organism group (no. of isolates)	No. of isolates at MIC (mg/L; cumulative %)												MIC_{50}	MIC_{90}
	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>		
Enterobacteriaceae (1,113)	1 0.1	15 1.4	314 29.6	373 63.2	217 82.7	78 89.7	25 91.9	25 94.2	21 96	16 97.5	5 97.9	23 100	0.25	2
Escherichia coli (216)	0 0	7 3.2	100 49.5	71 82.4	21 92.1	7 95.4	1 95.8	3 97.2	1 97.7	2 98.6	1 99.1	2 100	0.25	0.5
CRE (2)							0 0	1 50	0 50	1 100			4	
ESBL non-CRE (53)		0 0	5 9.4	19 45.3	15 73.6	6 84.9	1 86.8	2 90.6	1 92.5	1 94.3	1 96.2	2 100	0.5	4
Levofloxacin-nonsusceptible (82)	0 0	2 2.4	12 17.1	39 64.6	15 82.9	6 90.2	1 91.5	2 93.9	1 95.1	2 97.6	1 98.8	1 100	0.25	1
Klebsiella pneumoniae (296)	0 0	2 0.7	91 31.4	114 69.9	37 82.4	29 92.2	4 93.6	3 94.6	2 95.3	7 97.6	1 98	6 100	0.25	1
CRE (13)							0 0	1 7.7	1 15.4	4 46.2	1 53.8	6 100	32	>32
ESBL, non-CRE (37)		0 0	1 2.7	7 21.6	7 40.5	14 78.4	3 86.5	2 91.9	1 94.6	2 100			1	4
Levofloxacin-nonsusceptible (22)			0 0	2 9.1	5 31.8	2 40.9	2 50	1 54.5	1 59.1	3 72.7	1 77.3	5 100	2	>32
Pseudomonas aeruginosa (571)		0 0	6 1.1	60 11.6	336 70.4	108 89.3	30 94.6	19 97.9	2 98.2	1 98.4	2 98.8	7 100	0.5	2
β lactam-nonsusceptible (45)					0 0	11 24.4	12 51.1	11 75.6	2 80	1 82.2	1 84.4	7 100	2	>32
Meropenem-nonsusceptible (131)			0 0	1 0.8	45 35.1	45 69.5	16 81.7	12 90.8	2 92.4	1 93.1	2 94.7	7 100	1	4
Ceftazidime-nonsusceptible (100)				0 0	10 10	32 42	27 69	19 88	2 90	1 91	2 93	7 100	2	8

CLSI (2018) susceptible breakpoint indicated by shaded column.

CONCLUSIONS

- Ceftolozane-tazobactam demonstrated activity against gram-negative isolates from US patients hospitalized with pneumonia in the ICU
- Ceftolozane-tazobactam, amikacin, and colistin had >93% susceptibility for each of the 3 most common pathogens in pneumonia patients in the ICU: *E. coli, K. pneumoniae,* and *P. aeruginosa*
 - Ceftolozane-tazobactam was the only β-lactam that had >90% susceptibility for *E. coli, K. pneumoniae,* and *P. aeruginosa*
- For *P. aeruginosa,* ceftolozane-tazobactam was the most active β-lactam tested and second in activity to colistin
- Although clinical data are needed to confirm these findings, these data suggest that ceftolozane-tazobactam may be a useful treatment option against gram-negative pathogens obtained from ICU patients hospitalized with pneumonia

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ USA

References

Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition.* Wayne, PA: CLSI.

EUCAST (2018). Breakpoint tables for interpretation of MIC's and zone diameters. Version 8.0, January 2018. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf. Accessed January 2018.

Contact Information:

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Copies of this paper obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without permission of the authors.

https://bit.ly/2I8wk2O

Presented at ASM-Microbe 2018, June 7–11, 2018; Atlanta, Georgia

Copyright © 2018 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.