Sunday-393

Antimicrobial Activity of Ceftolozane-Tazobactam Tested against Contemporary (2015–2017) *P. aeruginosa* Isolates from a Global Surveillance Program

Dee Shortridge, Michael A. Pfaller, Leonard R. Duncan, Jennifer M. Streit, Robert K. Flamm
JMI Laboratories, North Liberty, Iowa, USA

INTRODUCTION

- Ceftolozane-tazobactam is an antibacterial combination of an antipseudomonal cephalosporin and a β-lactamase inhibitor
 - Ceftolozane-tazobactam is active against isolates with common β-lactam resistance mechanisms found in *Pseudomonas aeruginosa*, including AmpC production, up-regulated efflux pumps, and porin reductions
- Ceftolozane-tazobactam is approved in >50 countries, including the United States since 2014 and in Europe since 2015, for complicated urinary tract infections that include acute pyelonephritis and complicated intra-abdominal infections in combination with metronidazole
 - Clinical treatment trials for hospital-acquired bacterial pneumonia/ ventilator-associated bacterial pneumonia are currently in progress (clinicaltrials.gov Identifier: NCT02070757)
- The Program to Assess Ceftolozane-Tazobactam Susceptibility (PACTS) is a global surveillance program that monitors resistance of gram-negative isolates to ceftolozane-tazobactam
- PACTS data for *Pseudomonas aeruginosa* isolates consecutively collected from various infection types in hospitalized patients from 2015–2017 were analyzed for this study

MATERIALS AND METHODS

- A total of 7,115 *P. aeruginosa* isolates were collected from 104 hospitals from 40 countries on 4 continents and tested for susceptibility to ceftolozane-tazobactam by CLSI broth microdilution methodology at JMI Laboratories using CLSI (2018) breakpoints
 - Isolate distribution was: 2,715 from North America (US-only), 2,894 from Europe, 605 from Latin America, and 901 from Asia-Pacific (not including China or India)
- Other agents tested were amikacin, cefepime, ceftazidime, colistin, gentamicin, levofloxacin, meropenem, and piperacillin-tazobactam
 - Ceftolozane-tazobactam and piperacillin-tazobactam were tested with a fixed 4 mg/L concentration of tazobactam
- Antimicrobial resistance phenotypes analyzed included:
 - Ceftazidime-nonsusceptible (NS), colistin-NS, meropenem-NS, levofloxacin-NS, piperacillin-tazobactam-NS, and β-lactam-NS (BL-NS; NS to cefepime, ceftazidime, meropenem and piperacillin-tazobactam)
 - Multidrug-resistance (MDR) was identified as nonsusceptible to ≥1 drug in ≥3 drug classes
 - Extensively drug-resistant (XDR) was nonsusceptible to at least 1 agent in all but 2 or fewer antimicrobial classes
 - Classes tested included aminoglycosides, antipseudomonal carbapenems, antipseudomonal cephalosporins, quinolones, and antipseudomonal penicillin/β-lactamase inhibitor combination

Figure 1 Number of *P. aeruginosa* isolates by infection type and continent

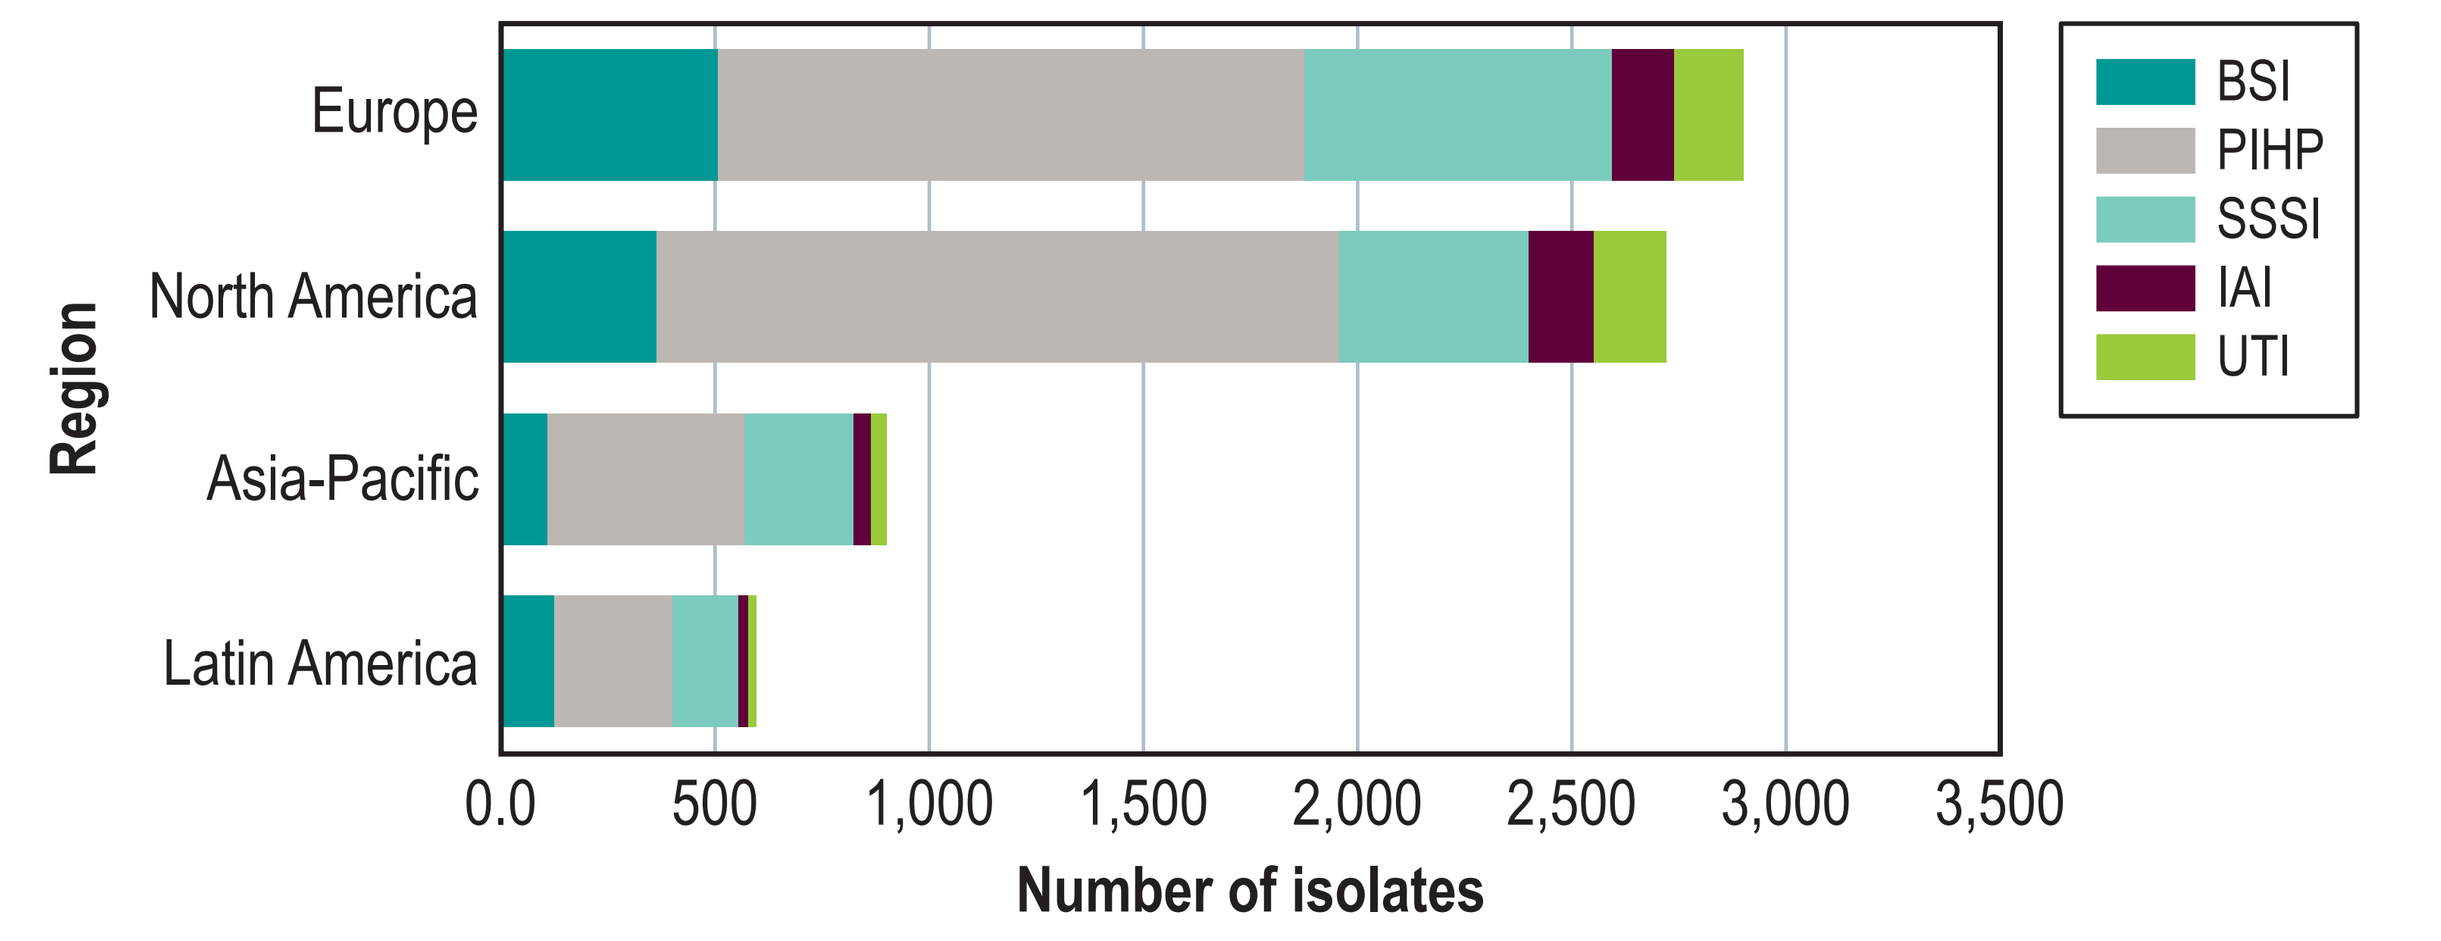
Europe
North America
Asia-Pacific
Latin America
Region
0.0
500
1,000
1,500
2,000
2,500
3,000
3,500
Number of isolates
BSI
PIHP
SSSI
IAI
UTI

RESULTS

- The most common infection was pneumonia (pneumonia in hospitalized patients [PIHP]; 51.9%) followed by skin and skin structure infection (SSSI; 22.0%) and bloodstream infection (BSI; 15.3%)
 - Other infection types included intra-abdominal infections (IAI) and urinary tract infections (UTI)
 - The distribution of *P. aeruginosa* isolates by infection type and continent is shown in Figure 1

Figure 2 Susceptibilities of ceftolozane-tazobactam and comparators by region

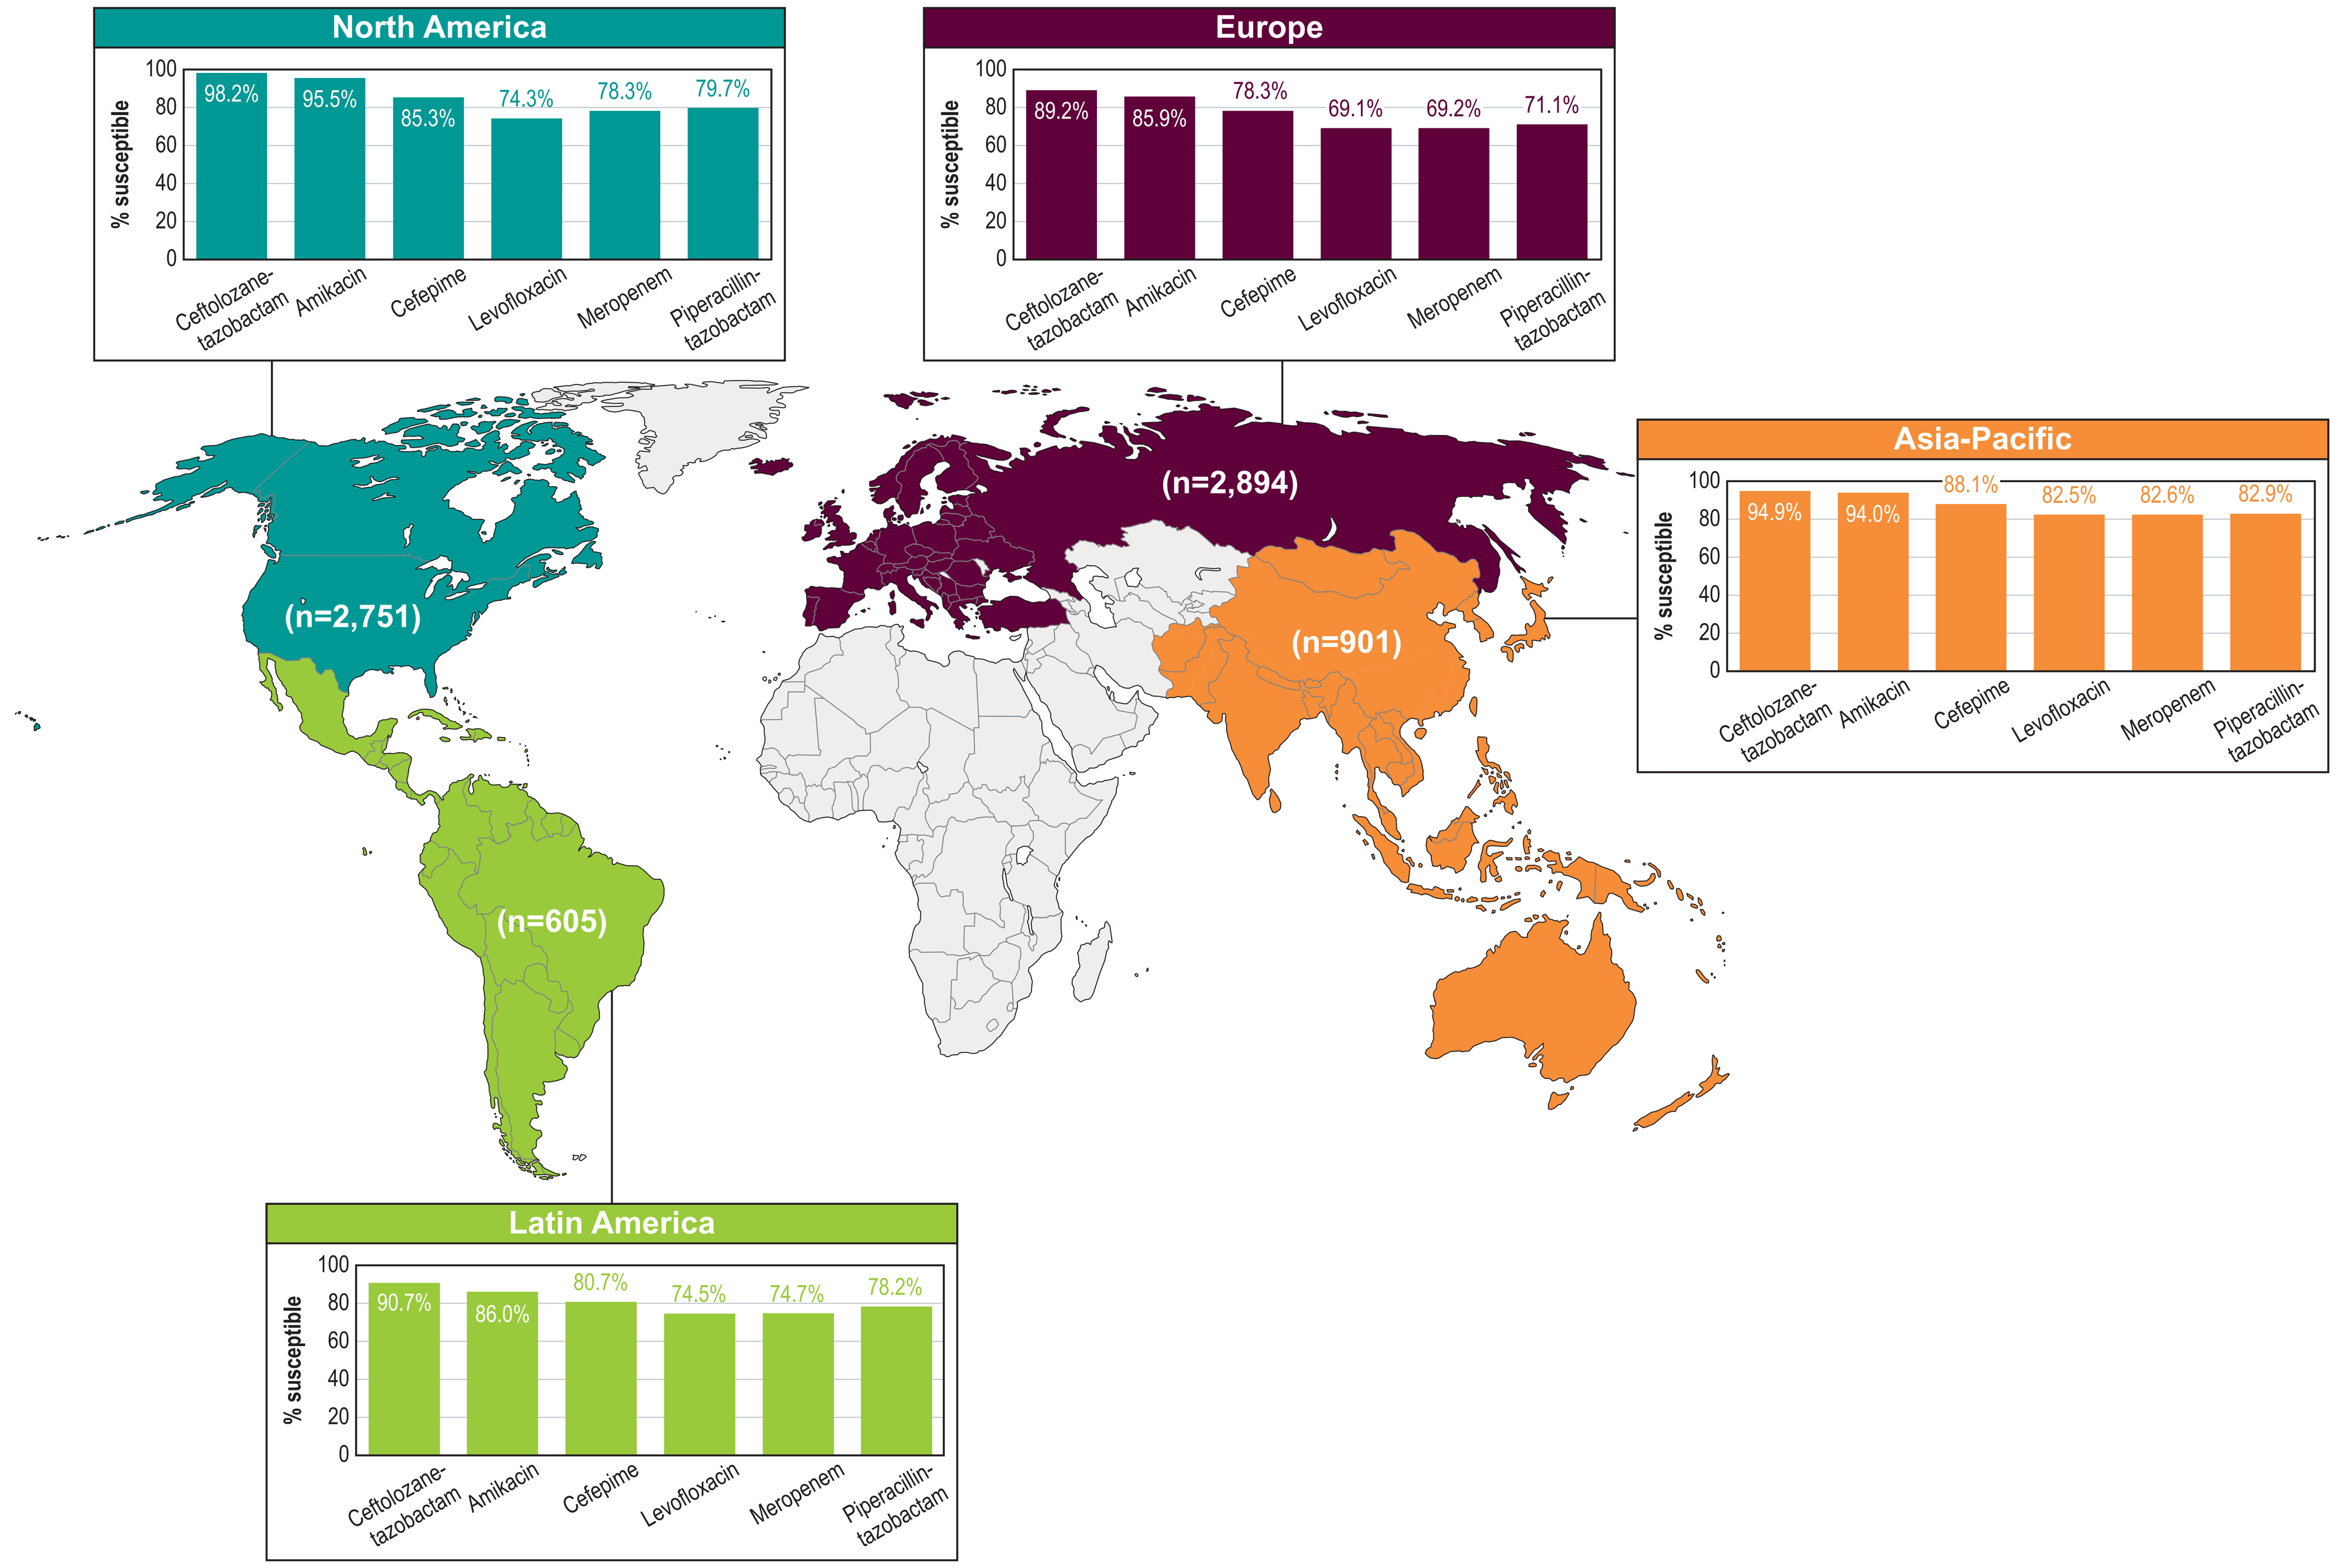
North America
% susceptible
100
80
60
40
20
0
98.2%
95.5%
85.3%
74.3%
78.3%
79.7%
Ceftolozane-tazobactam
Amikacin
Cefepime
Levofloxacin
Meropenem
Piperacillin-tazobactam
Europe
89.2%
85.9%
78.3%
69.1%
69.2%
71.1%
Asia-Pacific
94.9%
94.0%
88.1%
82.5%
82.6%
82.9%
Latin America
90.7%
86.0%
80.7%
74.5%
74.7%
78.2%
(n=2,751)
(n=2,894)
(n=901)
(n=605)

- Ceftolozane-tazobactam and comparators % susceptible against all isolates and the resistance phenotypes analyzed in this study are shown in Table 1
 - Ceftolozane-tazobactam showed >70% susceptibility to most phenotypes, including MDR, colistin-NS, and levofloxacin-NS
 - For isolates nonsusceptible to all other tested β-lactams, ceftolozane-tazobactam inhibited 47.5%
- Susceptibility for ceftolozane-tazobactam and comparators varied by region
 - 98.2% of isolates were ceftolozane-tazobactam-susceptible in North America, 94.9% in Asia-Pacific, 90.7% in Latin America, and 89.2% in Europe as shown in Table 2

Table 1 Susceptibility of ceftolozane-tazobactam and comparators to *P. aeruginosa* and resistance phenotypes

		% susceptible[a]						
P. aeruginosa	N	C-T	CAZ	MER	TZP	LVX	AMK	COL
All	7,115	93.5	74.5	71.9	70.8	61.6	84.3	99.3
MDR	1,789	74.3	37.1	17.7	25.5	18.9	66.0	99.4
XDR	1,013	57.5	16.7	3.4	6.6	4.8	49.0	99.1
Amikacin-NS	671	45.3	27.1	19.2	22.8	14.3	0.0	98.8
Colistin-NS	16	75.0	50.0	62.5	56.2	43.8	50.0	0.0
Levofloxacin-NS	1,904	77.1	52.0	36.4	43.0	0.0	69.8	99.5
Meropenem-NS	1,788	75.4	46.8	0.0	36.9	32.3	69.7	99.7
Ceftazidime-NS	1,385	67.5	0.0	31.3	5.3	34.0	64.7	99.4
Piperacillin-tazobactam-NS	1,671	73.7	21.5	32.4	0.0	35.1	69.0	99.6
β-lactam-NS[b]	753	47.5	0.0	0.0	0.0	13.5	46.9	99.5

[a] CLSI (2018).
[b] Nonsusceptible to 4 β-lactams: ceftazidime, cefepime, piperacillin-tazobactam, and meropenem.

Table 2 Antimicrobial activity of ceftolozane-tazobactam tested against the main organisms and organism groups of isolates

Organism / organism group (no. of isolates)	No. of isolates at MIC (mg/L; cumulative %)													MIC_{50}	MIC_{90}
	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>		
Pseudomonas aeruginosa (7,115)	1 <0.1	2 <0.1	17 0.3	60 1.1	1,041 15.8	3,782 68.9	1,227 86.2	342 91.0	179 93.5	59 94.3	37 94.8	49 95.5	319 100.0	0.5	2
MDR (1,789)		0 0.0	1 0.1	1 0.1	26 1.6	307 18.7	599 52.2	259 66.7	137 74.3	55 77.4	37 79.5	49 82.2	318 100.0	1	>32
XDR (1,013)				0 0.0	3 0.3	41 4.3	288 32.8	157 48.3	93 57.5	48 62.2	34 65.5	45 70.0	304 100.0	4	>32
β-lactam NS (753)					0 0.0	5 0.7	128 17.7	139 36.1	86 47.5	35 52.2	25 55.5	42 61.1	293 100.0	8	>32
Amikacin-nonsusceptible (671)			0 0.0	1 0.1	7 1.2	55 9.4	131 28.9	72 39.6	38 45.3	35 50.5	22 53.8	40 59.8	270 100.0	8	>32
Levofloxacin-nonsusceptible (1,904)		0 0.0	2 0.1	13 0.8	81 5.0	504 31.5	578 61.9	185 71.6	105 77.1	51 79.8	35 81.6	45 84.0	305 100.0	1	>32
Pseudomonas aeruginosa from North America (2,715)	0 0.0	1 <0.1	8 0.3	26 1.3	402 16.1	1,487 70.9	498 89.2	156 95.0	88 98.2	17 98.8	7 99.1	3 99.2	22 100.0	0.5	2
MDR from North America (627)				0 0.0	13 2.1	144 25.0	240 63.3	118 82.1	64 92.3	16 94.9	7 96.0	3 96.5	22 100.0	1	4
XDR from North America (282)				0 0.0	1 0.4	13 5.0	116 46.1	67 69.9	43 85.1	14 90.1	7 92.6	2 93.3	19 100.0	2	8
β-lactam NS from North America (188)					0 0.0	2 1.1	44 24.5	56 54.3	44 77.7	15 85.6	7 89.4	1 89.9	19 100.0	2	>32
Amikacin-nonsusceptible from North America (123)			0 0.0	1 0.8	2 2.4	18 17.1	37 47.2	27 69.1	11 78.0	10 86.2	5 90.2	1 91.1	11 100.0	2	16
Levofloxacin-nonsusceptible from North America (698)			0 0.0	4 0.6	45 7.0	235 40.7	238 74.8	83 86.7	48 93.6	15 95.7	7 96.7	2 97.0	21 100.0	1	4
Pseudomonas aeruginosa from Latin America (605)		0 0.0	1 0.2	8 1.5	71 13.2	342 69.8	96 85.6	17 88.4	14 90.7	7 91.9	6 92.9	5 93.7	38 100.0	0.5	4
MDR from Latin America (146)					0 0.0	20 13.7	46 45.2	10 52.1	14 61.6	7 66.4	6 70.5	5 74.0	38 100.0	2	>32
XDR from Latin America (86)					0 0.0	1 1.2	22 26.7	6 33.7	10 45.3	4 50.0	5 55.8	3 59.3	35 100.0	8	>32
β-lactam NS from Latin America (64)						0 0.0	13 20.3	4 26.6	6 35.9	2 39.1	3 43.8	3 48.4	33 100.0	>32	>32
Amikacin-nonsusceptible from Latin America (85)					0 0.0	10 11.8	13 27.1	8 36.5	9 47.1	7 55.3	3 58.8	2 61.2	33 100.0	8	>32
Levofloxacin-nonsusceptible from Latin America (154)			0 0.0	3 1.9	4 4.5	34 26.6	40 52.6	9 58.4	12 66.2	7 70.8	5 74.0	4 76.6	36 100.0	1	>32
Pseudomonas aeruginosa from Europe (2,894)	1 <0.1	0 <0.1	6 0.2	21 1.0	382 14.2	1,450 64.3	523 82.3	135 87.0	63 89.2	24 90.0	20 90.7	40 92.1	229 100.0	0.5	8
MDR from Europe (870)		0 0.0	1 0.1	1 0.2	10 1.4	122 15.4	269 46.3	108 58.7	48 64.3	23 66.9	20 69.2	40 73.8	228 100.0	2	>32
XDR from Europe (564)				0 0.0	1 0.2	22 4.1	134 27.8	74 41.0	30 46.3	23 50.4	19 53.7	39 60.6	222 100.0	8	>32
β-lactam NS from Europe (443)					0 0.0	2 0.5	61 14.2	68 29.6	30 36.3	12 39.1	12 41.8	37 50.1	221 100.0	32	>32
Amikacin-nonsusceptible from Europe (409)				0 0.0	5 1.2	24 7.1	71 24.4	33 32.5	14 35.9	14 39.4	11 42.1	36 50.9	201 100.0	32	>32
Levofloxacin-nonsusceptible from Europe (894)		0 0.0	2 0.2	6 0.9	21 3.2	195 25.1	257 53.8	78 62.5	35 66.4	23 69.0	20 71.3	38 75.5	219 100.0	1	>32
Pseudomonas aeruginosa from Asia-Pacific (901)	0 0.0	1 0.1	2 0.3	5 0.9	186 21.5	503 77.4	110 89.6	34 93.3	14 94.9	11 96.1	4 96.6	1 96.7	30 100.0	0.5	2
MDR from Asia-Pacific (146)				0 0.0	3 2.1	21 16.4	44 46.6	23 62.3	11 69.9	9 76.0	4 78.8	1 79.5	30 100.0	2	>32
XDR from Asia-Pacific (81)				0 0.0	1 1.2	5 7.4	16 27.2	10 39.5	10 51.9	7 60.5	3 64.2	1 65.4	28 100.0	4	>32
β-lactam NS from Asia-Pacific (58)					0 0.0	1 1.7	10 19.0	11 37.9	6 48.3	6 58.6	3 63.8	1 65.5	20 100.0	8	>32
Amikacin-nonsusceptible from Asia-Pacific (54)					0 0.0	3 5.6	10 24.1	4 31.5	4 38.9	4 46.3	3 51.9	1 53.7	25 100.0	16	>32
Levofloxacin-nonsusceptible from Asia-Pacific (158)				0 0.0	11 7.0	40 32.3	43 59.5	15 69.0	10 75.3	6 79.1	3 81.0	1 81.6	29 100.0	1	>32

CLSI (2018). Susceptible CLSI breakpoint (≤4 mg/L) is indicated by shaded column

- North America (US-only) generally had higher susceptibility rates for all comparators than other regions
- Europe generally had lower susceptibility rates than the other regions
- MIC distributions for ceftolozane-tazobactam against the resistance phenotypes for all isolates and by region are shown in Table 3

CONCLUSIONS

- Ceftolozane-tazobactam had activity against most *P. aeruginosa* isolated from patients hospitalized with various infections in 40 countries
- Ceftolozane-tazobactam was more active than all comparators, except colistin, and maintained activity against MDR and XDR isolates and isolates nonsusceptible to all 4 tested β-lactams
 - Ceftolozane-tazobactam was active against 12/16 colistin-NS isolates
 - Isolates nonsusceptible to levofloxacin were 77.1% susceptible to ceftolozane-tazobactam
- Ceftolozane-tazobactam may be a useful treatment for infections caused by *P. aeruginosa,* including infections with various resistant phenotypes

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ USA

References

Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition.* Wayne, PA: CLSI.

EUCAST (2018). Breakpoint tables for interpretation of MIC's and zone diameters. Version 8.0, January 2018. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf. Accessed January 2018.

Contact Information:

Dee Shortridge
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Copies of this paper obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without permission of the authors.
https://bit.ly/2G7tmFU

Presented at ASM-Microbe 2018, June 7–11, 2018; Atlanta, Georgia

Copyright © 2018 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.