ASM Microbe 2018

Saturday-645

Activity of Plazomicin and Comparator Agents Tested against Contemporary Clinical Isolates Collected Worldwide

M CASTANHEIRA, JM STREIT, SJR ARENDS, RK FLAMM

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/ASM-Microbe-2018-plazomicin-Enterobacteriaceae.pdf

Charges may apply.
No personal information is stored.

Amended Abstract

Background: Plazomicin (PLZ), a next-generation aminoglycoside developed to overcome common aminoglycoside-resistance (R) mechanisms, has completed phase 3 studies in complicated urinary tract infection and serious infections due to carbapenem-R *Enterobacteriaceae* (CRE). We evaluated the activity of PLZ and comparator agents against 6,417 clinical isolates collected worldwide during 2017.

Methods: A total of 5,658 *Enterobacteriaceae* (ENT), 438 gram-positive, 167 *P. aeruginosa* (PSA), and 154 *Acinetobacter* spp. (ASP) were susceptibility tested using reference broth microdilution method. CLSI and EUCAST interpretative criteria were applied.

Results: PLZ ($MIC_{50/90}$, 0.5/1 µg/mL) inhibited 96.7% and 99.0% of the ENT at ≤2 and ≤4 µg/mL, respectively. PLZ was active against *E. coli* (n=2,000; $MIC_{50/90}$, 1/1 µg/mL), *K. pneumoniae* (n=1,852; $MIC_{50/90}$, 0.25/0.5 µg/mL), and *E. cloacae* (n=180; $MIC_{50/90}$, 0.25/0.5 µg/mL). All *K. oxytoca* (n=344) and *E. aerogenes* (n=163) isolates had PLZ MIC values ≤2 µg/mL. *C. freundii* (n=229) and *Serratia* spp. (n=180) had only 1 isolate each with PLZ MIC values >4 µg/mL. PLZ MIC_{50} and MIC_{90} for *M. morganii* (n=176), *Providencia* spp. (n=82), and *Proteus* spp. (n=283) ranged from 1-2 and 2-8 µg/mL, respectively. PLZ ($MIC_{50/90}$, 0.25/4 µg/mL) inhibited 185/205 (90.2%) CRE at ≤4 µg/mL. PLZ inhibited 726/766 (94.8%) of the gentamicin-R, 673/709 (94.9%) of the tobramycin-R and 23/56 (41.1%) of the amikacin-R ENT at ≤4 µg/mL (CLSI breakpoints). The highest PLZ MIC value for staphylococci (n=225) was 1 µg/mL. PLZ displayed limited activity against *Enterococcus* spp. (n=110; $MIC_{50/90}$, 32/64 µg/mL) and *S. pneumoniae* (n=109; $MIC_{50/90}$, 32/64 µg/mL). PLZ displayed activity against some PSA (n=168; $MIC_{50/90}$, 8/16 µg/mL) and limited activity against ASP (n=155; $MIC_{50/90}$, 8/>128 µg/mL). There were some variations in PLZ activity observed among the different geographic regions analyzed (Table).

Conclusions: PLZ was active against contemporary ENT clinical isolates collected worldwide, including most CRE isolates where treatment options are limited. Further development of this agent for the potential treatment of serious infections due to ENT is warranted.

Isolates	$MIC_{50/90}$ in µg/mL (no. tested):			
	USA	Europe	Asia-Pacific	Latin America
ENT	0.5/1 (2,051)	0.5/1 (1,966)	0.5/1 (860)	0.5/2 (781)
CRE	0.25/1 (22)	0.25/2 (97)	0.25/128 (28)	0.25/>128 (57)
Gentamicin R	0.5/2 (164)	0.25/2 (264)	0.5/2 (136)	0.5/2 (202)
Tobramycin R	0.5/2 (117)	0.25/2 (312)	0.5/2 (102)	0.5/2 (117)
Amikacin R	0.5/ (2)	128/>128 (22)	>128/ (7)	8/>128 (25)

Introduction

- Aminoglycosides are broad-spectrum agents that have been used for several decades to treat serious infections caused by non-fastidious gram-negative bacteria, staphylococci, enterococci, and viridans group streptococci
 - Aminoglycosides are also used in combination with other agents displaying synergistic activity with this class, such as β-lactams, fluoroquinolones, polymyxins, and vancomycin
- Plazomicin is a semi-synthetic aminoglycoside developed from sisomicin that demonstrates activity against *Enterobacteriaceae*, including multidrug-resistant isolates, *Staphylococcus* spp., and some *Pseudomonas aeruginosa*
 - This agent contains structural modifications that allow it to retain activity in the presence of aminoglycoside-modifying enzymes (AMEs)
- Plazomicin has been evaluated in 2 phase 3 clinical trials, including a study to evaluate the efficacy of this aminoglycoside against carbapenem-resistant *Enterobacteriaceae* (CRE)
- We evaluated the activity of plazomicin and comparator antimicrobial agents against 6,417 clinical isolates collected worldwide during 2017

Materials and Methods

- A total of 6,417 clinical isolates were collected during 2017 from 90 hospitals worldwide as part of the ALERT (Antimicrobial Longitudinal Evaluation and Resistance Trends) program
 - Isolates identified as causative of infection were included in the study; isolates were limited to 1 per patient
 - Isolates included 5,658 *Enterobacteriaceae*, 438 gram-positive species, 167 *P. aeruginosa*, and 154 *Acinetobacter* spp.
 - The infection sources of isolates included in this study are summarized in Figure 1
- Isolates were susceptibility tested using the reference broth microdilution method described by the Clinical and Laboratory Standards Institute (CLSI)
 - Categorical interpretations for all comparator agents were those in the CLSI M100 document
 - Quality control (QC) was performed according to CLSI guidelines (M07, 2018), and all QC MIC results were within acceptable ranges as published in CLSI documents
- CRE was defined as any isolate exhibiting doripenem, imipenem, and/or meropenem MIC values at ≥2 µg/mL
 - *Proteus mirabilis* and indole-positive Proteeae were categorized as CRE if doripenem and/or meropenem MIC values were at ≥2 µg/mL due to intrinsically elevated imipenem MIC values

Results

- Plazomicin ($MIC_{50/90}$, 0.5/1 µg/mL) inhibited 96.7% and 99.0% of the *Enterobacteriaceae* at ≤2 and ≤4 µg/mL, respectively (Table 1)
- Plazomicin displayed similar activity against most common *Enterobacteriaceae* species, including *Escherichia coli* ($MIC_{50/90}$, 1/1 µg/mL), *Klebsiella pneumoniae* ($MIC_{50/90}$, 0.25/0.5 µg/mL), and *Enterobacter cloacae* ($MIC_{50/90}$, 0.25/0.5 µg/mL; Table 1)
 - All *Klebsiella oxytoca* and *Enterobacter aerogenes* isolates had plazomicin MIC values ≤2 µg/mL
 - *Citrobacter freundii* and *Serratia marcescens* had only 1 isolate each with plazomicin MIC values >4 µg/mL
- The plazomicin MIC_{50} and MIC_{90} values for *Morganella morganii*, *Providencia* spp., and *Proteus* spp. ranged from 1-2 and 2-8 µg/mL, respectively
- Plazomicin ($MIC_{50/90}$, 0.25/4 µg/mL) inhibited 90.2% (185/204) of CRE at ≤4 µg/mL
 - Amikacin, gentamicin, and tobramycin inhibited only 70.1%, 54.4%, and 15.2%, respectively, of the isolates at the current CLSI breakpoint (Figure 2)

Figure 1 Clinical isolates stratified by infection type

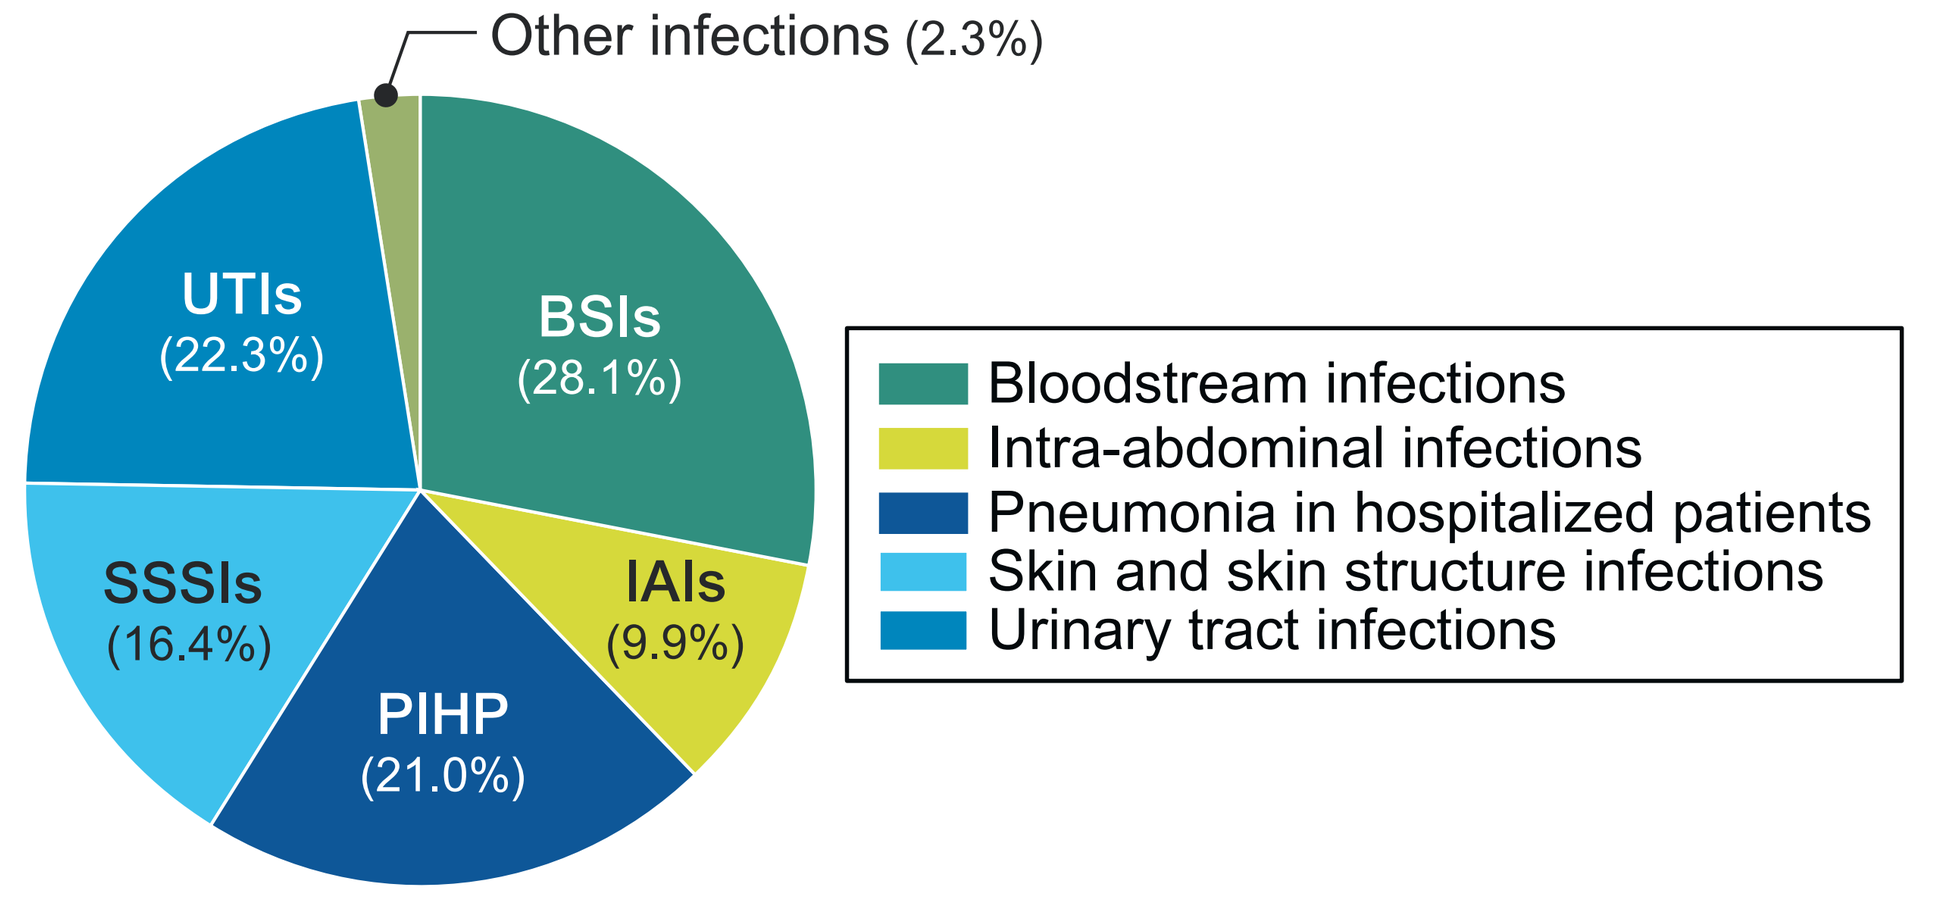

Other infections (2.3%)
BSIs (28.1%)
IAIs (9.9%)
PIHP (21.0%)
SSSIs (16.4%)
UTIs (22.3%)
Bloodstream infections
Intra-abdominal infections
Pneumonia in hospitalized patients
Skin and skin structure infections
Urinary tract infections

Figure 2 Comparative activity of plazomicin and other aminoglycosides tested against 204 CRE isolates collected worldwide

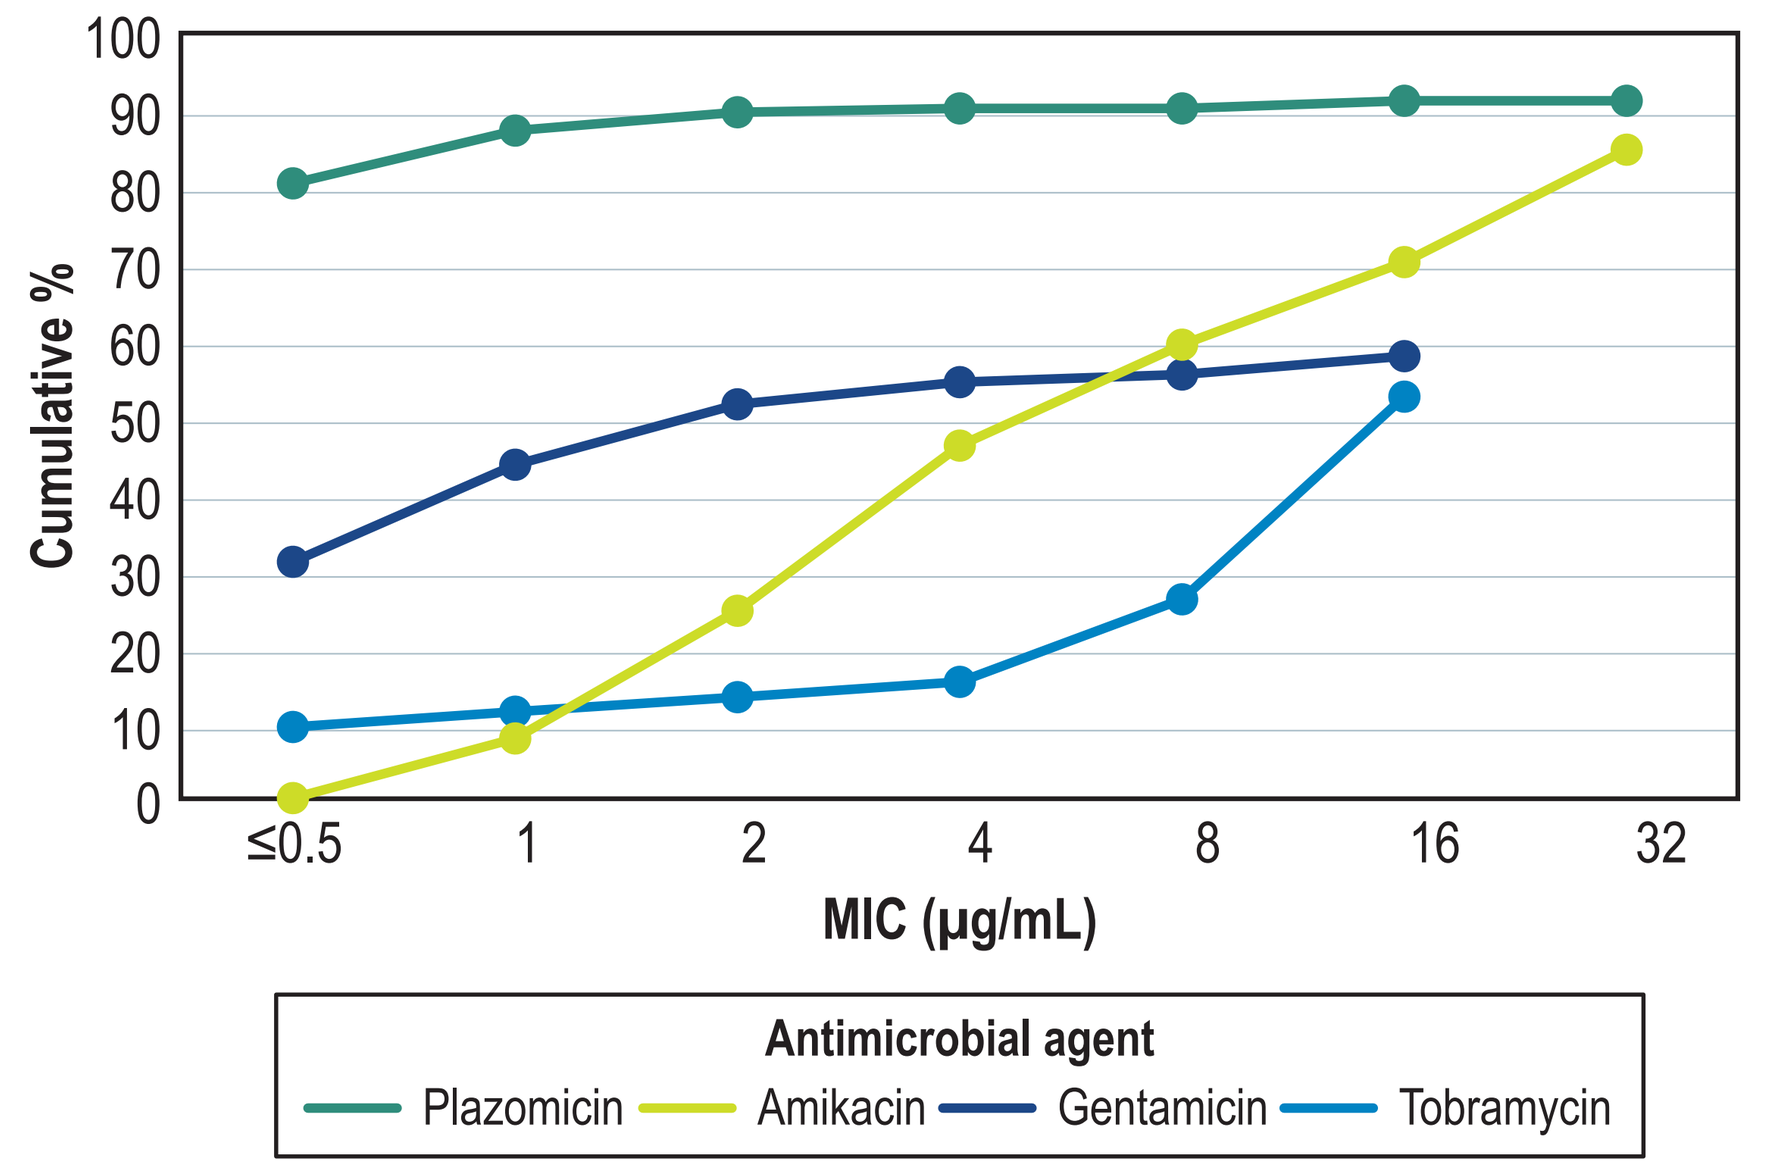

Cumulative %
MIC (µg/mL)
≤0.5
1
2
4
8
16
32
Antimicrobial agent
Plazomicin
Amikacin
Gentamicin
Tobramycin

Table 1 Antimicrobial activity of plazomicin against isolates tested

Organism/organism group (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %)													MIC_{50}	MIC_{90}
	≤0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	>128		
Enterobacteriaceae (5,658)	4 0.1	146 2.7	2,058 39.0	1,718 69.4	1,199 90.6	349 96.7	126 99.0	23 99.4	4 99.5	0 99.5	1 99.5	7 99.6	23 100.0	0.5	1
Carbapenem-resistant (204)	1 0.5	9 4.9	123 65.2	31 80.4	14 87.3	5 89.7	1 90.2	0 90.2	2 91.2	0 91.2	0 91.2	6 94.1	12 100.0	0.25	4
E. coli (2,000)	0 0.0	1 0.1	51 2.6	941 49.6	880 93.6	117 99.5	7 99.8	0 99.8	0 99.8	0 99.8	0 99.8	0 99.8	3 100.0	1	1
K. pneumoniae (1,850)	4 0.2	124 6.9	1,459 85.8	213 97.3	20 98.4	6 98.7	2 98.8	1 98.9	1 98.9	0 98.9	0 98.9	7 99.3	13 100.0	0.25	0.5
Carbapenem-resistant (179)	1 0.6	8 5.0	113 68.2	27 83.2	10 88.8	4 91.1	1 91.6	0 91.6	0 91.6	0 91.6	0 91.6	6 95.0	9 100.0	0.25	2
K. oxytoca (346)	0 0.0	3 0.9	139 41.0	185 94.5	17 99.4	2 100.0								0.5	0.5
E. cloacae species complex (180)	0 0.0	6 3.3	117 68.3	47 94.4	7 98.3	0 98.3	0 98.3	0 98.3	0 98.3	0 98.3	0 98.3	0 98.3	3 100.0	0.25	0.5
E. aerogenes (163)	0 0.0	2 1.2	86 54.0	66 94.5	7 98.8	2 100.0								0.25	0.5
S. marcescens (180)		0 0.0	6 3.3	68 41.1	95 93.9	8 98.3	2 99.4	0 99.4	0 99.4	0 99.4	0 99.4	0 99.4	1 100.0	1	1
C. freundii species complex (228)	0 0.0	2 0.9	78 35.1	121 88.2	26 99.6	0 99.6	0 99.6	0 99.6	0 99.6	0 99.6	0 99.6	0 99.6	1 100.0	0.5	1
C. koseri (170)	0 0.0	8 4.7	122 76.5	37 98.2	1 98.8	1 99.4	1 100.0							0.25	0.5
M. morganii (176)			0 0.0	17 9.7	70 49.4	71 89.8	13 97.2	5 100.0						2	4
Providencia spp. (82)			0 0.0	12 14.6	18 36.6	29 72.0	12 86.6	6 93.9	3 97.6	0 97.6	1 98.8	0 98.8	1 100.0	2	8
P. mirabilis (210)				0 0.0	18 8.6	94 53.3	88 95.2	9 99.5	0 99.5	0 99.5	0 99.5	0 99.5	1 100.0	2	4
P. vulgaris group (73)			0 0.0	11 15.1	40 69.9	19 95.9	1 97.3	2 100.0						1	2
P. aeruginosa (167)	1 0.6	2 1.8	5 4.8	4 7.2	3 9.0	7 13.2	52 44.3	55 77.2	25 92.2	8 97.0	2 98.2	3 100.0		8	16
Acinetobacter spp. (154)		0 0.0	3 1.9	17 13.0	26 29.9	13 38.3	5 41.6	14 50.6	7 55.2	13 63.6	4 66.2	1 66.9	51 100.0	8	>128
Coagulase-negative staphylococci (108)	26 24.1	29 50.9	43 90.7	7 97.2	3 100.0									0.12	0.25
S. aureus (113)	0 0.0	2 1.8	30 28.3	74 93.8	7 100.0									0.5	0.5
Methicillin-resistant (46)		0 0.0	14 30.4	30 95.7	2 100.0									0.5	0.5
S. pneumoniae (107)							0 0.0	1 0.9	8 8.4	59 63.6	37 98.1	2 100.0		32	64
Enterococcus spp. (110)					0 0.0	3 2.7	18 19.1	8 26.4	5 30.9	21 50.0	49 94.5	5 99.1	1 100.0	32	64

- Against isolates resistant to other aminoglycosides per CLSI breakpoints, plazomicin inhibited 94.8% (726/766) of the gentamicin-resistant, 94.9% (672/708) of the tobramycin-resistant, and 41.1% (23/56) of the amikacin-resistant *Enterobacteriaceae* isolates at ≤4 µg/mL (Figure 3)
 - The activity of plazomicin against gentamicin- and tobramycin-resistant isolates was similar in all geographic regions
- Against 167 *P. aeruginosa* isolates tested, the activity of plazomicin (MIC_{50} and MIC_{90}, 8 and 16 µg/mL) was similar to that of amikacin (MIC_{50} and MIC_{90}, 4 and 16 µg/mL; Table 1)

Figure 3 Activity of plazomicin tested against *Enterobacteriaceae* isolates resistant to other aminoglycosides applying CLSI breakpoints

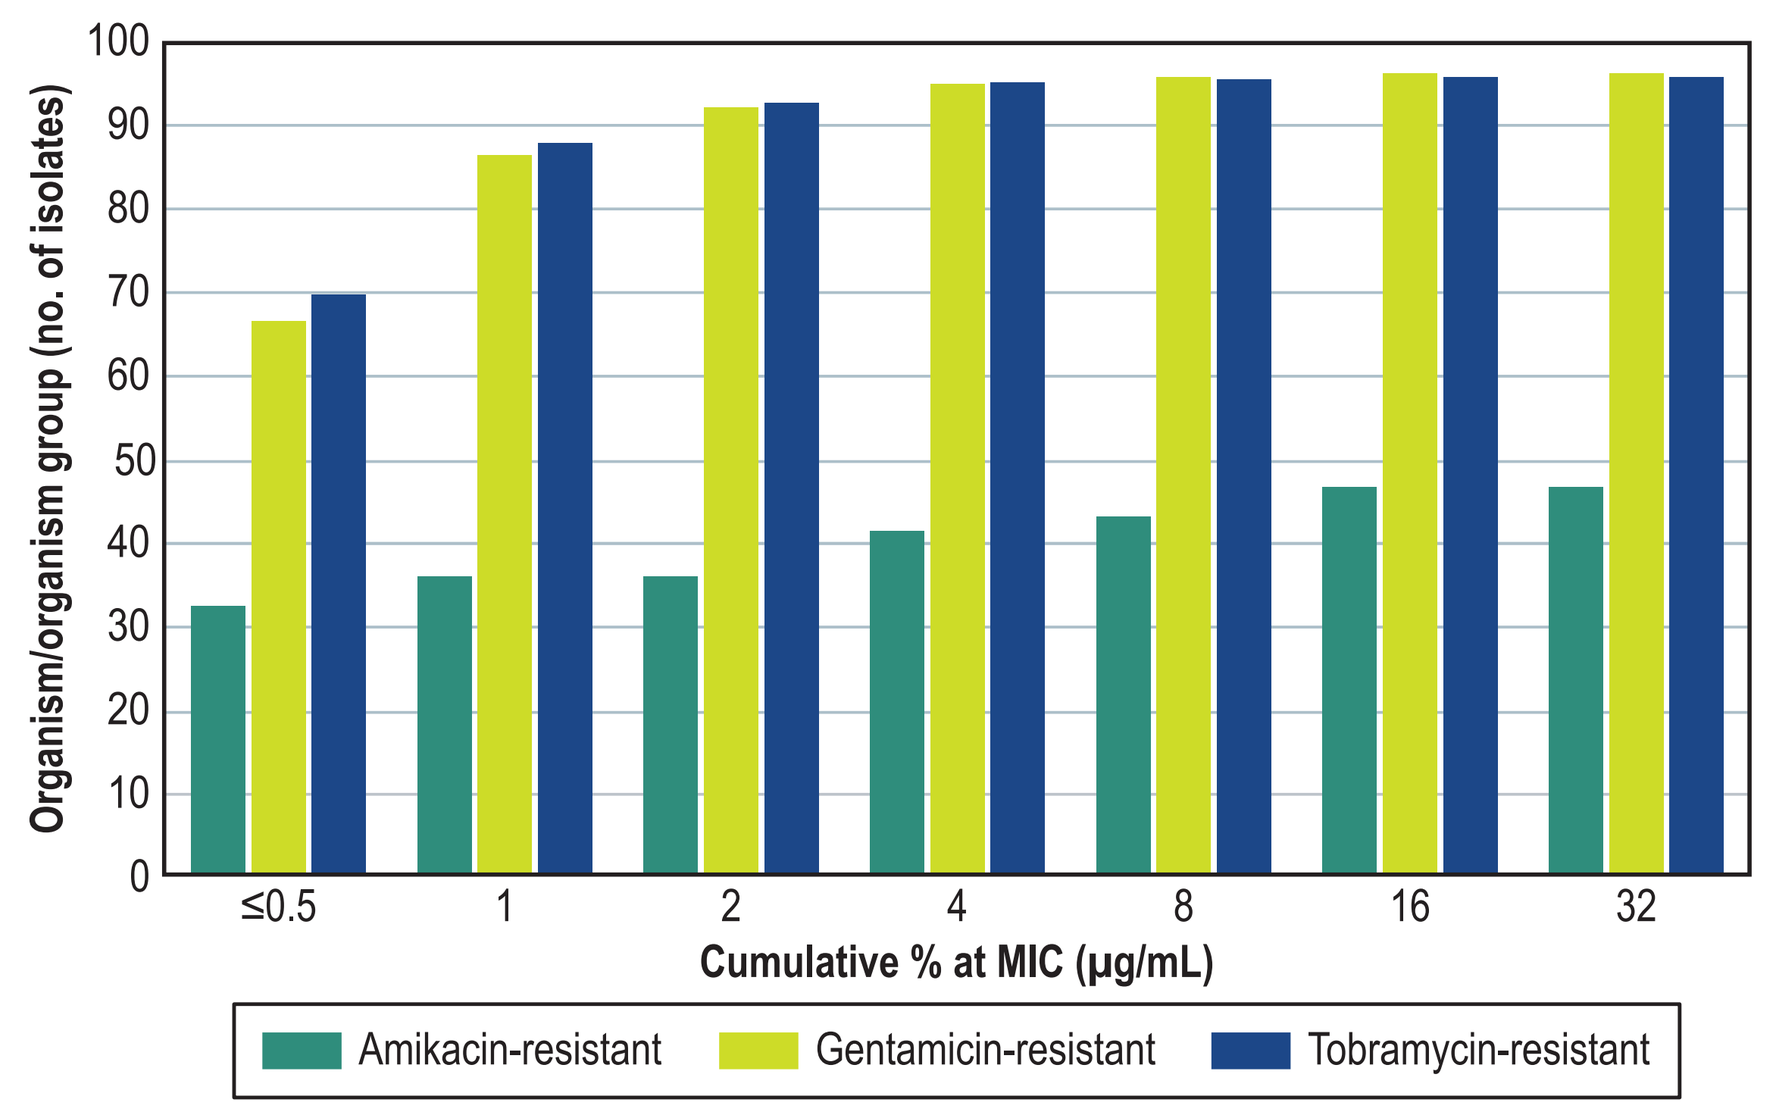

Organism/organism group (no. of isolates)
Cumulative % at MIC (µg/mL)
≤0.5
1
2
4
8
16
32
Amikacin-resistant
Gentamicin-resistant
Tobramycin-resistant

Figure 4 Comparative activity of plazomicin, amikacin, and gentamicin tested against CRE isolates by continent

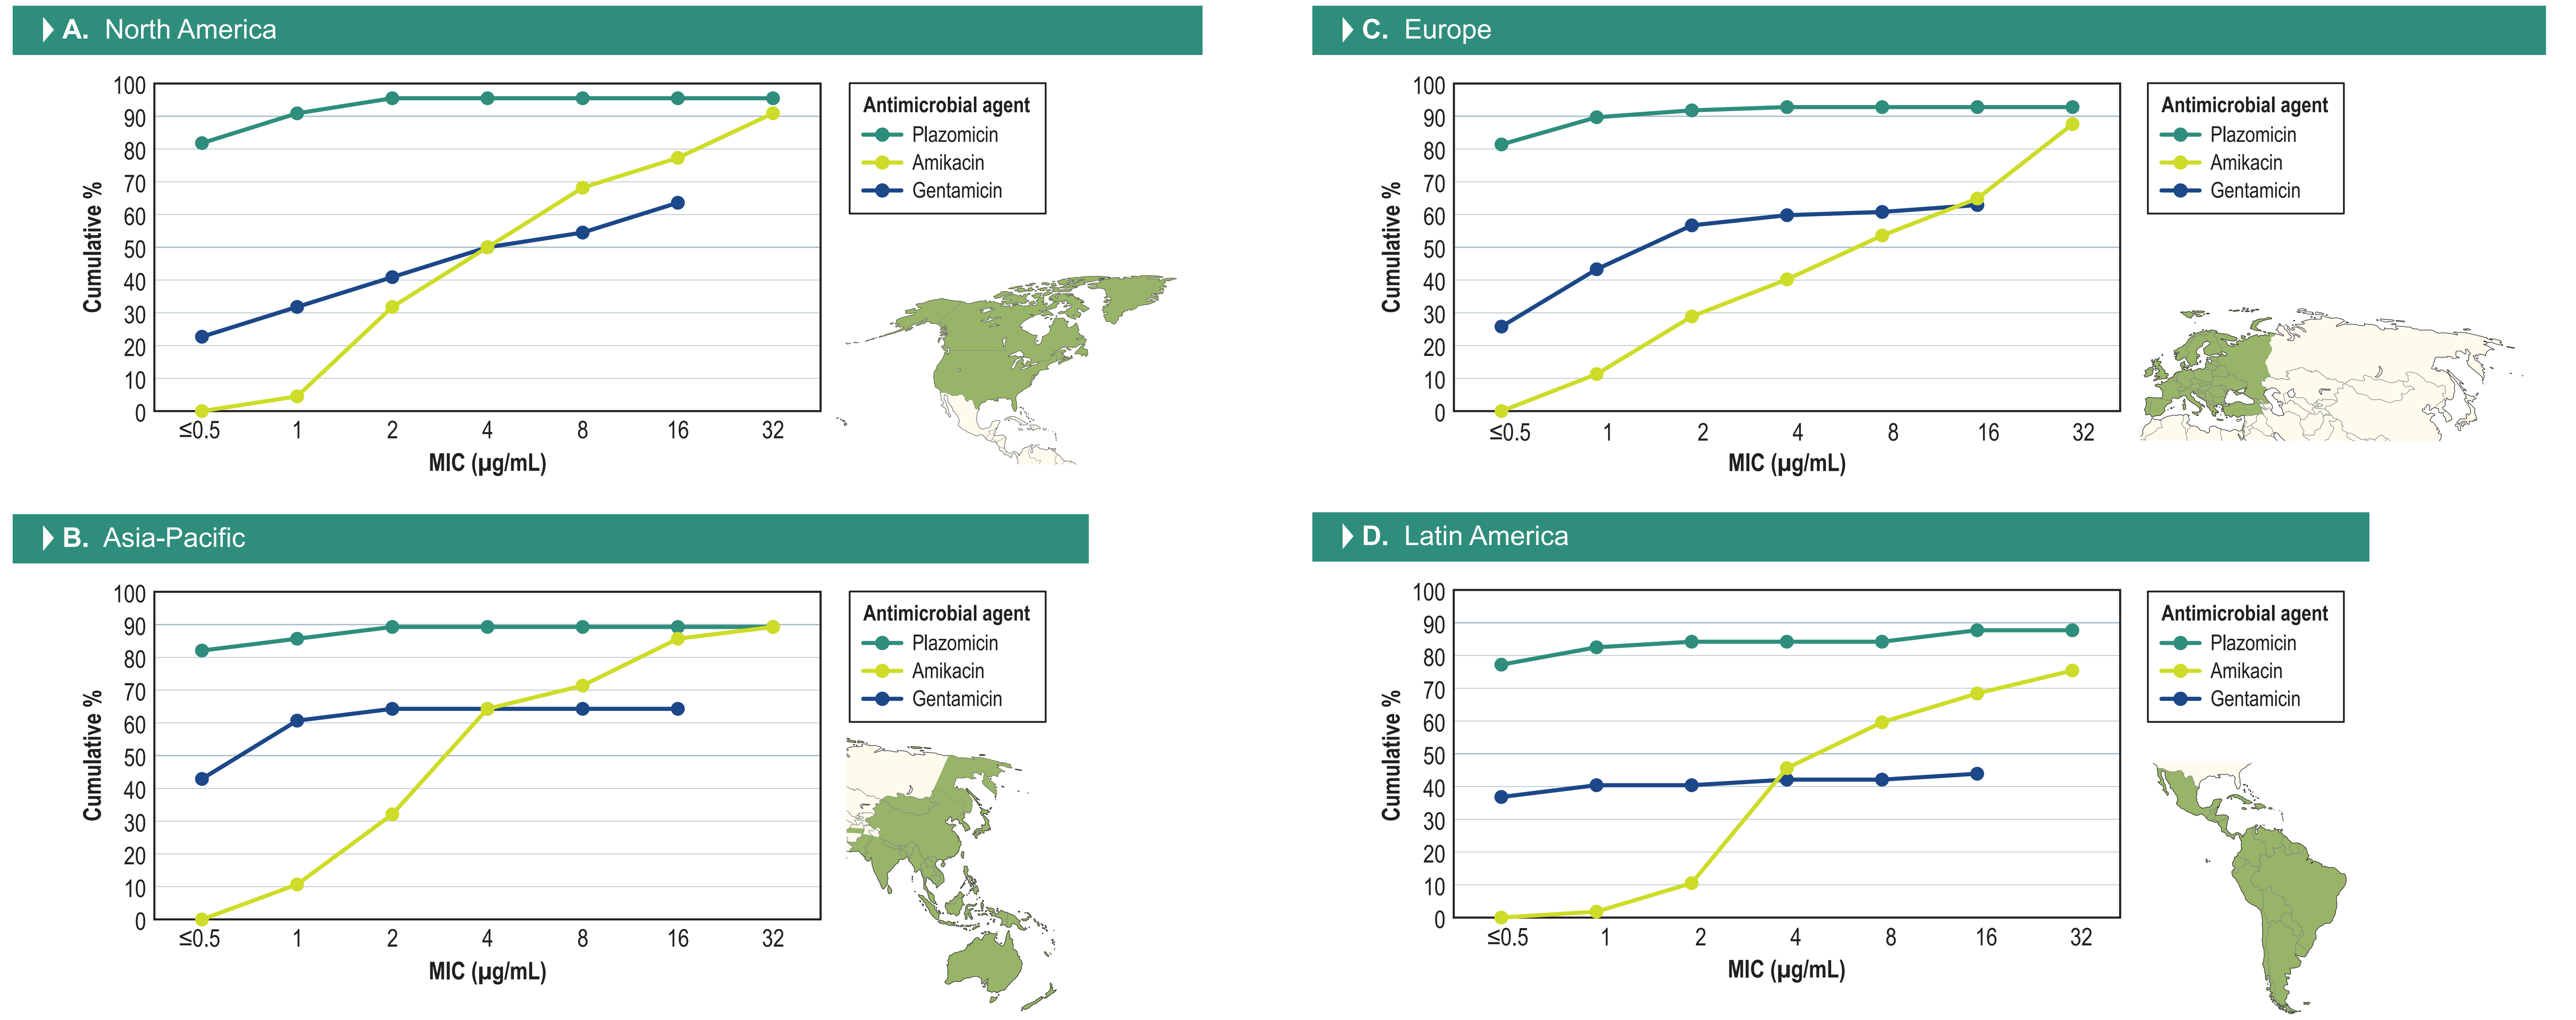

A. North America
B. Asia-Pacific
C. Europe
D. Latin America
Cumulative %
MIC (µg/mL)
≤0.5
1
2
4
8
16
32
Antimicrobial agent
Plazomicin
Amikacin
Gentamicin

- Plazomicin inhibited all 108 coagulase-negative staphylococci (MIC_{50} and MIC_{90}, 0.12 and 0.25 µg/mL) at ≤1 µg/mL and all 113 *Staphylococcus aureus* (MIC_{50} and MIC_{90}, 0.5 and 0.5 µg/mL) isolates at ≤1 µg/mL (Table 1), including methicillin-resistant (MRSA) isolates
 - Gentamicin inhibited only 63.0% of the coagulase-negative staphylococci isolates tested at the CLSI breakpoint (data not shown)
- Plazomicin displayed limited activity against *Acinetobacter* spp. ($MIC_{50/90}$, 8/>128 µg/mL), *Enterococcus* spp. ($MIC_{50/90}$, 32/64 µg/mL), and *Streptococcus pneumoniae* ($MIC_{50/90}$, 32/64 µg/mL)
- The activity of plazomicin against *Enterobacteriaceae* was similar in the different geographic regions with an MIC_{50} value of 0.5 µg/mL for all regions and MIC_{90} values ranging from 1 to 2 µg/mL
 - Variability in plazomicin activity against CRE isolates was noted among the different geographic regions analyzed with greater activity against isolates from North America and Europe when compared to Asia-Pacific and Latin America (Figure 4)

Conclusions

- Plazomicin inhibited 99.0% of the *Enterobacteriaceae* isolates, including CRE isolates, and >90.0% of the isolates resistant to gentamicin and tobramycin
 - Additionally, plazomicin inhibited >40% of the amikacin-resistant *Enterobacteriaceae*
- Plazomicin was less active against *P. aeruginosa* isolates when compared to *Enterobacteriaceae*
- Plazomicin demonstrated activity against *Staphylococcus* spp., including MRSA
- These results corroborate previous reports from the literature that describe plazomicin as a potential agent for the treatment of serious infections caused by *Enterobacteriaceae* isolates, including CRE, in patients who have limited or no alternative treatment options

Acknowledgements

This study was performed by JMI Laboratories and supported by Achaogen, which included funding for services related to preparing this poster.

This project has been funded in whole or in part with federal funds from the Biomedical Advanced Research and Development Authority, Office of the Assistant Secretary for Preparedness and Response, Office of the Secretary, Department of Health and Human Services, under Contract No. HHSO100201000046C.

References

Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition.* Wayne, PA: CLSI.

EUCAST (2018). Breakpoint tables for interpretation of MICs and zone diameters. Version 8.0, January 2018. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf. Accessed January 2018.

Walkty A, Adam H, Baxter M, et al. (2014). In vitro activity of plazomicin against 5,015 gram-negative and gram-positive clinical isolates obtained from patients in Canadian hospitals as part of the CANWARD study, 2011-2012. *Antimicrob Agents Chemother* 58: 2554-2563.

Armstrong ES, Miller GH. (2010). Combating evolution with intelligent design: the neoglycoside ACHN-490. *Curr Opin Microbiol* 13:565-573.

Karaiskos I, Souli M, Giamarellou H. (2015). Plazomicin: an investigational therapy for the treatment of urinary tract infections. *Expert Opin Investig Drugs* 24:1501-1511.

Zhanel GG, Lawson CD, Zelenitsky S, et al. (2012). Comparison of the next-generation aminoglycoside plazomicin to gentamicin, tobramycin and amikacin. *Expert Rev Anti Infect Ther* 10:459-473.