ASM Microbe 2019 | Poster Sunday-AAR-594

Activity of Meropenem-Vaborbactam and Characterization of Carbapenem Resistance Mechanisms among Carbapenem-Resistant *Enterobacteriaceae* from United States Hospitals (2016–2017)

Mariana Castanheira, Timothy B. Doyle, Valerie Kantro, Rodrigo E. Mendes, Dee Shortridge
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Carbapenems have been used for many years to treat serious infections caused by multidrug-resistant (MDR) *Enterobacteriaceae* isolates producing β-lactamases
- Using carbapenems resulted in the emergence of carbapenem-resistant isolates that include isolates producing KPCs, OXA-48, metallo-β-lactamases (MBLs) and those harboring permeability alterations and a β-lactamase that poorly hydrolyzes carbapenems
- In the United States (USA), isolates producing KPC enzymes have been detected in all states and isolates producing these enzymes are commonly detected in the New York City area and Texas
- Vaborbactam is a cyclic boronic acid β-lactamase inhibitor that has activity against Ambler class A, including KPC, and class C enzymes
 - Vaborbactam has been combined with meropenem and enhances the activity of this carbapenem against KPC-producing isolates when compared to meropenem tested alone
- Meropenem-vaborbactam has been approved in the USA and Europe for the treatment of complicated urinary tract infections
- We evaluated the activity of meropenem-vaborbactam and comparators against >9,000 *Enterobacteriaceae* isolates from US hospitals collected during 2016-2017 and characterized the carbapenemases carried by 105 carbapenem-resistant *Enterobacteriaceae* (CRE) isolates

Materials and Methods

- A total of 9,295 *Enterobacteriaceae* clinical isolates collected during 2016 (n=4,942) and 2017 (n=4,353) from 30 hospitals located in the USA were included in the study
- Isolates were limited to 1 per patient episode and were collected from bloodstream infections (n=2,548), intra-abdominal infections (n=796), pneumonia in hospitalized patients (n=1,846), skin and skin structure infections (n=1,340), urinary tract infections (n=2,690), and other sources (n=75)
- Species identification was confirmed by matrix-assisted laser desorption ionization-time of flight mass spectrometry when necessary and for all CRE isolates
- Isolates were susceptibility tested against meropenem-vaborbactam (inhibitor at fixed 8 µg/mL) and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI)
 - Quality control (QC) was performed according to CLSI guidelines (M100, 2019), and all QC MIC results were within acceptable ranges, as published in CLSI documents
 - Categorical interpretations for all comparator agents were those found in CLSI criteria in M100 (2019), EUCAST breakpoint tables (2019), and/or the US Food and Drug Administration (FDA) website (https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria)
- CRE was defined as any isolate exhibiting doripenem, imipenem, and/or meropenem MIC values at ≥2 µg/mL (*Proteus mirabilis* and indole-positive Proteeae used only meropenem due to intrinsically elevated imipenem MIC values)
 - CRE isolates were submitted to whole genome sequencing on a MiSeq (Illumina, San Diego, California, USA) instrument targeting a 30X coverage
 - Sequences were *de novo* assembled and searched for the presence of acquired carbapenemases using a curated library and applying criteria of >94% sequencing identity and 40% minimum length coverage
- MDR and extensively drug-resistant (XDR) isolates were defined as any isolate nonsusceptible (CLSI criteria) to ≥1 agent in ≥3 or ≥5, respectively, of the following antimicrobial classes: broad-spectrum cephalosporins, carbapenems, broad-spectrum penicillin combined with a β-lactamase-inhibitor, fluoroquinolones, aminoglycosides, glycylcyclines, and the polymyxins

Results

- Among 9,295 *Enterobacteriaceae* isolates collected at US hospitals in 2016 and 2017, a total of 105 (1.1%) were CREs, 749 (8.1%) were MDR, and 88 (0.9%) were XDR
 - Different rates for these resistant isolates were noted in the US census divisions (Figure 1)
 - Overall CRE rates ranged from 0% to 4.1%, MDR rates ranged from 3.2% to 14.0%, and XDR rates ranged from 0% to 3.3%
 - The Middle Atlantic division displayed higher rates of CRE, MDR, and XDR rates whereas the lowest rates were observed in the West North Central division
- Meropenem-vaborbactam inhibited >99.9% of the *Enterobacteriaceae* isolates at the US FDA susceptibility breakpoint of ≤4/8 µg/mL (Figure 2)
 - Meropenem alone inhibited 98.8% and other comparators inhibited 81.8% to 99.4% of these isolates
- Against 105 CRE isolates, meropenem-vaborbactam inhibited 99.0% of the isolates, while meropenem alone inhibited only 3.8%
 - CRE isolates were mainly *Klebsiella pneumoniae* (53/105), but also *Enterobacter cloacae* (19/105) and other 7 species (Figure 3)
 - Tigecycline and colistin were the only comparators to inhibit >80% of the CRE isolates (Figure 2)
- Among the CRE isolates, 94 (89.5%) carried carbapenemase genes that included 53 bla_{KPC-3}, 36 bla_{KPC-2}, 3 bla_{SME-4}, 1 bla_{KPC-4}, and 1 $bla_{OXA-232}$ (Figure 3)
 - Carbapenemase-producing isolates were observed in all census divisions, except West North Central where no CREs were noted
 - No carbapenemases were found in 11 CRE isolates
- Meropenem-vaborbactam inhibited all but 1 of the carbapenemase-producing isolates at ≤4/8 µg/mL (98.9% susceptible; data not shown)
 - The *K. pneumoniae* isolate carrying $bla_{OXA-232}$ displayed a meropenem-vaborbactam MIC at 8/8 µg/mL
- Meropenem-vaborbactam inhibited 99.9% of the 749 MDR (resistant to ≥3 drug classes) whereas meropenem inhibited 85.7% of these isolates and other compounds inhibited 12.6% to 92.7%
- Against XDR (resistant to ≥5 drug classes) isolates, meropenem-vaborbactam inhibited 98.9% of the 88 isolates while meropenem inhibited 31.8% and other comparators inhibited 0.0% to 76.1%

Conclusions

- Although the rates of resistant isolates varied in the US census divisions, MDR *Enterobacteriaceae* isolates were noted in all of them, and CRE and XDR isolates were noted in all but the West North Central division
- Meropenem-vaborbactam was the most active agent against CRE, MDR, and XDR isolates that have limited therapeutic options
- Meropenem-vaborbactam demonstrated an excellent safety profile in recent clinical trials, which when coupled with its potent activity, makes this combination an attractive agent for the treatment of these challenging organisms tested in this study that seem widespread in US hospitals

Acknowledgements

This research was funded by Melinta Therapeutics.

References

Castanheira M, Rhomberg PR, Flamm RK, Jones RN (2016). Effect of the beta-Lactamase inhibitor vaborbactam combined with meropenem against serine carbapenemase-producing *Enterobacteriaceae*. *Antimicrob Agents Chemother* 60: 5454–5458.

Castanheira M, Huband MD, Mendes RE, et al. (2017). Meropenem-vaborbactam tested against contemporary Gram-negative isolates collected worldwide during 2014, including carbapenem-resistant, KPC-producing, multidrug-resistant, and extensively drug-resistant *Enterobacteriaceae*. *Antimicrob Agents Chemother* 61: e00567.

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard- tenth edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement*. Wayne, PA: CLSI.

EUCAST (2019). Breakpoint tables for interpretation of MICs and zone diameters. Version 9.0, January 2019. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_9.0_Breakpoint_Tables.pdf. Accessed

Kaye KS, Bhowmick T, Metallidis S, et al. (2018). Effect of meropenem-vaborbactam vs piperacillin-tazobactam on clinical cure or improvement and microbial eradication in complicated urinary tract infection: The TANGO I Randomized Clinical Trial. *JAMA* 319: 788–799.

Pfaller MA, Huband MD, Mendes RE, et al. (2018). In vitro activity of meropenem-vaborbactam and characterisation of carbapenem resistance mechanisms among carbapenem-resistant Enterobacteriaceae from the 2015 meropenem/vaborbactam surveillance programme. *Int J Antimicrob Agents* 52: 144–150.

Sun D, Rubio-Aparicio D, Nelson K, et al. (2017). Meropenem-vaborbactam resistance selection, resistance prevention, and molecular mechanisms in mutants of KPC-producing *Klebsiella pneumoniae*. *Antimicrob Agents Chemother* 61: e01694.

Figure 1 Distribution of CRE, MDR, and XDR *Enterobacteriaceae* isolates in US census divisions

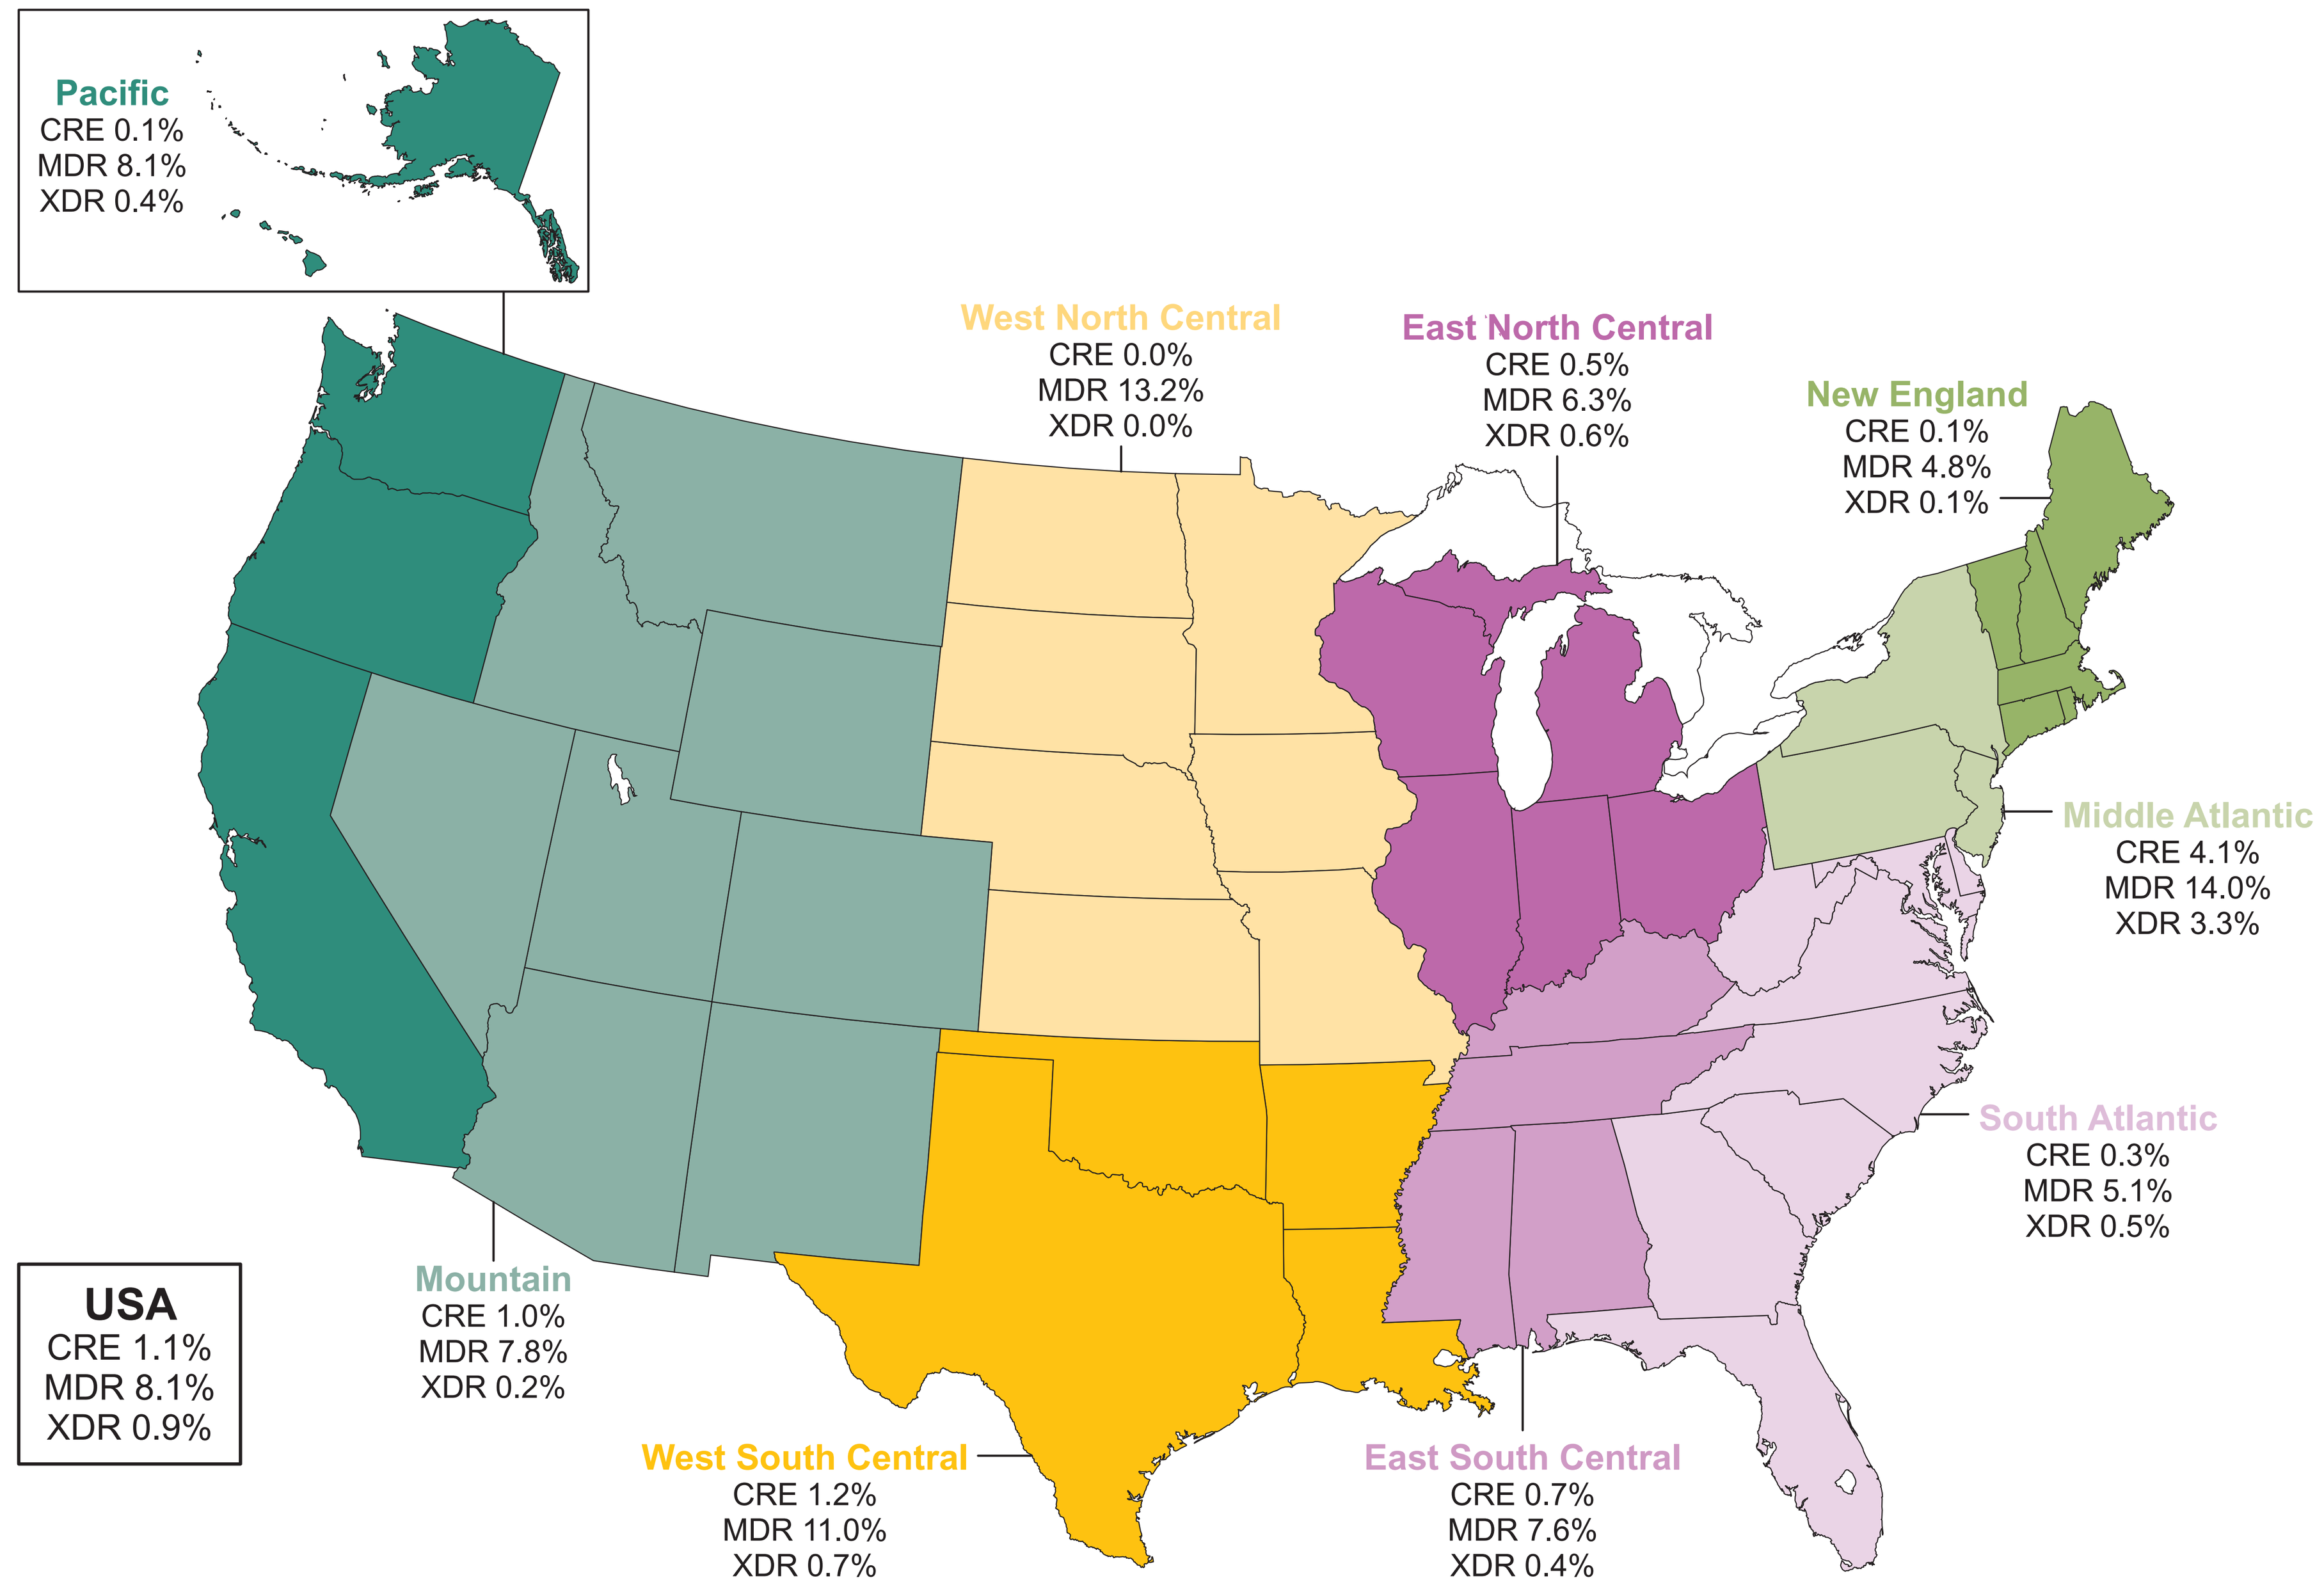
Pacific
CRE 0.1%
MDR 8.1%
XDR 0.4%
West North Central
CRE 0.0%
MDR 13.2%
XDR 0.0%
East North Central
CRE 0.5%
MDR 6.3%
XDR 0.6%
New England
CRE 0.1%
MDR 4.8%
XDR 0.1%
Middle Atlantic
CRE 4.1%
MDR 14.0%
XDR 3.3%
South Atlantic
CRE 0.3%
MDR 5.1%
XDR 0.5%
USA
CRE 1.1%
MDR 8.1%
XDR 0.9%
Mountain
CRE 1.0%
MDR 7.8%
XDR 0.2%
West South Central
CRE 1.2%
MDR 11.0%
XDR 0.7%
East South Central
CRE 0.7%
MDR 7.6%
XDR 0.4%

CRE, carbepenem-resistant *Enterobacteriaceae*; MDR, multidrug-resistant; XDR, extensively drug-resistant.

Figure 2 Activity of meropenem-vaborbactam and comparator agents tested against US *Enterobacteriaceae* isolates

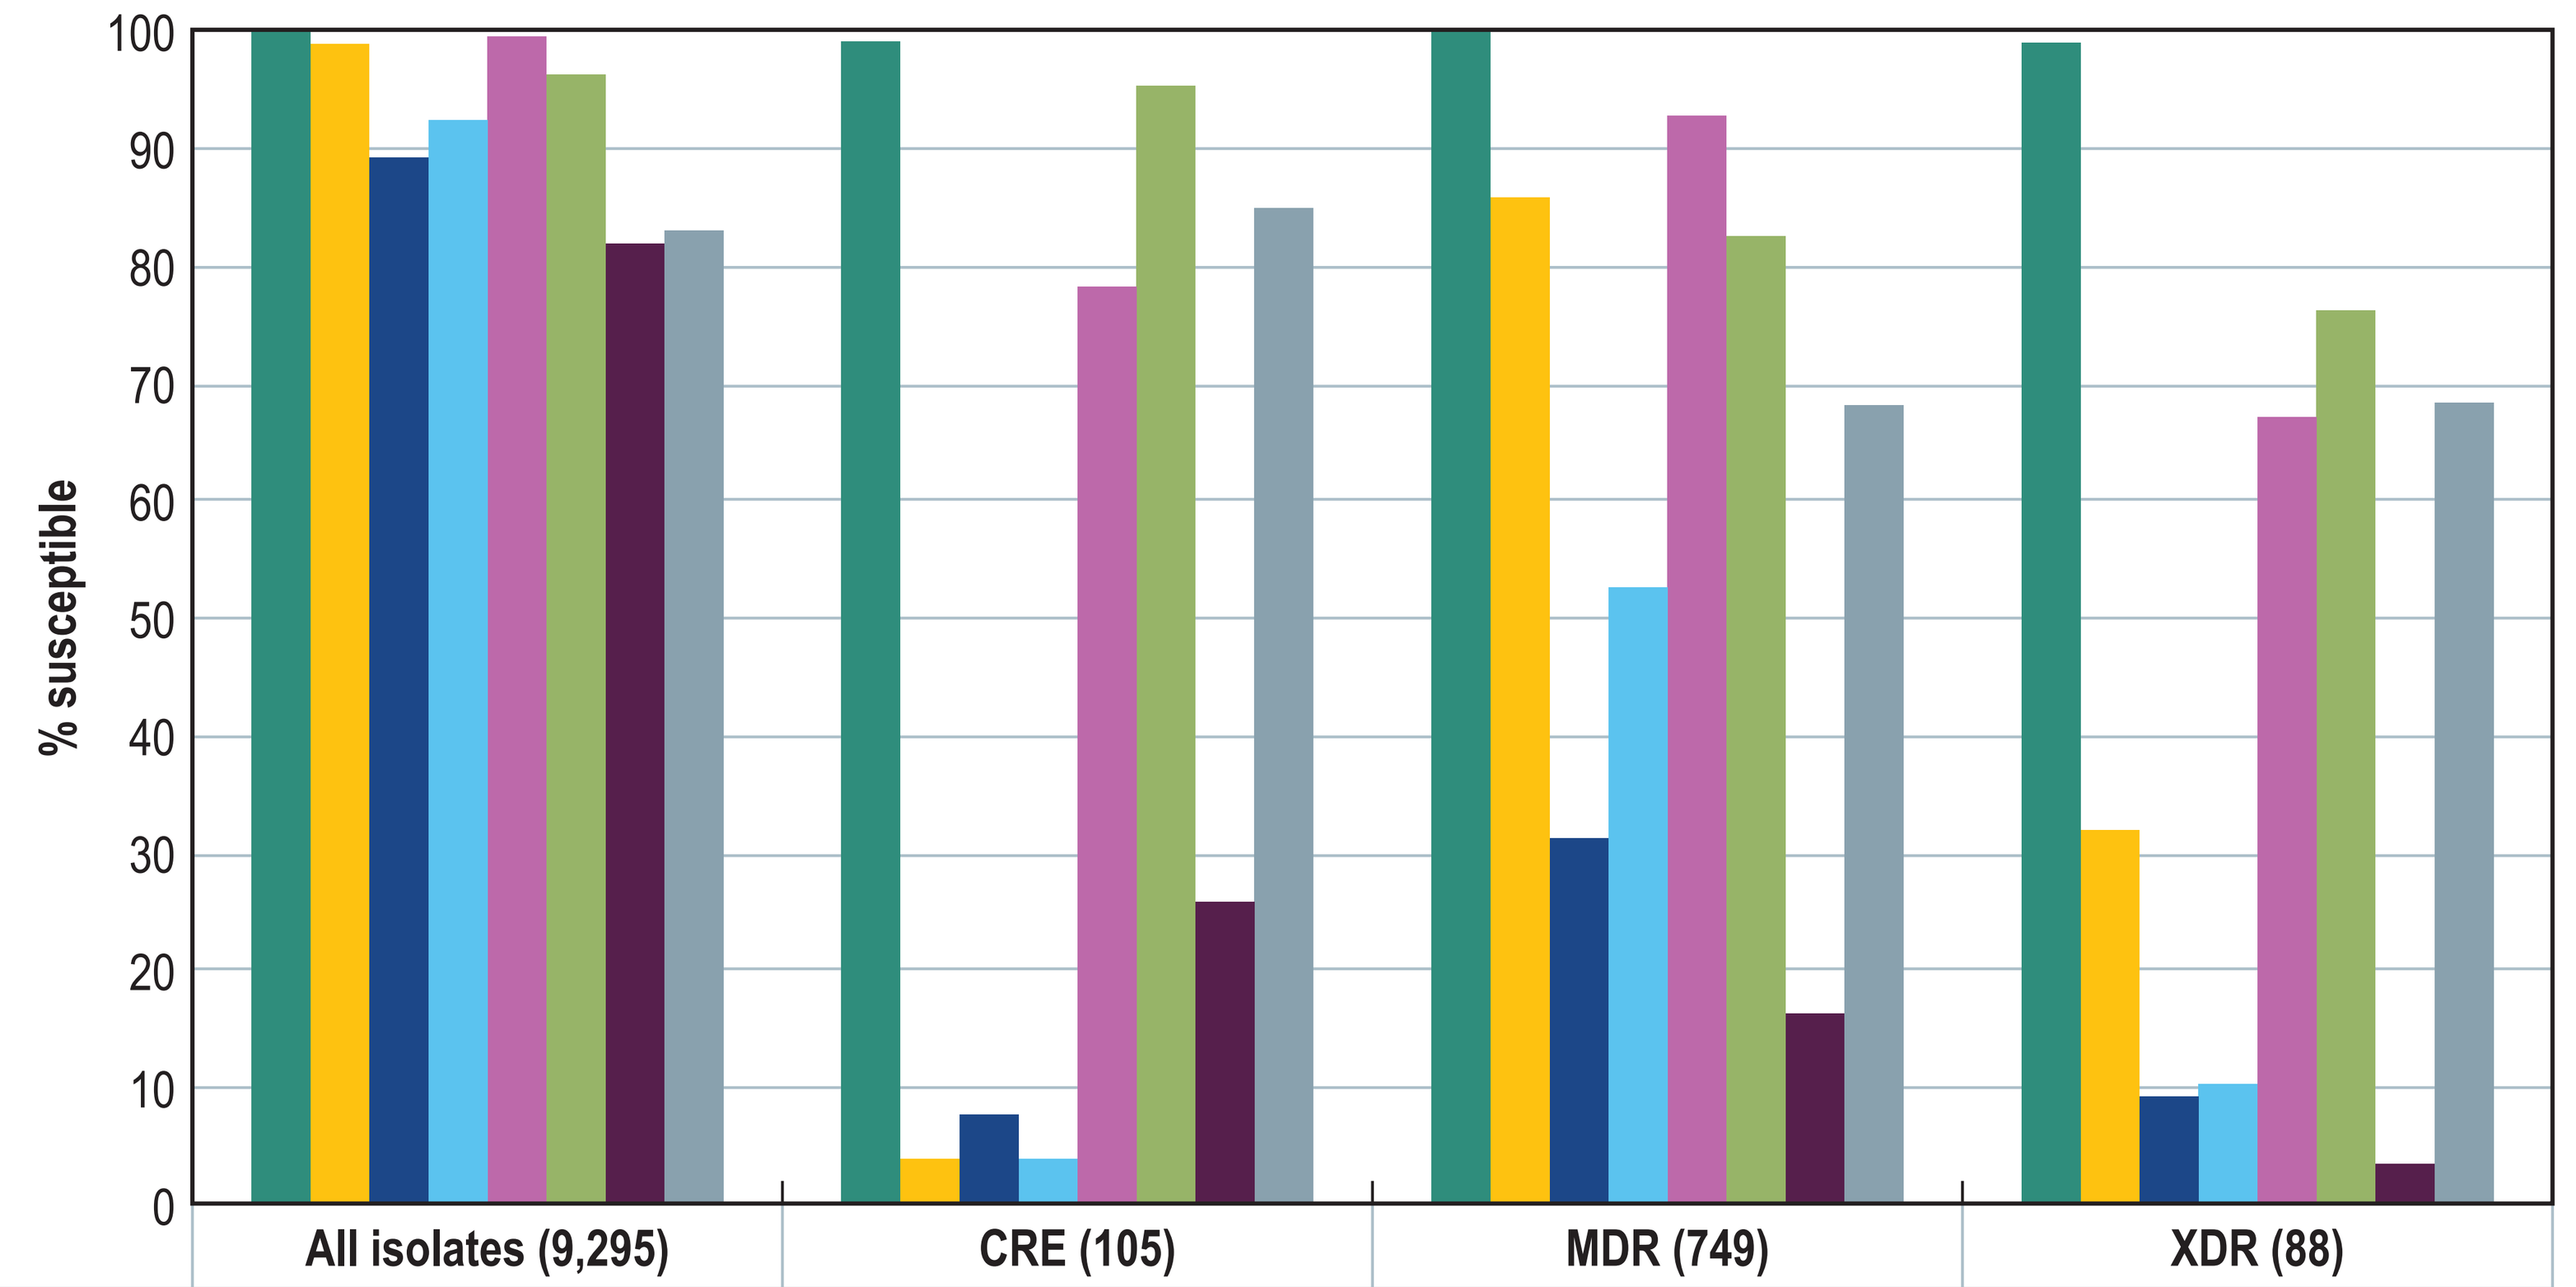
% susceptible
100
90
80
70
60
50
40
30
20
10
0

	All isolates (9,295)	CRE (105)	MDR (749)	XDR (88)
Meropenem-vaborbactam	>99.9	99.0	99.9	98.9
Meropenem	98.8	3.8	85.7	31.8
Cefepime	89.1	7.6	31.1	9.1
Piperacillin-tazobactam	92.3	3.8	52.5	10.2
Amikacin	99.4	78.1	92.7	67.0
Tigecycline[a]	96.2	95.2	82.4	76.1
Levofloxacin	81.8	25.7	16.2	3.4
Colistin[a]	82.9	84.8	68.0	68.2

[a] US FDA breakpoints were used for tigecycline and EUCAST breakpoints were applied for colistin.
CRE, carbapenem-resistant *Enterobacteriaceae*; MDR, multidrug-resistant; XDR, extensively drug-resistant.

Figure 3 Carbapenemase-producing and carbapenemase-negative *Enterobacteriaceae* isolates from US hospital

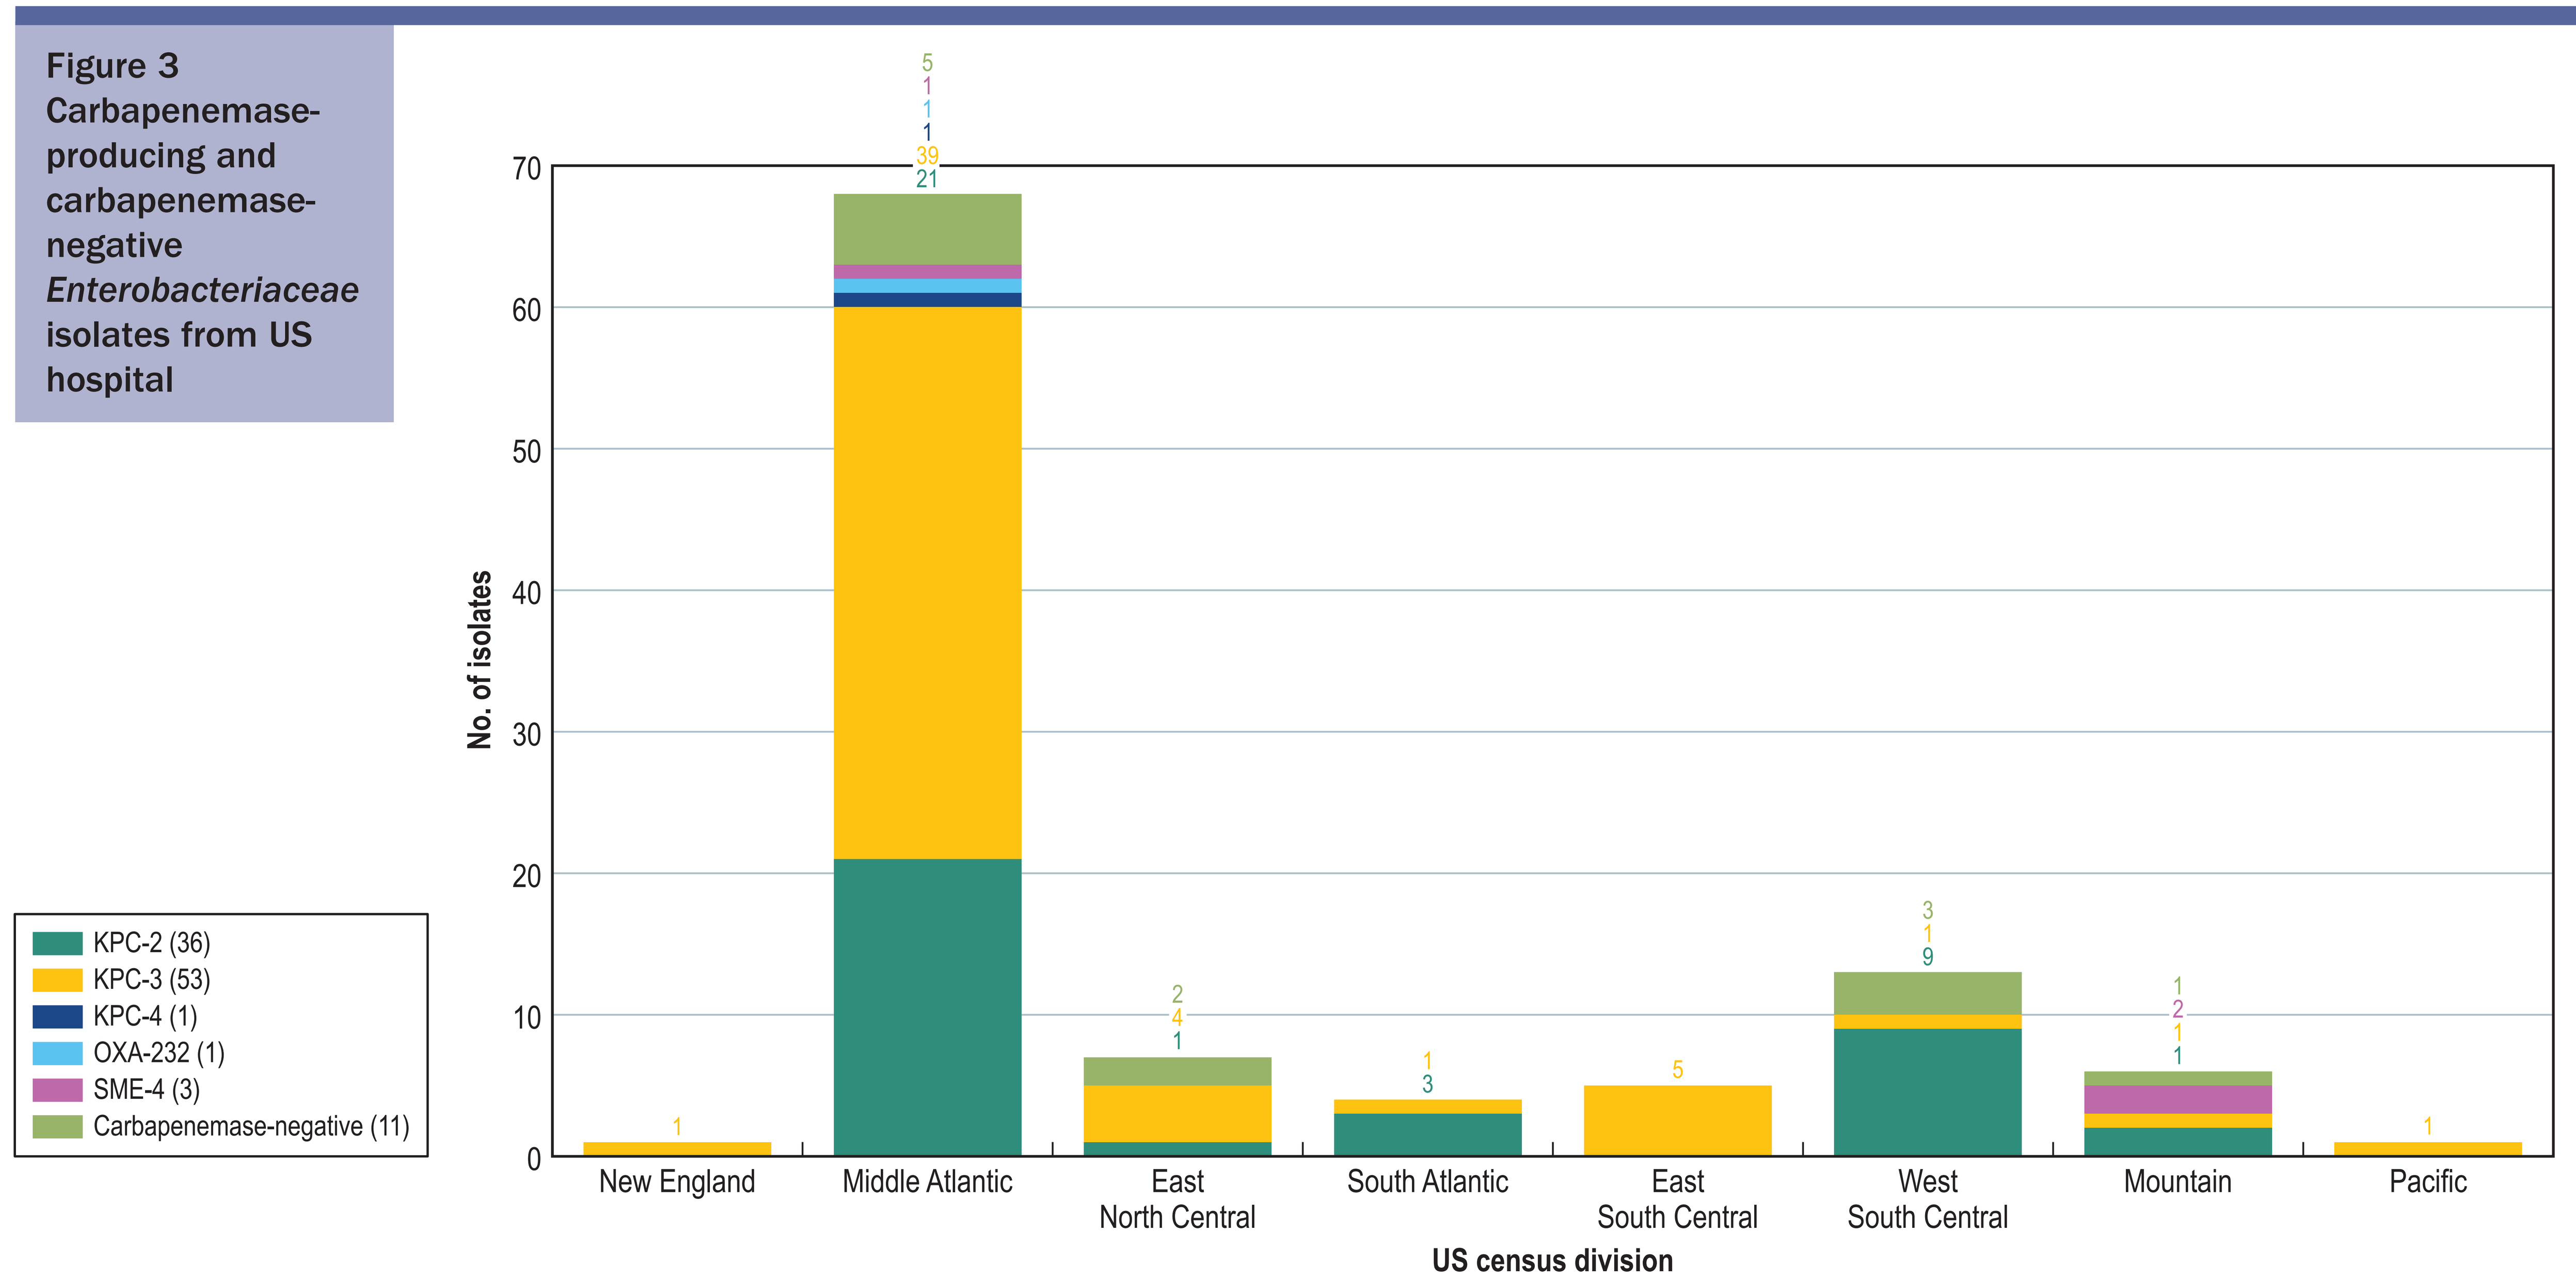
KPC-2 (36)
KPC-3 (53)
KPC-4 (1)
OXA-232 (1)
SME-4 (3)
Carbapenemase-negative (11)
No. of isolates
70
60
50
40
30
20
10
0
1
5
1
1
1
39
21
2
4
1
1
3
5
3
1
9
1
2
1
1
1
New England
Middle Atlantic
East North Central
South Atlantic
East South Central
West South Central
Mountain
Pacific
US census division

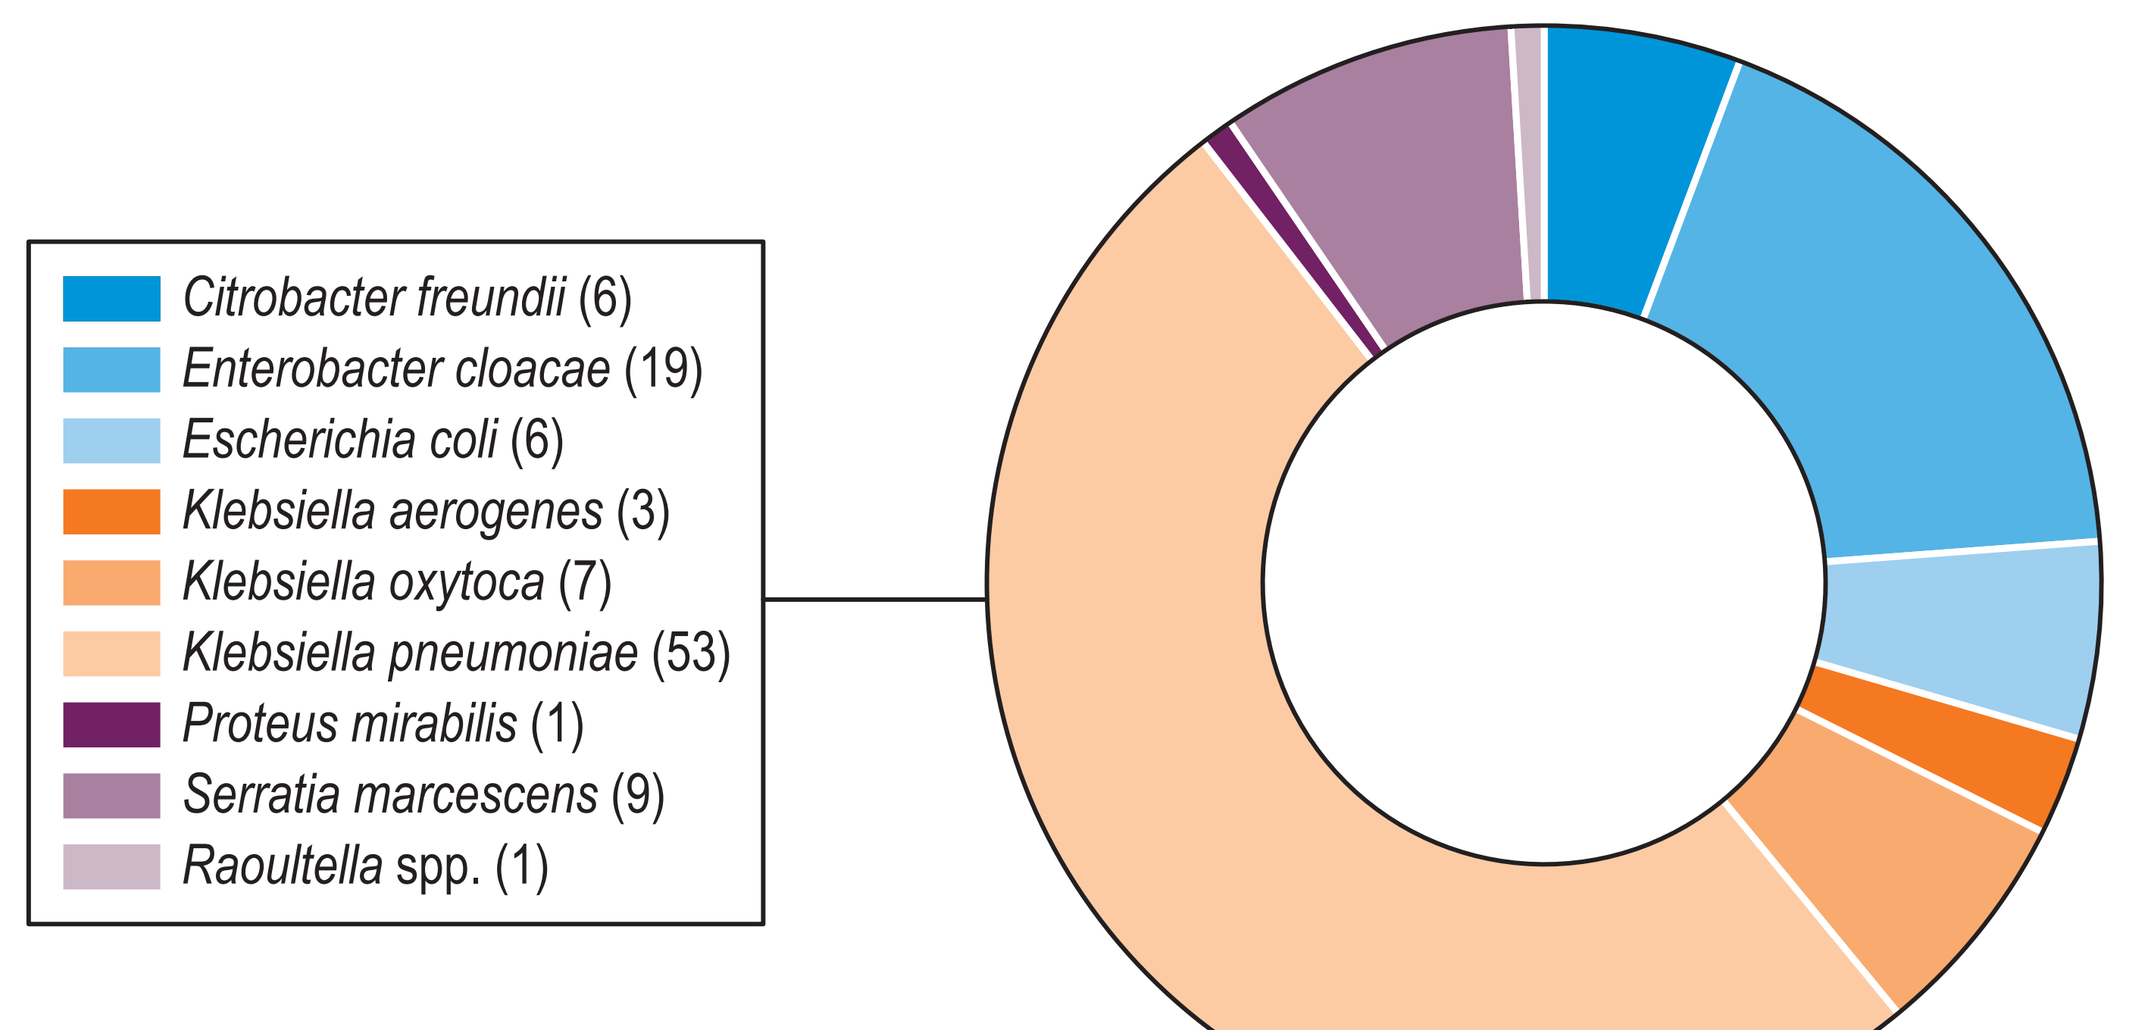
Citrobacter freundii (6)
Enterobacter cloacae (19)
Escherichia coli (6)
Klebsiella aerogenes (3)
Klebsiella oxytoca (7)
Klebsiella pneumoniae (53)
Proteus mirabilis (1)
Serratia marcescens (9)
Raoultella spp. (1)

Contact

Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ASM-Microbe19-meropenem-vaborbactam-CRE.pdf

Charges may apply. No personal information is stored.

ASM Microbe 2019, June 20–24, 2019, San Francisco, California