ASM Microbe 2019 | Poster Saturday-AAR-777

Monitoring the *In Vitro* Activity of Tebipenem, an Orally Available Carbapenem Agent, against a Current Collection of Surveillance *Enterobacterales* Clinical Isolates (2018)

Rodrigo E. Mendes, Helio S. Sader, Paul R. Rhomberg, Jill Lindley, Holly K. Huynh, Robert K. Flamm

JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Complicated and uncomplicated urinary tract infections (UTIs) are frequent in any given population
- Current guidelines recommend that empiric antibiotic therapy for UTIs should be based on local resistance data, drug availability, drug intolerance/allergy, and patient history
 - Nitrofurantoin or trimethoprim-sulfamethoxazole (TMP-SMX), if local resistance ≤20% can be used empirically for uncomplicated cystitis, while fluoroquinolones, ceftriaxone, aminoglycosides, and carbapenems are appropriate for pyelonephritis and complicated UTIs
- However, managing such infections is challenged by the dissemination of isolates producing extended-spectrum β-lactamases (ESBLs) and displaying cross-resistance to fluoroquinolones, aminoglycosides, and folate pathway inhibitors in the hospital and community settings
- Carbapenem agents represent good options for treating these patients with UTIs, who usually have a spectrum of illness severity, comorbidities, and clinical histories, and an oral carbapenem option would provide additional benefits
- Tebipenem, the active metabolite of tebipenem-pivoxil (SPR859), is an oral carbapenem introduced in Japan (2009) for pediatric respiratory and otolaryngologic infections
- SPR994 is a novel tebipenem formulation (tebipenem-pivoxil hydrobromide) that is under clinical development for treatment of UTIs
- This study evaluated the tebipenem activity and spectrum against targeted *Enterobacterales* species

Materials and Methods

Surveillance isolates

- *Enterobacterales* isolates included in this study were collected as part of the SENTRY Antimicrobial Surveillance Program
- A total of 1,031 isolates composed of target numbers of randomly selected species recovered from documented infections during 2018 were included (Table 1)
- Preference was given to isolates recovered from urinary tract samples (669 isolates; 64.9%), but those recovered from bloodstream infections (186 isolates; 18.0%) and lower respiratory tract of patients with pneumonia (176 isolates; 17.1%) were included, as well
- Isolates were collected in 113 medical centers located in 26 countries and 9 US census divisions: the United States (674 isolates [65.4%] from 62 medical centers), Europe (262 isolates [25.4%] from 32 medical centers), Latin America (44 isolates [4.3%] from 8 medical centers), and the Asia-Pacific region (51 isolates [4.9%] from 11 medical centers)
- Bacterial species were initially identified by the submitting laboratories and confirmed by JMI Laboratories using standard microbiology methods and matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany)

Antimicrobial susceptibility testing

- Susceptibility testing was performed for tebipenem and comparator agents by reference broth microdilution method
 - 96-well frozen-form broth microdilution panels with cation-adjusted Mueller-Hinton broth (CAMHB) were manufactured by JMI Laboratories (North Liberty, Iowa, USA) per the Clinical and Laboratory Standards Institute (CLSI) specifications described in the M07 (2018) document
 - Quality control strains were tested before and concomitantly with selected isolates, and bacterial inoculum density was monitored by counting the number of colony-forming units present in the inoculum material
 - Interpretations of MIC values obtained from clinical isolates used CLSI M100 (2019) breakpoint criteria, as available
- The ESBL phenotype was defined for *Escherichia coli*, *Klebsiella pneumoniae*, *Klebsiella oxytoca*, and *Proteus mirabilis* as an isolate that displayed MIC values ≥2 µg/mL for ceftriaxone, ceftazidime, and/or aztreonam

Results

- Overall, tebipenem showed MIC_{50} and MIC_{90} results of 0.06 µg/mL and 0.25 µg/mL, respectively, against all isolates in the study (Table 1)
 - These tebipenem ($MIC_{50/90}$, 0.06/0.25 µg/mL) MIC results were similar to meropenem ($MIC_{50/90}$, 0.06/0.12 µg/mL)
 - Isolates displaying tebipenem MIC values of >2 µg/mL were meropenem-nonsusceptible (Table 1)
- The lowest tebipenem MIC_{50} values (0.008 µg/mL) were obtained against *Citrobacter koseri* and *E. coli*, followed by *Citrobacter freundii*, *K. pneumoniae*, and *K. oxytoca* (MIC_{50}, 0.015 µg/mL) (Table 1)
- A tebipenem MIC_{50} value of 0.03 µg/mL was obtained against *Enterobacter cloacae* and *Enterobacter aerogenes* (Table 1), while the MIC_{50} value of 0.12 µg/mL was observed for other species tested (*Proteus* spp., *Morganella morganii*, *Providencia* spp., and *Serratia marcescens*)
- Agents other than carbapenems did not show high activity against *Enterobacterales*, except for piperacillin-tazobactam (91.0% susceptible) and the aminoglycosides (92.5–99.2% susceptible) (Figure 1)
- Carbapenems, amikacin, and tigecycline showed susceptibility rates >80% when tested against *Enterobacterales* isolates, meeting the MIC criteria for ESBL phenotype (Figure 2)
- Tebipenem MIC_{50} values within ±1 doubling dilution were obtained against the respective species of isolates with and without an ESBL phenotype (Table 2)
- Similar MIC_{50} results were observed for tebipenem when tested against *Enterobacterales* isolates showing nonsusceptible or susceptible phenotypes for levofloxacin or trimethoprim-sulfamethoxazole (Table 2)

Conclusions

- Tebipenem showed broad-spectrum and potent activity against this current and targeted collection of *Enterobacterales* clinical isolates and inhibited all but 14 isolates (98.6%) at ≤0.5 µg/mL
- Tebipenem was most active against *E. coli*, *K. pneumoniae*, *K. oxytoca*, *C. koseri*, and *C. freundii* (MIC_{50}, 0.008–0.015 µg/mL) followed by *Enterobacter* spp., *Proteus* spp., *M. morganii*, *Providencia* spp., and *S. marcescens* (MIC_{50}, 0.03–0.12 µg/mL)
- ESBL phenotype and/or nonsusceptibility to levofloxacin and/or trimethoprim-sulfamethoxazole did not adversely affect tebipenem activity
- These data suggest that tebipenem may be a convenient oral option for treating UTIs caused by carbapenem-susceptible *Enterobacterales* isolates and warrant its clinical development

Table 1 MIC distributions of tebipenem tested against surveillance *Enterobacterales* isolates

Species (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (µg/mL) of:													MIC_{50}	MIC_{90}
	≤0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4[a]	8[a]	>8[a]		
Enterobacterales (1,031)	2 0.2	113 11.2	174 28.0	161 43.6	195 62.6	229 84.8	125 96.9	18 98.6	0 98.6	1 98.7	3 99.0	2 99.2	8 100.0	0.06	0.25
E. coli (102)	1 1.0	67 66.7	30 96.1	4 100.0										0.008	0.015
C. koseri (55)	1 1.8	36 67.3	17 98.2	1 100.0										0.008	0.015
C. freundii (50)		4 8.0	32 72.0	11 94.0	2 98.0	0 98.0	0 98.0	0 98.0	0 98.0	0 98.0	0 98.0	1 100.0		0.015	0.03
K. pneumoniae (85)		1 1.2	42 50.6	34 90.6	4 95.3	1 96.5	0 96.5	0 96.5	0 96.5	0 96.5	0 96.5	0 96.5	3 100.0	0.015	0.03
K. oxytoca (16)			10 62.5	6 100.0										0.015	0.03
E. aerogenes (105)			15 14.3	44 56.2	35 89.5	9 98.1	2 100.0							0.03	0.12
E. cloacae (103)		5 4.9	27 31.1	25 55.3	26 80.6	9 89.3	5 94.2	0 94.2	0 94.2	0 94.2	2 96.1	0 96.1	4 100.0	0.03	0.25
M. morganii (107)				2 1.9	7 8.4	45 50.5	47 94.4	6 100.0						0.12	0.25
P. mirabilis (103)				5 4.9	15 19.4	39 57.3	38 94.2	6 100.0						0.12	0.25
P. rettgeri (49)				1 2.0	19 40.8	23 87.8	4 95.9	1 98.0	0 98.0	1 100.0				0.12	0.25
P. stuartii (50)				2 4.0	15 34.0	25 84.0	8 100.0							0.12	0.25
S. marcescens (103)			1 1.0	8 8.7	42 49.5	35 83.5	10 93.2	4 97.1	0 97.1	0 97.1	1 98.1	1 99.0	1 100.0	0.12	0.25

[a] Meropenem-nonsusceptible.

Table 2 MIC distributions of tebipenem tested against specific phenotypes

Phenotypic organism group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (µg/mL) of:													MIC_{50}	MIC_{90}
	≤0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4[a]	8[a]	>8[a]		
ESBL-phenotype *E. coli* (20)[b]		6 30.0	10 80.0	4 100.0										0.015	0.03
Non-ESBL-phenotype *E. coli* (82)	1 1.2	61 75.6	20 100.0											0.008	0.015
ESBL-phenotype *Klebsiella* spp. (25)[b]			12 48.0	10 88.0	0 88.0	0 88.0	0 88.0	0 88.0	0 88.0	0 88.0	0 88.0	0 88.0	3 100.0	0.03	>8
ESBL-phenotype carbapenem-susceptible *Klebsiella* spp. (22)[b]			12 54.5	10 100.0										0.015	0.03
Non-ESBL-phenotype *Klebsiella* spp. (76)		1 1.3	40 53.9	30 93.4	4 98.7	1 100.0								0.015	0.03
ESBL-phenotype *P. mirabilis* (7)[b]				1 14.3	1 28.6	3 71.4	2 100.0							0.12	
Non-ESBL-phenotype *P. mirabilis* (96)				4 4.2	14 18.8	36 56.2	36 93.8	6 100.0						0.12	0.25
Levofloxacin-susceptible *Enterobacterales* (894)[c]	1 0.1	93 10.5	157 28.1	148 44.6	179 64.7	191 86.0	103 97.5	16 99.3	0 99.3	1 99.4	1 99.6	0 99.6	4 100.0	0.06	0.25
Levofloxacin-non-susceptible *Enterobacterales* (135)	1 0.7	20 15.6	17 28.1	13 37.8	15 48.9	37 76.3	22 92.6	2 94.1	0 94.1	0 94.1	2 95.6	2 97.0	4 100.0	0.12	0.25
Trimethoprim-sulfamethoxazole-susceptible *Enterobacterales* (855)[d]	1 0.1	88 10.4	141 26.9	141 43.4	180 64.4	190 86.7	98 98.1	14 99.8	0 99.8	0 99.8	1 99.9	0 99.9	1 100.0	0.06	0.25
Trimethoprim-sulfamethoxazole-resistant *Enterobacterales* (176)	1 0.6	25 14.8	33 33.5	20 44.9	15 53.4	39 75.6	27 90.9	4 93.2	0 93.2	1 93.8	2 94.9	2 96.0	7 100.0	0.06	0.25

ESBL, extended-spectrum β-lactamase
[a] Meropenem-nonsusceptible.
[b] The ESBL phenotype was defined for *E. coli*, *K. pneumoniae*, *K. oxytoca*, and *P. mirabilis* as isolates that displayed MIC values of ≥2 µg/mL for ceftriaxone, ceftazidime, and/or aztreonam.
[c] Isolates with MIC results of ≤2 µg/mL.
[d] Isolates with MIC results of ≤2/38 µg/mL.

Figure 1 Antimicrobial activity of comparator agents tested against *Enterobacterales* and *Enterobacterales* displaying an extended-spectrum β-lactamase phenotype

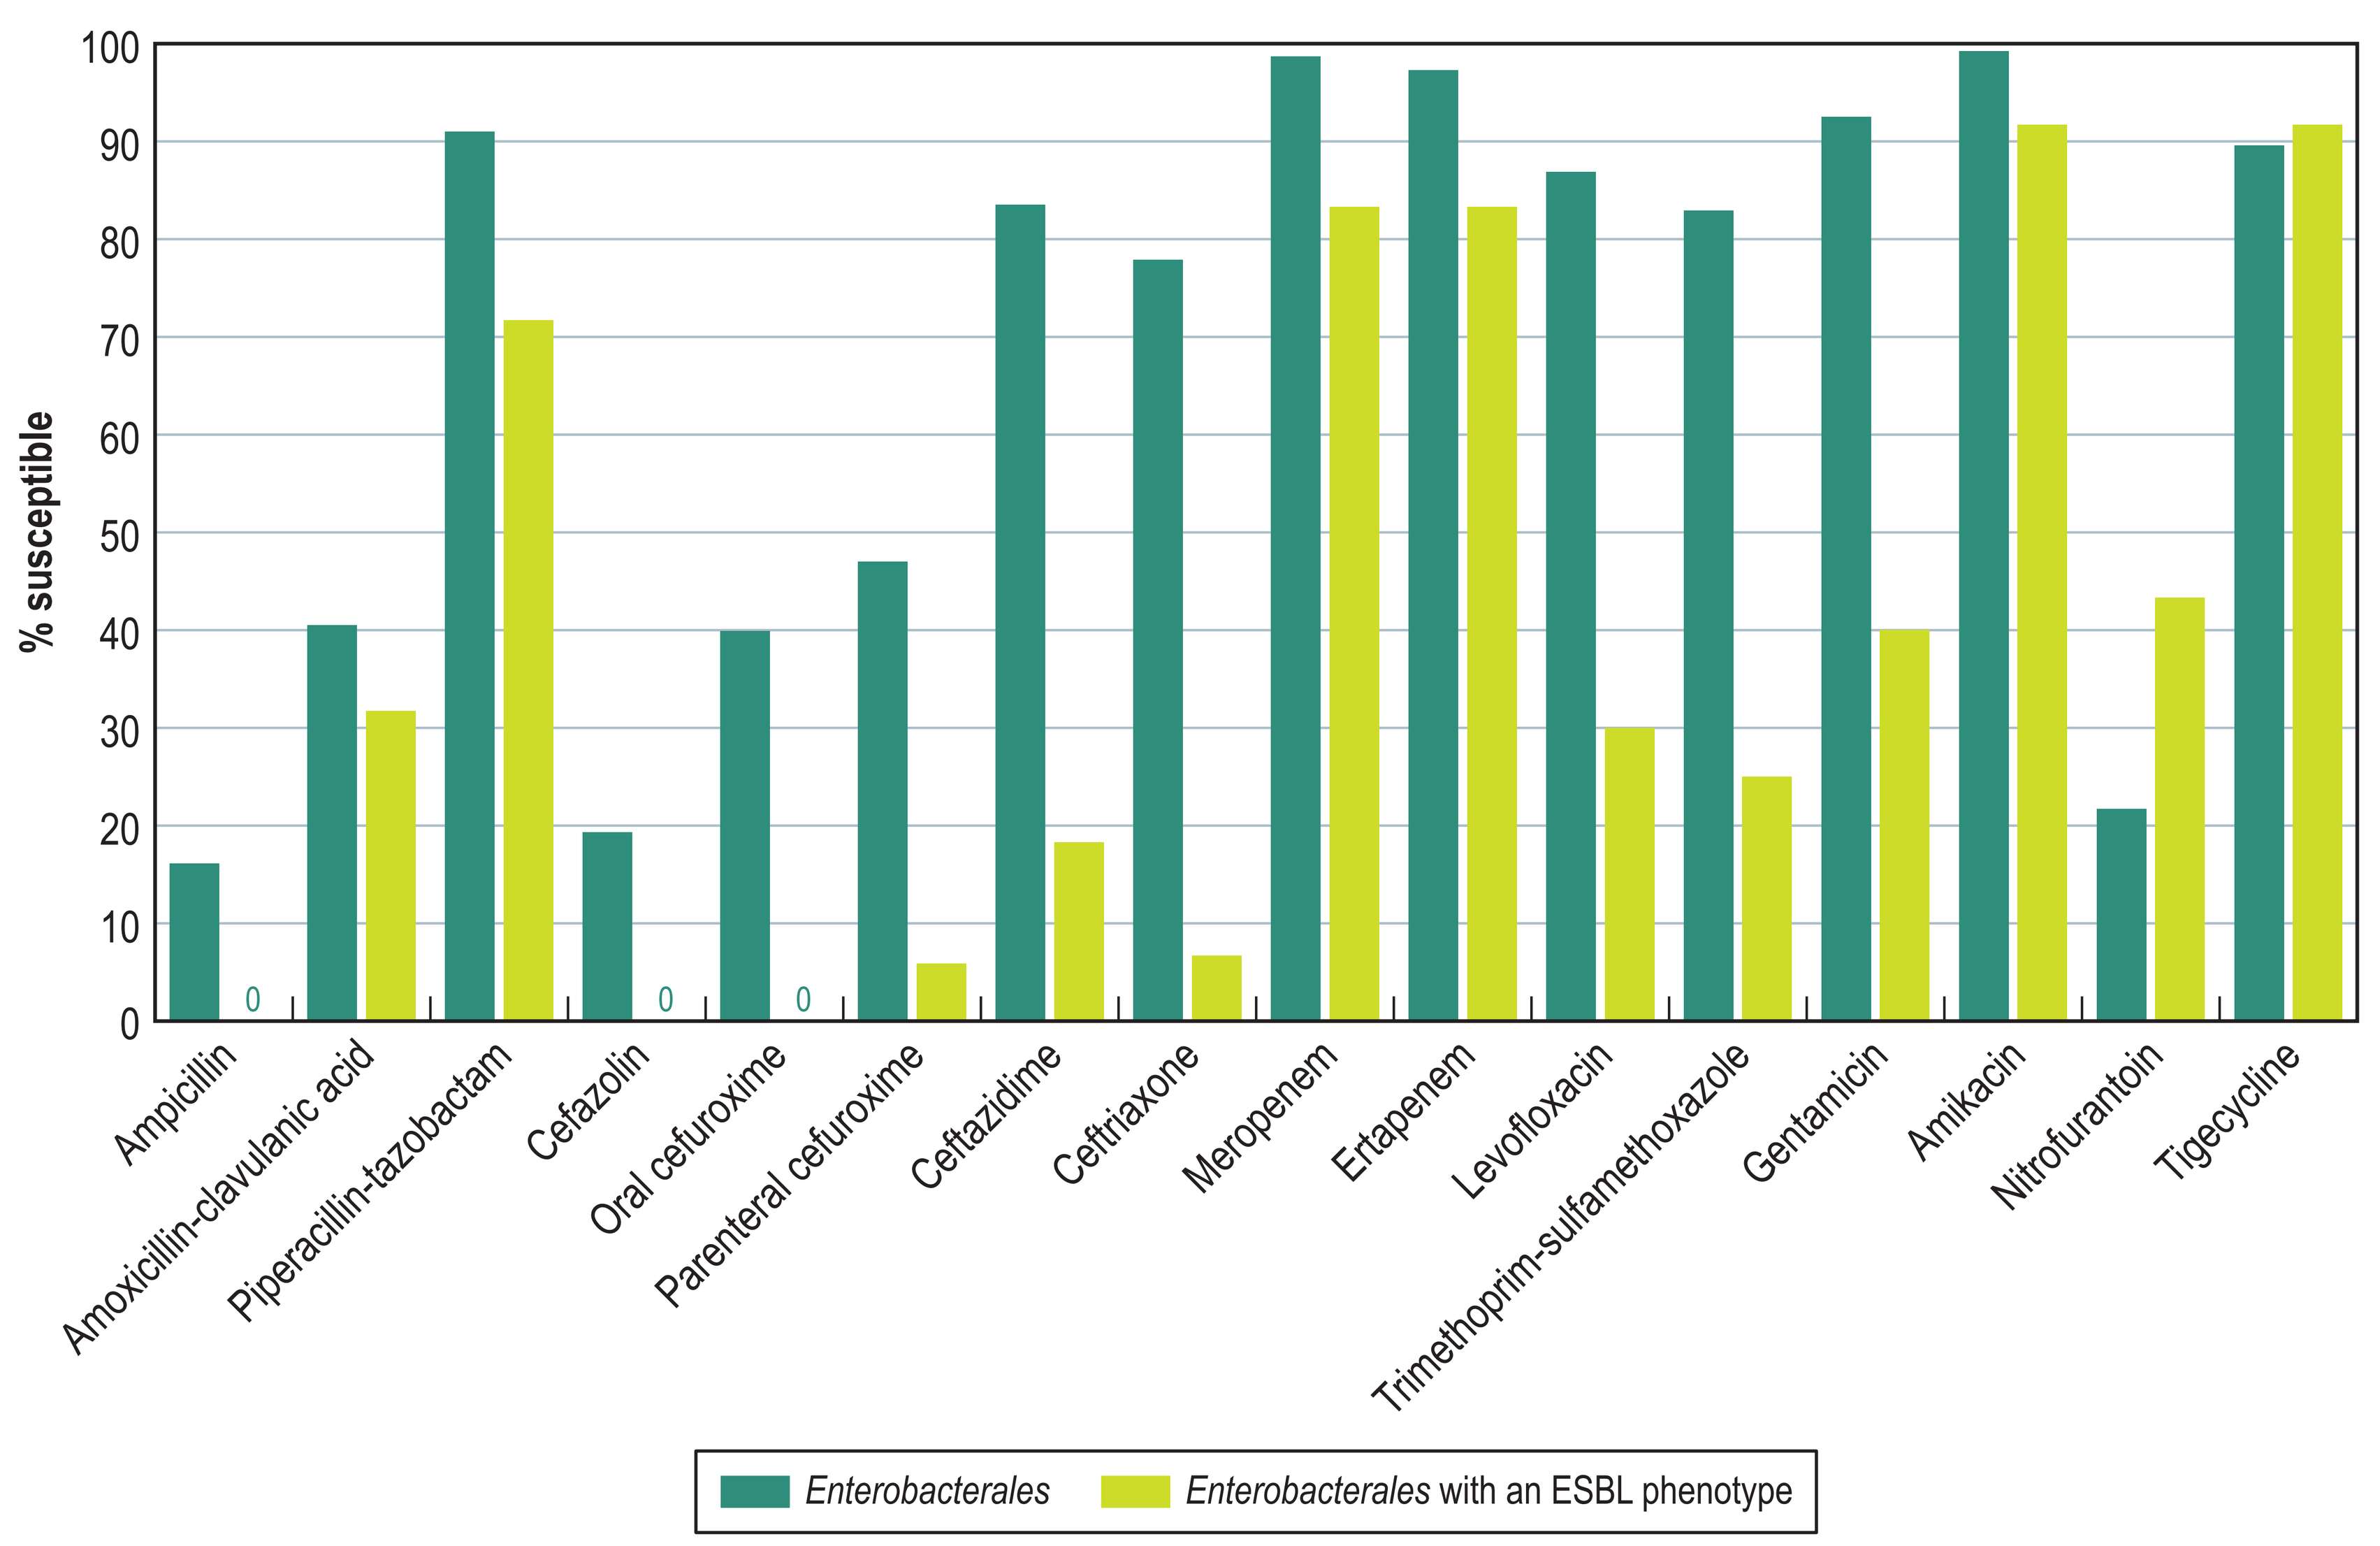
% susceptible
100
90
80
70
60
50
40
30
20
10
0
0
0
0
Ampicillin
Amoxicillin-clavulanic acid
Piperacillin-tazobactam
Cefazolin
Oral cefuroxime
Parenteral cefuroxime
Ceftazidime
Ceftriaxone
Meropenem
Ertapenem
Levofloxacin
Trimethoprim-sulfamethoxazole
Gentamicin
Amikacin
Nitrofurantoin
Tigecycline
Enterobacterales
Enterobacterales with an ESBL phenotype

Acknowledgements

This study and presentation were sponsored by Spero Therapeutics.

References

Gupta K, Hooton TM, Naber KG, et al. International clinical practice guidelines for the treatment of acute uncomplicated cystitis and pyelonephritis in women: A 2010 update by the Infectious Diseases Society of America and the European Society for Microbiology and Infectious Diseases. Clin Infect Dis 2011; 52:e103.

Khawcharoenporn T, Vasoo S, Singh K. Urinary Tract Infections due to Multidrug-Resistant Enterobacteriaceae: Prevalence and Risk Factors in a Chicago Emergency Department. Emerg Med Int 2013; 2013:258517.

United States Food and Drug Administration (2019). FDA-Recognized antimicrobial susceptibility test interpretive criteria. Available at https://www.fda.gov/drugs/development-resources /tigecycline-injection-products. Accessed April 2019.

Contact

Rodrigo E Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www .jmilabs.com/data/posters/ASM-Microbe19 -tebipenem-Enterobacterales.pdf

Charges may apply. No personal information is stored.

ASM Microbe 2019, June 20–24, 2019, San Francisco, California