ASM-Microbe 2019 | Poster Saturday-AAR-778

Evaluation of Tebipenem Activity Tested against a Collection of Isogenic *Escherichia coli* Strains Producing Various Clinically Relevant β-Lactamases

Lalitagauri M. Deshpande,[1] Ian A. Critchley,[2] Nicole Cotroneo,[2] Paul R. Rhomberg,[1] Robert K. Flamm,[1] Rodrigo E. Mendes[1]

[1] JMI Laboratories, North Liberty, Iowa, USA; [2] Spero Therapeutics, Cambridge, Massachusetts, USA

Introduction

- β-lactamases are important β-lactam resistance mechanisms in *Enterobacteriaceae*
- Carbapenems are widely used and potent antimicrobial agents with broad spectrum of activity, including against isolates producing extended-spectrum β-lactamases (ESBLs) and AmpC enzymes
- The high prevalence of fluoroquinolone- and extended-spectrum β-lactam-resistance among gram-negative pathogens in the hospital and community settings highlights the need for more effective therapies
- Tebipenem, the active metabolite of tebipenem-pivoxil, is an oral carbapenem introduced in Japan (2009) for pediatric respiratory and otolaryngologic infections and is under clinical development for treatment of complicated urinary tract infections (cUTIs) where it is administered as a prodrug, tebipenem pivoxil hydrobromide (SPR994)
- This study evaluates the *in vitro* antimicrobial activity of tebipenem and comparators when tested against a collection of isogenic *Escherichia coli* strains carrying clinically relevant and diverse β-lactamase-encoding genes

Materials and Methods

- Fifty *E. coli* isogenic strains carrying either native or recombinant plasmids containing narrow-spectrum β-lactamase (NSBL)-, ESBL-, AmpC-, or carbapenemase-encoding genes were included
- Isolates carrying engineered vectors providing different expression levels or combinations of β-lactamase genes were included, as well as genes encoding for mutations in the Ω loop of KPC-2
- *E. coli* laboratorial strains absent of β-lactamase genes were also included and tested to provide the respective baseline profile
- Susceptibility testing was performed by a reference broth microdilution method against tebipenem and comparators
 – 96-well frozen-form broth microdilution panels with cation-adjusted Mueller-Hinton broth were manufactured by JMI Laboratories (North Liberty, Iowa, USA) per the Clinical and Laboratory Standards Institute (CLSI) specifications described in the M07 (2018) document
 – Quality control strains that included *E. coli* ATCC 25922, *Klebsiella pneumoniae* ATCC BAA 1705, and *Pseudomonas aeruginosa* ATCC 27853 were tested before and concomitantly with selected isolates, and bacterial inoculum density was monitored by counting the number of colony-forming units present in the inoculum material
- Differences in fold MIC results for each antimicrobial agent tested against the baseline *E. coli* and respective isogenic strain carrying a β-lactamase-encoding gene were plotted in bar graphs (Figures 1 through 8)

Results

- Similar baseline MIC values for tebipenem (≤0.004–0.015 mg/L) and ertapenem (0.008–0.015 mg/L) were obtained against baseline laboratorial *E. coli* strains
- Tebipenem MIC results against strains producing narrow-spectrum SHV and TEM variants were similar to those obtained against isogenic baseline strains (≤4-fold differences; Figures 1 and 2)
 – MIC values for carbapenems other than tebipenem were generally also not affected by these NSBL enzymes, except for ertapenem (MIC, 0.06 mg/L) against SHV-1, for which the MIC was 8-fold higher than the baseline strain MIC (MIC, 0.008 mg/L) (Figure 1)
 – Cefazolin MIC values increased 64-, 16-, and 32-fold when tested against *E. coli* strains producing SHV-154, SHV-161, and TEM-2, respectively (Figure 1)
- Tebipenem MIC values against ESBLs were similar to those from baseline isogenic isolates regardless of ESBL variants (TEM, SHV, or CTX-M), transcription levels, or gene combinations (Figures 2 to 4)
 – Ertapenem MIC values increased 2- to 16-fold against isolates carrying TEM and SHV ESBL-encoding genes
- Tebipenem MIC values obtained against strains producing GES ESBL variants, PER-5, OXA-1, OXA-2, OXA-10, or plasmid-encoded AmpC enzymes remained similar to the respective baseline strain (Figures 5 and 6)
 – Ertapenem MIC values increased 16-fold when tested against a recombinant *E. coli* expressing OXA-10 (Figure 5)
- Tebipenem, ertapenem, and ceftazidime MIC values increased 8-fold against a strain producing the K1 *Klebsiella oxytoca* intrinsic enzyme compared to the baseline strain (Figure 5)
- Carbapenem MIC values were affected by NDM-1, KPC-2, KPC-3, SME-2, SME-4, OXA-48, and GES-11 (Figures 7 and 8)
 – The KPC-2 variants containing D179Y (KPC-33) or D176Y alterations had an effect over the MIC of carbapenem agents much lower than that observed for KPC-2 (Figure 7)

Conclusions

- The *in vitro* data presented here confirmed the activity of tebipenem against *E. coli* strains producing class A, C, or D non-carbapenemase enzymes, including ESBLs and plasmid-encoded AmpC enzymes
- Similar to other carbapenems, the tebipenem activity was adversely affected when tested against isolates expressing class A, B, and D carbapenemases
- Amino acids alterations (D176 or D179) in the Ω loop of KPC-2 restored the activity of carbapenem agents, including tebipenem
- These results confirm the stability of tebipenem against an array of β-lactamases and support its clinical development as an oral alternative for treating infections caused by isolates resistant to narrow- and extended-spectrum β-lactam agents

Acknowledgements

- The authors appreciate the expert technical assistance from Y. Edah and J. Lindley at JMI Laboratories. This study and poster presentation were supported by Spero Therapeutics.

References

- Arends SJR, Rhomberg PR, Cotroneo N, et al. (2019). Antimicrobial activity evaluation of tebipenem (SPR859), an orally available carbapenem, against a global set of *Enterobacteriaceae*, including a challenge set of organisms. *Antimicrob Agents Chemother* 63: e02618-18.
- Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard: Eleventh edition*. Wayne, PA, USA.
- Rice LB, Carias LL, Hujer AM, et al. (2000). High-level expression of chromosomally encoded SHV-1 beta-lactamase and an outer membrane protein change confer resistance to ceftazidime and piperacillin-tazobactam in a clinical isolate of *Klebsiella pneumoniae*. *Antimicrob Agents Chemother* 44: 362-367.
- Shields RK, Chen L, Cheng S, et al. (2017). Emergence of ceftazidime-avibactam resistance due to plasmid-borne bla_{KPC-3} mutations during treatment of carbapenem-resistant *Klebsiella pneumoniae* infections. *Antimicrob Agents Chemother* 61: e02097.

Figure 1 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying narrow-spectrum β-lactamase-encoding genes compared to the respective baseline strain

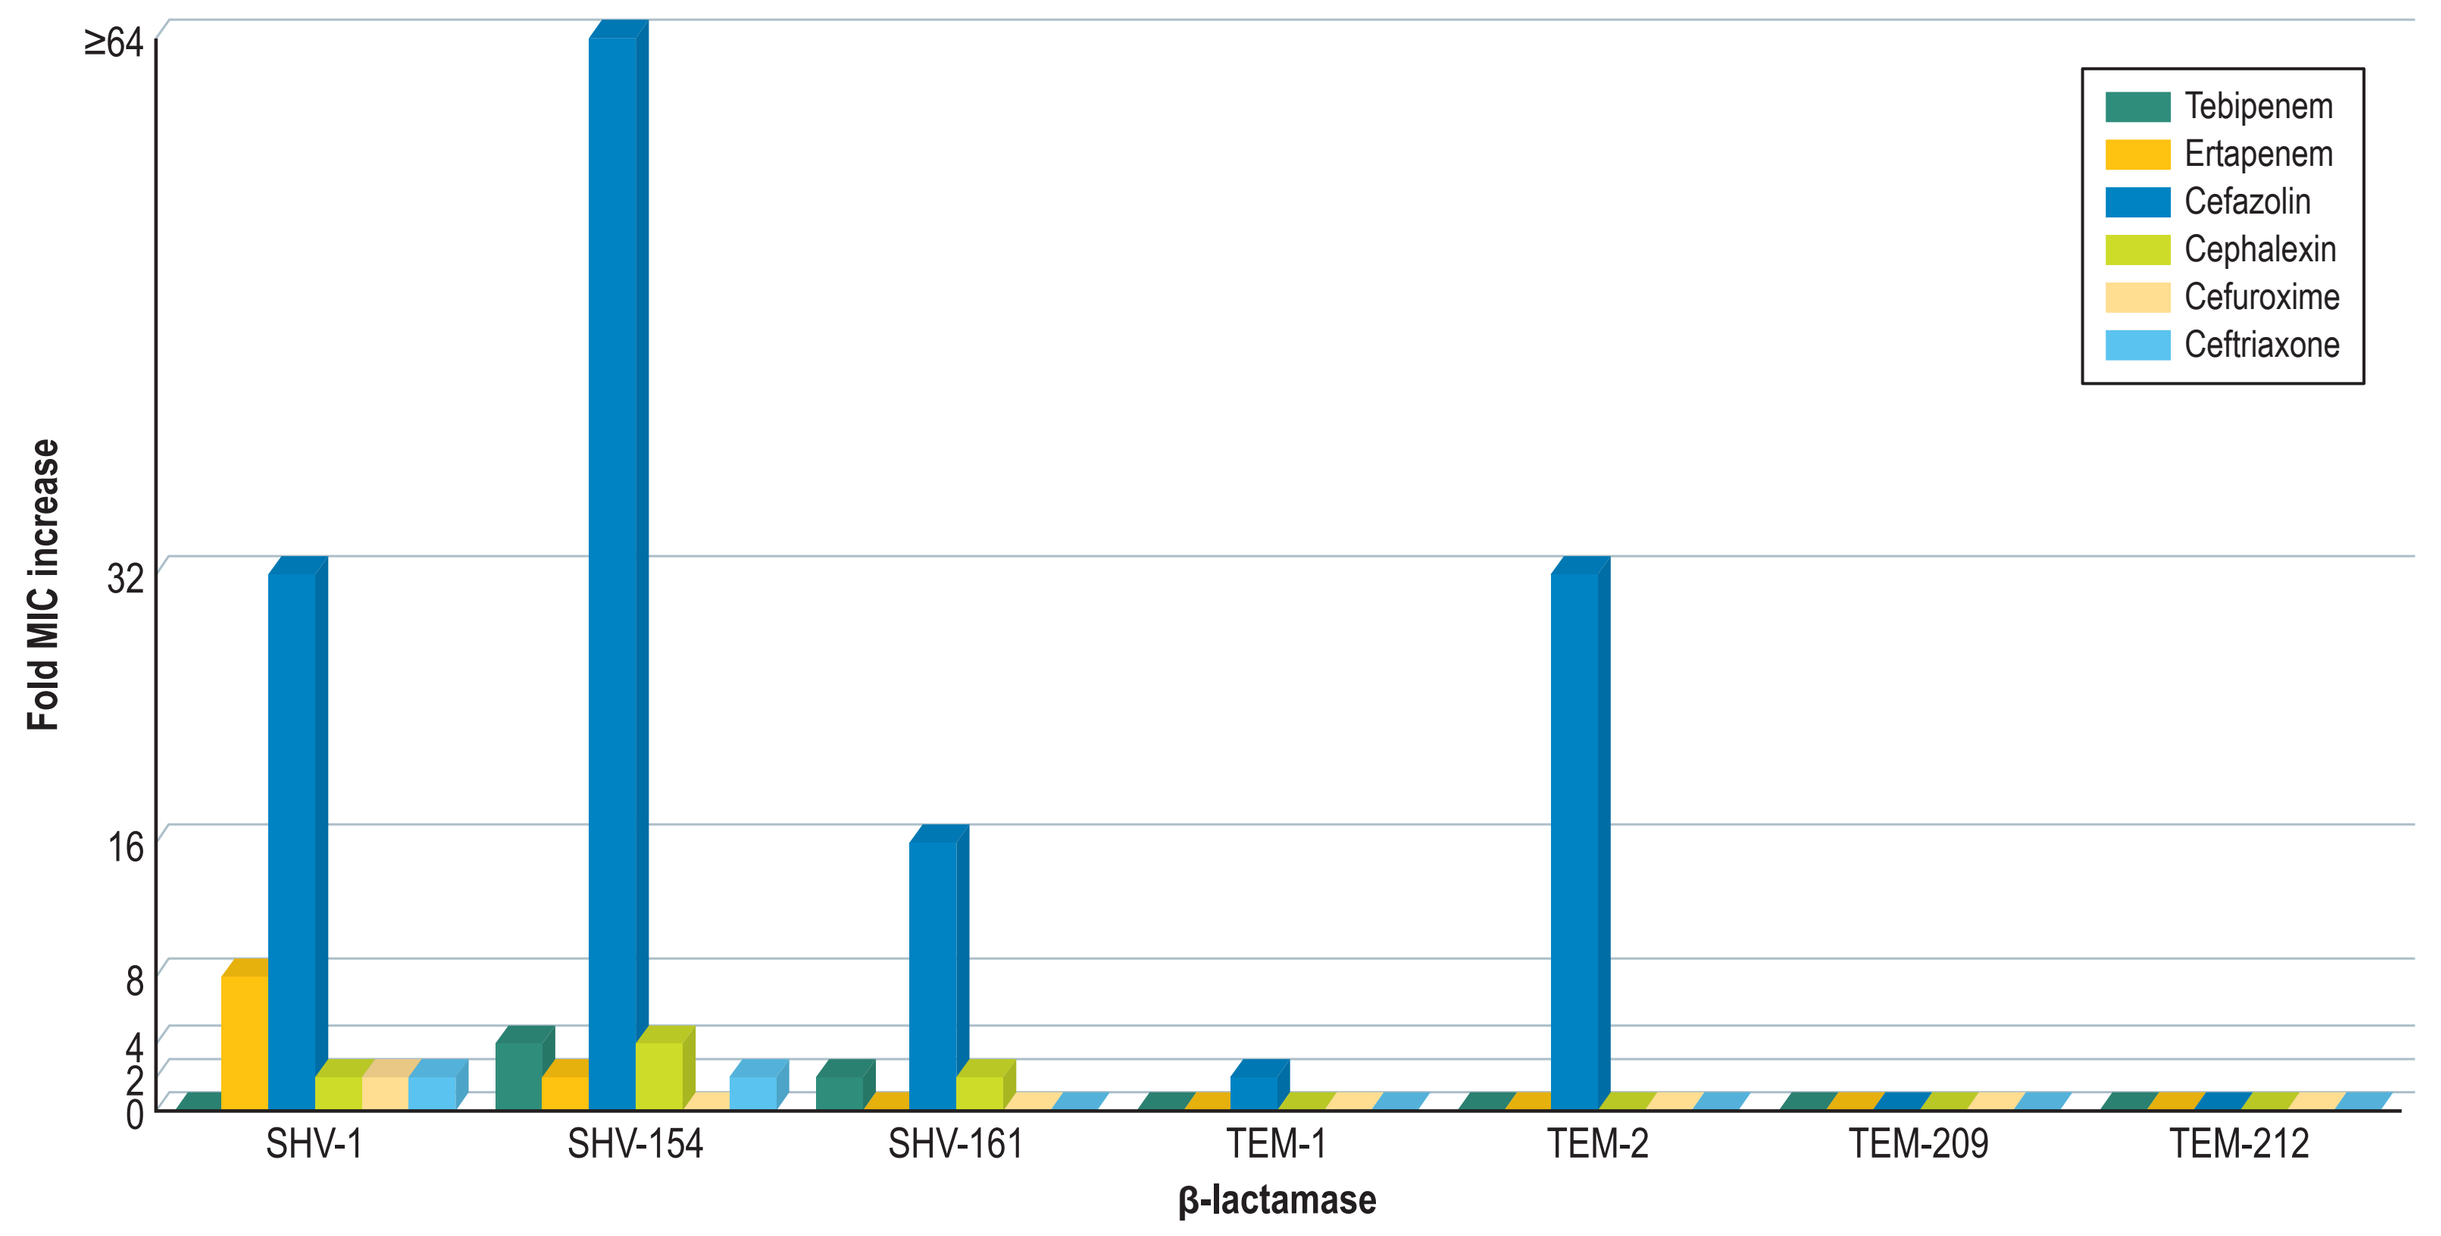
Fold MIC increase
β-lactamase
SHV-1
SHV-154
SHV-161
TEM-1
TEM-2
TEM-209
TEM-212
Tebipenem
Ertapenem
Cefazolin
Cephalexin
Cefuroxime
Ceftriaxone

Figure 2 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying TEM ESBL-encoding genes compared to the respective baseline strain

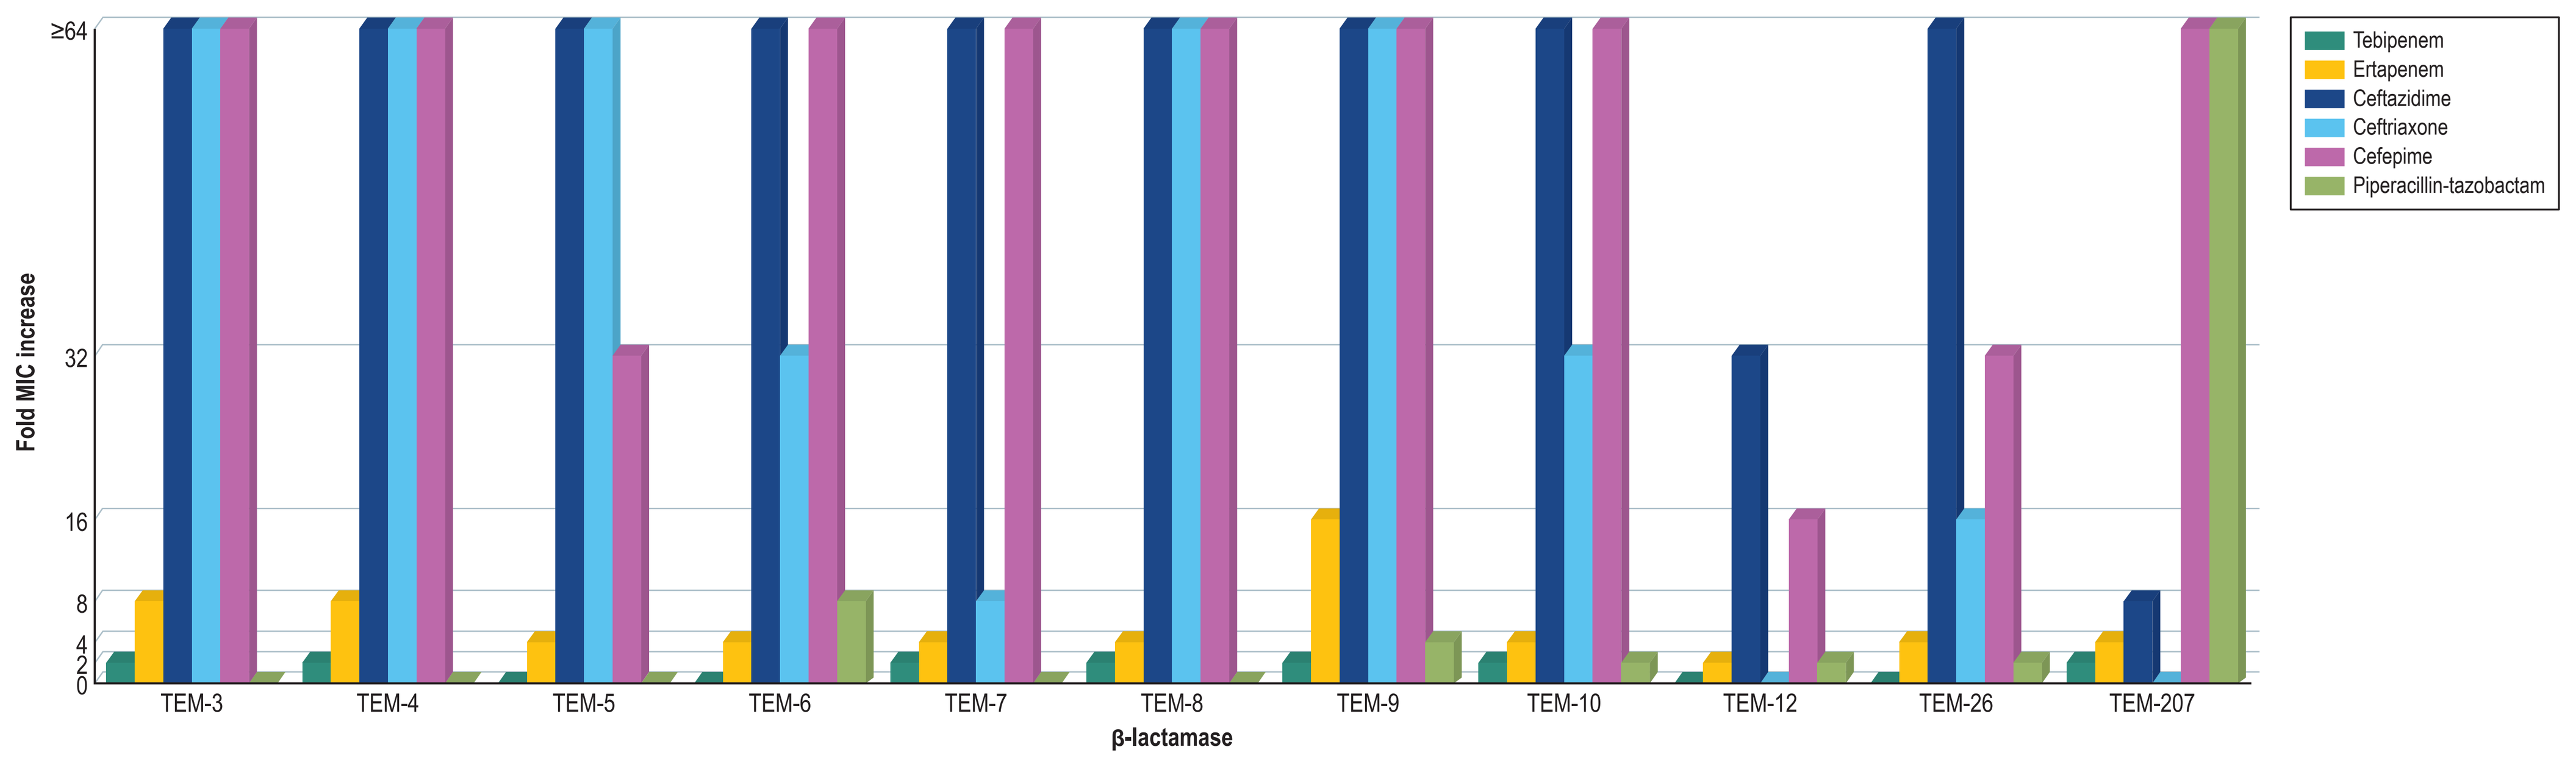
Fold MIC increase
β-lactamase
TEM-3
TEM-4
TEM-5
TEM-6
TEM-7
TEM-8
TEM-9
TEM-10
TEM-12
TEM-26
TEM-207
Tebipenem
Ertapenem
Ceftazidime
Ceftriaxone
Cefepime
Piperacillin-tazobactam

Figure 3 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying SHV- and CTX-M-encoding genes compared to the respective baseline strain

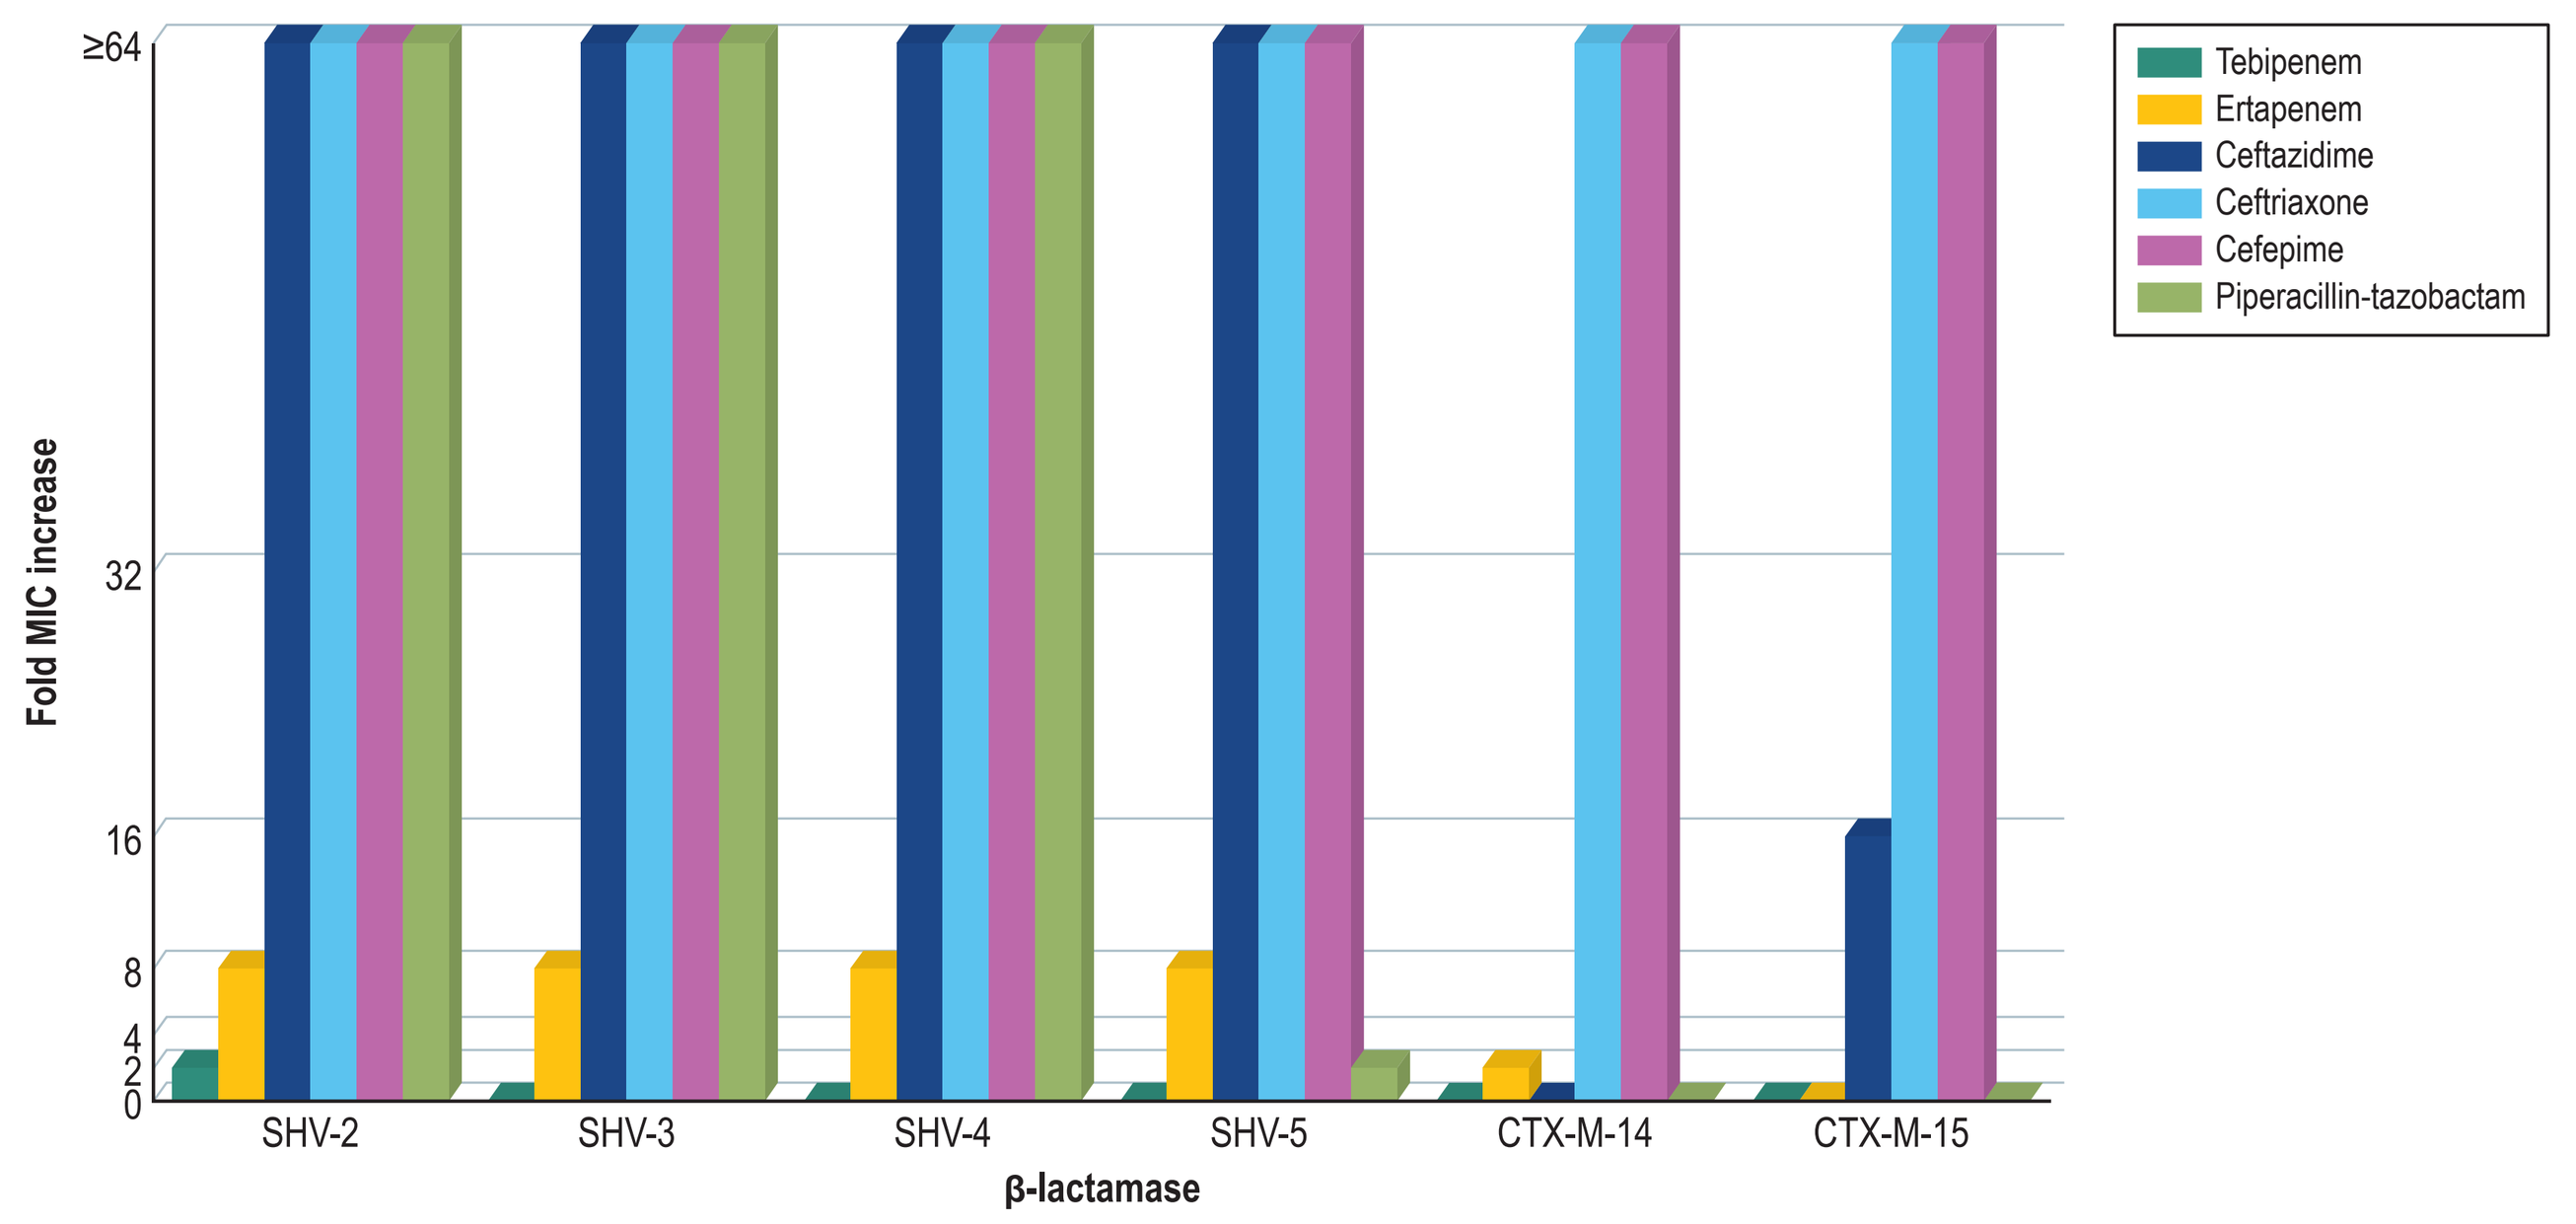
Fold MIC increase
β-lactamase
SHV-2
SHV-3
SHV-4
SHV-5
CTX-M-14
CTX-M-15
Tebipenem
Ertapenem
Ceftazidime
Ceftriaxone
Cefepime
Piperacillin-tazobactam

Figure 4 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying combinations of β-lactamase-encoding genes compared to the respective baseline strain

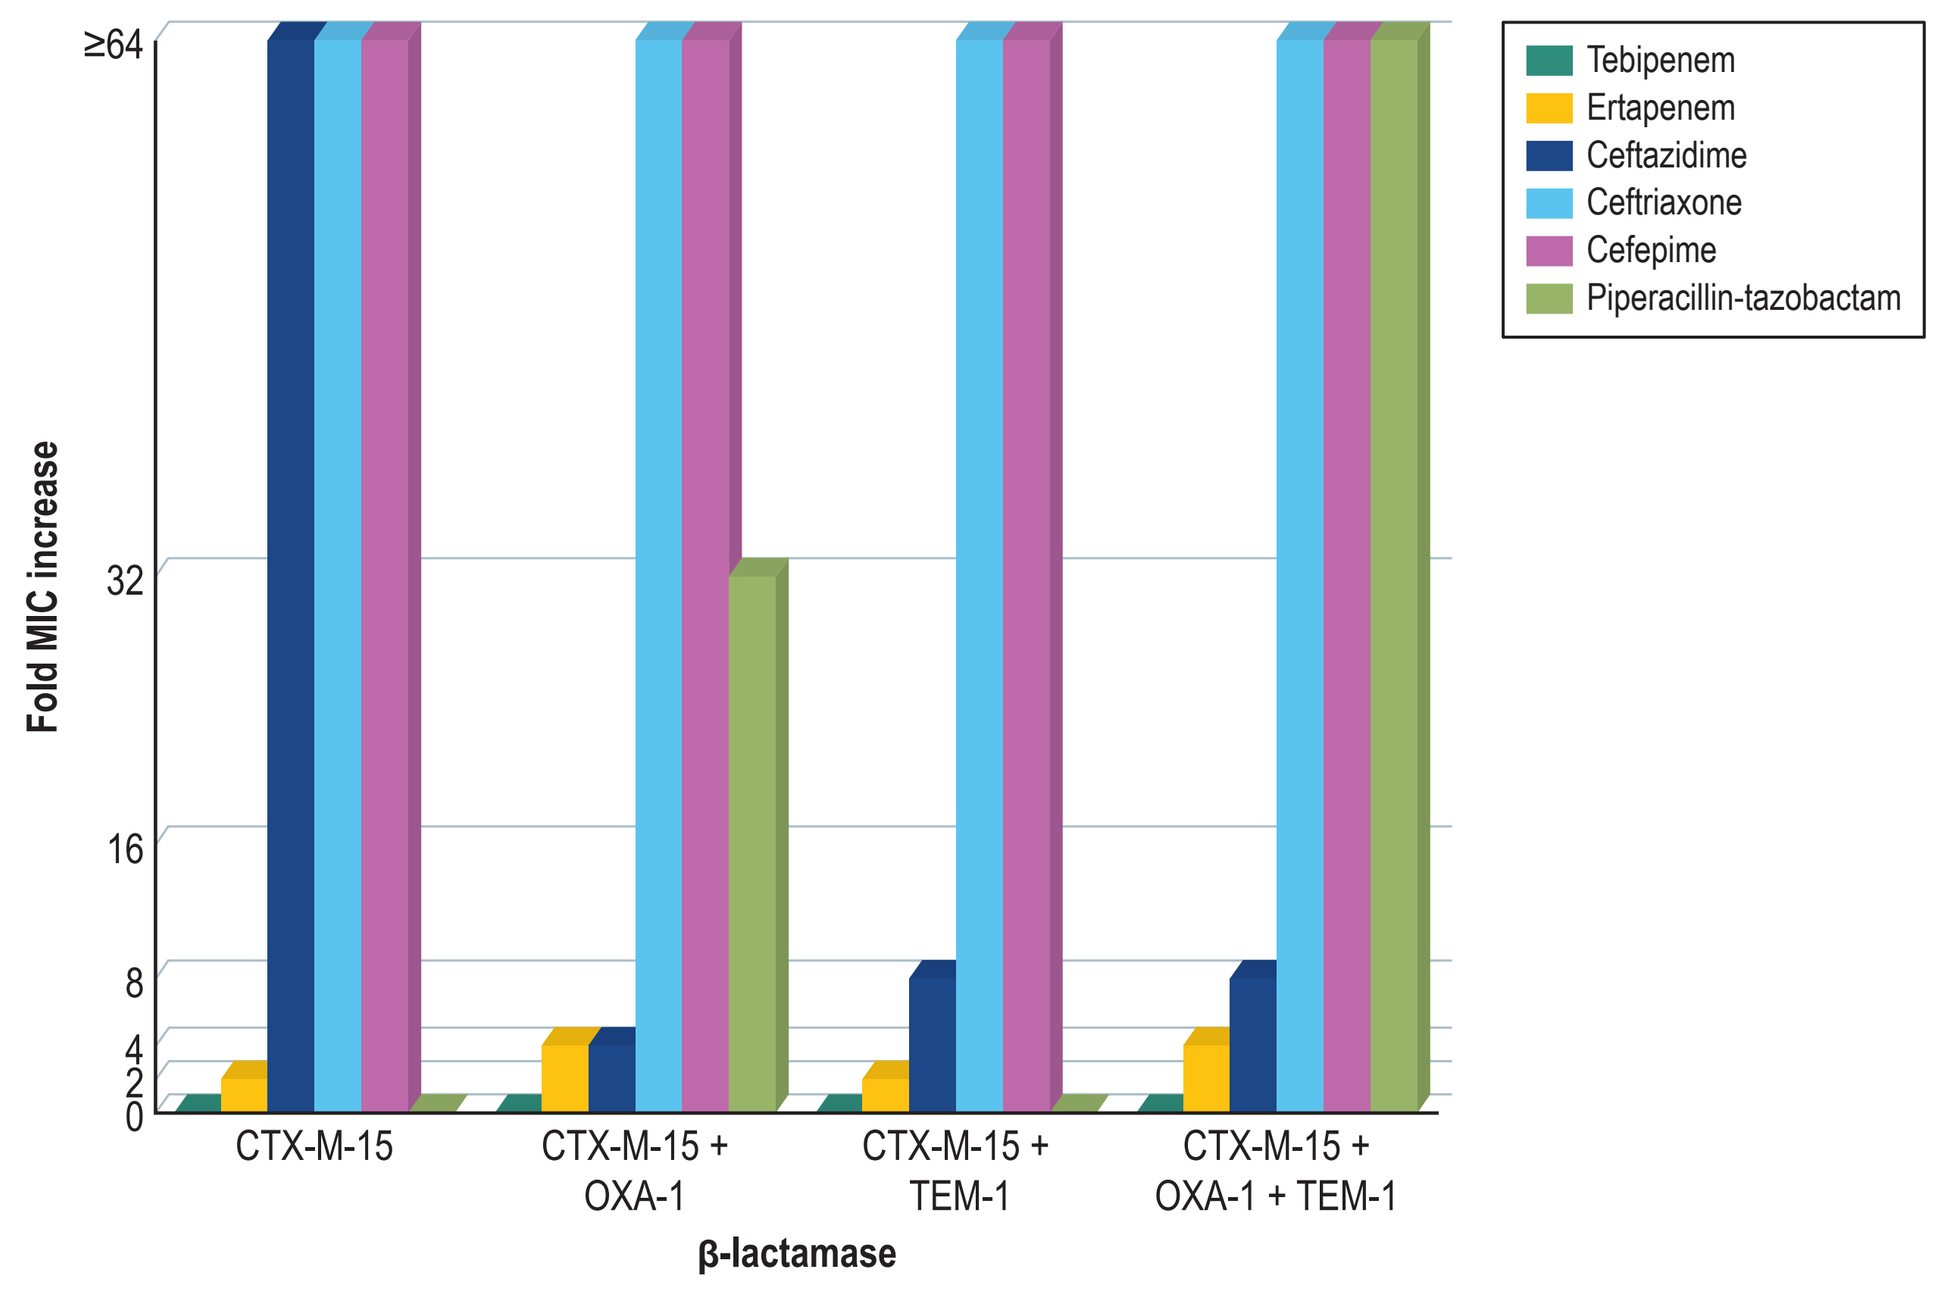
Fold MIC increase
β-lactamase
CTX-M-15
CTX-M-15 + OXA-1
CTX-M-15 + TEM-1
CTX-M-15 + OXA-1 + TEM-1
Tebipenem
Ertapenem
Ceftazidime
Ceftriaxone
Cefepime
Piperacillin-tazobactam

Figure 5 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying β-lactamase-encoding genes compared to the respective baseline strain

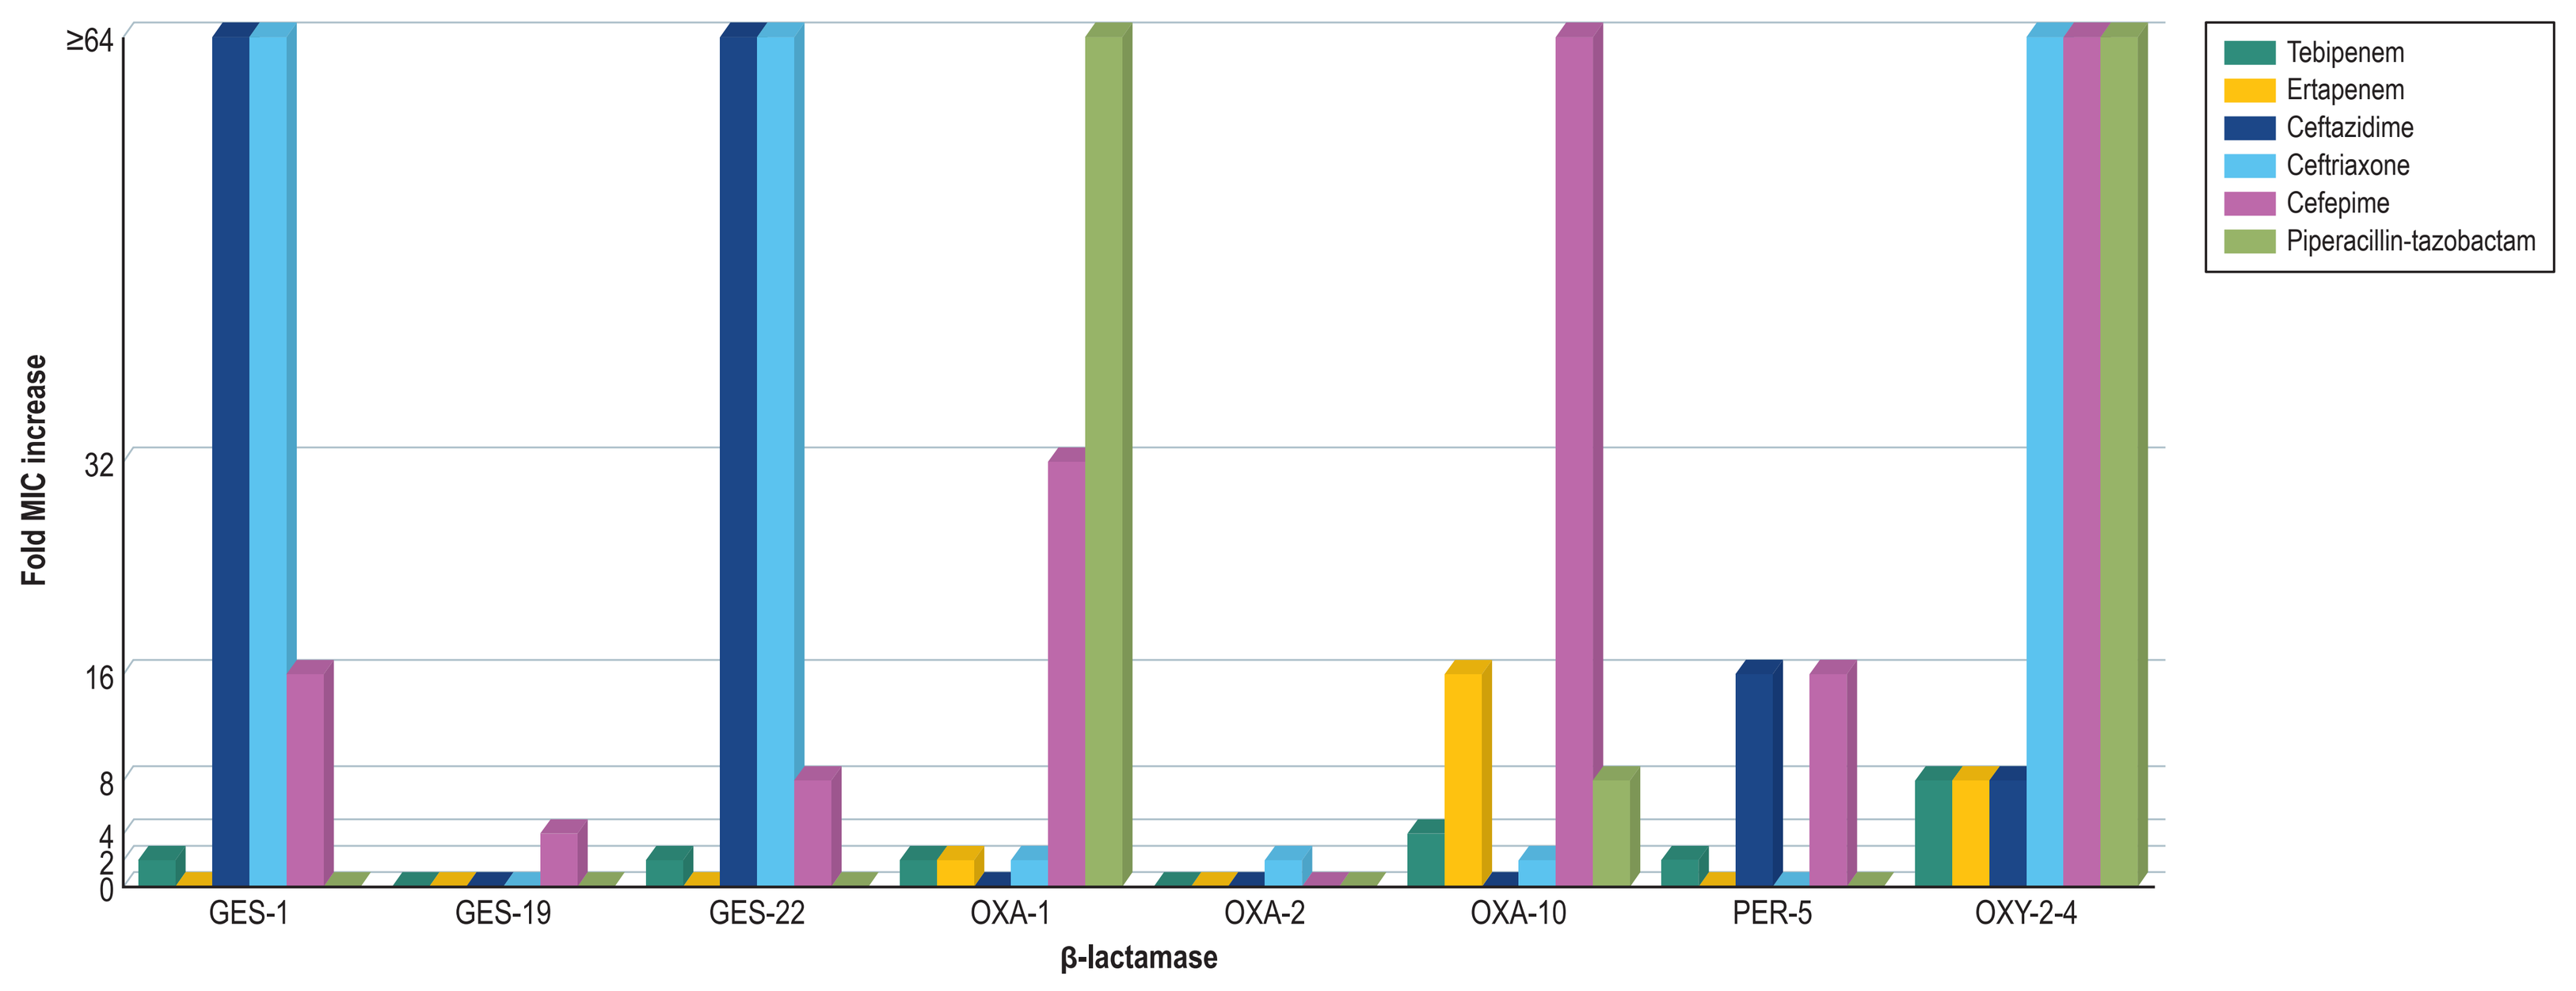
Fold MIC increase
β-lactamase
GES-1
GES-19
GES-22
OXA-1
OXA-2
OXA-10
PER-5
OXY-2-4
Tebipenem
Ertapenem
Ceftazidime
Ceftriaxone
Cefepime
Piperacillin-tazobactam

Figure 6 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying plasmid- and chromosomal (SRT, *Serratia marcescens*)-encoded AmpC genes compared to the respective baseline strain

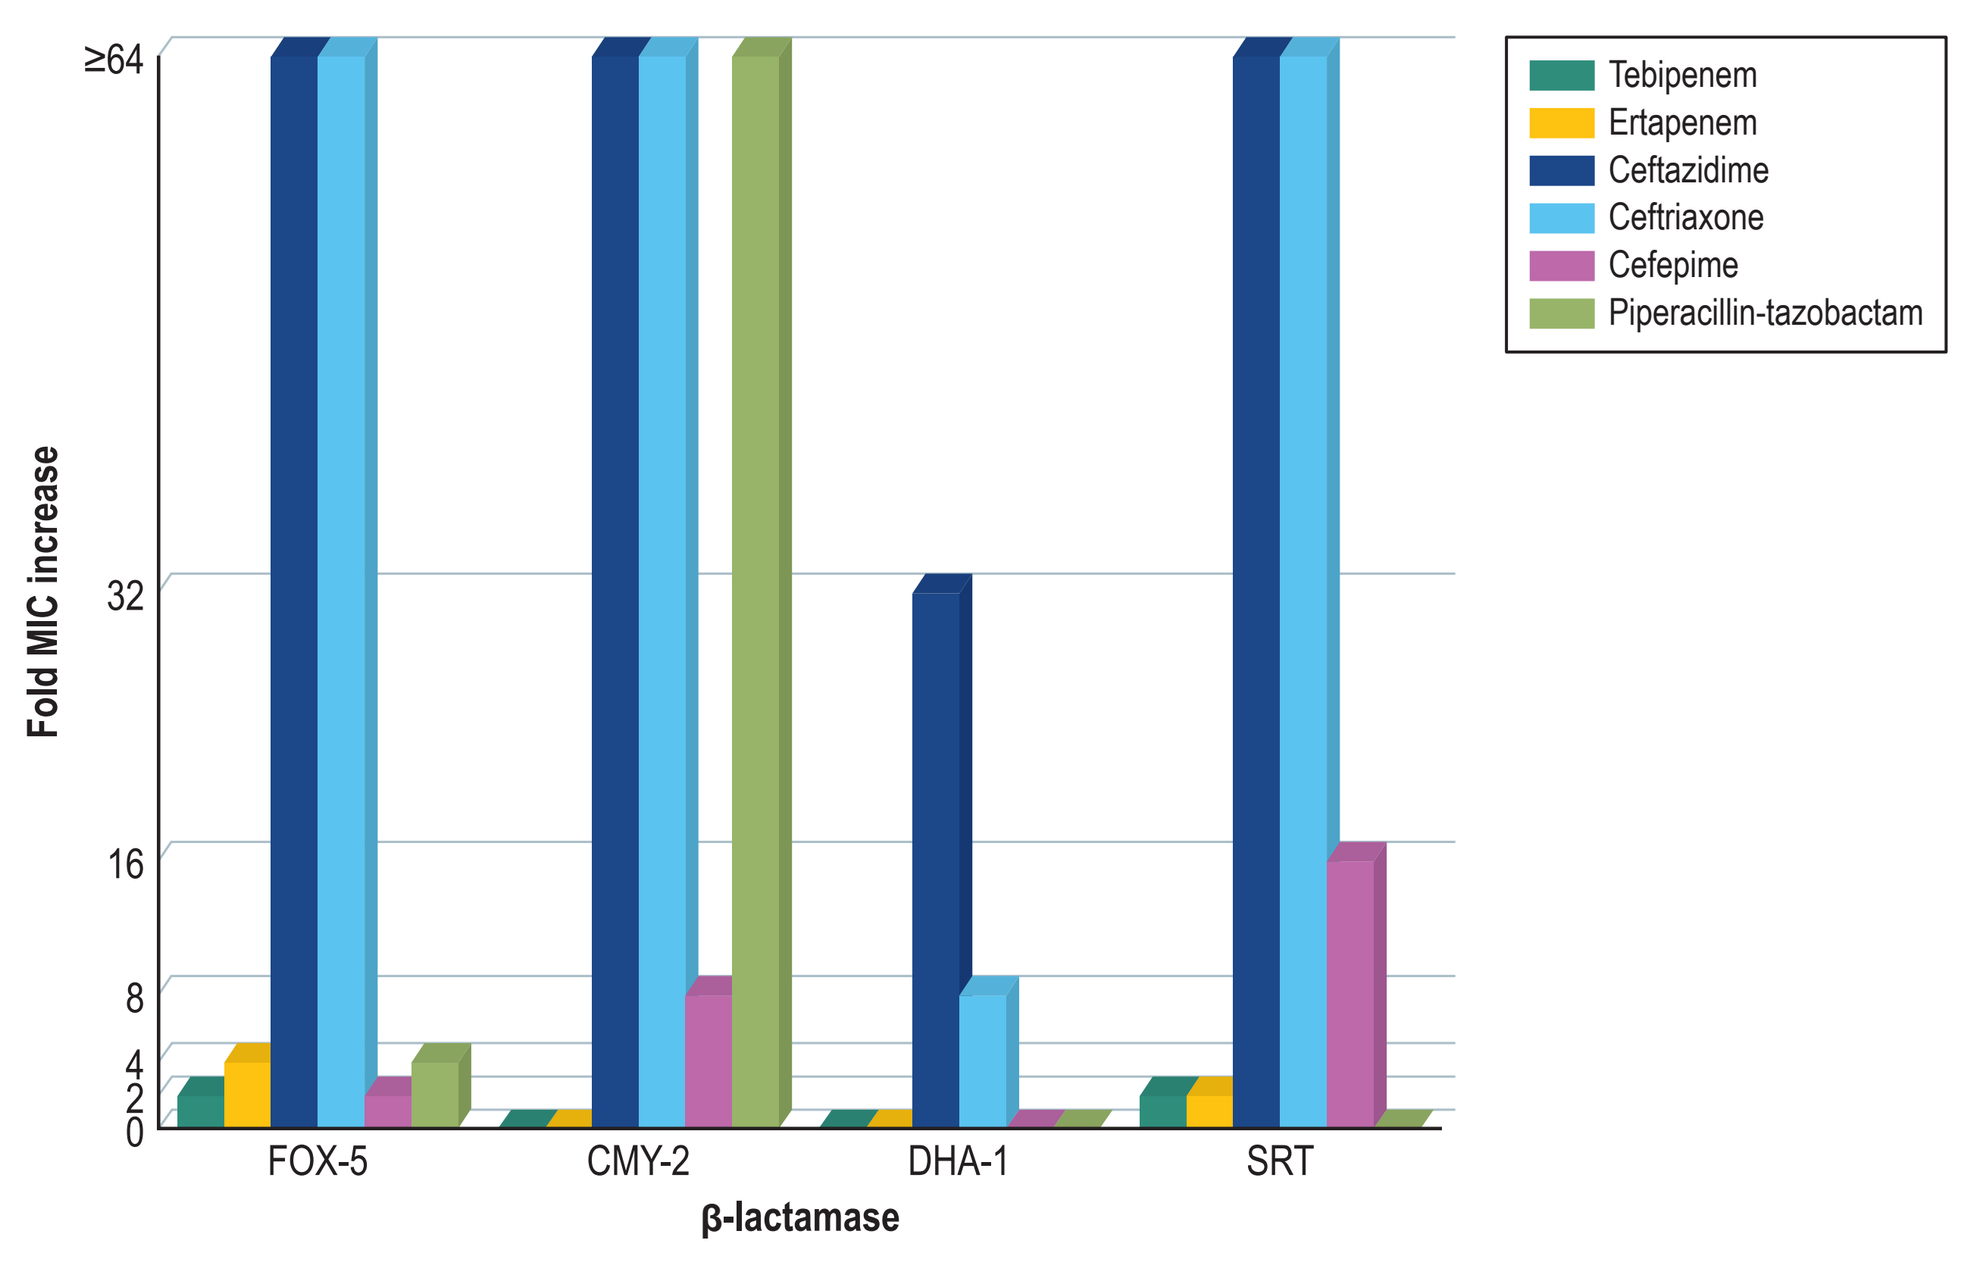
Fold MIC increase
β-lactamase
FOX-5
CMY-2
DHA-1
SRT
Tebipenem
Ertapenem
Ceftazidime
Ceftriaxone
Cefepime
Piperacillin-tazobactam

Figure 7 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying class A and B carbapenemase-encoding genes compared to the respective baseline strain

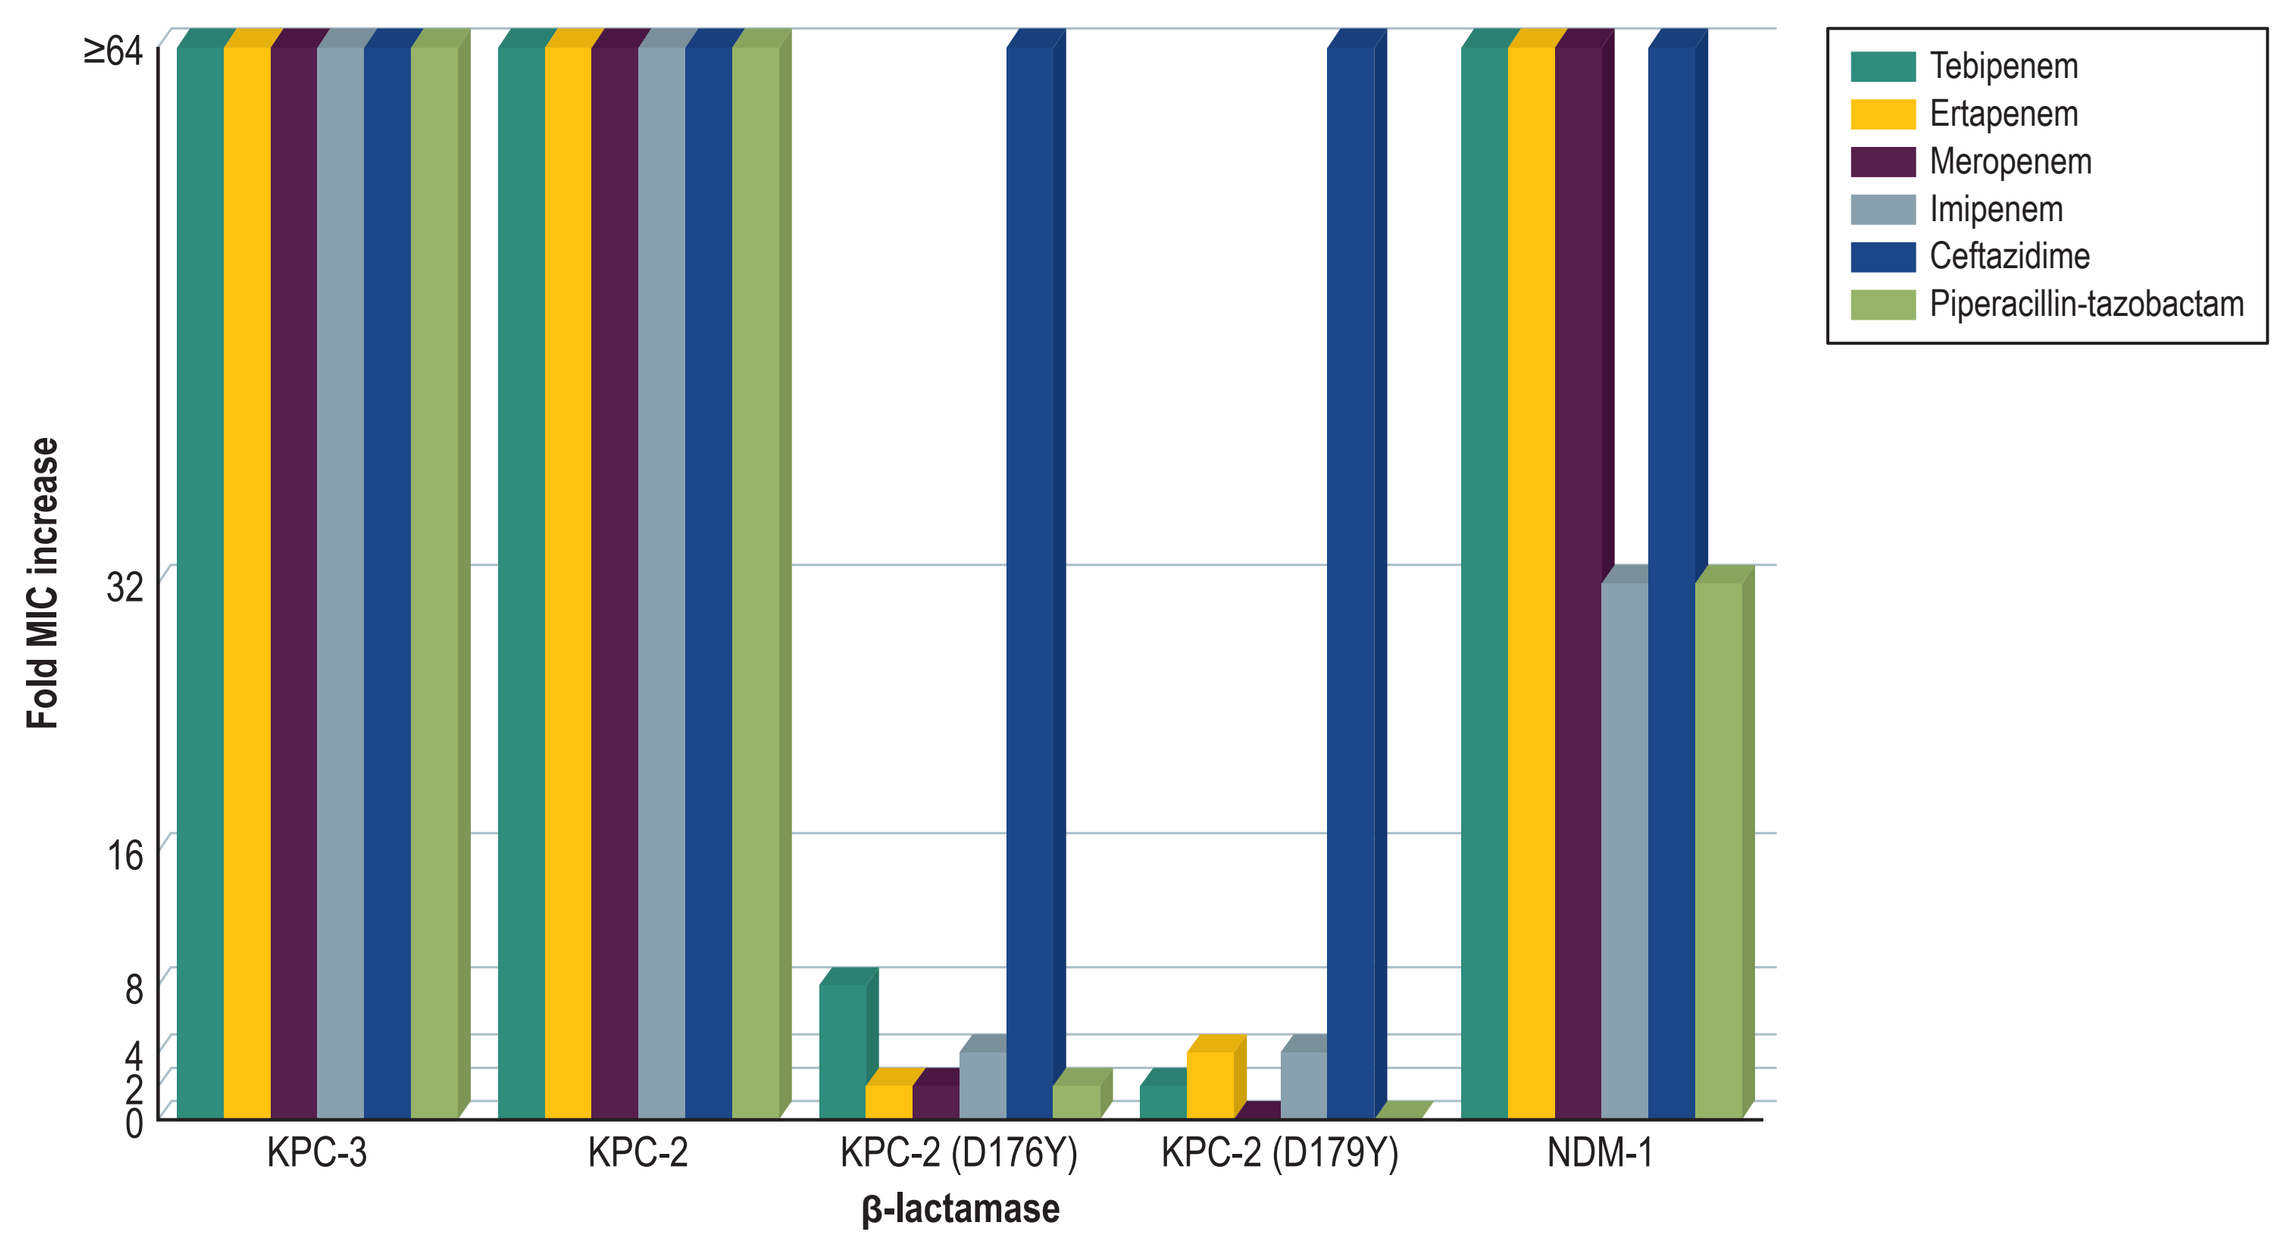
Fold MIC increase
β-lactamase
KPC-3
KPC-2
KPC-2 (D176Y)
KPC-2 (D179Y)
NDM-1
Tebipenem
Ertapenem
Meropenem
Imipenem
Ceftazidime
Piperacillin-tazobactam

Figure 8 Increase in fold MIC of antimicrobial agents tested against *E. coli* carrying other carbapenemase-encoding genes compared to the respective baseline strain

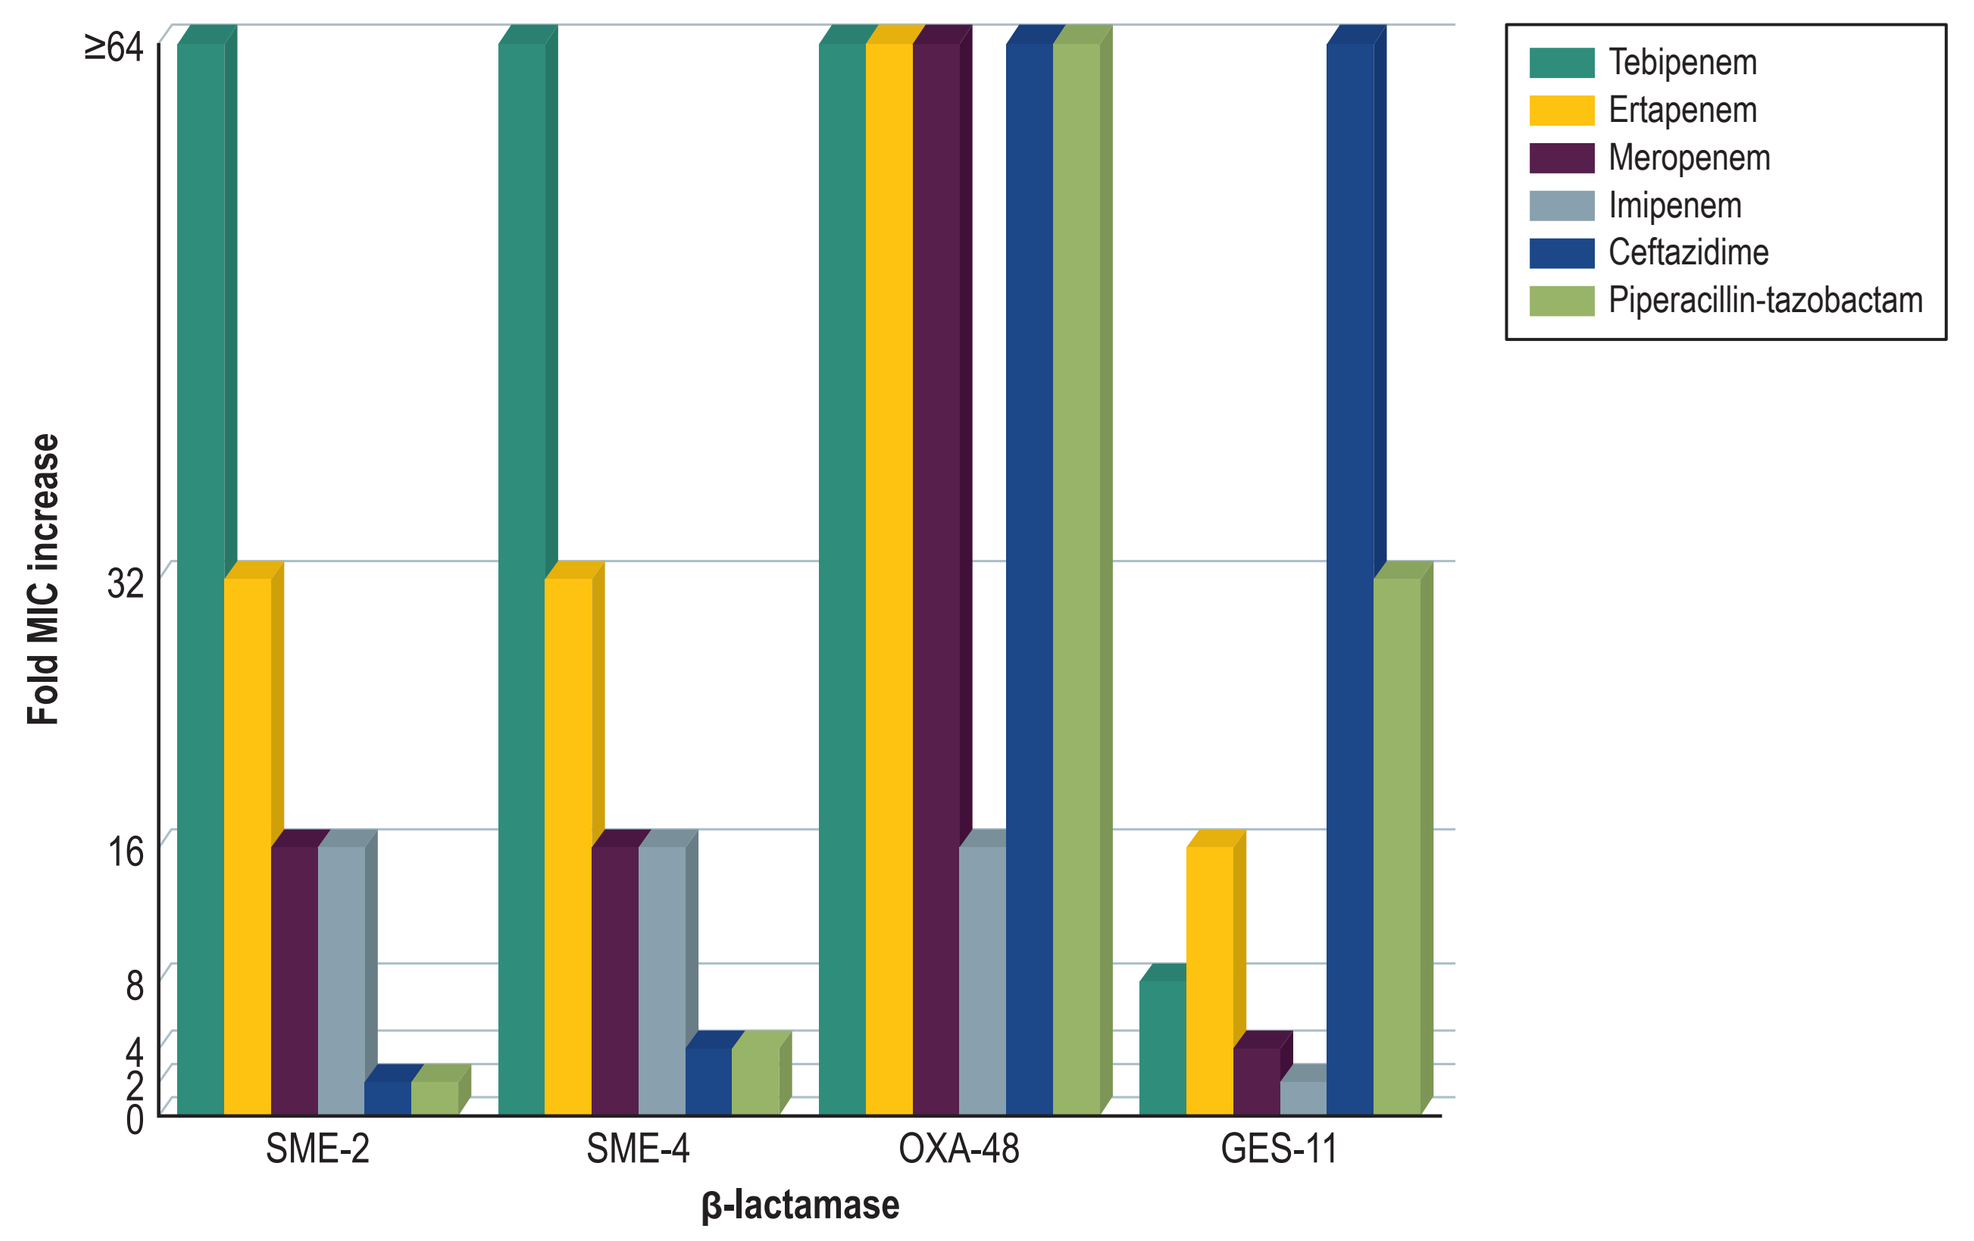
Fold MIC increase
β-lactamase
SME-2
SME-4
OXA-48
GES-11
Tebipenem
Ertapenem
Meropenem
Imipenem
Ceftazidime
Piperacillin-tazobactam

Contact

Rodrigo E. Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

To obtain a PDF of this poster:

https://www.jmilabs.com/data/posters/ASM-Microbe19-tebipenem-isogenic-E-coli.pdf

Charges may apply. No personal information is stored.

ASM Microbe 2019, June 20–24, 2019, San Francisco, California