C-296

Molecular Analysis of Resistance Markers and Clonality among Gram-negative Bacilli: Report from the 2006 MYSTIC Program (USA)

PR RHOMBERG, LM DESHPANDE, HS SADER, TR FRITSCHE, RN JONES
JMI Laboratories, North Liberty, IA

ASM 2007
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370
319.665.3371 fax
ronald-jones@jmilabs.com

MYSTIC
Meropenem Yearly Susceptibility Test Information Collection

ABSTRACT

Background:
The MYSTIC Program is a global surveillance network now monitoring >100 medical centers (MC) actively using full spectrum carbapenems (CARBS) for serious infection therapy. In the USA, 15 MC participated in 2006 by submitting 200 consecutive, non-duplicate clinical isolates.
Methods:
2,841 isolates including 640 *E. coli* (EC), 619 *Klebsiella* spp. (KSP), 606 *P. aeruginosa* (PSA), 906 Gram-positive (GP) strains (*E. faecalis*, streptococci, and oxacillin-susceptible *S. aureus*), and 70 GP anaerobes (ANA) were tested by a central monitor using CLSI broth and agar dilution susceptibility (S) methods and interpretative criteria. Ribotyping (RT) and/or PFGE were performed on strains exhibiting multidrug-resistance (MDR) or identical antibiograms to determine clonal dissemination. R mechanisms were characterized using PCR and sequencing techniques.
Results:
CARBS had the highest S rate (100%) against EC and fluoroquinolones (FQ) the lowest (72.4-72.9%). Gentamicin had the highest S rate (92.4%) for KSP followed by CARBS (90.1-91.0%). MEM was 2-fold more potent than imipenem ($MIC_{50/90}$, 0.5/8 vs. 1/16 µg/ml) and had a higher S rate (86.5 vs. 80.2%). Confirmed ESBLs among EC and KSP were 4.8 and 5.0%, respectively. Epidemic PSA clusters were detected (19 isolates) in 5 MCs among 42 isolates. Four EC clusters were detected from 2 MCs in New York. KPC carbapenemases were found in KSP from 3 New York City area MCs (rates 22.2-49.2%), and 1 MC in Ohio (2.4%). Mobile AmpC enzymes, FOX-5 (5 isolates) and CMY-2 (EC and KSP, 8 isolates) were observed in ESBL confirmatory test-negative strains. Among 283 FQ-R EC and KSP tested, *qnr* (A, B, and S) were documented in 6 (2.1%) isolates. Only 1 MßL-positive strain was found.
Conclusions:
RT/PFGE detected clonally related outbreaks of MDR PSA isolates negatively influencing R rates in 5 sites in the 2006 USA MYSTIC Program. CARB-R due to KPCs has become prevalent in the New York City area. Meropenem had excellent activity against ESBL-, AmpC-, and *qnr*-(+) isolates. Continued surveillance of these MYSTIC sites appears warranted to monitor the activity of the CARBS in areas of higher utilization.

INTRODUCTION

The Meropenem Yearly Susceptibility Test Information Collection (MYSTIC) Program is an international resistance surveillance study, now having greater than 100 participant sites located in Europe, North America, Latin America and Asia. This study's primary objective is to monitor the in vitro activity of meropenem and other broad-spectrum comparator agents against bacteria isolated from serious infection specimens in hospitals utilizing carbapenems. Fifteen medical centers have been monitored in the United States (USA) by a central laboratory (JMI Laboratories, North Liberty, Iowa, USA) since 1999 using reference broth microdilution susceptibility testing methods.

Antimicrobial resistance surveillance studies are very useful to monitor 1) the emergence of new resistance mechanisms, 2) rates of occurrence for existing mechanisms and 3) the dissemination of an antimicrobial resistance mechanism within a medical center, in a geographic region, or on a global scale. Surveillance studies can help identify resistant clones and then aid in the control and minimization of the spread while providing valuable information to clinicians for selecting empiric or directed therapy for serious infections.

We report the molecular testing results of antimicrobial resistant clones and mechanisms of resistances identified from the antimicrobial susceptibility testing results of the 2006 USA MYSTIC Program isolates.

MATERIALS AND METHODS

Specimen collection: The 2006 USA MYSTIC Program sampled 15 geographically dispersed medical centers with each center requested to submit 200 bacterial isolates recovered from serious infections to fill protocol quotas among *E. coli*, *Klebsiella* spp. and *P. aeruginosa* among Gram-negative bacilli as well as *S. aureus*, *E. faecalis*, streptococci, clostridia and peptostreptococci among Gram-positive species. All isolates were shipped to the central monitoring laboratory (JMI Laboratories) on provided transport swabs.

A total of 2,841 isolates were processed (range 115 to 230 isolates per site) for a compliance rate of 94.8%. Identification of the strains was performed locally with confirmation at the central laboratory using colonial morphology, biochemical tests (Remel, Lenexa, KS) and/or the Vitek System identification cards (bioMerieux, Hazelwood, MO), as required.

Susceptibility testing: MIC testing was performed with validated dry-form panels (TREK Diagnostics, Cleveland, OH) for all strains using CLSI reference quality methods for 11 antimicrobial agents (Table 1). CLSI (M100-S17) criteria were utilized for categorization of susceptibility and resistance for aerobic isolates. The CLSI M11-A7 (2007) methods were performed for four agents against the anaerobic isolates. Quality control was assured utilizing appropriate American Type Culture Collection (ATCC) strains with all results observed within published CLSI ranges.

The CLSI extended spectrum ß-lactamase (ESBL) MIC screening criteria (≥2 µg/ml for ceftazidime or ceftriaxone) was applied to *E. coli,* and *Klebsiella* spp. to determine phenotypic ESBL rates. Confirmation of screen-positive isolates was made by using Etest ESBL strips (AB BIODISK, Solna, Sweden) methods. *Klebsiella* spp. isolates with elevated carbapenem MIC results (MIC ≥2 µg/ml) were screened for the presence of either a serine carbapenemase or metallo-ß-lactamase using disk approximation methods followed by an Etest metallo-ß-lactamase strip (imipenem ± EDTA), and/or PCR sequencing. *P. aeruginosa* isolates demonstrating resistance to carbapenems (imipenem and meropenem MIC, ≥16 µg/ml) and ceftazidime (MIC, ≥32 µg/ml) were further screened for the presence of a metallo-ß-lactamase.

Molecular testing: Clonality was assessed for strains demonstrating multi-drug resistance or highly similar antibiograms by molecular procedures utilizing a Riboprinter Microbial Characterization System® (Qualicon, Wilmington, DE) followed by further epidemiologic discrimination of common ribotypes using CHEF-DRII pulsed-field gel electrophoresis (PFGE; BioRad Laboratories, Hercules, CA).

RESULTS

- Against *E. coli* isolates, only the carbapenems demonstrated a 100% susceptibility rate with meropenem and ertapenem having an eight- to 16-fold lower MIC_{90} result compared to imipenem (Table 1).
- Gentamicin had the highest susceptibility rate (92.4%) against the *Klebsiella* spp. isolates followed by the carbapenems and cefepime (90.1 – 91.0%; Table 1).
- Overall ESBL-phenotype/ESBL-confirmed rates for *Klebsiella* spp. and *E. coli* isolates were 17.1/5.0 and 7.8/4.8%, respectively. Elevated ESBL-phenotype rates were observed for *Klebsiella* spp. due to the presence of serine carbapenemase-producing isolates exhibiting the same phenotype.
- The fluoroquinolones demonstrated the lowest susceptibility rates among the broad-spectrum agents tested against *E. coli* (72.4 – 72.9%), *Klebsiella* spp. (81.9 – 82.7%) and *P. aeruginosa* (71.8 – 73.9%) isolates (Table 1).
- A total of 57 *Klebsiella* spp. isolates from four medical centers were identified that produced KPC carbapenemases with individual site rates of occurrence at 50.8, 32.5, 26.7, and 2.4% (Table 2).
- Among ESBL-confirmatory test-negative strains, the acquired AmpC enzymes CMY-2 (eight strains) and FOX-5 (five strains) were detected from the nine recognized AmpC phenotypes screened using a multiplex PCR strategy and follow-up sequencing (Table 3).
- The presence of *qnr* genes (A, B, and S) was identified in six of 283 (2.1%) fluoroquinolone-resistant Enterobacteriaceae isolates received from five medical centers (Table 4).
- Multidrug-resistant *P. aeruginosa* isolates from five medical centers were observed to be clonally related with clusters of 2 to 5 isolates per site. Clonally related fluoroquinolone-resistant *E. coli* isolates (12) were also identified from two medical centers (Table 5).

Table 1. Antimicrobial activity of meropenem and ten comparator agents tested against *E. coli*, *Klebsiella* spp. and *P. aeruginosa* isolates in the USA MYSTIC Program (2006).

Organism (no. tested)/	MIC (µg/ml)			
antimicrobial agent	50%	90%	Range	% susceptible/resistant[a]
E. coli (641)				
Meropenem	≤0.015	0.03	≤0.015-0.06	100.0/0.0
Imipenem	0.12	0.25	0.06-1	100.0/0.0
Ertapenem	≤0.015	≤0.015	≤0.015-1	100.0/0.0
Ceftriaxone	≤0.25	≤0.25	≤0.25->32	95.0/3.4 (6.7/4.5)[b]
Ceftazidime	≤0.12	0.5	≤0.12->16	95.0/3.3 (7.2/4.4)[b]
Cefepime	≤0.12	0.25	≤0.12->16	97.5/1.6
Piperacillin/Tazobactam	≤8	≤8	≤8->64	96.4/1.7
Gentamicin	≤4	>8	≤4->8	86.9/11.4
Tobramycin	≤1	8	≤1->8	86.4/9.0
Ciprofloxacin	≤0.25	>2	≤0.25->2	72.4/27.5
Levofloxacin	≤0.5	>4	≤0.5->4	72.9/26.4
Klebsiella spp.[c] (619)				
Meropenem	0.03	0.5	≤0.015->32	91.0/8.2 (8.7)[d]
Imipenem	0.25	1	0.03->32	90.6/6.8 (8.7)[d]
Ertapenem	≤0.015	2	≤0.015->32	90.1/9.5
Ceftriaxone	≤0.25	>32	≤0.25->32	85.1/10.3 (15.7/4.8)[b]
Ceftazidime	≤0.12	>16	≤0.12->16	86.4/12.9 (16.5/4.5)[b]
Cefepime	≤0.12	8	≤0.12->16	90.1/9.2
Piperacillin/Tazobactam	≤8	>64	≤8->64	84.7/14.1
Gentamicin	≤4	≤4	≤4->8	92.4/6.3
Tobramycin	≤1	>8	≤1->8	85.9/13.4
Ciprofloxacin	≤0.25	>2	≤0.25->2	81.9/17.3
Levofloxacin	≤0.5	>4	≤0.5->4	82.7/16.8
P. aeruginosa (606)				
Meropenem	0.5	8	≤0.015->32	86.5/6.4[e]
Imipenem	1	16	0.06->32	80.2/10.7[e]
Ceftazidime	2	>16	0.25->16	82.0/12.9[e]
Cefepime	4	16	≤0.12->16	84.2/5.6
Piperacillin/Tazobactam	≤8	>64	≤8->64	88.6/11.4
Gentamicin	≤4	>8	≤4->8	84.2/11.7
Tobramycin	≤1	8	≤1->8	89.9/7.9
Ciprofloxacin	≤0.25	>2	≤0.25->2	73.9/20.6
Levofloxacin	≤0.5	>4	≤0.5->4	71.8/21.8

a. Criteria as published by the CLSI M100-S17 [CLSI, 2007].
b. ESBL phenotype using CLSI screening criteria of ≥2 µg/ml for ceftriaxone or ceftazidime / confirmed rate using Etest with ≥2 $\log_2$ dilution MIC decrease in the presence of clavulanate.
c. Includes: *Klebsiella ornithinolytica* (1 strain), *K. oxytoca* (89 strains), and *K. pneumoniae* (529 strains).
d. Bush group 2f carbapenemase screening concentration of ≥2 µg/ml for meropenem or imipenem. Serine carbapenemase (KPC) production confirmed in 57 strains.
e. Eighteen strains demonstrated R to both carbapenems (≥16 µg/ml) and ceftazidime (≥32 µg/ml).

- Eighteen *P. aeruginosa* isolates conformed to the Senda et al. (1996) criteria (≥32 µg/ml for ceftazidime and ≥16 µg/ml for imipenem and meropenem) as possible producers of metallo-ß-lactamase, but a confirmatory disk approximation test and MßL Etest result were only positive for a single strain. That isolate was not confirmed by PCR testing [IMP(-), VIM(-)].

Table 2. KPC carbapenemase-producing *Klebsiella* spp. isolates identified within the USA MYSTIC Program (2006).

				MIC range (µg/ml)	
Organism	State	# of strains/total	Rate	Meropenem	Imipenem
K. pneumoniae (56 strains)	New York	31/61	50.8%	16->32	8->32
	New York	12/45	26.7%	16-32	8->32
	New Jersey	13/40	32.5%	4->32	8->32
K. oxytoca (1 strain)	Ohio	1/42	2.4%	2	8

Table 3. Presence of AmpC enzymes identified among ESBL confirmatory test negative isolates of *E. coli* and *Klebsiella* spp. in the 2006 USA MYSTIC Program.

Organism	Site	AmpC Enzyme	# of isolates
Escherichia coli	02	CMY-2	2
	02	FOX-5	1
	04	FOX-5	3
	11	FOX-5	1
	14	CMY-2	1
	24	CMY-2	1
	29	CMY-2	2
Klebsiella pneumoniae	20	CMY-2	1
	29	CMY-2	1

Table 4. Presence of *qnr*-mediated fluoroquinolone resistance among *E. coli* and *Klebsiella* spp. in the 2006 USA MYSTIC Program.

Organism	Site	*qnr* type	# of isolates
Escherichia coli	01	*qnr*A	1
	16	*qnr*B	1
	29	*qnr*B	1
Klebsiella pneumoniae	02	*qnr*B	1
	25	*qnr*B	1
	29	*qnr*S	1

Table 5. Ribogroup and/or pulsed-field gel electrophoresis results for clonally related multidrug-resistant isolates identified in the 2006 USA MYSTIC Program.

Organism	Site	# of isolates	Ribogroup	PFGE[a]
Escherichia coli	02	4	NT	EC2A
	02	2	NT	EC2C
	02	4	NT	EC2F
	04	2	NT	EC4B
Pseudomonas aeruginosa	02	4	105.1600.1	PSA2C
	11	4	105.780.4	PSA11A
	16	4	258.256.7	PSA16A, A1
	23	2	105.566.6	PSA23RR1
	28	5	258.132.4	PSA28A, A1, A2

a. NT = not tested.

CONCLUSIONS

- The carbapenems continue to show excellent antimicrobial activity against *E. coli* and are the second most active agents against *Klebsiella* among the broad-spectrum agents tested in the MYSTIC Program. Meropenem was at least two-fold more potent than imipenem against Enterobacteriaceae and *P. aeruginosa* isolates.
- The fluoroquinolone susceptibility rates were the lowest of all tested broad-spectrum agents and showed a continuing trend of decreasing susceptibility compared to prior USA MYSTIC Program results.
- ESBL, AmpC and *qnr* resistance mechanisms were identified in Enterobacteriaceae isolates, however, all isolates remained susceptible to carbapenem class agents.
- The rates of KPC-producing *Klebsiella* spp. isolated in or near New York City continues to increase.
- Continued surveillance of Enterobacteriaceae, especially *Klebsiella* spp., as well as non-fermentative Gram-negative bacilli is becoming more important to monitor for the steady emergence and potential spread of resistance mechanisms affecting the therapeutic utility of carbapenem class agents.

ACKNOWLEDGEMENTS

This study was supported by an educational/research grant from AstraZeneca Pharmaceuticals. The 2006 MYSTIC Program sites/participants in the USA were: Arkansas Children's Hospital (T. Beavers-May/G. Schutze); Children's Hospital of Orange County (A. Arrieta/O. Vargas); Cleveland Clinic (G. Hall/D. Wilson/M Tuohy); Clinical Laboratories of Hawaii (F. Pien/L. Ikei-Canter); Columbia Presbyterian Medical Center (P. Della-Latta/S. Whittier/S. Mittman); Creighton University, St. Joseph Hospital (S. Cavalieri/M. Hostetter); Iowa Methodist Medical Center (A. Herring/L. Roller); Ochsner Clinic Foundation (G. Pankey/D. Ashcraft); Robert Wood Johnson Medical School (M. Weinstein/J. Rothberg); University of Colorado Hospital (N. Madinger/J. Monahan); University of Kentucky Hospital (J. Ribes/S. Overman); University of Texas, MD Anderson Cancer Center (K. Rolston/R. Prince); University of Utah, ARUP Laboratories, Inc. (M. Bale/A. Croft); University of Washington (A. Limaye/S. Swanzy); and Winthrop University Hospital (P. Schoch).

SELECTED REFERENCES

Clinical and Laboratory Standards Institute. (2007). *Performance standards for antimicrobial susceptibility testing, 17th informational supplement M100-S17*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute. (2006). *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically, 7th ed. Approved Standard M7-A7*. Wayne, PA: CLSI, 2006.

Jones, RN, Mendes, C, Turner, PJ, Masterton, R (2005). An overview of the Meropenem Yearly Susceptibility Test Information Collection (MYSTIC) Program: 1997-2004. *Diagn Microbiol Infect Dis* 53: 247-256.

Klugman, KP (2003). The role of clonality in the global spread of fluoroquinolone-resistant bacteria. *Clin Infect Dis* 36: 783-785.

Masterton, RG, Turner, PJ (2005). Overview of the Meropenem Yearly Susceptibility Test Information Collection (1997-2004). *Diagn Microbiol Infect Dis* 53: 245-246.

Tenover, FC, Arbeit, RD, Goering, RV, Mickelsen, PA, Murray, BE, Persing, DH, Swaminathan, B (1995). Interpreting chromosomal DNA restriction patterns produced by pulsed-field gel electrophoresis: Criteria for bacterial strain typing. *J Clin Microbiol* 33: 2233-2239.

Turner, PJ (2005). Use of a program-specific website to disseminate surveillance data obtained from the Meropenem Yearly Susceptibility Test Information Collection (MYSTIC) Study. *Diagn Microbiol Infect Dis* 53: 273-279.

Unal, S, Garcia-Rodriguez, JA (2005). Activity of meropenem and comparators against *Pseudomonas aeruginosa* and *Acinetobacter* spp. isolated in the MYSTIC Program, 2002-2004. *Diagn Microbiol Infect Dis* 53: 265-271.