C-040
ASM 2008
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
ronald-jones@jmilabs.com

Proposed Susceptibility Testing Criteria for *H. influenzae* (HI) Tested Against Piperacillin/Tazobactam (P/T): Report Using Results from the SENTRY Program and Japan Surveillance Isolates

Y HIRAKATA, PG AMBROSE, PR RHOMBERG, RN JONES
Tohoku Univ. Sch. Med., Miyagi, Japan; Ordway Research Inst., Albany, NY, USA; JMI Laboratories, North Liberty, IA, USA

SENTRY ANTIMICROBIAL SURVEILLANCE

AMENDED ABSTRACT

Background: ß-lactam resistance (R) among HI has recently increased, especially in some geographic areas (Japan, EU, Australia), mediated by altered PBP3 targets (so-called ampicillin-R ß-lactamase-negative [BLNAR] strains). CLSI has disk and/or MIC S testing breakpoints for multiple agents, but P/T has only a MIC breakpoint (≤1 µg/ml) and a disclaimer comment that BLNAR HI should be considered R regardless of MIC results. Two separate studies re-evaluated P/T potency against HI isolates.

Methods: Seven ß-lactams (includes P/T) and ciprofloxacin (CIP) were tested by CLSI M2-A9 and M7-A7 methods, interpreted by M100-S18 (2008). SENTRY Program BLNAR (ampicillin [AMP] MIC, ≥2 µg/ml), 96 from 17 countries and 400 HI from Japan (100 BLNAR) were tested. HI controls (100 AMP-S, 100 AMP-R ß-lactamase-positive [BL+], 100 BL+ amoxicillin/clavulanate-R [ACR]) were also analyzed; and pharmacodynamic (PD) principals for the routine 13.5 gram P/T daily dose were applied.

Results: Rates of BLNAR in the SENTRY Program (among 12,694 HI) were: Asia-Pacific (7.9%; 76% from Japan), EU (0.5%), Latin America (0.4%) and USA (0.4%); however only 31.7% of these had AMP MIC results at ≥4 µg/ml. Scattergrams of SENTRY Program and Japanese BLNAR strains showed all P/T MIC values at ≤0.5 µg/ml (93.4% at ≤0.12 µg/ml); all zones at ≥21 mm (median, 32 mm). BL+ACR HI were also P/T-S (MICs, ≤0.5 µg/ml), as were other HI isolate subsets from Japan. PD parameters predicting P/T treatment success (T>MIC) showed 100% target attainment with 3.375 gram Q6 dosing (C_{max}, 242 µg/ml; trough, 1.0 µg/ml). US-FDA P/T package insert lists HI indications for community and nosocomial pneumonia, including BL+ strains at a breakpoint of ≤1 µg/ml. Other tested agents had some discords between test method results (false-S or R) requiring CLSI re-evaluation.

Conclusion: HI tested against P/T demonstrated uniformly high S (MICs, ≤0.5 µg/ml) including BLNAR strains worldwide. Modified S only breakpoints are proposed for P/T at ≤1 µg/ml (current US-FDA and CLSI criteria) and ≥21 mm for DD that provides 100% intermethod categorical agreement and shows BLNAR isolates as P/T-S.

INTRODUCTION

Haemophilus influenzae is a common cause of community-acquired respiratory tract infections (CA-RTI) and serious invasive disease (bacteremia and meningitis), although the latter types of infection were markedly diminished by vaccine introduction in some nations. Resistances to frequently used therapies remain unusual except for ß-lactam hydrolysis mediated by a TEM-type enzyme. Standardized susceptibility methods for *H. influenzae* were developed by the Clinical and Laboratory Standards Institute (CLSI; formerly NCCLS) in the early 1990s based on the use of a modified Mueller-Hinton medium (Haemophilus Test medium or HTM). With the exception of ß-lactamase-producing isolates, few resistant *H. influenzae* strains were available for method development and validation; ß-lactamase-negative ampicillin-resistant (BLNAR) strains occurred at rates of <0.1%. Subsequently, these BLNAR strains have become more frequent worldwide and particularly in the Western Pacific and some European nations. Also, fluoroquinolone-resistant *H. influenzae* having target mutations have emerged, leading to confusion about the interpretation of non-susceptibility as tested/interpreted by CLSI methods.

Initial CLSI interpretive criteria for ampicillin were published between 1990 and 1992. As newer ß-lactams (oral cephems, carbapenems, penicillin/ß-lactamase inhibitor combinations) and other antimicrobial classes were developed, the CLSI added interpretive criteria to supplemental tables. However, no systemic re-evaluation of the *H. influenzae* breakpoints has been forthcoming as demanded by the greater frequency of BLNAR isolates and resistance to other drugs (fluoroquinolones) used to treat CA-RTI. Furthermore, discords between CLSI testing interpretations by MIC and disk diffusion methods have been observed, additional problems with HTM lot-to-lot quality, suspected flaws in quality control ranges and unexpected favorable clinical outcomes using "supposedly inactive" agents casts doubt on the contemporary utility of CLSI standards/breakpoints for *H. influenzae*. Finally, the appearance and acceptance of pharmacodynamic (PD) principals as a significant contributor to breakpoint selection, places the current CLSI breakpoints in doubt, especially for those agents having interpretive criteria established prior to 2001.

In this study, we examined two (2) highly selected collections of *H. influenzae* strains from the global SENTRY Antimicrobial Surveillance Program and from numerous hospitals in Japan. These 496 strains (with PBP3 gene data) were tested to determine intermethod accuracy of CLSI tests and interpretations for ampicillin, six other ß-lactams and ciprofloxacin. Particularly, piperacillin/tazobactam was reassessed for possible addition to the CLSI disk diffusion interpretive tables or modification of US-FDA product package insert.

MATERIALS AND METHODS

The SENTRY Program *H. influenzae* isolates (1998-2004) were screened for ß-lactamase-negative strains having elevated ampicillin MIC values (≥2 µg/ml). These organisms (96; 85 from Japan) were confirmed by repeat testing with reference broth microdilution methods of the CLSI (M7-A7) and additional processing against ampicillin/sulbactam, amoxicillin/clavulanate, ceftriaxone, imipenem, meropenem, piperacillin/tazobactam and a non-ß-lactam control (ciprofloxacin). Disk diffusion testing was simultaneously performed using the CLSI M2-A9 method for the same antimicrobial agents. Interpretations of the test results used criteria published in M100-S18 (2008).

Similarly, 400 recent *H. influenzae* clinical isolates (2001-2007) from 138 Japanese hospitals and clinics were categorized into BLNAR, ß-lactamase-negative ampicillin-susceptible (BLNAS), ß-lactamase-positive ampicillin-resistant (BLPAR) and ß-lactamase-positive amoxicillin/clavulanate-resistant (BLPACR) groups of 100 strains each (provided by Mitsubishi Clinical Medicine Corp., Tokyo, Japan). All tests for susceptibility used CLSI methods (M7-A7, M2-A9) and the same comparison agents were utilized.

Results of MIC and disk diffusion testing were compared by scattergram analysis (Figure 1 A-E; piperacillin/tazobactam only) with calculations of intermethod error for all tested agents in the SENTRY Program series. Limits of acceptable error were derived from CLSI M23-A2 where the occurrence of false susceptibility (very major error) was ≤1.5%, false resistance (major error) was ≤3.0% and minor error (intermediate by one test and resistant or susceptible by the other) was not to exceed 10.0%. An intermediate category criterion has not been established for the studied agents other than ampicillin. Furthermore, only susceptible breakpoints are currently published for ceftriaxone, the carbapenems and ciprofloxacin.

Please note that a warning that BLNAR *H. influenzae* strains should be considered resistant to some ß-lactam compounds is found in M2-A2/M100-S18 tables (amoxicillin/clavulanate, ampicillin/sulbactam, cefaclor, cefetamet, cefonicid, cefprozil, cefuroxime and loracarbef) and in M7-A7/M100-S18 tables piperacillin/tazobactam and cefamandole were additionally listed, many of those statements existing for nearly two decades. The *H. influenzae* quality control strain (ATCC 49247), a BLNAR organism, was utilized concurrently with all results recorded within CLSI ranges. Of note, piperacillin/tazobactam control ranges were 0.06-0.5 µg/ml and 33-38 mm; values considered to indicate high susceptibility.

ACKNOWLEDGEMENTS

This project was jointly funded by the JMI Laboratories surveillance platform and Taiho Pharmaceuticals research grants.

RESULTS

- Piperacillin/tazobactam was active against all SENTRY Program (BLNAR) strains as well as recent Japanese strains (BLNAR, BLNAS, BLPACR, BLPAR) of *H. influenzae*; see Tables 1 and 2. This level of activity (MIC_{90}, ≤0.12 µg/ml) was comparable or superior to ceftriaxone, meropenem and ciprofloxacin.
- In contrast to the existing disclaimer against piperacillin/tazobactam use for BLNAR *H. influenzae* strains, this ß-lactamases inhibitor combination appears to be an "agent of choice" based on potency and calculated PD parameters (T>MIC shows 100% target attainment with a C_{max} of 242 µg/ml and trough concentrations at the breakpoint of 1 µg/ml).
- *ftsI* typing by PCR revealed group II and III mutations and resultant ß-lactam MIC values that span the currently applied CLSI breakpoints for several agents. Also, fluoroquinolone resistance (MIC, ≥0.12 µg/ml) was noted among the Japanese BLNAR and BLPACR *H. influenzae* strains.
- Disk diffusion breakpoint zone diameters (susceptible at ≥21 mm) for interpretation were proposed and correlations with the ≤1 µg/ml MIC breakpoint was without intermethod error (Figures 1A-E).
- Evaluations of the intermethod accuracy of seven existing agents in the CLSI documents detected categorical agreements ranging from only 55.2% (ampicillin) to 100.0% (ciprofloxacin and piperacillin/tazobactam; Table 3). Some serious errors were noted including false-susceptible rates of 7.3, 8.3 and 12.5% for meropenem, ampicillin/sulbactam and amoxicillin/clavulanate, respectively. False-resistant errors of >20.0% were recorded for four drugs.

Table 1. MIC distributions for piperacillin/tazobactam and seven comparison agents tested against 400 *H. influenzae* (100 each of BLNAR, BLNAS, BLPAR, BLPACR) from Japanese medical centers (2001-2007).

	Cumulative % inhibited at MIC (µg/ml)								
Antimicrobial agent	≤0.12	0.25	0.5	1	2	4	8	16	% susceptible[a]
Piperacillin/tazobactam	91	99	100	100	100	100	100	100	100
Ampicillin	3	19	23	25	31	42	-[b]	-	25
Ampicillin/sulbactam	3	19	25	38	49	65	97	100	49
Amoxicillin/clavulanate	0	8	33	41	48	60	99	100	40
Ceftriaxone	70	96	99	100	100	100	100	-	100
Imipenem	6	16	34	76	94	98	-	-	98
Meropenem	61	86	99	100	100	100	-	-	99
Ciprofloxacin	97	99	99	99	100	-	-	-	99

a. CLSI MIC breakpoints only.
b. - = untested concentrations.

Table 2. MIC distributions for piperacillin/tazobactam and seven comparison agents tested against 96 BLNAR *H. influenzae* strains from the SENTRY Program worldwide.

	Cumulative % inhibited at MIC (µg/ml):								
Antimicrobial agent	≤0.12	0.25	0.5	1	2	4	8	16	% susceptible[a]
Piperacillin/tazobactam	97	100	100	100	100	100	100	100	100
Ampicillin	0	0	0	0	61	94	-[b]	-	0
Ampicillin/sulbactam	0	0	0	13	67	96	100	100	67
Amoxicillin/clavulanate	0	0	0	2	9	55	94	100	55
Ceftriaxone	58	99	100	100	100	100	100	-	100
Imipenem	1	3	19	66	86	97	-	-	97
Meropenem	7	63	93	100	100	100	-	-	93
Ciprofloxacin	100	100	100	100	100	-	-	-	100

a. CLSI MIC breakpoints only.
b. - = untested concentration.

Table 3. Intermethod categorical agreement (%) for the CLSI disk diffusion interpretive criteria when testing BLNAR isolates from the SENTRY Program (96 strains).

	Error rate (%) by category			
Antimicrobial agent	Minor[a]	Major[a]	Very major[a]	Categorical agreement (%)
Ampicillin	42.7	0	2.1	55.2
Ampicillin/sulbactam	NA[b]	21.9	8.3	69.8
Amoxicillin/clavulanate	NA	29.2	12.5	58.3
Ceftriaxone	NA	27.1	0.0	72.9
Imipenem	NA	20.8	1.0	78.1
Meropenem	NA	4.2	7.3	88.5
Piperacillin/tazobactam[c]	NA	0.0	0.0	100.0
Ciprofloxacin	NA	0.0	0.0	100.0

a. Very major = false-susceptible by disk diffusion method, major = false-resistant and minor = intermediate by one method and susceptible or resistant by the other (see ampicillin results only).
b. NA = not applicable due to unpublished intermediate criteria by the CLSI.
c. Results using proposed disk diffusion susceptible breakpoint at ≥21 mm to correlate with published MIC breakpoint (≤1 µg/ml) by CLSI and US-FDA.

Figure 1. Scattergrams showing CLSI and US-FDA piperacillin/tazobactam MIC (solid horizontal lines) breakpoint criteria for *H. influenzae* when testing the SENTRY Program BLNAR collection (A) and the four Japan organism subsets of BLNAS (B), BLPAR (C), BLNAR (D) and BLPACR (E). Broken vertical lines represent the proposed disk diffusion breakpoints (≥21 mm), recently approved by the CLSI (2008).

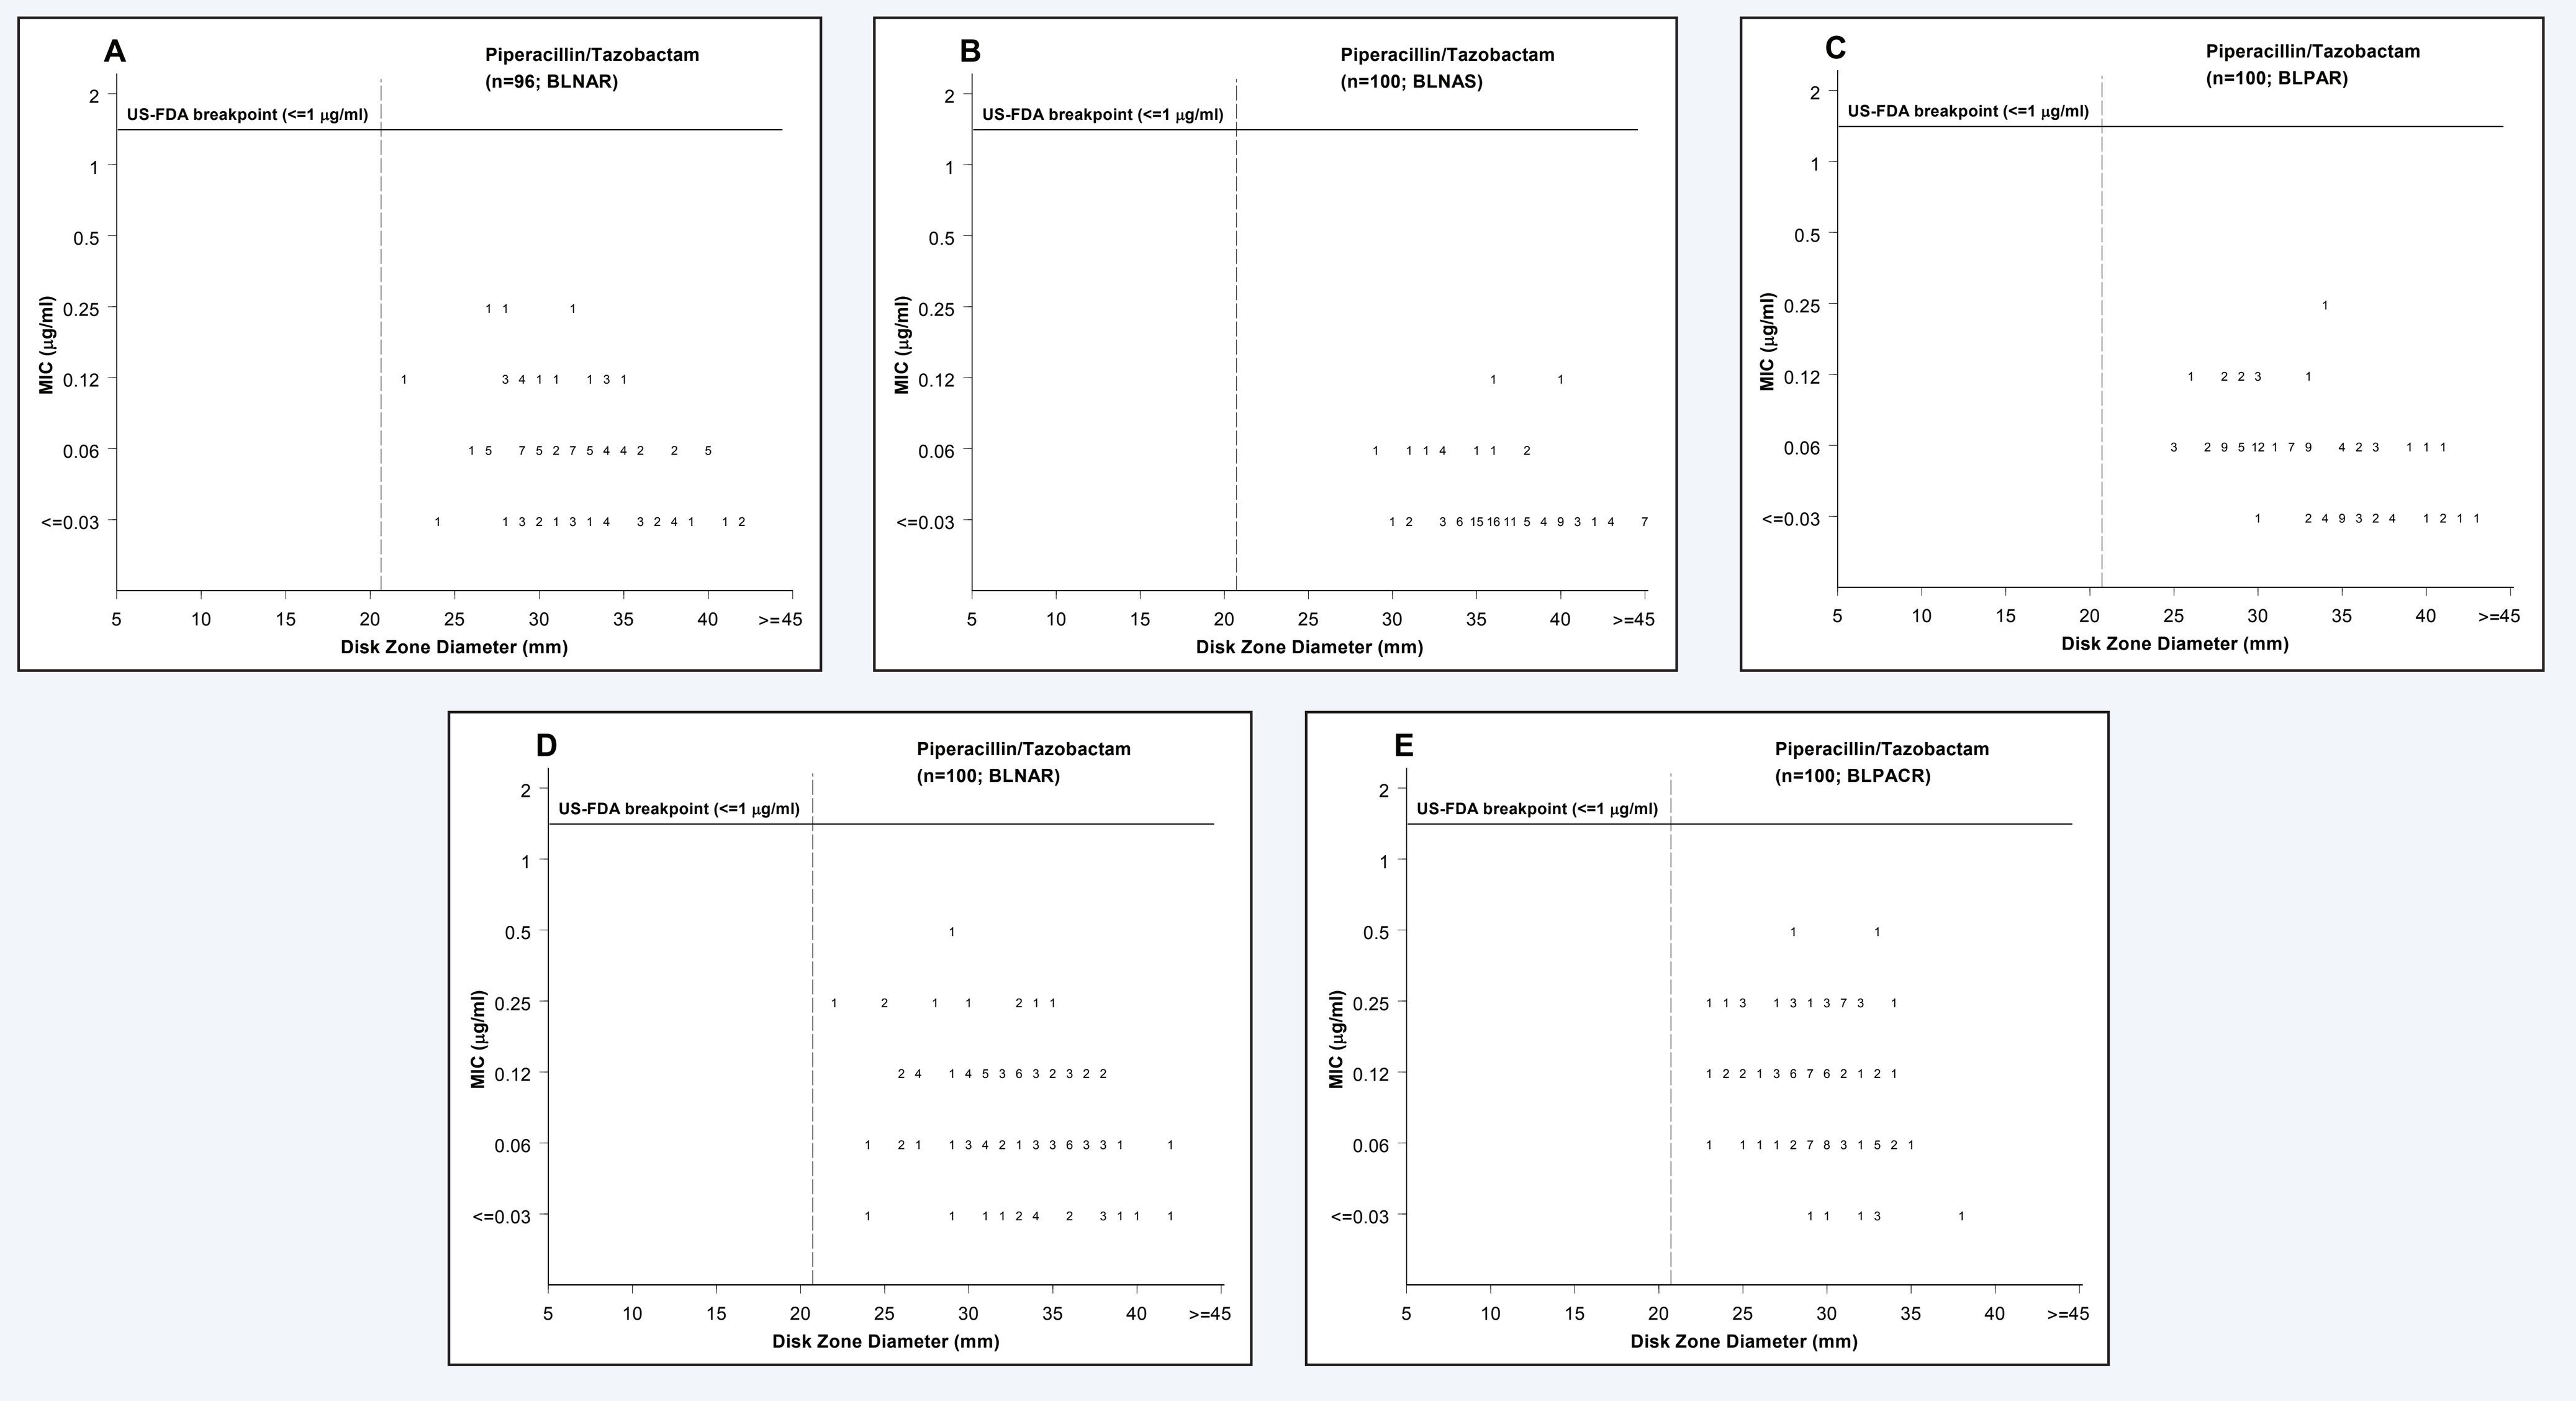
A
Piperacillin/Tazobactam (n=96; BLNAR)
B
Piperacillin/Tazobactam (n=100; BLNAS)
C
Piperacillin/Tazobactam (n=100; BLPAR)
D
Piperacillin/Tazobactam (n=100; BLNAR)
E
Piperacillin/Tazobactam (n=100; BLPACR)
US-FDA breakpoint (<=1 µg/ml)
MIC (µg/ml)
2
1
0.5
0.25
0.12
0.06
<=0.03
Disk Zone Diameter (mm)
5
10
15
20
25
30
35
40
>=45

CONCLUSIONS

- BLNAR and BLPACR *H. influenzae* are occurring in clinical practice with greater regularity, especially in Japan.
- Some agents previously considered as inactive against BLNAR *H. influenzae* (due to limited clinical data) have emerged as having excellent potency (piperacillin/tazobactam) and should be given accurate susceptible breakpoints as proposed here (MIC at ≤1 µg/ml [US-FDA] and correlate disk diffusion zones at ≥21 mm), and removal of any disclaimer regarding BLNAR strains. These conclusions are supported by PD target attainment simulations for the current MIC breakpoints.
- Proposed piperacillin/tazobactam breakpoints were without error when applied to the SENTRY Program and recent Japanese *H. influenzae* collections (496 strains).
- Current CLSI disk diffusion testing methods for several agents when testing *H. influenzae* do not accurately categorize the susceptibility of BLNAR isolates. A systematic re-evaluation appears necessary to prevent misguiding chemotherapy (see Table 3).

SELECTED REFERENCES

1. Alou L, Gimenez MJ, Sevillano D, Aguilar L, Gonzalez N, Echeverria O, Torrico M, Coronel P, Prieto J (2007). Are ß-lactam breakpoints adequate to define non-susceptibility for all *Haemophilus influenzae* resistance phenotypes from a pharmacodynamic point of view? *J Antimicrob Chemother* 59: 652-657.
2. Clinical and Laboratory Standards Institute. (2006). *M7-A7, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - seventh edition.* Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute. (2008). *M100-S18, Performance standards for antimicrobial susceptibility testing, 18th informational supplement.* Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute. (2006). *M2-A9. Performance standards for antimicrobial disk susceptibility tests; approved standard - ninth edition.* Wayne, PA: CLSI.
5. Doern GV, Jorgensen JH, Thornsberry C, Snapper H (1990). Disk diffusion susceptibility testing of *Haemophilus influenzae* using *Haemophilus* Test Medium. *Eur J Clin Microbiol Infect Dis* 9: 329-336.
6. Garcia-de-Lomas J, Lerma M, Cebrian L, Juan-Banon JL, Coronel P, Gimenez MJ, Aguilar L (2007). Influence of *Haemophilus influenzae* ß-lactamase production and/or *ftsI* gene mutations on in vitro activity of and susceptibility rates to aminopenicillins and second- and third-generation cephalosporins. *Int J Antimicrob Agents* 30: 190-192.
7. Gomi K, Watanabe A, Aoki S, Kikuchi T, Fuse K, Nukiwa T, Kurokawa I, Fujimura S (2007). Antibacterial activity of carbapenems against clinically isolated respiratory bacterial pathogens in Japan between 2005 and 2006. *Int J Antimicrob Agents* 29: 586-592.
8. Hasegawa K, Yamamoto K, Chiba N, Kobayashi R, Nagai K, Jacobs MR, Appelbaum PC, Sunakawa K, Ubukata K (2003). Diversity of ampicillin-resistance genes in *Haemophilus influenzae* in Japan and the United States. *Microb Drug Resist* 9: 39-46.
9. Hasegawa K, Chiba N, Kobayashi R, Murayama SY, Iwata S, Sunakawa K, Ubukata K (2004). Rapidly increasing prevalence of ß-lactamase-nonproducing, ampicillin-resistant *Haemophilus influenzae* type b in patients with meningitis. *Antimicrob Agents Chemother* 48: 1509-1514.
10. Hotomi M, Fujihara K, Billal DS, Suzuki K, Nishimura T, Baba S, Yamanaka N (2007). Genetic characteristics and clonal dissemination of ß-lactamase-negative ampicillin-resistant *Haemophilus influenzae* strains isolated from the upper respiratory tract of patients in Japan. *Antimicrob Agents Chemother* 51: 3969-3976.
11. Jones RN, Thornsberry C, Hindler J, Hugh Gerlach E, Doern GV (1992). The re-evaluation of the NCCLS ampicillin susceptibility tests with *Haemophilus* test medium: New interpretive criteria for the disk diffusion method. *Antimicrobic Newsletter* 8: 31-35.
12. Markowitz SM (1980). Isolation of an ampicillin-resistant, non-ß-lactamase-producing strain of *Haemophilus influenzae. Antimicrob Agents Chemother* 17: 80-83.
13. Morikawa Y, Kitazato M, Mitsuyama J, Mizunaga S, Minami S, Watanabe Y (2004). In vitro activities of piperacillin against ß-lactamase-negative ampicillin-resistant *Haemophilus influenzae. Antimicrob Agents Chemother* 48: 1229-1234.
14. Takahata S, Ida T, Senju N, Sanbongi Y, Miyata A, Maebashi K, Hoshiko S (2007). Horizontal gene transfer of *ftsI*, encoding penicillin-binding protein 3, in *Haemophilus influenzae. Antimicrob Agents Chemother* 51: 1589-1595.