ASM Microbe 2017
Friday - 57

Activity of Ceftazidime-Avibactam and Comparator Agents Tested against Isolates Harboring β-Lactamase Genes Detected Using Whole Genome Sequencing Analysis

M Castanheira, AP Davis, LM Deshpande, RE Mendes, HS Sader

JMI Laboratories, North Liberty, Iowa, USA

Contact Info
Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

AMENDED ABSTRACT

Background: An evolving scenario of β-lactamase (BL)-producing isolates was recently documented in US hospitals, highlighting the importance of surveillance. We used whole genome sequencing analysis (WGS) to screen for BLs in 909 *Escherichia coli* (EC) and *Klebsiella* spp. isolates collected from US hospitals during 2016. Additionally, we evaluated the activity of ceftazidime-avibactam (CAZ-AVI) and comparators tested against these isolates.

Methods: Isolates were susceptibility tested by CLSI reference broth microdilution methods. Isolates submitted to WGS displayed MIC values ≥2 µg/mL for at least 2 of the following agents: ceftazidime, ceftriaxone, aztreonam, or cefepime. *De novo* assembly and screening for BL genes and multilocus sequence typing were evaluated using an in-house-developed pipeline.

Results: Among 558 EC, 292 *K. pneumoniae* (KPN), and 59 *K. oxytoca* (KOX), 661 isolates carried *bla*$_{\text{CTX-M}}$ (478 EC, 172 KPN, and 9 KOX). Three isolates carried 2 *bla*$_{\text{CTX-M}}$ alleles. The most common genes were *bla*$_{\text{CTX-M-15}}$ (467 isolates), *bla*$_{\text{CTX-M-27}}$ (91), *bla*$_{\text{CTX-M-14}}$ (59), and *bla*$_{\text{CTX-M-55}}$ (23), but ≥13 other variants were detected. The *bla*$_{\text{OXA-1/30}}$ was noted among 313 isolates (312 carrying other BL genes). SHV ESBL genes (11 variants) were detected among 73 isolates, and SHV was the only ESBL gene among 14 isolates. Transferable AmpC genes were detected among 65 isolates and *bla*$_{\text{CMY-2}}$ was the most common (41 isolates). The *bla*$_{\text{KPC-2}}$ and *bla*$_{\text{KPC-3}}$ were observed among 31 and 54 isolates, respectively. Isolates included 74 KPN, 57 of them belonging to clonal complex 11 (ST258-like). One KPN isolate carried *bla*$_{\text{OXA-232}}$. Another 23 BL genes were detected. The highest CAZ-AVI MIC for all isolates tested was 8 µg/mL. Meropenem (MER), tigecycline (TIG), and colistin (COL) were active against all isolates carrying genes encoding CTX-M group 9, SHV ESBL, and CMY-2 isolates, but CAZ-AVI was the only antimicrobial that inhibited all isolates producing KPC or CTX-M group 1.

Conclusions: CTX-M enzymes continue to be the most prevalent ESBL in US hospitals. KPC producers, including ST258-like KPN, were also prevalent. CAZ-AVI inhibited all tested isolates at ≤8 µg/mL.

INTRODUCTION

- The prevalence of β-lactamases in the US differs from other countries regarding the occurrence and distribution of β-lactamase-producing isolates and enzyme types
- Isolates carrying *bla*$_{\text{CTX-M}}$ were considered endemic in European and Asian nosocomial and community settings a decade ago, but the first studies showing the dissemination of isolates harboring *bla*$_{\text{CTX-M}}$ in the US date from 2007
 - Isolates harboring *bla*$_{\text{CTX-M-15}}$-like and *bla*$_{\text{CTX-M-14}}$-like rapidly spread in US hospitals after the initial reports
 - Currently the rates of isolates harboring *bla*$_{\text{CTX-M}}$ are becoming more similar to those observed in other nations
- The carbapenemase-emerging scenario in US hospitals is also different
 - Isolates carrying *bla*$_{\text{KPC}}$ were first described in North Carolina and later in New York City where these isolates became endemic in the latter geographic region and surrounding areas
 - Metallo-β-lactamase (MBL)-producing isolates were uncommon in US hospitals; however, this could rapidly change with the dissemination of *Enterobacteriaceae* isolates harboring *bla*$_{\text{NDM-1}}$ that has been reported in 22 US states
- In this study, we evaluated the presence of β-lactamases among 909 isolates of *Escherichia coli*, *Klebsiella oxytoca*, and *K. pneumoniae* collected during 2016 from US hospitals participating in the International Network For Optimal Resistance Monitoring (INFORM) program
 - We used a whole genome sequencing analysis approach to detect β-lactamase encoding genes and evaluate multilocus sequence typing (MLST) for selected isolates

MATERIALS AND METHODS

- A total of 6,432 *E. coli* (n=558), *K. oxytoca* (n=59), and *K. pneumoniae* (292) clinical isolates collected during 2016 from 84 US hospitals participating in the INFORM program were evaluated
- Species identification was confirmed by matrix-assisted laser desorption ionization-time of flight mass spectrometry, when needed
- Isolates were susceptibility tested against ceftazidime-avibactam (inhibitor tested at fixed 4 µg/mL) and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI)
 - Quality control (QC) was performed according to CLSI guidelines (M100-S27), and all QC MIC results were within acceptable ranges, as published in CLSI documents
 - Categorical interpretations for all comparator agents were those found in CLSI criteria in M100-S27 (2017), EUCAST breakpoint tables (version 7.0, January 2017), and/or United States Food and Drug Administration (US FDA) package inserts
- A total of 909 isolates displaying MIC values ≥2 µg/mL for at least 2 β-lactams (ie, ceftazidime, ceftriaxone, aztreonam, or cefepime) were further evaluated
 - Selected isolates were submitted to whole genome sequencing on a MiSeq (Illumina, San Diego, California, US) instrument targeting a 30X coverage
 - Sequences were *de novo* assembled and searched for the presence of acquired β-lactamases using a curated library and applying criteria of >94% sequencing identity and 40% minimum length coverage
 - MLST data was extracted and sequence types (STs) were analyzed

RESULTS

- A total of 909 isolates, including 558 (14.8% for this species) *E. coli*, 59 (12.3%) *K. oxytoca*, and 292 (13.3%) *K. pneumoniae,* were evaluated for the presence of β-lactamases
- Overall, *bla*$_{\text{CTX-M}}$ (661 positive results) was the most common β-lactamase gene detected among *E. coli* (478) and *K. pneumoniae* (172)
 - 504 (55.4%) of the isolates tested carried *bla*$_{\text{CTX-M}}$ group 1, and the vast majority of the genes detected within this group were *bla*$_{\text{CTX-M-15}}$ (471 isolates; 51.8% overall), but 7 other variants were observed
 - *bla*$_{\text{CTX-M}}$ group 9 was observed among 156 isolates and included 91 and 59 isolates carrying *bla*$_{\text{CTX-M-27}}$ or *bla*$_{\text{CTX-M-14}}$, respectively
 - 1 *bla*$_{\text{CTX-M-2}}$ and 1 *bla*$_{\text{CTX-M-8}}$ were also observed
- *bla*$_{\text{SHV}}$ and *bla*$_{\text{TEM}}$ variants were noted among 73 and 6 isolates, respectively
 - At least 11 *bla*$_{\text{SHV}}$ ESBL-encoding genes were detected, and *bla*$_{\text{SHV-12}}$-like genes (36 *bla*$_{\text{SHV-12}}$ and 14 *bla*$_{\text{SHV-12}}$-like) corresponded to 50/73 positive results
 - Most isolates carrying *bla*$_{\text{SHV}}$ ESBL genes were *K. pneumoniae* (57/73)
- 313 isolates carried *bla*$_{\text{OXA-1}}$, also known as *bla*$_{\text{OXA-30}}$; however, this gene was detected alone in only 1 *E. coli* isolate and it was commonly observed in isolates harboring *bla*$_{\text{CTX-M}}$ genes
- 2 *K. pneumoniae* isolates carrying *bla*$_{\text{GES-17}}$ or *bla*$_{\text{VEB-9}}$ were detected
- Genes encoding transferrable AmpCs were detected among 65 (7.2%) *E. coli* and *Klebsiella* spp. isolates
 - *bla*$_{\text{CMY-2}}$ was the most common (50 positive results) and was mainly detected among *E. coli* (45 positive results
 - 10 isolates harbored *bla*$_{\text{DHA-1}}$ and 5 harbored *bla*$_{\text{FOX-5}}$-like
- 86 (9.5% of the isolates tested) of the *E. coli* (4) and *Klebsiella* spp. (82) isolates exhibiting elevated MIC values for cephalosporins and aztreonam carried genes encoding carbapenemases, including 85 *bla*$_{\text{KPC}}$ (31 *bla*$_{\text{KPC-2}}$ and 54 *bla*$_{\text{KPC-3}}$) and 1 *K. pneumoniae* isolate carrying *bla*$_{\text{OXA-232}}$
 - 3.4% (75/2,201) of all *K. pneumoniae* isolates harbored carbapenemase genes
 - Among 74 *K. pneumoniae* carrying *bla*$_{\text{KPC}}$, 57 were ST258-like
 - 3 isolates that carried *bla*$_{\text{KPC}}$ and 1 *K. pneumoniae* harboring *bla*$_{\text{OXA-232}}$ displayed low carbapenem MIC values
- Ceftazidime-avibactam inhibited all 909 isolates at the current breakpoint (≤8 µg/mL)
 - All isolates carrying *bla*$_{\text{CTX-M}}$ were inhibited at ≤2 µg/mL and categorized as susceptible to ceftazidime-avibactam
 - Ceftazidime-avibactam (MIC$_{50}$ and MIC$_{90}$, 0.25 and 1 µg/mL) displayed good activity against 49 isolates carrying *bla*$_{\text{SHV}}$ encoding ESBL enzymes alone
 - Isolates carrying transferrable AmpC genes alone (63) were susceptible to ceftazidime-avibactam (MIC$_{50}$ and MIC$_{90}$, 0.25 and 1 µg/mL)
- Isolates carrying ESBL and transferrable AmpCs were mostly susceptible to carbapenems
- Ceftazidime-avibactam (MIC$_{50}$ and MIC$_{90}$, 0.5 and 2 µg/mL) was the most active agent tested against 86 isolates carrying genes encoding carbapenemases, including 85 isolates carrying *bla*$_{\text{KPC}}$
 - This combination inhibited all isolates carrying carbapenemase genes at ≤4 µg/mL and was more active when compared to tigecycline (98.8% susceptible by US FDA breakpoints) or colistin (81.4% susceptible by EUCAST criteria)
 - Isolates carrying carbapenemase genes were highly resistant to comparator agents, including all β-lactams (1.2–9.3% susceptible) except ceftazidime-avibactam, fluoroquinolones (14.0–23.3% susceptible), gentamicin (43.0% susceptible) and trimethoprim-sulfamethoxazole (12.8% susceptible; CLSI breakpoints)
 - Amikacin inhibited 67.4% of the isolates harboring carbapenemase genes when applying the CLSI breakpoints

CONCLUSIONS

- As seen in other studies, *bla*$_{\text{CTX-M}}$ was the most common β-lactamase gene detected among *E. coli* and *Klebsiella* spp. isolates collected in US hospitals during 2016
 - Isolates harboring *bla*$_{\text{SHV}}$ and *bla*$_{\text{CMY-2}}$-like genes were also frequently detected
 - Ceftazidime-avibactam inhibited all isolates harboring *bla*$_{\text{CTX-M}}$, *bla*$_{\text{SHV}}$, and *bla*$_{\text{CMY-2}}$-like and other ESBL enzymes that can hydrolyze broad-spectrum cephalosporins or aztreonam without carbapenemases at the current breakpoint (≤8 µg/mL, susceptible)
- Carbapenemases were detected among 86 isolates, and 85 of these isolates harbored *bla*$_{\text{KPC}}$ enzymes, including 57 ST258-like isolates
 - Ceftazidime-avibactam inhibited all isolates carrying carbapenemase genes at ≤4 µg/mL

Figure 1 Results by organism and β-lactamase genes for 909 *E. coli* and *Klebsiella* spp. isolates collected during 2016 in US as part of the INFORM surveillance program displaying elevated MIC values (≥2 µg/mL) for at least 2 of the following β-lactams: ceftazidime, ceftriaxone, aztreonam, cefepime

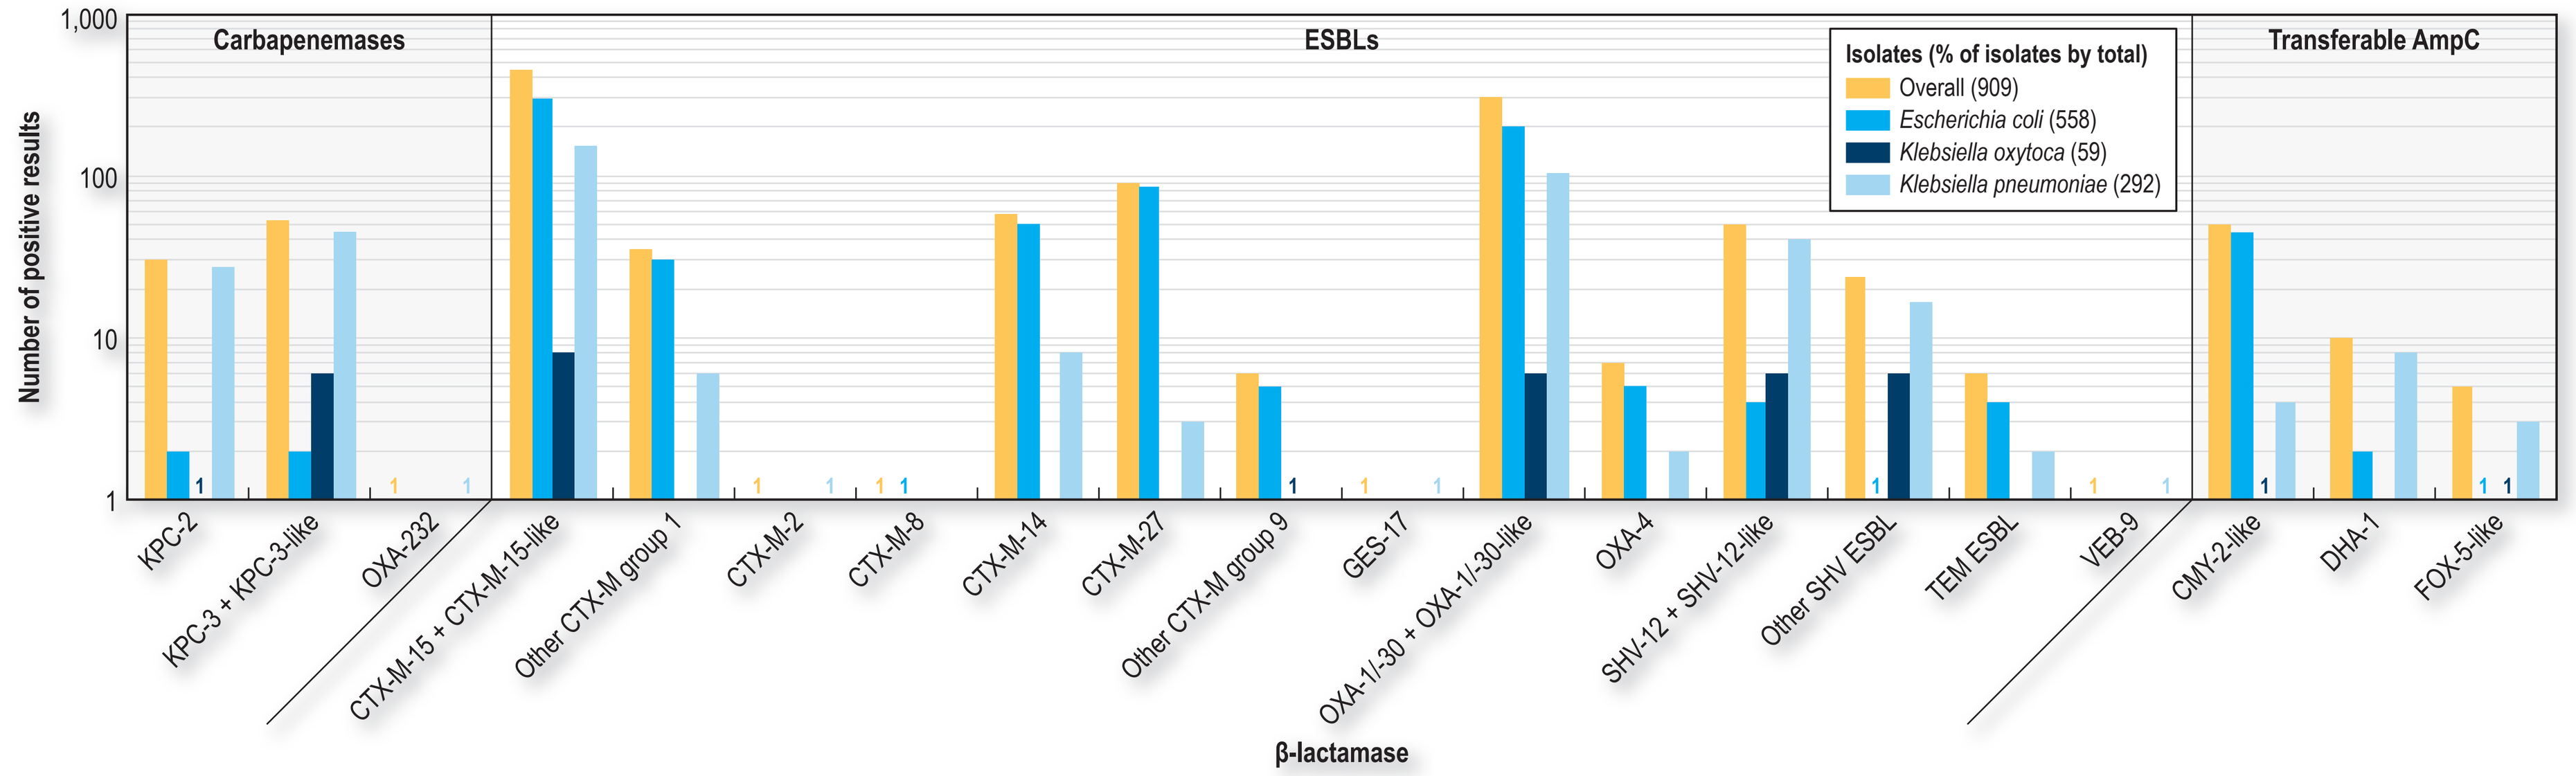
Number of positive results
1,000
100
10
1
Carbapenemases
ESBLs
Transferable AmpC
Isolates (% of isolates by total)
Overall (909)
Escherichia coli (558)
Klebsiella oxytoca (59)
Klebsiella pneumoniae (292)
KPC-2
KPC-3 + KPC-3-like
OXA-232
CTX-M-15 + CTX-M-15-like
Other CTX-M group 1
CTX-M-2
CTX-M-8
CTX-M-14
CTX-M-27
Other CTX-M group 9
GES-17
OXA-1/-30 + OXA-1/-30-like
OXA-4
SHV-12 + SHV-12-like
Other SHV ESBL
TEM ESBL
VEB-9
CMY-2-like
DHA-1
FOX-5-like
β-lactamase

Figure 2 Activity of ceftazidime-avibactam and comparator agents against isolates producing β-lactamases collected during 2016 in US as part of the INFORM surveillance program

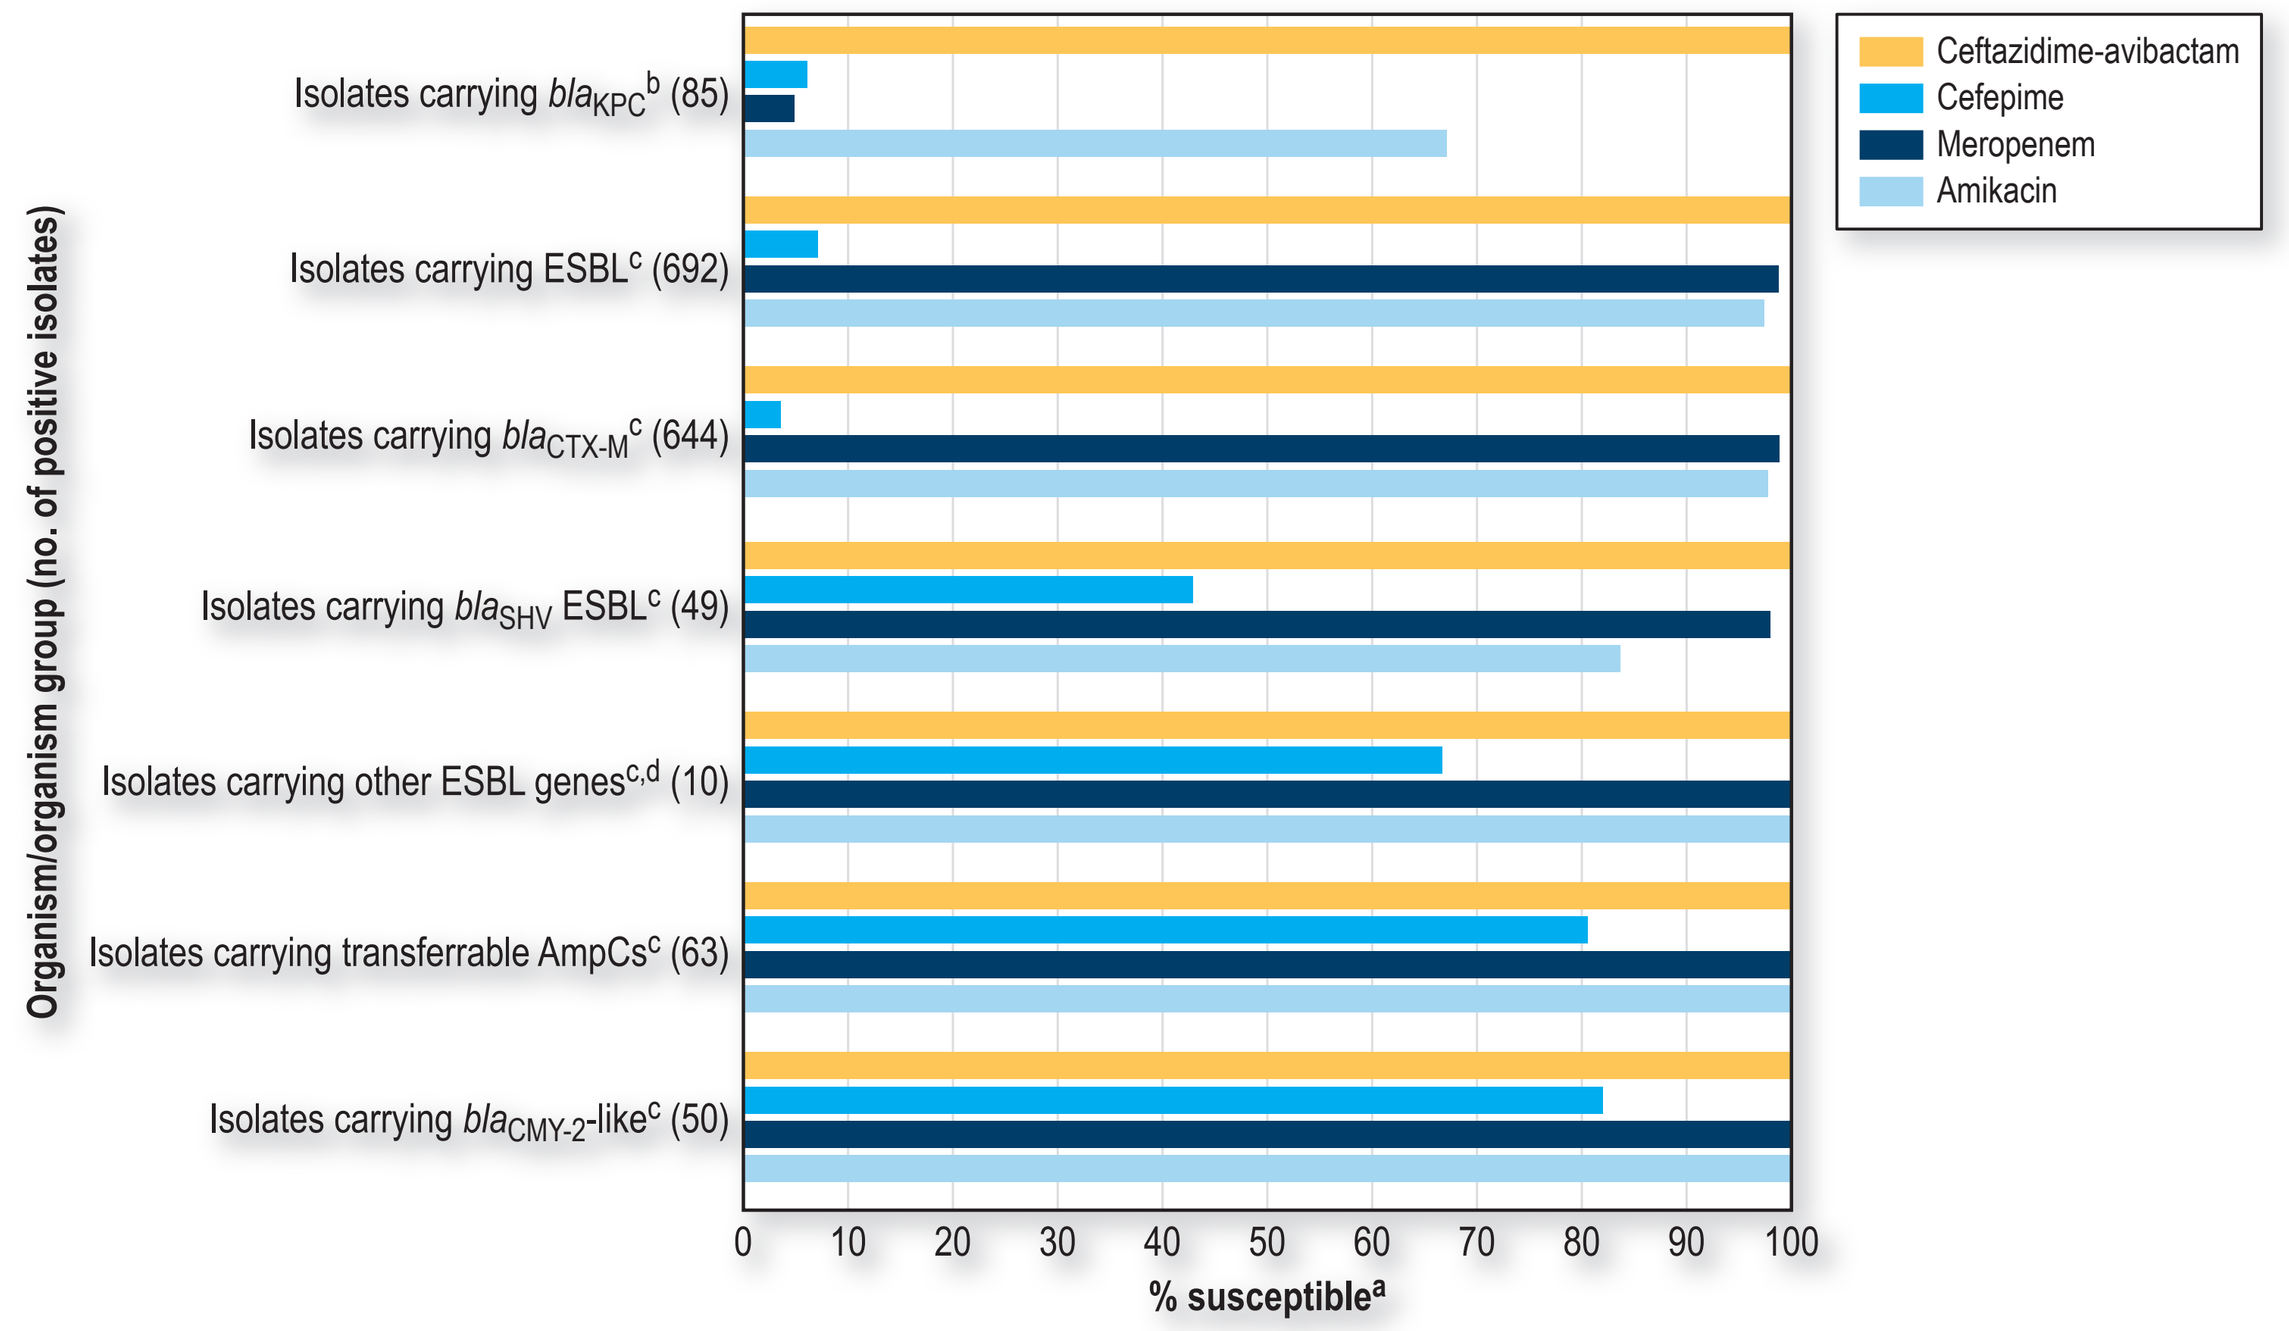
Organism/organism group (no. of positive isolates)
Isolates carrying blaKPC b (85)
Isolates carrying ESBL c (692)
Isolates carrying blaCTX-M c (644)
Isolates carrying blaSHV ESBL c (49)
Isolates carrying other ESBL genes c,d (10)
Isolates carrying transferrable AmpCs c (63)
Isolates carrying blaCMY-2-like c (50)
0 10 20 30 40 50 60 70 80 90 100
% susceptible a
Ceftazidime-avibactam
Cefepime
Meropenem
Amikacin

ACKNOWLEDGEMENTS

The authors acknowledge the contributions of the US hospitals participating on the International Network For Optimal Resistance Monitoring program.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation fees for services in relation to preparing the abstract and poster. Allergan was not involved in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2017). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi. Accessed February 2017.
2. Castanheira M, Farrell SE, Deshpande LM, et al. (2013). Prevalence of β-lactamase encoding genes among *Enterobacteriaceae* bacteremia isolates collected in 26 USA hospitals: Report from the SENTRY Antimicrobial Surveillance Program (2010). *Antimicrob Agents Chemother* 57: 3012–3020.
3. Castanheira M, Farrell SE, Krause KM, et al. (2014). Contemporary diversity of β-lactamases among Enterobacteriaceae in the nine United States census regions and ceftazidime-avibactam activity tested against isolates producing the most prevalent β-lactamase groups. *Antimicrob Agents Chemother* 58: 833–838.
4. Castanheira M, Mendes RE, Jones RN, et al. (2016). Changes in the frequencies of beta-lactamase genes among *Enterobacteriaceae* isolates in U.S. Hospitals, 2012 to 2014: Activity of ceftazidime-avibactam tested against beta-lactamase-producing isolates. *Antimicrob Agents Chemother* 60: 4770–4777.
5. CDC (2015). Tracking CRE infections. Available at: http://www.cdc.gov/hai/organisms/cre/TrackingCRE.html. Accessed April 2016.
6. Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.
7. EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.

Visit http://www.allergancongressposters.com/189733

Allergan

ASM Microbe 2017, June 1–5 in New Orleans, Louisiana