Friday - 76

ASM Microbe 2017
Contact Information:
Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Susceptibility of Ceftolozane-Tazobactam against Isolates Collected from Intensive Care Unit Patients in US Hospitals from 2014–2016

MA Pfaller, D Shortridge, M Castanheira, JM Streit, RK Flamm

JMI Laboratories, North Liberty, Iowa, USA

http://merck.creative.studios.s3-website-us-east-1.amazonaws.com/ASM_Microbe_2017/ASM_Microbe_2017-Shortridge_Susceptibility_Ceftolozane_FRIDAY-76.pdf

Introduction

- Ceftolozane is a novel oxyimino-aminothiazolyl cephalosporin with potent activity against *Enterobacteriaceae* (similar to other oxyimino-aminothiazolyl cephalosporins) and has demonstrated greater activity than ceftazidime against *Pseudomonas aeruginosa*
 - Ceftolozane maintains its stability against many *P. aeruginosa* resistance mechanisms, including AmpC hyperproduction and efflux mechanisms
 - Porin deficiency has little effect on ceftolozane
 - However, as with other oxyimino-aminothiazolyl cephalosporins, ceftolozane's activity can be compromised in bacteria producing extended-spectrum β-lactamases (ESBLs), stably derepressed AmpC β-lactamases, and carbapenemases
- Tazobactam, a penicillanic acid sulfone, is a well-established β-lactamase inhibitor that extends the spectrum of β-lactam agents
- Ceftolozane-tazobactam was approved by the US Food and Drug Administration in 2014 and the European Medicines Agency in 2015 to treat patients with complicated intra-abdominal infections in combination with metronidazole and to treat complicated urinary tract infections, including pyelonephritis, and is in a Phase 3 clinical study for hospital-acquired and ventilator-associated pneumonia
- For the past decade, nosocomial infections in intensive care unit patients caused by *P. aeruginosa* and *Enterobacteriaceae*, as well as antimicrobial resistance to them, have increased in prevalence worldwide; associated morbidity and mortality rates have also increased
- Empiric and targeted therapies to treat infections caused by these organisms are becoming increasingly limited in efficacy
- In this study, the activity of C-T and comparators was assessed against gram-negative pathogens from intensive care unit (ICU) patients in 29 United States (US) hospitals participating in the Program to Assess Ceftolozane-Tazobactam Susceptibility (PACTS)

Materials and Methods

- A total of 2,956 GN isolates were collected in 2014–2016 from 29 US hospitals' ICU patients and tested for susceptibility by the CLSI broth microdilution method at a central laboratory (JMI Laboratories, North Liberty, Iowa, USA)
- Isolates were from documented infections and only 1 isolate per patient-infection episode was included
- The most common infection type for this study was pneumonia followed by bloodstream; the sources of isolates in this study are shown in Figure 1
- In addition to C-T, amikacin (AMK), cefepime (FEP), ceftazidime (CAZ), colistin (COL), levofloxacin (LVX), meropenem (MER), and piperacillin-tazobactam (TZP) were tested
- Antibiotic-resistant phenotypes identified using CLSI (2017) clinical breakpoints included extended-spectrum beta-lactamase (ESBL), carbapenem-resistant *Enterobacteriaceae* (ENT; CRE), ceftazidime-nonsusceptible (CAZ-NS), meropenem-nonsusceptible (MER-NS), and multidrug-resistant (MDR)
 - Given that the ESBL phenotype may also include carbapenemase producing *Enterobacteriaceae*, the ESBL isolates analyzed were non-carbapenem resistant (ESBL, nonCRE)
 - ESBL screening criteria used was according to CLSI M100
 - MDR classification was according to Magiorakos et al. (2012), where the isolate is nonsusceptible to ≥3 drug classes

Figure 1 Most common infection types found in US ICUs in this study

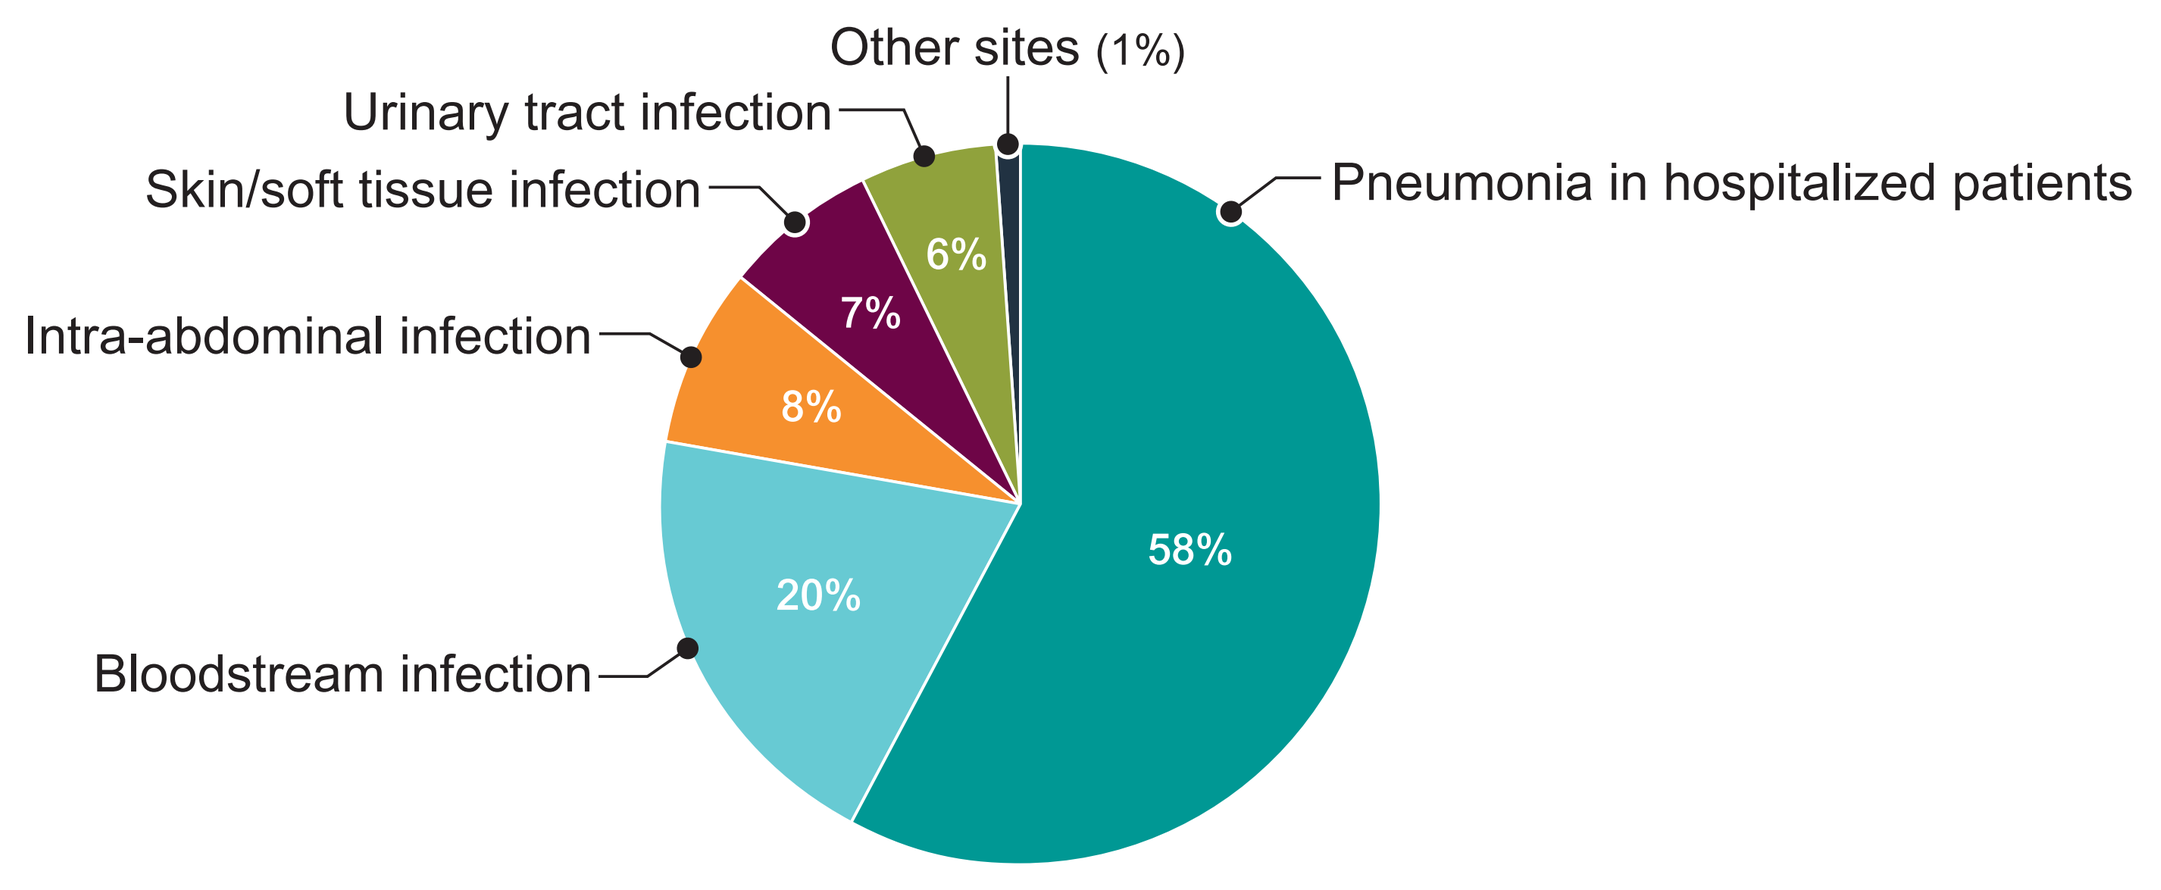
Other sites (1%)
Urinary tract infection
Skin/soft tissue infection
Intra-abdominal infection
Bloodstream infection
Pneumonia in hospitalized patients
6%
7%
8%
20%
58%

- CLSI (2017) C-T breakpoints for ENT are ≤2.0 mg/L susceptible (S), 4.0 mg/L intermediate (I) and ≥8.0 mg/L resistant (R); *P. aeruginosa* C-T breakpoints are ≤4.0 mg/L for S, 8.0 mg/L for I, and ≥16.0 mg/L for R (Figures 2 and 3)
- EUCAST (2017) C-T breakpoints for ENT are ≤1.0 mg/L for S and >1.0 mg/L R; *Pseudomonas* spp. C-T breakpoints are ≤4.0 mg/L for S and >4mg/L for R
- EUCAST (2017) COL breakpoints are used for ENT

Results

- The 5 most common organisms isolated from ICU patients were *P. aeruginosa* (726 isolates), *E. coli* (651), *K. pneumoniae* (512), *E. cloacae* species complex (316), and *Serratia marcescens* (198) (Table 1)
- The C-T MIC distributions for the most common enteric species are shown in Figure 2
- The *P. aeruginosa* MIC distributions for C-T and 2 comparators are shown in Figure 3
- Activities of C-T and 7 comparators against the common species are shown in Table 2
- Overall, C-T had potent activity (92.4%S) against *Enterobacteriaceae*; only AMK and MER were more active
 - C-T had excellent activity against *E. coli* (98.2%S), including ESBL-phenotype, nonCRE *E. coli* (92.4%S)
 - Against *K. pneumoniae*, C-T was slightly less active (92.2%S)
- For *P. aeruginosa*, C-T was the most active beta-lactam (97.2%S)
 - Among all comparators, only COL was more active (99.0%S), while 97.2% of isolates were S to AMK and 77.5% were S to MER
 - For MER-NS *P. aeruginosa*, 87.7% were S to C-T; for CAZ-NS, 83.3% were S to C-T; for TZP-NS, 89.3% were S to C-T; and for MDR, 87.4% were S to C-T

Table 1 The 5 most common gram-negative species in ICU infections

Species	Number
Enterobacter cloacae	316
Escherichia coli	651
Klebsiella pneumoniae	512
Pseudomonas aeruginosa	726
Serratia marcescens	198

Table 2 Activity of ceftolozane-tazobactam and comparator antimicrobial agents when tested against US 2014–2016 isolates

Organism group (no. tested) antimicrobial agent	CLSI† %S	EUCAST† %S	$MIC_{50/90}$
Enterobacteriaceae (2,230)‡			
Ceftolozane-tazobactam	92.4	90.0	0.25 / 2
Amikacin	99.5	99.1	2 / 4
Cefepime	89.1	87.2	≤0.5 / 4
Ceftazidime	84.1	81.6	0.25 / 32
Colistin		80.7	≤0.5 / >8
Levofloxacin	84.6	81.0	≤0.12 / >4
Meropenem	97.3	98.0	0.03 / 0.06
Piperacillin-tazobactam	88.1	84.3	2 / 32
CRE (56)			
Ceftolozane-tazobactam	10.7	7.1	>32 / >32
Amikacin	82.1	75.0	4 / 32
Cefepime	17.9	10.7	>16 / >16
Ceftazidime	7.1	7.1	>32 / >32
Colistin		87.0	≤0.5 / >8
Levofloxacin	39.3	26.8	>4 / >4
Meropenem	3.6	19.6	8 / >32
Piperacillin-tazobactam	7.1	7.1	>64 / >64
ESBL phenotype, nonCRE (212)			
Ceftolozane-tazobactam	86.8	77.8	0.5 / 8
Amikacin	99.1	97.2	2 / 8
Cefepime	26.9	22.6	16 / >16
Ceftazidime	25.5	11.3	16 / >32
Colistin		96.7	≤0.5 / ≤0.5
Levofloxacin	40.1	33.5	>4 / >4
Meropenem	97.6	100.0	0.03 / 0.06
Piperacillin-tazobactam	69.8	55.7	8 / >64
MDR *Enterobacteriaceae* (191)			
Ceftolozane-tazobactam	50.3	43.5	2 / >32
Amikacin	93.7	90.6	2 / 8
Cefepime	30.9	24.1	16 / >16
Ceftazidime	17.8	13.1	32 / >32
Colistin		68.8	≤0.5 / >8
Levofloxacin	37.7	24.6	>4 / >4
Meropenem	69.6	77.0	0.06 / 16
Piperacillin-tazobactam	36.6	27.7	64 / >64
Escherichia coli (651)			
Ceftolozane-tazobactam	98.2	96.8	0.12 / 0.5
Amikacin	99.7	98.9	2 / 4
Cefepime	84.5	83.7	≤0.5 / >16
Ceftazidime	84.5	82.0	0.25 / 16
Colistin		99.7	≤0.5 / ≤0.5
Levofloxacin	64.7	63.7	≤0.12 / >4
Meropenem	99.5	99.7	≤0.015 / 0.03
Piperacillin-tazobactam	93.1	90.2	2 / 8
ESBL phenotype, nonCRE *E. coli* (119)			
Ceftolozane-tazobactam	92.4	85.7	0.5 / 2
Amikacin	98.3	95.0	2 / 8
Cefepime	18.5	16.8	>16 / >16
Ceftazidime	17.6	4.2	16 / >32
Colistin		98.3	≤0.5 / ≤0.5
Levofloxacin	16.8	15.1	>4 / >4
Meropenem	99.2	100.0	≤0.015 / 0.03
Piperacillin-tazobactam	86.6	72.3	4 / 64
MDR *E. coli* (47)			
Ceftolozane-tazobactam	80.9	74.5	0.5 / 16
Amikacin	95.7	91.5	4 / 8
Cefepime	14.9	12.8	>16 / >16
Ceftazidime	8.5	6.4	32 / >32

Organism group (no. tested) antimicrobial agent	CLSI† %S	EUCAST† %S	$MIC_{50/90}$
Colistin		95.7	≤0.5 / ≤0.5
Levofloxacin	6.4	4.3	>4 / >4
Meropenem	93.6	95.7	0.03 / 0.12
Piperacillin-tazobactam	59.6	38.3	16 / >64
Klebsiella pneumoniae (512)			
Ceftolozane-tazobactam	92.2	90.4	0.25 / 1
Amikacin	98.2	97.7	1 / 2
Cefepime	86.3	84.8	≤0.5 / 16
Ceftazidime	85.0	82.8	0.25 / 32
Colistin		98.4	≤0.5 / ≤0.5
Levofloxacin	90.2	85.5	≤0.12 / 2
Meropenem	94.3	95.9	0.03 / 0.03
Piperacillin-tazobactam	87.1	80.7	4 / 64
CRE *K. pneumoniae* (26)			
Ceftolozane-tazobactam	3.8	0.0	>32 / >32
Amikacin	65.4	53.8	8 / 32
Cefepime	15.4	7.7	>16 / >16
Ceftazidime	0.0	0.0	>32 / >32
Colistin		88.0	≤0.5 / >8
Levofloxacin	23.1	19.2	>4 / >4
Meropenem	3.8	19.2	8 / >32
Piperacillin-tazobactam	0.0	0.0	>64 / >64
ESBL-phenotype, nonCRE *K. pneumoniae* (65)			
Ceftolozane-tazobactam	76.9	66.2	1 / 16
Amikacin	100.0	100.0	2 / 4
Cefepime	26.2	16.9	16 / >16
Ceftazidime	21.5	4.6	32 / >32
Colistin		98.5	≤0.5 / ≤0.5
Levofloxacin	60.0	47.7	1 / >4
Meropenem	93.8	100.0	0.03 / 0.12
Piperacillin-tazobactam	53.8	35.4	16 / >64
MDR *K. pneumoniae* (57)			
Ceftolozane-tazobactam	38.6	28.1	8 / >32
Amikacin	84.2	78.9	4 / 32
Cefepime	12.3	7.0	>16 / >16
Ceftazidime	5.3	0.0	>32 / >32
Colistin		92.9	≤0.5 / 1
Levofloxacin	35.1	22.8	>4 / >4
Meropenem	49.1	63.2	2 / 32
Piperacillin-tazobactam	24.6	14.0	64 / >64
Enterobacter cloacae species complex (316)			
Ceftolozane-tazobactam	77.8	75.0	0.25 / 16
Amikacin	100.0	99.7	1 / 2
Cefepime	88.3	81.0	≤0.5 / 4
Ceftazidime	69.3	66.8	0.5 / >32
Colistin		77.8	≤0.5 / >8
Levofloxacin	94.6	91.1	≤0.12 / 0.5
Meropenem	95.6	96.2	0.03 / 0.06
Piperacillin-tazobactam	76.6	72.5	2 / >64
Pseudomonas aeruginosa (726)			
Ceftolozane-tazobactam	97.2	97.2	0.5 / 2
Amikacin	97.2	92.6	4 / 8
Cefepime	85.5	85.5	2 / 16
Ceftazidime	83.5	83.5	2 / 32
Colistin	99.0	99.0	1 / 2
Levofloxacin	75.8	67.1	0.5 / >4
Meropenem	77.5	77.5	0.5 / 8
Piperacillin-tazobactam	79.3	79.3	4 / 64

Organism group (no. tested) antimicrobial agent	CLSI† %S	EUCAST† %S	$MIC_{50/90}$
Meropenem nonsusceptible (MIC, ≥4 mg/L) *P. aeruginosa* (163)			
Ceftolozane-tazobactam	87.7	87.7	1 / 8
Amikacin	89.0	76.7	4 / 32
Cefepime	52.8	52.8	8 / >16
Ceftazidime	56.4	56.4	8 / >32
Colistin	98.2	98.2	1 / 2
Levofloxacin	33.7	21.5	>4 / >4
Meropenem	0.0	0.0	8 / 32
Piperacillin-tazobactam	44.8	44.8	32 / >64
Ceftazidime nonsusceptible (MIC, ≥16 mg/L) *P. aeruginosa* (120)			
Ceftolozane-tazobactam	83.3	83.3	2 / 16
Amikacin	87.5	77.5	4 / 32
Cefepime	33.3	33.3	16 / >16
Ceftazidime	0.0	0.0	32 / >32
Colistin	98.3	98.3	1 / 2
Levofloxacin	48.3	36.7	4 / >4
Meropenem	40.8	40.8	4 / 32
Piperacillin-tazobactam	8.3	8.3	64 / >64
Piperacillin-tazobactam nonsusceptible (≥32 mg/L) *P. aeruginosa* (150)			
Ceftolozane-tazobactam	89.3	89.3	2 / 8
Amikacin	91.3	80.7	4 / 16
Cefepime	39.6	39.6	16 / >16
Ceftazidime	26.7	26.7	32 / >32
Colistin	98.7	98.7	1 / 2
Levofloxacin	46.0	31.3	4 / >4
Meropenem	40.0	40.0	8 / 32
Piperacillin-tazobactam	0.0	0.0	64 / >64
MDR *P. aeruginosa* (159)			
Ceftolozane-tazobactam	87.4	87.4	1 / 8
Amikacin	88.1	73.6	8 / 32
Cefepime	43.4	43.4	16 / >16
Ceftazidime	47.2	47.2	16 / >32
Colistin	97.5	97.5	1 / 2
Levofloxacin	20.1	9.4	>4 / >4
Meropenem	16.4	16.4	8 / 32
Piperacillin-tazobactam	34.0	34.0	32 / >64

† Criteria as published by CLSI [2016] and EUCAST [2016]
‡ Organisms include: *Citrobacter amalonaticus* (3), *Citrobacter braakii* (4), *C. freundii* (21), *C. freundii* species complex (16), *C. koseri* (36), *C. youngae* (1), *Enterobacter aerogenes* (123), *E. asburiae* (7), *E. cloacae* (185), *E. cloacae* species complex (131), *E. kobei* (1), *E. taylorae* (1), *E. coli* (7651), *E. hermannii* (1), *Hafnia alvei* (5), *Klebsiella oxytoca* (180), *K. pneumoniae* (512), *K. variicola* (3), *Morganella morganii* (23), *Pantoea agglomerans* (2), *P. dispersa* (1), *Proteus mirabilis* (86), *P. penneri* (1), *P. vulgaris* (3), *Providencia rettgeri* (7), *P. stuartii* (9), *Serratia liquefaciens* (7), *Raoultella ornithinolytica* (4), *R. planticola* (2), *S. marcescens* (198), *S. odorifera* (1), *S. plymuthica* (1), unspeciated *Raoultella* (2)

Figure 2 MIC distribution of C-T for the most frequently isolated *Enterobacteriaceae* from US hospitals (PACTS, 2014–2016)

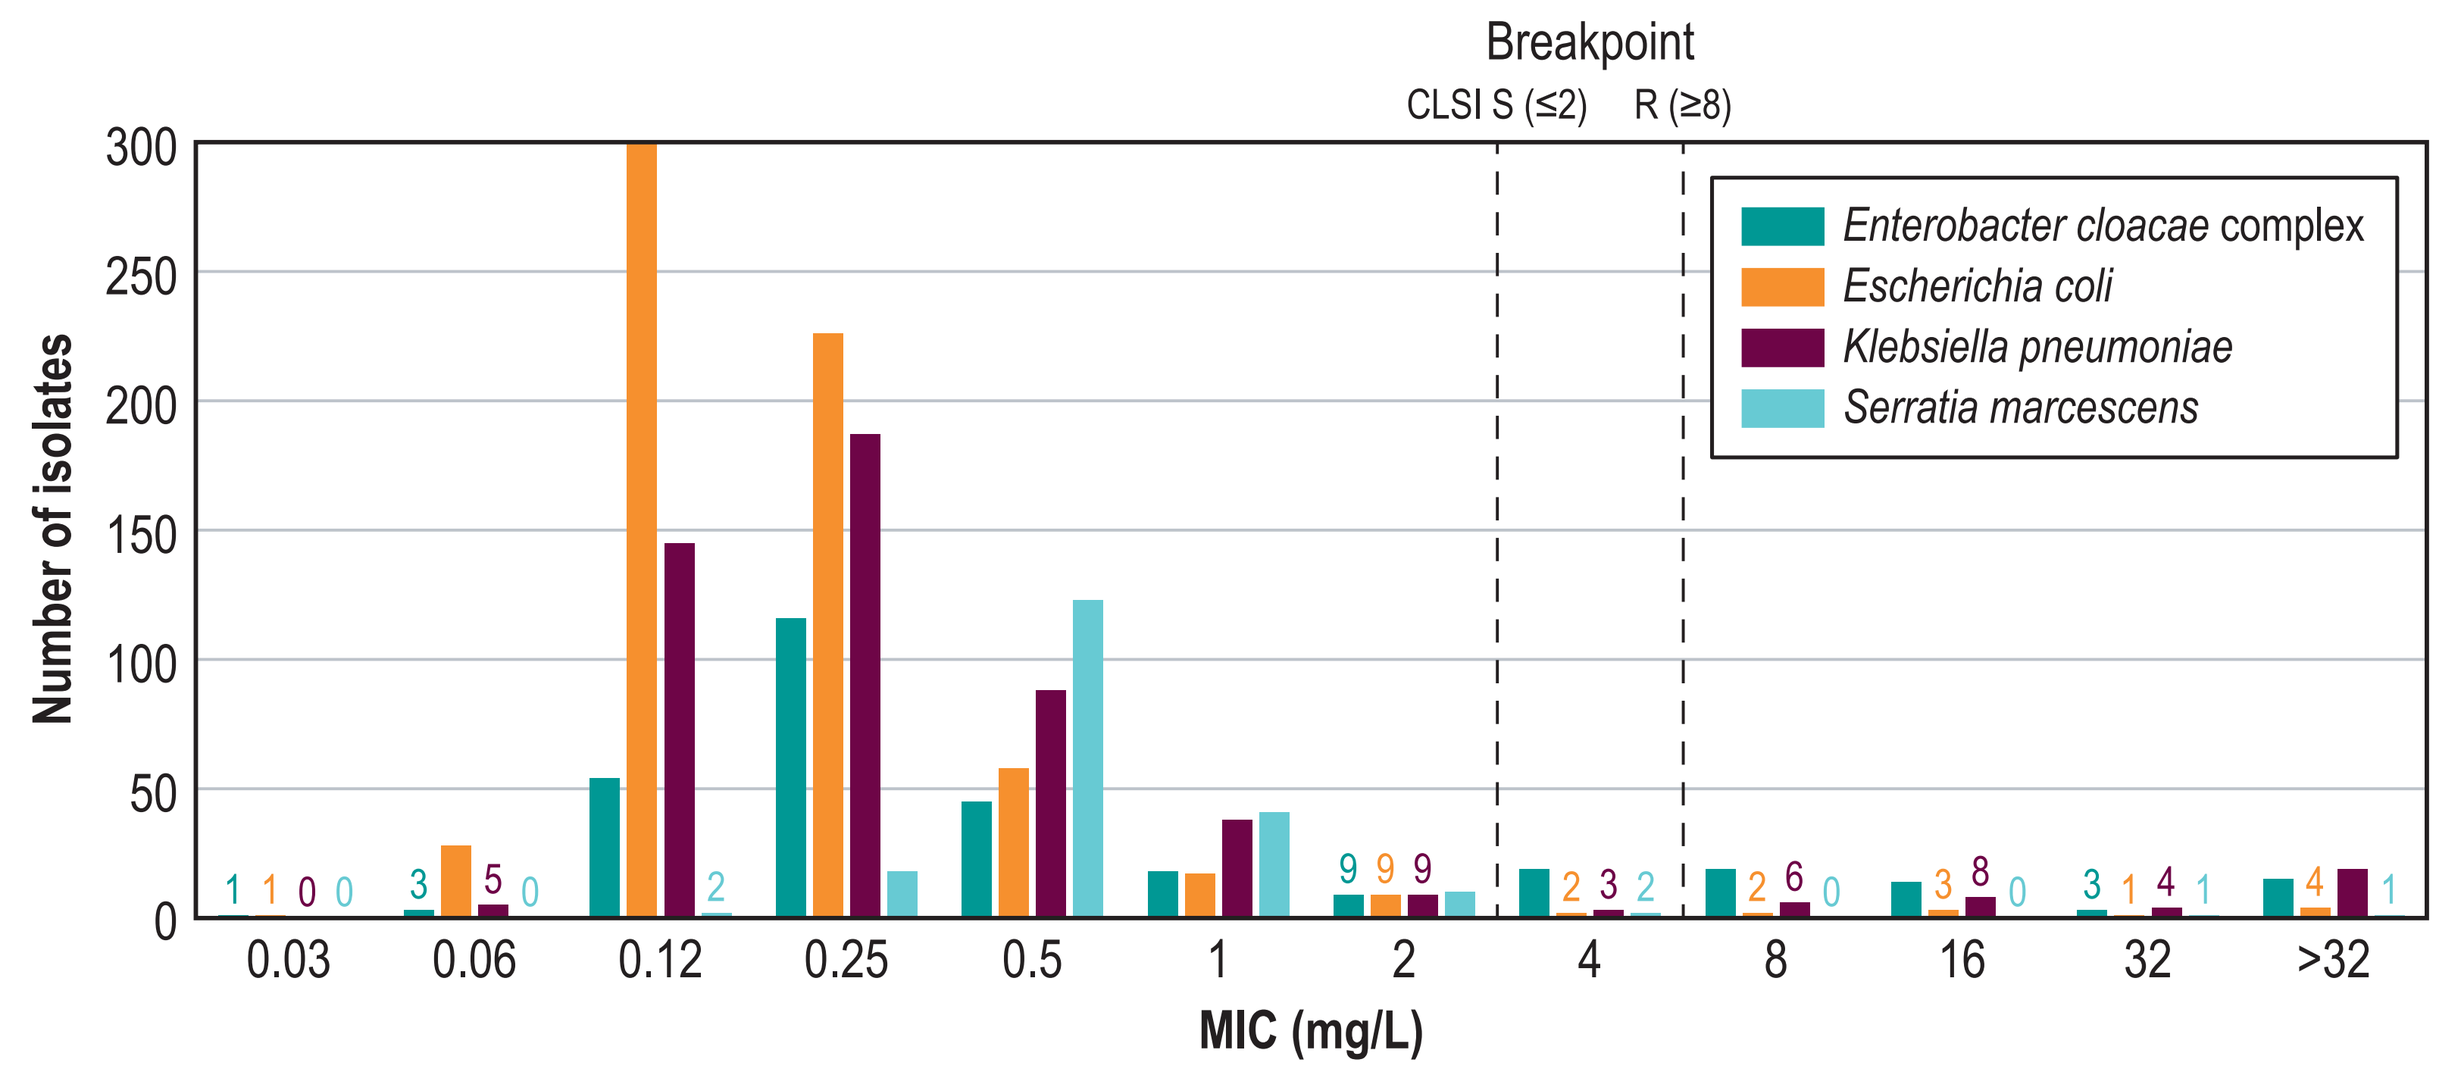
Breakpoint
CLSI S (≤2) R (≥8)
Enterobacter cloacae complex
Escherichia coli
Klebsiella pneumoniae
Serratia marcescens
Number of isolates
MIC (mg/L)
0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 >32

CLSI S (≤2 mg/L) and R (≥8 mg/L) breakpoints are indicated

Figure 3 The MIC distribution of ceftolozane-tazobactam (C-T), meropenem (MER), and piperacillin-tazobactam (TZP) for *P. aeruginosa* ICU isolates from US hospitals (PACTS, 2014–2016)

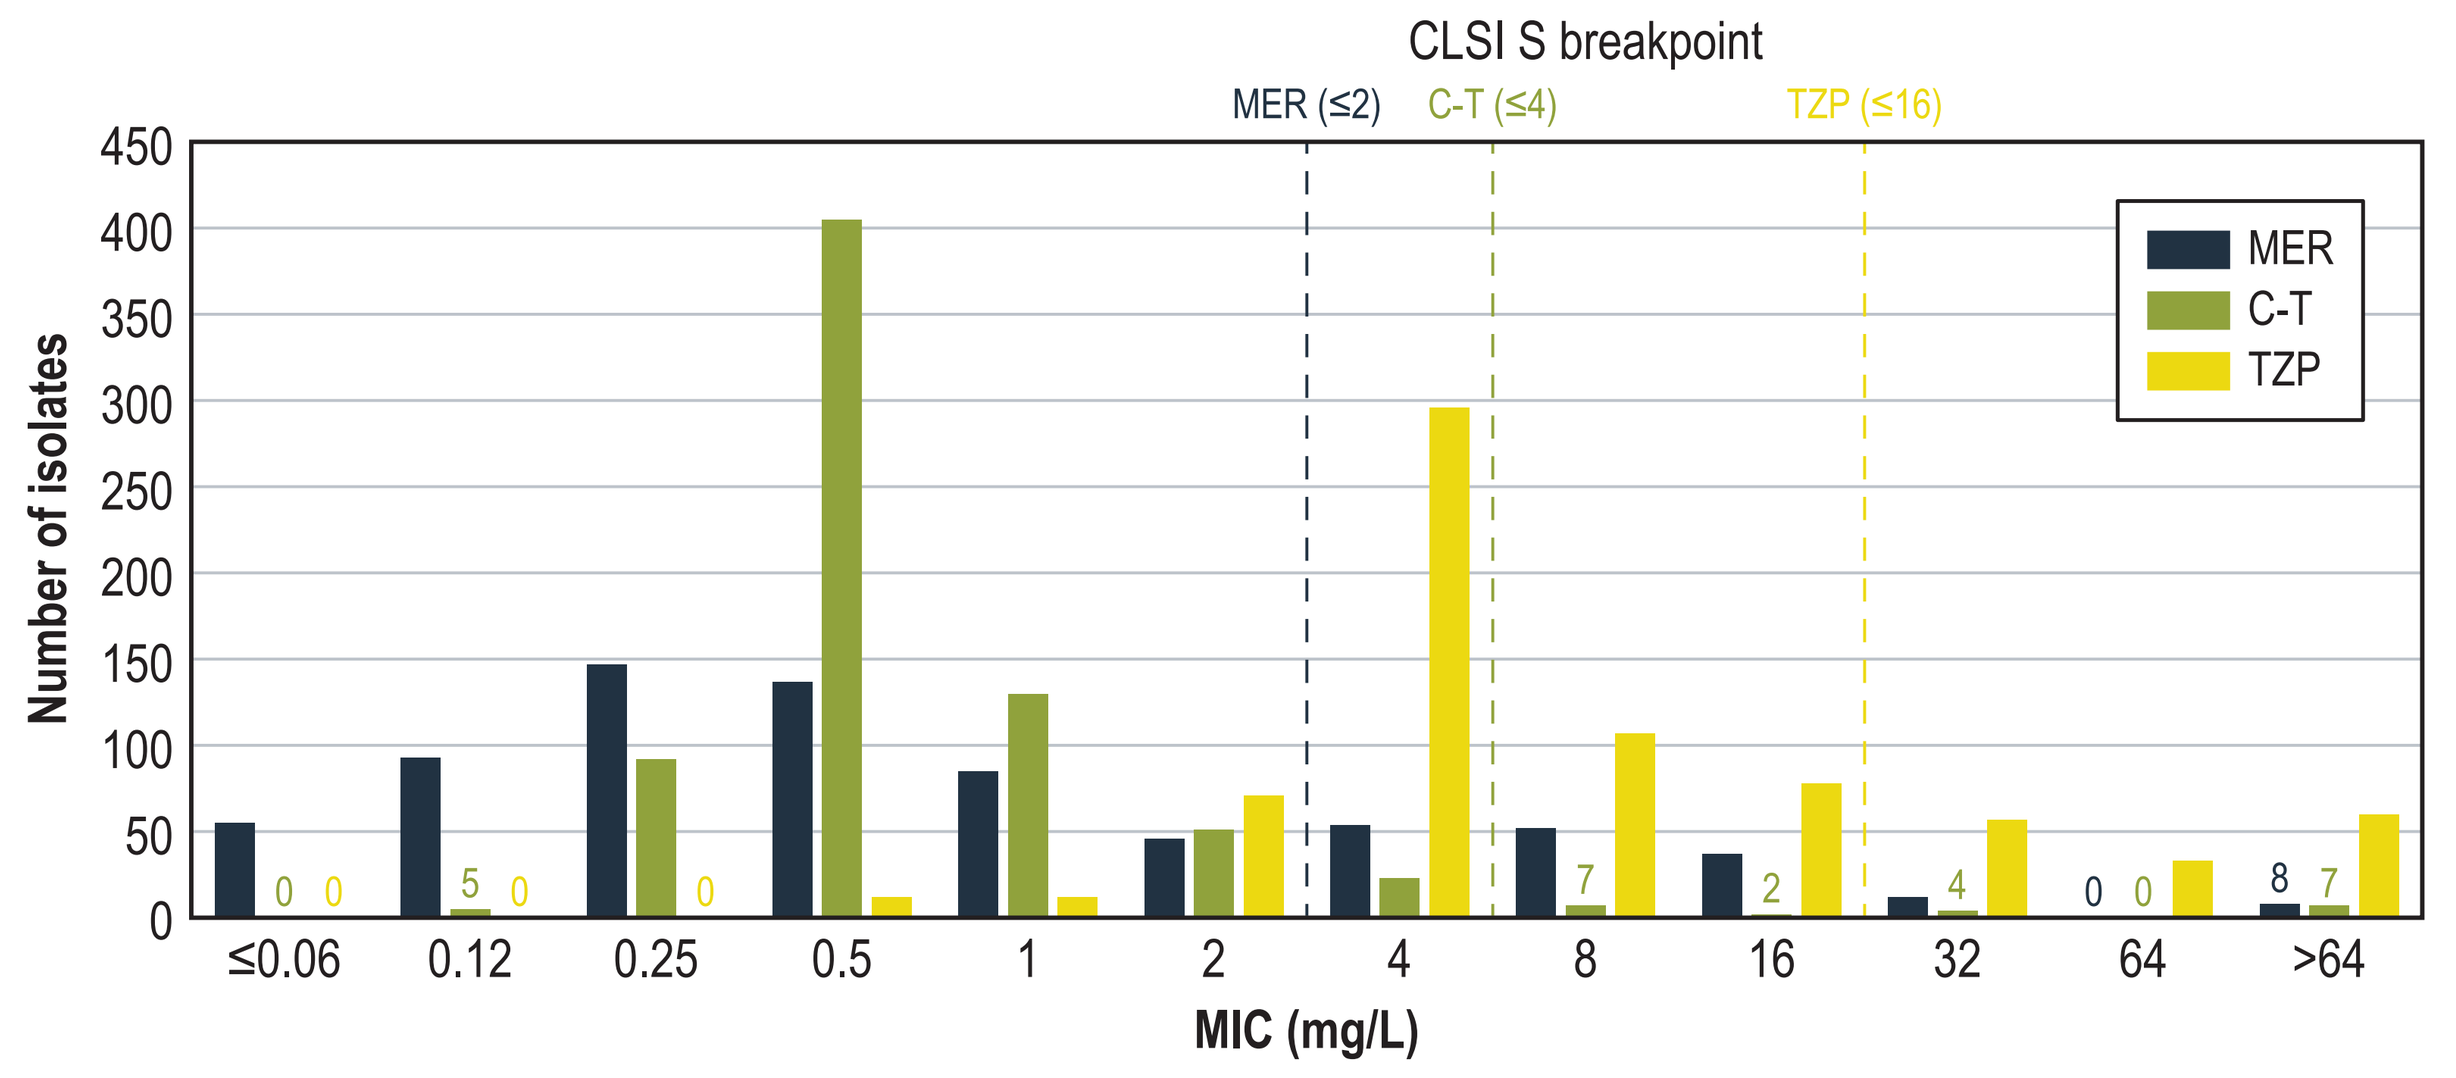
CLSI S breakpoint
MER (≤2) C-T (≤4) TZP (≤16)
MER
C-T
TZP
Number of isolates
MIC (mg/L)
≤0.06 0.12 0.25 0.5 1 2 4 8 16 32 64 >64

CLSI S breakpoint indicated for MER (≤2 mg/L), C-T (≤4 mg/L) and TZP (≤16 mg/L)

Conclusions

- Ceftolozane-tazobactam activity was assessed against 2,956 isolates from ICU patients collected in 29 US hospitals as a part of PACTS from 2014–2016
- Ceftolozane-tazobactam had very good activity against most ICU isolates, particularly *P. aeruginosa*, which was the most frequently isolated pathogen in this study
- Among the antimicrobials tested, ceftolozane-tazobactam was the most active β-lactam against *P. aeruginosa* and, except for the carbapenem, was also the most active β-lactam against *Enterobacteriaceae*
- Ceftolozane-tazobactam may represent a valuable treatment option for serious gram-negative infections in ICU patients

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ USA

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.

Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect.* 2012;18:268–281.

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.