Friday - 50

ASM Microbe 2017
Contact Information:
Dee Shortridge, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Antimicrobial Activity of Ceftolozane-Tazobactam Tested against Contemporary (2014-2016) *Pseudomonas aeruginosa* Isolates with Various Resistant Phenotypes from US Hospitals

D Shortridge, M Castanheira, MA Pfaller, JM Streit, RK Flamm

JMI Laboratories, North Liberty, Iowa, USA

http://merck.creative.studios.s3-website-us-east-1.amazonaws.com/ASM_Microbe_2017/ASM_Microbe_2017-Shortridge_AntimicrobialActivity_FRIDAY-50.pdf

Introduction

- Ceftolozane-tazobactam (C-T) is a combination of a novel antipseudomonal cephalosporin and a well-described β-lactamase inhibitor
 - C-T retains activity against most *Pseudomonas aeruginosa* with derepressed AmpC or up-regulated efflux, including ceftazidime-, cefepime-, or meropenem-resistant strains
- C-T was approved by the United States (US) Food and Drug Administration in 2014 for complicated urinary tract infections, including acute pyelonephritis and complicated intra-abdominal infections (in combination with metronidazole) at a dose of 1.5 g (1 g C + 0.5 g T) q8h
- C-T is currently in a clinical trial for treatment of hospital-acquired bacterial pneumonia, including ventilator-associated bacterial pneumonia (3 g [2 g C + 1 g T] q8h)
- The Program to Assess Ceftolozane-Tazobactam Susceptibility (PACTS) monitors C-T resistance to gram-negative (GN) isolates worldwide
- In this study, we tested recent *P. aeruginosa* isolates collected from US hospitalized patients in all 9 US census divisions
 - Activities of C-T and 7 other agents were compared

Materials and Methods

- A total of 2,656 *P. aeruginosa* (PSA) isolates were collected from patients in 30 US hospitals throughout the 9 US census divisions in 2014-2016 and were tested for susceptibility (S) to C-T by CLSI broth microdilution methodology at JMI Laboratories (North Liberty, Iowa, USA)
 - Only 1 isolate per patient per infection episode was collected
 - The most frequent infection type was pneumonia (56.0%) followed by skin and skin structure infections (18.5%) and bloodstream infections (12.8%)
- Other antibiotics tested included amikacin, cefepime (FEP), ceftazidime (CAZ), colistin (COL), meropenem (MER), levofloxacin, and piperacillin-tazobactam (TZP)
- Resistant phenotypes analyzed were: CAZ+FEP nonsusceptible (CAZ+FEP-NS), MER-NS, TZP-NS, CAZ+FEP+TZP+MER-NS (beta-lactam [BL]-NS), and COL-NS according to CLSI criteria
- Multidrug resistant (MDR) and extensively drug R (XDR) phenotypes were defined according to Magiorakos et al (2012)
- CLSI (2017) and EUCAST (2017) interpretive criteria were used
- The US divisions were: 1) New England, 2) Middle Atlantic, 3) East North Central, 4) West North Central, 5) South Atlantic, 6) East South Central, 7) West South Central, 8) Mountain, and 9) Pacific

Results

- For all PSA isolates, 97.7% were S to C-T (Table 1)
 - C-T was more active than other comparators, except COL (99.4%S)
- The C-T MIC distributions and $MIC_{50/90}$ of all isolates tested against the resistant phenotypes are shown in Table 2
 - A total of 16 (0.6%) isolates were COL-NS (Table 2) and 93.8% were S to C-T
 - 165 (6.2%) isolates were BL-NS of which 72.1% were S to C-T
 - 248 (9.3%) were XDR isolates (79.8%S to C-T)
 - 546 (20.6%) were MDR isolates (89.2%S to C-T)
- The %S to C-T for various resistance phenotypes for each division are shown in Figure 1
- The %S for C-T by division ranged from 99.7% (West North Central) to 93.0% (Pacific) for all PSA
 - The West North Central division and East South Central had the highest %S to C-T for all resistant phenotypes, including isolates resistant to other 4 beta-lactam agents tested (CAZ, FEP, MER, and TZP) where 90.9% and 95.0% were S to C-T
 - The Pacific division had the lowest %S to C-T for the resistant phenotypes

Table 1 Activity of ceftolozane-tazobactam and comparator antimicrobial agents when tested against 2,656 PSA isolates

Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI			EUCAST		
			%S	%I	%R	%S	%I	%R
Ceftolozane-tazobactam	0.5	2	97.7	1	1.3	97.7		2.3
Amikacin	4	8	96.5	1.2	2.2	92.2	4.3	3.5
Cefepime	2	16	87.4	8.6	4	87.4		12.6
Ceftazidime	2	32	85.4	4.5	10.1	85.4		14.6
Colistin	1	2	99.4		0.6	99.4		0.6
Levofloxacin	0.5	>4	76.3	7	16.7	67.1		32.9
Meropenem	0.5	8	81	5.7	13.3	81	12.1	6.9
Piperacillin-tazobactam	4	64	81.9	9.8	8.3	81.9		18.1

Table 2 Antimicrobial activity of ceftolozane-tazobactam tested against the main organisms and organism groups of isolates (µg/mL)

Organism / organism group (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %)													MIC_{50}	MIC_{90}
	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>		
PSA (2,656)	0 0.0	1 <0.1	4 0.2	24 1.1	393 15.9	1446 70.3	502 89.2	144 94.7	81 97.7	27 98.7	6 98.9	5 99.1	23 100.0	0.5	2
TZP-NS PSA (480)				0 0.0	3 0.6	72 15.6	162 49.4	108 71.9	80 88.5	24 93.5	5 94.6	3 95.2	23 100.0	2	8
CAZ- or FEP-NS PSA (474)				0 0.0	2 0.4	43 9.5	165 44.3	122 70.0	81 87.1	27 92.8	6 94.1	5 95.1	23 100.0	2	8
CAZ+FEP-NS (248)					0 0	3 1.2	46 19.8	67 46.8	75 77.0	26 87.5	6 89.9	3 91.1	22 100.0	4	32
MER-NS PSA (504)				0 0.0	10 2.0	138 29.4	173 63.7	77 79.0	51 89.1	22 93.5	6 94.6	5 95.6	22 100.0	1	8
COL-NS PSA (16)					0 0.0	9 56.2	4 81.2	2 93.8	0 93.8	0 93.8	1 100.0			0.5	2
MDR PSA (546)				0 0.0	12 2.2	118 23.8	201 60.6	100 78.9	56 89.2	25 93.8	6 94.9	5 95.8	23 100.0	1	8
XDR PSA (248)				0 0.0	1 0.4	9 4.0	90 40.3	54 62.1	44 79.8	19 87.5	6 89.9	3 91.1	22 100.0	2	32
BL-NS PSA (165)					0 0.0	2 1.2	32 20.6	40 44.8	45 72.1	19 83.6	5 86.7	1 87.3	21 100.0	4	>32

Susceptible breakpoint for C-T (CLSI) is ≤4 µg/mL.

Conclusions

- Against PSA, C-T demonstrated potent activity versus isolates with various resistant phenotypes, including BL-NS, COL-NS, MDR, and XDR isolates
- Divisions varied for C-T %S
- C-T was the most active beta-lactam tested and had activity second only to COL among all antimicrobials tested
- These results demonstrate that C-T is an important treatment for PSA infections in the US

Acknowledgements

This study was sponsored by Merck & Co., Inc.

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.

Magiorakos, A. P., A. Srinivasan, R. B. Carey, Y. Carmeli, M. E. Falagas, C. G. Giske, S. Harbarth, J. F. Hindler, G. Kahlmeter, B. Olsson-Liljequist, D. L. Paterson, L. B. Rice, J. Stelling, M. J. Struelens, A. Vatopoulos, J. T. Weber, and D. L. Monnet. "Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance." (2012) *Clin Microbiol Infect* **18**(3): 268-281.

Figure 1 Percent susceptibility of C-T against various resistant phenotypes (NS to TZP, MER, CAZ+FEP, and BLs) by US division

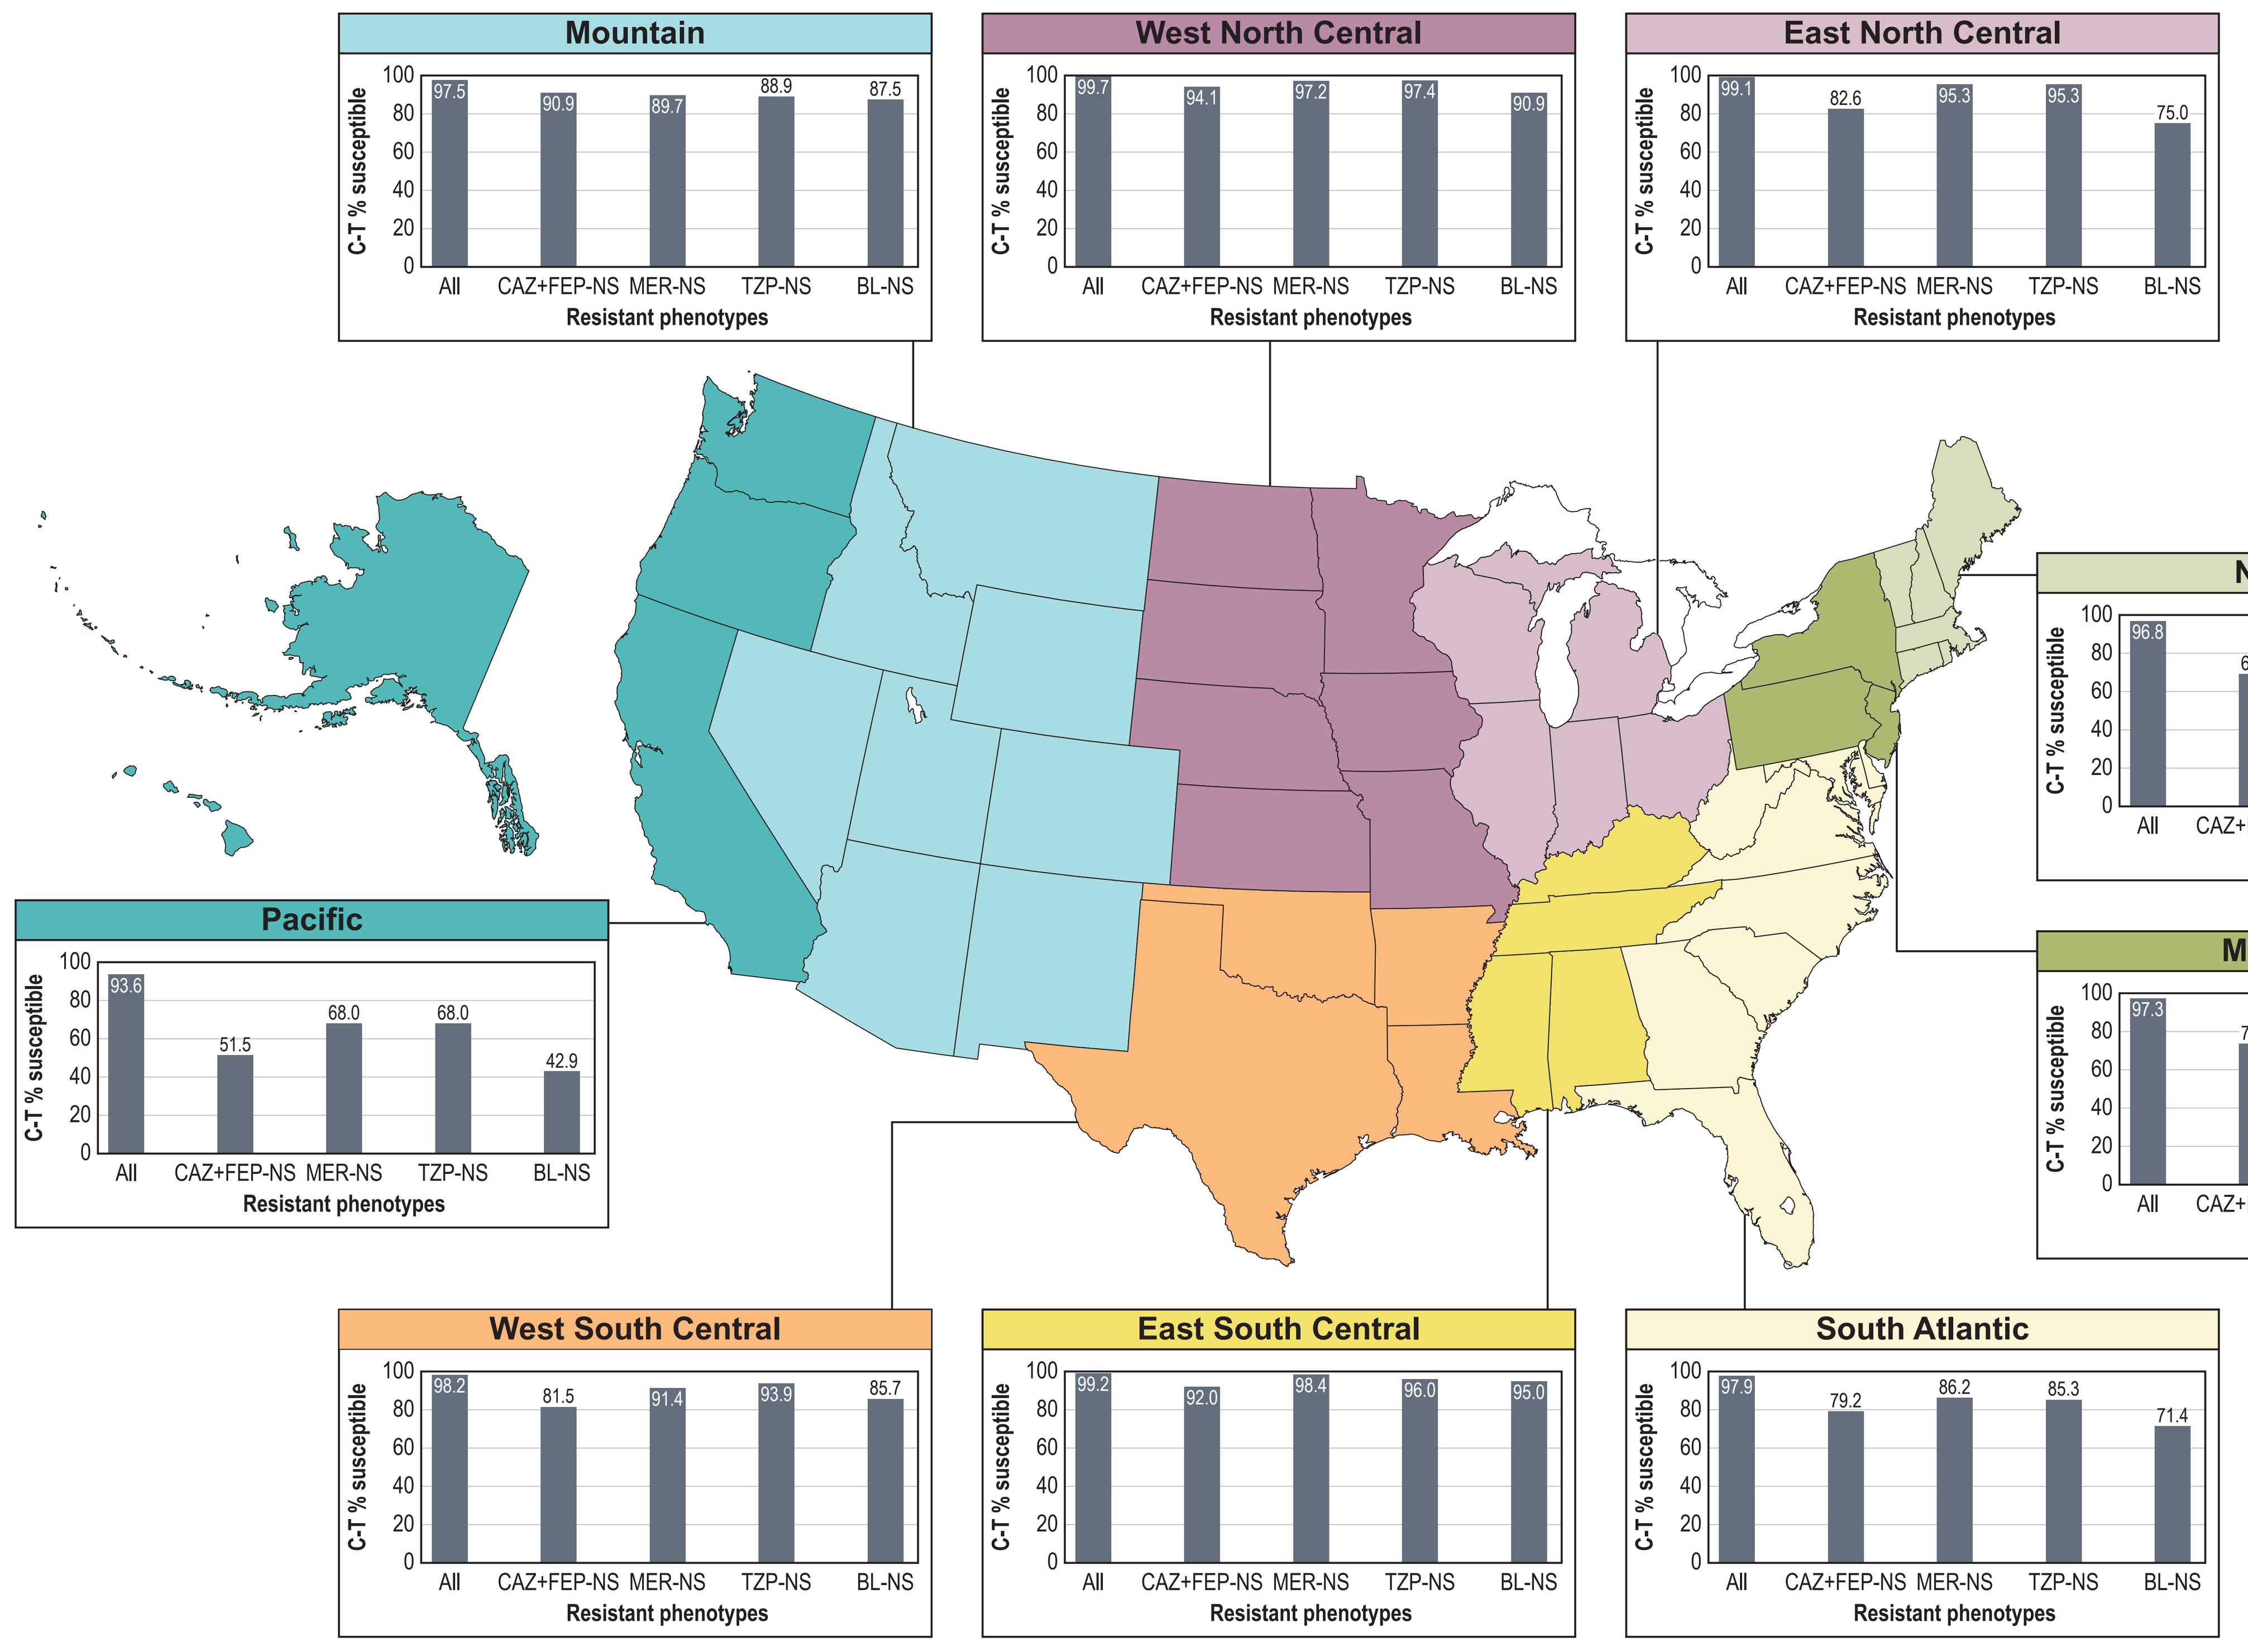
Mountain
97.5 90.9 89.7 88.9 87.5
West North Central
99.7 94.1 97.2 97.4 90.9
East North Central
99.1 82.6 95.3 95.3 75.0
New England
96.8 69.2 81.0 85.7 53.8
Middle Atlantic
97.3 73.7 90.6 88.0 71.4
Pacific
93.6 51.5 68.0 68.0 42.9
West South Central
98.2 81.5 91.4 93.9 85.7
East South Central
99.2 92.0 98.4 96.0 95.0
South Atlantic
97.9 79.2 86.2 85.3 71.4
C-T % susceptible
All CAZ+FEP-NS MER-NS TZP-NS BL-NS
Resistant phenotypes

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.