ASM Microbe 2017

Saturday - 56

Activity of Fusidic Acid against Recent Clinical Isolates of Staphylococci Collected from United States Hospitals in 2016 as Part of the SENTRY Antimicrobial Surveillance Program

D SHORTRIDGE, JM STREIT, PR RHOMBERG, RK FLAMM

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Dee Shortridge, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

Amended Abstract

Introduction: Fusidic acid (FA) is being studied in the United States as a targeted treatment for acute bacterial skin and skin structure infection and other infections caused by gram-positive pathogens. This study used 1,378 staphylococci isolates collected from 38 US hospitals in 2016 as part of the SENTRY Antimicrobial Surveillance Program.

Methods: Tested isolates from skin and skin structure infections and bloodstream infections were composed of 1,064 *Staphylococcus aureus* (SA) and 314 coagulase-negative staphylococci (CoNS). A central monitoring laboratory (JMI Laboratories) used the CLSI broth microdilution method to determine susceptibilities to FA and comparators. Other agents tested included vancomycin (VAN), daptomycin (DAP), clindamycin (CLN), trimethoprim-sulfamethoxazole (T-S) and linezolid (LZD). Interpretive criteria used for FA were EUCAST (2016), and CLSI (2016) criteria were used for the other comparators.

Results: FA and comparator susceptibilities (S) are shown in the table. For SA that included methicillin-resistant SA (MRSA), 99.9% had FA MICs ≤1.0 µg/mL, the EUCAST breakpoint for staphylococci. Against CoNS, 94.9% had FA MICs ≤1.0 µg/mL. For MR-CoNS, 93.2% had MICs ≤1.0 µg/mL compared to CLN and T-S, which had 52.2% S and 51.2%S, respectively, with CLSI breakpoints

Conclusions: Against staphylococci, FA was among the most active oral agents and had activity for SA similar to VAN, DAP, and LZD. These data show that FA is a potent agent against SA in the US and support its continued development to treat staphylococcal infections.

$MIC_{50/90}$ (µg/mL) and % susceptibility of FA and comparators

Organism	No.	FA[a]	VAN[b]	DAP[b]	CLN[b]	T-S[b]	LZD[b]
SA	1,064	0.12/0.12 (99.9)	0.5/1.0 (100.0)	0.25/0.5 (100.0)	≤0.25/>2 (85.5)	≤0.5/≤0.5 (98.0)	1.0/1.0 (100.0)
MRSA	451	0.06/0.12 (100.0)	0.5/1.0 (100.0)	0.25/0.5 (100.0)	≤0.25/>2 (70.7)	≤0.5/≤0.5 (96.7)	0.5/1.0 (100.0)
CoNS	314	0.12/0.12 (94.9)	1.0/2.0 (100.0)	0.25/0.5 (100.0)	≤0.25/>2 (65.6)	≤0.5/>4 (63.7)	0.5/1.0 (96.8)
MR-CoNS	207	0.12/0.12 (93.2)	1.0/2.0 (100.0)	0.25/0.5 (100.0)	≤0.25/>2 (52.2)	2.0/>4 (51.2)	0.5/1.0 (95.2)

[a] EUCAST (2016)
[b] CLSI (2016)

Introduction

- Fusidic acid (CEM-102) is a steroidal antibiotic that possesses a well-characterized potency against gram-positive bacteria such as staphylococci, including methicillin-resistant *Staphylococcus aureus* (MRSA) and coagulase-negative staphylococcal species (CoNS)
- Europe has used fusidic acid (FA) for many years for acute therapy of skin and skin structure infections, and for long-term treatment of bone and joint infections.
- FA has never been used in the US and an oral formulation is currently being studied in clinical trials as a targeted treatment for acute bacterial skin and skin structure infections and other infections caused by staphylococci
 - Resistance rates for staphylococci to other commonly used outpatient therapies such as cephalexin, trimethoprim-sulfamethoxazole (T-S) and clindamycin (CLN) are increasing in the US
- In this study, FA and comparators were tested against 1,378 staphylococci isolates collected from 38 US hospitals in 2016 as part of the SENTRY Antimicrobial Surveillance Program

Materials and Methods

- A total of 1,378 non-duplicated staphylococci isolates were collected prospectively from 38 US medical centers
- Tested isolates from skin and skin structure infections and bloodstream infections were composed of 1,064 *Staphylococcus aureus* (SA) and 314 coagulase-negative staphylococci (CoNS)
- Submitting laboratories identified isolates, and JMI Laboratories confirmed isolate identification by using standard techniques that included biochemical tests, matrix-assisted laser desorption ionization-time of flight mass spectrometry (MALDI-TOF), and DNA sequencing, when necessary
- The CLSI broth microdilution method was used to determine susceptibilities (S) to FA and comparators
 - Other agents tested included: vancomycin (VAN), daptomycin (DAP), clindamycin (CLN), trimethoprim-sulfamethoxazole (T-S) and linezolid (LZD)
 - EUCAST (2017) interpretive criteria were used for FA (S ≤1 and R >1 mg/L), and CLSI (2017) criteria were used for other comparators

Results

- FA and comparators susceptibilities are shown in Table 1
- FA MIC distributions for MRSA, methicillin-susceptible SA (MSSA), MR-CoNS and methicillin-susceptible CoNS (MS-CoNS) are shown in Figure 1
- For SA including MRSA, 99.9% had FA MICs ≤1.0 µg/mL, the EUCAST breakpoint for staphylococci
 - One MSSA isolate was resistant (R) to FA with an MIC of 2.0 µg/mL
 - The highest MRSA MIC was 0.5 µg/mL
- For SA, 85.5% were S to CLN, and 70.7% of MRSA were S to CLN
- 98.0% of SA and 96.7% of MRSA were S to T-S

Table 1 Activities of fusidic acid and comparators tested against staphylococci from SENTRY 2016

Organism/ antimicrobial agent (no. of isolates)	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %I	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %I	EUCAST[a] %R
Staphylococcus aureus (n=1,064)								
Fusidic acid	0.12	0.12				99.9		0.1
Clindamycin	≤0.25	>2	85.5	0	14.5	85.4	0.1	14.5
Vancomycin	0.5	1	100	0	0	100		0
Linezolid	1	1	100		0	100		0
Tetracycline	≤0.5	≤0.5	97.4	0.4	2.3	95.1	1.3	3.6
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	98		2	98	0.3	1.7
Daptomycin	0.25	0.5	100			100		0
MRSA (n=451)								
Fusidic acid	0.06	0.12				100		0
Clindamycin	≤0.25	>2	70.7	0	29.3	70.7	0	29.3
Vancomycin	0.5	1	100	0	0	100		0
Linezolid	0.5	1	100		0	100		0
Tetracycline	≤0.5	≤0.5	97.3	0.2	2.4	94.7	2.4	2.9
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	96.7		3.3	96.7	0.4	2.9
Daptomycin	0.25	0.5	100			100		0
Coagulase-negative staphylococci[a] (n=314)								
Fusidic acid	0.12	0.12				94.9		5.1
Clindamycin	≤0.25	>2	65.6	1.3	33.1	64.6	1	34.4
Vancomycin	1	2	100	0	0	100		0
Linezolid	0.5	1	96.8		3.2	96.8		3.2
Tetracycline	≤0.5	>8	84.7	0	15.3	83.1	1.3	15.6
Trimethoprim-sulfamethoxazole	≤0.5	>4	63.7		36.3	63.7	23.9	12.4
Daptomycin	0.25	0.5	100			100		0
MR-CoNS[b] (n=207)								
Fusidic acid	0.12	0.12				93.2		6.8
Clindamycin	≤0.25	>2	52.2	1.4	46.4	51.7	0.5	47.8
Vancomycin	1	2	100	0	0	100		0
Linezolid	0.5	1	95.2		4.8	95.2		4.8
Tetracycline	≤0.5	>8	79.2	0	20.8	77.8	1.4	20.8
Trimethoprim-sulfamethoxazole	2	>4	51.2		48.8	51.2	32.9	15.9
Daptomycin	0.25	0.5	100			100		0

[a] Organisms include: *Staphylococcus capitis* (19), *S. cohnii* (2), *S. epidermidis* (216), *S. haemolyticus* (13), *S. hominis* (43), *S. lugdunensis* (11), *S. petrasii* (1), *S. pettenkoferi* (5), *S. schleiferi* (2), *S. warneri* (2)
[b] Organisms include: *Staphylococcus capitis* (3), *S. cohnii* (2), *S. epidermidis* (166), *S. haemolyticus* (10), *S. hominis* (21), *S. lugdunensis* (1), *S. pettenkoferi* (4)

- No isolates were R to VAN, DAP, or LZD
- Against CoNS, 94.9% had FA MICs of ≤1.0 µg/mL with 16 R isolates, using EUCAST breakpoints
 - 14 MR-CoNS had MICs in the range of 2 to 8 µg/mL
 - 2 MS-CoNS had MICs of 8 µg/mL
 - 65.6% were S to CLN and 63.7% were S to T-S
- For MR-CoNS, 93.2% had FA MICs of ≤1.0 µg/mL compared to CLN and T-S, which had 52.2%S and 51.2%S, with CLSI breakpoints, respectively

Figure 1 MIC distribution of fusidic acid for MRSA, MSSA, MR-CoNS, and MS-CoNS

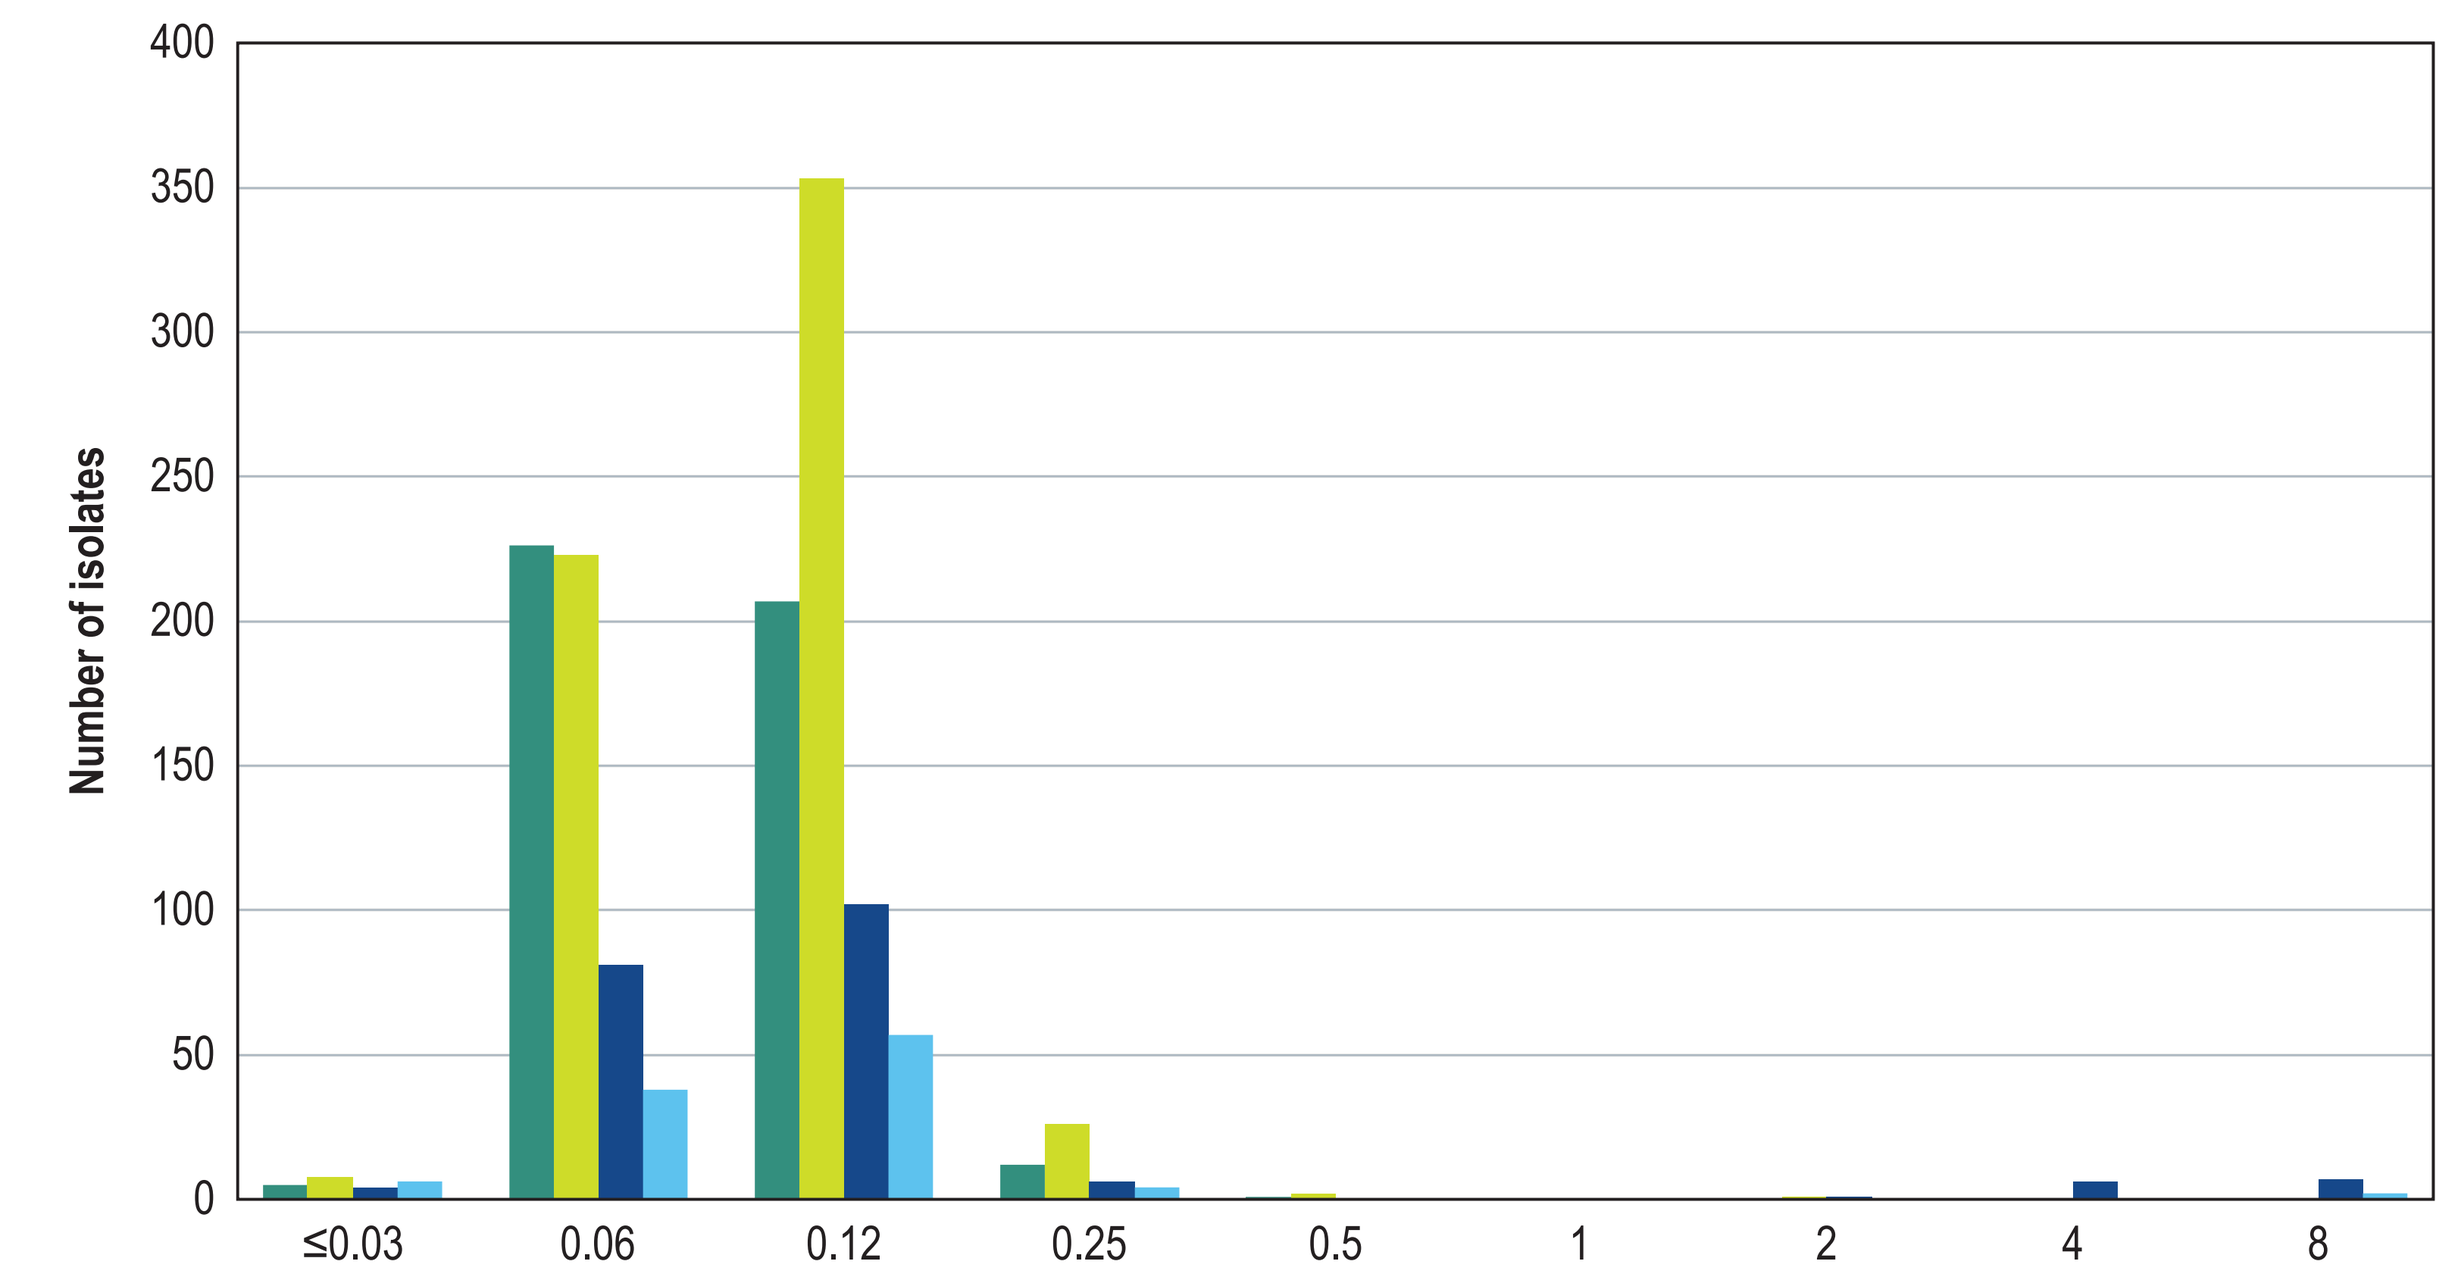
Number of isolates
400
350
300
250
200
150
100
50
0

	≤0.03	0.06	0.12	0.25	0.5	1	2	4	8
MRSA	5	226	207	12	1	0	0	0	0
MSSA	8	223	353	26	2	0	1	0	0
MR-CoNS	4	81	102	6	0	0	1	6	7
MS-CoNS	6	38	57	4	0	0	0	0	2

MRSA, methicillin-resistant S. aureus; MSSA, methicillin-susceptible S. aureus; MR-CoNS, methicillin-resistant coagulase-negative staphylococci; MS-CoNS, methicillin-susceptible coagulase-negative staphylococci

Conclusions

- Against staphylococci, FA was among the most potent oral agents tested
 - FA had activity for SA similar to VAN, DAP, and LZD
- Only 1.2% of staphylococci had FA MICs >1 µg/mL, suggesting that R to FA (based on EUCAST breakpoints) in the US is very low
 - 1 SA and 16 CoNS had FA MICs >1 µg/mL (Figure 1)
- These data show that FA is a potent agent against SA in the US and support continued development to treat staphylococcal infections

Acknowledgements

This study was supported by Cempra, Inc.

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.

https://www.jmilabs.com/data/posters/ASMMicrobe17-fusidic-acid.pdf