ASM Microbe 2017
Friday - 129

Serotype Distributions and Analysis of Susceptibility Profiles of *Streptococcus pneumoniae* Causing Infections in Adult Patients in the United States (2009–2015)

RE MENDES[1], HL SINGS[2], SJR ARENDS[1], TB DOYLE[1], LN WOOSLEY[1], RK FLAMM[1], RE ISTURIZ[2]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Pfizer Inc., Collegeville, Pennsylvania, USA

Contact Information:
Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

Abstract

Background: A pneumococcal polysaccharide vaccine (PPSV23) has been recommended for adults ≥65 years old since 1997 with vaccination rates ~65% in the past decade. A pneumococcal conjugate vaccine (PCV13) has been recommended for adults ≥65 years old since 2014. This study evaluated the serotype distribution and susceptibility profiles of pneumococci causing infections in adult patients (≥18 years old) in the United States (US) between 2009 and 2015.

Methods: A total of 6,148 *S. pneumoniae* isolates originated from patients seen/hospitalized in 91 US centers. Isolates were recovered primarily (81.3%) from respiratory tract specimens. Serotyping was performed by *cpsB* sequencing, multiplex PCR, and/or Quellung reaction. Serotype distribution was also analyzed by patient age group (18-49, 50-64, and ≥65 years old). Susceptibility testing applied CLSI methods, and interpretation of MICs applied CLSI criteria.

Results: Overall, PCV13 serotypes comprised 38.6% of serotypes in 2009 and declined to 23.5% in 2015. A decline in PCV13 serotypes was observed in all 3 age groups (18-49, 50-64, and ≥65 years old), with the decline most pronounced in those ≥65 years old (19.9% difference vs 11.7% and 13.0%, respectively). Serotype 19A decreased from 17.5% to 6.6% with a similar decreasing trend in all age groups (8.6% to 12.7%). Decreasing trends were also noted for serotype 7F (from 4.3% to 1.2%), 6A (from 1.6% to 0.3%), and 6B (from 0.6% to 0.1%). Prevalence of serotype 3 and 19F remained unchanged over the study period. The proportion of non-vaccine serotypes increased during the study period overall and in each age group (from 36.8% to 48.9%). Among the non-vaccine serotypes, 35B (11.6%) was the most common serotype in 2015. Other prevalent non-vaccine serotypes in 2015 were 23B (6.7%), 23A (6.6%), and 15A/15F (4.7%). Serotypes unique to PPSV23 showed a slight increasing trend (from 22.9% to 26.5%). Susceptibility rates for penicillin (from 85.8% to 96.1%), ceftriaxone (from 88.3% to 97.9%), amoxicillin-clavulanate (from 84.8% to 94.3%), and clindamycin (from 80.3% to 87.0%) increased over time.

Conclusion: PCV13 rates declined significantly over the study period. In contrast, the susceptibility rates for several antimicrobial agents increased. Non-vaccine and PPSV23 serotypes increased in prevalence in the older US population.

Introduction

- *Streptococcus pneumoniae* remains an important pathogen responsible for community-acquired bacterial pneumonia (CABP), bacteremia, meningitis, and otitis media and continues to be a major cause of morbidity and mortality worldwide
- After introducing the 7-valent and 13-valent pneumococcal conjugate vaccines (PCV7, PCV13) in National Immunization Programs (NIPs) globally, invasive pneumococcal disease (IPD) and non-IPD incidences have declined among children <5 years of age
- Furthermore, the universal introduction of PCV13 for infant use has been associated with significant reductions in nasopharyngeal carriage of PCV13 serotypes and declines in pneumococcal infections (herd effects) in the older, non-vaccinated, population
- Once NIPs are implemented, monitoring the impact and magnitude of its effect on IPD and non-IPD among children and adults is important, as well as monitorial antimicrobial resistance profiles
- This study was conducted to determine the prevalence and serotype distribution of *S. pneumoniae* clinical isolates recovered predominantly from non-sterile sites (mostly sputum or lower respiratory tract secretions) and associated with disease among adult patients (≥18 years of age) in the United States from 2009 through 2015
 - Antimicrobial susceptibility profiles between periods were also analyzed

Materials and Methods

Clinical isolates

- 6,148 *S. pneumoniae* clinical isolates recovered from adult patients (≥18 years old) seen/hospitalized in 91 US centers were included in this study
- Isolates were collected primarily (78.4%; 4,823/6,148) from lower respiratory tract specimens and submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA), as part of the SENTRY Antimicrobial Surveillance Program
- Participating microbiology laboratories identified bacteria
- The central monitoring laboratory confirmed bacterial identification by colony morphology and biochemical algorithms
- Isolates were subjected to a PCR assay for further identification when bacterial identification was questionable after using phenotypic methods or an untypeable serotyping result was obtained by the applied methodology

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility by broth microdilution methods according to the recommendations of the Clinical and Laboratory Standards Institute (CLSI)
- MIC values were validated by concurrently testing quality control (QC) strain *S. pneumoniae* American Type Culture Collection (ATCC) 49619
- Inoculum density was monitored by colony counts to ensure an adequate number of cells for each testing event
- MIC interpretations were based on the CLSI breakpoint criteria

Pneumococcal serotyping

- Isolates were subjected to PCR assays to amplify the *cpsB* gene
- Amplicons were sequenced on both strands, and the nucleotide sequences were analyzed using the Lasergene software package (DNASTAR, Madison, Wisconsin, USA)
- Sequences were compared to others available via PubMed (http://www.ncbi.nlm.nih.gov/blast/)
- Due to sequence homology among certain serotypes, those showing nucleotide sequence similarity greater than 99% were grouped (eg, 9V/9A, 7F/7A, 11A/11D, 15A/15F, 22F/22A, 15B/15C)
- All isolates determined to be serogroup 6 by sequencing analysis were subjected to multiplex PCR assays for confirmation and discrimination between 6A/6B and 6C/6D
- Isolates determined to be serogroup 6A/6B and 7F/7A were serotyped by the capsular swelling method using commercially available antisera according to manufacturer's instructions (Statens Serum Institut, Copenhagen, Denmark)

Figure 1 Relative percentages of *S. pneumoniae* vaccine serotypes observed in the US adult population (≥18 years old) during 2009–2015

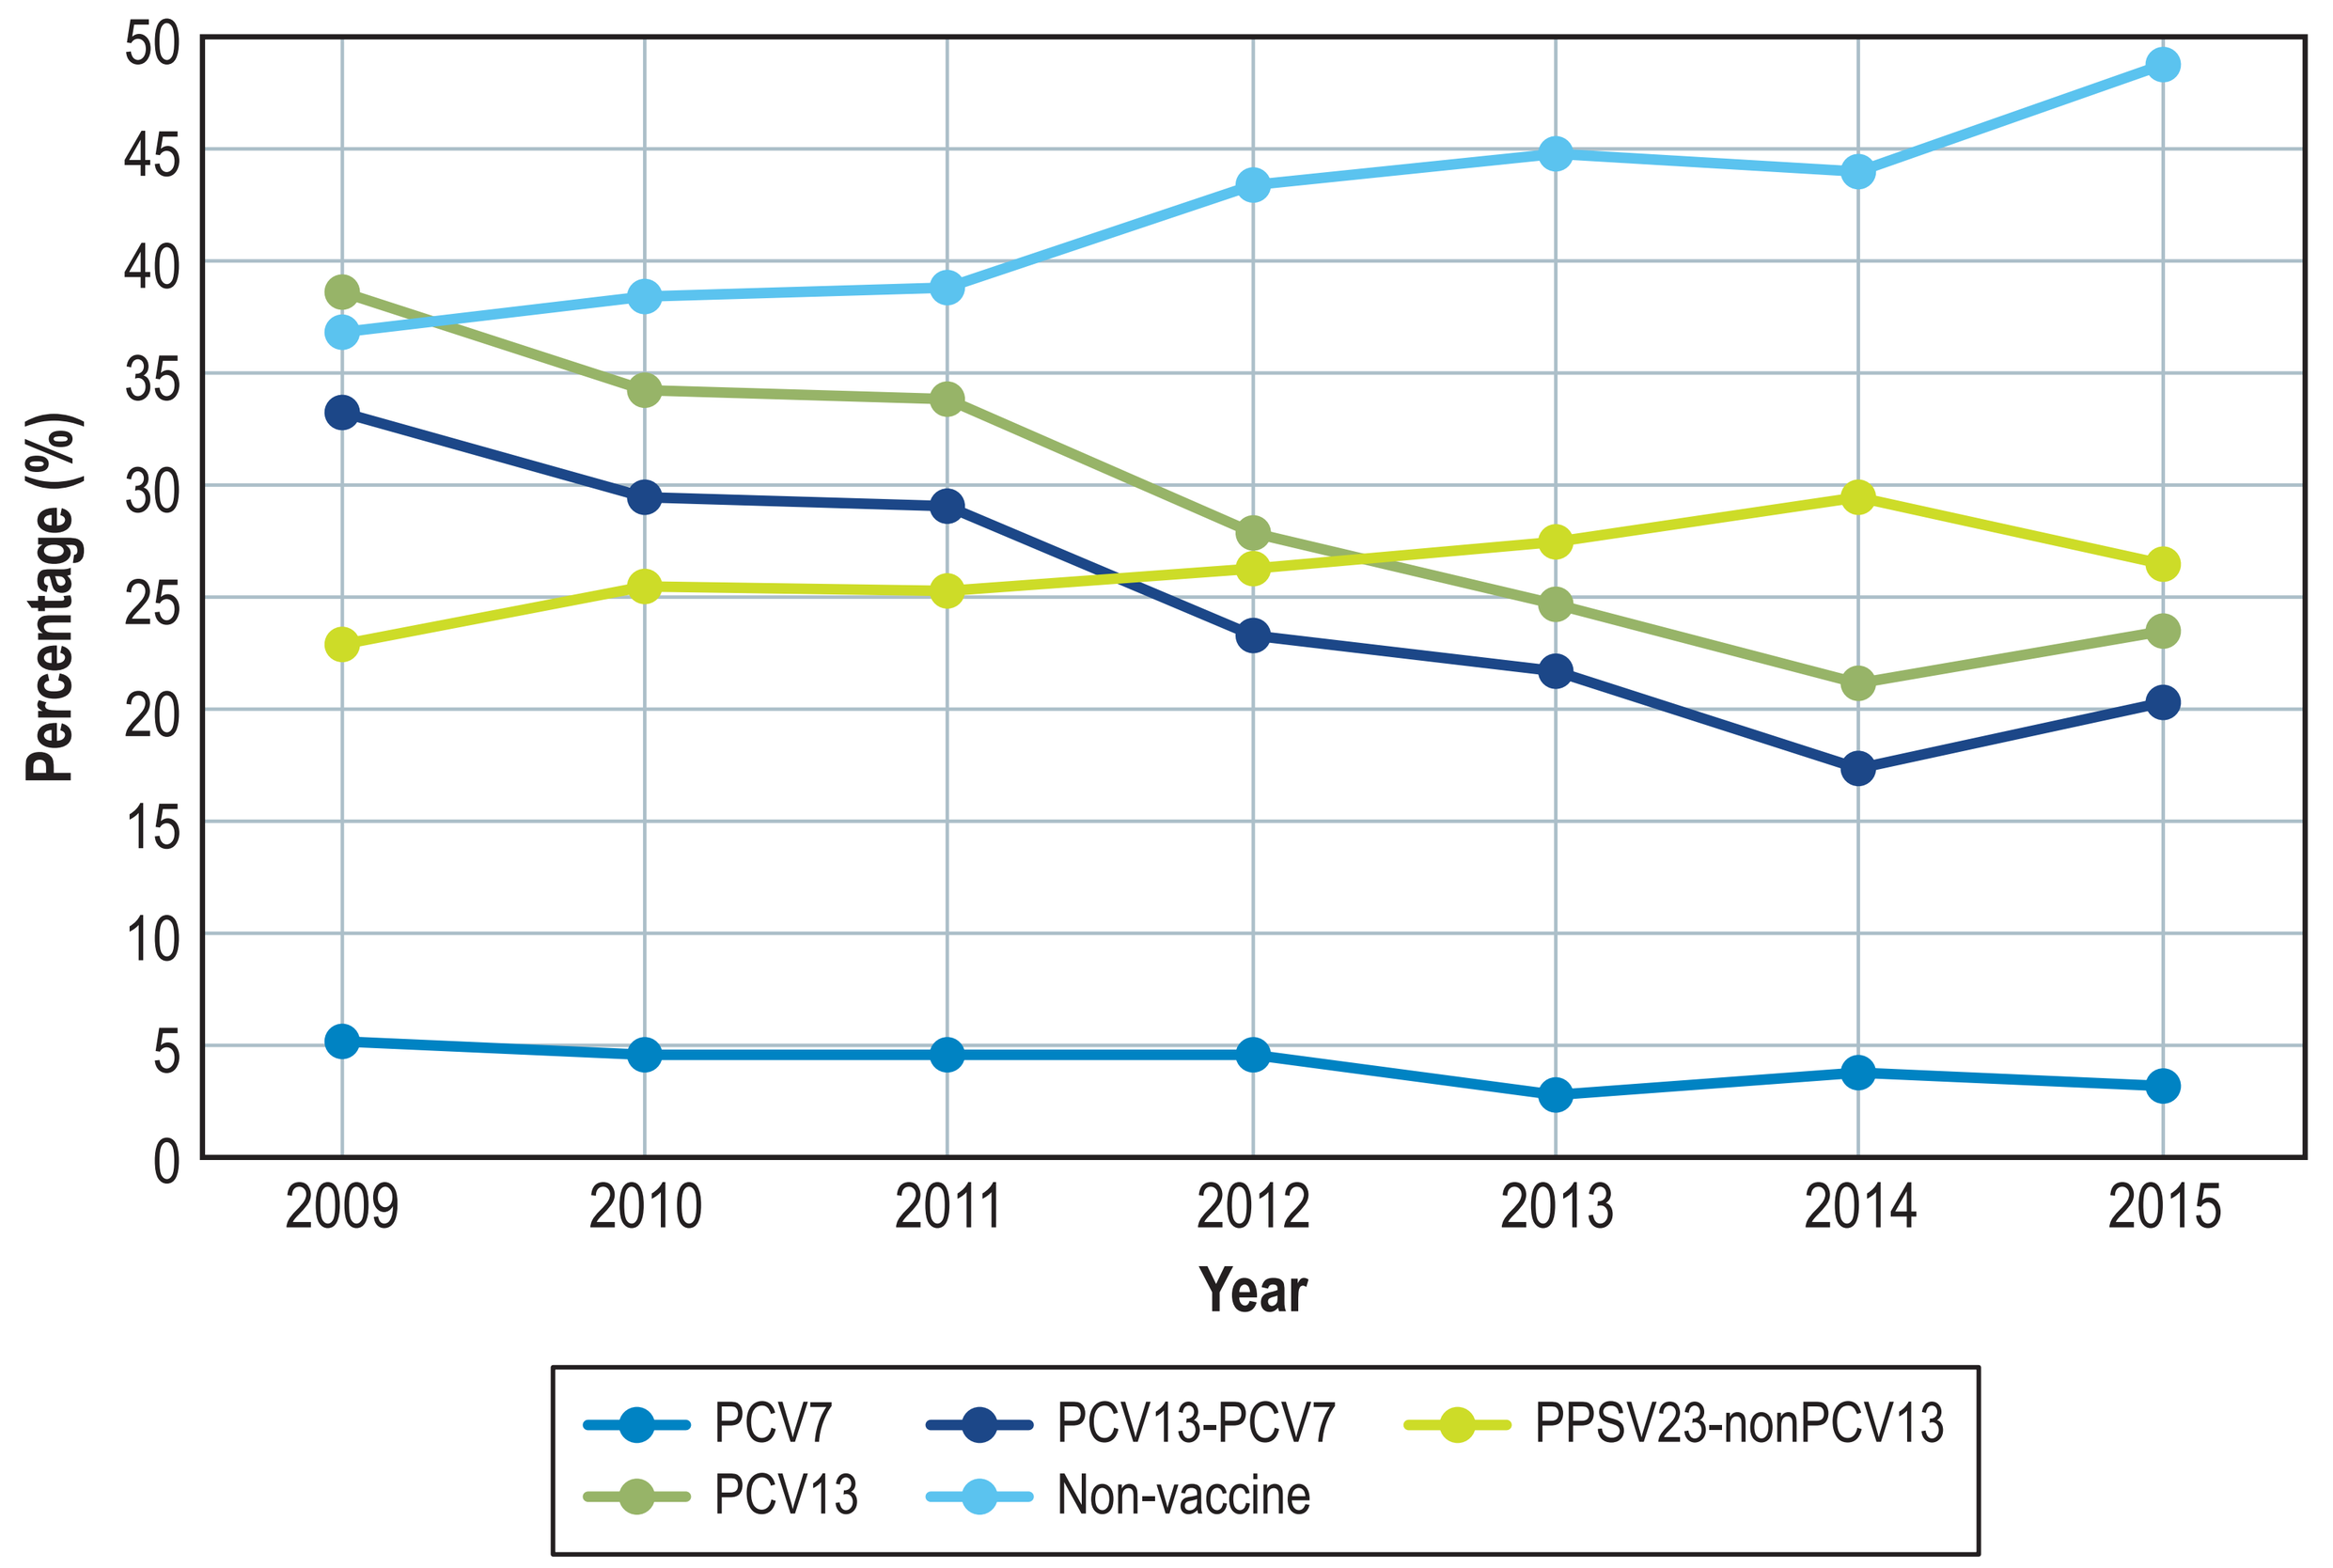

Percentage (%)
50
45
40
35
30
25
20
15
10
5
0
2009
2010
2011
2012
2013
2014
2015
Year
PCV7
PCV13
PCV13-PCV7
Non-vaccine
PPSV23-nonPCV13

Results

- Overall, PCV7, PCV13-nonPCV7, and PCV13 serotypes comprised 4.1%, 24.9%, and 29.0% of all isolates, respectively, with serotypes 19F (2.7%) predominating among PCV7-type isolates and serotypes 19A (12.6%) and 3 (9.0%) prevailing among PCV13-type isolates (Table 1)
- Rates for PCV7 serotypes remained stable (4.6%–5.2%) during 2009–2012, declined slightly in 2013–2015 (2.8%–3.8%; Table 1 and Figure 1)
- PCV13 (from 38.6% to 23.5%) and PCV13-nonPCV7 (from 33.3% to 20.3%) showed a consistent decreasing trend over the study period (Table 1 and Figure 1)
 - Within PCV13, serotypes 19A, 7F, and 6A showed prevalence rates in 2013 lower than those observed in 2009
 - Only serotype 3 showed a slight increase from 9.9% (2009) to 12.3% (2015; Table 1)
- The decline in PCV13 STs was observed in all 3 age groups (18-49, 50-64, and ≥65 years), with the decline most pronounced in those ≥65 years old (19.9% difference versus 11.7% and 13.0%, respectively; Table 2)
- Overall, a total of 26.4% and 42.2% of isolates consisted of PPSV23-nonPCV13 and nonvaccine serotypes, respectively (Table 1)
 - A consistent upward trend (from 22.9% in 2009 to 29.6% in 2014, with a decrease to 26.5% in 2015) was noted for PPSV23-nonPCV13, which was more evident within the 50-64 age group (Tables 1 and 2; Figure 1)

Table 1 Distribution of serogroups/types of *S. pneumoniae* causing infections in the US adult population during 2009–2015

Serogroup/type	Number (%) of *S. pneumoniae* serogroups/types							
	2009	2010	2011	2012	2013	2014	2015	All years
PCV7	35 (5.2)	39 (4.7)	54 (4.7)	39 (4.6)	29 (2.8)	34 (3.8)	24 (3.2)	254 (4.1)
19F	19 (2.8)	24 (2.9)	34 (3.0)	27 (3.2)	17 (1.7)	26 (2.9)	22 (2.9)	169 (2.7)
9V/9A	5 (0.7)	0 (0.0)	7 (0.6)	2 (0.2)	4 (0.4)	1 (0.1)	0 (0.0)	19 (0.3)
6B	4 (0.6)	2 (0.2)	4 (0.3)	2 (0.2)	2 (0.2)	2 (0.2)	1 (0.1)	17 (0.3)
23F	3 (0.4)	4 (0.5)	3 (0.3)	3 (0.4)	2 (0.2)	1 (0.1)	0 (0.0)	16 (0.3)
4	0 (0.0)	3 (0.4)	5 (0.4)	1 (0.1)	4 (0.4)	2 (0.2)	1 (0.1)	16 (0.3)
18(18A/18B/18C/18F)	3 (0.4)	2 (0.2)	0 (0.0)	2 (0.2)	0 (0.0)	2 (0.2)	0 (0.0)	9 (0.1)
14	1 (0.1)	4 (0.5)	1 (0.1)	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)	8 (0.1)
PCV13	258 (38.6)	282 (34.3)	387 (33.9)	238 (28.0)	252 (24.7)	189 (21.2)	176 (23.5)	1,782 (29.0)
PCV13 -PCV7	223 (33.3)	243 (29.5)	333 (29.1)	199 (23.4)	223 (21.8)	155 (17.4)	152 (20.3)	1,528 (24.9)
19A	117 (17.5)	123 (14.9)	196 (17.1)	108 (12.7)	116 (11.4)	68 (7.6)	49 (6.6)	777 (12.6)
3	66 (9.9)	70 (8.5)	100 (8.7)	69 (8.1)	84 (8.2)	73 (8.2)	92 (12.3)	554 (9.0)
7F	29 (4.3)	45 (5.5)	32 (2.8)	14 (1.6)	18 (1.8)	10 (1.1)	9 (1.2)	157 (2.6)
6A	11 (1.6)	5 (0.6)	4 (0.3)	8 (0.9)	4 (0.4)	2 (0.2)	2 (0.3)	36 (0.6)
1	0 (0.0)	0 (0.0)	1 (0.1)	0 (0.0)	1 (0.1)	2 (0.2)	0 (0.0)	4 (0.1)
5	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
PPSV23-nonPCV13	153 (22.9)	210 (25.5)	289 (25.3)	224 (26.3)	282 (27.6)	264 (29.6)	198 (26.5)	1,620 (26.4)
11A/11D	38 (5.7)	47 (5.7)	59 (5.2)	55 (6.5)	63 (6.2)	73 (8.2)	47 (6.3)	382 (6.2)
22A/22F	33 (4.9)	48 (5.8)	67 (5.9)	41 (4.8)	61 (6.0)	48 (5.4)	40 (5.3)	338 (5.5)
15B/15C	18 (2.7)	39 (4.7)	53 (4.6)	54 (6.3)	45 (4.4)	39 (4.4)	39 (5.2)	287 (4.7)
9N/9L	15 (2.2)	23 (2.8)	33 (2.9)	22 (2.6)	33 (3.2)	27 (3.0)	32 (4.3)	185 (3.0)
10A	15 (2.2)	8 (1.0)	22 (1.9)	11 (1.3)	24 (2.4)	17 (1.9)	17 (2.3)	114 (1.9)
33F/33A	10 (1.5)	10 (1.2)	20 (1.7)	13 (1.5)	28 (2.7)	27 (3.0)	1 (0.1)	109 (1.8)
17F	16 (2.4)	13 (1.6)	18 (1.6)	11 (1.3)	15 (1.5)	13 (1.5)	8 (1.1)	94 (1.5)
8	4 (0.6)	6 (0.7)	8 (0.7)	6 (0.7)	9 (0.9)	10 (1.1)	9 (1.2)	52 (0.8)
20	2 (0.3)	9 (1.1)	6 (0.5)	10 (1.2)	4 (0.4)	10 (1.1)	4 (0.5)	45 (0.7)
12F/12A/44/46	2 (0.3)	7 (0.9)	3 (0.3)	1 (0.1)	0 (0.0)	0 (0.0)	1 (0.1)	14 (0.2)
Non-vaccine	246 (36.8)	317 (38.5)	444 (38.8)	370 (43.5)	456 (44.8)	394 (44.1)	366 (48.9)	2,593 (42.2)
35B	26 (3.9)	54 (6.6)	82 (7.2)	79 (9.3)	73 (7.1)	87 (9.7)	87 (11.6)	488 (7.9)
23A	38 (5.7)	37 (4.5)	64 (5.6)	50 (5.9)	86 (8.4)	64 (7.2)	49 (6.6)	388 (6.3)
6C/6D	51 (7.6)	53 (6.4)	77 (6.7)	63 (7.4)	52 (5.1)	46 (5.2)	24 (3.2)	366 (6.0)
15A/15F	35 (5.2)	45 (5.5)	50 (4.4)	36 (4.2)	55 (5.4)	38 (4.3)	35 (4.7)	294 (4.8)
23B	25 (3.7)	28 (3.4)	43 (3.8)	46 (5.4)	42 (4.1)	52 (5.8)	50 (6.7)	286 (4.7)
16F	18 (2.7)	28 (3.4)	22 (1.9)	16 (1.9)	31 (3.0)	29 (3.2)	16 (2.1)	160 (2.6)
31	17 (2.5)	19 (2.3)	31 (2.7)	22 (2.6)	26 (2.5)	18 (2.0)	13 (1.7)	146 (2.4)
35F/47F	10 (1.5)	11 (1.3)	16 (1.4)	9 (1.1)	17 (1.7)	16 (1.8)	0 (0.0)	79 (1.3)
7C/7B/40	9 (1.3)	12 (1.5)	12 (1.0)	8 (0.9)	11 (1.1)	7 (0.8)	15 (2.0)	74 (1.2)
34	7 (1.0)	17 (2.1)	17 (1.5)	14 (1.6)	14 (1.4)	0 (0.0)	4 (0.5)	73 (1.2)
21	3 (0.4)	2 (0.2)	9 (0.8)	9 (1.1)	23 (2.3)	11 (1.2)	11 (1.5)	68 (1.1)
38/25F/25A	1 (0.1)	3 (0.4)	13 (1.1)	6 (0.7)	10 (1.0)	3 (0.3)	5 (0.7)	41 (0.7)
13	3 (0.4)	4 (0.5)	3 (0.3)	8 (0.9)	2 (0.2)	3 (0.3)	1 (0.1)	24 (0.4)
Other	3 (0.4)	4 (0.5)	5 (0.4)	4 (0.5)	14 (1.4)	20 (2.2)	56 (7.5)	106 (1.7)
Untypeable	12 (1.8)	14 (1.7)	23 (2.0)	19 (2.2)	31 (3.0)	46 (5.2)	8 (1.1)	153 (2.5)

- An increase from 36.8% in 2009 to 48.9% in 2015 for non-vaccine serotypes was observed over the study period (Table 1 and Figure 1), and this trend was present among patients from all age groups (Table 2)
 - Among non-vaccine serotypes, 35B (11.6%) was the most common in 2015, followed by 23B (6.7%), 23A (6.6%), 15A/15F (4.7%), and 6C/6D (3.2%)
 - Serotype 6C/6D showed a decreased prevalence from 7.6% in 2009 to 3.2% in 2015, whereas 35B rates increased during the study period from 3.9% in 2009 to 11.6% in 2015 (Table 1)
- Overall, susceptibility rates for penicillin (MIC, ≤2 µg/mL; +7.4%) and amoxicillin-clavulante (+7.2%) increased and erythromycin (-2.5%) susceptibility decreased slightly between the 2009–2011 and 2013–2015 periods
 - Other antimicrobial agents showed smaller increases in susceptibility or similar overall susceptibility rates between study periods (Table 3)

Table 2 Vaccine type distribution of noninvasive *S. pneumoniae* according to subject age group during 2009–2015 in the US

Age group	Number (%) of vaccine-type by age group and year			
Year	PCV7	PCV13	PPSV23-nonPCV13	Non-vaccine
18–49				
2009	9 (4.2)	77 (36.0)	61 (28.5)	71 (33.2)
2010	9 (3.3)	91 (33.1)	72 (26.2)	109 (39.6)
2011	16 (4.2)	140 (36.9)	88 (23.2)	142 (37.5)
2012	14 (5.1)	78 (28.3)	70 (25.4)	121 (43.8)
2013	8 (2.5)	83 (25.5)	93 (28.6)	141 (43.4)
2014	12 (5.0)	49 (20.3)	65 (27.0)	111 (46.1)
2015	7 (3.7)	46 (24.3)	44 (23.3)	97 (51.3)
50–64				
2009	14 (6.2)	82 (36.4)	51 (22.7)	86 (38.2)
2010	11 (4.0)	98 (35.8)	64 (23.4)	106 (38.7)
2011	20 (5.3)	119 (31.6)	101 (26.9)	149 (39.6)
2012	8 (2.7)	88 (30.0)	81 (27.6)	122 (41.6)
2013	13 (3.7)	91 (25.7)	96 (27.1)	161 (45.5)
2014	11 (3.5)	66 (21.2)	90 (28.9)	145 (46.6)
2015	15 (5.2)	68 (23.4)	86 (29.6)	133 (45.7)
≥65				
2009	12 (5.2)	99 (43.0)	41 (17.8)	89 (38.7)
2010	19 (6.9)	93 (33.9)	74 (27.0)	102 (37.2)
2011	18 (4.6)	128 (33.0)	100 (25.8)	153 (39.4)
2012	17 (6.0)	72 (25.5)	73 (25.9)	127 (45.0)
2013	8 (2.3)	78 (22.8)	93 (27.2)	154 (45.0)
2014	11 (3.2)	74 (21.7)	109 (32.0)	138 (40.5)
2015	2 (0.7)	62 (23.1)	68 (25.4)	136 (50.7)

Table 3 Antimicrobial activity of antimicrobial agents against 5,290 *S. pneumoniae* isolates collected in the US during 2009–2011 and 2013–2015

Antimicrobial agent[a]	MIC (µg/mL):			%Susceptible/%Intermediate/%Resistant[b]:		
period	MIC_{50}	MIC_{90}	Range			
Penicillin[c]						
2009-2011	≤0.06	4	≤0.06 to >4	86.7	11.6	1.7
2013-2015	≤0.06	2	≤0.06 to >4	94.1	5.6	0.3
A-C[d]						
2009-2011	≤1	>4	≤1 to >4	83.6	3.8	12.6
2013-2015	≤1	2	≤1 to >4	90.8	3.3	5.9
Ceftriaxone[e]						
2009-2011	≤0.25	2	≤0.25 to >2	89.9	8.5	1.6
2013-2015	≤0.25	1	≤0.25 to >2	94.6	4.8	0.5
Erythromycin						
2009-2011	≤0.25	>2	≤0.25 to >2	59.6	0.5	39.9
2013-2015	≤0.25	>2	≤0.25 to >2	56.9	0.7	42.4
Clindamycin						
2009-2011	≤0.25	>1	≤0.25 to >1	79.5	0.5	20.0
2013-2015	≤0.25	>1	≤0.25 to >1	84.1	0.8	15.2
Levofloxacin						
2009-2011	1	1	≤0.5 to >4	98.8	<0.1	1.2
2013-2015	1	1	≤0.5 to >4	98.6	0.2	1.2
Tetracycline						
2009-2011	≤2	>8	≤2 to >4	76.4	0.2	23.3
2013-2015	≤0.5	>4	≤2 to >4	79.0	0.4	20.7
TMP-SMX						
2009-2011	≤0.5	>2	≤0.5 to >2	67.9	7.7	24.4
2013-2015	≤0.5	4	≤0.5 to >2	72.3	11.6	16.1
Linezolid						
2009-2011	1	1	≤0.12 to 4	>99.9	—	—
2013-2015	1	1	≤0.12 to 2	100.0	—	—
Vancomycin						
2009-2011	≤1	≤1	≤1 to ≤1	100.0	—	—
2013-2015	≤1	≤1	≤1 to ≤1	100.0	—	—

[a] A-C, amoxicillin-clavulanate; TMP-SMX, trimethoprim-sulfamethoxazole
[b] Breakpoint criteria were those from CLSI (2016); "—" breakpoint not available
[c] Parenteral non-meningitis (≤2 µg/mL for susceptible; 4 µg/mL for intermediate; and ≥8 µg/mL for resistant)
[d] Non-meningitis (≤2/1 µg/mL for susceptible; 4/2 µg/mL for intermediate; and ≥8/4 µg/mL for resistant)
[e] Non-meningitis (≤1 µg/mL for susceptible; 2 µg/mL for intermediate; and ≥4 µg/mL for resistant)

Conclusions

- PCV7 serotypes causing lower respiratory tract infections in adults have shown persistent rates (4.6–5.2%) during the first 4 years of the study, and rates finally declined further in 2013–2015 (2.8%–3.8%)
- The yearly prevalence of PCV13 serotypes showed signs of decline, which seemed to be led by decreasing rates of serotype 19A and 7F, whereas serotype 3 rates have not declined over the study period
- Clear trends for increased prevalence over time were documented for PPSV23-nonPCV13 and especially for non-vaccine serotypes across all age groups
- Continued surveillance remains important for monitoring the impact of immunization programs in the US adult population

Acknowledgements

The research and publication process was supported by Pfizer Inc., Collegeville, Pennsylvania.

References

Ben-Shimol S, Givon-Lavi N, Leibovitz E, Raiz S, Greenberg D, Dagan R (2016). Impact of widespread introduction of pneumococcal conjugate vaccines on pneumococcal and nonpneumococcal otitis media. *Clin Infect Dis* 63: 611–618.

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: Tenth edition*. Wayne, PA, USA.

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

Desai AP, Sharma D, Crispell EK, Baughman W, Thomas S, Tunali A, Sherwood L, Zmitrovich A, Jerris R, Satola SW, Beall B, Moore MR, Jain S, Farley MM (2015). Decline in pneumococcal nasopharyngeal carriage of vaccine serotypes after the introduction of the 13-valent pneumococcal conjugate vaccine in children in Atlanta, Georgia. *Pediatr Infect Dis J* 34: 1168–1174.

Griffin MR, Mitchel E, Moore MR, Whitney CG, Grijalva CG; Centers for Disease Control and Prevention (CDC) (2014). Declines in pneumonia hospitalizations of children aged <2 years associated with the use of pneumococcal conjugate vaccines—Tennessee, 1998–2012. *Morb Mortal Wkly Rep* 63: 995-998.

Mendes RE, Hollingsworth RC, Costello A, Jones RN, Isturiz RE, Hewlett D Jr, Farrell DJ (2015). Noninvasive *Streptococcus pneumoniae* serotypes recovered from hospitalized adult patients in the United States in 2009 to 2012. *Antimicrob Agents Chemother* 59: 5595–5601.

Musher DM, Sampath R, Rodriguez-Barradas MC (2011). The potential role for protein-conjugate pneumococcal vaccine in adults: what is the supporting evidence? *Clin Infect Dis* 52: 633–640.

Pilishvili T, Bennett NM (2015). Pneumococcal disease prevention among adults: Strategies for the use of pneumococcal vaccines. *Vaccine* 33 Suppl 4: D60–D65.

Smith KJ, Wateska AR, Nowalk MP, Raymund M, Nuorti JP, Zimmerman RK (2012). Cost-effectiveness of adult vaccination strategies using pneumococcal conjugate vaccine compared with pneumococcal polysaccharide vaccine. *JAMA* 307: 804–812.

Weycker D, Sato R, Strutton D, Edelsberg J, Atwood M, Jackson LA (2012). Public health and economic impact of 13-valent pneumococcal conjugate vaccine in US adults aged >/=50 years. *Vaccine* 30: 5437–5444.

https://www.jmilabs.com/data/posters/ASMMicrobe17-serotype.pdf