Poster #P1808

Activity of Telavancin against a Global Collection of Multidrug-Resistant Gram-Positive Isolates and Ceftaroline-Nonsusceptible *Staphylococcus aureus* (2015-2017)

LR Duncan, HS Sader, MD Huband, RK Flamm, RE Mendes

JMI Laboratories, North Liberty, Iowa, USA

Leonard R. Duncan, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: leonard-duncan@jmilabs.com

INTRODUCTION AND PURPOSE

- Telavancin is a once-daily parenteral bactericidal lipoglycopeptide antimicrobial agent[1]
- Telavancin exhibits a dual mechanism of action that involves inhibition of peptidoglycan synthesis and disruption of bacterial cell membrane function[2]
- Telavancin was approved in Europe for the treatment of adult patients with nosocomial pneumonia (including VABP), known or suspected to be caused by methicillin-resistant *S. aureus* (MRSA) in situations where other alternatives are not suitable[3]
- Telavancin is approved in the United States for the treatment of adult patients with complicated skin and skin-structure infections (cSSSIs) and hospital-acquired and ventilator-associated bacterial pneumonia (HABP/VABP) caused by susceptible isolates of *Staphylococcus aureus* when alternative treatments are not suitable[1]
- Comparable efficacy has been demonstrated between telavancin and vancomycin in a limited number of patients with either cSSSI or HABP/VABP and concurrent *S. aureus* bacteremia[1]
- This study evaluated telavancin activity against a global collection of multidrug-resistant (MDR) Gram-positive and ceftaroline-nonsusceptible *S. aureus* isolates collected from 2015 to 2017

MATERIALS AND METHODS

Bacterial strain collection

- A total of 24,408 Gram-positive isolates were collected from 144 sites located in 34 countries from 2015 to 2017
 - North American isolates (n = 19,617) were collected from 2015 to 2017
 - Isolates from Europe (n = 3,706), Asia-Pacific (n = 547), and Latin America (n = 538) were collected from 2015 to 2016
- Isolates were principally from cSSSIs (42.2%), bloodstream infections (21.2%), and pneumonia in hospitalized patients (21.1%)

Antimicrobial susceptibility test methods and MDR definitions

- Isolates were tested for susceptibility by current Clinical and Laboratory Standards Institute (CLSI) methods, and minimal inhibitory concentration (MIC) interpretations used current CLSI and European Committee on Antimicrobial Susceptibility Testing criteria[4-6]
- Telavancin broth microdilution MIC testing followed the CLSI-approved method, which includes supplementation with 0.002% polysorbate-80
- Bacterial inoculum densities were monitored by colony counts
- MIC values were validated by concurrently tested CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213, *Enterococcus faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619)
- An isolate was considered MDR if it was nonsusceptible to ≥3 of the following antimicrobials (using CLSI interpretive criteria, where applicable):
 - MRSA: ceftaroline, clindamycin, daptomycin, erythromycin, gentamicin, levofloxacin, linezolid, teicoplanin, tetracycline, trimethoprim-sulfamethoxazole, and vancomycin
 - Coagulase-negative staphylococci (CoNS): ceftaroline, clindamycin, daptomycin, erythromycin, gentamicin, levofloxacin, linezolid, oxacillin, teicoplanin, tetracycline, trimethoprim-sulfamethoxazole, and vancomycin
 - *Streptococcus* spp.: clindamycin, daptomycin, erythromycin, levofloxacin, linezolid, penicillin, teicoplanin, tetracycline, and vancomycin
 - *Enterococcus faecalis*: ampicillin, daptomycin, levofloxacin, linezolid, teicoplanin, tetracycline, and vancomycin

RESULTS

- Telavancin activity against the major tested groups of pathogens is shown in **Table 1** and **Figure 1**
- Telavancin activity was identical for the full *S. aureus* isolate set and the MDR MRSA isolate subset (14.3% of all *S. aureus* and 32.2% of all MRSA) (**Table 1**)
- Telavancin ($MIC_{50/90}$, 0.03/0.06 mg/L; 100% susceptible) exhibited MIC_{50}/MIC_{90} values 8- to 32-fold lower than ceftaroline ($MIC_{50/90}$, 1/2 mg/L), daptomycin ($MIC_{50/90}$, 0.25/0.5 mg/L), linezolid ($MIC_{50/90}$, 1/1 mg/L), and vancomycin ($MIC_{50/90}$, 1/1 mg/L) against MDR MRSA isolates (**Table 2** and **Figure 2**)
- Ceftaroline nonsusceptible isolates (MIC, ≥2 mg/L) comprised 2.7% of all *S. aureus* isolates (18.2% of MDR MRSA), and telavancin maintained potent activity against this subset ($MIC_{50/90}$, 0.06/0.06 mg/L; 100% susceptible) (**Table 1**)
- No significant differences in telavancin activity against MDR MRSA were observed when the MIC data were stratified by continent (**Figure 3**); however, ceftaroline susceptibility varied by geographic region from a low of 39.1% in Asia-Pacific isolates to 85.7% in North American isolates
- Telavancin MIC_{50}/MIC_{90} values for the MDR β-haemolytic streptococci ($MIC_{50/90}$, 0.03/0.03 mg/L; 100.0% susceptible), MDR CoNS ($MIC_{50/90}$, 0.06/0.06 mg/L), and MDR *S. pneumoniae* isolates ($MIC_{50/90}$, 0.008/0.015 mg/L) were unchanged compared to the corresponding full isolate sets (**Table 1**)
- Telavancin MIC_{50}/MIC_{90} values for the MDR viridans group streptococci increased by 2-fold ($MIC_{50/90}$, 0.03/0.06 mg/L; 99.4% susceptible) compared to the full viridans group streptococci isolate set (**Table 1**)
- As expected, telavancin exhibited good activity against vancomycin-susceptible *Enterococcus faecalis* ($MIC_{50/90}$, 0.12/0.25 mg/L; 99.9% susceptible), but was not active against MDR *E. faecalis* ($MIC_{50/90}$, >2/>2 mg/L) (**Table 1**), which were 100% resistant to vancomycin

Table 1. Antimicrobial Activity of Telavancin against Major Organism Groups Tested

Organism group (number tested)	MIC (mg/L)			CLSI[a]	EUCAST[a]
	MIC_{50}	MIC_{90}	Range	%S	%S
***S. aureus* (17,632)**	0.03	0.06	≤0.004–0.12	100.0	100.0[b]
MRSA (7,829)	0.03	0.06	≤0.004–0.12	100.0	100.0[b]
MDR MRSA (2,520)	0.03	0.06	≤0.004–0.12	100.0	100.0[b]
Ceftaroline-NS (484)	0.06	0.06	0.015–0.12	100.0	100.0[b]
CoNS (1,023)	0.06	0.06	0.008–0.25		
MDR CoNS (480)	0.06	0.06	0.015–0.12		
BHS (1,839)	0.03	0.03	≤0.002–0.12	100.0	
MDR (295)	0.03	0.03	0.008–0.12	100.0	
VGS (817)	0.015	0.03	≤0.002–0.25	99.0	
MDR (163)	0.03	0.06	0.004–0.12	99.4	
***S. pneumoniae* (1,232)**	0.008	0.015	0.004–0.03		
MDR (190)	0.008	0.015	0.004–0.03		
VAN-S *E. faecalis* (1,102)	0.12	0.25	≤0.015–0.5	99.9	
MDR *E. faecalis* (52)	>2	>2	0.12–>2	7.7[c]	

[a] Criteria as published by CLSI 2018 and EUCAST 2018.
[b] Breakpoint applied to all *S. aureus*, but approved for MRSA isolates only.
[c] All MDR *E. faecalis* isolates were vancomycin-resistant. Breakpoint applied to all *E. faecalis*, but approved for vancomycin-susceptible isolates only.
BHS, β-haemolytic streptococci; CLSI, Clinical and Laboratory Standards Institute; CoNS, coagulase-negative staphylococci; EUCAST, European Committee on Antimicrobial Susceptibility Testing; MDR, multidrug-resistant; MIC, minimal inhibitory concentration; MRSA, methicillin-resistant *Staphylococcus aureus*; NS, nonsusceptible; S, susceptible; VAN-S, vancomycin-susceptible; VGS, viridans group streptococci.

Table 2. Activity of Telavancin and Comparator Agents against Multidrug-Resistant MRSA Isolates (n = 2,520)

Antimicrobial agent	MIC (mg/L)			CLSI[a]			EUCAST[a]		
	MIC_{50}	MIC_{90}	Range	%S	%I	%R	%S	%I	%R
Telavancin	0.03	0.06	≤0.004 to 0.12	100.0			100.0		0.0[b]
Ceftaroline	1	2	0.25 to >8	81.8	18.0	0.2	81.8	18.0	0.2[c]
Clindamycin	>2	>2	≤0.25 to >2	14.9	0.8	84.3	14.8	0.2	85.1
Daptomycin	0.25	0.5	≤0.12 to 2	99.9			99.9		0.1
Erythromycin	>8	>8	≤0.06 to >8	0.8	1.8	97.3	0.9	0.5	98.6
Gentamicin	≤1	>8	≤1 to >8	83.4	0.4	16.2	83.2		16.8
Levofloxacin	>4	>4	0.12 to >4	1.9	0.7	97.4	1.9		98.1
Linezolid	1	1	≤0.12 to >8	99.9		0.1	99.9		0.1
Teicoplanin	≤0.5	1	≤0.5 to 8	100.0	0.0	0.0	98.9		1.1
Tetracycline	≤0.5	>8	≤0.5 to >8	83.1	1.6	15.3	80.0	2.6	17.4
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 to >4	92.7		7.3	92.7	1.3	6.0
Vancomycin	1	1	≤0.12 to 2	100.0	0.0	0.0	100.0		0.0

[a] Criteria as published by CLSI 2018 and EUCAST 2018.
[b] Breakpoint applied to all *S. aureus*, but approved for MRSA isolates only.
[c] Using other than pneumonia breakpoints.
CLSI, Clinical and Laboratory Standards Institute; EUCAST, European Committee on Antimicrobial Susceptibility Testing; I, intermediate; MIC, minimal inhibitory concentration; MRSA, methicillin-resistant *Staphylococcus aureus*; R, resistant; S, susceptible.

Figure 1. Telavancin Activity against Global Collections of Multidrug-Resistant β-Haemolytic Streptococci, Coagulase-Negative Staphylococci, MRSA, *S. pneumoniae*, and Viridans Group Streptococci

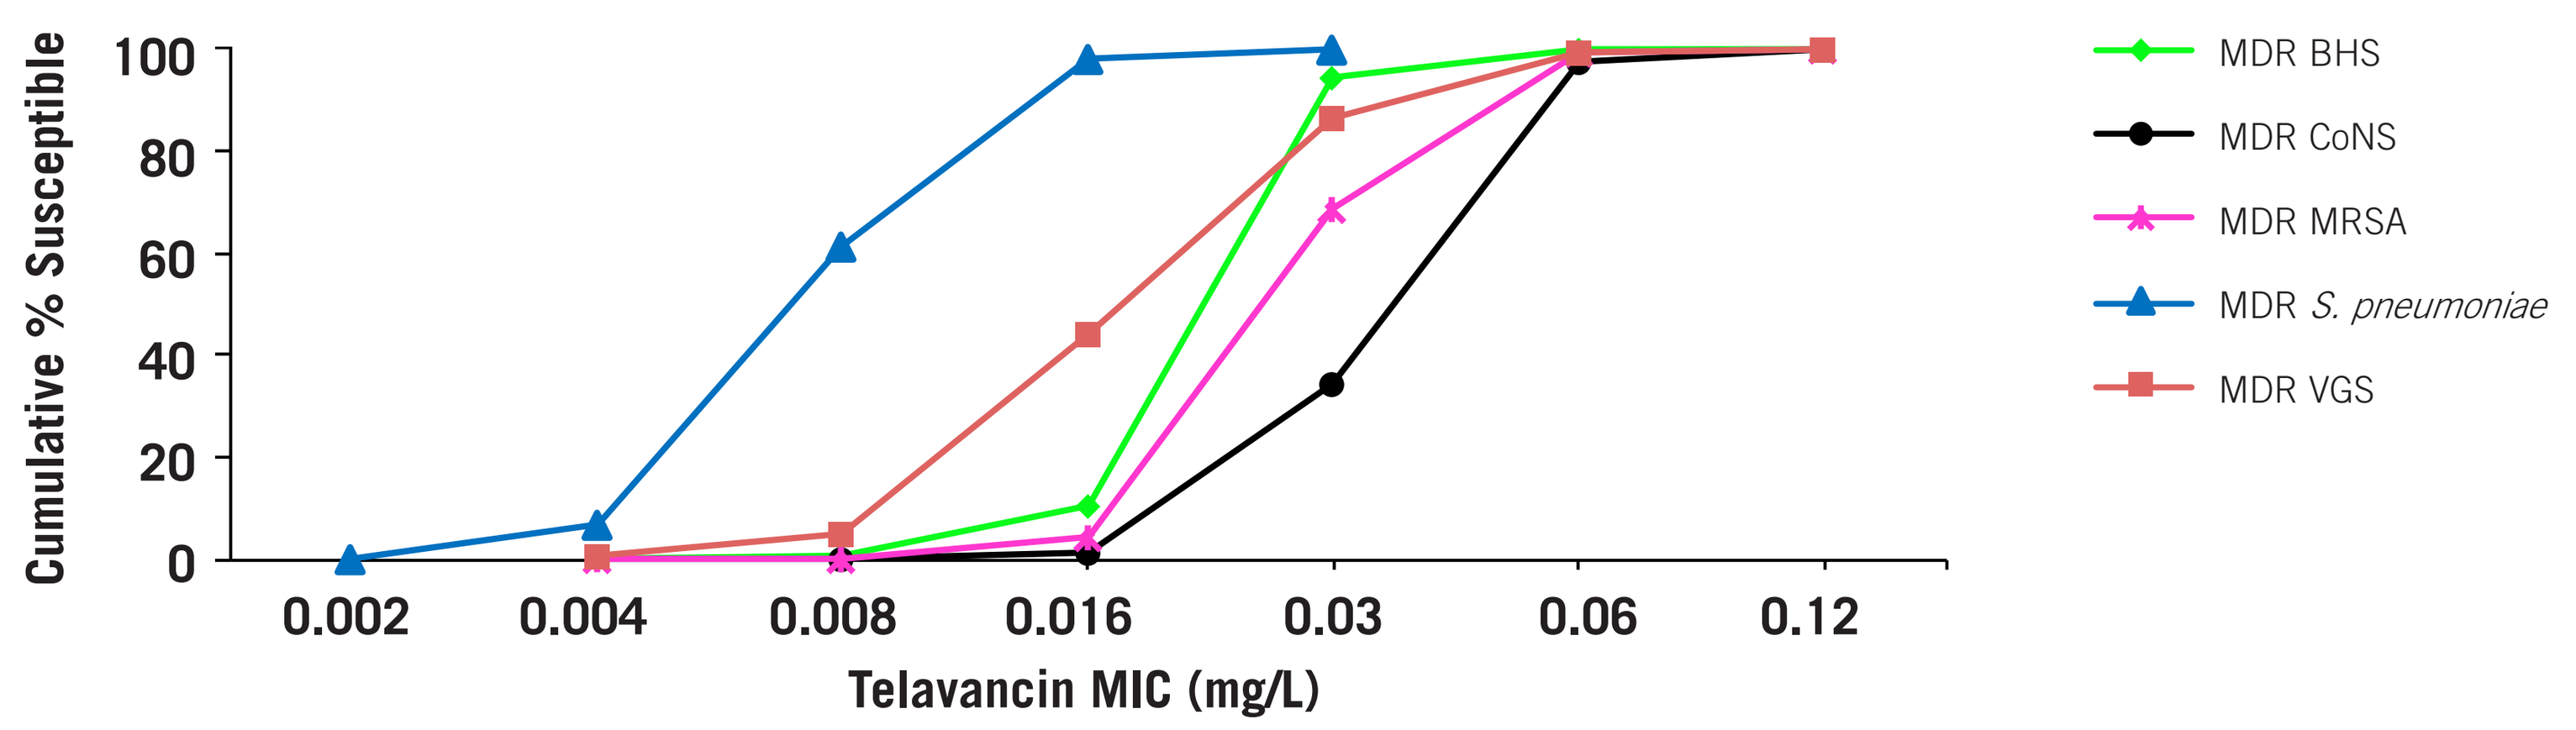

Cumulative % Susceptible
100
80
60
40
20
0
0.002
0.004
0.008
0.016
0.03
0.06
0.12
Telavancin MIC (mg/L)
MDR BHS
MDR CoNS
MDR MRSA
MDR S. pneumoniae
MDR VGS

BHS, β-haemolytic streptococci; CoNS, coagulase-negative staphylococci; MDR, multidrug-resistant; MIC, minimal inhibitory concentration; MRSA, methicillin-resistant *Staphylococcus aureus*; VGS, viridans group streptococci.

Figure 2. Telavancin is More Potent than Tested Comparator Agents against Multidrug-Resistant MRSA

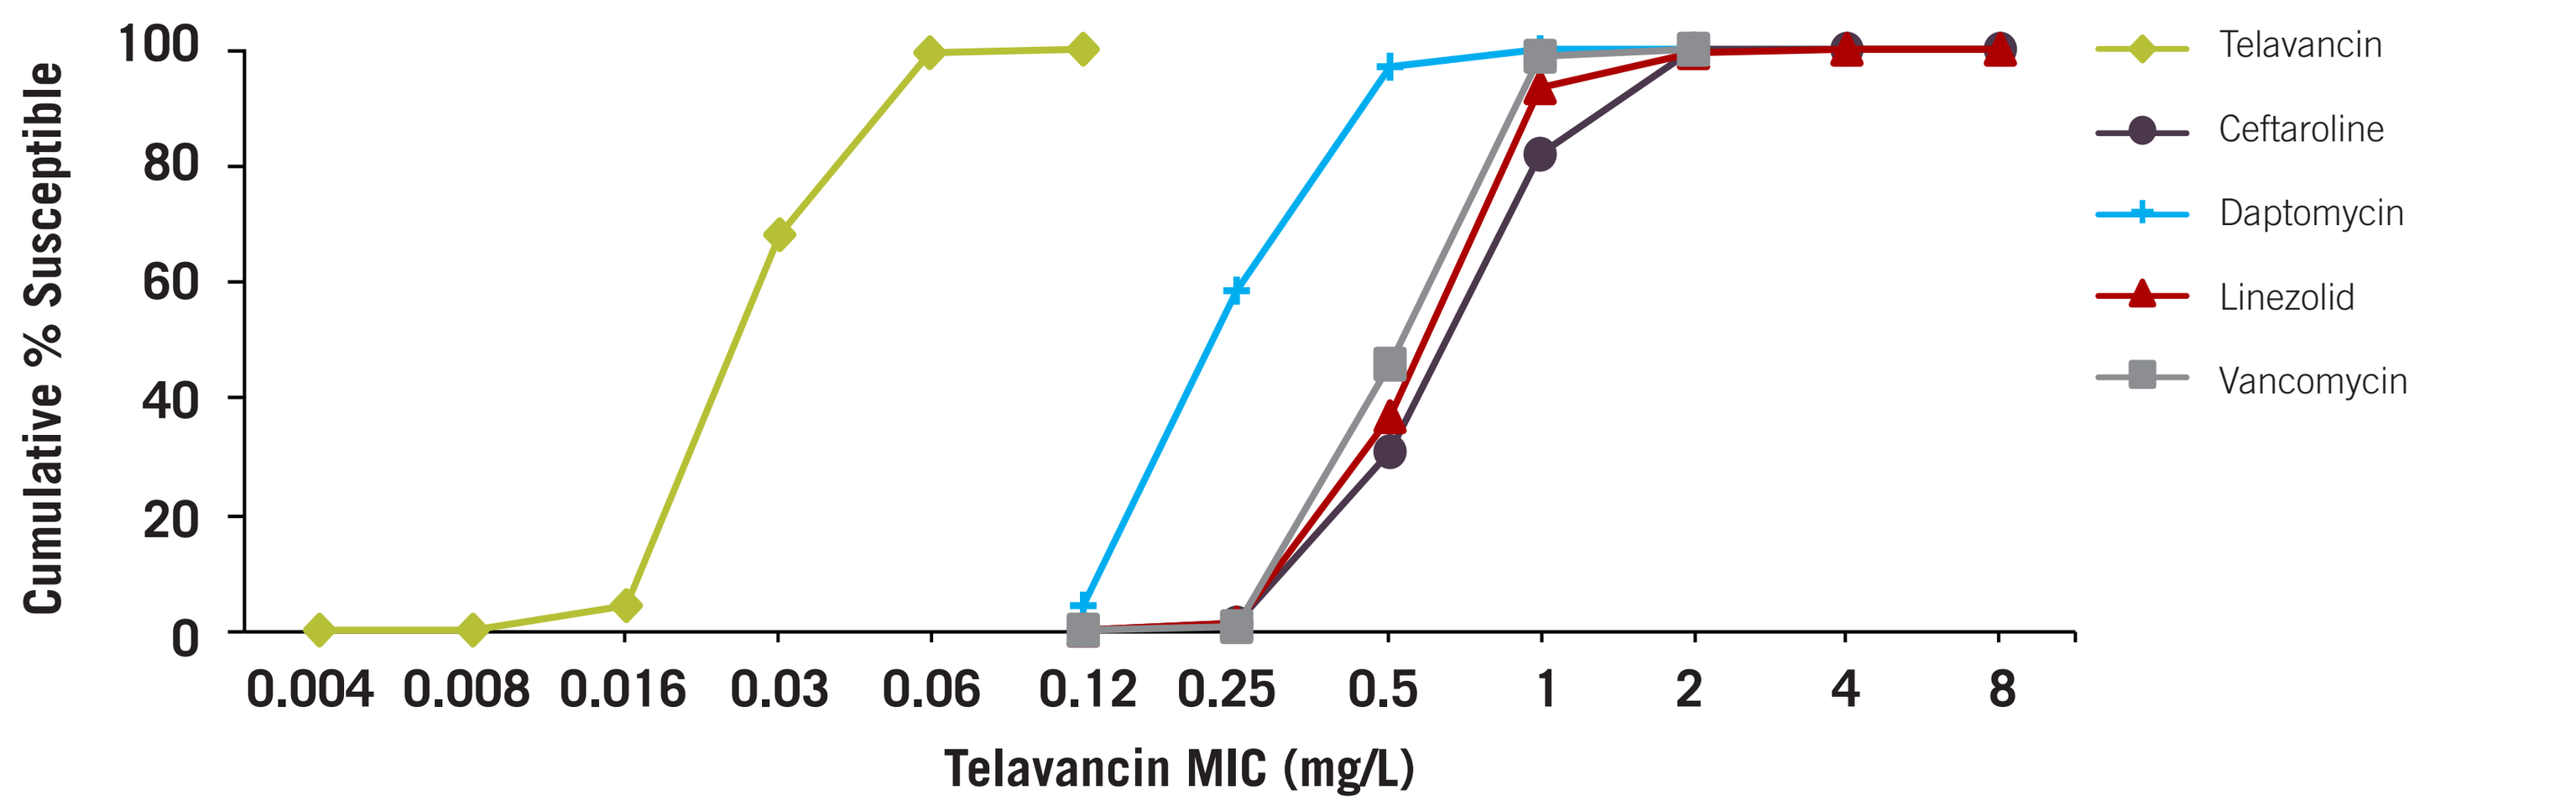

Cumulative % Susceptible
100
80
60
40
20
0
0.004
0.008
0.016
0.03
0.06
0.12
0.25
0.5
1
2
4
8
Telavancin MIC (mg/L)
Telavancin
Ceftaroline
Daptomycin
Linezolid
Vancomycin

MIC, minimal inhibitory concentration; MRSA, methicillin-resistant *Staphylococcus aureus*.

Figure 3. Telavancin and Ceftaroline Activity against Multidrug-Resistant MRSA Isolates by Region

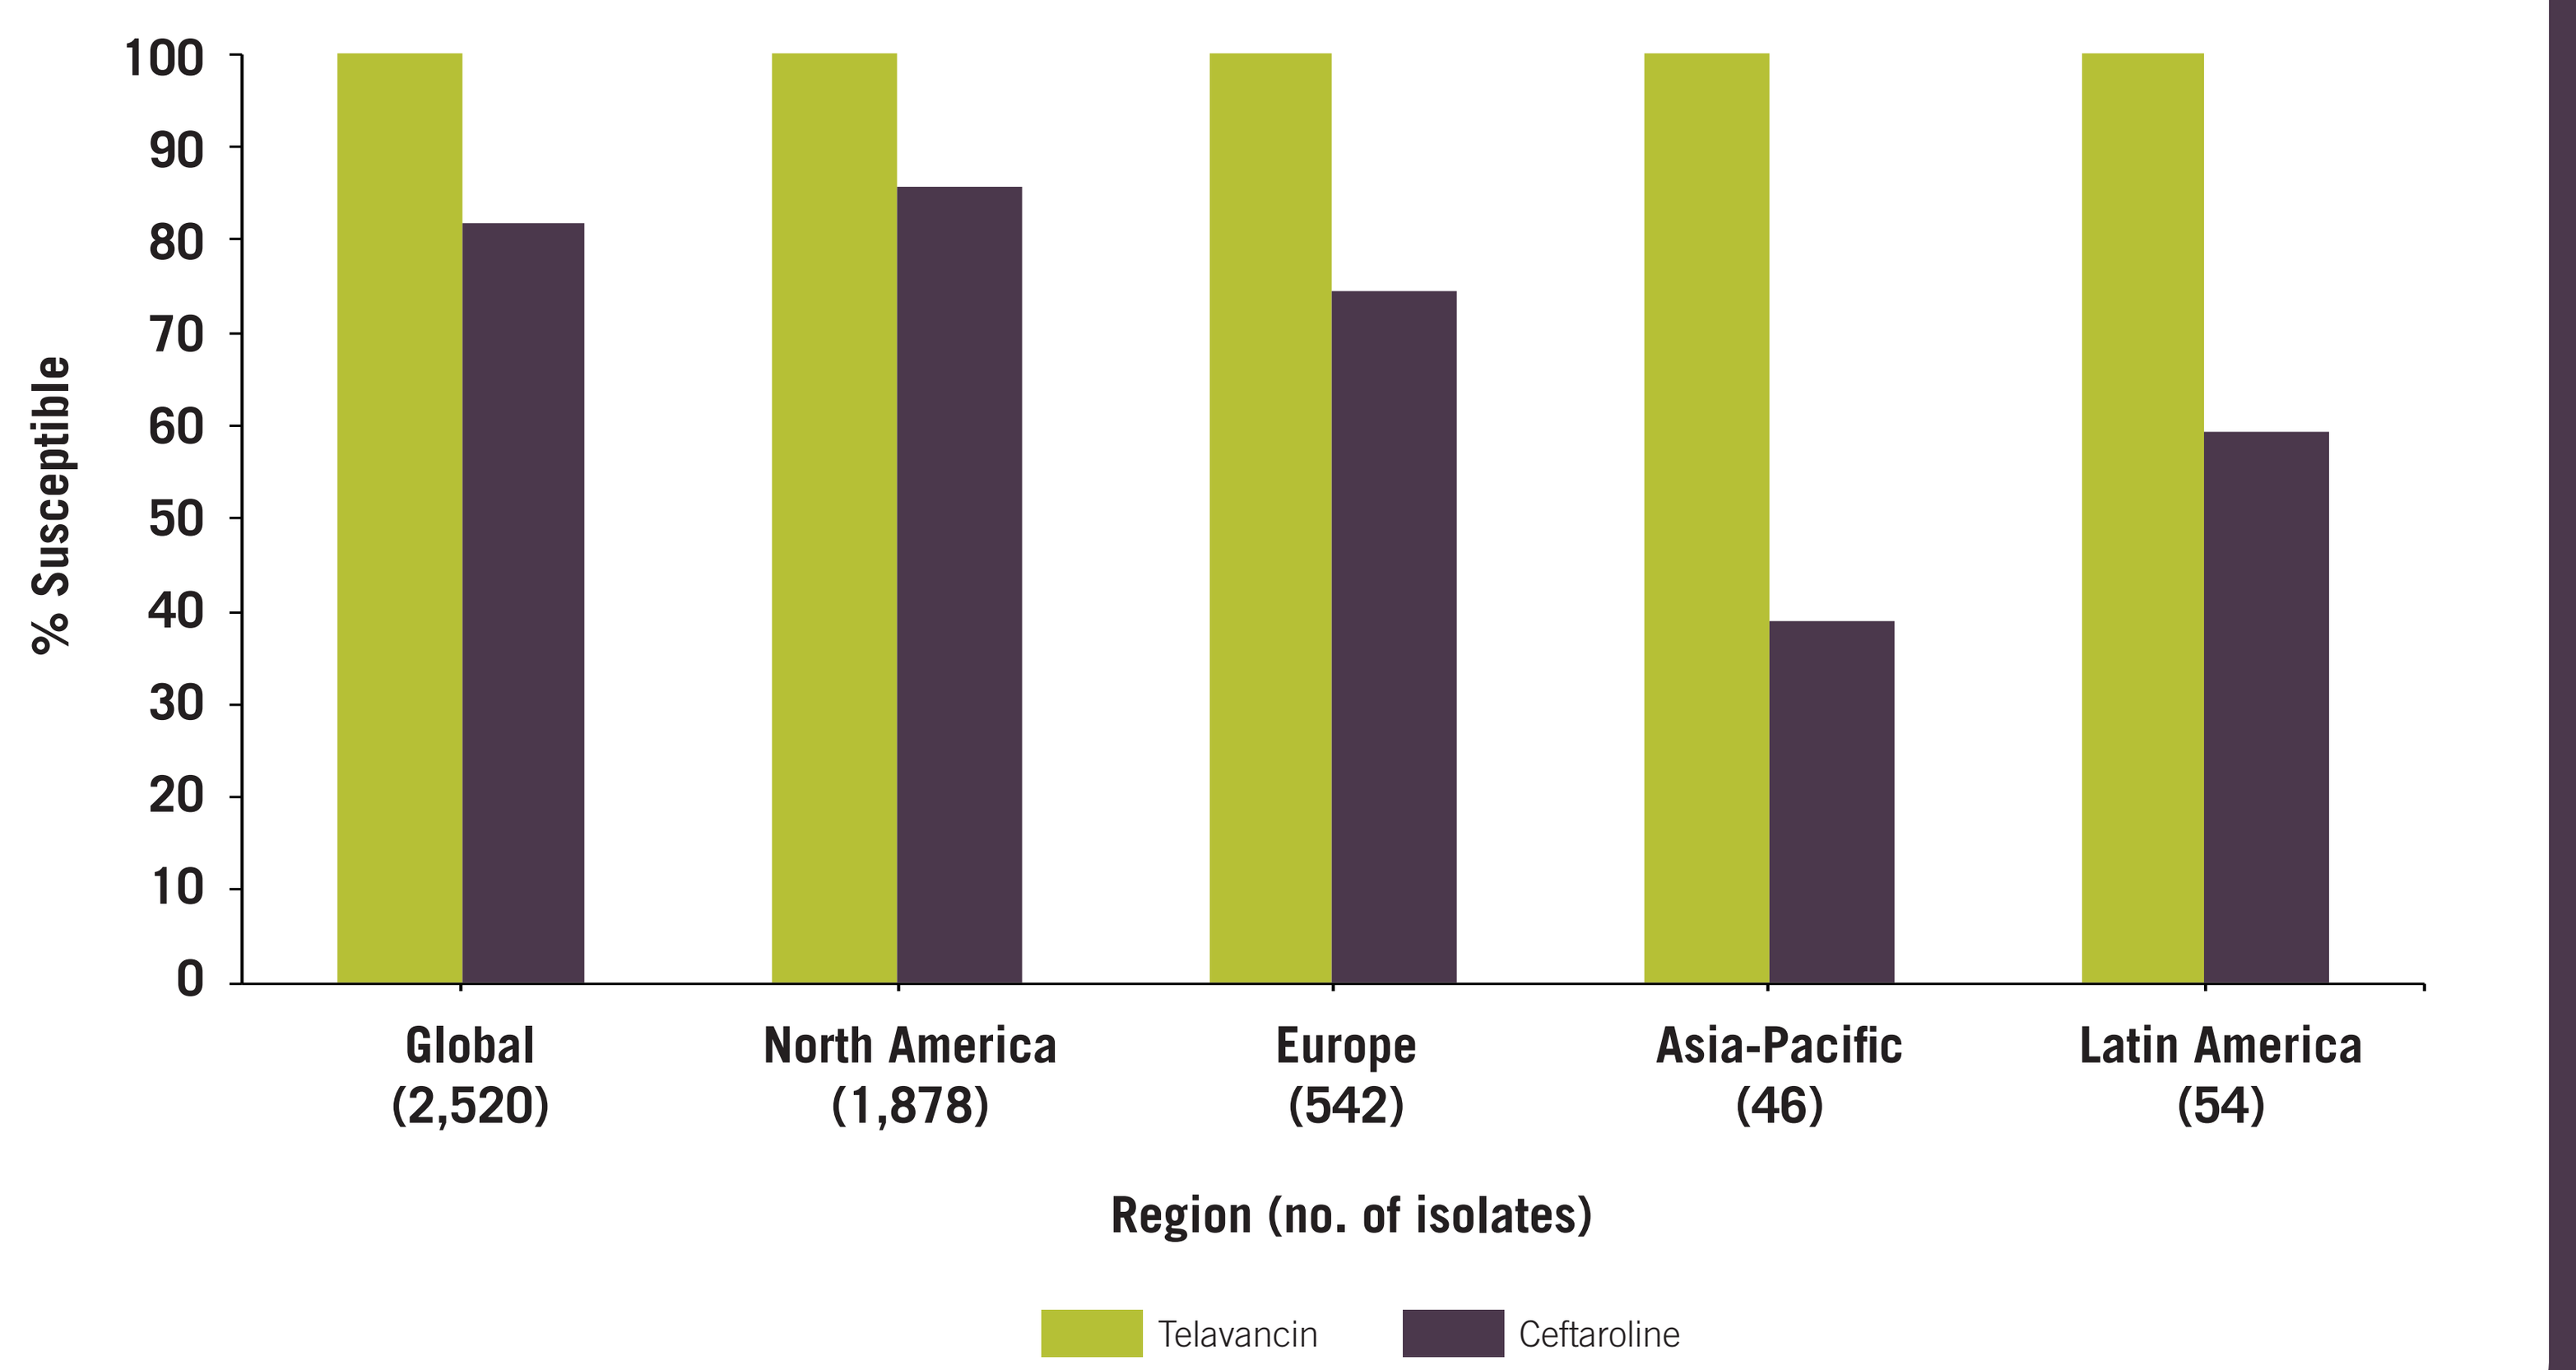

% Susceptible
100
90
80
70
60
50
40
30
20
10
0
Global (2,520)
North America (1,878)
Europe (542)
Asia-Pacific (46)
Latin America (54)
Region (no. of isolates)
Telavancin
Ceftaroline

MRSA, methicillin-resistant *Staphylococcus aureus*.

CONCLUSIONS

- Telavancin showed potent *in vitro* activity against a global collection of Gram-positive clinical isolates from 2015 to 2017, including ceftaroline-nonsusceptible *S. aureus*
- Telavancin activity was unaffected by the MDR status of the isolates, with the exception of vancomycin-resistant *E. faecalis*
- These data indicate that telavancin may be an attractive option for treating cSSSI and pneumonia that are caused by *S. aureus*, regardless of resistance phenotype

REFERENCES

1. VIBATIV® (telavancin), USP [package insert]. South San Francisco, CA: Theravance Biopharma US, Inc.; 2016.
2. Higgins LD, Chang R, Debabov DV, et al. *Antimicrob Agents Chemother*. (2005) 49:1127-1134.
3. Summary of Product Characteristics (VIBATIV 250 mg powder for concentrate for solution for infusion). 2011. EMA. Withdrawal notice filed by the sponsor was approved by EMA in March 2018.
4. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
5. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibilty tests for bacteria that grow aerobically; approved standard - eleventh edition.* Wayne, PA: CLSI.
6. EUCAST (2018). Breakpoint tables for interpretation of MICs and zone diameters. Version 8.0, January 2018. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf.

ACKNOWLEDGEMENTS

The research and publication process was supported by Theravance Biopharma R&D, Inc. Poster production support was coordinated by AlphaBioCom and funded by Theravance Biopharma US, Inc.

Presented at the 2018 ECCMID Meeting, Madrid, Spain, April 21–24, 2018