Antimicrobial Activity of Ceftazidime-Avibactam Tested against Contemporary (2013–2015) Gram-Negative Organisms Collected from United States Medical Centres

ECCMID 2017
Poster #P1291

HS Sader, M Castanheira, D Shortridge, RE Mendes, RK Flamm

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

AMENDED ABSTRACT

Background: Avibactam is a non-β-lactam β-lactamase inhibitor that inhibits enzymes (eg, ESBL, KPC, and AmpC) in Ambler classes A and C and some in class D. Ceftazidime-avibactam was approved by the US Food and Drug Administration in early 2015 and by the European Medicine Agency in mid-2016.

Methods: A total of 36,260 gram-negative bacilli (GNB), including 25,922 *Enterobacteriaceae*, 5,486 *Pseudomonas aeruginosa* (PSA), and 797 *Acinetobacter* spp. (ASP) isolates were collected from 77 US hospitals and tested for susceptibility by reference broth microdilution methods in a central monitoring laboratory (JMI Laboratories). *Enterobacteriaceae* strains with elevated ceftazidime-avibactam MIC values (≥16 mg/L) were evaluated for the presence of genes encoding ESBLs, KPC, NDM, and transferable AmpC enzymes using a microarray-based assay.

Results: An ESBL phenotype was observed among 14.0% of *Escherichia coli* (EC) and 14.7% of *Klebsiella* spp. (KSP) isolates. Ceftazidime-avibactam inhibited 99.9% of all *Enterobacteriaceae* at the susceptible breakpoint of ≤8 mg/L and was highly active against KSP, including ESBL phenotype (n=989; $MIC_{50/90}$, 0.25/1 mg/L; 99.6% susceptible), carbapenem-resistant *Enterobacteriaceae* (CRE; n=392; $MIC_{50/90}$, 0.5/2 mg/L; 97.4% susceptible), multidrug-resistant *Enterobacteriaceae* (MDR; n=2,070; $MIC_{50/90}$, 0.25/1 mg/L; 99.1% susceptible), and extensively drug-resistant *Enterobacteriaceae* (XDR; n=326; $MIC_{50/90}$, 0.5/2 mg/L; 97.9% susceptible). Only 64.4% of ESBL phenotype *K. pneumoniae* (ESBL-KPN) were meropenem-susceptible. Among *Enterobacter cloacae* (ECL; 20.9% ceftazidime-nonsusceptible [CAZ-NS]), 99.9% of isolates, including 99.3% of CAZ-NS strains, were ceftazidime-avibactam-susceptible. Only 18 of 25,923 *Enterobacteriaceae* (0.07%) isolates were ceftazidime-avibactam-nonsusceptible (8 MBL-producers and 1 KPC-2-producing strain with porin alteration; the remaining 9 strains showed negative results for all β-lactamases tested). Ceftazidime-avibactam was very active against PSA ($MIC_{50/90}$, 2/4 mg/L; 97.0% susceptible), including isolates nonsusceptible to meropenem (MEM-NS; 85.8% ceftazidime-avibactam-susceptible), piperacillin-tazobactam (85.2% ceftazidime-avibactam-susceptible) or ceftazidime (80.5% ceftazidime-avibactam-susceptible). Furthermore, 69.9% of PSA isolates nonsusceptible to meropenem, piperacillin-tazobactam, and ceftazidime (n=432) were ceftazidime-avibactam-susceptible.

Conclusions: Ceftazidime-avibactam demonstrated potent activity against a large collection (n=36,273) of contemporary GNB isolated from patients in US hospitals, including organisms resistant to most currently available agents, such as KPC-producing *Enterobacteriaceae* and meropenem-nonsusceptible PSA.

	Cumulative % inhibited at ceftazidime-avibactam MIC (mg/L) of:								
Organism (no. tested)	≤0.12	0.25	0.5	1	2	4	8	16	≥32
Enterobacteriaceae (25,923)	81.3	93.7	98.1	99.4	99.8	99.9	99.9	>99.9	100.0
CRE (392)	13.8	28.1	54.6	82.7	94.4	97.2	97.4	98.2	100.0
MDR (2,070)	44.5	64.1	82.6	93.3	97.4	98.6	99.1	99.6	100.0
XDR (326)	14.4	27.0	55.5	83.4	93.9	96.6	97.9	98.8	100.0
ESBL-KPN (856)	35.7	58.1	80.4	93.0	98.4	99.5	99.5	99.6	100.0
CAZ-NS ECL (568)	7.9	34.3	76.9	94.9	98.2	99.3	99.3	99.6	100.0
P. aeruginosa (5,486)	<0.1	1.9	7.1	44.8	77.6	91.6	97.0	98.7	100.0
MEM-NS (988)	<0.1	0.2	0.8	9.8	34.0	65.6	85.8	93.5	100.0

INTRODUCTION

- As a group, aerobic gram-negative bacilli are the most common causes of nosocomial infections and the most common causes of infection in the intensive care unit; these organisms are very efficient at upregulating and acquiring antimicrobial resistance genes
- β-lactamase production remains the major factor in the rise of antimicrobial resistance in gram-negative bacteria, and the most clinically relevant β-lactamases produced by gram-negative organisms isolated from US medical centres are the ESBLs CTX-M and SHV, and the carbapenemase KPC
- Ceftazidime-avibactam is a combination agent consisting of the β-lactamase inhibitor avibactam and the broad-spectrum cephalosporin ceftazidime. Avibactam effectively inactivates class A (ESBLs and KPC), class C (AmpC), and some class D (such as OXA-48) β-lactamases
- As part of the International Network for Optimal Resistance Monitoring (INFORM) program, we evaluated the activity of ceftazidime-avibactam against a large collection of contemporary (2013–2015) gram-negative organisms causing infection in patients from United States (US) medical centres

MATERIALS AND METHODS

Organism collection

- 36,260 gram-negative bacilli, including 25,922 *Enterobacteriaceae*, 5,486 *Pseudomonas aeruginosa*, 797 *Acinetobacter* spp., 2,603 *Haemophilus influenzae*, 187 *H. parainfluenzae*, and 1,265 *Moraxella catarrhalis*
- The organisms were collected from 77 medical centres in 37 states from all 9 US Census divisions from 2013–2015 as part of the INFORM program
- The isolates were collected from bloodstream, respiratory tract, skin/soft tissue, urinary tract, and intra-abdominal infections according to defined protocols. Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program
- Species identification was confirmed by standard biochemical tests and using the MALDI Biotyper (Bruker Daltonics, Billerica, Massachusetts, USA) according to the manufacturer instructions, where necessary

Susceptibility testing

- Broth microdilution test methods conducted according to the Clinical and Laboratory Standards Institute (CLSI) were performed to determine the antimicrobial susceptibility of ceftazidime-avibactam (inhibitor at fixed concentration of 4 mg/L) and comparator agents

Resistant subsets

- *Escherichia coli*, *Klebsiella pneumoniae*, *K. oxytoca*, and *Proteus mirabilis* isolates were grouped as "ESBL screening-positive phenotype" based on the CLSI screening criteria for ESBL production, ie, MIC of >1 mg/L for ceftazidime, ceftriaxone, and/or aztreonam (CLSI, 2017) for the purpose of susceptibility testing results analysis. Although other β-lactamases, such as AmpC and KPC, may also produce an "ESBL screening-positive phenotype," these strains were grouped together because they usually demonstrate resistance to various broad-spectrum β-lactam compounds
- Carbapenem-resistant *Enterobacteriaceae* (CRE) were defined as isolates displaying imipenem and/or meropenem MIC values at ≥8 mg/L (EUCAST, 2017) and/or ≥4 mg/L for doripenem
 - Imipenem was not applied to *P. mirabilis* and indole-positive Proteeae due to the intrinsically elevated MIC values
- Multidrug-resistant (MDR), extensively drug-resistant (XDR), and pandrug-resistant (PDR) *Enterobacteriaceae* strains were classified according to recommended guidelines (Magiorakos et al., 2012)
 - *Enterobacteriaceae* analysis included the following antimicrobial classes and drug representatives: Broad-spectrum cephalosporins (ceftriaxone, ceftazidime, and cefepime), carbapenems (imipenem, meropenem, and doripenem), broad-spectrum penicillin combined with β-lactamase-inhibitor (piperacillin-tazobactam), fluoroquinolone (ciprofloxacin and levofloxacin), aminoglycosides (gentamicin, tobramycin, and amikacin), glycylcyclines (tigecycline), and the polymyxins (colistin; EUCAST criteria)
 - *P. aeruginosa* analysis included the following antimicrobial classes and drug representatives: Antipseudomonal cephalosporins (ceftazidime and cefepime), carbapenems (imipenem, meropenem, and doripenem), broad-spectrum penicillins combined with β-lactamase-inhibitor (piperacillin-tazobactam), fluoroquinolones (ciprofloxacin and levofloxacin), aminoglycosides (gentamicin, tobramycin, and amikacin), and the polymyxins (colistin)
- Classifications were based on the following recommended parameters:
 - MDR = nonsusceptible (NS; CLSI breakpoints) to at least 3 antimicrobial classes
 - XDR = susceptible (S) to 2 or fewer antimicrobial classes
 - PDR = NS to all antimicrobial classes (Magiorakos et al., 2012)
- CLSI and EUCAST susceptibility interpretive criteria were used to determine susceptibility/resistance rates for comparator agents. Furthermore, the US Food and Drug Administration (FDA) and EUCAST breakpoint criteria were applied for ceftazidime-avibactam when testing *Enterobacteriaceae* and *P. aeruginosa*, ie, susceptible at ≤8 mg/L and resistant at ≥16 mg/L

Screening for β-lactamases

- *Enterobacteriaceae* isolates displaying elevated ceftazidime-avibactam MIC values (≥16 mg/L) were tested for β-lactamase-encoding genes using the microarray based assay Check-MDR CT101 kit (Check-Points, Wageningen, Netherlands)
- The assay was performed according to the manufacturer's instructions. This kit has the capabilities to detect CTX-M groups 1, 2, 8+25, and 9, TEM wildtype (WT) and ESBL, SHV WT and ESBL, ACC, ACT/MIR, CMYII, DHA, FOX, KPC, and NDM-1

RESULTS

- Ceftazidime-avibactam MIC distributions for all organism groups and resistant subsets are summarized in Table 1, and the activities of ceftazidime-avibactam and comparator agents tested against selected organism groups are presented in Table 2 and Figures 1 and 2
- Ceftazidime-avibactam inhibited 99.9% of all *Enterobacteriaceae* isolates (n=25,923) at the susceptible breakpoint of ≤8 mg/L (Tables 1 and 2) and was highly active against MDR (n=2,070; $MIC_{50/90}$, 0.25/1 mg/L; 99.1% susceptible), XDR isolates (n=326; $MIC_{50/90}$, 0.5/2 mg/L; 97.9% susceptible), and CRE (n=392) isolates $MIC_{50/90}$, 0.5/2 mg/L; 97.4% susceptible; Tables 1 and 2)
- The most active compound tested against MDR and XDR *Enterobacteriaceae* isolates was ceftazidime-avibactam (99.1% and 97.9% susceptible, respectively [EUCAST and US FDA]), followed by tigecycline (79.8% and 86.8% susceptible [EUCAST], respectively), amikacin (83.4% and 43.4% [EUCAST], respectively), and meropenem (83.1% and 22.1% susceptible [EUCAST], respectively; Table 2)
- Among CRE, 97.4% of isolates were susceptible to ceftazidime-avibactam (Table 2 and Figure 1), whereas only 78.6% were susceptible to colistin (Table 2)
- Only 18 of 25,923 *Enterobacteriaceae* (0.07%) were ceftazidime-avibactam-nonsusceptible; 5 NDM-1-producing strains (3 *E. coli* and 2 *K. pneumoniae*), 1 VIM-1-producing *E. cloacae*, 1 VIM-4-producing *K. pneumoniae*, 1 IMP-27-producing *Morganella morganii*, 1 KPC-producing *K. pneumoniae*, and 9 isolates (3 *E. cloacae*, 1 *E. aerogenes*, 3 *Providencia stuartii*, and 2 *Serratia marcescens*) with negative results for all β-lactamases tested (data not shown)
- Ceftazidime-avibactam was very active against *P. aeruginosa* (n=5,486; $MIC_{50/90}$, 2/4 mg/L; 97.0% susceptible), including most MDR (n=1,024; $MIC_{50/90}$, 4/16 mg/L; 85.2% susceptible) and XDR (n=465; $MIC_{50/90}$, 8/32 mg/L; 72.9% susceptible; Tables 1 and 2)
- Ceftazidime-avibactam retained potent *in vitro* activity against *P. aeruginosa* isolates nonsusceptible to meropenem ($MIC_{50/90}$, 4/16 mg/L; 85.8% susceptible), piperacillin-tazobactam ($MIC_{50/90}$, 4/16 mg/L; 85.2% susceptible), or ceftazidime ($MIC_{50/90}$, 4/16 mg/L; 80.5% susceptible), as well as isolates nonsusceptible to meropenem, piperacillin-tazobactam, and ceftazidime ($MIC_{50/90}$, 8/32 mg/L; 69.9% susceptible; Table 1)

Figure 1 Antimicrobial activities of ceftazidime-avibactam (CAZ-AVI) and meropenem against selected resistant subsets of *Enterobacteriaceae* isolates

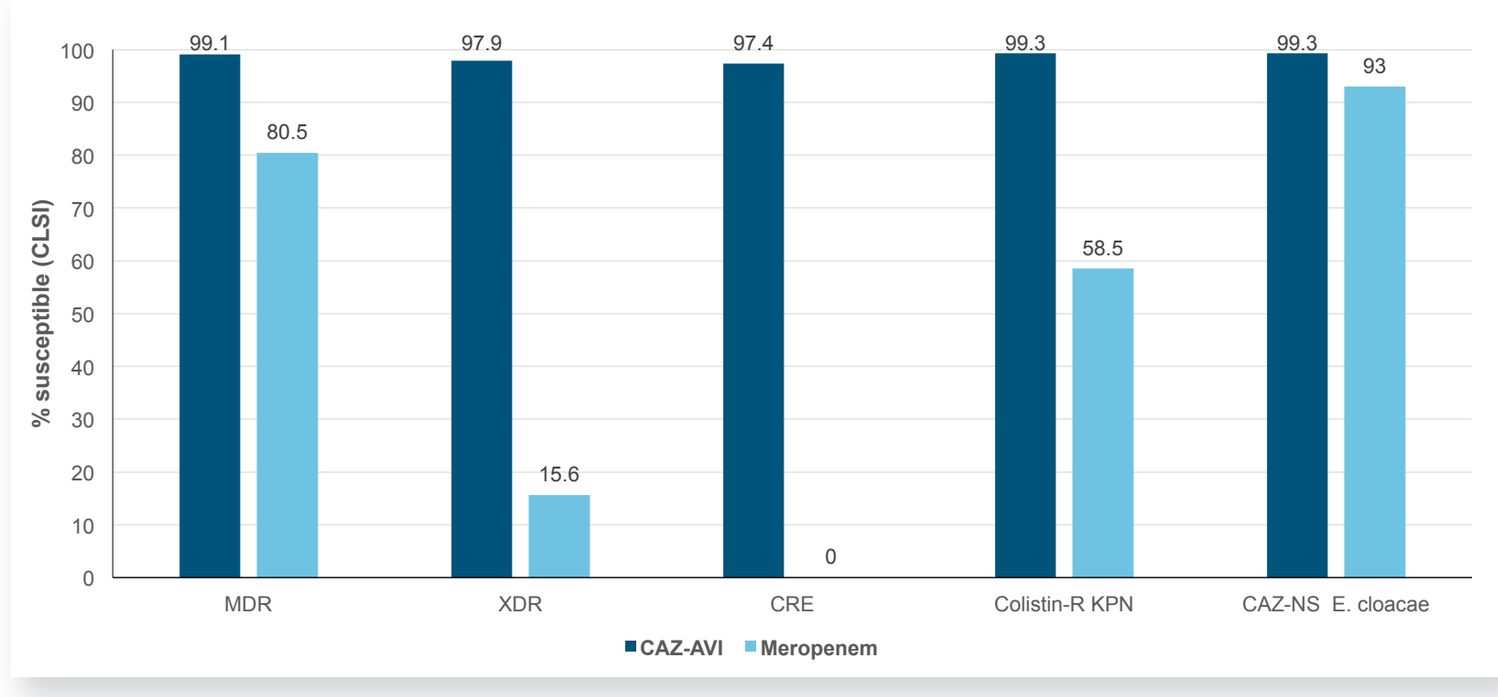
% susceptible (CLSI)
99.1
80.5
97.9
15.6
97.4
0
99.3
58.5
99.3
93
MDR
XDR
CRE
Colistin-R KPN
CAZ-NS E. cloacae
CAZ-AVI
Meropenem

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; CRE, carbapenem-resistant *Enterobacteriaceae*; R, resistant; KPN, *Klebsiella pneumoniae*; CAZ, ceftazidime; NS, nonsusceptible

Figure 2 Antimicrobial activities of ceftazidime-avibactam (CAZ-AVI) and meropenem against *P. aeruginosa*

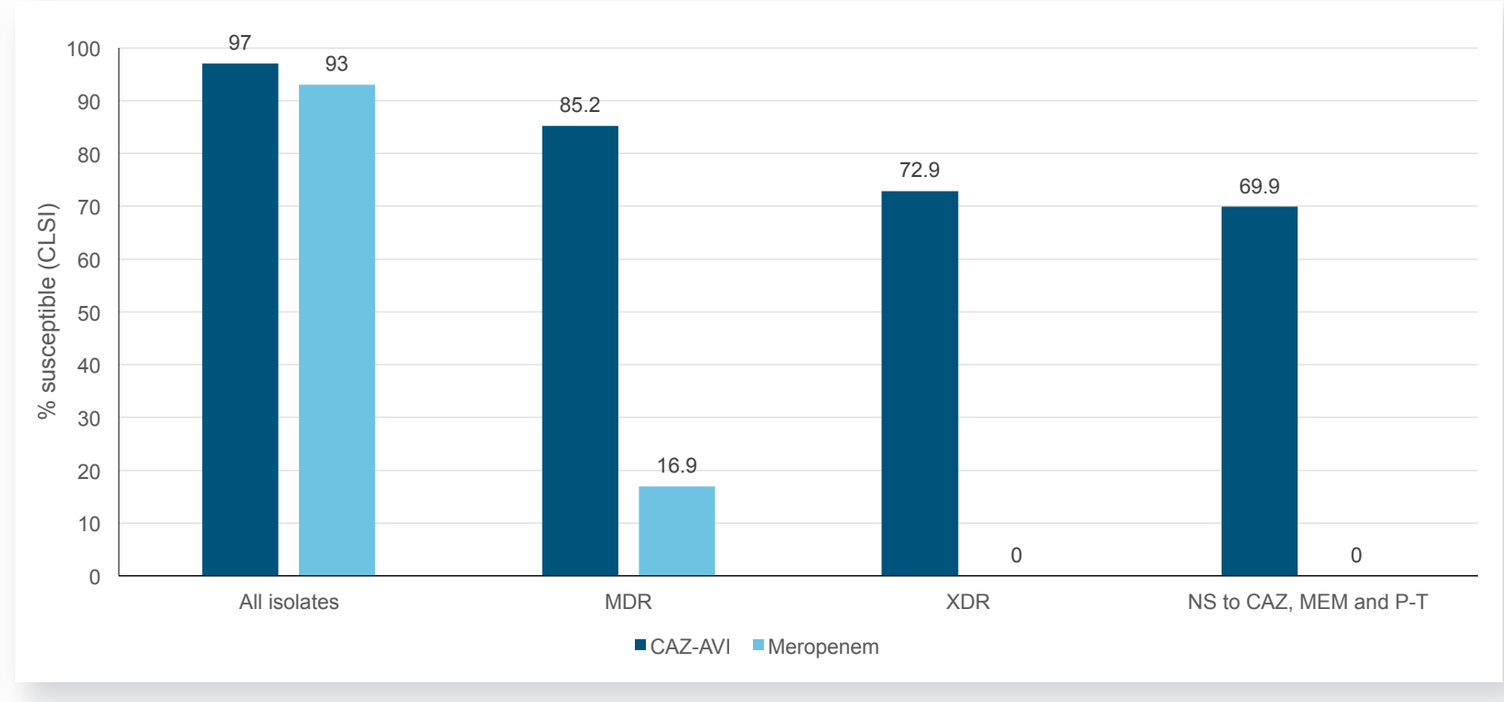
% susceptible (CLSI)
97
93
85.2
16.9
72.9
0
69.9
0
All isolates
MDR
XDR
NS to CAZ, MEM and P-T
CAZ-AVI
Meropenem

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; NS, nonsusceptible; CAZ, ceftazidime; MEM, meropenem; P-T, piperacillin-tazobactam

CONCLUSIONS

- Ceftazidime-avibactam demonstrated potent activity against a large collection (n=36,273) of contemporary gram-negative bacilli isolated from patients in US hospitals, including organisms resistant to most currently available agents, such as KPC-producing *Enterobacteriaceae* and meropenem-nonsusceptible *P. aeruginosa*
- Notably, ceftazidime-avibactam remained highly active against CRE as well as *Enterobacteriaceae* and *P. aeruginosa* isolates with MDR and XDR phenotypes

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the International Network for Optimal Resistance Monitoring (INFORM) program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results and JMI Laboratories received compensation fees for services in relation to preparing the abstract and poster. Allergan was not involved in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi. Accessed January 2017.
2. Castanheira M, Mendes RE, Sader HS (2017). Low frequency of ceftazidime-avibactam resistance among Enterobacteriaceae isolates carrying bla_{KPC} collected in hospitals from the United States from 2012 to 2015. *Antimicrob Agents Chemother in press.*
3. Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.
4. EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.
5. Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268-281.
6. Sader HS, Castanheira M, Flamm RK (2017). Antimicrobial activity of ceftazidime-avibactam when tested against gram-negative bacteria isolated from patients hospitalized with pneumonia in United States medical centers (2011-2015). *Antimicrob Agents Chemother in press.*
7. Sader HS, Huband MD, Castanheira M, Flamm RK (2017). Antimicrobial susceptibility of *Pseudomonas aeruginosa:* Results from four years (2012-2015) of the International Network for Optimal Resistance Monitoring (INFORM) Program in the United States. *Antimicrob Agents Chemother in press.*
8. Zavicefta (2016). Zavicefta® Package Insert. AstraZeneca AB. Available at http://www.ema.europa.eu/docs/en_GB/document_library/EPAR_-_Product_Information/human/004027/WC500210234.pdf. Accessed September 2016.

Table 1 Antimicrobial activity of ceftazidime-avibactam tested against antimicrobial resistant gram-negative organisms

Organisms / Organism groups	No. of isolates at MIC (mg/L; cumulative %)														
	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32	MIC_{50}	MIC_{90}
Enterobacteriaceae (25,923)	868 (3.3)	3,430 (16.6)	8,126 (47.9)	8,651 (81.3)	3,225 (93.7)	1,137 (98.1)	320 (99.4)	103 (99.8)	34 (99.9)	11 (99.9)	10 (>99.9)	1 (>99.9)	7 (100.0)	0.12	0.25
MDR (2,070)	63 (3.0)	161 (10.8)	276 (24.2)	422 (44.5)	404 (64.1)	384 (82.6)	221 (93.3)	85 (97.4)	26 (98.6)	10 (99.1)	10 (99.6)	1 (99.7)	7 (100.0)	0.25	1
XDR (326)	10 (3.1)	5 (4.6)	13 (8.6)	19 (14.4)	41 (27.0)	93 (55.5)	91 (83.4)	34 (93.9)	9 (96.6)	4 (97.9)	3 (98.8)	0 (98.8)	4 (100.0)	0.5	2
CRE (392)	12 (3.1)	4 (4.1)	12 (7.1)	26 (13.8)	56 (28.1)	104 (54.6)	110 (82.7)	46 (94.4)	11 (97.2)	1 (97.4)	3 (98.2)	0 (98.2)	7 (100.0)	0.5	2
E. coli (9,191)	659 (7.2)	1,054 (18.6)	3,500 (56.7)	3,130 (90.8)	680 (98.2)	129 (99.6)	25 (99.8)	9 (99.9)	2 (>99.9)	0 (>99.9)	0 (>99.9)	0 (>99.9)	3 (100.0)	0.06	0.12
ESBL-phenotype (1,289)	67 (5.2)	44 (8.6)	206 (24.6)	556 (67.7)	299 (90.9)	79 (97.1)	24 (98.9)	9 (99.6)	2 (99.8)	0 (99.8)	0 (99.8)	0 (99.8)	3 (100.0)	0.12	0.25
MEM-NS (21)	1 (4.8)	0 (4.8)	3 (19.0)	3 (33.3)	7 (66.7)	3 (81.0)	0 (81.0)	1 (85.7)	0 (85.7)	0 (85.7)	0 (85.7)	0 (85.7)	3 (100.0)	0.25	>32
K. pneumoniae (5,310)	94 (1.8)	267 (6.8)	1,853 (41.7)	2,010 (79.5)	643 (91.7)	273 (96.8)	110 (98.9)	46 (99.7)	10 (99.9)	0 (99.9)	1 (99.9)	0 (99.9)	3 (100.0)	0.12	0.25
ESBL-phenotype (856)	31 (3.6)	17 (5.6)	52 (11.7)	206 (35.7)	191 (58.1)	191 (80.4)	108 (93.0)	46 (98.4)	10 (99.5)	0 (99.5)	1 (99.6)	0 (99.6)	3 (100.0)	0.25	1
MEM-NS (306)	12 (3.9)	4 (5.2)	6 (7.2)	19 (13.4)	43 (27.5)	88 (56.2)	82 (83.0)	39 (95.8)	9 (98.7)	0 (98.7)	1 (99.0)	0 (99.0)	3 (100.0)	0.5	2
Colistin-resistant (147)	4 (2.7)	2 (4.1)	15 (14.3)	36 (38.8)	19 (51.7)	32 (73.5)	24 (89.8)	10 (96.6)	4 (99.3)	0 (99.3)	0 (99.3)	0 (99.3)	1 (100.0)	0.25	2
K. oxytoca (1,422)	5 (0.4)	70 (5.3)	650 (51.0)	518 (87.4)	116 (95.6)	48 (98.9)	11 (99.7)	2 (99.9)	1 (99.9)	1 (100.0)				0.06	0.25
ESBL-phenotype (133)	1 (0.8)	1 (1.5)	20 (16.5)	47 (51.9)	29 (73.7)	21 (89.5)	10 (97.0)	2 (98.5)	1 (99.2)	1 (100.0)				0.12	1
Proteus mirabilis (1,992)	38 (1.9)	1,267 (65.5)	623 (96.8)	49 (99.2)	9 (99.7)	3 (99.8)	2 (99.9)	0 (99.9)	0 (99.9)	1 (100.0)				0.03	0.06
ESBL-phenotype (115)	2 (1.7)	37 (33.9)	56 (82.6)	11 (92.2)	4 (95.7)	2 (97.4)	2 (99.1)	0 (99.1)	0 (99.1)	1 (100.0)				0.06	0.12
E. cloacae (2,719)	18 (0.7)	21 (1.4)	160 (7.3)	1,227 (52.4)	817 (82.5)	330 (94.6)	114 (98.8)	22 (99.6)	6 (99.9)	0 (99.9)	2 (99.9)	1 (>99.9)	1 (100.0)	0.12	0.5
CAZ-NS (568)	9 (1.6)	1 (1.8)	6 (2.8)	29 (7.9)	150 (34.3)	242 (76.9)	102 (94.9)	19 (98.2)	6 (99.3)	0 (99.3)	2 (99.6)	1 (99.8)	1 (100.0)	0.5	1
E. aerogenes (957)	17 (1.8)	35 (5.4)	267 (33.3)	418 (77.0)	158 (93.5)	47 (98.4)	7 (99.2)	2 (99.4)	5 (99.9)	0 (99.9)	1 (100.0)			0.12	0.25
CAZ-NS (193)	5 (2.6)	5 (5.2)	11 (10.9)	51 (37.3)	77 (77.2)	30 (92.7)	6 (95.9)	2 (96.9)	5 (99.5)	0 (99.5)	1 (100.0)			0.25	0.5
Morganella morganii (718)	2 (0.3)	305 (42.8)	293 (83.6)	70 (93.3)	26 (96.9)	13 (98.7)	7 (99.7)	1 (99.9)	0 (99.9)	0 (99.9)	1 (100.0)			0.06	0.12
Citrobacter koseri (563)	17 (3.0)	65 (14.6)	280 (64.3)	149 (90.8)	42 (98.2)	9 (99.8)	0 (99.8)	1 (100.0)						0.06	0.12
C. freundii (708)	9 (1.3)	12 (3.0)	135 (22.0)	325 (67.9)	148 (88.8)	60 (97.3)	15 (99.4)	1 (99.6)	2 (99.9)	1 (100.0)				0.12	0.5
Serratia marcescens (1,278)	0 (0.0)	4 (0.3)	44 (3.8)	551 (46.9)	485 (84.8)	167 (97.9)	18 (99.3)	4 (99.6)	1 (99.7)	2 (99.8)	2 (100.0)			0.25	0.5
Proteus vulgaris (414)	3 (0.7)	186 (45.7)	181 (89.4)	38 (98.6)	2 (99.0)	4 (100.0)								0.06	0.12
Providencia spp. (649)	5 (0.8)	144 (23.0)	140 (44.5)	165 (70.0)	99 (85.2)	54 (93.5)	11 (95.2)	15 (97.5)	7 (98.6)	6 (99.5)	3 (100.0)			0.12	0.5
P. aeruginosa (5,486)					105 (1.9)	285 (7.1)	2,065 (44.8)	1,801 (77.6)	770 (91.6)	296 (97.0)	95 (98.7)	31 (99.3)	38 (100.0)	2	4
MDR (1,024)					4 (0.4)	7 (1.1)	83 (9.2)	232 (31.8)	329 (64.0)	217 (85.2)	85 (93.5)	30 (96.4)	37 (100.0)	4	16
XDR (465)			0 (0.0)	1 (0.2)	0 (0.2)	1 (0.4)	19 (4.5)	66 (18.7)	115 (43.4)	137 (72.9)	64 (86.7)	26 (92.3)	36 (100.0)	8	32
CAZ-NS (838)			0 (0.0)	1 (0.1)	1 (0.2)	7 (1.1)	62 (8.5)	206 (33.1)	234 (61.0)	164 (80.5)	94 (91.8)	31 (95.5)	38 (100.0)	4	16
MEM-NS (988)					2 (0.2)	6 (0.8)	89 (9.8)	239 (34.0)	312 (65.6)	200 (85.8)	76 (93.5)	27 (96.3)	37 (100.0)	4	16
PT-NS (1,023)			0 (0.0)	1 (0.1)	1 (0.2)	12 (1.4)	76 (8.8)	228 (31.1)	321 (62.5)	233 (85.2)	87 (93.7)	29 (96.6)	35 (100.0)	4	16
NS to CAZ, MEM, and P/T (432)						0 (0.0)	14 (3.2)	62 (17.6)	112 (43.5)	114 (69.9)	70 (86.1)	26 (92.1)	34 (100.0)	8	32
Acinetobacter spp. (797)					0 (0.0)	2 (0.3)	6 (1.0)	30 (4.8)	135 (21.7)	142 (39.5)	153 (58.7)	123 (74.2)	206 (100.0)	16	>32
MEM-NS (399)						0 (0.0)	1 (0.3)	1 (0.5)	18 (5.0)	37 (14.3)	82 (34.8)	75 (53.6)	185 (100.0)	31	>32
H. influenzae (2,603)	1,696 (65.2)	757 (94.2)	123 (99.0)	25 (99.9)	2 (100.0)									≤0.015	0.03
β-lactamase-positive (690)	468 (67.8)	200 (96.8)	13 (98.7)	7 (99.7)	2 (100.0)									≤0.015	0.03
H. parainfluenzae (187)	158 (84.5)	22 (96.3)	7 (100.0)											≤0.015	0.03
M. catarrhalis (1,265)	204 (16.1)	235 (34.7)	592 (81.5)	178 (95.6)	55 (99.9)	1 (100.0)								0.06	0.12

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; CRE, carbapenem-resistant *Enterobacteriaceae*; ESBL, extended-spectrum β-lactamase phenotype (ESBL-screen-positive phenotype per CLSI, 2017); MEM-NS, meropenem-nonsusceptible (MIC, ≥4 mg/L for *P. aeruginosa* and ≥2 mg/L for *Enterobacteriaceae*); P-T-NS, piperacillin-tazobactam-nonsusceptible (MIC, ≥16 mg/L for *P. aeruginosa* and *Enterobacteriaceae*); CAZ-NS, ceftazidime-nonsusceptible (MIC, ≥16 mg/L for *P. aeruginosa* and ≥8 mg/L for *Enterobacteriaceae*)

Table 2 Activity of ceftazidime-avibactam and comparator antimicrobial agents when tested against selected gram-negative organisms and resistance phenotypes

Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %R
Enterobacteriaceae (25,923)						
Ceftazidime-avibactam	0.12	0.25	99.9[b]	0.1[b]	99.9	0.1
Ceftazidime	0.12	8	89.1	9.5	86.6	10.9
Ceftriaxone	≤0.06	>8	85.7	13.2	85.7	13.2
Cefepime	≤0.03	1	91.5	6.4[c]	90.2	7.5
Piperacillin-tazobactam	2	16	92.8	4.1	90.0	7.2
Meropenem	≤0.06	≤0.06	98.4	1.4	98.6	0.8
Levofloxacin	≤0.12	>4	82.4	15.9	80.9	17.6
Gentamicin	≤1	4	91.1	7.7	90.0	8.9
Amikacin	2	4	99.2	0.2	98.4	0.8
Tigecycline	0.25	1	98.5[b]	<0.1[b]	93.5	1.5
Colistin	≤0.5	>8	—	—	77.9	22.1
MDR *Enterobacteriaceae* (2,070)[c]						
Ceftazidime-avibactam	0.25	1	99.1	0.9[b]	99.1	0.9
Ceftazidime	32	>32	28.7	64.6	21.3	71.3
Ceftriaxone	>8	>8	18.3	78.4	18.3	78.4
Cefepime	16	>64	36.0	51.5[c]	30.5	58.8
Piperacillin-tazobactam	16	>64	50.9	31.7	43.8	49.1
Meropenem	≤0.06	>8	80.5	16.9	83.1	10.1
Levofloxacin	>4	>4	19.8	71.8	14.1	80.2
Gentamicin	>8	>8	37.4	51.5	34.0	62.6
Amikacin	2	16	90.2	2.3	83.4	9.8
Tigecycline	0.5	2	91.6[b]	0.3[b]	79.8	8.4
Colistin	≤0.5	>8	—	—	61.7	38.3
XDR *Enterobacteriaceae* (326)[d]						
Ceftazidime-avibactam	0.5	2	97.9	2.1[b]	97.9	2.1
Ceftazidime	>32	>32	3.7	94.5	2.5	96.3
Ceftriaxone	>8	>8	1.2	98.5	1.2	98.5
Cefepime	32	>64	4.9	84.5[c]	1.9	92.2
Piperacillin-tazobactam	>64	>64	4.6	86.5	4.0	95.4
Meropenem	>8	>8	15.6	77.9	22.1	52.1
Levofloxacin	>4	>4	8.0	87.4	3.7	92.0
Gentamicin	8	>8	29.8	49.1	25.2	70.2
Amikacin	16	32	58.2	8.6	43.4	41.8
Tigecycline	0.5	2	93.3[b]	0.3[b]	86.8	6.7
Colistin	≤0.5	>8	—	—	63.0	37.0
CRE (392)[e]						
Ceftazidime-avibactam	0.5	2	97.4	2.6[b]	97.4	2.6
Ceftazidime	>32	>32	4.3	93.6	2.3	95.7
Ceftriaxone	>8	>8	2.3	97.4	2.3	97.4
Cefepime	32	>64	8.5	76.0[c]	3.9	86.8
Piperacillin-tazobactam	>64	>64	3.1	92.1	2.6	96.9
Meropenem	>8	>8	2.3	90.6	9.4	54.3
Levofloxacin	>4	>4	22.2	75.0	18.1	77.8
Gentamicin	4	>8	51.5	33.7	45.7	48.5
Amikacin	16	32	66.6	6.9	49.2	33.4
Tigecycline	0.5	1	99.5[b]	0.0[b]	92.1	0.5
Colistin	≤0.5	>8	—	—	78.6	21.4
Meropenem-nonsusceptible *K. pneumoniae* (306)[f]						
Ceftazidime-avibactam	0.5	2	98.7	1.3[b]	98.7	1.3
Ceftazidime	>32	>32	1.3	97.7	0.3	98.7
Levofloxacin	>4	>4	13.4	84.0	12.4	86.6
Gentamicin	4	>8	52.3	35.0	45.1	47.7
Amikacin	16	32	59.5	7.5	38.2	40.5
Tigecycline	0.5	1	99.3[b]	0.0[b]	93.8	0.7
Colistin	≤0.5	>8	—	—	79.7	20.3
Colistin-resistant *K. pneumoniae* (147)[g]						
Ceftazidime-avibactam	0.25	2	99.3	0.7[b]	99.3	0.7
Ceftazidime	>32	>32	38.1	60.5	38.1	61.9
Ceftriaxone	>8	>8	39.5	59.2	39.5	59.2
Cefepime	32	>64	33.3	63.6[c]	33.3	63.6
Piperacillin-tazobactam	32	>64	49.7	46.9	41.5	50.3
Meropenem	≤0.06	>8	58.5	41.5	58.5	33.3
Levofloxacin	>4	>4	43.5	53.7	42.9	56.5
Gentamicin	≤1	>8	69.4	27.2	63.9	30.6
Amikacin	2	32	78.2	6.8	66.0	21.8
Tigecycline	0.5	1	100.0[b]	0.0[b]	94.6	0.0
Ceftazidime-nonsusceptible *E. cloacae* (568)[h]						
Ceftazidime-avibactam	0.5	1	99.3	0.7[b]	99.3	0.7
Ceftriaxone	>8	>8	0.0	99.6	0.0	99.6
Cefepime	2	32	63.2	20.4[c]	40.8	26.4
Piperacillin-tazobactam	64	>64	21.8	36.5	14.7	78.2
Meropenem	≤0.06	0.5	93.0	4.9	95.1	1.6
Levofloxacin	≤0.12	>4	81.5	13.4	77.8	18.5
Gentamicin	≤1	>8	83.8	12.1	82.4	16.2
Amikacin	1	4	99.5	0.4	98.1	0.5
Tigecycline	0.25	2	98.4	0.0[e]	88.6	1.6
Colistin	≤0.5	>8	—	—	84.4	15.6
P. aeruginosa (5,486)						
Ceftazidime-avibactam	2	4	97.0	3.0[b]	97.0	3.0
Ceftazidime	2	32	84.7	11.2	84.7	15.3
Cefepime	2	16	85.9	5.6	85.9	14.1
Piperacillin-tazobactam	4	64	81.3	9.5	81.3	18.7
Meropenem	0.5	8	82.0	12.0	82.0	6.0
Levofloxacin	0.5	>4	74.8	18.6	65.7	25.2
Gentamicin	≤1	8	88.1	8.1	88.1	11.9
Amikacin	2	8	96.8	2.0	92.7	3.2
Colistin	1	2	99.7	0.1	99.9	0.1
MDR *P. aeruginosa* (1,024)						
Ceftazidime-avibactam	4	16	85.2	14.8[b]	85.2	14.8
Ceftazidime	16	>32	41.2	44.9	41.2	58.8
Cefepime	16	>16	40.7	25.7	40.7	59.3
Piperacillin-tazobactam	64	>64	30.1	38.3	30.1	69.9
Meropenem	8	>8	16.9	58.6	16.9	30.2
Levofloxacin	>4	>4	17.0	67.6	9.2	83.0
Gentamicin	4	>8	58.6	32.8	58.6	41.4
Amikacin	4	32	88.1	7.7	77.0	11.9
Colistin	1	2	99.5	0.2	99.8	0.2
XDR *P. aeruginosa* (465)						
Ceftazidime-avibactam	8	32	72.9	27.1[b]	72.9	27.1
Ceftazidime	32	>32	18.7	66.0	18.7	81.3
Cefepime	16	>16	15.9	41.9	15.9	84.1
Piperacillin-tazobactam	>64	>64	6.9	56.6	6.9	93.1
Meropenem	8	>8	0.0	79.8	0.0	49.7
Levofloxacin	>4	>4	4.1	80.0	2.2	95.9
Gentamicin	8	>8	42.2	49.0	42.2	57.8
Amikacin	8	>32	81.7	11.6	66.7	18.3
Colistin	1	2	99.6	0.2	99.8	0.2
P. aeruginosa nonsusceptible to ceftazidime, meropenem, and piperacillin-tazobactam (432)[i]						
Ceftazidime-avibactam	8	32	69.9	30.1[b]	69.9	30.1
Cefepime	16	>16	17.4	44.7	17.4	82.6
Levofloxacin	>4	>4	20.1	67.6	14.1	79.9
Gentamicin	4	>8	56.7	37.5	56.7	43.3
Amikacin	4	>32	83.8	10.6	73.1	16.2
Colistin	1	2	99.5	0.2	99.8	0.2
Acinetobacter spp. (797)						
Ceftazidime-avibactam	16	>32	—	—	—	—
Ceftazidime	16	>32	46.4	48.3	—	—
Cefepime	16	>16	43.5	46.8	—	—
Ampicillin-sulbactam	8	>32	53.8	32.0	—	—
Piperacillin-tazobactam	>64	>64	40.3	50.4	—	—
Meropenem	4	>8	49.8	48.4	49.8	45.0
Levofloxacin	>4	>4	44.9	52.9	44.2	55.1
Gentamicin	4	>8	55.0	40.0	55.0	45.0
Amikacin	4	>32	70.0	26.1	66.6	30.0
Colistin	1	2	94.5	5.5	94.5	5.5

[a] Criteria as published by CLSI and EUCAST
[b] Breakpoints from US FDA Package Insert
[c] Organisms include: *Citrobacter freundii* (39), *Enterobacter aerogenes* (31), *E. cloacae* (197), *Escherichia coli* (554), *Klebsiella oxytoca* (31), *K. pneumoniae* (598), *Morganella morganii* (123), *Proteus mirabilis* (267), *P. vulgaris* (5), *Providencia rettgeri* (12), *P. stuartii* (133), *Serratia marcescens* (80)
[d] Organisms include: *Citrobacter freundii* (3), *Enterobacter aerogenes* (1), *E. cloacae* (29), *Escherichia coli* (11), *Klebsiella oxytoca* (5), *K. pneumoniae* (234), *Morganella morganii* (11), *Proteus mirabilis* (9), *Providencia stuartii* (10), *Serratia marcescens* (13)
[e] Organisms include: *Citrobacter freundii* (2), *Enterobacter aerogenes* (8), *E. cloacae* (40), *Escherichia coli* (19), *Klebsiella oxytoca* (9), *K. pneumoniae* (294), *Proteus mirabilis* (1), *Providencia stuartii* (1), *Serratia marcescens* (18)
[f] According to CLSI criteria; ie, ≥2 mg/L
[g] According to EUCAST criteria; ie, ≥4 mg/L
[h] According to CLSI criteria; ie, ≥8 mg/L
[i] According to CLSI and EUCAST criteria

To obtain a PDF of this poster:
- Scan the QR code

OR
- Visit www.allergancongressposters.com/410342

Charges may apply.
No personal information is stored.

Allergan

European Congress of Clinical Microbiology and Infectious Diseases (ECCMID), April 22–25, 2017, Vienna, Austria