ECCMID 2017
Poster #P1748

Monitoring Echinocandin and Azole Susceptibility in a Global Collection of Invasive Fungal Isolates

M Castanheira, LM Deshpande, AP Davis, PR Rhomberg, MA Pfaller
JMI Laboratories, North Liberty, Iowa, USA

Contact information:
Mariana Castanheira, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

Amended Abstract

Background: Continuous monitoring of antifungal susceptibility patterns and understanding resistance mechanisms against these agents seems prudent after reports of breakthrough infections, emerging resistance mechanisms, and increasing prevalence of uncommon species associated with higher resistance. We evaluated the activity of 7 antifungals against 3,557 invasive yeasts and moulds collected worldwide, and we screened resistance mechanisms among *Candida* spp. (CANS) displaying elevated echinocandin MICs and azole-resistant *Candida albicans* (CA).

Materials/methods: Fungal isolates collected during 2014-2015 in 66 hospitals in 29 countries were susceptibility tested by CLSI broth microdilution methods. CLSI interpretive criteria (clinical breakpoints and epidemiological cutoff values [ECV]) were applied. CANS isolates displaying echinocandin MIC>ECV were sequenced for *fks* hotspot (HS) mutations using PCR/sequencing. Five CA isolates were submitted to quantitative RT-PCR for *Erg11*, *CDR1*, *CDR2*, and *MDR1* and to whole genome sequencing analysis for alterations in genes associated to azole resistance.

Results: Susceptibility rates for the most common CANS are displayed in the table. Among 20 CANS isolates screened for *fks* HS mutations, 11 *C. glabrata* (CGLA) and 2 CA (2 *fks1*HS1 S645P) displayed alterations. Among CGLA, 3 isolates had double substitutions with *fks1*HS1 F625S (n=3), S629P (n=3), *fks1*HS2 F659S/Y (n=4), and S663P (n=4) being the most common alterations. All azole-resistant CA belonged to novel and unrelated multilocus sequence types. One isolate displaying fluconazole resistance and elevated MICs for other azoles displayed Erg11 alterations (G450E and G464S) associated with azole resistance. Elevated expression of MDR1 or CDR2 was observed among 3 and 1 isolates, respectively. Polymorphisms on MMR1, TAC1, and UPC2 were noted, but the correlation with azole resistance is uncertain. *C. dubliniensis* (n=58) and *C. lusitaniae* (n=39) isolates displayed wild-type (WT) MICs for anidulafungin and micafungin based on ECV. The highest MIC results for fluconazole, voriconazole, and posaconazole against *C. neoformans var. grubii* (n=79) were 8, 0.12, and 0.25 mg/L, respectively. Among *Aspergillus fumigatus*, only 2 isolates displayed itraconazole MICs >4 mg/L and 1 of those isolates also displayed a voriconazole MIC at >8 mg/L. Other *Aspergillus* species were WT for mould-active azoles and caspofungin according to recently published ECVs (CLSI M59).

Conclusions: Echinocandin and azole resistance was uncommon among contemporary fungal isolates; however, genetic mechanisms encoding resistance to antifungal agents were observed among CANS isolates, showing that resistance can emerge and should be monitored.

Organism (no. tested)	% susceptible using CLSI clinical breakpoints				
	Anidulafungin	Caspofungin	Micafungin	Fluconazole	Voriconazole
C. albicans (1,310)	99.9	99.8	99.8	99.6	99.9
C. glabrata (514)	95.9	96.9	97.5	NA[a]	NA
C. parapsilosis (417)	88.7	100.0	100.0	95.7	96.4
C. tropicalis (264)	100.0	100.0	100.0	96.2	97.0
C. krusei (93)	100.0	100.0	100.0	NA	100.0

[a] NA = not available

Introduction

- Invasive fungal infections (IFIs) are associated with high morbidity and mortality rates and elevated hospital care costs
- *Candida* and *Aspergillus* species are among the most frequent causes of IFI, and although isolates displaying resistance to clinically available antifungal agents are still uncommon, these isolates are increasingly reported worldwide
- Monitoring antifungal susceptibility patterns and resistance mechanisms to clinically used antifungal agents is important
- Echinocandin resistance among *Candida* isolates is encoded by alterations in the target of these antifungal agents, the 1,3 β-D-glucan synthase (GS) encoded by *fks*
- Azole resistance among *Candida* and *Aspergillus* isolates can be encoded by target alterations in the ergosterol synthesis pathway and increased efflux of the antifungal molecules
- In this study, we evaluated the susceptibility patterns against antifungal agents tested using the CLSI reference broth microdilution method against 3,557 fungal clinical isolates collected during 2014-2015 in 29 countries
- Additionally, we investigated resistance mechanisms among *Candida* spp. isolates displaying non-wild-type MIC values for echinocandins and *Candida albicans* displaying elevated fluconazole MIC values

Materials and Methods

- A total of 3,557 fungal consecutive nonduplicated clinical isolates were collected as part of a global surveillance initiative in 66 hospitals located in 29 countries (Figure 1)
- Susceptibility testing was performed for anidulafungin, caspofungin, micafungin, fluconazole, posaconazole, voriconazole, and amphotericin B using the CLSI reference broth microdilution method
- CLSI clinical breakpoints were used for the most common species of *Candida*, and CLSI M59 recently published epidemiologic cutoff values (ECVs) were applied for less common species of *Candida* and *Aspergillus* species
- Quality control was performed as recommended in CLSI documents M27-A3 and M38-A2 and all results were within established ranges
- *Candida* spp. isolates with MIC values higher than the ECV for the echinocandin compounds were screened for *fks* hotspot mutations as previously described
- Five *C. albicans* isolates displaying fluconazole MIC values ≥4 mg/L were submitted to whole genome sequencing on a MiSeq Sequencer (Illumina, San Diego, CA, USA). *Erg11*, *Erg3*, *UPC2*, *MDR1* including the promoter region, *MRR1*, and *TAC1* were compared to sequences from *C. albicans* ATCC 90028. *CDR1* and *CDR2* were compared to *C. albicans* ATCC 10261 sequences
- The expression of *Erg11, CDR1, CDR2*, and *MDR1* was determined by quantitative real-time PCR (qRT-PCR) using high quality DNA-free mRNA preparations. Relative quantification of target genes was performed in triplicate by normalization to an endogenous reference gene (18S). Transcription levels were considered significantly different if a 10-fold difference was noted compared with *C. albicans* ATCC 90028

Figure 1 Species and infection sources distributions of 3,557 invasive fungal isolates collected as part of a global surveillance initiative in 66 hospitals located in 29 countries during 2014–2015

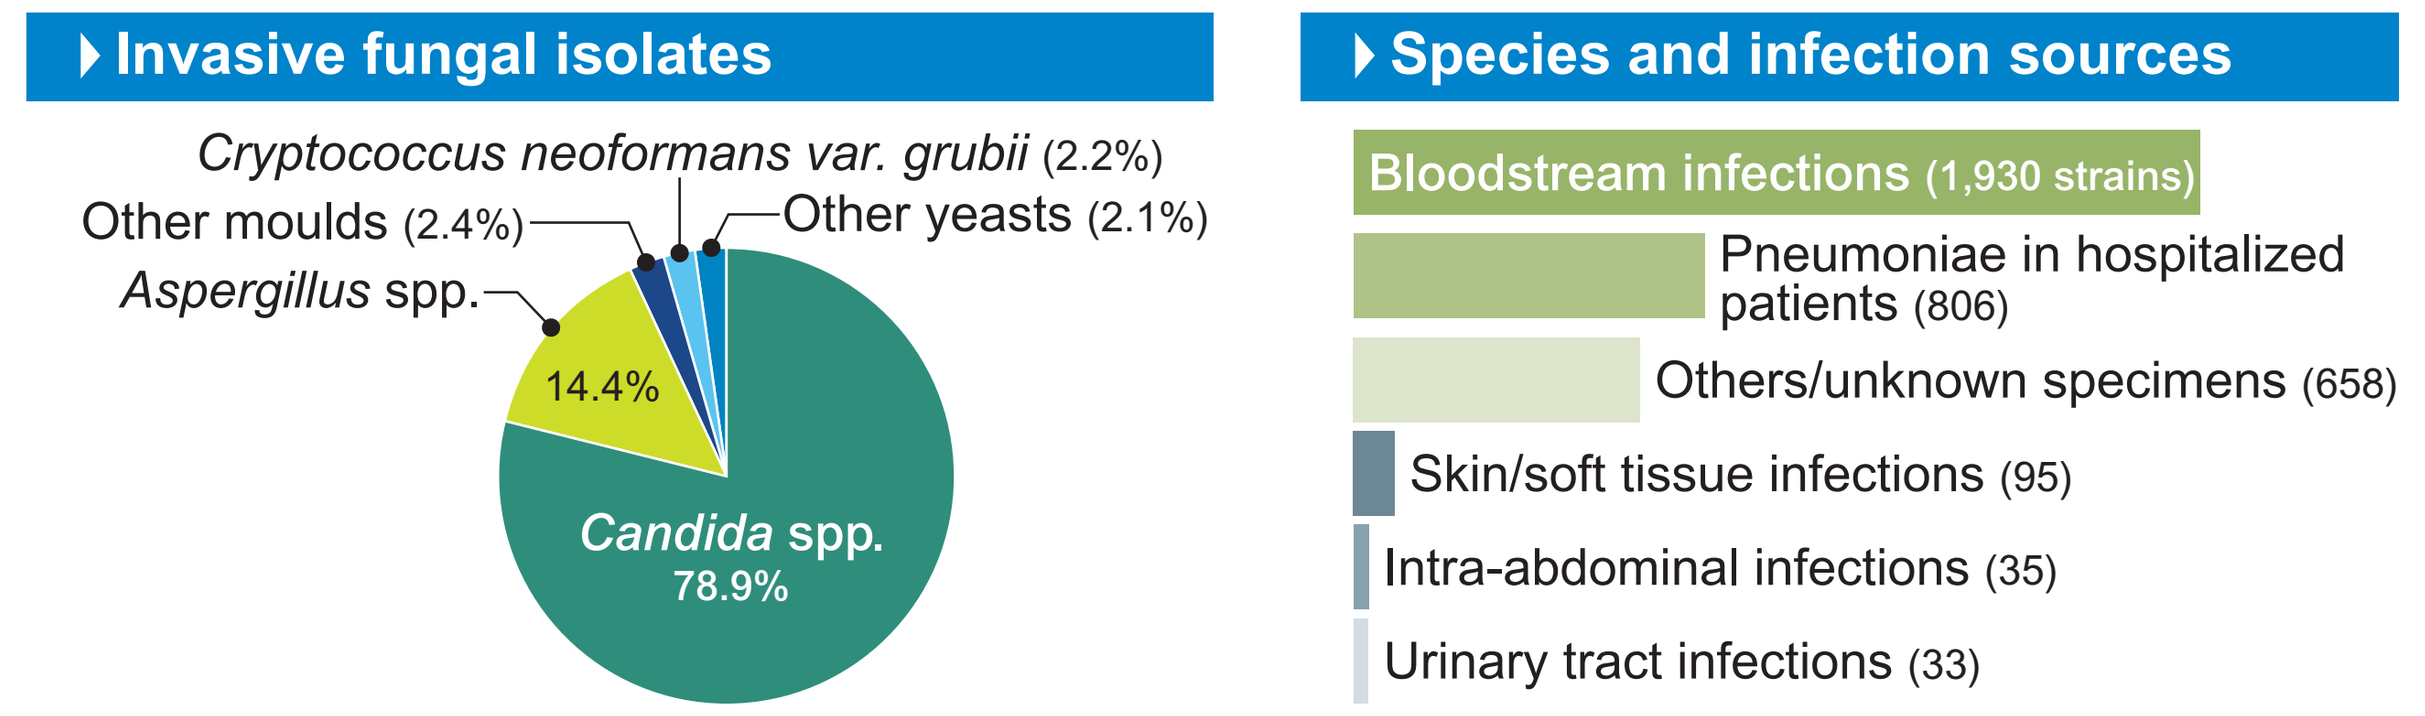
Invasive fungal isolates
Cryptococcus neoformans var. grubii (2.2%)
Other moulds (2.4%)
Other yeasts (2.1%)
Aspergillus spp.
14.4%
Candida spp.
78.9%
Species and infection sources
Bloodstream infections (1,930 strains)
Pneumoniae in hospitalized patients (806)
Others/unknown specimens (658)
Skin/soft tissue infections (95)
Intra-abdominal infections (35)
Urinary tract infections (33)

Results

- Figure 2 summarizes the CLSI broth microdilution susceptibility results against echinocandins, azoles, and amphotericin B for the species for which clinical breakpoints and/or ECVs have been established in CLSI documents M27-S4 and M59
 - Anidulafungin, caspofungin, and micafungin displayed good activity against the 9 *Candida* species
 - Resistance to the fluconazole was observed among 0.1% of *C. albicans*, 3.8% of *C. parapsilosis*, 2.7% of *C. tropicalis*, and 8.0% of *C. glabrata*
 - Voriconazole resistance was observed among <0.1% of *C. albicans*, 0.7% of *C. parapsilosis*, and 2.3% of *C. tropicalis*
 - All *C. krusei* isolates were susceptible to voriconazole
- Recently published CLSI ECVs (M59 document) were applied for the *Candida* species-antifungal combinations that do not have clinical breakpoints
 - All *C. lusitaneae* and *C. dubliniensis* isolates were considered wild type for anidulafungin and micafungin
 - All *C. albicans*, *C. glabrata*, *C. parapsilosis*, and *C. tropicalis* isolates were wild type to amphotericin B (Figure 2)
- ECVs for various *Aspergillus* species were applied for *sensu stricto* of *A. fumigatus*, *A. niger*, *and A. terreus* isolates (Figure 2)
 - Two (0.3%) *A. fumigatus* isolates displayed nonwild-type itraconazole MIC results
- Azoles were very active against *Cryptococcus neoformans var. grubii*, exhibiting $MIC_{50/90}$ values of 2/4, 0.03/0.06, and 0.25/0.25 µg/mL for fluconazole, voriconazole, and posaconazole, respectively (data not shown)
- Among 15 *C. glabrata* isolates displaying nonwild-type results for anidulafungin and micafungin MIC values, 11 (73.3% of the isolates screened) harbored *fks* HS alterations
 - Mutations included: *fks2* HS1 S663P (4 isolates), *fks1* HS1 S629P (3 isolates), *fks1* HS1 F625S (3 isolates) or *fks2* HS1 F659S/Y (3 isolates)
 - Three isolates carried double mutations that were either *fks1* HS1 S629P/*fks2* HS1 S663P or *fks1* HS1 F625S/*fks2* HS1 F659Y

Figure 2 Activity of antifungal agents tested against main organism groups using CLSI reference broth microdilution method with CLSI clinical breakpoints (M27-S4 document) and epidemiological cutoff values (M59 document) applied

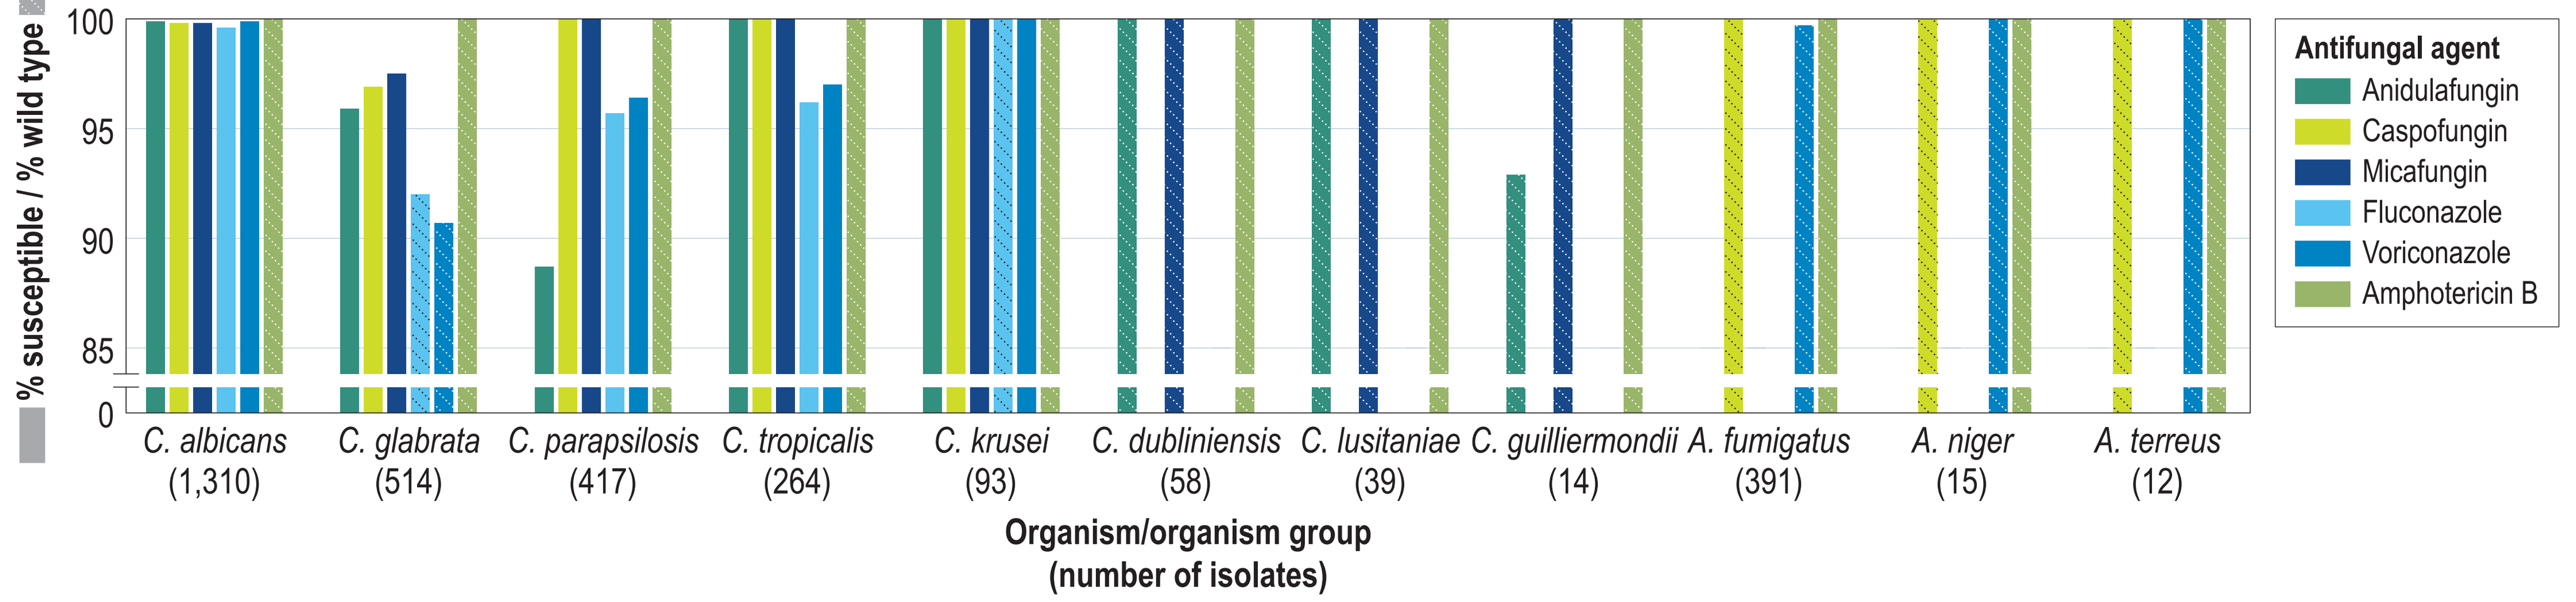
% susceptible / % wild type
100
95
90
85
0
C. albicans (1,310)
C. glabrata (514)
C. parapsilosis (417)
C. tropicalis (264)
C. krusei (93)
C. dubliniensis (58)
C. lusitaniae (39)
C. guilliermondii (14)
A. fumigatus (391)
A. niger (15)
A. terreus (12)
Organism/organism group (number of isolates)
Antifungal agent
Anidulafungin
Caspofungin
Micafungin
Fluconazole
Voriconazole
Amphotericin B

- *C. glabrata* isolates carrying *fks* HS alterations displayed greater MIC values for all 3 echinocandins when compared to those not carrying mutations (Figure 3)
- Two out of 3 *C. albicans* isolates tested harbored mutations encoding a *fks*1 HS1 alteration S645P
- One *C. tropicalis* isolate did not harbor mutations on *fks1* HSs
- Five fluconazole nonsusceptible *C. albicans* isolates were genetically unrelated
- Gain of function homozygous mutations on *Erg11* was noted in 1 isolate resistant to fluconazole and voriconazole (Table 1)
- One isolate had heterozygous gain of function mutations on *Erg3* and a slightly elevated expression of *MDR1* associated with an *MDR* promoter allele mutation previously described
- Elevated expression of *MDR1* was observed among 3 fluconazole-nonsusceptible isolates, and 1 isolate displayed elevated expression of CDR2

Table 1 Results for 5 *C. albicans* isolates displaying fluconazole MIC values ≥4 mg/L

State, Country	MLST	MIC (mg/L)[a]		Gain of function alterations[b]			Relative quantification of the mRNA expression levels			
		FLU	VOR	*Erg11*	*Erg3*	MDR promoter	*Erg11*	*MDR1*	*CDR1*	*CDR2*
Romania	3212	4	0.015		A351V, A353T		0.52	11.21	0.23	2.13
France	3213	8	0.015				0.83	86.87	0.48	5.33
United Kingdom	3216	8	0.06				1.15	200.64	2.55	0.92
Washington, USA	3214	8	0.12				0.80	0.58	1.84	27.74
New York, USA	3215	64	1	G450E, G464S		A/A	1.07	0.28	1.15	8.24

[a] FLU = fluconazole; VOR = voriconazole
[b] Only gain of function mutations previously associated with azole resistance are listed

Figure 3 Distribution of *C. glabrata* isolates harbouring *fks* HS mutations and those with wild-type *fks* HS according to echinocandin MIC

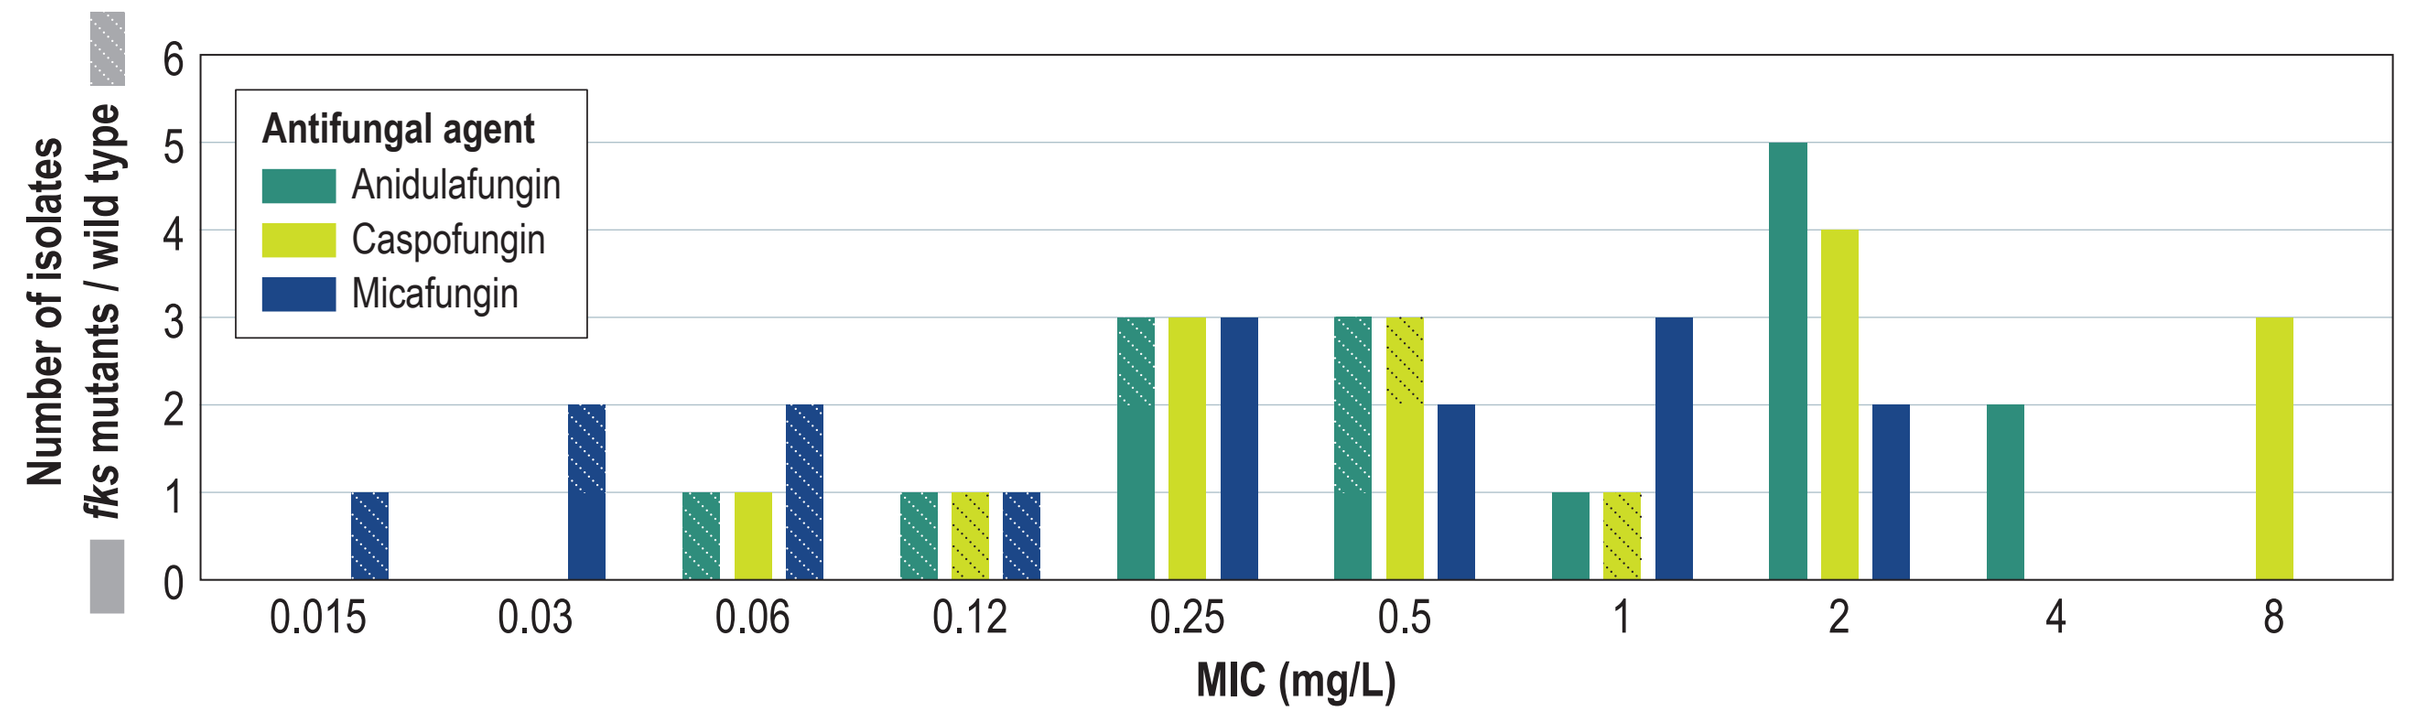
Number of isolates
fks mutants / wild type
0
1
2
3
4
5
6
Antifungal agent
Anidulafungin
Caspofungin
Micafungin
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
MIC (mg/L)

Isolates that displayed an echinocandin MIC>ECV were selected. Mutations were *fks2* HS1 S663P (2 isolates), *fks1* HS1 S629P (1 isolate), *fks1* HS1 F625S (2 isolates), *fks2* HS1 F659Y (2 isolates), and *fks2* HS1 P667T (1 isolate). Double mutations *fks1* HS1 S629P/*fks2* HS1 S663P (2 isolates) and *fks1* HS1 F625S/*fks2* HS1 F659Y (1 isolate) were also observed

Conclusions

- Echinocandins, azoles, and amphotericin B exhibited good activity against common *Candida* and *Aspergillus* species evaluated
- Azoles were very active against *C. neoformans var. grubii* isolates tested
- Most isolates carrying *fks* HS mutations displayed resistance to 1 or more of the echinocandins
- Isolates exhibiting MIC values above the ECV but below the resistant breakpoints did not carry *fks* HS alterations
- We observed diverse resistance mechanisms among 4 azole-resistant *C. albicans*
- Erg11 and Erg3 alterations were detected in 1 isolate each
- Three isolates had overexpression of efflux pumps
- Monitoring of antifungal susceptibility patterns and understanding resistance mechanisms against antifungal agents seems prudent due to increasing reports of breakthrough infections, increasing prevalence of uncommon species refractory to clinically available antifungal agents, and emerging resistance mechanisms

Acknowledgements

This study was performed by JMI Laboratories and supported by Pfizer, which included funding for services related to preparing this poster.

The authors would like to acknowledge the contribution of the SENTRY Antifungal Surveillance Program participants and the technical assistance of SA Messer and RD Dietrich performing the susceptibility testing assays.

References

Arendrup MC (2014). Update on antifungal resistance in *Aspergillus* and *Candida*. *Clin Microbiol Infect* 20 Suppl 6: 42-48.

Bruzual I, Kumamoto CA (2011). An *MDR1* promoter allele with higher promoter activity is common in clinically isolated strains of *Candida albicans*. *Mol Genet Genomics* 286: 347-357.

Clinical and Laboratory Standards Institute (2012). *M27-S4. Reference method for broth dilution antifungal susceptibility testing of yeasts: 4th informational supplement*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2016). *M59. Epidemiological cutoff values for antifungal susceptibility testing, 1st edition*. Wayne, PA: CLSI.

Garnaud C, Botterel F, Sertour N, et al. (2015). Next-generation sequencing offers new insights into the resistance of *Candida* spp. to echinocandins and azoles. *J Antimicrob Chemother* 70: 2556-2565.

Pfeiffer CD, Garcia-Effron G, Zaas AK, Perfect JR, Perlin DS, Alexander BD (2010). Breakthrough invasive candidiasis in patients on micafungin. *J Clin Microbiol* 48: 2373-2380.

Sanglard D, Odds FC (2002). Resistance of *Candida* species to antifungal agents: molecular mechanisms and clinical consequences. *Lancet Infect Dis* 2: 73-85.

Shields RK, Nguyen MH, Press EG, et al. (2012). The presence of an *FKS* mutation rather than minimum inhibitory concentration is an independent risk factor for failure of echinocandin therapy among patients with invasive candidiasis due to *Candida glabrata*. *Antimicrob Agents Chemother* 56: 4862-4869.