ECCMID 2017
Poster #P1280

Activity of Plazomicin and Comparator Agents Tested against Recent Clinical Isolates Collected in Asia-Pacific, Europe, and Latin America

M Castanheira, JM Streit, LM Deshpande, RE Mendes, RK Flamm
JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Mariana Castanheira, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

Amended Abstract

Background: Plazomicin is a next-generation aminoglycoside that was developed to overcome common aminoglycoside resistance mechanisms and is under development for the treatment of patients with serious bacterial infections due to MDR *Enterobacteriaceae*, including ESBL-producing and carbapenem-resistant *Enterobacteriaceae* (CRE). We evaluated the activity of plazomicin and comparators tested against 3,830 clinical isolates collected in hospitals from the Asia-Pacific (APAC) region, Europe, and Latin America (LATAM) during 2015.

Materials/methods: A total of 3,375 *Enterobacteriaceae*, 252 gram-positive cocci, 101 *Pseudomonas aeruginosa* (PSA), and 102 *Acinetobacter* spp. were collected in hospitals in APAC (n=851), Europe (n=2,365), and LATAM (n=614). Isolates were susceptibility (S) tested using the reference broth microdilution method. CLSI and EUCAST interpretive criteria were applied. CRE isolates were screened for carbapenemase encoding genes by PCR/sequencing.

Results: Overall, plazomicin ($MIC_{50/90}$, 0.5/1 mg/L) inhibited 90.9% and 95.8% of *Enterobacteriaceae* at ≤1 and ≤2 mg/L, respectively. Plazomicin displayed good activity against the main *Enterobacteriaceae* species, including *E. coli* ($MIC_{50/90}$, 0.5/1 mg/L), *K. pneumoniae* ($MIC_{50/90}$, 0.25/0.5 mg/L), and *E. cloacae* ($MIC_{50/90}$, 0.25/0.5 mg/L). Against ESBL-phenotype isolates that were not CRE ($MIC_{50/90}$, 0.25/1 mg/L), plazomicin inhibited 94.1% at ≤1 mg/L and 95.8% at ≤2 mg/L. Among 187 CRE isolates, 52 carried bla_{KPC-2}, 42 bla_{KPC-3}, 41 bla_{NDM-1}, 37 bla_{OXA-48}-like, and 1 bla_{VIM-1}. Plazomicin ($MIC_{50/90}$, 0.25/>128 mg/L for CRE) inhibited 83/94 isolates carrying bla_{KPC}, 32/30 isolates carrying bla_{OXA-48}-like, and 11/42 isolates carrying metallo-beta-lactamase genes. The activity of plazomicin ($MIC_{50/90}$, 4/8 mg/L) against PSA was similar to the activity of amikacin ($MIC_{50/90}$, 2/16 mg/L) and lower than the activity of gentamicin ($MIC_{50/90}$, 1/8 mg/L) against these isolates. Plazomicin demonstrated good activity against coagulase-negative staphylococci (CoNS; $MIC_{50/90}$, 0.25/0.5 mg/L) and *S. aureus* ($MIC_{50/90}$, 0.5/1 mg/L). Plazomicin activity was limited against *Acinetobacter* spp. ($MIC_{50/90}$, 8/>128 mg/L), *Enterococcus* spp. ($MIC_{50/90}$, 32/64 mg/L), and *S. pneumoniae* ($MIC_{50/90}$, 32/64 mg/L). The activity of plazomicin and other aminoglycosides displayed slight variability according to geographic region (Table).

Conclusions: Plazomicin was active against *Enterobacteriaceae* isolates, including isolates displaying an ESBL phenotype and carrying genes encoding serine-carbapenemases. These data support the current development plan for plazomicin to treat serious infections caused by resistant *Enterobacteriaceae* where treatment options are limited.

Organism/Group (no. tested)	Asia-Pacific PLZ	Asia-Pacific AMK	Asia-Pacific GEN	Europe PLZ	Europe AMK	Europe GEN	Latin America PLZ	Latin America AMK	Latin America GEN
	MIC_{50}/MIC_{90} in mg/L								
Enterobacteriaceae (3,375)	0.5/1	2/4	0.5/>8	0.5/1	2/8	0.5/>8	0.5/2	2/8	0.5/>8
ESBL-phenotype non-CRE (663)	0.5/1	2/8	1/>8	0.25/1	2/8	1/>8	0.5/1	2/8	>8/>8
CRE (187)	0.25/1	4/16	0.5/>8	0.25/>128	16/>32	>8/>8	0.25/>128	16/>32	>8/>8
P. aeruginosa (101)	4/8	4/4	1/4	4/8	2/16	1/>8	4/16	4/16	1/>8
S. aureus (65)	0.5/1	—	≤1/>8	0.5/1	—	≤1/≤1	0.5/0.5	—	≤1/>8
CoNS (64)	0.25/0.5	—	≤1/>8	0.25/0.5	—	≤1/>8	0.25/0.5	—	8/>8

PLZ, plazomicin; AMK, amikacin; GEN, gentamicin

Introduction

- Aminoglycosides are broad spectrum antimicrobial agents that bind to the 16S ribosomal RNA leading to inhibition of polypeptide synthesis and subsequent cell death
- Resistance to aminoglycosides may occur due to a variety of different resistance mechanisms
- The main mechanism in gram-positive and -negative bacteria is the enzymatic modification and inactivation of the aminoglycosides, mediated by aminoglycoside modifying enzymes (AME)
- Plazomicin is a semi-synthetic aminoglycoside derived from sisomicin and contains structural modifications that allow it to retain activity in the presence of AMEs
- Plazomicin is very active against *Enterobacteriaceae*, some *Pseudomonas aeruginosa*, and *Staphylococcus* spp., including methicillin-resistant (MRSA) isolates
- In this study, we evaluated the activity of plazomicin tested against a collection of 3,830 clinical isolates collected in Asia-Pacific, Europe, and Latin America during 2015 that comprised 3,375 *Enterobacteriaceae*, including molecular characterized carbapenem-resistant (CRE) isolates, 252 gram-positive cocci, 101 *P. aeruginosa,* and 102 *Acinetobacter* spp.

Materials and Methods

- A total of 3,830 bacterial isolates deemed cause of infection limited to 1 per patient episode were included in the study
- Isolates were collected during 2015 from 59 hospitals in Asia-Pacific (n=851), Europe (n=2,365), and Latin America (n=614) and included 3,375 *Enterobacteriaceae*, 252 gram-positive cocci, 101 *Pseudomonas aeruginosa*, and 102 *Acinetobacter* spp.
- Species identification was confirmed when necessary by matrix-assisted laser desorption ionization-time of flight mass spectrometry (MALDI-TOF MS)
- Isolates were susceptibility tested against plazomicin and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI)
- Quality control (QC) was performed according to CLSI guidelines (M100-S27) and all QC MIC results were within acceptable ranges as published in CLSI documents
- Categorical interpretations for all comparator agents were those found in CLSI criteria in M100-S27 (2017), EUCAST breakpoint tables (version 7.0, January 2017), or United States Food and Drug Administration (US FDA) package insert (tigecycline)
- ESBL-phenotype criterion was applied for *Escherichia coli*, *Klebsiella pneumoniae*, *K. oxytoca*, and *P. mirabilis* displaying an MIC value ≥2 mg/L for ceftriaxone, ceftazidime, or aztreonam (M100-S27)
- Carbapenem-resistant *Enterobacteriaceae* (CRE) was defined as any isolate exhibiting doripenem, imipenem, and/or meropenem MIC values at ≥2 mg/L
 - *Proteus mirabilis* and indole-positive *Proteeae* were categorized as CRE if meropenem or doripenem MIC values at ≥2 mg/L due to intrinsically elevated imipenem MIC values
- CRE were screened for acquired carbapenemase encoding genes by PCR using custom primers
 - Amplicons were sequenced on both strands and nucleotide sequences obtained were analyzed using the Lasergene® software package (DNAStar; Madison, Wisconsin, USA) and compared to available sequences via NCBI BLAST search (http://www.ncbi.nlm.nih.gov/blast/)

Results

- Plazomicin ($MIC_{50/90}$, 0.5/1 mg/L) inhibited 95.8% at ≤2 mg/L and 97.7% at ≤4 mg/L of 3,375 *Enterobacteriaceae* isolates collected in all regions (Figure 1)
- The activity of plazomicin was similar in all 3 regions, MIC_{50} values were 0.5 mg/L for all 3 regions, and MIC_{90} values were 1 mg/L (Asia-Pacific and Europe) or 2 mg/L (Latin America; Figure 1)
- Against *Enterobacteriaceae*, plazomicin (MIC_{90}, 1 mg/L) was more active than amikacin (MIC_{90}, 4 mg/L), gentamicin (MIC_{90}, >8 mg/L), and tobramycin (MIC_{90}, >8 mg/L; Table 1)
- Plazomicin was very active against ESBL-phenotype isolates, displaying no resistance to the carbapenems (ESBL non-CRE; $MIC_{50/90}$, 0.25/1 mg/L) and inhibiting 95.8% and 96.8% at ≤2 and ≤4 mg/L, respectively (Figure 1)
- A total of 187 CRE isolates were observed in this collection and plazomicin ($MIC_{50/90}$, 0.25/>128 mg/L) was considerably more active against these isolates when compared to amikacin ($MIC_{50/90}$, 16/>32 mg/L), gentamicin, or tobramycin ($MIC_{50/90}$, >8/>8 mg/L for both; Table 1)
- The majority of CRE isolates carried bla_{KPC} (94 isolates, 52 bla_{KPC-2}, and 42 bla_{KPC-3}; Figure 2) and plazomicin ($MIC_{50/90}$, 0.25/128 mg/L) displayed greater activity against these isolates when compared to amikacin ($MIC_{50/90}$, 16/>32 mg/L), gentamicin ($MIC_{50/90}$, 2/>8 mg/L) or tobramycin ($MIC_{50/90}$, >8/>8 mg/L; Table 2)
- A total of 30 isolates from Europe harboured bla_{OXA-48}, 41 isolates carried bla_{NDM-1}, 7 harboured $bla_{OXA-232}$, and bla_{VIM-1} was detected in 1 isolate (Figure 2)
- The activity of plazomicin and all other aminoglycosides (Table 2) was limited against NDM-1-producing organisms, but plazomicin ($MIC_{50/90}$, 0.25/128 mg/L) was more active against OXA-48-producers when compared to amikacin ($MIC_{50/90}$, 4/>32 mg/L) or gentamicin ($MIC_{50/90}$, >8/>8 mg/L)
- Plazomicin ($MIC_{50/90}$, 4/8 mg/L) activity was similar to the activity of amikacin ($MIC_{50/90}$, 2/16 mg/L) and four-fold lower than that of gentamicin ($MIC_{50/90}$, 1/8 mg/L) against *P. aeruginosa* isolates
- Plazomicin demonstrated activity against *Staphylococcus* spp. isolates ($MIC_{50/90}$, 0.5/1 mg/L; Figure 1), including coagulase-negative staphylococci ($MIC_{50/90}$, 0.25/0.5 mg/L) and *S. aureus* ($MIC_{50/90}$, 0.5/1 mg/L; data not shown)
- The activity of plazomicin was limited against *Acinetobacter* spp. ($MIC_{50/90}$, 8/>128 mg/L), *Enterococcus* spp. ($MIC_{50/90}$, 32/64 mg/L), and *S. pneumoniae* ($MIC_{50/90}$, 32/64 mg/L) isolates (data not shown)

Conclusions

- Plazomicin was active against *Enterobacteriaceae* isolates, including resistance groups such as cephalosporin-resistant isolates (ESBL non-CRE) and CRE
- The *in vitro* activity of plazomicin was greater compared to the activity of aminoglycosides against cephalosporin-resistant isolates and CRE, including isolates producing KPC or OXA-48
- Plazomicin displayed activity against *Staphylococcus* spp. and some *P. aeruginosa*, but its activity was limited against *Acinetobacter* spp., *Enterococcus* spp., and *S. pneumoniae*
- Plazomicin seems to be a valuable option for the treatment of infections caused by *Enterobacteriaceae* isolates, including isolates that may produce ESBLs and those carrying serine-carbapenemases

Acknowledgements

This study was performed by JMI Laboratories and supported by Achaogen, which included funding for services related to preparing this poster.

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 7.0, January 2017. Available at http://www.eucast.org/clinical_breakpoints/. Accessed January 2017.

Walkty A, Adam H, Baxter M, et al. (2014). In vitro activity of plazomicin against 5,015 gram-negative and gram-positive clinical isolates obtained from patients in Canadian hospitals as part of the CANWARD study, 2011-2012. *Antimicrob Agents Chemother* 58: 2554-2563.

Zhanel GG, Lawson CD, Zelenitsky S, et al. (2012). Comparison of the next-generation aminoglycoside plazomicin to gentamicin, tobramycin and amikacin. *Expert Rev Anti Infect Ther* 10: 459-473.

Figure 1 Activity of plazomicin against main organism groups for geographic regions evaluated

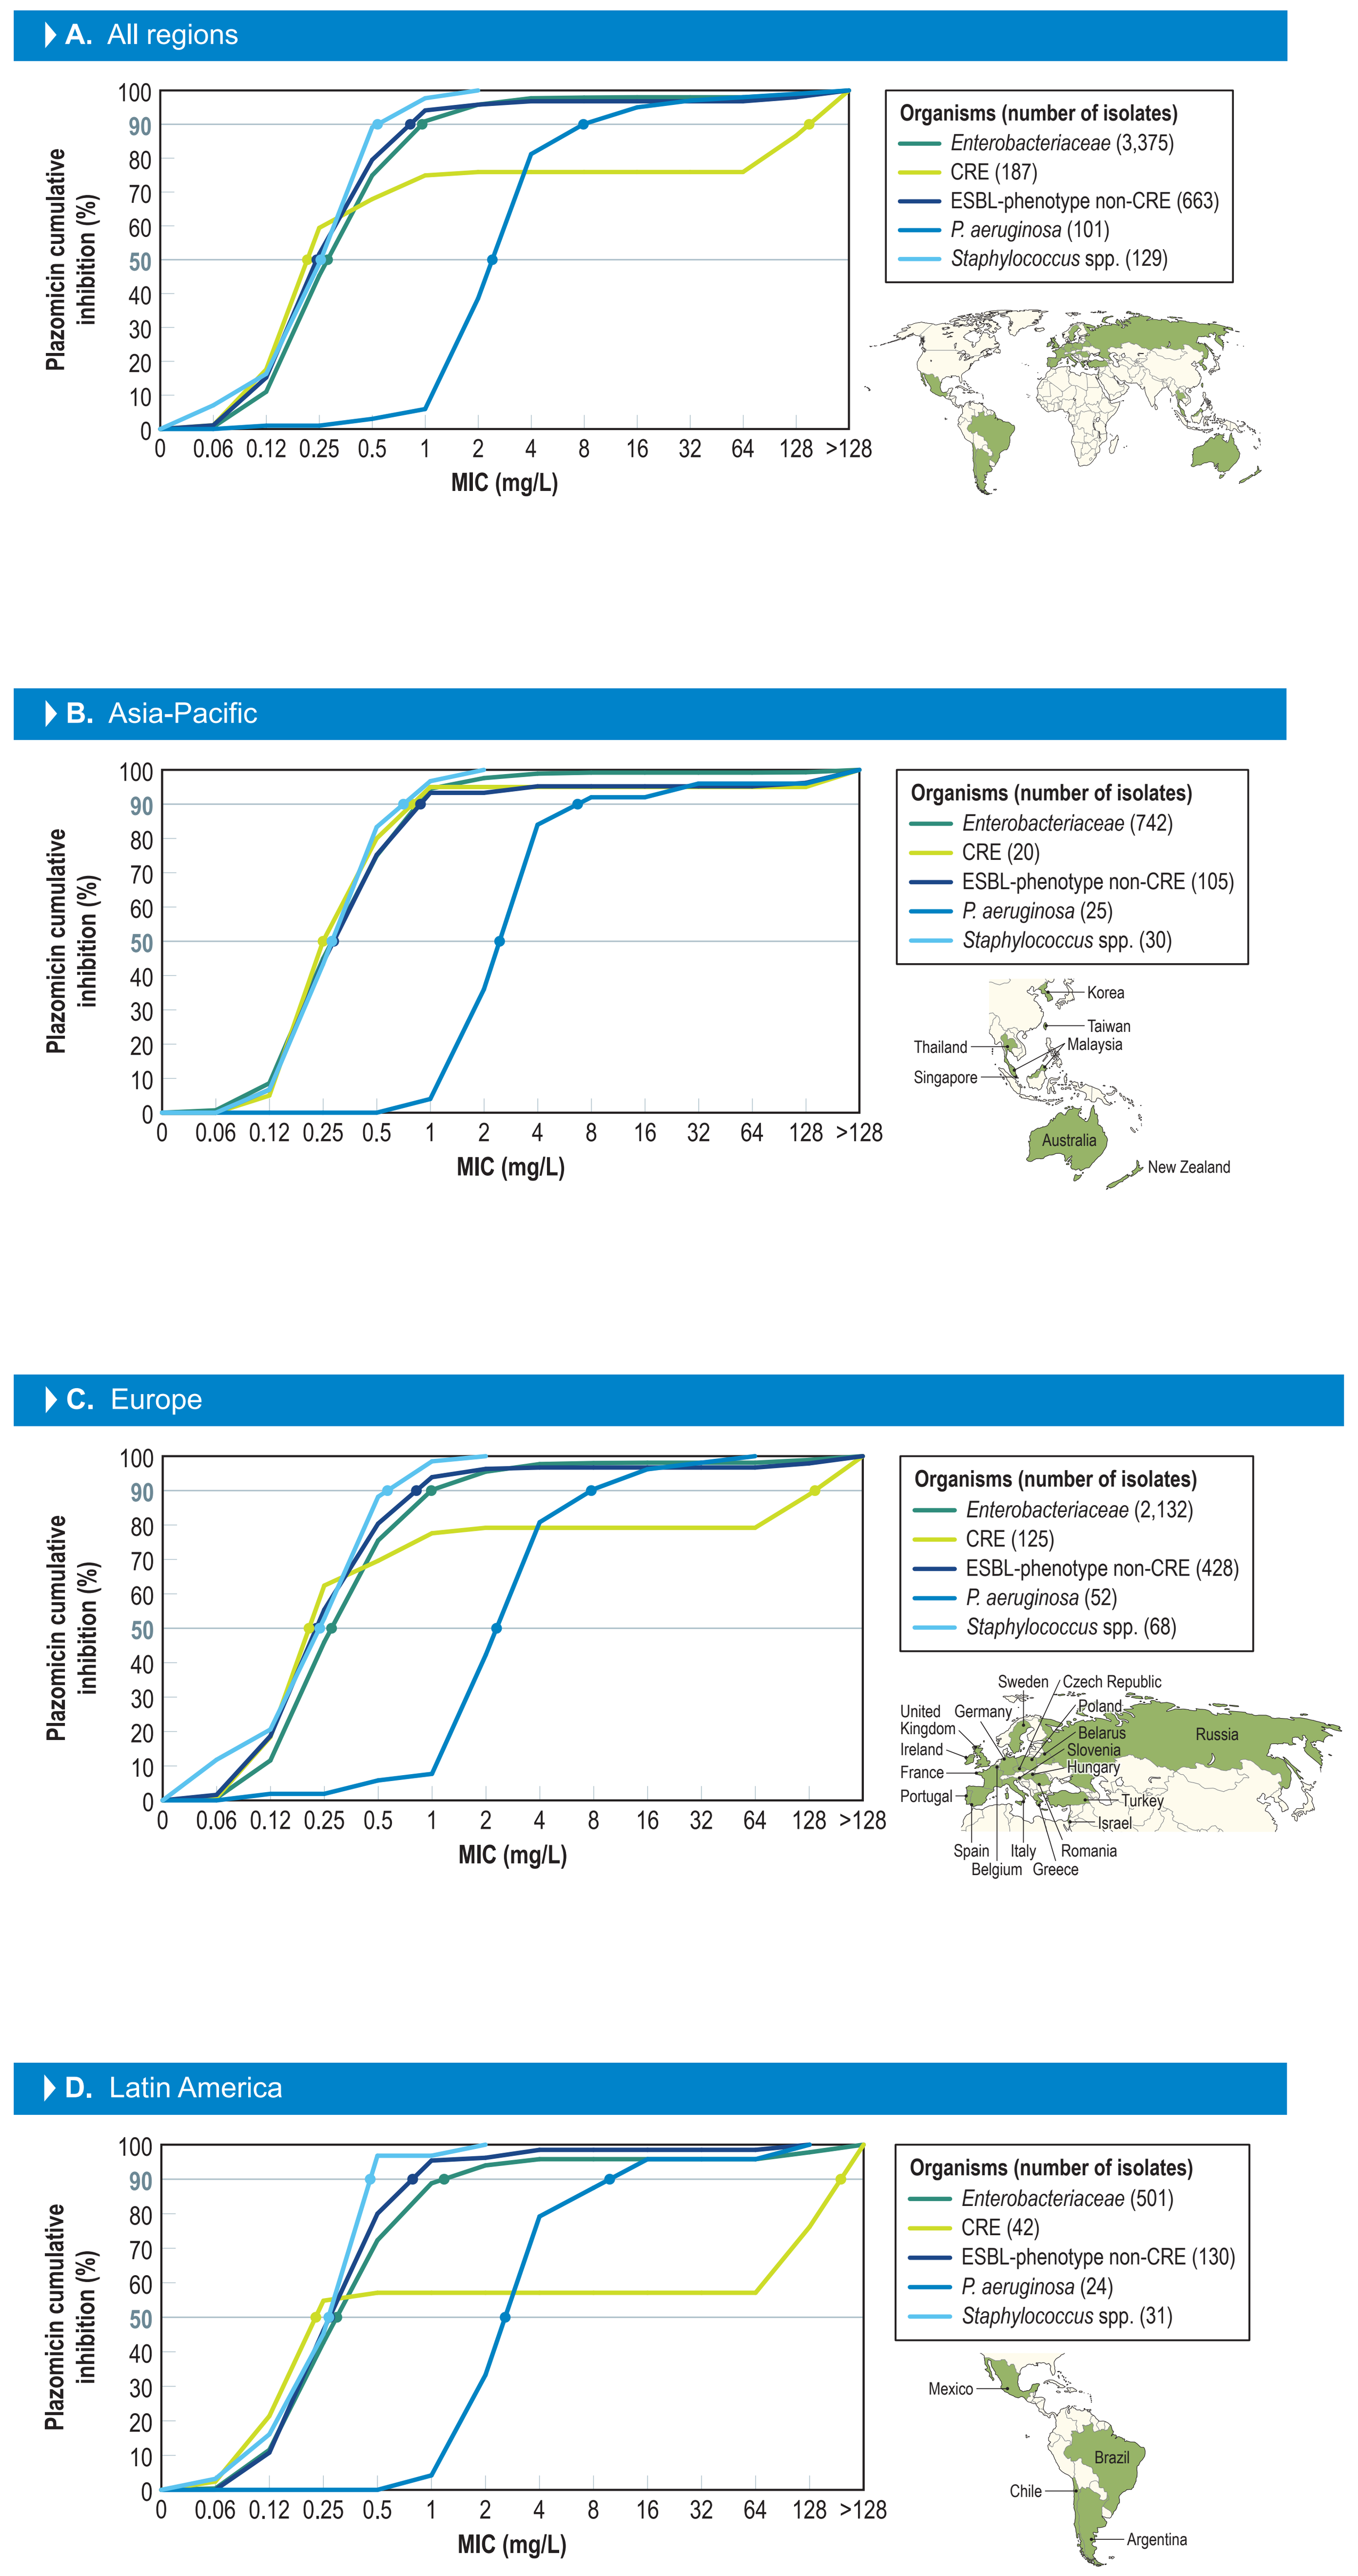
A. All regions
Organisms (number of isolates)
Enterobacteriaceae (3,375)
CRE (187)
ESBL-phenotype non-CRE (663)
P. aeruginosa (101)
Staphylococcus spp. (129)
B. Asia-Pacific
Organisms (number of isolates)
Enterobacteriaceae (742)
CRE (20)
ESBL-phenotype non-CRE (105)
P. aeruginosa (25)
Staphylococcus spp. (30)
Korea
Taiwan
Malaysia
Thailand
Singapore
Australia
New Zealand
C. Europe
Organisms (number of isolates)
Enterobacteriaceae (2,132)
CRE (125)
ESBL-phenotype non-CRE (428)
P. aeruginosa (52)
Staphylococcus spp. (68)
Sweden
Czech Republic
Poland
United Kingdom
Germany
Ireland
Belarus
Slovenia
Hungary
France
Portugal
Turkey
Israel
Spain
Italy
Romania
Belgium
Greece
Russia
D. Latin America
Organisms (number of isolates)
Enterobacteriaceae (501)
CRE (42)
ESBL-phenotype non-CRE (130)
P. aeruginosa (24)
Staphylococcus spp. (31)
Mexico
Brazil
Chile
Argentina
Plazomicin cumulative inhibition (%)
MIC (mg/L)
0
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
128
>128

Figure 2 Distribution of CREs and carbapenemase genes by region

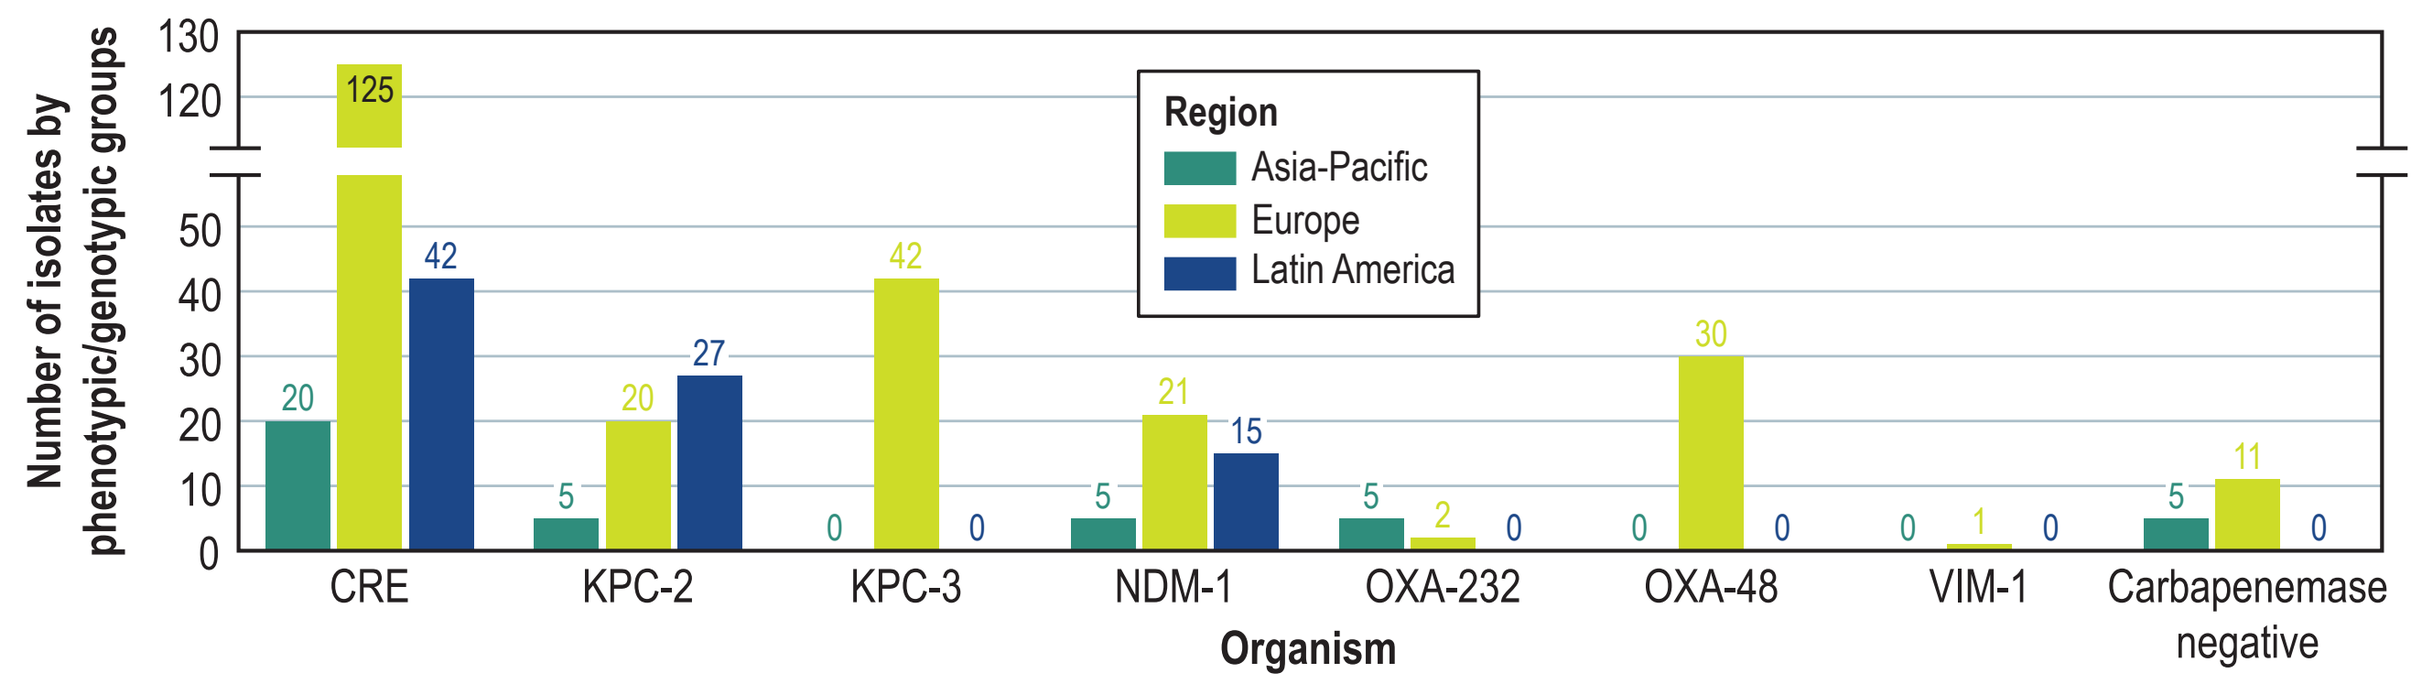
Number of isolates by phenotypic/genotypic groups
Region
Asia-Pacific
Europe
Latin America
CRE
KPC-2
KPC-3
NDM-1
OXA-232
OXA-48
VIM-1
Carbapenemase negative
Organism

Table 1 Activity of plazomicin and comparator agents against main bacterial species analyzed

Organism antimicrobial agent	Overall	Asia-Pacific	Europe	Latin America
	Region (no. tested) $MIC_{50/90}$ (mg/L)			
Enterobacteriaceae	3,375	742	2,132	501
Plazomicin	0.5 / 1	0.5 / 1	0.5 / 1	0.5 / 2
Amikacin	2 / 4	2 / 4	2 / 8	2 / 8
Gentamicin	0.5 / >8	0.5 / >8	0.5 / >8	0.5 / >8
Tobramycin	0.5 / >8	0.5 / 8	0.5 / >8	1 / >8
Ceftazidime	0.25 / >32	0.12 / 32	0.25 / >32	0.25 / >32
Meropenem	0.03 / 0.06	0.03 / 0.06	0.03 / 0.12	0.03 / 1
Piperacillin-tazobactam	2 / 128	2 / 16	2 / 128	2 / >128
Colistin	0.12 / >8	0.12 / >8	0.12 / >8	0.12 / >8
Tigecycline	0.25 / 1	0.25 / 0.5	0.25 / 1	0.25 / 1
ESBL non-CRE	663	105	428	130
Plazomicin	0.25 / 1	0.5 / 1	0.25 / 1	0.5 / 1
Amikacin	2 / 8	2 / 8	2 / 8	2 / 8
Gentamicin	1 / >8	1 / >8	1 / >8	>8 / >8
Tobramycin	8 / >8	4 / >8	8 / >8	>8 / >8
Ceftazidime	32 / >32	16 / >32	32 / >32	32 / >32
Meropenem	0.03 / 0.12	0.03 / 0.06	0.03 / 0.12	0.03 / 0.5
Piperacillin-tazobactam	8 / >128	8 / >128	8 / >128	8 / >128
Colistin	0.12 / 0.25	0.12 / 0.25	0.12 / 0.25	0.12 / 1
Tigecycline	0.25 / 1	0.25 / 1	0.25 / 1	0.25 / 1
CRE	187	20	125	42
Plazomicin	0.25 / >128	0.25 / 1	0.25 / >128	0.25 / >128
Amikacin	16 / >32	4 / 16	16 / >32	16 / >32
Gentamicin	>8 / >8	0.5 / >8	>8 / >8	>8 / >8
Tobramycin	>8 / >8	8 / >8	>8 / >8	>8 / >8
Ceftazidime	>32 / >32	>32 / >32	>32 / >32	>32 / >32
Meropenem	32 / >32	16 / >32	32 / >32	32 / >32
Piperacillin-tazobactam	>128 / >128	>128 / >128	>128 / >128	>128 / >128
Colistin	0.25 / >8	0.12 / >8	0.25 / >8	0.25 / >8
Tigecycline	0.5 / 2	0.25 / 0.5	0.5 / 2	0.5 / 2
P. aeruginosa	101	25	52	24
Plazomicin	4 / 8	4 / 8	4 / 8	4 / 16
Amikacin	2 / 16	4 / 4	2 / 16	4 / 16
Gentamicin	1 / 8	1 / 4	1 / >8	1 / >8
Tobramycin	0.5 / 4	0.5 / 1	0.5 / 8	0.5 / >8
Ceftazidime	2 / 16	2 / >32	2 / 16	2 / 16
Meropenem	0.5 / 16	0.25 / 8	0.25 / 16	0.5 / 16
Piperacillin-tazobactam	4 / >64	4 / >64	4 / >64	4 / 64
Colistin	≤0.5 / 1	1 / 1	≤0.5 / 1	≤0.5 / 1

Table 2 Activity of plazomicin and comparator agents against main carbapenemase groups

Organism antimicrobial agent	Overall	Asia-Pacific	Europe	Latin America
	Region (no. tested) $MIC_{50/90}$ (mg/L)			
KPC producers	94	5	62	27
Plazomicin	0.25 / 128	0.5 /	0.25 / 2	0.25 / >128
Amikacin	16 / >32	2 /	32 / >32	4 / >32
Gentamicin	2 / >8	0.5 /	1 / >8	>8 / >8
Tobramycin	>8 / >8	0.5 /	>8 / >8	>8 / >8
Meropenem	>32 / >32	>32 /	>32 / >32	>32 / >32
Piperacillin-tazobactam	>128 / >128	>128 /	>128 / >128	>128 / >128
Colistin	0.5 / >8	0.25 /	0.25 / >8	8 / >8
Tigecycline	0.5 / 2	0.5 /	0.5 / 2	0.5 / 1
OXA-48-producers	30	0	30	0
Plazomicin	0.25 / 128		0.25 / 128	
Amikacin	4 / >32		4 / >32	
Gentamicin	>8 / >8		>8 / >8	
Tobramycin	>8 / >8		>8 / >8	
Meropenem	16 / 32		16 / 32	
Piperacillin-tazobactam	>128 / >128		>128 / >128	
Colistin	1 / >8		1 / >8	
Tigecycline	0.5 / 1		0.5 / 1	
NDM producers	41	5	21	15
Plazomicin	128 / >128	0.25 /	>128 / >128	128 / >128
Amikacin	>32 / >32	16 /	>32 / >32	>32 / >32
Gentamicin	>8 / >8	1 /	>8 / >8	>8 / >8
Tobramycin	>8 / >8	>8 /	>8 / >8	>8 / >8
Meropenem	32 / >32	>32 /	>32 / >32	32 / >32
Piperacillin-tazobactam	>128 / >128	>128 /	>128 / >128	>128 / >128
Colistin	0.12 / >8	0.12 /	0.12 / 0.5	0.25 / >8
Tigecycline	0.5 / 2	0.5 /	0.5 / 2	1 / 2