Poster #P1344

ECCMID 2017

Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

Activity of Tedizolid against Methicillin-Susceptible and -Resistant *Staphylococcus aureus* Pathogens Isolated from Patients in European Centres and Surrounding Regions

R.E. Mendes, D. Shortridge,
H.S. Sader, M. Castanheira,
R.K. Flamm

JMI Laboratories,
North Liberty, Iowa, USA

http://tinyurl.com/hd9xr94

Abstract

Background: Tedizolid has regulatory approvals for the treatment of acute bacterial skin and skin structure infections in the United States (US), European Union, Canada, and other countries. The activities of tedizolid and comparators were evaluated against *Staphylococcus aureus* pathogens from medical centres across Europe and surrounding areas in 2015.

Material/methods: A total of 2,007 nonduplicate, single-patient *S. aureus* (454 methicillin-resistant [MRSA]) were collected from 13 European countries (27 sites), including Russia (3 sites), Turkey (2 sites), and Israel (1 site). Isolates were submitted to a monitoring laboratory as part of the Surveillance of Tedizolid Activity and Resistance (STAR) program. Identification was confirmed and susceptibility testing was performed by CLSI methods. MIC interpretation used the EUCAST criteria. Isolates displaying a resistance phenotype to at least 3 classes of antibacterials (in addition to methicillin) were considered multidrug-resistant (MDR).

Results: MRSA rates were lowest in Sweden (1.0%), Turkey (5.0%), the United Kingdom (10.3%), and France (14.1%) and were highest in Portugal (63.2%), Greece (45.5%), Italy (40.7%), Ireland (36.8%), and Israel (30.0%). Overall, tedizolid inhibited all isolates at ≤0.25 mg/L (100.0% susceptible) and had MIC_{50} and MIC_{90} results of 0.12 and 0.12 mg/L, respectively, against the MRSA and MDR subsets. Equivalent tedizolid MIC results ($MIC_{50/90}$, 0.12/0.12 mg/L) were obtained against methicillin-susceptible *S. aureus* (MSSA) isolates. Tedizolid and tigecycline ($MIC_{50/90}$, 0.06/0.12 mg/L; 100.0% susceptible) were similarly active against the MRSA population, and tedizolid had MIC results 4- to 16-fold lower than daptomycin ($MIC_{50/90}$, 0.25/0.5 mg/L; 100.0% susceptible), vancomycin ($MIC_{50/90}$, 0.5/1 mg/L; 100.0% susceptible), linezolid ($MIC_{50/90}$, 1/1 mg/L; 100.0% susceptible), and ceftaroline ($MIC_{50/90}$, 1/2 mg/L; 85.5% susceptible). These comparators and tedizolid showed equivalent MIC values against MDR isolates, respectively, compared to MRSA. The only exception was tigecycline ($MIC_{50/90}$, 0.12/0.25 mg/L) that had MIC values against MDR isolates 2-fold higher than the MRSA subset. Tedizolid (MIC_{90}, 0.12 mg/L) and linezolid (MIC_{90}, 1 mg/L) showed consistent MIC_{90} values against MRSA from each region evaluated, while these values varied ±2-fold for other agents.

Conclusion: MRSA rates remained high in some European regions. Tedizolid demonstrated potent and consistent in vitro activity against MRSA and MDR clinical isolates from these European regions. Tedizolid activity was also greater than comparator agents.

Introduction

- *Staphylococcus aureus* remains an important bacterial pathogen that causes a variety of human infections in the community and nosocomial environment[1]
- *S. aureus* has proven to be a highly adaptable pathogen, fully capable of acquiring multiple resistance mechanisms as well as increased virulence[1]
- The multidrug-resistant (MDR) capacity of *S. aureus*, especially those lineages expressing methicillin resistance (MRSA), pose a challenge to the empirical treatment of serious infections[1]
- Concerns regarding the adequacy of vancomycin in the treatment of complicated staphylococcal infections have prompted the development of several new agents with potent activity against MRSA, methicillin-susceptible *S. aureus* (MSSA), and MDR MRSA, including some strains with elevated minimal inhibitory concentration (MIC) values to vancomycin, which has become the cornerstone for treating MRSA infections[2]
- Tedizolid is an oxazolidinone derivative that exhibits potency greater than linezolid when tested against gram-positive cocci, including MDR phenotypes such as MRSA, vancomycin-resistant enterococci (VRE), and linezolid-resistant phenotypes[3]
- Tedizolid demonstrates activity against linezolid-resistant bacterial strains harbouring the horizontally transferable *cfr* gene[3,4]
- Tedizolid was approved in the United States (US), European Union, Canada, and other countries for the treatment of acute bacterial skin and skin structure infections (ABSSSI)[3]
- Tedizolid is also under investigation for the treatment of hospital-acquired bacterial pneumonia, ventilated nosocomial pneumonia, and bone and joint infections[4, 5]
- In this study, the activities of tedizolid and comparators were evaluated against *S. aureus* pathogens from medical centres across Europe and surrounding areas in 2015

Materials and Methods

- 2,007 nonduplicate, single-patient *S. aureus* (454 MRSA) isolates were included
 - Collected from 10 European countries (27 sites) and Russia (3 sites), Turkey (2 sites), and Israel (1 site)
 - Part of The Surveillance of Tedizolid Activity and Resistance (STAR) Program during 2015 for Europe and adjacent regions (Figure 1)
- Isolates were initially identified by the participating laboratory and submitted to a central monitoring facility (JMI Laboratories, North Liberty, Iowa, USA) where bacterial identifications were confirmed using standard algorithms and supported by matrix assisted laser desorption ionization time of flight technology mass spectrometry (MALDI-TOF MS; Bruker Daltonics, Bremen, Germany)
- Isolates were tested for susceptibility by broth microdilution following guidelines from the Clinical and Laboratory Standards Institute (CLSI) M07-A10 document[6]
 - Tests performed used reference 96-well panels manufactured by JMI Laboratories (North Liberty, Iowa, USA)
- MIC reading for tedizolid and linezolid was performed according to the CLSI guidelines – ie, the first well at which trailing begins without regard for pinpoint trailing in the wells[7]
- Quality assurance was performed by concurrent testing of CLSI-recommended QC reference strains (*S. aureus* ATCC 29213 and *Enterococcus faecalis* 29212)[7]
 - All QC results were within published acceptable ranges
- Breakpoint criteria for tedizolid and comparator agents were those from EUCAST (2016)[8]
- Tigecycline MIC breakpoints were those found in the US Food and Drug Administration-approved package insert[9]
- MRSA isolates displaying a resistance phenotype to at least 3 classes of antibacterials (in addition to methicillin) were considered MDR

Figure 1. Methicillin resistance rates observed among *S. aureus* collected from each country/region participating in the surveillance program for Europe and adjacent regions in 2015

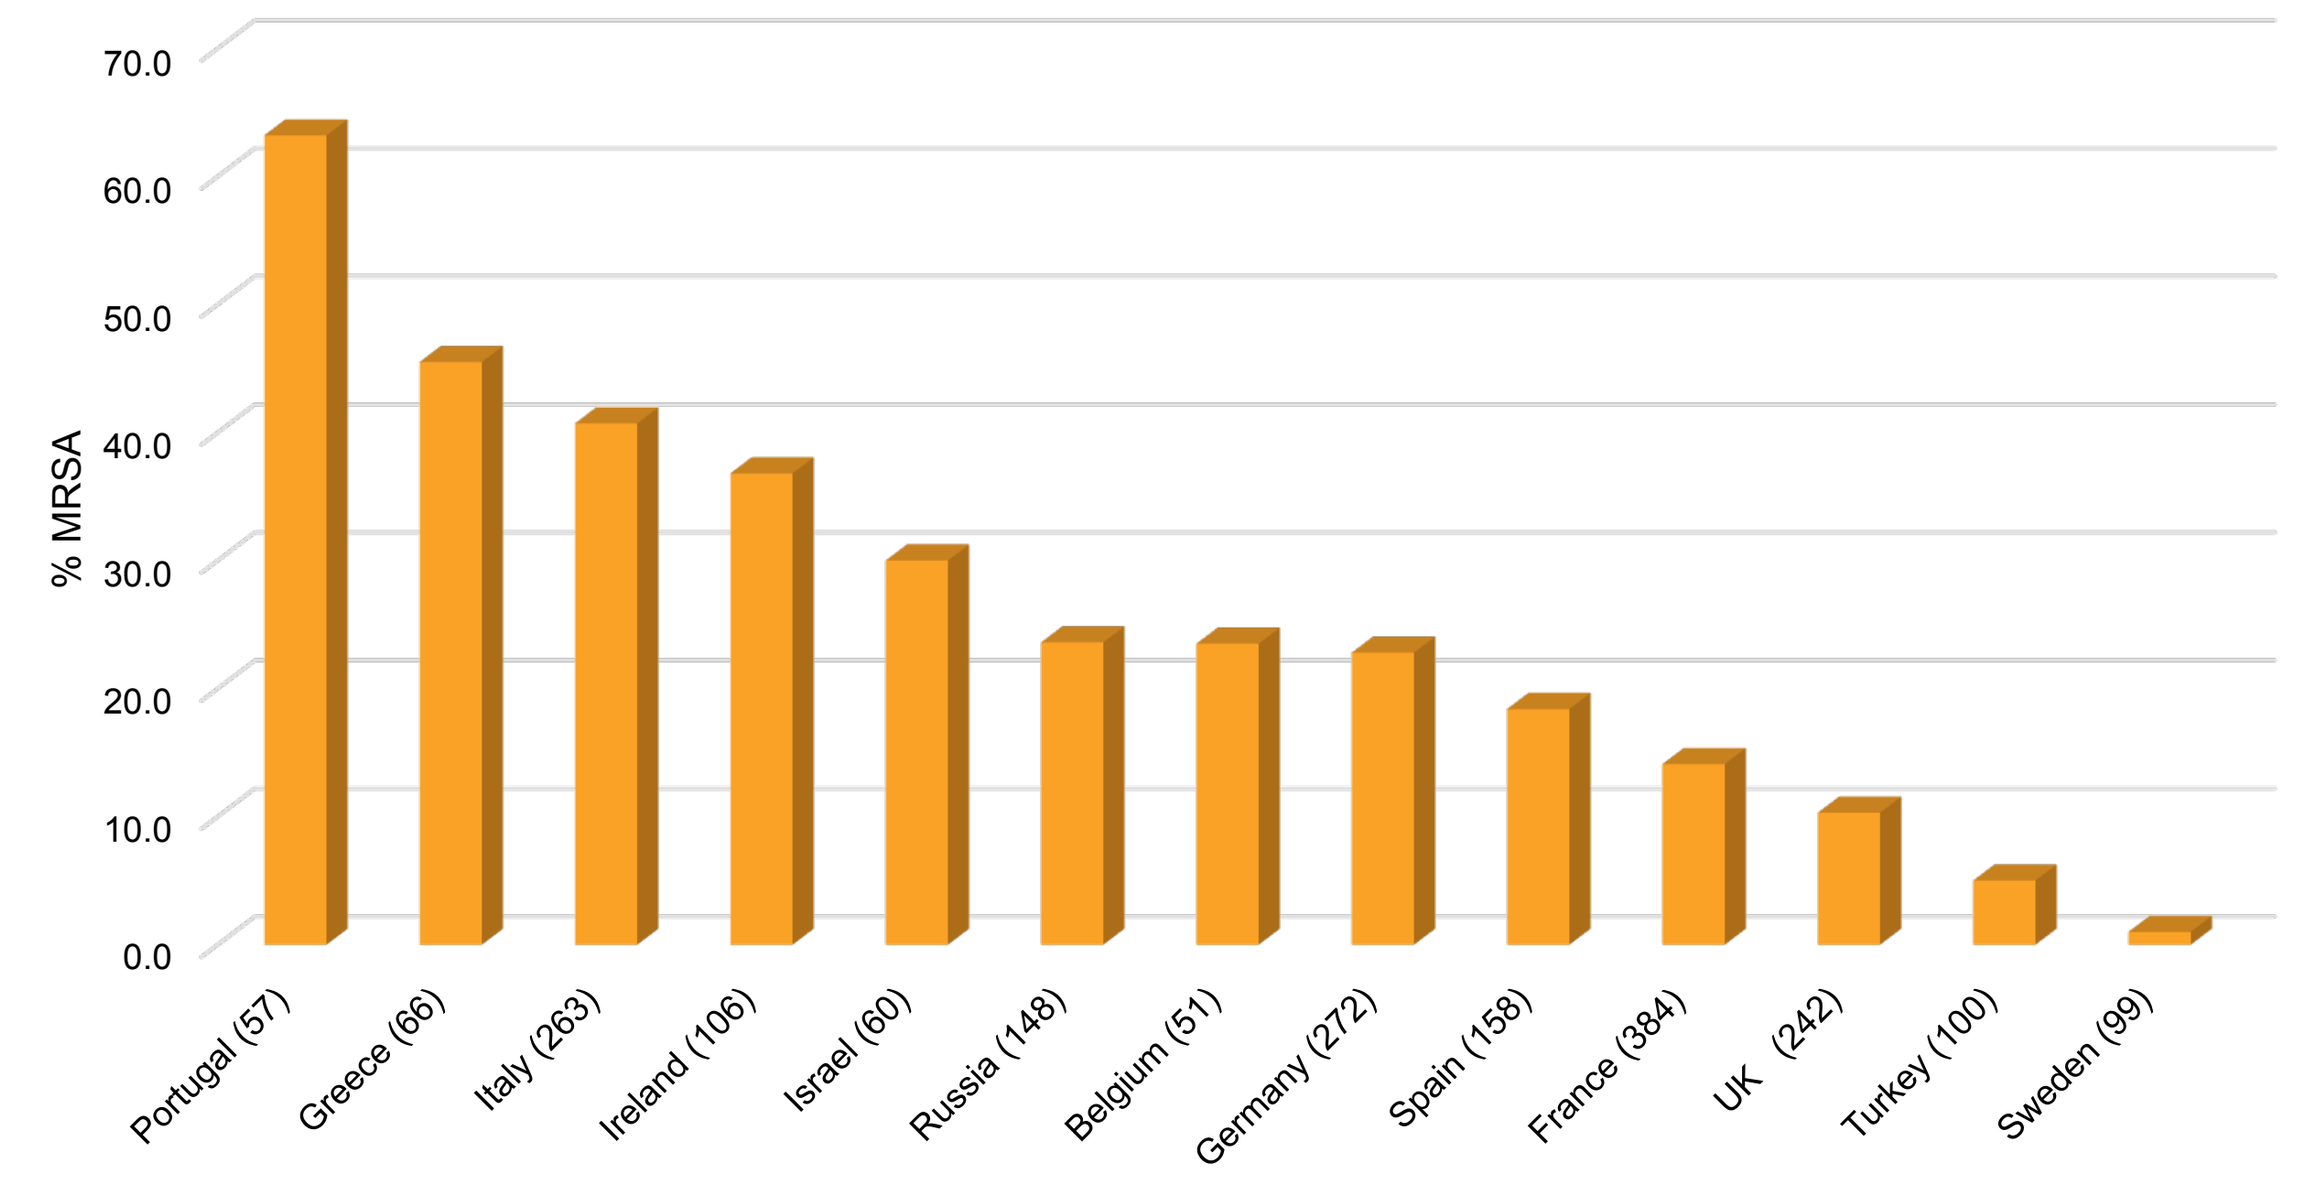
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
% MRSA
Portugal (57)
Greece (66)
Italy (263)
Ireland (106)
Israel (60)
Russia (148)
Belgium (51)
Germany (272)
Spain (158)
France (384)
UK (242)
Turkey (100)
Sweden (99)

Results

- MRSA rates were lowest in Sweden (1.0%), Turkey (5.0%), the United Kingdom (10.3%), and France (14.1%) and were highest in Portugal (63.2%), Greece (45.5%), Italy (40.7%), Ireland (36.8%), and Israel (30.0%) during the surveillance sampling year of 2015 (Figure 1)
- Overall, tedizolid inhibited all isolates at ≤0.25 mg/L (100.0% susceptible) and had MIC_{50} and MIC_{90} results of 0.12 and 0.12 mg/L, respectively, against the MSSA and MRSA subsets (Table 1)
 - Tedizolid MIC values ($MIC_{50/90}$, 0.12/0.12 mg/L) were 8-fold lower than linezolid ($MIC_{50/90}$, 1/1 mg/L) against MRSA and MSSA isolates
- Tedizolid ($MIC_{50/90}$, 0.12/0.12 mg/L; 100.0% susceptible) and tigecycline ($MIC_{50/90}$, 0.06/0.12 mg/L; 100.0% susceptible) were similarly active against the MRSA population (Table 2 and Figure 2)
 - Other agents, such as daptomycin, vancomycin, teicoplanin, and linezolid were also active (99.3%–100.0% susceptible) against MRSA, but had MIC_{90} results 4- to 16-fold higher than tedizolid

Table 1. Activity of tedizolid and the direct comparator linezolid tested against contemporary *S. aureus* isolates from Europe and adjacent regions

S. aureus / phenotype	Number (cumulative %) inhibited at tedizolid MIC (mg/L) of†: 0.03	0.06	0.12	0.25	0.5	1	2	MIC_{50}	MIC_{90}
All (2,007)									
Tedizolid	16 (0.8%)	484 (24.9%)	**1,368 (93.1%)**	139 (100.0%)				0.12	0.12
Linezolid			2 (0.1%)	21 (1.1%)	643 (33.2%)	**1,320 (99.0%)**	21 (100.0%)	1	1
MSSA (1,553)									
Tedizolid	10 (0.6%)	346 (22.9%)	**1,075 (92.1%)**	122 (100.0%)				0.12	0.12
Linezolid			2 (0.1%)	14 (1.0%)	455 (30.3%)	**1,065 (98.9%)**	17 (100.0%)	1	1
MRSA (454)									
Tedizolid	6 (1.3%)	138 (31.7%)	**293 (96.3%)**	17 (100.0%)				0.12	0.12
Linezolid				7 (1.5%)	188 (43.0%)	**255 (99.1%)**	4 (100.0%)	1	1
MRSA MDR (180)									
Tedizolid	3 (1.7%)	50 (29.4%)	**119 (95.6%)**	8 (100.0%)				0.12	0.12
Linezolid				5 (2.8%)	**89 (52.2%)**	86 (100.0%)		0.5	1

MSSA = methicillin-susceptible *S. aureus*; MRSA = methicillin-resistant *S. aureus*; MDR = multidrug-resistant (ie, MRSA showing a resistance phenotype to 3 drug classes in addition to β-lactams)
† Bold values represent modal MIC results

Table 2. Antimicrobial activity of tedizolid and comparator agents against contemporary *S. aureus* clinical isolates causing infections in Europe and adjacent regions

Organism/phenotype (no. tested) Antimicrobial Agent	MIC (µg/mL): MIC_{50}	MIC_{90}	Range	%Susceptible	%Intermediate	%Resistant†
MSSA (1,553)						
Tedizolid	0.12	0.12	0.03 – 0.25	100.0	—	0.0
Linezolid	1	1	≤0.12 – 2	100.0	—	0.0
Ceftaroline	0.25	0.25	≤0.06 – 0.5	100.0	—	0.0
Clindamycin	≤0.25	≤0.25	≤0.25 – >2	98.1	0.3	1.6
Daptomycin	0.25	0.5	≤0.12 – 1	100.0	—	0.0
Erythromycin	0.25	>8	≤0.06 – >8	83.4	1.4	15.3
Levofloxacin	0.12	0.25	0.06 – >4	95.8	—	4.2
Teicoplanin	≤0.5	≤0.5	≤0.5 – 2	100.0	—	0.0
Tetracycline	≤0.5	≤0.5	≤0.5 – >8	94.9	0.2	4.9
Tigecycline	0.06	0.12	≤0.015 – 0.25	100.0	—	0.0
TMP-SMX	≤0.5	≤0.5	≤0.5 – 4	99.9	0.1	0.0
Vancomycin	0.5	1	0.03 – 0.25	100.0	—	0.0
MRSA (454)						
Tedizolid	0.12	0.12	0.03 – 0.25	100.0	—	0.0
Linezolid	1	1	0.25 – 2	100.0	—	0.0
Ceftaroline	1	2	0.25 – 2	85.5	—	14.5
Clindamycin	≤0.25	>2	≤0.25 – >2	67.6	0.2	32.2
Daptomycin	0.25	0.5	≤0.12 – 1	100.0	—	0.0
Erythromycin	>8	>8	≤0.06 – >8	30.5	2.0	67.5
Levofloxacin	>4	>4	0.12 – >4	15.0	0.2	84.8
Teicoplanin	≤0.5	1	≤0.5 – 8	99.3	—	0.7
Tetracycline	≤0.5	>8	≤0.5 – >8	86.3	0.2	13.4
Tigecycline	0.06	0.12	0.03 – 0.5	100.0	—	0.0
TMP-SMX	≤0.5	≤0.5	≤0.5 – >4	99.6	0.2	0.2
Vancomycin	0.5	1	≤0.12 – 2	100.0	—	0.0
MDR MRSA (180)						
Tedizolid	0.12	0.12	0.03 – 0.25	100.0	—	0.0
Linezolid	0.5	1	0.25 – 1	100.0	—	0.0
Ceftaroline	1	2	0.25 – 2	68.3	—	31.7
Clindamycin	>2	>2	≤0.25 – >2	18.3	0.6	81.1
Daptomycin	0.25	0.5	≤0.12 – 1	100.0	—	0.0
Erythromycin	>8	>8	2 – >8	0.0	0.6	99.4
Levofloxacin	>4	>4	0.25 – >4	0.6	0.6	98.9
Teicoplanin	≤0.5	1	≤0.5 – 8	98.9	—	1.1
Tetracycline	≤0.5	>8	≤0.5 – >8	83.9	0.6	15.6
Tigecycline	0.12	0.25	0.03 – 0.5	100.0	—	0.0
TMP-SMX	≤0.5	≤0.5	≤0.5 – >4	98.9	0.6	0.6
Vancomycin	0.5	1	≤0.12 – 2	100.0	—	0.0

MSSA = methicillin-susceptible *S. aureus*; MRSA = methicillin-resistant *S. aureus*; MDR = multidrug-resistant (ie, MRSA showing a resistance phenotype to three drug classes in addition to β-lactams); TMP-SMX = trimethoprim-sulfamethoxazole.
† Breakpoint criteria for tedizolid and comparator agents were those from EUCAST (2016), as available. "—" breakpoint not available. Interpretation for tigecycline MIC results utilized breakpoints approved by the US Food and Drug Administration.

- Tedizolid ($MIC_{50/90}$, 0.12/0.12 mg/L; 100.0% susceptible) displayed MIC values 4- to 8-fold lower than linezolid ($MIC_{50/90}$, 0.5/1 mg/L; 100.0% susceptible) against MDR MRSA (Tables 1 and 2)
 - The respective in vitro activity of tedizolid and linezolid against MDR isolates remained comparable to those obtained against MSSA
- Daptomycin, teicoplanin, tigecycline, trimethoprim-sulfamethoxazole, and vancomycin showed activity against MDR isolates similar to that obtained against the susceptible counterpart MSSA isolates (Table 2 and Figure 2)
- Overall, MDR isolates had high resistance rates to ceftaroline (31.7%), clindamycin (81.1%), erythromycin (99.4%), levofloxacin (98.9%), and tetracycline (15.6%; Table 2 and Figure 2)
- Tedizolid (MIC_{90}, 0.12 mg/L) and linezolid (MIC_{90}, 1 mg/L) showed consistent MIC_{90} values against MRSA isolates from each region evaluated, while these values varied ±2-fold for other agents (data not shown)

Figure 2. Antimicrobial susceptibility profiles of MSSA, MRSA, and MDR MRSA tested against tedizolid and comparator agents

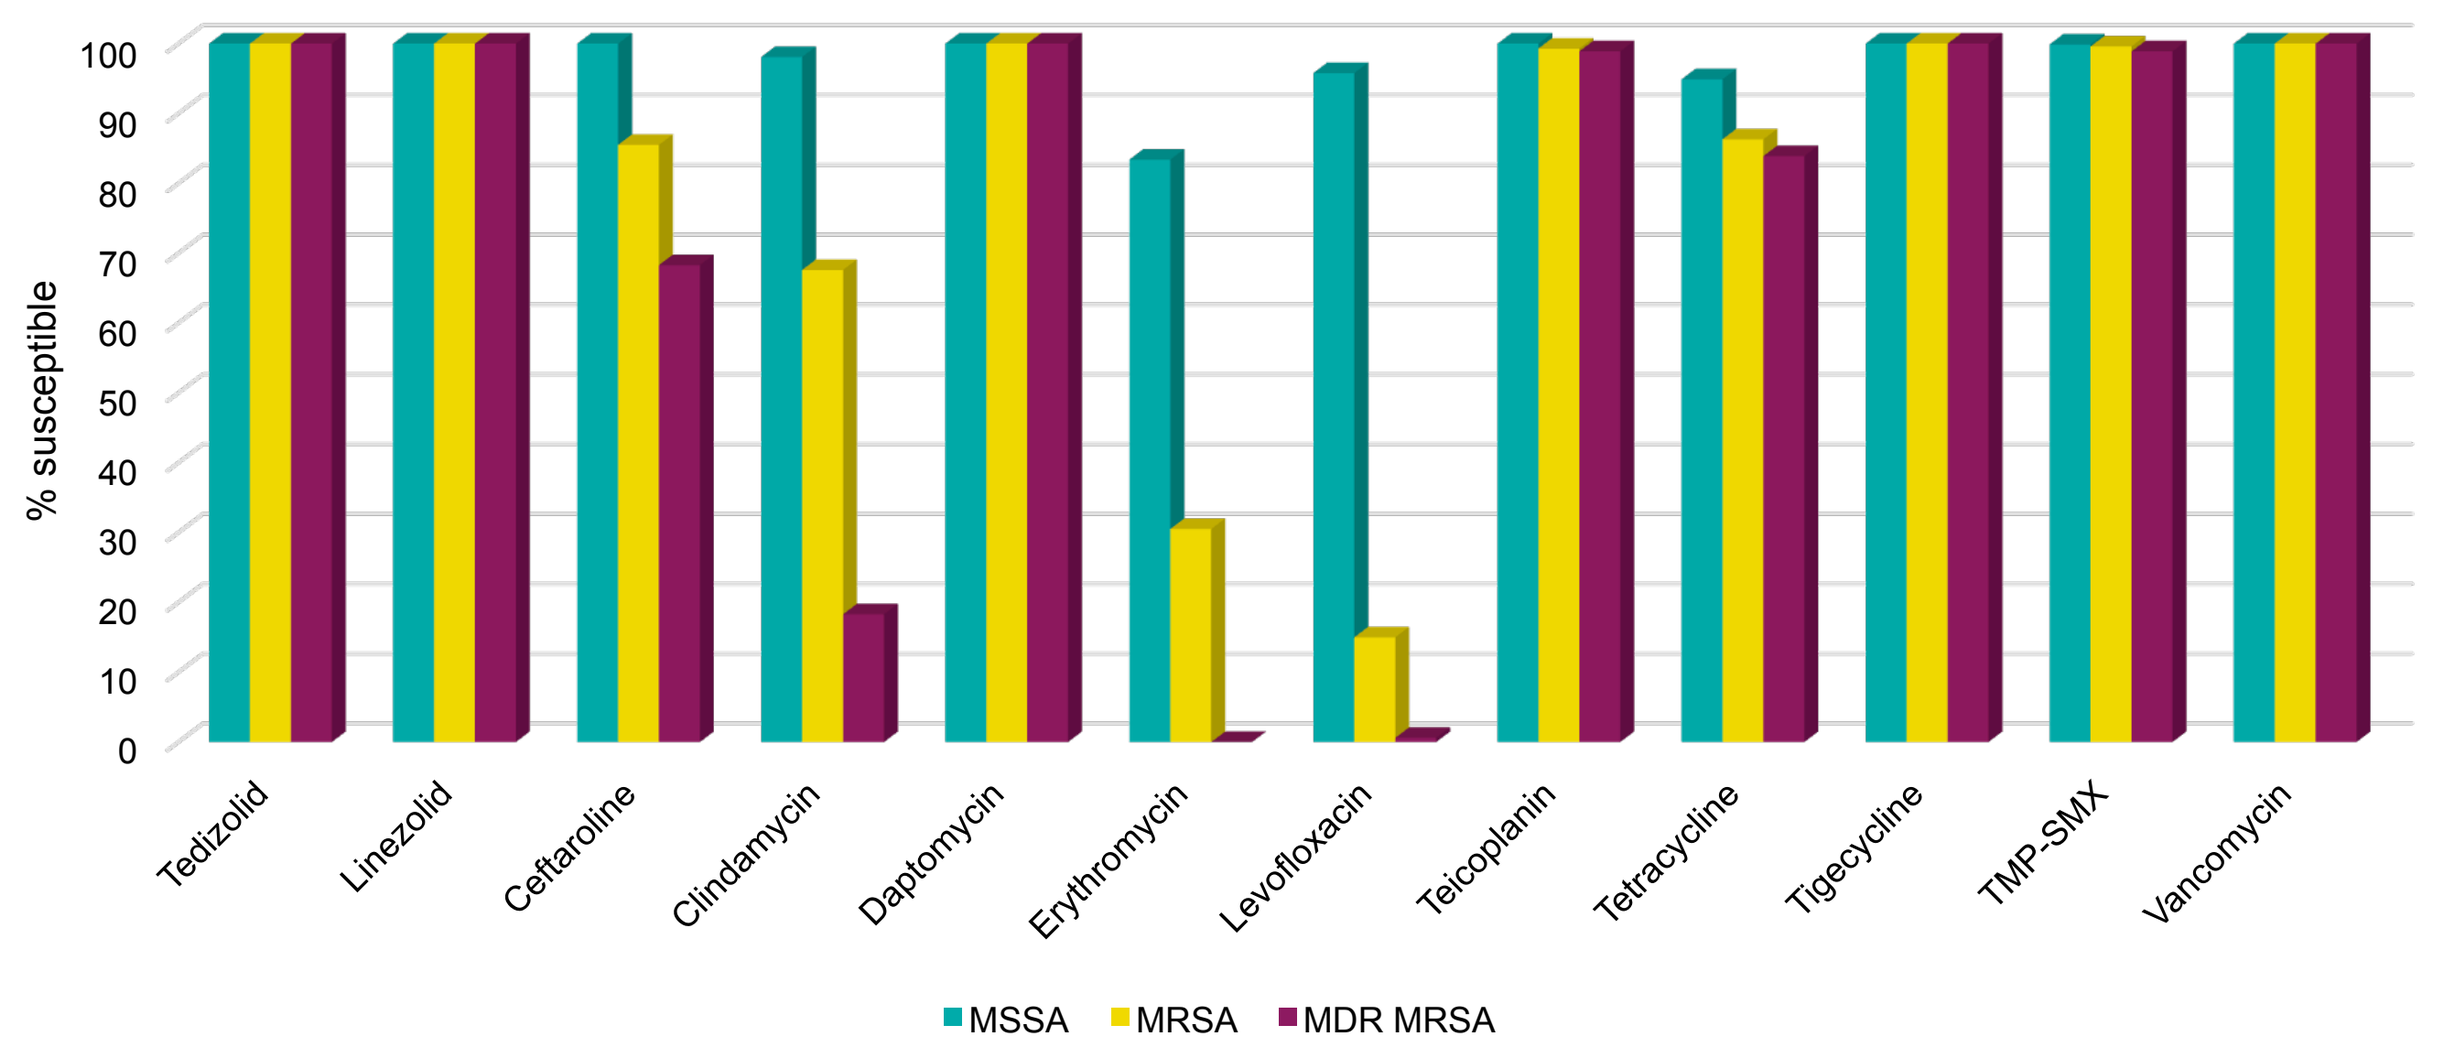
100
90
80
70
60
50
40
30
20
10
0
% susceptible
Tedizolid
Linezolid
Ceftaroline
Clindamycin
Daptomycin
Erythromycin
Levofloxacin
Teicoplanin
Tetracycline
Tigecycline
TMP-SMX
Vancomycin
MSSA
MRSA
MDR MRSA

Conclusions

- Methicillin resistance rates varied considerably among the European regions investigated and reached rates as high as 60% in Portugal and as low as 1% in Sweden
- Tedizolid demonstrated potent in vitro activity against both aggregate populations of MRSA and MDR from these European regions
- Tedizolid activity was consistent across MRSA and MDR isolates from each geographic region

Acknowledgements

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ, USA. Editorial assistance was provided by Tracy Cao, PhD, of ApotheCom, Yardley, PA, USA. This assistance was funded by Merck & Co., Inc., Kenilworth, NJ, USA.

References

1. Kraker ME, Jarlier V, Monen JC, Heuer OE, van de Sande N, Grundmann H (2013). The changing epidemiology of bacteraemias in Europe: trends from the European Antimicrobial Resistance Surveillance System. *Clin Microbiol Infect* 19: 860 - 868.
2. Kalil AC, Van Schooneveld TC, Fey PD, Rupp ME (2014). Association between vancomycin minimum inhibitory concentration and mortality among patients with *Staphylococcus aureus* bloodstream infections: A systematic review and meta-analysis. *JAMA* 312: 1552-1564.
3. SIVEXTRO Package Insert (2014). Available at http://www.accessdata.fda.gov/drugsatfda_docs/label/2014/205435s000lbl.pdf. Accessed April 2015.
4. Pfaller MA, Flamm RK, Jones RN, Farrell DJ, Mendes RE (2016). Activities of tedizolid and linezolid determined by the reference broth microdilution method against 3,032 Gram-positive bacterial isolates collected in Asia-Pacific, Eastern Europe, and Latin American countries in 2014. *Antimicrob Agents Chemother* 60: 5393 - 5399.
5. ClinicalTrials.gov. Tolerability, safety, and efficacy of tedizolid as oral treatment for bone and joint infections. Available at https://clinicaltrials.gov/ct2/show/NCT03009045. Accessed March 2017.
6. Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard - Tenth edition*. Wayne, PA, USA.
7. Clinical and Laboratory Standards Institute (2016). *M100-S26. Performance standards for antimicrobial susceptibility testing: 26th Informational Supplement.* Wayne, PA, USA.
8. EUCAST (2017). Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, January 2016. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2016.
9. Tygacil (2016). Tygacil ® Package Insert. Wyeth Pharmaceuticals. Available at www.tygacil.com. Accessed March 1, 2016.

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.