ECCMID 2019 | Poster #P1843

Antimicrobial Activity of POL7306 Tested against Clinical Isolates of Gram-Negative Bacteria Collected Worldwide

Helio S. Sader[1], Paul R. Rhomberg[1], Leonard R. Duncan[1], Hans H. Locher[2], Glenn E. Dale[2], Robert K. Flamm[1]

[1] JMI Laboratories, North Liberty, Iowa, USA; [2] Polyphor Ltd, Hegenheimermattweg 125, CH-4123 Allschwil, SwitzerlandK

Introduction

- POL7306 belongs to a novel class of peptidomimetic outer membrane protein targeting antibiotics (OMPTAs) with a novel mechanism of action and potent activity against gram-negative bacteria
- See also ECCMID posters P1838, P1839, P1840 and P1843

Materials and Methods

Organism collection

- A total of 891 isolates were collected by the SENTRY Antimicrobial Surveillance Program
- Isolates were from 134 medical centres in Europe (n=424; 41 centres in 18 nations), United States (USA; 411 isolates from 67 centres), Asia-Pacific region (APAC; n=24; 15 centres in 6 nations), and Latin America (LATAM; n=32; 11 centres in 9 nations)
- The collection comprised 558 *Enterobacterales*, 310 non-fermenters, and 23 fastidious organisms

Susceptibility testing

- Susceptibility testing was performed by reference broth microdilution method
- MIC panels were manufactured at JMI Laboratories
- Media was supplemented with 0.002% polysorbate 80 (P-80) for POL7306 tests
- Resistant subsets were characterized by whole genome sequencing

Results

- POL7306 was very active against *Enterobacterales*, except against *Proteus mirabilis*, indole-positive Proteeae (IPP), and *Serratia* spp., which are organisms intrinsically resistant to cationic antimicrobial peptides (**Table 1** and **Figure 1**)
- POL7306 ($MIC_{50/90}$, 0.06/0.25 mg/L; highest MIC, 1 mg/L) was slightly more active than colistin ($MIC_{50/90}$, 0.12/0.25 mg/L; highest MIC, >8 mg/L) against randomly selected *Enterobacterales* (excluding the intrinsically resistant species) (**Table 1** and **Figure 1**)
- POL7306 was also highly active against carbapenem-resistant ($MIC_{50/90}$, 0.06/0.25 mg/L), extended-spectrum β-lactamase (ESBL)-producing ($MIC_{50/90}$, 0.06/0.12 mg/L), KPC-producing ($MIC_{50/90}$, 0.12/0.25 mg/L), and metallo-β-lactamase-producing ($MIC_{50/90}$, 0.06/0.25 mg/L) *Enterobacterales* (**Table 1** and **Figure 2**)
- The only β-lactamase-producing *Enterobacterales* isolate with an elevated POL7306 MIC result was a VIM-like-producing *Serratia marcescens*, a species intrinsically resistant to cationic antimicrobial peptides (**Figure 2**)
- POL7306 ($MIC_{50/90}$, 0.12/0.25 mg/L) was 32-fold more potent than colistin ($MIC_{50/90}$, 4/8 mg/L) when tested against *mcr*-positive *Enterobacterales* and retained good activity ($MIC_{50/90}$, 0.5/2 mg/L) against colistin-resistant (MIC >2 mg/L) *mcr*-negative *Klebsiella pneumoniae* (**Figure 3**)
- The *Pseudomonas aeruginosa* collection (n=154) included 102 extensively drug-resistant (XDR) isolates, and the highest POL7306 MIC value was only 0.5 mg/L ($MIC_{50/90}$, 0.25/0.25 mg/L; **Table 2** and **Figure 1**)
- POL7306 exhibited good activity against XDR *Acinetobacter baumannii* (n=105; $MIC_{50/90}$, 0.06/0.12 mg/L) and *Stenotrophomonas maltophilia* (n=51; $MIC_{50/90}$, 0.06/0.25 mg/L **Table 2** and **Figure 1**)

Figure 1 POL7306 MIC distributions when tested against *Enterobacterales* (non-intrinsically resistant species) and non-fermentative gram-negative bacilli

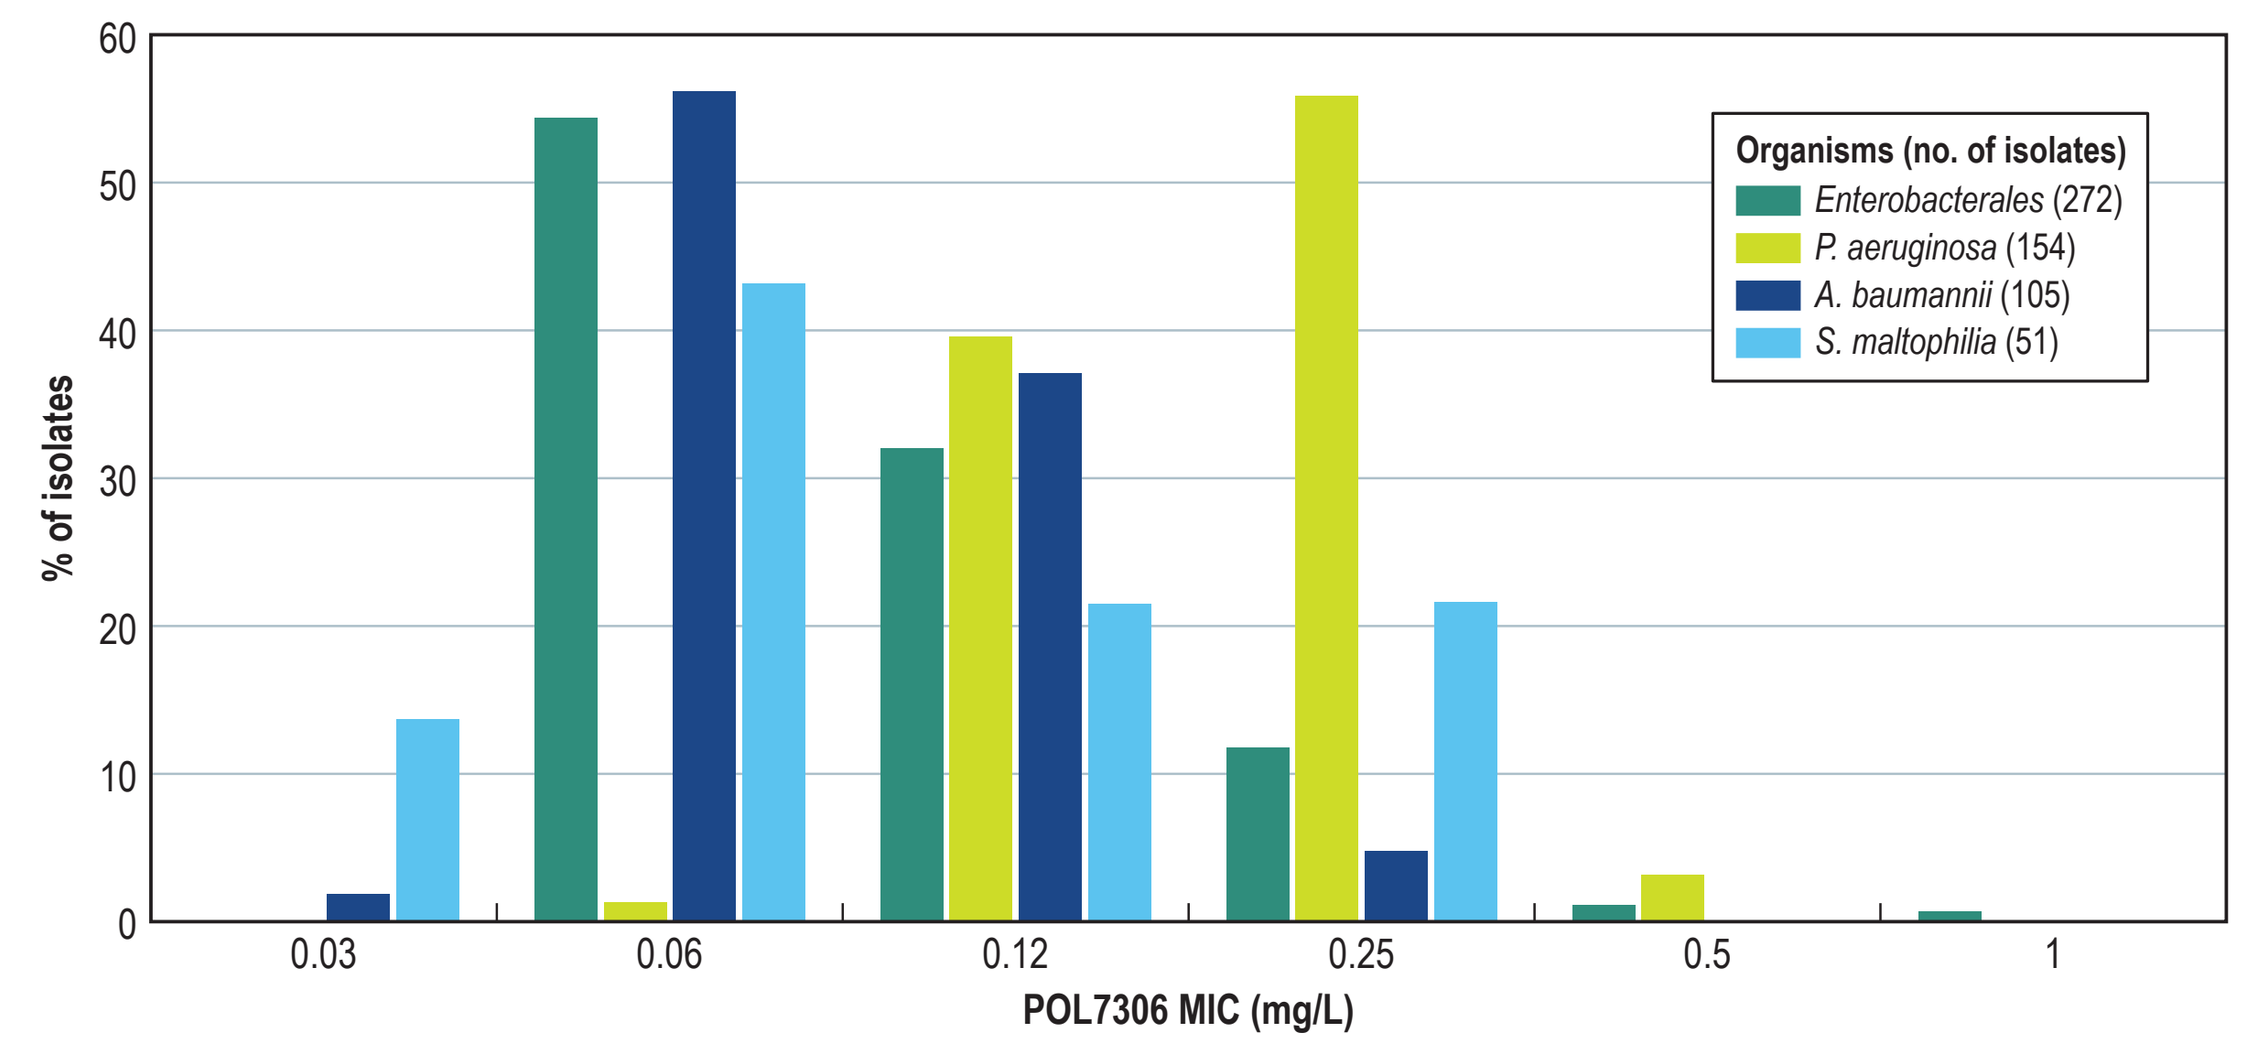
% of isolates
POL7306 MIC (mg/L)
0.03
0.06
0.12
0.25
0.5
1
Organisms (no. of isolates)
Enterobacterales (272)
P. aeruginosa (154)
A. baumannii (105)
S. maltophilia (51)

Figure 2 POL7306 MIC distributions when tested against resistant subsets of *Enterobacterales*

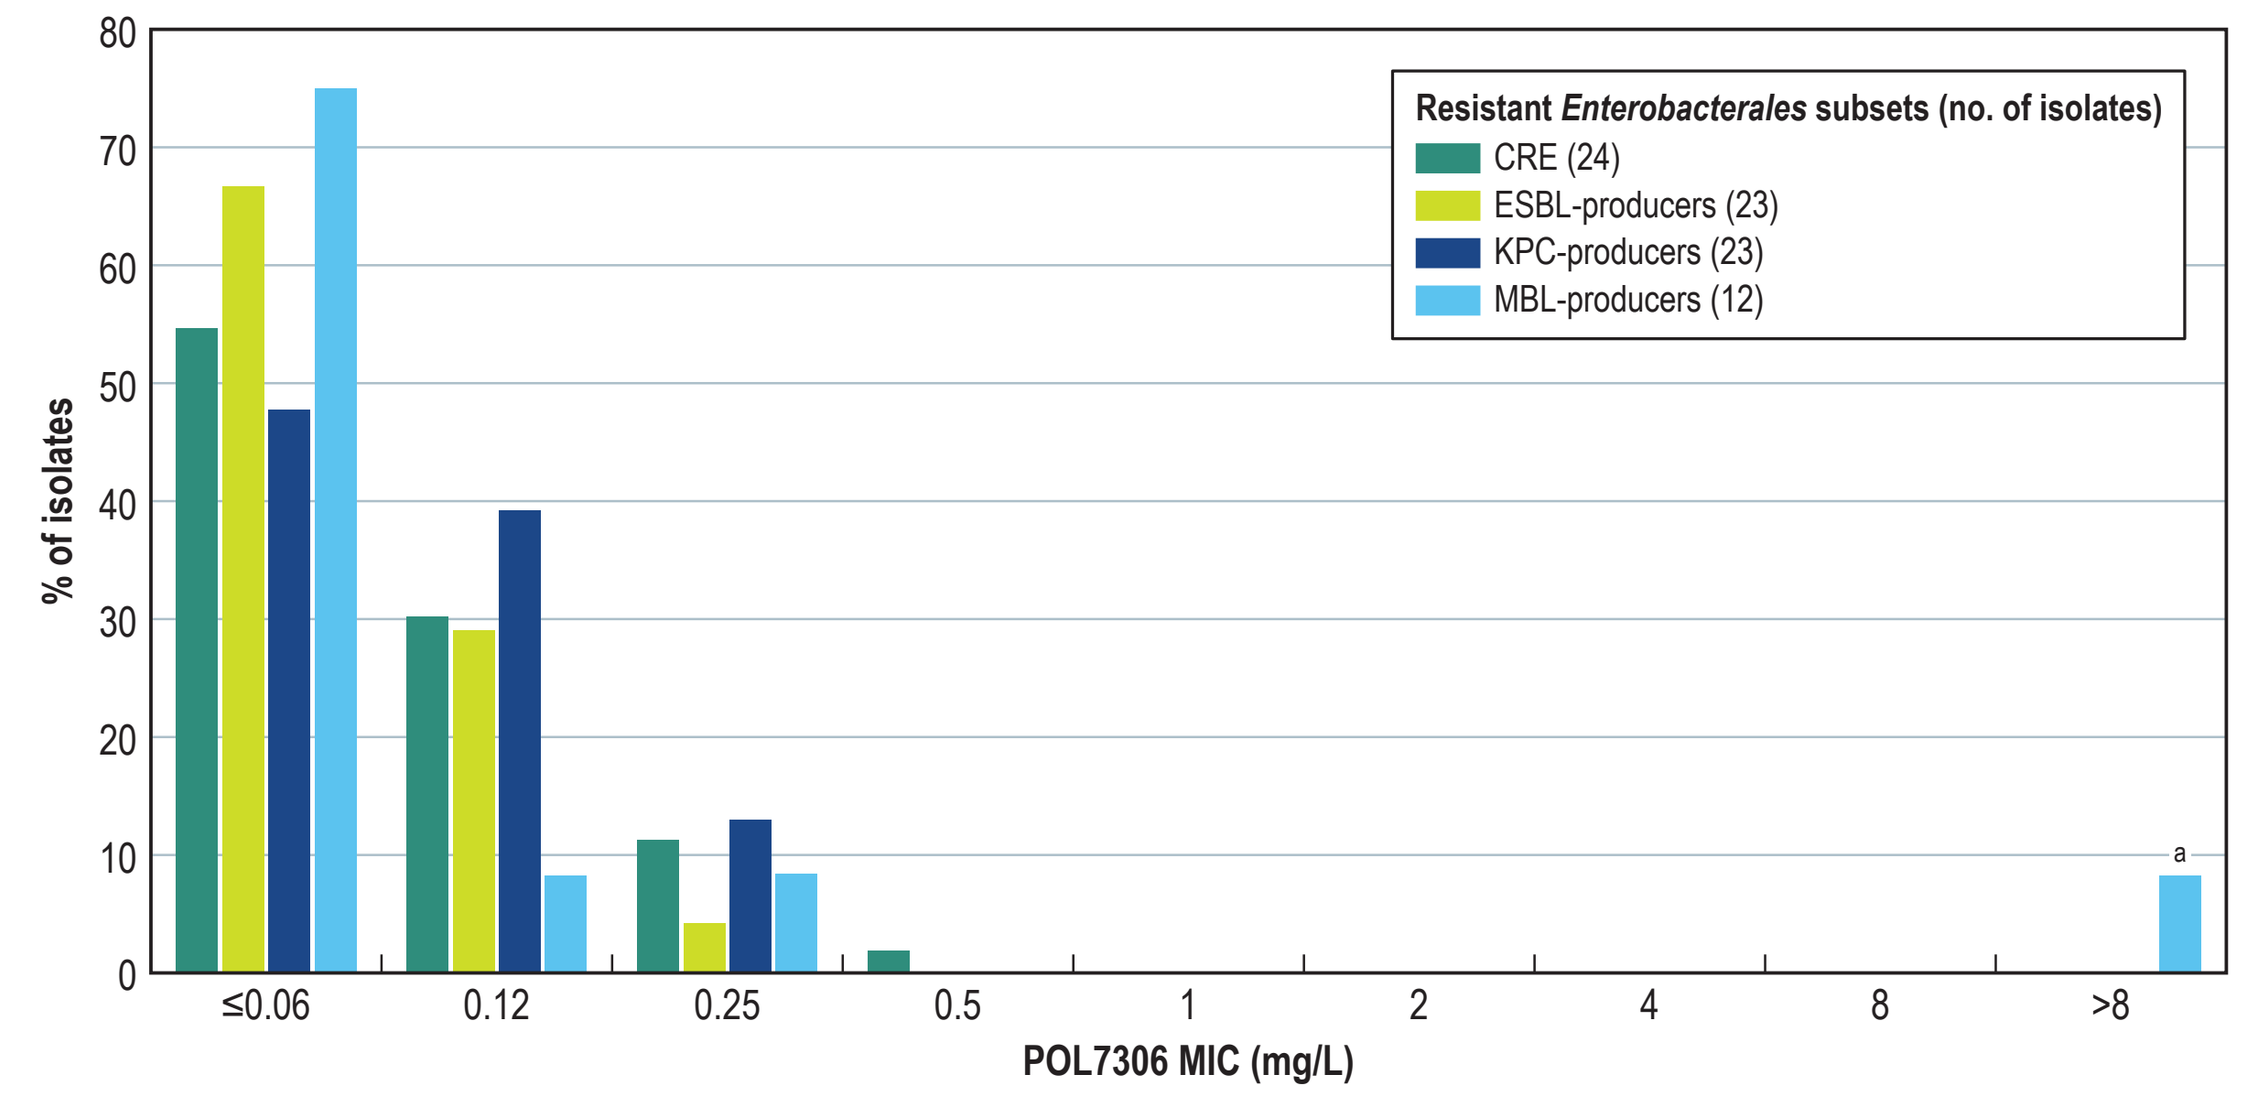
% of isolates
POL7306 MIC (mg/L)
≤0.06
0.12
0.25
0.5
1
2
4
8
>8
a
Resistant Enterobacterales subsets (no. of isolates)
CRE (24)
ESBL-producers (23)
KPC-producers (23)
MBL-producers (12)

[a] A VIM-like-producing *S. marcescens*.
CRE, carbapenem-resistant *Enterobacterales*; ESBL, extended-spectrum β-lactamase; KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamase.

Figure 3 POL7306 MIC distributions when tested against colistin-nonsusceptible *Enterobacterales* isolates

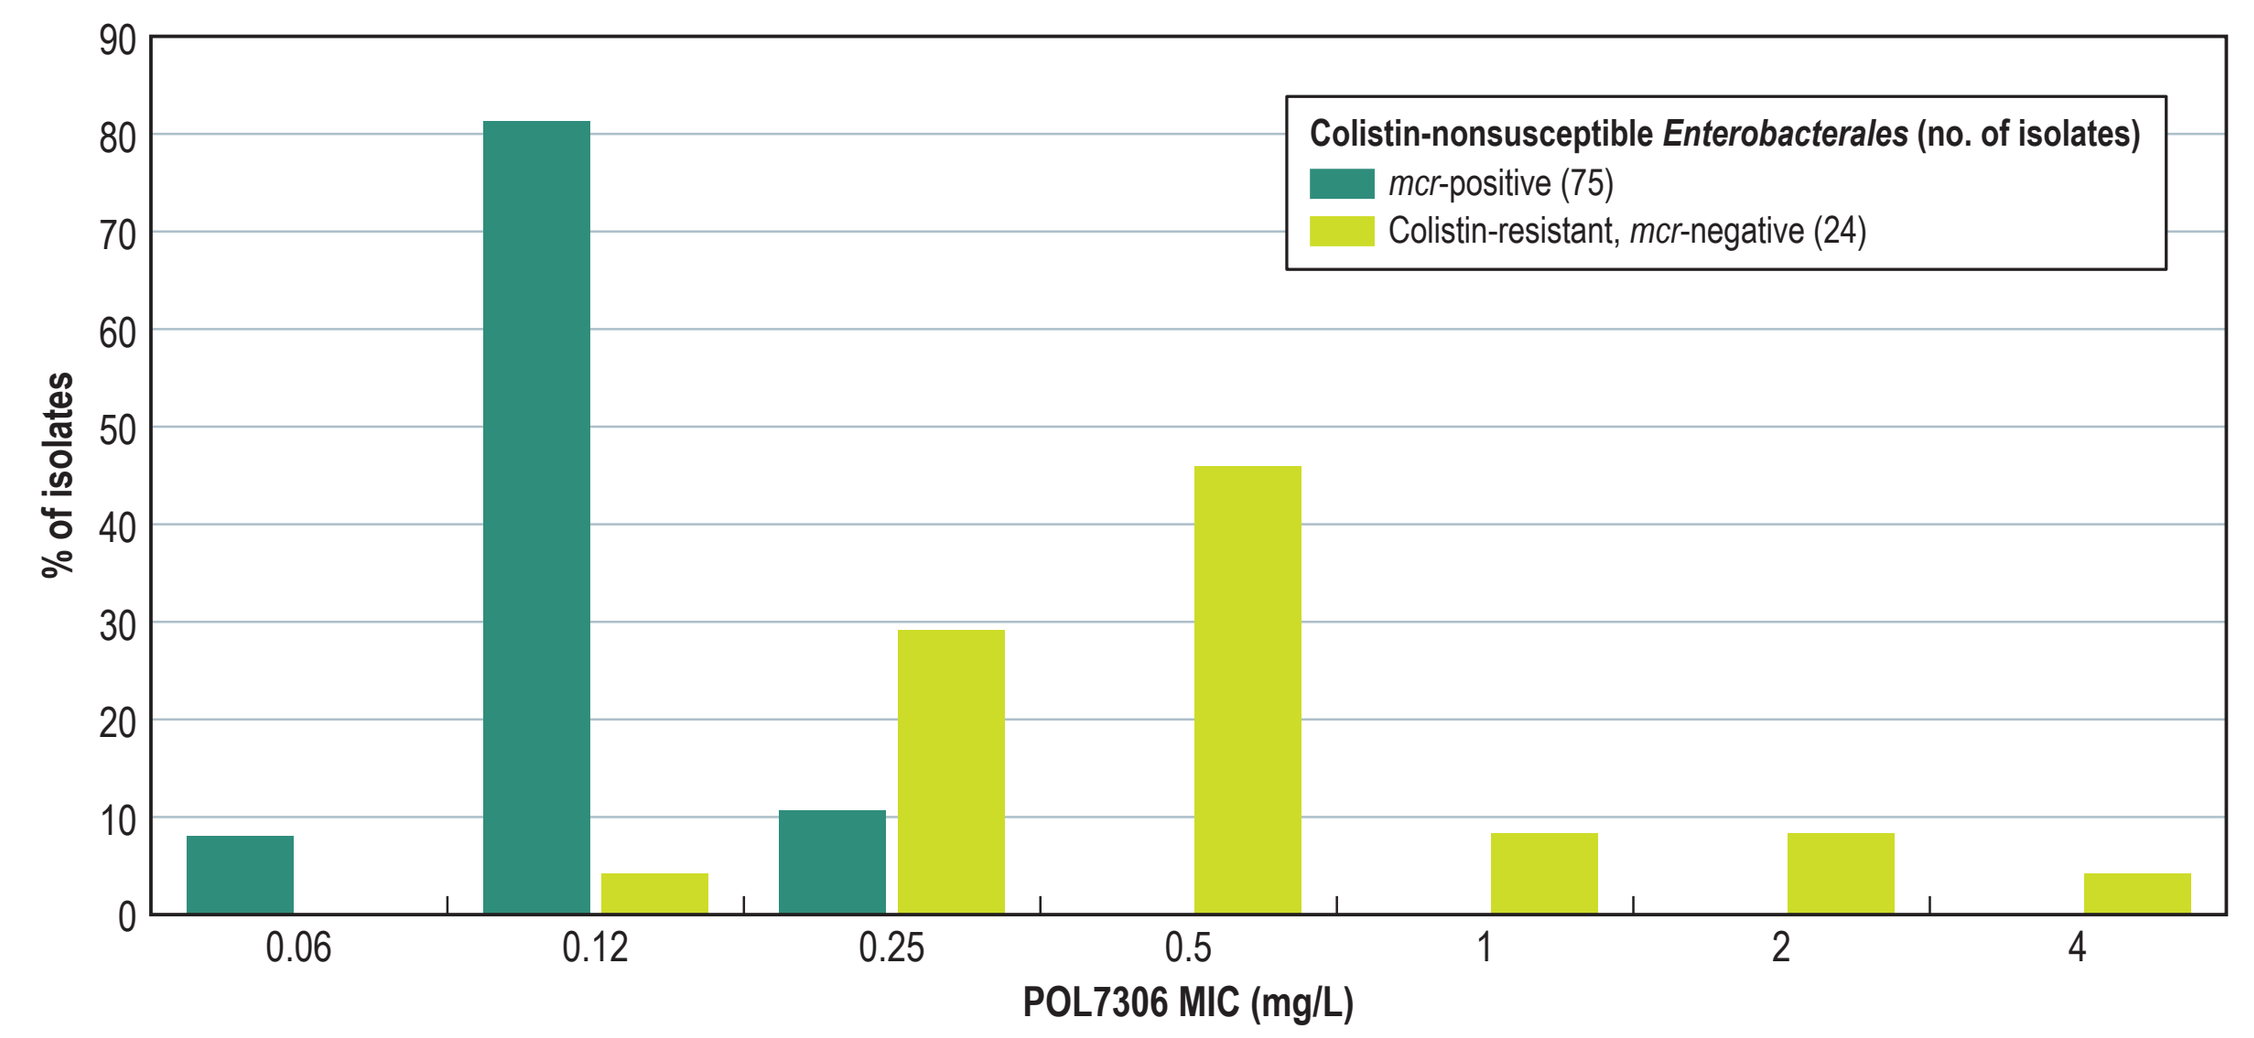
% of isolates
POL7306 MIC (mg/L)
0.06
0.12
0.25
0.5
1
2
4
Colistin-nonsusceptible Enterobacterales (no. of isolates)
mcr-positive (75)
Colistin-resistant, mcr-negative (24)

Table 1 Antimicrobial activity of POL7306 and comparator agents tested against *Enterobacterales* isolates

Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %R
Enterobacterales (272)[b]						
POL7306[c]	0.06	0.25				
Colistin	0.12	0.25			96.0	4.0
Ceftriaxone	0.12	>8	79.0	20.2	79.0	20.2
Ceftazidime	≤0.25	>16	83.5	16.2	79.8	16.5
Ceftazidime-avibactam	0.12	0.5	100.0	0.0	100.0	0.0
Piperacillin-tazobactam	2	64	85.6	7.7	82.3	14.4
Meropenem	≤0.12	≤0.12	98.9	0.7	99.3	0.7
Levofloxacin	≤0.5	>4	83.8	11.0	83.5	14.7
Gentamicin	≤1	2	91.9	8.1	91.5	8.1
Amikacin	2	4	98.9	0.7	98.5	1.1
CRE (53)[d]						
POL7306[c]	0.06	0.25				
Colistin	0.12	0.25	89.6[e]		90.6	9.4
Ceftriaxone	>8	>8	1.9	96.2	1.9	96.2
Ceftazidime	>16	>16	5.7	94.3	1.9	94.3
Ceftazidime-avibactam	2	8	90.6	9.4	90.6	9.4
Piperacillin-tazobactam	>128	>128	0.0	100.0	0.0	100.0
Meropenem	16	>32	0.0	100.0	0.0	50.9
Levofloxacin	16	>16	20.8	75.5	11.3	86.8
Gentamicin	4	>16	52.8	37.7	49.1	47.2
Amikacin	4	32	81.1	7.5	66.0	18.9
mcr-positive *Enterobacterales* (75)[f]						
POL7306[c]	0.12	0.25				
Colistin	4	8	6.7[b]		6.7	93.3
Ceftazidime	≤0.25	>16	70.7	24.0	64.0	29.3
Ceftriaxone	≤0.25	>8	58.7	40.0	58.7	40.0
Ceftazidime-avibactam	0.12	0.25	100.0	0.0	100.0	0.0
Piperacillin-tazobactam	2	>64	84.0	10.7	80.0	16.0
Meropenem	≤0.12	≤0.12	98.7	1.3	98.7	0.0
Levofloxacin	>4	>4	29.3	61.3	21.3	73.3
Gentamicin	≤2	>8	61.3	38.7	60.0	38.7
Amikacin	2	4	95.9	4.1	93.2	4.1
Colistin-R, *mcr*-negative *K. pneumoniae* (24)						
POL7306 [c]	0.5	2				
Colistin	>8	>8	8.3[b]		8.3	91.7
Ceftazidime	>16	>16	4.2	91.7	4.2	95.8
Ceftriaxone	>8	>8	8.3	87.5	8.3	87.5
Ceftazidime-avibactam	0.5	4	95.8	4.2	95.8	4.2
Piperacillin-tazobactam	>64	>64	16.7	83.3	8.3	83.3
Meropenem	8	>32	33.3	58.3	41.7	50.0
Levofloxacin	>4	>4	12.5	87.5	8.3	87.5
Gentamicin	8	>8	45.8	45.8	45.8	54.2
Amikacin	16	>32	58.3	16.7	45.8	41.7

[a] Criteria as published by CLSI 2018 and EUCAST 2018.
[b] Organisms include: *Citrobacter freundii* species complex (31), *Enterobacter aerogenes* (30), *E. cloacae* (10), *E. cloacae* species complex (20), *Escherichia coli* (50), group B *Salmonella enterica* (3), group D *Salmonella enterica* (1), *Klebsiella oxytoca* (32), *K. pneumoniae* (50), *Salmonella enterica* (2), *S. enterica* subsp. *enterica serovar paratyphi* (1), *S. enterica* subsp. *enterica serovar typhi* (2), *Shigella flexneri* (7), *S. sonnei* (15), unspeciated *Salmonella* (16), unspeciated *Shigella* (2).
[c] Tested in media supplemented with polysorbate 80.
[d] CRE, carbapenem-resistant *Enterobacterales*. Organisms include: *Citrobacter freundii* species complex (3), *Enterobacter aerogenes* (7), *E. cloacae* species complex (14), *Escherichia coli* (7), *Klebsiella oxytoca* (2), *K. pneumoniae* (20).
[e] Percentage of wild type based on epidemiologic cutoff value. CLSI M100 (2018).
[f] Includes *Escherichia coli* (71), *Klebsiella pneumoniae* (4).

Table 2 Antimicrobial activity of POL7306 and comparator agents tested against non-fermentative gram-negative bacilli

Organism / antimicrobial agent (no. tested)	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %R
P. aeruginosa (154)						
POL7306[b]	0.25	0.25				
Colistin	1	1	98.7	1.3	98.7	1.3
Ceftazidime	16	>16	46.1	35.7	46.1	53.9
Ceftazidime-avibactam	4	32	77.9	22.1	77.9	22.1
Piperacillin-tazobactam	32	>128	35.7	31.8	35.7	64.3
Meropenem	8	>32	39.6	53.2	39.6	44.2
Levofloxacin	8	>16	33.8	57.1	26.6	73.4
Gentamicin	8	>16	49.4	27.3	49.4	50.6
Amikacin	8	>32	78.6	13.6	63.6	21.4
A. baumannii (105)						
POL7306[b]	0.06	0.12				
Colistin	0.25	2	91.4	8.6	91.4	8.6
Ceftazidime	>16	>16	6.7	91.4		
Ceftazidime-avibactam	32	>32				
Piperacillin-tazobactam	>128	>128	0.0	98.1		
Meropenem	>32	>32	4.8	92.4	4.8	92.4
Levofloxacin	>16	>16	1.9	96.2	0.0	100.0
Gentamicin	>16	>16	11.4	79.0	11.4	88.6
Amikacin	>32	>32	25.7	69.5	23.8	74.3
S. maltophilia (51)						
POL7306[b]	0.06	0.25				
Colistin	8	>8				
Ceftazidime	>16	>16	17.6	74.5		
Ceftazidime-avibactam	32	>32				
Levofloxacin	1	4	78.4	9.8		
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	96.1	3.9	98.0	2.0

[a] Criteria as published by CLSI 2018 and EUCAST 2018.
[b] Tested in media supplemented with polysorbate 80.

Conclusions

- POL7306 demonstrated potent activity against a large collection of gram-negative organisms collected worldwide
- POL7306 spectrum of activity included colistin-resistant, XDR, and ESBL- and carbapenemase-producing *Enterobacterales* isolates for which there are currently limited treatment options
- POL7306 was also highly active against MDR and XDR isolates of *P. aeruginosa*, *A. baumannii*, and *S. maltophilia*

Acknowledgements

This study was supported by Polyphor Ltd. (Switzerland) and the Novo REPAIR Impact Fund.

References

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibilty tests for bacteria that grow aerobically; approved standard - eleventh edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.

EUCAST (2018). Breakpoint tables for interpretation of MIC's and zone diameters. Version 8.0, January 2018. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_8.0_Breakpoint_Tables.pdf. Accessed January 2018.

Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clinical Microbiololy and Infection* 18:268-281.

Srinivas N, Jetter P, Ueberbacher BJ, et al. (2010). Peptidomimetic antibiotics target outer-membrane biogenesis in *Pseudomonas aeruginosa*. *Science* 327: 1010-1013.

Contact

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID19-POL7306-global-gram-negatives.pdf

Charges may apply. No personal information is stored.

ECCMID 2019, April 13–16, 2019, Amsterdam, the Netherlands