ECCMID 2019 | Poster #P1170

In Vitro Activity of the Extended-Spectrum ß-Lactamase Inhibitor Enmetazobactam (formerly AAI101), in Combination with Cefepime, against 90 Molecularly Characterized Enterobacteriaceae Isolates Expressing a Variety of Non-ß-Lactam Resistance Mechanisms

Michael D. Huband[1], Amy A. Watters[1], Jill M. Lindley[1], Lalitagauri M. Deshpande[1], A Belley[2], Philipp Knechtle[2], Robert K. Flamm[1]
[1] JMI Laboratories, North Liberty, Iowa, USA; [2] Allecra Therapeutics SAS, Saint-Louis, France

Introduction

- Development of resistance due to the spread of extended-spectrum ß-lactamases (ESBLs) among Enterobacteriaceae has created the need for new therapeutic modalities
- Enmetazobactam (formerly AAI101) is a novel extended-spectrum ß-lactamase inhibitor that is highly active against ESBLs and other ß-lactamases
- *In vitro*, cefepime-enmetazobactam outperforms piperacillin-tazobactam against ESBL-producing isolates of Enterobacteriaceae, and is as potent as meropenem
- Enmetazobactam, in combination with cefepime, is intended as empiric treatment of serious Gram-negative infections in settings with an elevated prevalence of ESBL-producing Enterobacteriaceae
- Cefepime (2 g) combined with enmetazobactam (0.5 g) administered q8h has recently entered Phase 3 clinical trials in patients with complicated urinary infections, including pyelonephritis
- In this study, the combination of cefepime-enmetazobactam (enmetazobactam at fixed 8 mg/L) and comparator agents were tested against 90 molecularly characterized Enterobacteriaceae isolates containing a variety of non ß-lactam resistance mechanisms
- Non-ß-lactam resistance mechanisms included: aminoglycoside resistance, fluoroquinolone resistance, colistin resistance (intrinsic and plasmid-mediated), tetracycline-resistance, and resistance due to porin alterations/deletions
- Non ß-lactam resistance isolates also expressed ß-lactam resistance mechanisms including: ESBLs (phenotype/genotype), AmpCs (plasmid encoded), and OXA-type ß-lactamases

Materials and Methods

- A challenge set of 90 molecularly characterized Enterobacteriaceae isolates exhibiting phenotypic cross-resistance to numerous antibiotic classes were recovered from patients with documented infections during 2013–2017 and consisted of 43 *Escherichia coli*, 34 *Klebsiella pneumoniae*, 3 *Citrobacter freundii* species complex, 3 *Proteus mirabilis*, 2 *Enterobacter cloacae* species complex, 2 *Providencia stuartii*, 2 *Serratia marcescens*, and 1 *Klebsiella oxytoca*
- Isolates were collected in 56 medical centres located in 19 countries and 9 US census divisions: the United States (29 medical centres; 48 isolates; 53.3% overall), Europe (14 medical centres; 25 isolates; 27.8% overall), Latin America (7 medical centres; 9 isolates; 10.0% overall), and the Asia-Pacific region (6 medical centres; 8 isolates; 8.9% overall)
- Organisms were identified by matrix-assisted laser desorption ionization–time of flight mass spectrometry and molecularly characterized via next-generation sequencing
- Reference broth microdilution susceptibility testing was conducted using frozen-form panels produced by JMI Laboratories according to CLSI M07 (2018) guidelines using cation-adjusted Mueller-Hinton broth
- Isolates were tested for susceptibility to cefepime-enmetazobactam (enmetazobactam fixed at 8 mg/L), cefepime, ceftazidime, ciprofloxacin, colistin, gentamicin, meropenem, piperacillin-tazobactam, tetracycline, and trimethoprim-sulfamethoxazole
- Isolates containing class A carbapenemases and/or class B metallo-ß-lactamases were excluded from the study due to the limited activity of cefepime-enmetazobactam against these resistance mechanisms
- For comparison purposes, CLSI susceptible-dose-dependent (SDD) breakpoint interpretive criteria for cefepime (susceptible MIC ≤8 mg/L) were also applied to cefepime-enmetazobactam (fixed 8 mg/L) combinations

Results

- Cefepime-enmetazobactam ($MIC_{50/90}$, 0.12/2 mg/L) was the most active compound tested based on MIC_{90} values against 90 molecularly characterized Enterobacteriaceae isolates (Table 1)
- Applying the CLSI SDD breakpoint of 8 mg/L to cefepime-enmetazobactam (fixed 8 mg/L) resulted in 97.8% (88/90) of the isolates testing susceptible (S) compared to 50.0%S for cefepime alone, 86.7%S for meropenem, and 58.9%S for piperacillin-tazobactam (Table 2)
- Cefepime-enmetazobactam (fixed 8 mg/L) was the most active compound tested against aminoglycoside-R ($MIC_{50/90}$, 0.12/2 mg/L; 100.0%S), colistin-R ($MIC_{50/90}$, 0.25/8 mg/L; 92.0%S), fluoroquinolone-R ($MIC_{50/90}$, 0.12/4 mg/L; 95.9%S), and tetracycline-R ($MIC_{50/90}$, 0.12/2 mg/L; 98.0%S) Enterobacteriaceae isolates (Table 2 and Figures 1–4)
- Cefepime-enmetazobactam (fixed 8 mg/L) was the most active compound tested against molecularly characterized Enterobacteriaceae isolates containing an AmpC cephalosporinase ($MIC_{50/90}$, 0.25/4 mg/L; 94.1%S), ESBL phenotype/genotype ($MIC_{50/90}$, 0.12/2 mg/L; 96.8%S), OXA-ß-lactamase ($MIC_{50/90}$, 0.12/4 mg/L; 94.1%S), and/or porin alterations/deletions ($MIC_{50/90}$, 0.5/16 mg/L; 87.5%S) (Table 2)
- Cefepime-enmetazobactam (fixed 8 mg/L; 97.8%S) outperformed cefepime, meropenem, and piperacillin-tazobactam against 90 molecularly characterized Enterobacteriaceae isolates
 - MIC_{90} values for cefepime, meropenem, and piperacillin-tazobactam were in the resistant interpretive category with values of >64 mg/L (50.0%S), 4 mg/L (86.7%S), and >512 mg/L (58.9%S), respectively
- 37.8% of the Enterobacteriaceae isolates tested were aminoglycoside resistant (R), 27.8% were colistin R, 54.4% were fluoroquinolone R, and 55.6% were tetracycline R (Table 2)
- 18.9% of the 90 molecularly characterized isolates contained an AmpC cephalosporinase, 68.9% had an ESBL phenotype or genotype, 37.8% had an OXA-type ß-lactamase, and 17.8% contained porin mutations (Table 2)

Figure 1 Cumulative percent inhibition results for cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 34 aminoglycoside-resistant Enterobacteriaceae isolates

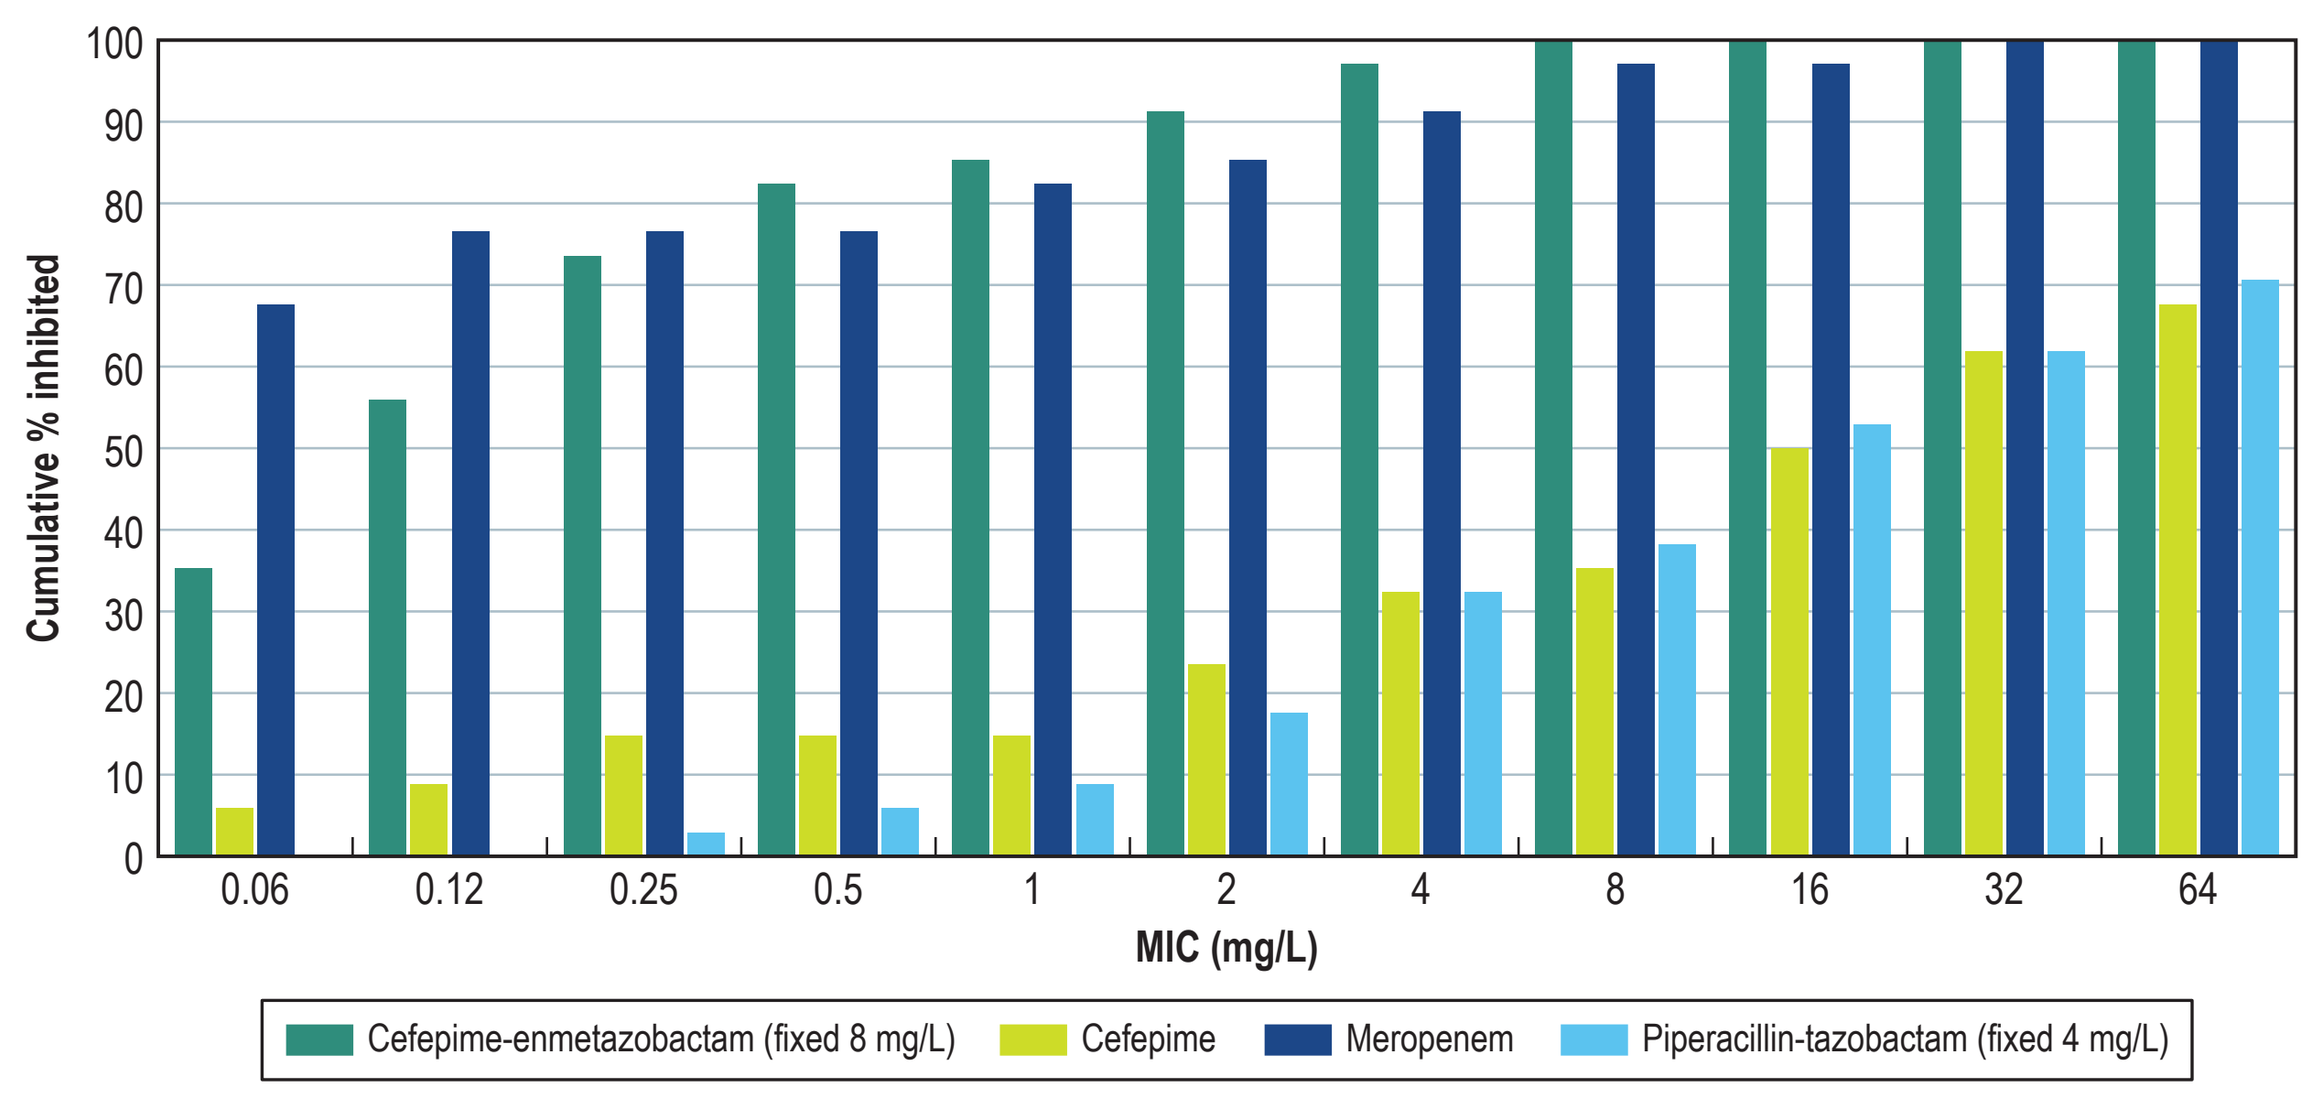
Cumulative % inhibited
0
10
20
30
40
50
60
70
80
90
100
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
MIC (mg/L)
Cefepime-enmetazobactam (fixed 8 mg/L)
Cefepime
Meropenem
Piperacillin-tazobactam (fixed 4 mg/L)

Figure 2 Cumulative percent inhibition results for cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 25 colistin-resistant Enterobacteriaceae isolates

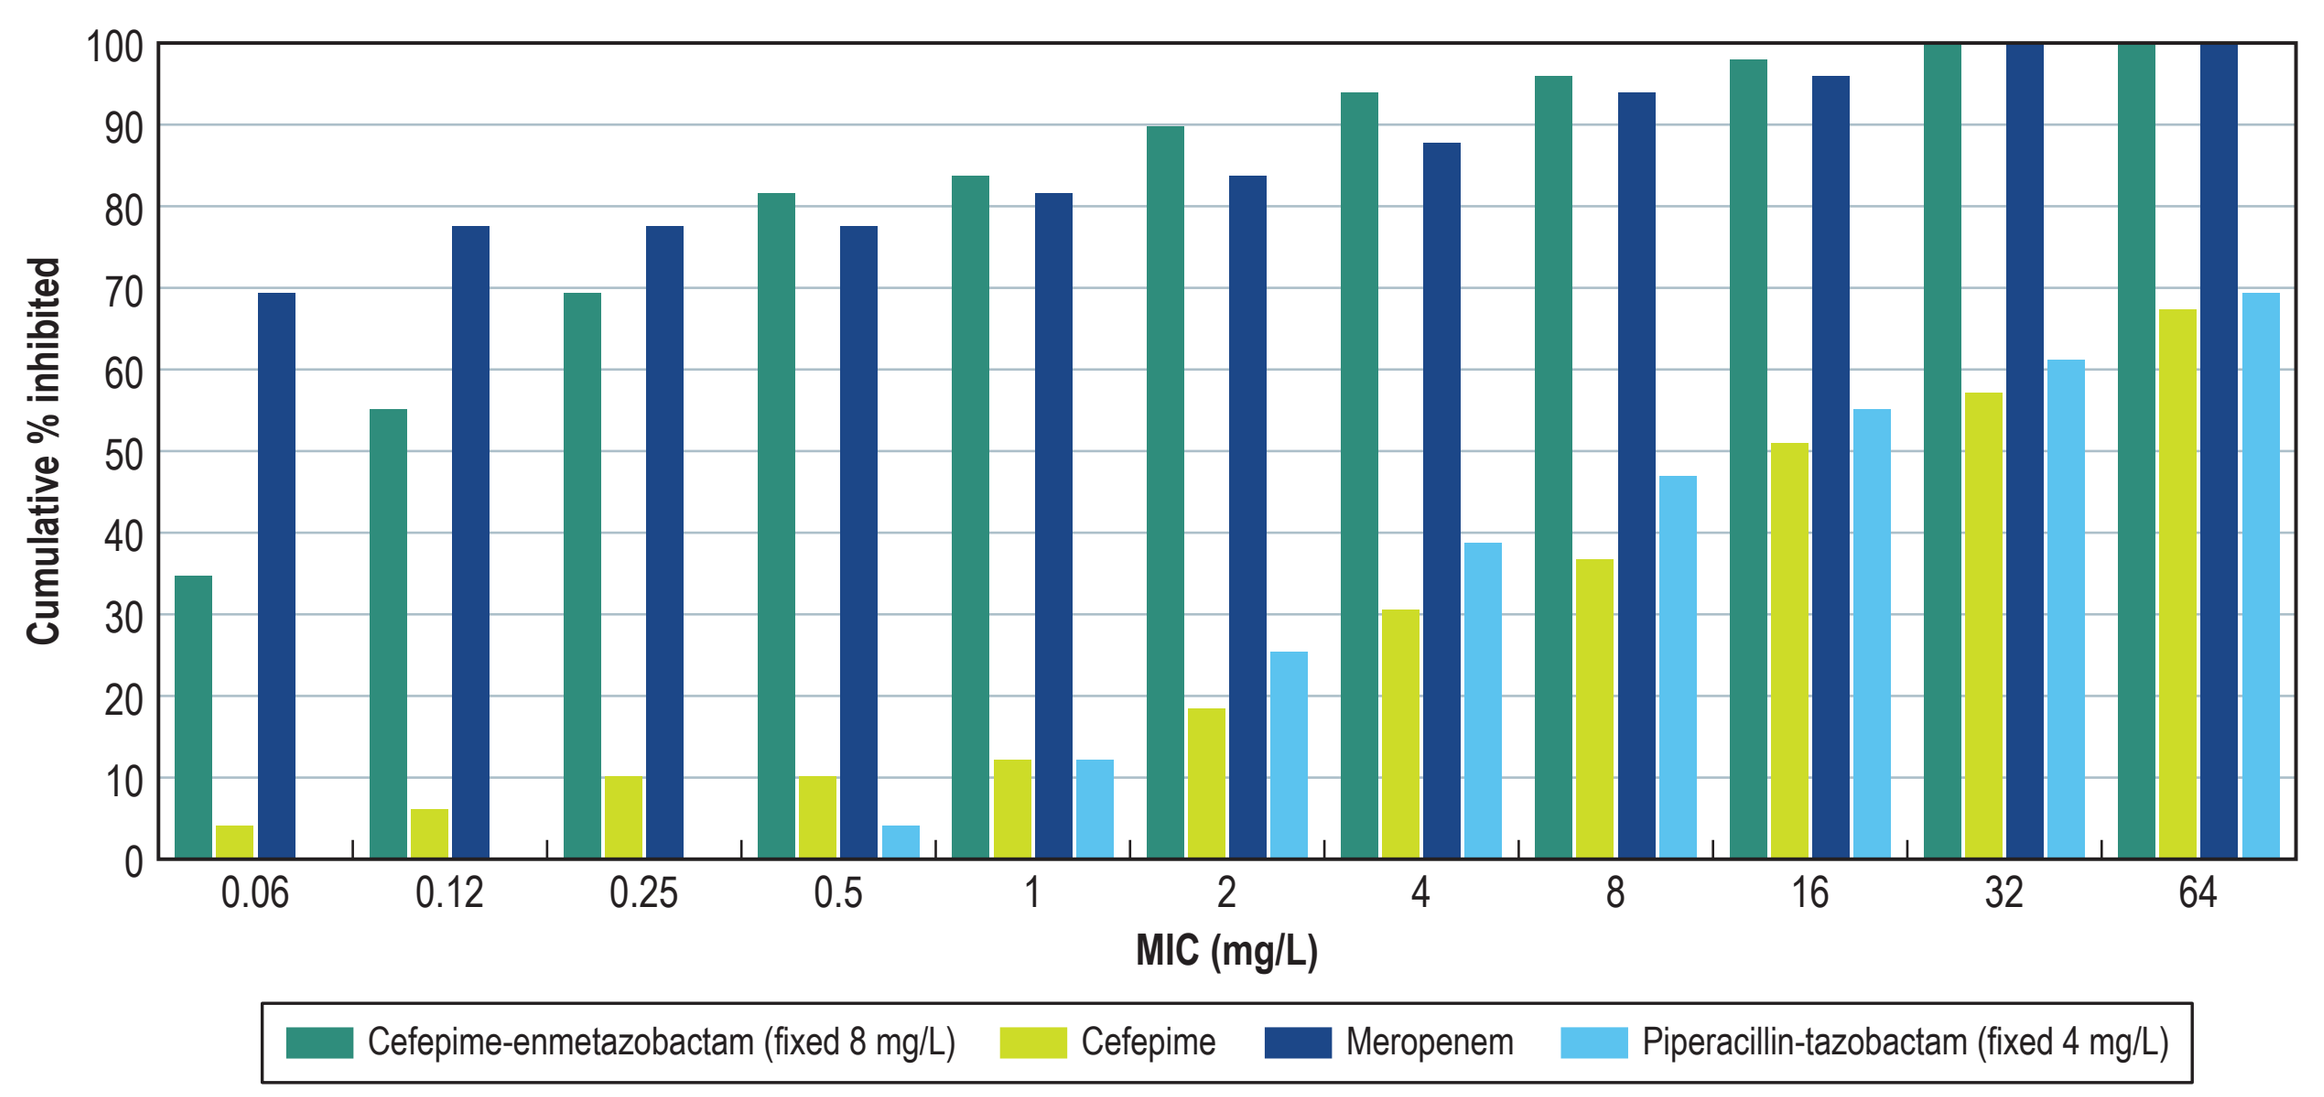
Cumulative % inhibited
0
10
20
30
40
50
60
70
80
90
100
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
MIC (mg/L)
Cefepime-enmetazobactam (fixed 8 mg/L)
Cefepime
Meropenem
Piperacillin-tazobactam (fixed 4 mg/L)

Figure 3 Cumulative percent inhibition results for cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 49 fluoroquinolone-resistant Enterobacteriaceae isolates

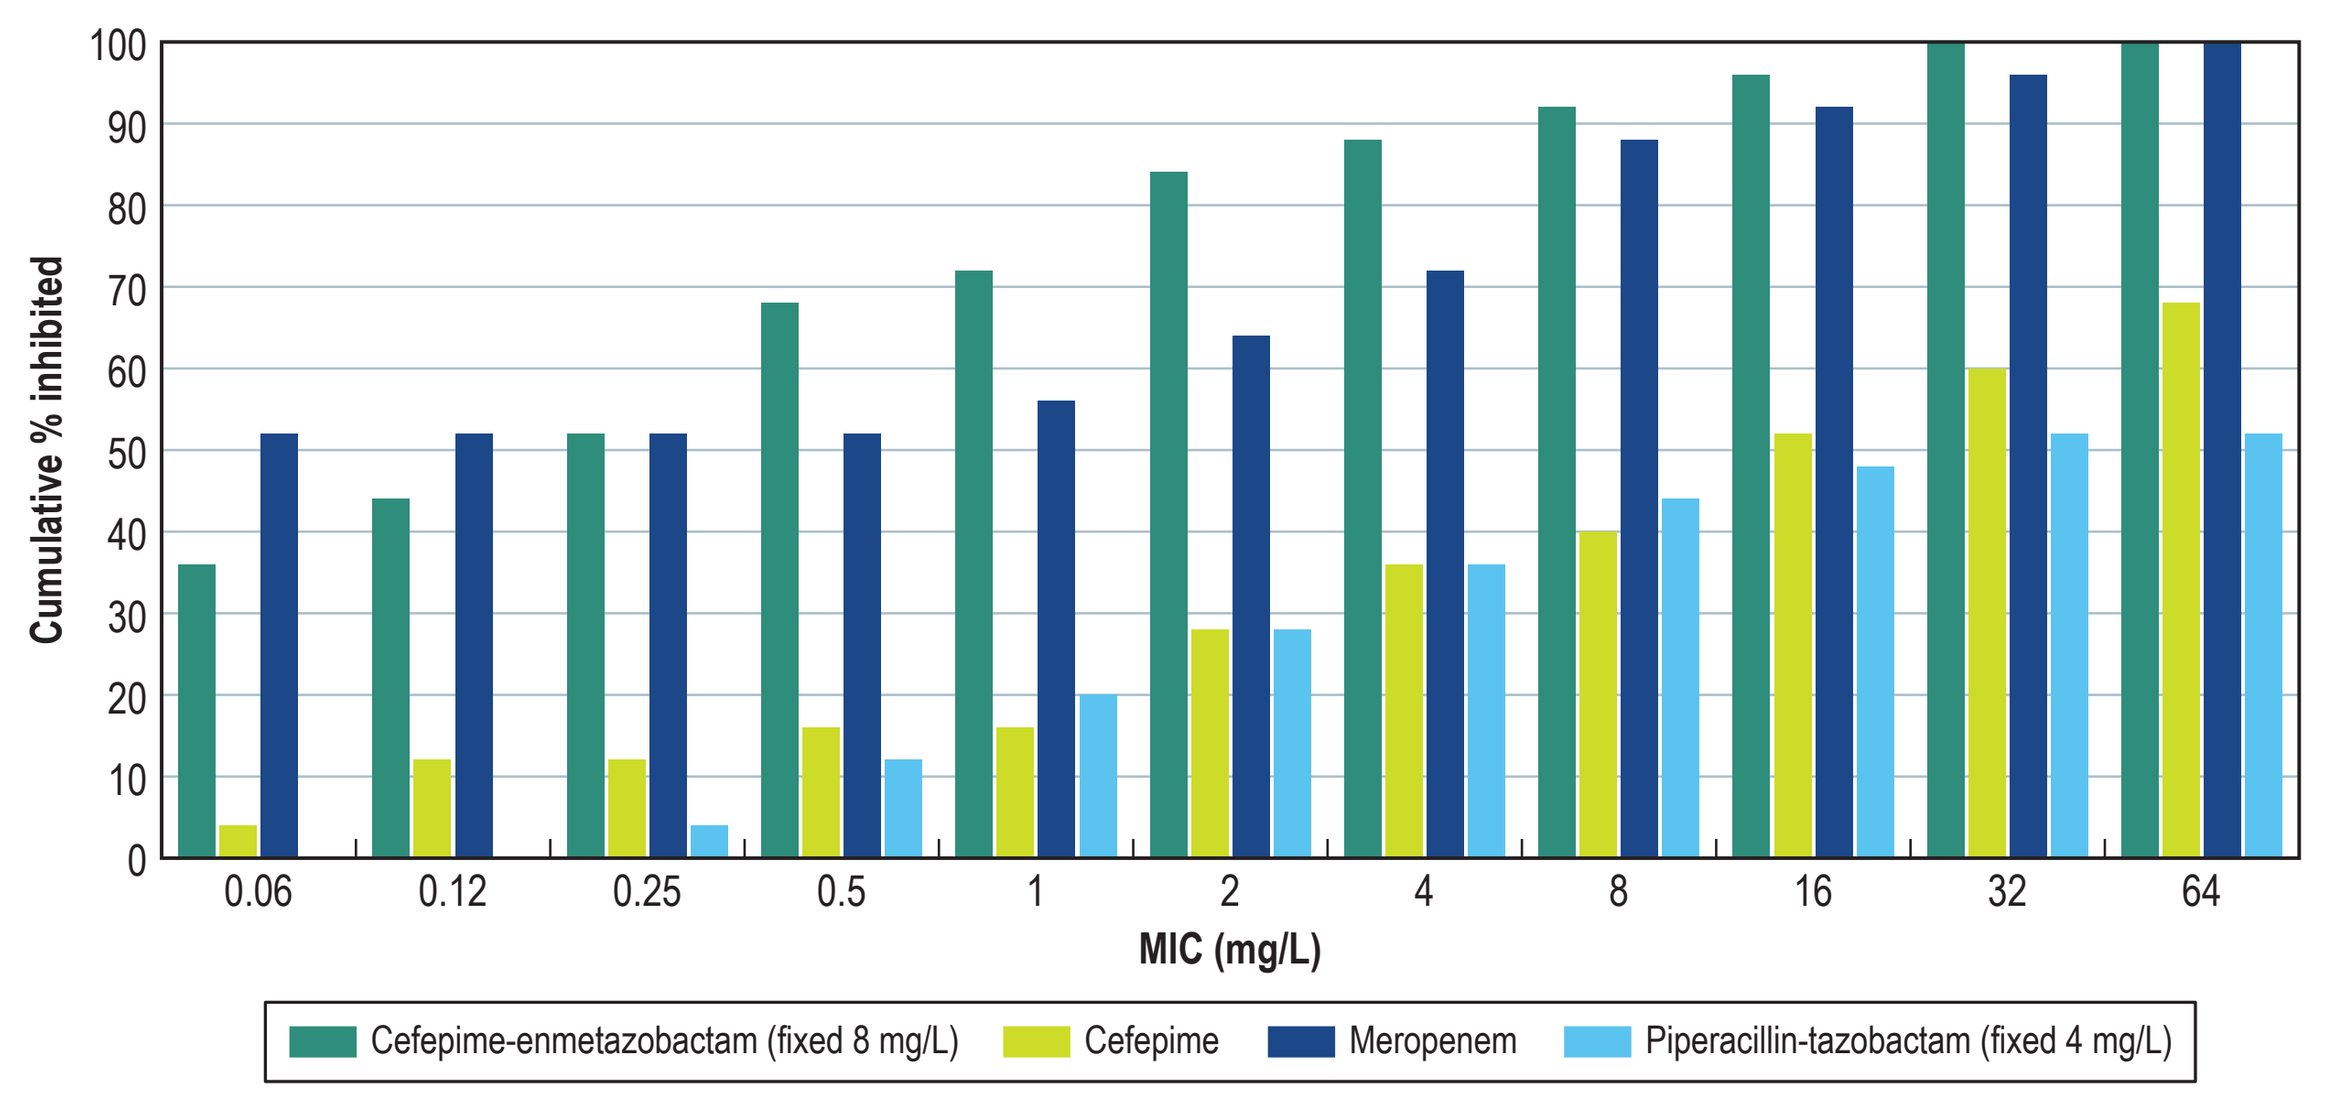
Cumulative % inhibited
0
10
20
30
40
50
60
70
80
90
100
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
MIC (mg/L)
Cefepime-enmetazobactam (fixed 8 mg/L)
Cefepime
Meropenem
Piperacillin-tazobactam (fixed 4 mg/L)

Figure 4 Cumulative percent inhibition results for cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 50 tetracycline-resistant Enterobacteriaceae isolates

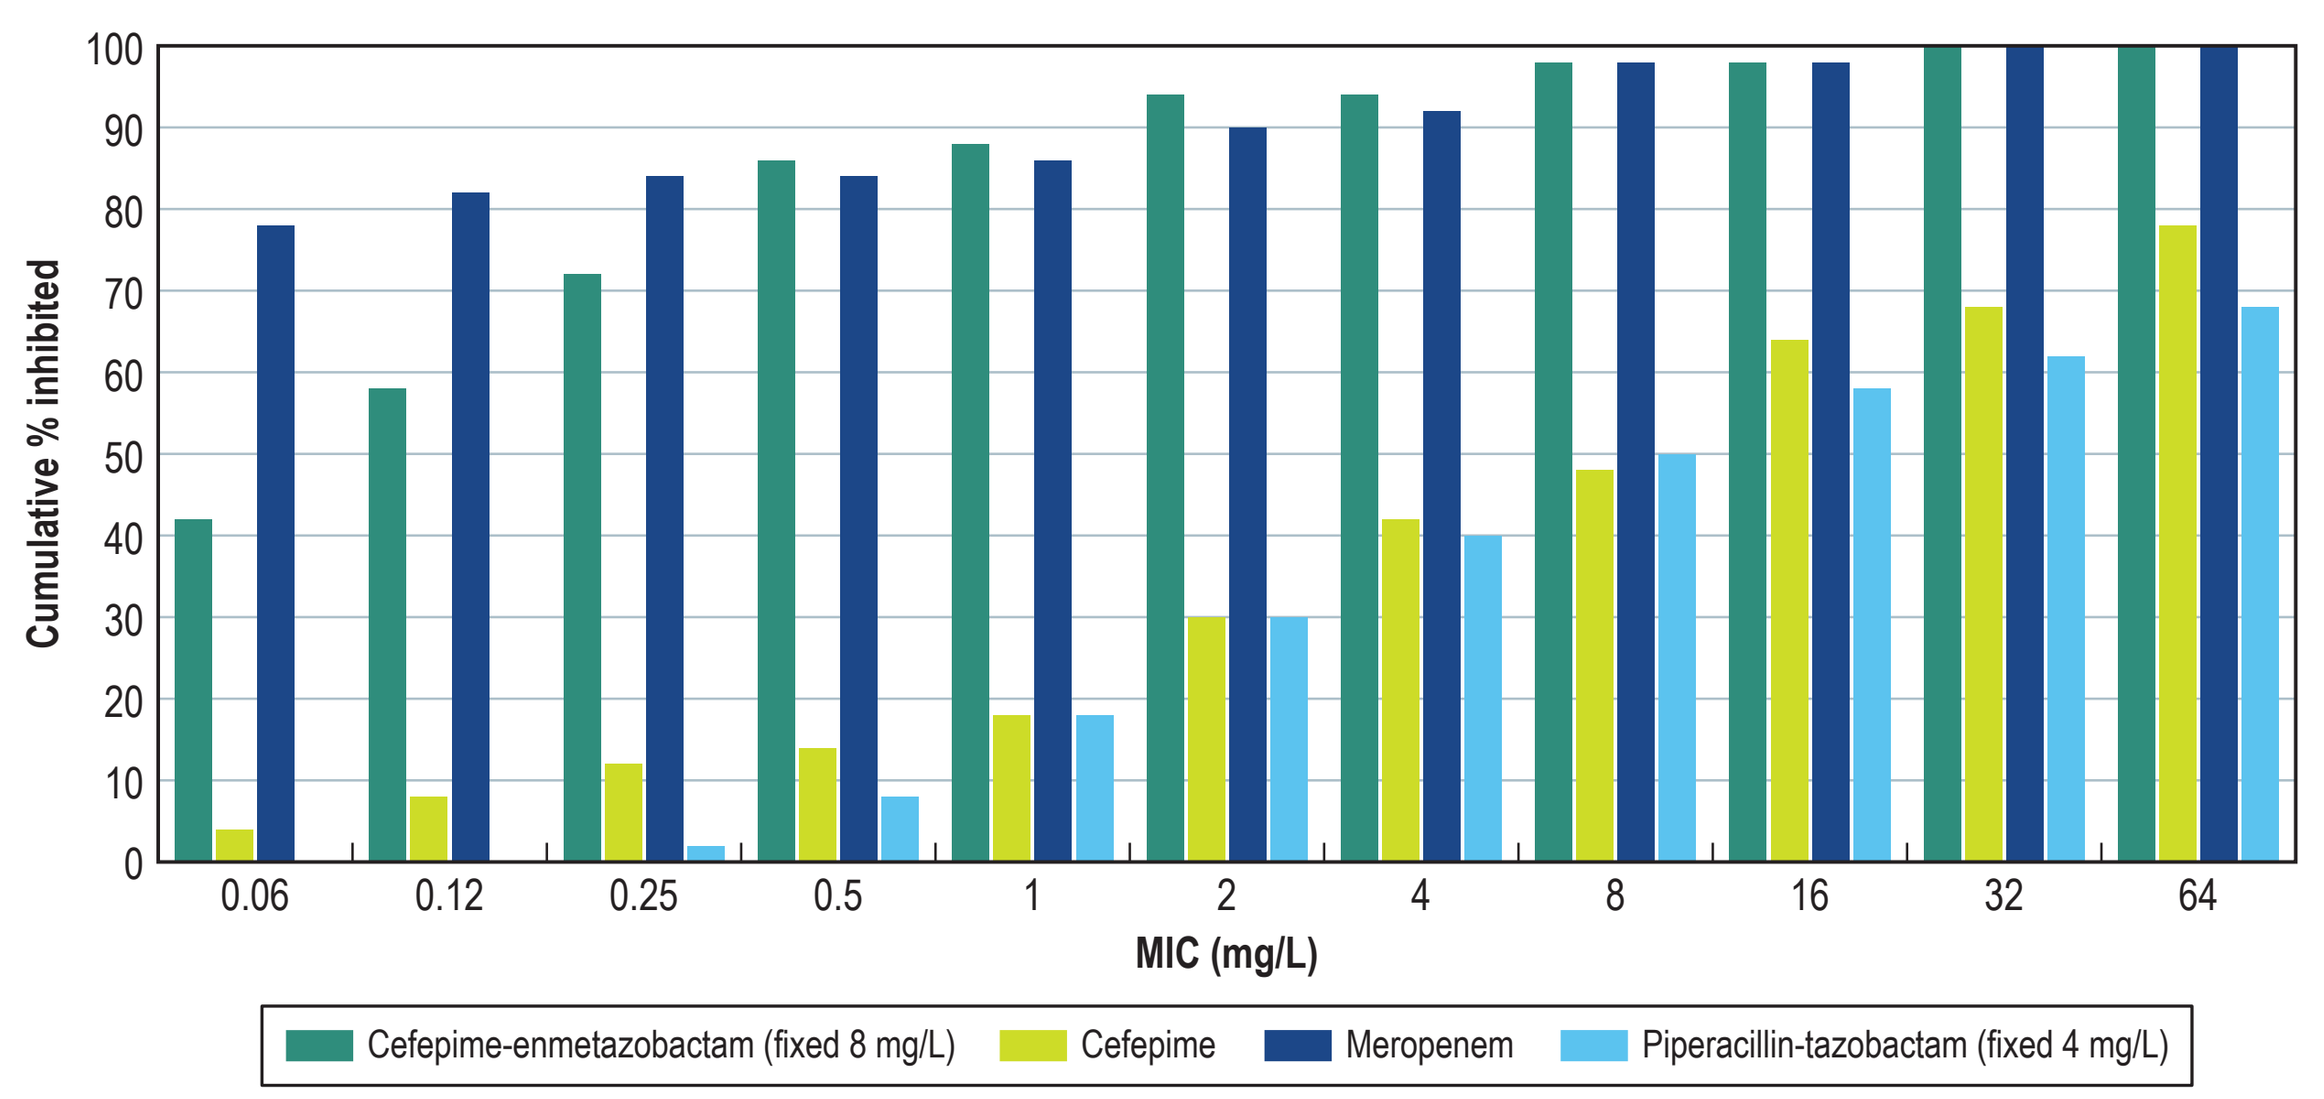
Cumulative % inhibited
0
10
20
30
40
50
60
70
80
90
100
0.06
0.12
0.25
0.5
1
2
4
8
16
32
64
MIC (mg/L)
Cefepime-enmetazobactam (fixed 8 mg/L)
Cefepime
Meropenem
Piperacillin-tazobactam (fixed 4 mg/L)

Table 1 Cumulative percent inhibition results for cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 90 molecularly characterized Enterobacteriaceae isolates

Organism/organism group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of:																			MIC_{50}	MIC_{90}[c]
	≤0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	64	128	256	512	>[a]		
All Enterobacteriaceae (90)																					
Cefepime-enmetazobactam (fixed 8 mg/L) (90)		0 0.0	2 2.2	13 16.7	26 45.6	17 64.4	9 74.4	10 85.6	1 86.7	**5 92.2**	3 95.6	2 97.8	1 98.9	1 100.0						0.12	2
Cefepime (90)			0 0.0	1 1.1	3 4.4	3 7.8	6 14.4	4 18.9	2 21.1	9 31.1	8 40.0	9 50.0	14 65.6	4 70.0	7 77.8				20 100.0	8	>64
Ciprofloxacin (90)	0 0.0	5 5.6	8 14.4	2 16.7	3 20.0	2 22.2	3 25.6	6 32.2	7 40.0	5 45.6	2 47.8	3 51.1	6 57.8						38 100.0	8	>16
Colistin (90)						1 1.1	49 55.6	14 71.1	0 71.1	1 72.2	3 75.6	4 80.0	4 84.4						14 100.0	0.25	>16
Gentamicin (90)						2 2.2	15 18.9	26 47.8	9 57.8	1 58.9	0 58.9	3 62.2	4 66.7	11 78.9	**10 90.0**				9 100.0	1	64
Meropenem (90)		0 0.0	11 12.2	35 51.1	20 73.3	6 80.0	1 81.1	2 83.3	3 86.7	2 88.9	**2 91.1**	4 95.6	1 96.7	2 98.9	1 100.0					0.03	4
Piperacillin-tazobactam (90)							1 1.1	3 4.4	6 11.1	17 30.0	13 44.4	7 52.2	6 58.9	5 64.4	5 70.0	4 74.4	5 80.0	5 85.6	13 100.0	8	>512
Tetracycline (90)								0 0.0	7 7.8	23 33.3	6 40.0	4 44.4	3 47.8	0 47.8	7 55.6				40 100.0	64	>64
Trimethoprim-sulfamethoxazole (90)							36 40.0	9 50.0	5 55.6	0 55.6	3 58.9	0 58.9							37 100.0	0.5	>8

[a] MIC value is greater than the highest concentration tested.
[b] Composed of 43 *Escherichia coli*, 34 *Klebsiella pneumoniae*, 3 *Citrobacter freundii* species complex, 3 *Proteus mirabilis*, 2 *Enterobacter cloacae* species complex, 2 *Providencia stuartii*, 2 *Serratia marcescens*, and 1 *Klebsiella oxytoca*.
[c] MIC_{90} value listed in bold.

Table 2 Activity of cefepime-enmetazobactam (fixed 8 mg/L) and comparators against 90 recent molecularly characterized Enterobacteriaceae isolates

Organism/group (n)	$MIC_{50/90}$ (mg/L)/% susceptible (CLSI)[a]								
	FPE[b]	FEP[c]	CIP	COL[d]	GEN	MEM	PIP-TAZ	TMP-SMX	TET
Enterobacteriaceae (90)	0.12 / 2 **97.8%**[e]	8 / >64 50.0%	8 / >16 40.0%	0.25 / >16 72.2%	1 / 64 58.9%	0.03 / 4 86.7%	8 / >512 58.9%	0.5 / >8 55.6%	64 / >64 40.0%
Aminoglycoside resistant (34)[f]	0.12 / 2 **100.0%**	16 / >64 35.3%	>16 / >16 8.8%	0.5 / >16 55.9%	64 / >64 0.0%	0.06 / 4 82.4%	16 / >512 52.9%	>8 / >8 32.4%	64 / >64 35.3%
Colistin resistant (25)[g]	0.25 / 8 **92.0%**	16 / >64 40.0%	16 / >16 20.0%	>16 / >16 0.0%	16 / >64 36.0%	0.06 / 16 56.0%	32 / >512 48.0%	>8 / >8 36.0%	64 / >64 24.0%
Fluoroquinolone resistant (49)[h]	0.12 / 4 **95.9%**	16 / >64 36.7%	>16 / >16 0.0%	0.5 / >16 63.3%	32 / >64 38.8%	0.06 / 8 81.6%	16 / >512 55.1%	>8 / >8 38.8%	>64 / >64 34.7%
Tetracycline resistant (50)[i]	0.12 / 2 **98.0%**	16 / >64 48.0%	16 / >16 32.0%	0.5 / >16 64.0%	1 / 64 56.0%	0.03 / 2 86.0%	8 / >512 58.0%	>8 / >8 44.0%	>64 / >64 0.0%
Amp-C (17)[j]	0.25 / 4 **94.1%**	2 / >64 70.6%	1 / >16 52.9%	0.25 / >16 70.6%	1 / 64 70.6%	0.06 / 8 76.5%	128 / >512 35.3%	1 / >8 52.9%	16 / >64 47.1%
ESBL (62)[k]	0.12 / 2 **96.8%**	16 / >64 29.0%	>16 / >16 29.0%	0.25 / >16 74.2%	1 / >64 54.8%	0.03 / 8 85.5%	16 / >512 56.5%	1 / >8 51.6%	64 / >64 41.9%
OXA-1/30, 2, 9, 10, and/or 48 (34)[l]	0.12 / 4 **94.1%**	16 / >64 41.2%	>16 / >16 20.6%	0.5 / >16 58.8%	32 / >64 32.4%	0.06 / 8 73.5%	64 / >512 35.3%	>8 / >8 35.3%	16 / >64 38.2%
Porin mutations (16)[m]	0.5 / 16 87.5%	64 / >64 31.2%	>16 / >16 6.2%	16 / >16 31.2%	16 / >64 37.5%	0.06 / 16 56.2%	256 / >512 25.0%	>8 / >8 25.0%	16 / >64 37.5%
Multidrug-resistant (46)[n]	0.25 / 4 **95.7%**	16 / >64 34.8%	>16 / >16 10.9%	0.5 / >16 50.0%	32 / >64 23.9%	0.06 / 8 76.1%	32 / >512 41.3%	>8 / >8 34.8	>64 / >64 26.1%

[a] FPE, cefepime-enmetazobactam; FEP, cefepime; CIP, ciprofloxacin; COL, colistin; GEN, gentamicin; MEM, meropenem; PIP-TAZ, piperacillin-tazobactam; TMP-SMX, trimethoprim-sulfamethoxazole; TET, tetracycline; ESBL, extended-spectrum ß-lactamase.
[b] Cefepime susceptible-dose-dependent breakpoint for Enterobacteriaceae applied to cefepime-enmetazobactam (fixed 8 mg/L) for comparison purposes only.
[c] Cefepime susceptible-dose-dependent breakpoint applied.
[d] Percentage of wild type based on the epidemiological cutoff value for colistin.
[e] Susceptibility ≥90.0% listed in bold.
[f] Composed of *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (1), *Escherichia coli* (12), *Klebsiella pneumoniae* (16), *Proteus mirabilis* (2), and *Providencia stuartii* (2).
[g] Composed of *E. coli* (7), *K. pneumoniae* (11), *P. mirabilis* (3), *P. stuartii* (2), and *Serratia marcescens* (2).
[h] Composed of *C. freundii* species complex (1), *Escherichia coli* (25), *K. pneumoniae* (19), *P. mirabilis* (2), *P. stuartii* (2).
[i] Composed of *C. freundii* species complex (2), *E. cloacae* species complex (1), *Escherichia coli* (25), *K. pneumoniae* (16), *P. mirabilis* (3), *P. stuartii* (2), and *S. marcescens* (1)
[j] Composed of *E. coli* (10), *Klebsiella oxytoca* (1), and *K. pneumoniae* (6).
[k] Composed of *C. freundii* species complex (1), *E. cloacae* species complex (2), *Escherichia coli* (29), *K. oxytoca* (1), *K. pneumoniae* (28), and *P. mirabilis* (1).
[l] Composed of *E. cloacae* species complex (2), *Escherichia coli* (7), *K. oxytoca* (1), *K. pneumoniae* (22), *P. mirabilis* (1), and *S. marcescens* (1).
[m] Composed of *C. freundii* species complex (1), *E. coli* (4), *K. pneumoniae* (10), and *P. stuartii* (1).
[n] Composed of *C. freundii* species complex (1), *E. cloacae* species complex (1), *Escherichia coli* (16), *K. pneumoniae* (22), *P. mirabilis* (3), *P. stuartii* (2), and *S. marcescens* (1).

Conclusions

- The novel extended-spectrum ß-lactamase inhibitor enmetazobactam, combined with cefepime, was highly active against 90 recent molecularly characterized Enterobacteriaceae isolates expressing a variety of ß-lactam and non-ß-lactam resistance mechanisms
- Cefepime-enmetazobactam (enmetazobactam fixed at 8 mg/L) activity was unaffected by resistance mechanisms targeting aminoglycoside, fluoroquinolone, colistin, and tetracycline classes of antibacterial agents
- Cefepime-enmetazobactam (fixed 8 mg/L) outperformed cefepime, meropenem, and piperacillin-tazobactam against this collection of molecularly characterized Enterobacteriaceae isolates, including isolates containing AmpC, ESBL, and/or OXA-ß-lactamases
- Cefepime-enmetazobactam may represent a novel empiric option for treatment of serious Gram-negative infections in settings that have high rates of ESBL-producing Enterobacteriaceae and that restrict carbapenem use as part of antimicrobial stewardship measures

Acknowledgements

This study and poster presentation were funded by a grant from Allecra Therapeutics SAS (Saint-Louis, France).

References

Clinical and Laboratory Standards Institute (2018). *M100-S28. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07-A11. Methods for dilution antimicrobial susceptibilty tests for bacteria that grow aerobically; approved standard - eleventh edition*. Wayne, PA: CLSI.

Contact

Michael D. Huband
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID19-enmetazobactam-cefepime.pdf

Charges may apply. No personal information is stored.

ECCMID 2019, April 13–16, 2019, Amsterdam, the Netherlands