Debio 1452 MIC Quality Control Range with *Staphylococcus aureus* ATCC 29213 Using a Multi-laboratory Study Design

EV0763

JE ROSS, RN JONES, RK FLAMM
JMI Laboratories, North Liberty, IA, USA

ECCMID 2015
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
robert-flamm@jmilabs.com

ABSTRACT

Objectives: To conduct a study to establish MIC quality control (QC) ranges for Debio 1452 (formerly AFN-1252), the active form of the prodrug Debio 1450 (formerly AFN-1720) using the reference CLSI broth microdilution (BMD) method. This FabI inhibitor is being developed for the treatment of staphylococcal infections including those caused by methicillin-resistant *Staphylococcus aureus* (MRSA). Debio 1452 specifically targets the FabI enzyme in *Staphylococcus* spp. and demonstrates limited activity against other bacterial species.

Methods: An eight laboratory study design was compliant with CLSI M23-A3 guidelines. One QC strain was tested (*S. aureus* ATCC 29213 [SA]) using four media lots (three manufacturers) of cation-adjusted Mueller-Hinton broth (MHB). Frozen-form broth microdilution panels were manufactured at TREK Diagnostics (Cleveland, Ohio, USA). Ten replicate tests were performed generating 320 BMD MIC values. Rifampin was used as a control agent and all results were within the published QC limits (320 values). Colony counts were performed by each participating laboratory.

Results: A four log_2 dilution QC range of 0.002 – 0.015 mg/L was required for *S. aureus* with Debio 1452 due to a dominant "shoulder" MIC at 0.004 mg/L, which had 70.6% of the MIC values compared to the modal occurrences at 0.008 mg/L. Only one value was outside of the proposed range at 0.03 mg/L with 99.7% of all values included in the proposed range. Average colony count of all laboratories was 3.4 x 10^5 CFU/ml. No significant MIC differences were noted among MHB lots for Debio 1452. All media lot modes were either 0.004 (1) or 0.008 (3) mg/L. The results were analyzed using the Range Finder statistical program to identify outlier laboratories, but none were identified. Quality control results for the control agent, rifampin, ranged from 0.004-0.015 mg/L in complete agreement with the published CLSI QC range. The mode was 0.004 mg/L (67.8% of results) and 31.9% of values were at 0.008 mg/L. All media lots exhibited the identical mode for rifampin of 0.004 mg/L. The CLSI Subcommittee on Antimicrobial Susceptibility Testing approved this Debio 1452 QC range for *S. aureus* ATCC 29213 in January 2011 for publication after the selection of the compound's official chemical name.

Conclusions: Proposed MIC QC range for Debio 1452 should accurately guide microbiologic testing in research laboratories and in clinical or reference laboratories participating in the testing of clinical trial staphylococcal isolates when applying the CLSI BMD method.

INTRODUCTION

Debio 1452 (formerly AFN-1252) is an investigational FabI inhibitor active *in vitro* against staphylococci including *Staphylococcus aureus* (methicillin-susceptible and -resistant strains); see **Figure 1**. MIC_{90} values for Debio 1452 have been reported at 0.008 and 0.12 mg/L for *S. aureus* and coagulase-negative staphylococci, respectively. Currently, Debio 1450 (formerly AFN-1720; the prodrug of Debio 1452) is in clinical development for staphylococcal infections by Debiopharm International, SA.

Debio 1452 selectively inhibits bacterial enoyl-ACP reductase, an essential step in the elongation cycle of bacterial fatty acid biosynthesis. Among the four known enzyme forms of enoyl-ACP (FabI, FabK, FabL and FabV), an essential FabI is present in *S. aureus, S. epidermidis* and other coagulase-negative staphylococci. This specific-spectrum of Debio 1452 is beneficial to minimize the effect on normal bacterial flora thus reducing adverse events. In addition, Debio 1452 exhibits a low rate of single-step resistance and its targeted spectrum limits cross-resistance to other bacterial classes.

A Clinical Laboratory and Standards Institute (CLSI) M23 style quality control (QC) study was performed to establish broth microdilution QC ranges for *S. aureus* ATCC 29213 to assist clinical laboratories in monitoring the activity of this investigational compound during the clinical trials.

MATERIALS / METHODS

Study design: Eight experienced laboratories (seven required by CLSI M23-A3 guidelines) were used in this broth microdilution study to establish the Debio 1452 QC range. Laboratories followed the CLSI procedure for broth microdilution methods. The sites participating, laboratory director in parenthesis were: Wheaton Franciscan Laboratory, Wauwatosa, Wisconsin, USA (E. Munson); JMI Laboratories, North Liberty, Iowa, USA (R. N. Jones); Thermo Fisher Scientific, Cleveland, Ohio, USA (C. Knapp); University of Alberta, Edmonton, Alberta, Canada (R. Rennie); University of Washington, Seattle, Washington, USA (S. Swanzy); Cleveland Clinic Foundation, Cleveland, Ohio, USA (G. Hall/G. Procop); Massachusetts General Hospital, Boston, Massachusetts, USA, (M.J. Ferraro); Duke University Medical Center, Durham, North Carolina, USA, (S. Mirrett).

Susceptibility methods: Reference frozen-form broth microdilution panels were prepared by Thermo Fisher Scientific according to Good Manufacturing Practice (GMP) guidelines and shipped frozen to all sites. Panels contained four lots of cation-adjusted Mueller-Hinton broth (Oxoid, Hampshire UK; BBL, Sparks, Maryland, USA; and Difco [two lots], Detroit, Michigan, USA). Rifampin was utilized as an internal control agent.

Colony counts of the inoculum were performed on drug-free agar media and resulted in the following average counts for *S. aureus* ATCC 29213: 3.4 x 10^5 CFU/mL (Range 0.3 x 10^5 to 7.3 x 10^5 CFU/mL)

RESULTS

- The Debio 1452 MIC results from eight laboratories for *S. aureus* ATCC 29213 are shown in **Table 1** and **Figure 2**. Using M23 criteria to establish MIC QC ranges, 99.7% of all reported results would be in the proposed limits for Debio 1452 of 0.002 – 0.015 mg/L. Due to a shoulder MIC (0.004 mg/L) occurrence of greater than 60% (108 results; 70.6%) of that of the modal MIC (0.008 mg/L; 153 results), the CLSI procedure recommends a four dilution QC range.
- All media lots (modes at 0.004 or 0.008 mg/L) and laboratories (modes at 0.004 [3], 0.008 [4], and 0.015 mg/L [1]) failed to exhibit any significant skewing of results and geometric means varied only from 0.004 to 0.014 mg/L.
- The MIC data from this study was also analyzed by the Range Finder program. The Range Finder statistical analysis confirmed the proposed range with no significant outlier laboratories.
- All rifampin results (320/320; 100.0%) were within published QC ranges for the MIC panel tested providing a valid internal control result (**Table 2**).

Figure 1. Chemical structure of Debio 1452.

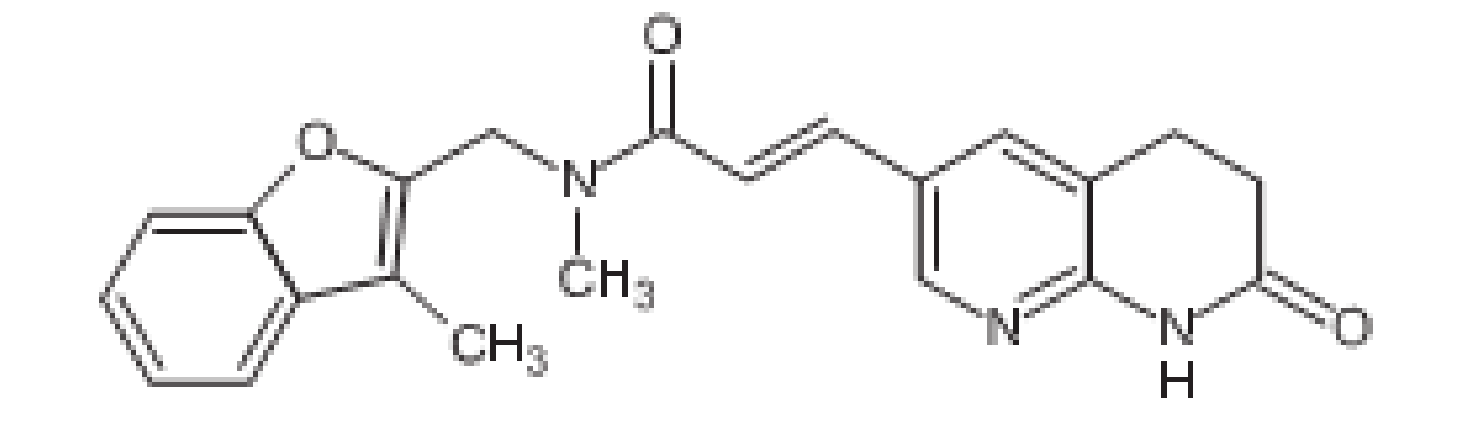
O
N
CH3
CH3
O
N
N
H
O

Figure 2. Debio 1452 MIC distributions for *S. aureus* ATCC 29213.

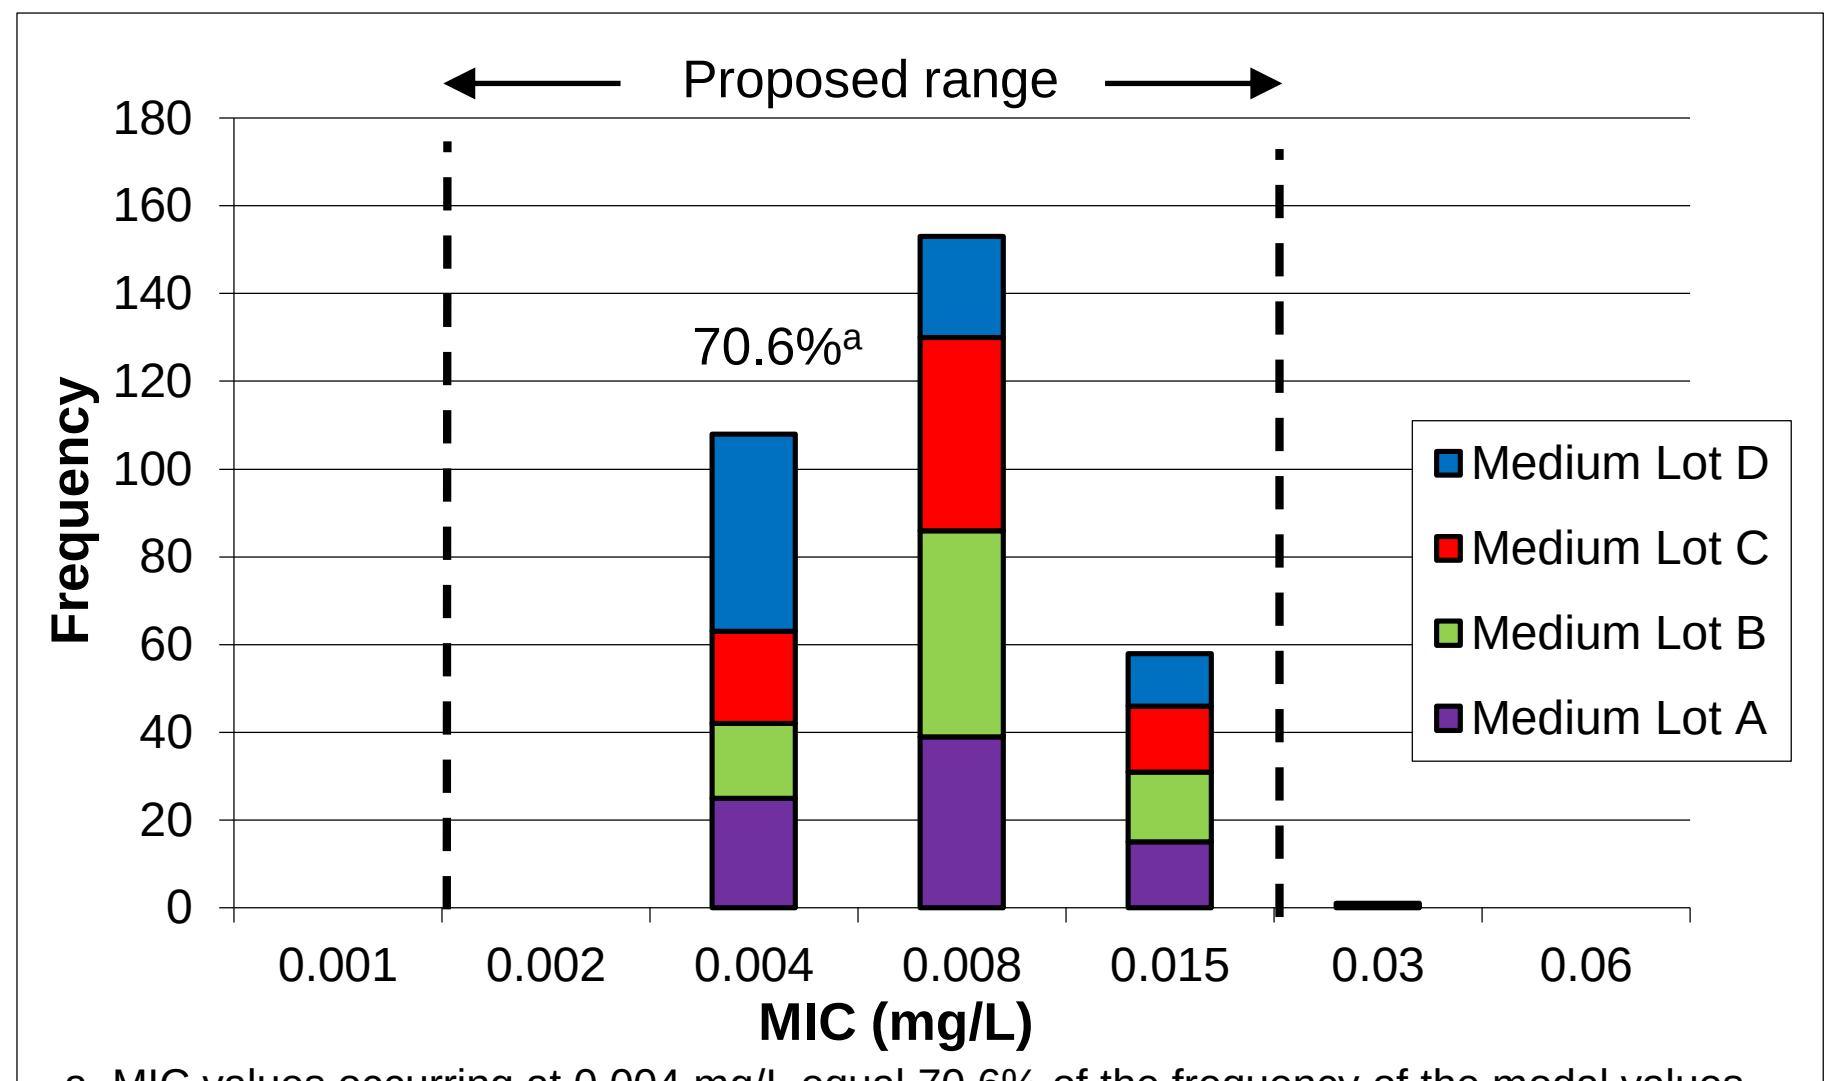
Proposed range
180
160
140
120
100
80
60
40
20
0
Frequency
70.6%a
Medium Lot D
Medium Lot C
Medium Lot B
Medium Lot A
0.001
0.002
0.004
0.008
0.015
0.03
0.06
MIC (mg/L)

a. MIC values occurring at 0.004 mg/L equal 70.6% of the frequency of the modal values at 0.008 mg/L.

Table 1. Media lot comparisons and inter- and intra-laboratory comparisons of Debio 1452 MIC results versus *S. aureus* ATCC 29213 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008).

MIC (mg/L)	Occurrences by lot:				Laboratory code (occurrences):								Total
	A	B	C	D	A	B	C	D	E	F	G	H	
0.002													0[a]
0.004	25	17	21	45	4		2	24	12	24	4	38	108[a]
0.008	39	47	44	23	36	4	23	16	28	15	29	2	153[a]
0.015	15	16	15	12		35	15			1	7		58[a]
0.03	1					1							1
Total	80	80	80	80	40	40	40	40	40	40	40	40	320
Mode	0.008	0.008	0.008	0.004	0.008	0.015	0.008	0.004	0.008	0.004	0.008	0.004	0.008
Geomean	0.007	0.008	0.008	0.006	0.008	0.014	0.010	0.005	0.007	0.005	0.008	0.004	0.007
Range	4	3	3	3	2	3	3	2	2	3	3	2	4

a. 99.7% of qualified results in proposed QC range (0.002 – 0.015mg/L).

Table 2. Media lot comparisons and inter- and intra-laboratory comparisons of rifampin MIC results versus *S. aureus* ATCC 29213 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008).

MIC (mg/L)	Occurrences by lot:				Laboratory code (occurrences):								Total
	A	B	C	D	A	B	C	D	E	F	G	H	
0.002													
0.004	46	49	75	47	39	8	32	37	14	37	10	40	217
0.008	33	31	5	33	1	32	8	3	26	3	29		102
0.015	1										1		1
0.03													
Total	80	80	80	80	40	40	40	40	40	40	40	40	320
Mode	0.004	0.004	0.004	0.004	0.004	0.008	0.008	0.008	0.008	0.004	0.008	0.004	0.004
Geomean	0.005	0.005	0.004	0.005	0.004	0.007	0.005	0.004	0.006	0.004	0.007	0.004	0.005
Range	3	2	2	2	2	2	2	2	2	2	3	1	3

a. 100.0% of control agent results in the published QC range (0.004 – 0.015 mg/L).

CONCLUSIONS

- These results (**Table 1**, 0.002 – 0.015 mg/L) provide Tier II level QC ranges for Debio 1452 for routine susceptibility testing using the broth microdilution method, as this new FabI inhibitor is developed for treatment of staphylococcal infections.
- The CLSI Subcommittee on Antimicrobial Susceptibility Testing approved this MIC QC range at the January 2011 meeting for publication, which will occur only after the selection of the compound's chemical name.

ACKNOWLEDGEMENTS

This study was sponsored by an educational/research grant from Affinium Pharmaceuticals Inc (Toronto, Canada). JER, RKF and RNJ are employees of JMI Laboratories that received grant funds to study Debio 1452 (formerly AFN-1252) and were paid consultants to Debiopharm International SA in connection with the development of this presentation. We also thank the eight contributing laboratories (personnel and directors) for their excellent support of this protocol.

The sponsor may be contacted at the following email and website address:
Email: hub1450@debiopharm.com
Website: www.debiopharm.com

REFERENCES

1. Clinical and Laboratory Standards Institute (2008). *M23-A3. Development of in vitro susceptibility testing criteria and quality control parameters: third edition*. Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2015). *M100-S25. Performance standards for antimicrobial susceptibility testing: 25th informational supplement.* Wayne, PA: CLSI.
3. EUCAST (2015). Breakpoint tables for interpretation of MICs and zone diameters. Version 5.0, January 2015. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2015.
4. Flamm RK, Rhomberg P, Kaplan N, Jones RN, Farrell DJ (2015). Activity of Debio1452, a FabI inhibitor with potent activity against *S. aureus* and coagulase-negative *Staphylococcus* spp., including multidrug-resistant strains. *Antimicrob Agents Chemother in press*.
5. Kaplan N, Albert M, Awrey D, Bardouniotis E, Berman J, Clarke T, Dorsey M, Hafkin B, Ramnauth J, Romanov V, Schmid MB, Thalakada R, Yethon J, Pauls HW (2012). Mode of action, in vitro activity, and in vivo efficacy of AFN-1252, a selective antistaphylococcal FabI inhibitor. *Antimicrob Agents Chemother* 56: 5865-5874.
6. Karlowsky JA, Kaplan N, Hafkin B, Hoban DJ, Zhanel GG (2009). AFN-1252, a FabI inhibitor, demonstrates a Staphylococcus-specific spectrum of activity. *Antimicrob Agents Chemother* 53: 3544-3548.
7. Turnidge J, Bordash G (2007). Statistical methods for establishing quality control ranges for antibacterial agents in Clinical and Laboratory Standards Institute susceptibility testing. *Antimicrob Agents Chemother* 51: 2483-2488.