ECCMID 2021 | Poster #01606

Activity of Cefiderocol and Comparators against US isolates of *Pseudomonas aeruginosa*, *Acinetobacter baumannii-calcoaceticus* species complex, and *Stenotrophomonas maltophilia*, including Carbapenem-Resistant Isolates

Dee Shortridge, Jennifer M. Streit, Rodrigo Mendes, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Cefiderocol is a novel siderophore-conjugated cephalosporin with broad activity against aerobic Gram-negative bacteria, including multidrug-resistant (MDR) organisms.
- *Acinetobacter baumannii-calcoaceticus* species complex, *Pseudomonas aeruginosa* and *Stenotrophomonas maltophilia* can be extensively-drug resistant (XDR) and present serious treatment challenges.
- Cefiderocol was approved by the EMA for the treatment of infections caused by Gram-negative bacteria in adult patients with limited treatment options and the FDA for complicated urinary tract infection (UTI), hospital-acquired bacterial pneumonia, and ventilator-associated bacterial pneumonia.
- The susceptibility of cefiderocol and comparators was investigated against US Gram-negative non-fermentative isolates collected in 2020 as a part of the SENTRY Antimicrobial Surveillance Program.

Materials and Methods

- A total of 1,536 isolates were consecutively collected from multiple specimen types in 29 US hospitals during 2020.
- Susceptibility testing was performed using the broth microdilution method.
 - Cefiderocol was tested in iron-depleted Mueller-Hinton broth.
- FDA, CLSI, and/or EUCAST (2021) breakpoints were used as available.
- For cefiderocol, the following breakpoints were used for the organism groups in this study:
 - For *P. aeruginosa*: cefiderocol CLSI breakpoints are ≤4/8/≥16 mg/L (susceptible, intermediate, and resistant, respectively), FDA breakpoints are ≤1/2/≥/4 mg/L, and EUCAST breakpoints are ≤2/-/>2 mg/L.
 - For *A. baumannii-calcoaceticus* species complex: cefiderocol CLSI breakpoints are ≤4/8/≥16 mg/L (susceptible/ intermediate/ resistant), FDA breakpoints are ≤1/2/≥4 mg/L, and EUCAST non-species specific PK/PD breakpoints are ≤2/-/>2 mg/L.
 - For *S. maltophilia*: cefiderocol CLSI breakpoints are ≤4/8/≥16 mg/L and will be updated in 2022 to ≤1/-/- mg/L and EUCAST non-species-specific PK/PD breakpoints are ≤2/-/>2 mg/L.
- XDR was defined as susceptible to ≤2 drug classes.
 - Other agents tested included the beta-lactam/beta-lactamase inhibitor (BL/BLI) combinations ceftazidime-avibactam, ceftolozane-tazobactam, imipenem-relebactam, meropenem-vaborbactam, and meropenem.

Results

- The most common non-fermenting Gram-negative organism was *Pseudomonas aeruginosa* (n=999), followed by *A. baumannii-calcoacetius* species complex (n=230) and *S. maltophilia* (n=186; Figure 1).
- The most common infection type was pneumonia (n=795), followed by skin/skin structure infection (n=257).
- The cumulative MIC distributions of cefiderocol and comparators are shown for *P. aeruginosa* (Figure 2) and *A. baumannii-calcoacetius* species complex (Figure 3).
- Cefiderocol and comparator BL/BLI susceptibilities against *P. aeruginosa* were >96.0%, except for meropenem-vaborbactam (89.2%, Table 1).
- Cefiderocol was the most active drug tested against XDR *P. aeruginosa* isolates (97.3/96.4/97.3% susceptibility according to CLSI, FDA, EUCAST respectively). The susceptibilities of BL/BLI comparators ranged from 40.5-83.8% (Table 1).
 - Cefiderocol was also active against meropenem-resistant *P. aeruginosa* (≥96.7%).
 - Cefiderocol remained active against 8 isolates resistant to all BL/BLI tested showing 87.5% susceptibility according to breakpoints provided by CLSI, FDA, and EUCAST, respectively.
- Against all *A. baumannii-calcoacetius* species complex, cefiderocol was very active (97.0/92.2/96.5%, CLSI/FDA/EUCAST; Table 2).
 - Cefiderocol susceptibility rates against the XDR and meropenem-resistant *A. calcoacetius-baumannii* species complex subsets were 94.2/81.2/92.8% and 93.2/81.8/92.0%, respectively (CLSI/FDA/EUCAST).
- Cefiderocol was highly active against *S. maltophilia*, with 100.0/98.9% susceptibility at MIC values ≤4 mg/L and ≤2 mg/L according to breakpoints provided by CLSI and EUCAST, respectively (Table 2).
 - CLSI *S. maltophilia* breakpoints will be updated in 2022 to ≤1 mg/L S; cefiderocol was active against 97.3% of isolates at MIC values of ≤1 mg/L.

Figure 1. Frequency of the most commonly isolated non-fermentative organisms

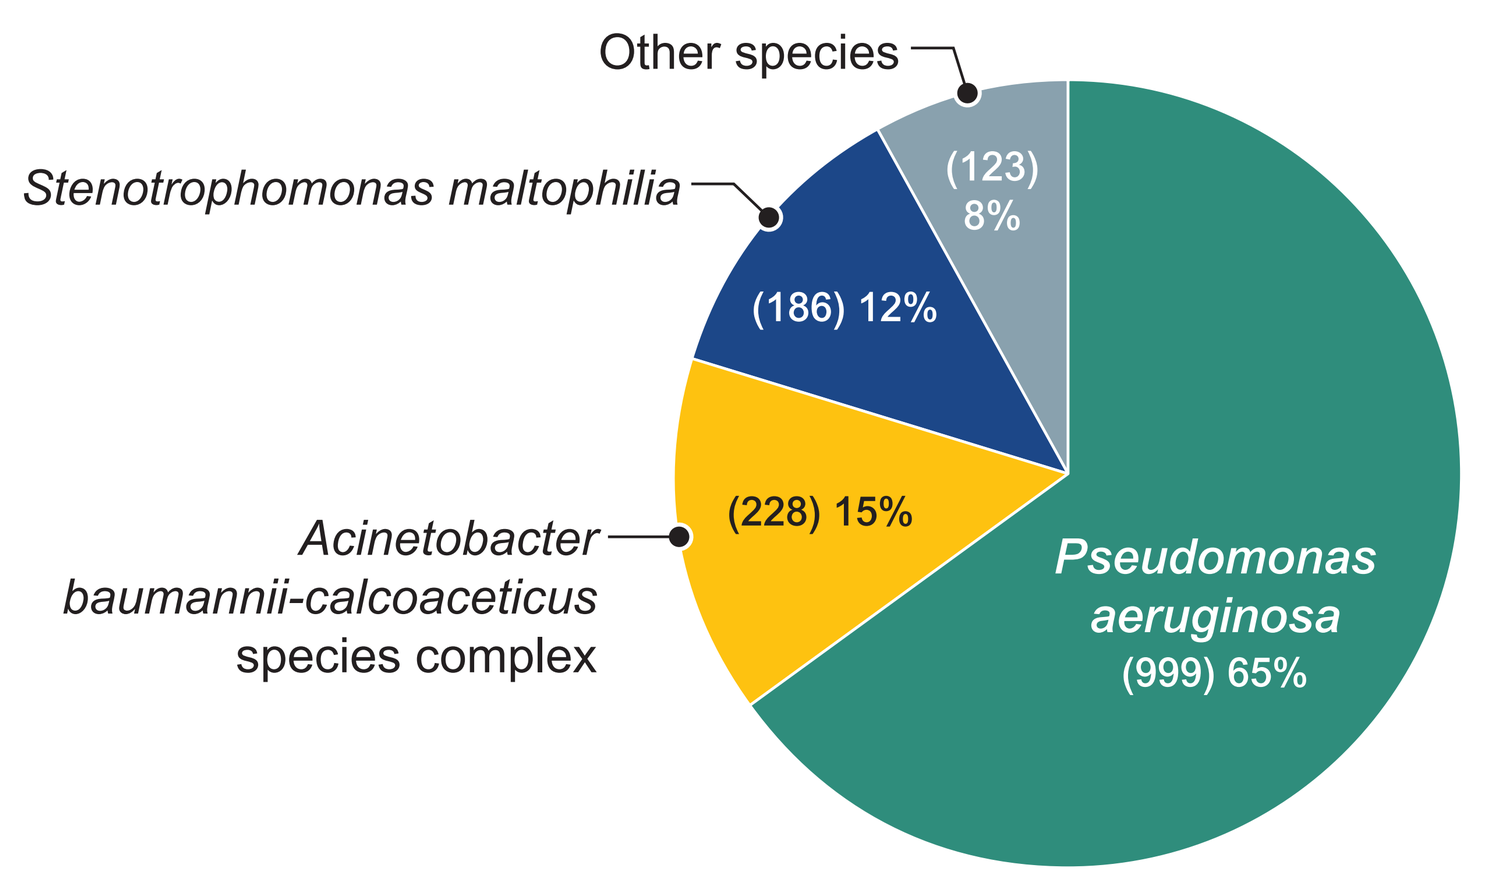
Other species
(123) 8%
Stenotrophomonas maltophilia
(186) 12%
Acinetobacter baumannii-calcoaceticus species complex
(228) 15%
Pseudomonas aeruginosa (999) 65%

Figure 2. Cumulative percent MIC distribution of cefiderocol and comparators for 999 US isolates of *P. aeruginosa* (n=999)

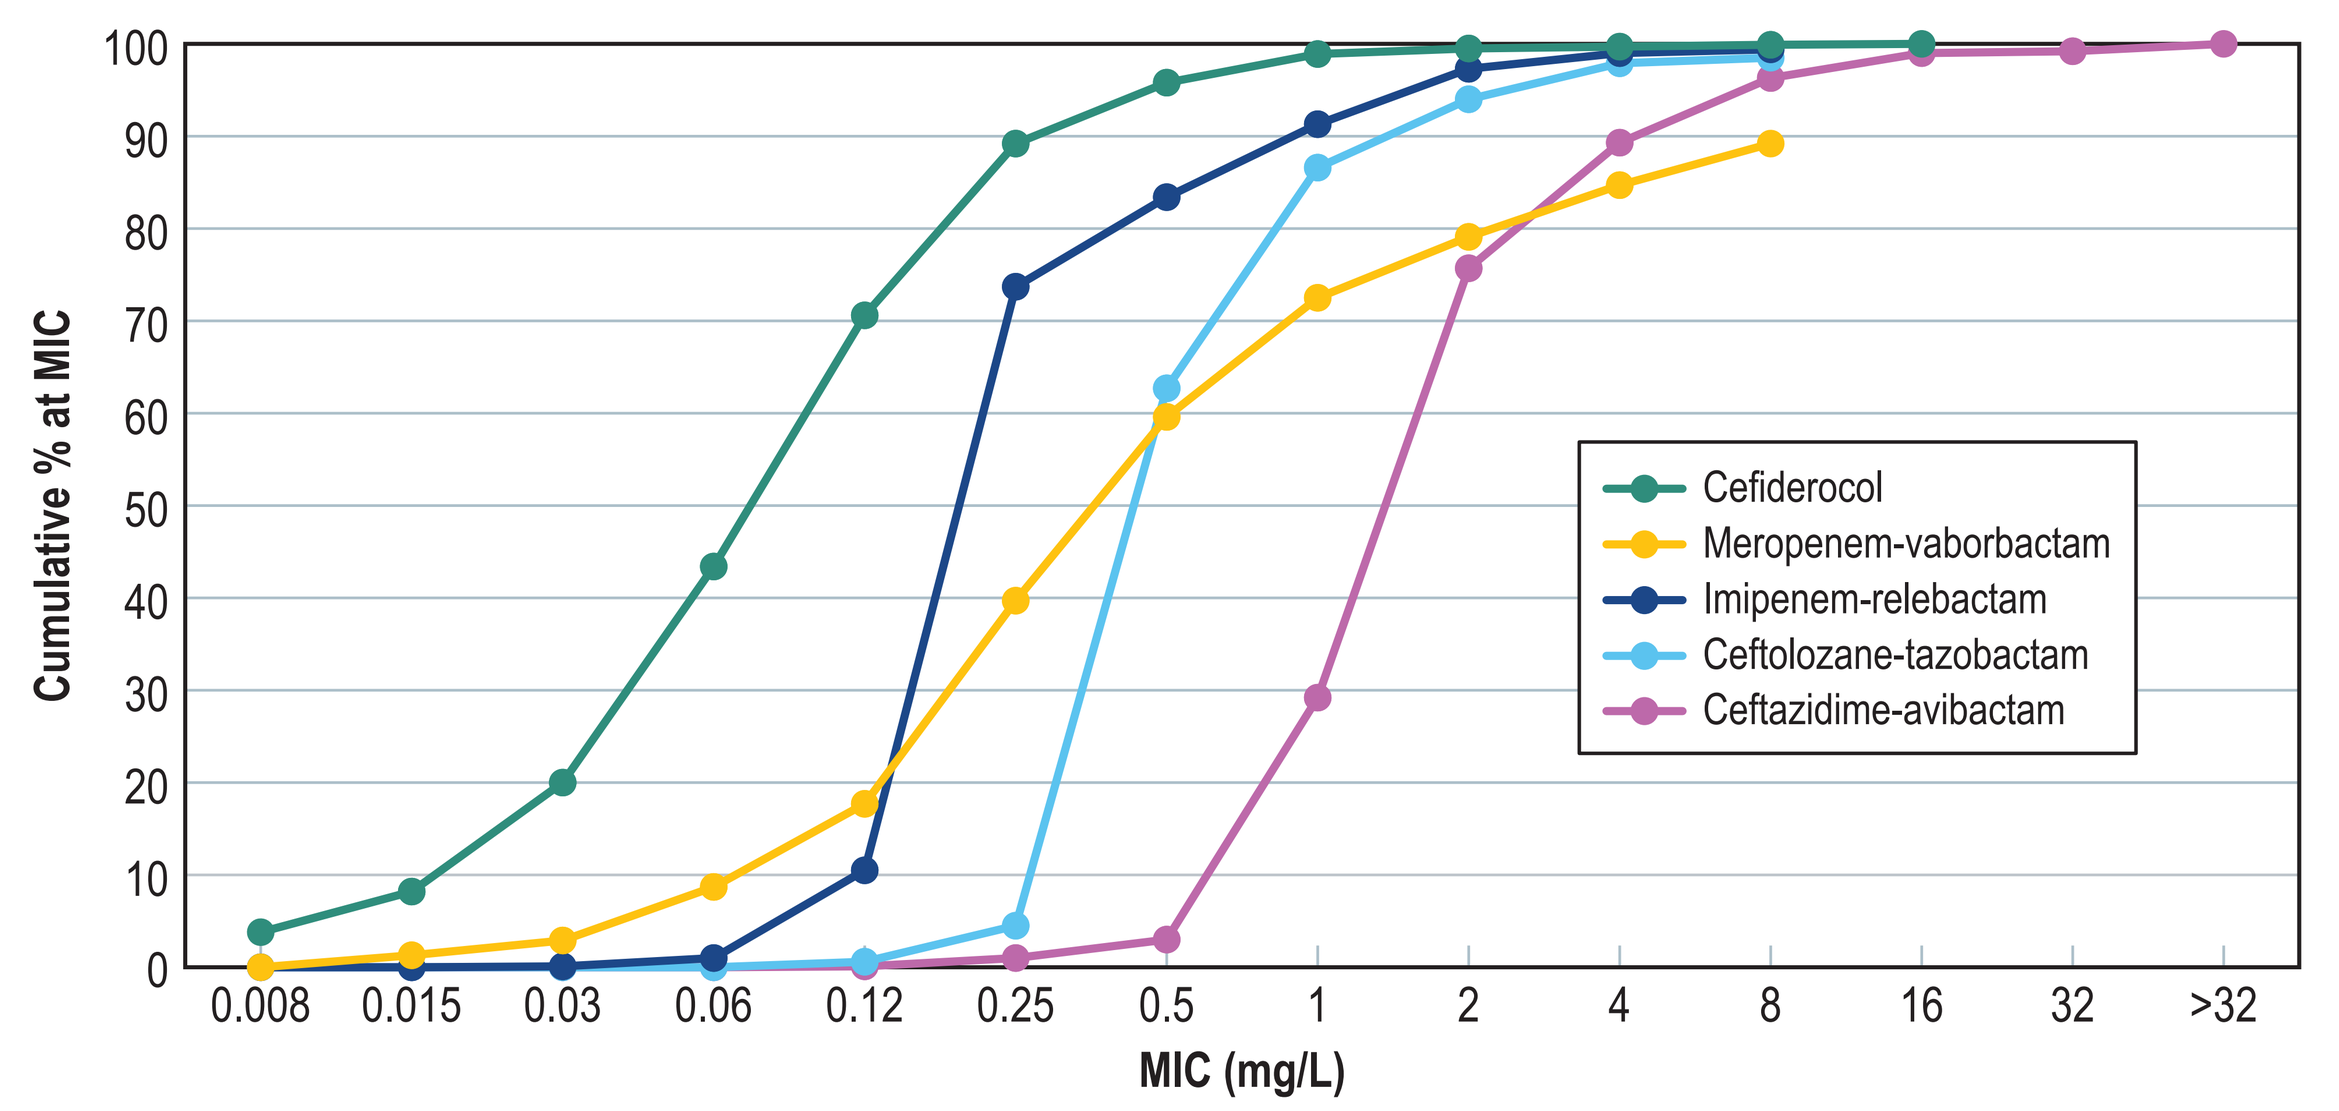
Cumulative % at MIC
MIC (mg/L)
0.008 0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 >32
Cefiderocol
Meropenem-vaborbactam
Imipenem-relebactam
Ceftolozane-tazobactam
Ceftazidime-avibactam

Table 1. Susceptibilities of *P. aeruginosa* and resistant subgroups tested against cefiderocol and comparators

Antimicrobial agent	No. of isolates	mg/L MIC$_{50}$	mg/L MIC$_{90}$	CLSI[a] %S	FDA %S	EUCAST[a] %S
All						
Cefiderocol	999	0.12	0.5	99.7	98.9	99.5
Meropenem-vaborbactam	992	0.5	>8			89.2
Imipenem-relebactam	999	0.25	1	97.3	97.3	97.3
Ceftolozane-tazobactam	997	0.5	2	97.9		97.9
Ceftazidime-avibactam	999	2	8	96.3		96.3
XDR [b]						
Cefiderocol	111	0.12	1	97.3	96.4	97.3
Meropenem-vaborbactam	111	>8	>8			40.5
Imipenem-relebactam	111	2	4	81.1	81.1	81.1
Ceftolozane-tazobactam	111	2	16	83.8		83.8
Ceftazidime-avibactam	111	8	16	72.1		72.1
Meropenem-R						
Cefiderocol	152	0.12	0.5	98.7	96.7	97.4
Meropenem-vaborbactam	152	>8	>8			30.9
Imipenem-relebactam	152	2	4	82.2	82.2	82.2
Ceftolozane-tazobactam	152	2	8	88.2		88.2
Ceftazidime-avibactam	152	4	16	79.6		79.6
Meropenem-vaborbactam -R						
Cefiderocol	107	0.12	0.5	98.1	97.2	98.1
Meropenem-vaborbactam	107	>8	>8			0.0
Imipenem-relebactam	107	2	4	77.6	77.6	77.6
Ceftolozane-tazobactam	107	2	16	85		85
Ceftazidime-avibactam	107	8	16	74.8		74.8
Imipenem-relebactam -R						
Cefiderocol	10	0.12	0.25	100.0	100.0	100.0
Meropenem-vaborbactam	10	>8	>8			0.0
Imipenem-relebactam	10	>8	>8	0.0	0.0	0.0
Ceftolozane-tazobactam	10	4	>16	50.0		50.0
Ceftazidime-avibactam	10	16	>32	20.0		20.0
Ceftolozane-tazobactam -R						
Cefiderocol	15	0.5	8	80.0	66.7	73.3
Meropenem-vaborbactam	15	>8	>8			20.0
Imipenem-relebactam	15	2	>8	53.3	53.3	53.3
Ceftolozane-tazobactam	15	>16	>16	0.0		0.0
Ceftazidime-avibactam	15	32	>32	20.0		20.0
Ceftazidime-avibactam -R						
Cefiderocol	37	0.25	4	91.9	89.2	89.2
Meropenem-vaborbactam	37	>8	>8			27.0
Imipenem-relebactam	37	2	>8	59.5	59.5	59.5
Ceftolozane-tazobactam	37	4	>16	62.2		62.2
Ceftazidime-avibactam	37	16	>32	0.0		0.0
BL/BLI- R [c]						
Cefiderocol	8	0.25	N/A	87.5	87.5	87.5
Meropenem-vaborbactam	8	>8	N/A	0.0		0.0
Imipenem-relebactam	8	>8	N/A	0.0	0.0	0.0
Ceftolozane-tazobactam	8	16	N/A	0.0		0.0
Ceftazidime-avibactam	8	32	N/A	0.0		0.0

[a] Breakpoints as published by CLSI, FDA, or EUCAST (2021). Cefiderocol CLSI breakpoints are ≤4/8/≥16 mg/L (susceptible, intermediate, and resistant, respectively), FDA breakpoints are ≤1/2/≥/4 mg/L, and EUCAST PK/PD breakpoints are ≤2/-/>2 mg/L.
[b] XDR, extensively-drug resistant; R, resistant using CLSI breakpoints (2021).
[c] BL/BLI-R are resistant to all 4 BL/BLI combinations.

Figure 3. Cumulative percent MIC distribution of cefiderocol and comparators for 228 US isolates of *A. baumannii-calcoaceticus* species complex (n=228)

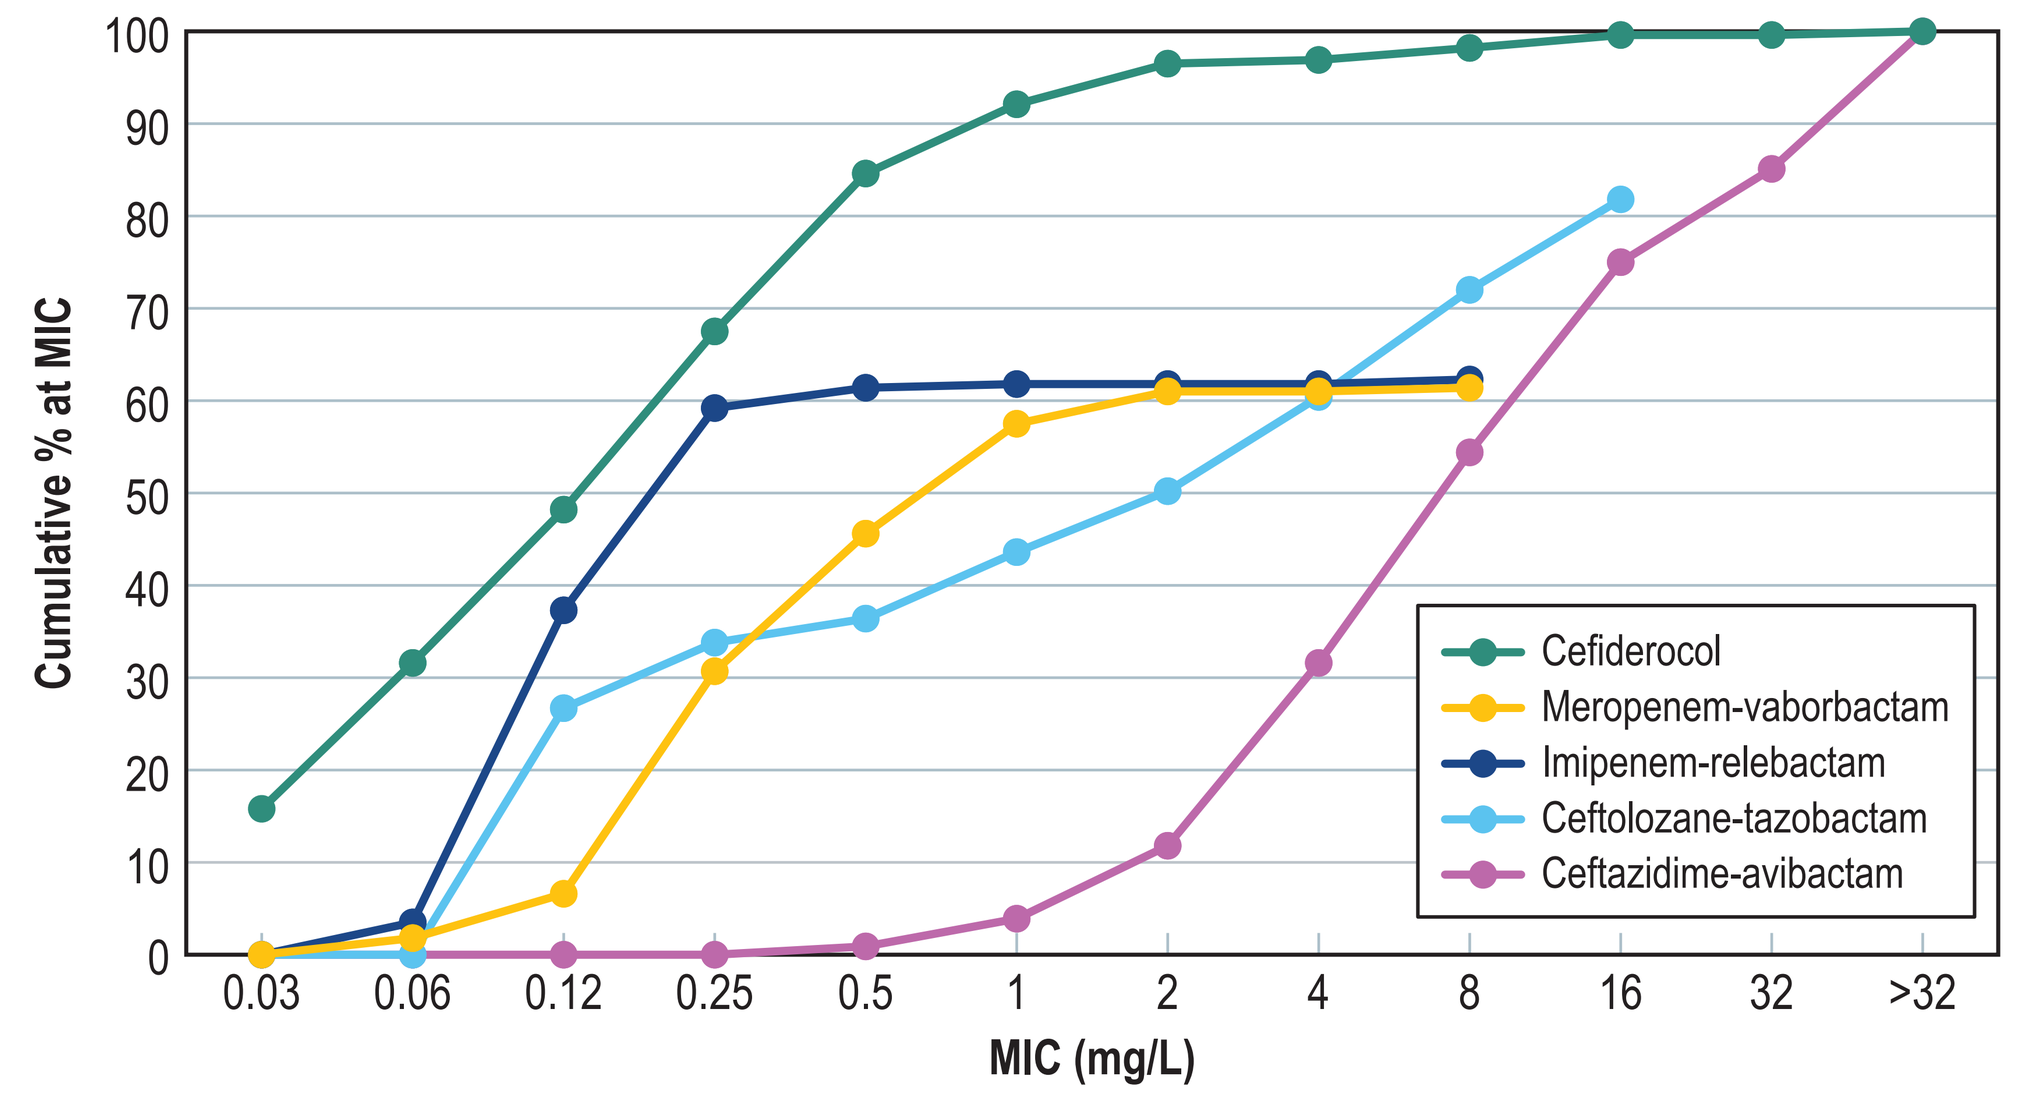
Cumulative % at MIC
MIC (mg/L)
0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 >32
Cefiderocol
Meropenem-vaborbactam
Imipenem-relebactam
Ceftolozane-tazobactam
Ceftazidime-avibactam

Table 2. Susceptibilities of *Acinetobacter baumannii-calcoaceticus* species complex and resistant groups as well as *Stenotrophomonas maltophilia* tested against cefiderocol and comparators

Antimicrobial agent	No. of isolates	mg/L MIC$_{50}$	mg/L MIC$_{90}$	CLSI[a] %S	FDA[a] %S	EUCAST[a] %S
All *A. baumannii-calcoacetius* species complex						
Cefiderocol [b]	230	0.25	1	97.0	92.2	96.5
Meropenem-vaborbactam	230	1	>8			
Imipenem-relebactam	230	0.25	>8	[c]	62.2	62.2
Ceftolozane-tazobactam	227	2	>16			
Ceftazidime-avibactam	230	8	>32			
XDR [c]						
Cefiderocol	69	0.5	2	94.2	81.2	92.8
Meropenem-vaborbactam	69	>8	>8			
Imipenem-relebactam	69	>8	>8		4.3	4.3
Ceftolozane-tazobactam	68	16	>16			
Ceftazidime-avibactam	69	32	>32			
Meropenem-R						
Cefiderocol	88	0.5	2	93.2	81.8	92.0
Meropenem-vaborbactam	88	>8	>8			
Imipenem-relebactam	88	>8	>8		1.1	1.1
Ceftolozane-tazobactam	87	16	>16			
Ceftazidime-avibactam	88	16	>32			
Imipenem-relebactam-R						
Cefiderocol	87	0.5	2	93.1	81.6	92.0
Meropenem-vaborbactam	87	>8	>8			
Imipenem-relebactam	87	>8	>8		0.0	0.0
Ceftolozane-tazobactam	86	16	>16			
Ceftazidime-avibactam	87	16	>32			
S. maltophilia [d]						
Cefiderocol	186	0.12	0.5	100.0/97.3		98.9
Meropenem-vaborbactam	186	>8	>8			
Imipenem-relebactam	186	>8	>8			
Ceftolozane-tazobactam	186	>16	>16			
Ceftazidime-avibactam	186	32	>32			

[a] CLSI/FDA or EUCAST (2021).
[b] Breakpoints for *A. calcoacetius-baumannii* species complex were: Cefiderocol CLSI breakpoints, ≤4/8/≥16 mg/L (susceptible/ intermediate/ resistant); FDA breakpoints, ≤1/2/≥4 mg/L; EUCAST PK/PD breakpoints, ≤2/-/>2 mg/L.
[c] XDR, extensively drug resistant; R, resistant using CLSI breakpoints (2021).
[d] Breakpoints for *S. maltophilia*: cefiderocol CLSI breakpoints currently are ≤4/8/≥16 mg/L and in 2022, the breakpoints will be updated to ≤1/-/-; cefiderocol EUCAST PK/PD breakpoints are ≤2/-/>2 mg/L.

Conclusions

- Cefiderocol had potent activity against *P. aeruginosa*, including XDR isolates and isolates resistant to other BL/BLI combination therapies.
- Cefiderocol also had excellent activity against *A. baumannii-calcoacetius* species complex, including XDR and meropenem-resistant isolates.
- All *S. maltophilia* were inhibited by cefiderocol at an MIC of ≤4 mg/L.
- These *in vitro* data suggest that cefiderocol is an important treatment option for infections caused by non-fermenting species, including resistant pathogens for which there are limited therapeutic choices.

Acknowledgements

This study was sponsored by Shionogi & Co., LTD.

References

CLSI. *M07 Eleventh edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically.* Clinical and Laboratory Standards Institute, Wayne, PA, 2018.

CLSI. *M100 31st edition. Performance Standards for Antimicrobial Susceptibility Testing.* Clinical and Laboratory Standards Institute, Wayne, PA, 2021.

EUCAST. The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters, v 11.0, January 1, 2021. http://www.eucast.org. Accessed Jan 31, 2021.

FDA Susceptibility Test Interpretive Criteria. https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed May 2021.

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2021_CefiderocolVMolecularEnteros.pdf

Charges may apply. No personal information is stored.

ECCMID 2021, July 9–12, 2021