ECCMID 2021 | Poster #1499

Assessment of Resistance Development to KBP-7072 by Serial-Passage and Single-Step Mutational Selection

MD Huband[1], RE Mendes[1], Q Liu[2], Li Li[2], J Zhang[3], J Thompson[1], GE Strand[1], and M Castanheira[1]
[1] JMI Laboratories, North Liberty, Iowa, USA; [2] KBP Biosciences Co., Ltd., Jinan, China; [3] KBP Biosciences USA Inc., Princeton, NJ, USA

Introduction

- KBP-7072 is a third-generation tetracycline (aminomethylcycline) antibacterial in clinical development for the treatment of acute bacterial skin and skin structure infection (ABSSSI), community-acquired bacterial pneumonia (CABP), and complicated intra-abdominal infection (cIAI).
- This study assessed the potential for resistance to KBP-7072 to develop in tetracycline-susceptible (S) and tetracycline-resistant (R) *Acinetobacter baumannii*, *Escherichia coli, Staphylococcus aureus,* and *Streptococcus pneumoniae* clinical isolates and quality control (QC) strains during single-step selection and 10-day serial-passage (multi-step selection) studies.

Methods and Materials

- Bacterial isolate identifications were confirmed by JMI Laboratories using matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).
- Broth microdilution and agar dilution susceptibility testing was performed in triplicate according to Clinical and Laboratory Standards Institute (CLSI) M07 (2018) and M100 (2021) guidelines.
 - The modal MIC values obtained became the baseline broth microdilution and agar dilution MIC values.
 - American Type Culture Collection (ATCC) QC strains recommended by CLSI were incorporated into each testing event.
- Single-step resistance testing was conducted using 4x, 8x, and 16x the baseline KBP-7072 agar dilution MIC value.
 - Inoculated agar plates were incubated at 35°C in ambient atmosphere and examined for growth at 24 hours.
 - Plates lacking visible colonies at 24 hours were incubated further and re-examined at 48 hours.
 - Putative resistant colonies were counted and compared to the starting inoculum to calculate the single-step frequency of resistance.
 - Representative colonies were selected from the agar plates demonstrating the highest KBP-7072 MIC values, transferred twice on drug-free agar plates, followed by confirmatory broth microdilution susceptibility testing against KBP-7072 and comparator agents.
- The modal broth microdilution KBP-7072 MIC value served as the baseline MIC for the serial-passage resistance testing.
 - Each strain was tested for susceptibility to KBP-7072 in 100 µL volumes using an initial starting inoculum of approximately 5×10^{5} CFU/mL.
 - Subsequent daily passages utilized the entire well contents from the last well demonstrating visible growth from the previous day's test.
 - This process was repeated for up to 10-daily transfers or until the KBP-7072 MIC value reached >64 mg/L.

Results

- KBP-7072 single-step frequency of resistance values for *S. aureus* ATCC 29213 and *S. aureus* 1088771 (tetracycline-resistant) were very low at $<9.4 \times 10^{-10}$ and $<8.6 \times 10^{-10}$, respectively.
 - No resistant mutants were recovered at 4x, 8x, or 16x the baseline KBP-7072 MIC value (Table 1)
- KBP-7072 MIC values remained stable (no increase in MIC value) against *S. aureus* ATCC 29213 and increased 4-fold (highest MIC, 0.25 mg/L) for *S. aureus* 1088771 over the course of 10-daily serial passages (Figures 1–2).
- Single-step frequency of resistance values for *S. pneumoniae* ATCC 49619 and *S. pneumoniae* 1119350 (penicillin- and tetracycline-resistant) ranged from $>2.3 \times 10^{-6}$ to $<8.2 \times 10^{-9}$ at 4x to 16x the baseline KBP-7072 MIC (Table 1).
- A ≤4-fold increase in KBP-7072 MIC values (highest MIC, 0.03 mg/L) were observed following drug free passage and confirmatory MIC testing, indicating that the single-step mutants obtained with *S. pneumoniae* ATCC 49619 (8x and 16x the baseline KBP-7072 MIC) and *S. pneumoniae* 1119350 (4x, 8x, and 16x the baseline KBP-7072 MIC) were not stably resistant (Table 1)
- KBP-7072 MIC values increased modestly (up to 4-fold; highest MIC, 0.03 mg/L) against *S. pneumoniae* ATCC 49619 and *S. pneumoniae* 1119350 following 10-daily serial passages (Figures 3–4).
- KBP-7072 single-step frequency of resistance was low, ranging from 9.0×10^{-8} at 4x MIC to $<9.6 \times 10^{-10}$ at 16x MIC for *A. baumannii* NCTC 13304 and from 2.7×10^{-7} at 4x MIC to $<8.7 \times 10^{-10}$ at 16x MIC for *A. baumannii* 1044704 (tetracycline-, levofloxacin-, and carbapenem-resistant) (Table 1).
- In serial passage testing, KBP-7072 MIC values increased 8-fold (0.12 to 1 mg/L) for *A. baumannii* NCTC 13304 and 4-fold (0.5 to 2 mg/L) for *A. baumannii* 1044704 over the course of 10-daily serial passages (Figures 5–6).
 - KBP-7072 MIC values remained elevated by ≤8-fold against *A. baumannii* NCTC 13304 and *A. baumannii* 1044704 following drug-free passage and confirmatory MIC testing (data not shown).
 - These mutants also demonstrated elevated MIC values to other tetracycline class agents (data not shown).
- Single-step frequency of resistance to KBP-7072 was very low ($<8.3 \times 10^{-10}$) against *E. coli* ATCC 25922 and no resistant mutants were recovered (Table 1).
 - Similarly, single-step frequency of resistance to KBP-7072 was low against a *E. coli* 1093450 (tetracycline- and levofloxacin-resistant; ESBL-phenotype), ranging from 1.6×10^{-7} at 4x MIC to $<2.6 \times 10^{-10}$ at 16x MIC (Table 1).
- In serial passage testing, KBP-7072 MIC values increased 8-fold (0.12 to 1 mg/L) for *E. coli* ATCC 25922 and >128-fold (0.5 to >64 mg/L) for *E. coli* 1093450 over the course of 10-daily serial passages (Figures 7–8).
 - KBP-7072 and tetracycline-class comparator agent MIC values remained elevated against *E. coli* following drug-free passage and confirmatory MIC testing (data not shown).

Figure 1. KBP-7072 serial-passage results for *S. aureus* ATCC 29213 (CLSI QC strain)

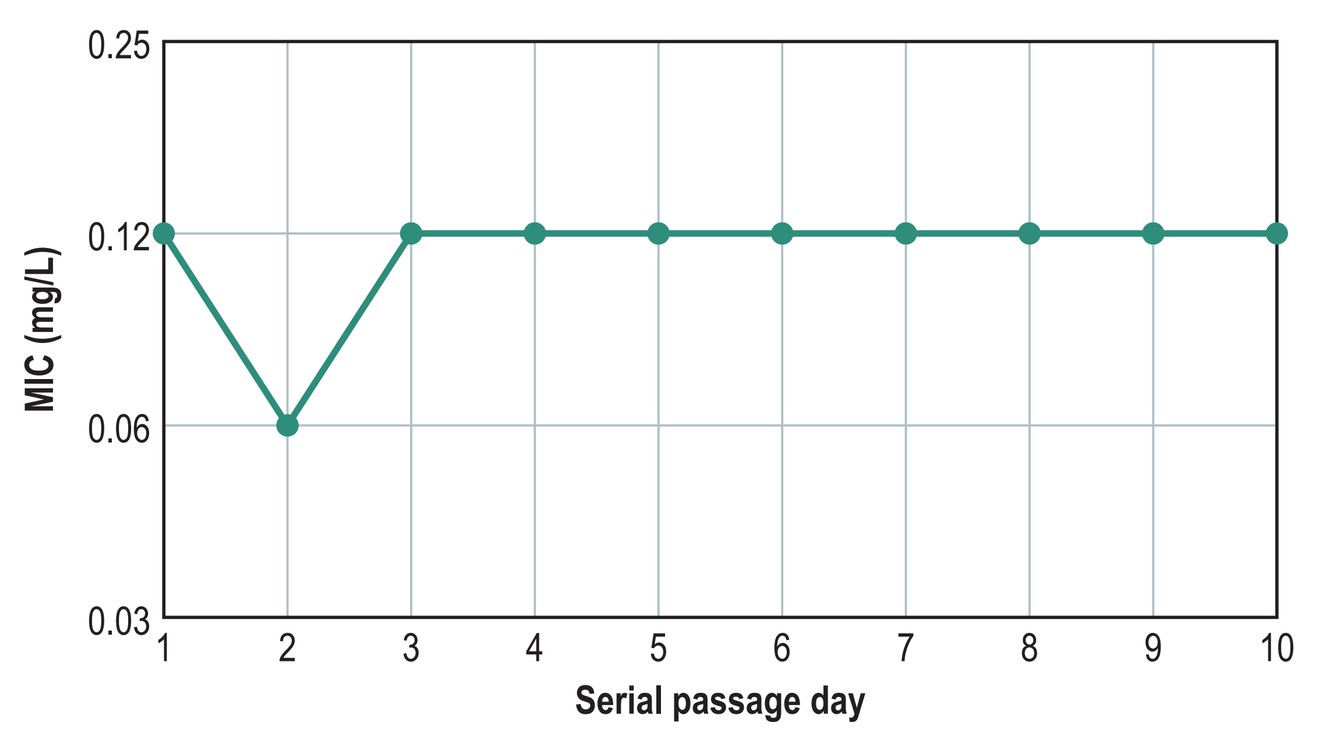
MIC (mg/L)
0.25
0.12
0.06
0.03
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 2. KBP-7072 serial-passage results for *S. aureus* 1088771 (resistant to tetracycline, clindamycin, levofloxacin, and oxacillin)

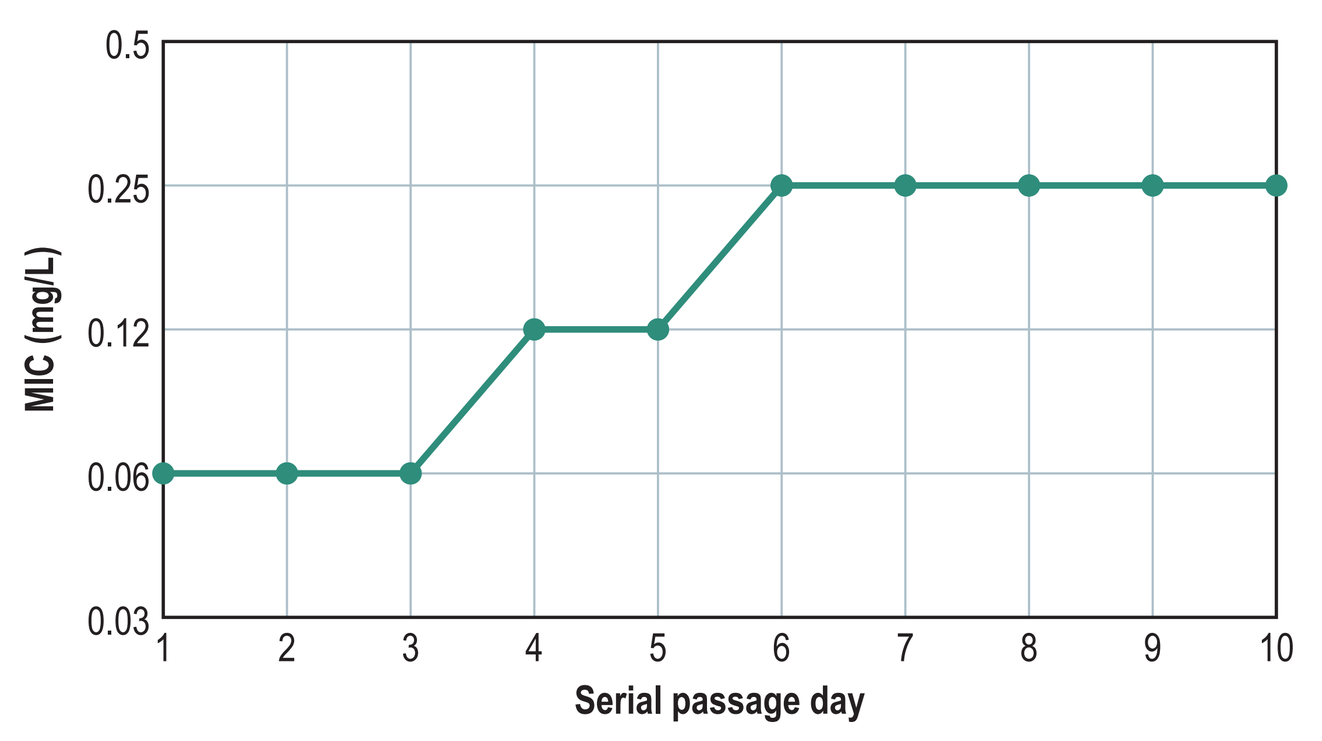
MIC (mg/L)
0.5
0.25
0.12
0.06
0.03
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 3. KBP-7072 serial-passage results for *S. pneumoniae* ATCC 49619 (CLSI QC strain)

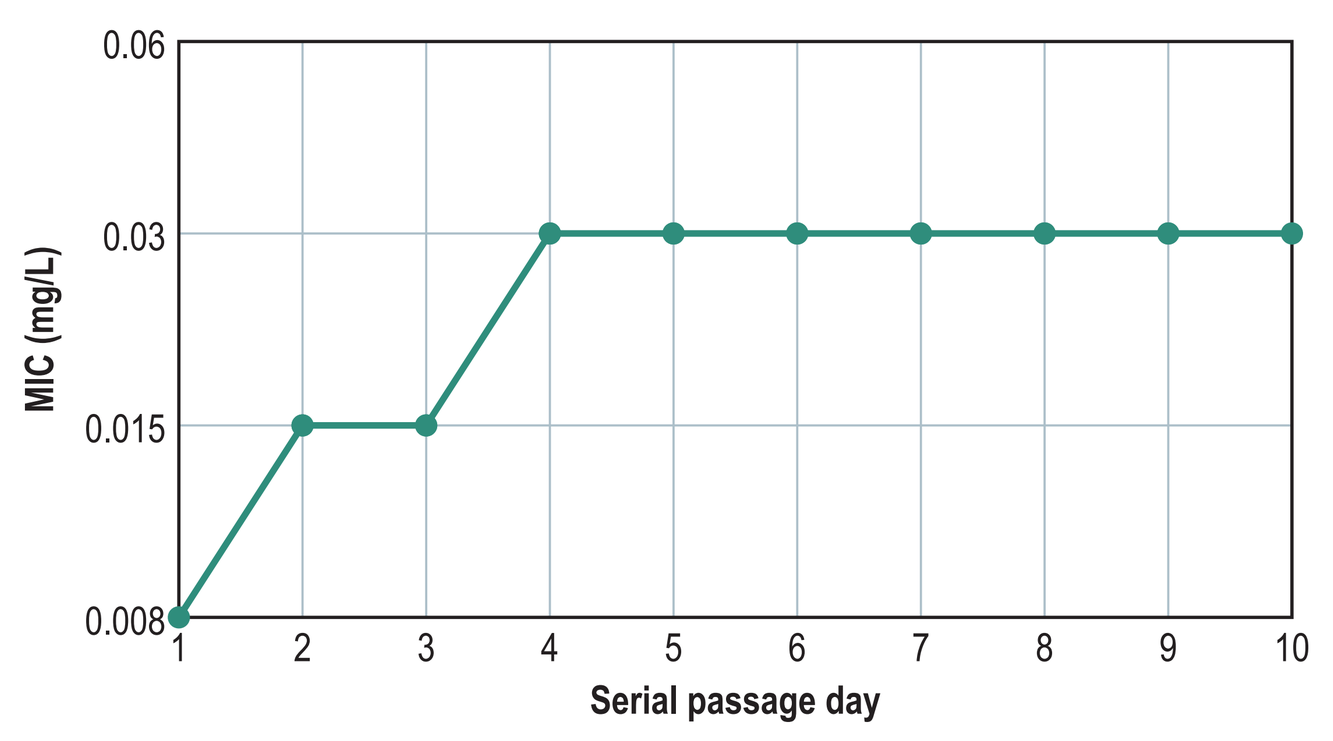
MIC (mg/L)
0.06
0.03
0.015
0.008
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 4. KBP-7072 serial-passage results for *S. pneumoniae* 1119350 (resistant to tetracycline and penicillin)

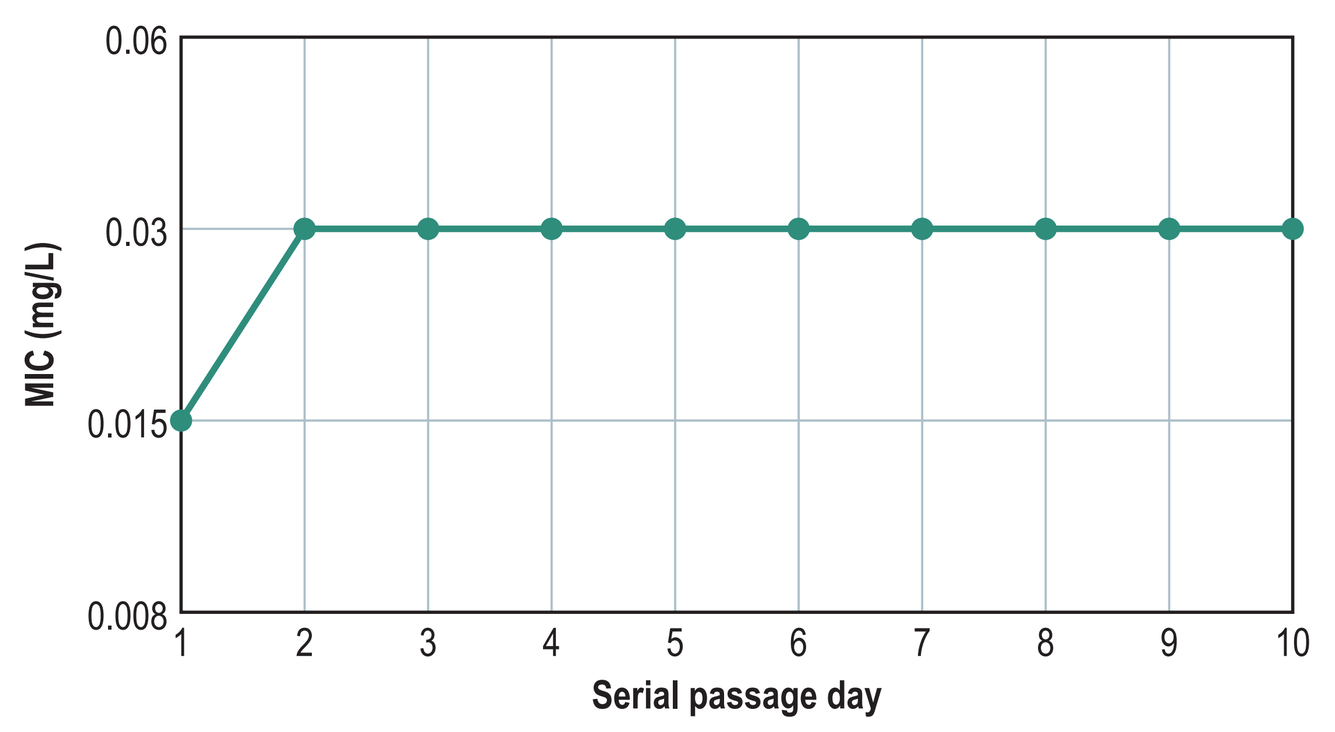
MIC (mg/L)
0.06
0.03
0.015
0.008
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 5. KBP-7072 serial-passage results for *A. baumannii* NCTC 13304 (CLSI QC strain)

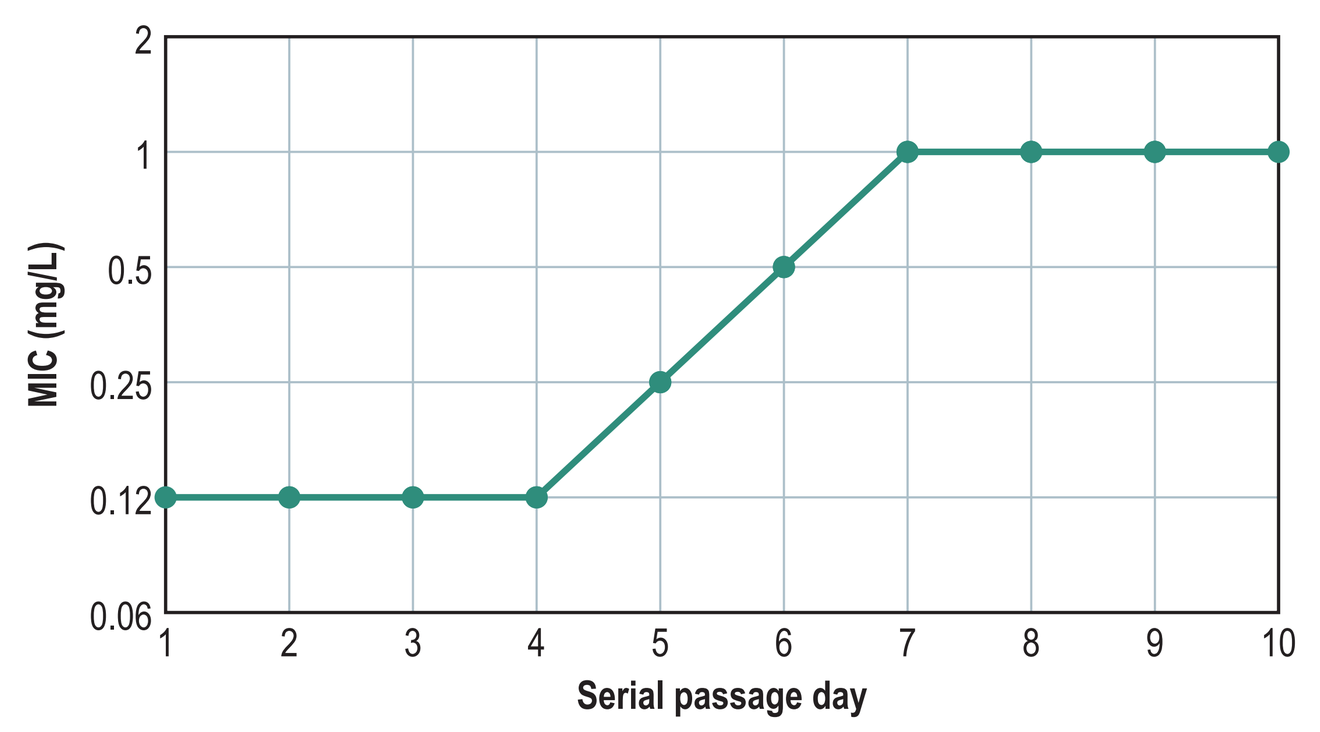
MIC (mg/L)
2
1
0.5
0.25
0.12
0.06
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 6. KBP-7072 serial-passage results for *A. baumannii* 1044704 (resistant to tetracycline, levofloxacin, and carbapenems)

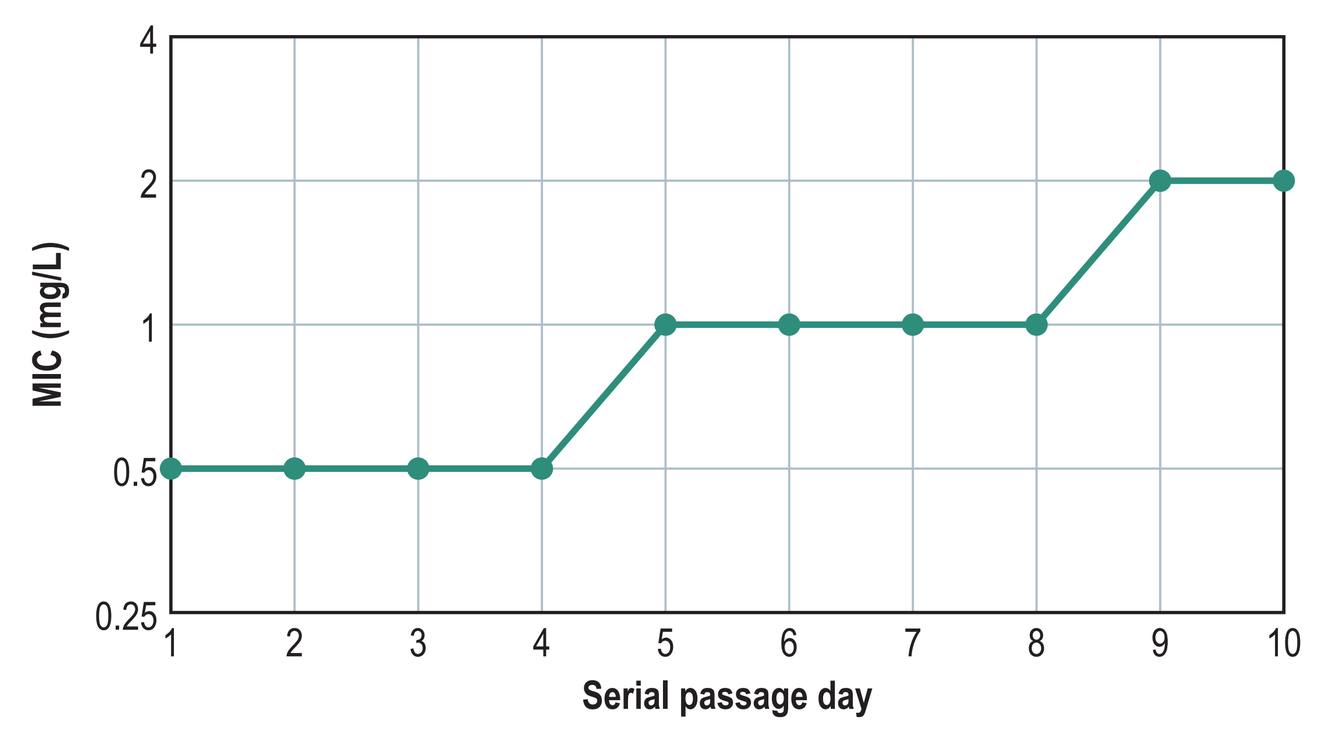
MIC (mg/L)
4
2
1
0.5
0.25
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 7. KBP-7072 serial-passage results for *E. coli* ATCC 25922 (CLSI QC strain)

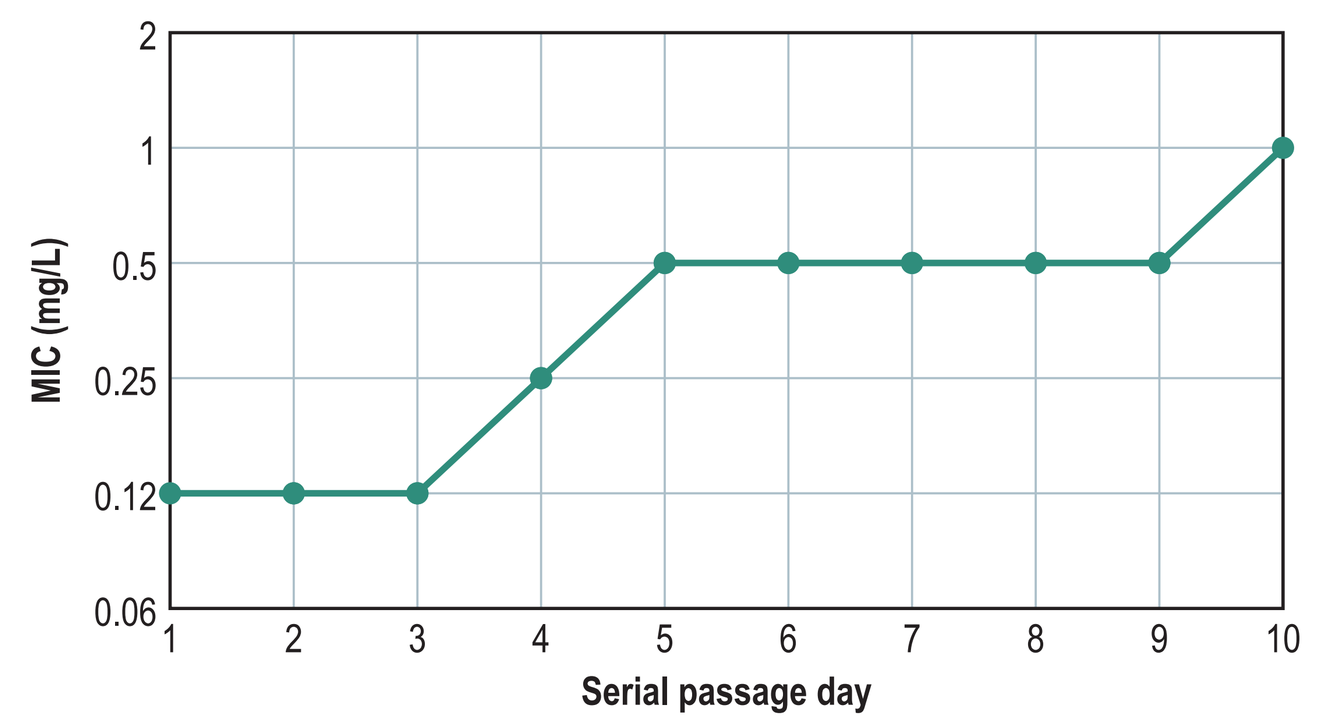
MIC (mg/L)
2
1
0.5
0.25
0.12
0.06
1 2 3 4 5 6 7 8 9 10
Serial passage day

Figure 8. KBP-7072 serial-passage results for *E. coli* 1093450 (ESBL-phenotype; resistant to tetracycline and levofloxacin)

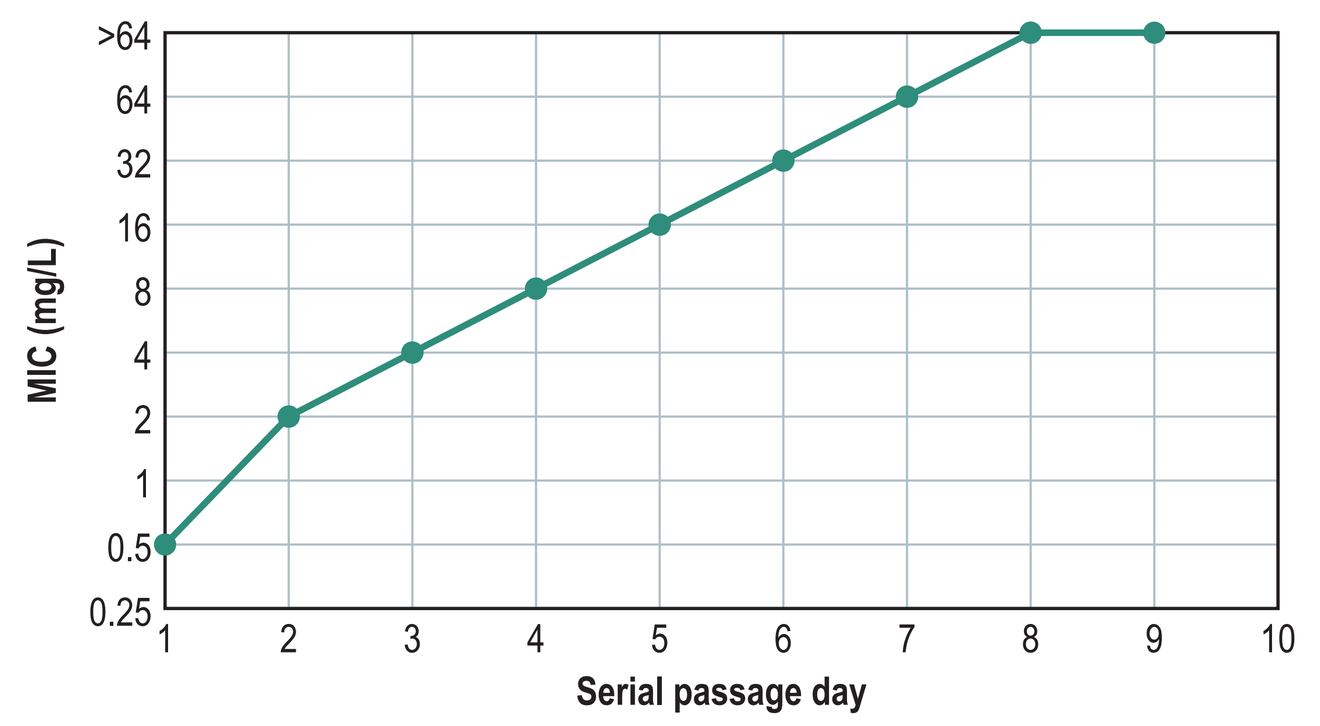
MIC (mg/L)
>64
64
32
16
8
4
2
1
0.5
0.25
1 2 3 4 5 6 7 8 9 10
Serial passage day

Table 1. KBP-7072 single-step frequency of resistance development

Isolate	KBP-7072 Agar dilution MIC (mg/L)	Starting Inoculum (CFU/mL	Volume Plated (mL)	Mutation frequency 4x MIC	Mutation frequency 8x MIC	Mutation frequency 16x MIC
A. baumannii	0.12	1.0×10^{9}	1 mL	9.0×10^{-8} [a]	1.4×10^{-8}	$<9.6 \times 10^{-10}$
(NCTC 13304)			0.1 mL	9.6×10^{-9}	$<9.6 \times 10^{-9}$	$<9.6 \times 10^{-9}$
A. baumannii	0.5	1.2×10^{9}	1 mL	2.7×10^{-7}	1.1×10^{-7}	$<8.7 \times 10^{-10}$
(1044704)			0.1 mL	2.4×10^{-7}	$<8.7 \times 10^{-9}$	$<8.7 \times 10^{-9}$
E. coli	0.12	1.2×10^{9}	1 mL	$<8.3 \times 10^{-10}$	$<8.3 \times 10^{-10}$	$<8.3 \times 10^{-10}$
(ATCC 25922)			0.1 mL	$<8.3 \times 10^{-9}$	$<8.3 \times 10^{-9}$	$<8.3 \times 10^{-9}$
E. coli	0.5	3.8×10^{9}	1 mL	2.1×10^{-8}	5.2×10^{-10}	$<2.6 \times 10^{-10}$
(1093450)			0.1 mL	1.6×10^{-7}	$<2.6 \times 10^{-9}$	$<2.6 \times 10^{-9}$
S. aureus	0.12	1.1×10^{9}	1 mL	$<9.4 \times 10^{-10}$	$<9.4 \times 10^{-10}$	$<9.4 \times 10^{-10}$
(ATCC 29213)			0.1 mL	$<9.4 \times 10^{-9}$	$<9.4 \times 10^{-9}$	$<9.4 \times 10^{-9}$
S. aureus	0.12	1.2×10^{9}	1 mL	$<8.6 \times 10^{-10}$	$<8.6 \times 10^{-10}$	$<8.6 \times 10^{-10}$
(1088771)			0.1 mL	$<8.6 \times 10^{-9}$	$<8.6 \times 10^{-9}$	$<8.6 \times 10^{-9}$
S. pneumoniae	0.008	1.2×10^{9}	1 mL	$>2.5 \times 10^{-7}$ [a]	$>2.5 \times 10^{-7}$ [a]	$>2.5 \times 10^{-7}$ [a]
(ATCC 49619)			0.1 mL	$>2.5 \times 10^{-6}$ [a]	8.2×10^{-9}	$<8.2 \times 10^{-9}$
			0.01 mL	6.6×10^{-7}	$<8.2 \times 10^{-8}$	$<8.2 \times 10^{-8}$
S. pneumoniae	0.015	1.3×10^{9}	1 mL	$>2.3 \times 10^{-7}$ [a]	$>2.3 \times 10^{-7}$ [a]	$>2.3 \times 10^{-7}$ [a]
(1119350)			0.1 mL	$>2.3 \times 10^{-6}$ [a]	$>2.3 \times 10^{-6}$ [a]	$>2.3 \times 10^{-6}$ [a]
			0.01 mL	$<7.6 \times 10^{-8}$	$<7.6 \times 10^{-8}$	$<7.6 \times 10^{-8}$

[a] A hazy lawn of growth was observed (no individual colonies), therefore a value of 301 was used to estimate the frequency of resistance value.

Conclusions

- ***S. aureus*** – KBP-7072 demonstrated a very low propensity for resistance development in single-step selection studies, and no resistant mutants were recovered. In serial passage testing, KBP-7072 MIC values remained unchanged or increased modestly (up to 4-fold; highest MIC, 0.25 mg/L) over the course of 10-daily serial passages.
- ***S. pneumoniae*** – In single-step selection studies, KBP-7072 demonstrated a low rate of resistance development with no more than a 4-fold MIC increase (highest MIC, 0.03 mg/L) following drug free passage and confirmatory MIC testing. In serial passage testing, KBP-7072 MIC values increased modestly (2- to 4-fold; highest MIC, 0.03 mg/L) over the course of 10-daily passages.
- ***A. baumannii*** – KBP-7072 demonstrated a low propensity for resistance development against *A. baumannii* in single-step selection studies. In serial passage testing, KBP-7072 MIC values increased 4- to 8-fold (highest MIC, 2 mg/L) over the course of 10-daily serial passages. Putative resistant mutants from the single-step and serial passage studies against *A. baumannii* demonstrated stably elevated MIC values to KBP-7072 and other tetracycline class agents.
- ***E. coli*** – In single-step selection studies, KBP-7072 demonstrated a low propensity for resistance development against *E. coli*. In serial passage testing, KBP-7072 MIC values increased 8- to >128-fold (highest MIC, >64 mg/L) over the course of 10-daily serial passages. Putative resistant mutants from the single-step and serial passage studies against *E. coli* demonstrated stably elevated MIC values to KBP-7072 and other tetracycline class agents.

Acknowledgements

This study was performed by JMI Laboratories and supported by KBP Biosciences Co., Ltd., which included funding for services related to preparing this poster.

References

CLSI. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. 11th ed. CLSI standard M07. Wayne, PA: Clinical and Laboratory Standards Institute; 2018.

CLSI. Performance standards for antimicrobial susceptibility testing. 31st ed. CLSI supplement M100. Clinical and Laboratory Standards Institute; 2021.

Contact

Michael D. Huband, Associate Director
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: michael-huband@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2021_KBP7072wSMAandSP.pdf

Charges may apply. No personal information is stored.

ECCMID 2021, July 9–12, 2021