ECCMID 2021 | Poster #01047

Activity of Meropenem-Vaborbactam and Comparators against European Carbapenem-Resistant *Enterobacterales* Isolates without a Carbapenemase

ECCMID 2021 TOP RATED POSTER

Dee Shortridge, Lalitagauri M. Deshpande, Jennifer M. Streit, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Meropenem-vaborbactam is a combination of a carbapenem and a β-lactamase inhibitor that is active against serine β-lactamases including carbapenemases.
 - Meropenem-vaborbactam has been approved in Europe for the treatment of the following infections in adults: complicated urinary tract infection (cUTI), including acute pyelonephritis, complicated intra-abdominal infection (cIAI), hospital-acquired bacterial pneumonia (HAP), ventilator-associated pneumonia (VAP), and bacteremia (BSI) that occurs in association with or is suspected to be associated with any of the infections listed above.
 - Meropenem-vaborbactam is also indicated for the treatment of infections due to aerobic Gram-negative organisms in adults with limited treatment options.
- Carbapenem-resistant *Enterobacterales* (CRE) isolates have disseminated worldwide.
 - These isolates typically produce a carbapenemase.
 - However, some CRE isolates are negative for known carbapenemases.
- In this study, we evaluated the activity of meropenem-vaborbactam and comparators against CREs without a carbapenemase (non-CP CREs) collected from European hospitals from 2016 to 2019.

Materials and Methods

- 23,043 *Enterobacterales* clinical isolates were consecutively collected from 41 European hospitals in 20 countries over the 4-year period.
 - Participating laboratories submitted 1 isolate per patient per infection episode.
- Susceptibility (S) testing was performed using the broth microdilution method.
- EUCAST (2021) interpretive criteria were used.
- 978 CREs were identified under the criteria of having a MIC >2 mg/L to doripenem, imipenem and/or meropenem. Imipenem was not used for *Proteus, Providencia,* or *Morganella* spp.
- Whole genome sequencing was performed on each CRE isolate.
- 125 CRE isolates were negative for known carbapenemase genes, including bla_{KPC}, bla_{NDM}, bla_{IMP}, bla_{VIM}, and $bla_{OXA\text{-}48\text{-}like}$.
- NonCP CRE isolates were analysed for the presence of other beta-lactamases, multilocus sequence typing (ST), increased expression of AmpC or efflux pumps, and disruptions in or increased expression of outer membrane protein (OMP) sequences.

Results

- The most common infections were pneumonia (n=37), UTI (n=26), cIAI (n=23), and BSI (n=22).
- The rate of CREs during this period was 4.2%.
 - 12.8% of the CREs lacked a known carbapenemase gene.
- The most common species of nonCP CRE were *Klebsiella pneumoniae* (n=97, 77.6%), *Enterobacter cloacae* species complex (n=11, 8.8%) and 10 *K. aerogenes* (8.0%; Figure 1).
- 84.0% of nonCP CRE (n=105) were from Poland, including 90.7% of nonCP CRE *K. pneumoniae* (n=88; Figure 1).
- Among the 92 *K. pneumoniae* isolates with an ST identified, 30 belonged to ST11, 18 to ST152, and 17 to ST147, but at least 13 other STs were observed (Figure 2).
 - The *K. pneumoniae* isolates from Poland were primarily ST11 (n=29), ST152 (n=18), and ST147 (n=17; Figure 2).
- Most (n=91, 72.8%) isolates carried $bla_{CTX\text{-}M\text{-}15}$, including 86 of 97 *K. pneumoniae* isolates.
 - Other acquired β-lactamases included bla_{SHV}, $bla_{OXA\text{-}1}$, bla_{TEM}, $bla_{DHA\text{-}1}$.
 - 96.8% of the isolates had two or more acquired β-lactamases.
- OMP disruptions were noted among the *Klebsiella pneumoniae*: 23 isolates had disruptions of both OmpK35 and OmpK36, 7 had only OmpK35 disrupted, and 43 had only OmpK36 disrupted.
- Meropenem-vaborbactam susceptibility was 97.6% while meropenem was 8.0% (Table 1 and Figure 3).
 - The MIC distribution of all isolates and *K. pneumoniae* with OMP disruptions or alterations is shown in Figure 3.
 - Two of the 3 meropenem-vaborbactam resistant isolates were *K. pneumoniae* and had alterations or disruptions in both OmpK35 and 36. These isolates also contained $bla_{CTX\text{-}M\text{-}15}$, $bla_{SHV\text{-}12}$, $bla_{OXA\text{-}1,\ OXA\text{-}10}$, and $bla_{TEM\text{-}57}$.
 - The third meropenem-vaborbactam resistant isolate was a *K. aerogenes* with elevated AmpC expression and an OmpC disruption.
- NonCP CRE isolates were resistant to many comparator agents. The most active agents were colistin and amikacin (susceptibility was 74.8% and 65.6%).

Table 1. Activity of meropenem-vaborbactam and comparator antimicrobial agents tested against 125 CRE, nonCP European isolates (2016–2019)

Antimicrobial agent	No. of isolates	mg/L MIC$_{50}$	MIC$_{90}$	MIC range	EUCAST[a] %S	%I	%R
Meropenem-vaborbactam	125	1	4	0.03 to 16	97.6		2.4
Meropenem	125	8	16	0.12 to 32	8.0 [b] 8.0 [c]	 80.0	92.0 12.0
Imipenem	125	4	>8	0.5 to >8	48.8	24.0	27.2
Amikacin	125	8	32	0.5 to >32	65.6 [d]		34.4
Aztreonam	125	>16	>16	2 to >16	0.0	3.2	96.8
Cefepime	125	>16	>16	0.5 to >16	0.8	8.8	90.4
Ceftazidime	125	>32	>32	2 to >32	0.0	2.4	97.6
Colistin	123	0.25	>8	≤0.06 to >8	74.8		25.2
Gentamicin	125	2	>8	≤0.12 to >8	56.0 [e]		44.0
Levofloxacin	125	>4	>4	≤0.03 to >4	11.2	5.6	83.2
Piperacillin-tazobactam	125	>64	>64	8 to >64	0.8		99.2

[a] Criteria as published by CLSI (2021) and EUCAST (2021).
[b] Using meningitis breakpoints.
[c] Using non-meningitis breakpoints.
[d] For infections originating from the urinary tract. For systemic infections, aminoglycosides must be used in combination with other active therapy.
[e] Intermediate is interpreted as susceptible-dose dependent.
Organisms include: *Enterobacter cloacae* species complex (11), *Escherichia coli* (3), *Hafnia alvei* (2), *Klebsiella aerogenes* (10), *K. oxytoca* (1), *K. pneumoniae* (97), and *Serratia marcescens* (1).

Figure 1. Country and species distributions of nonCP CREs in Europe

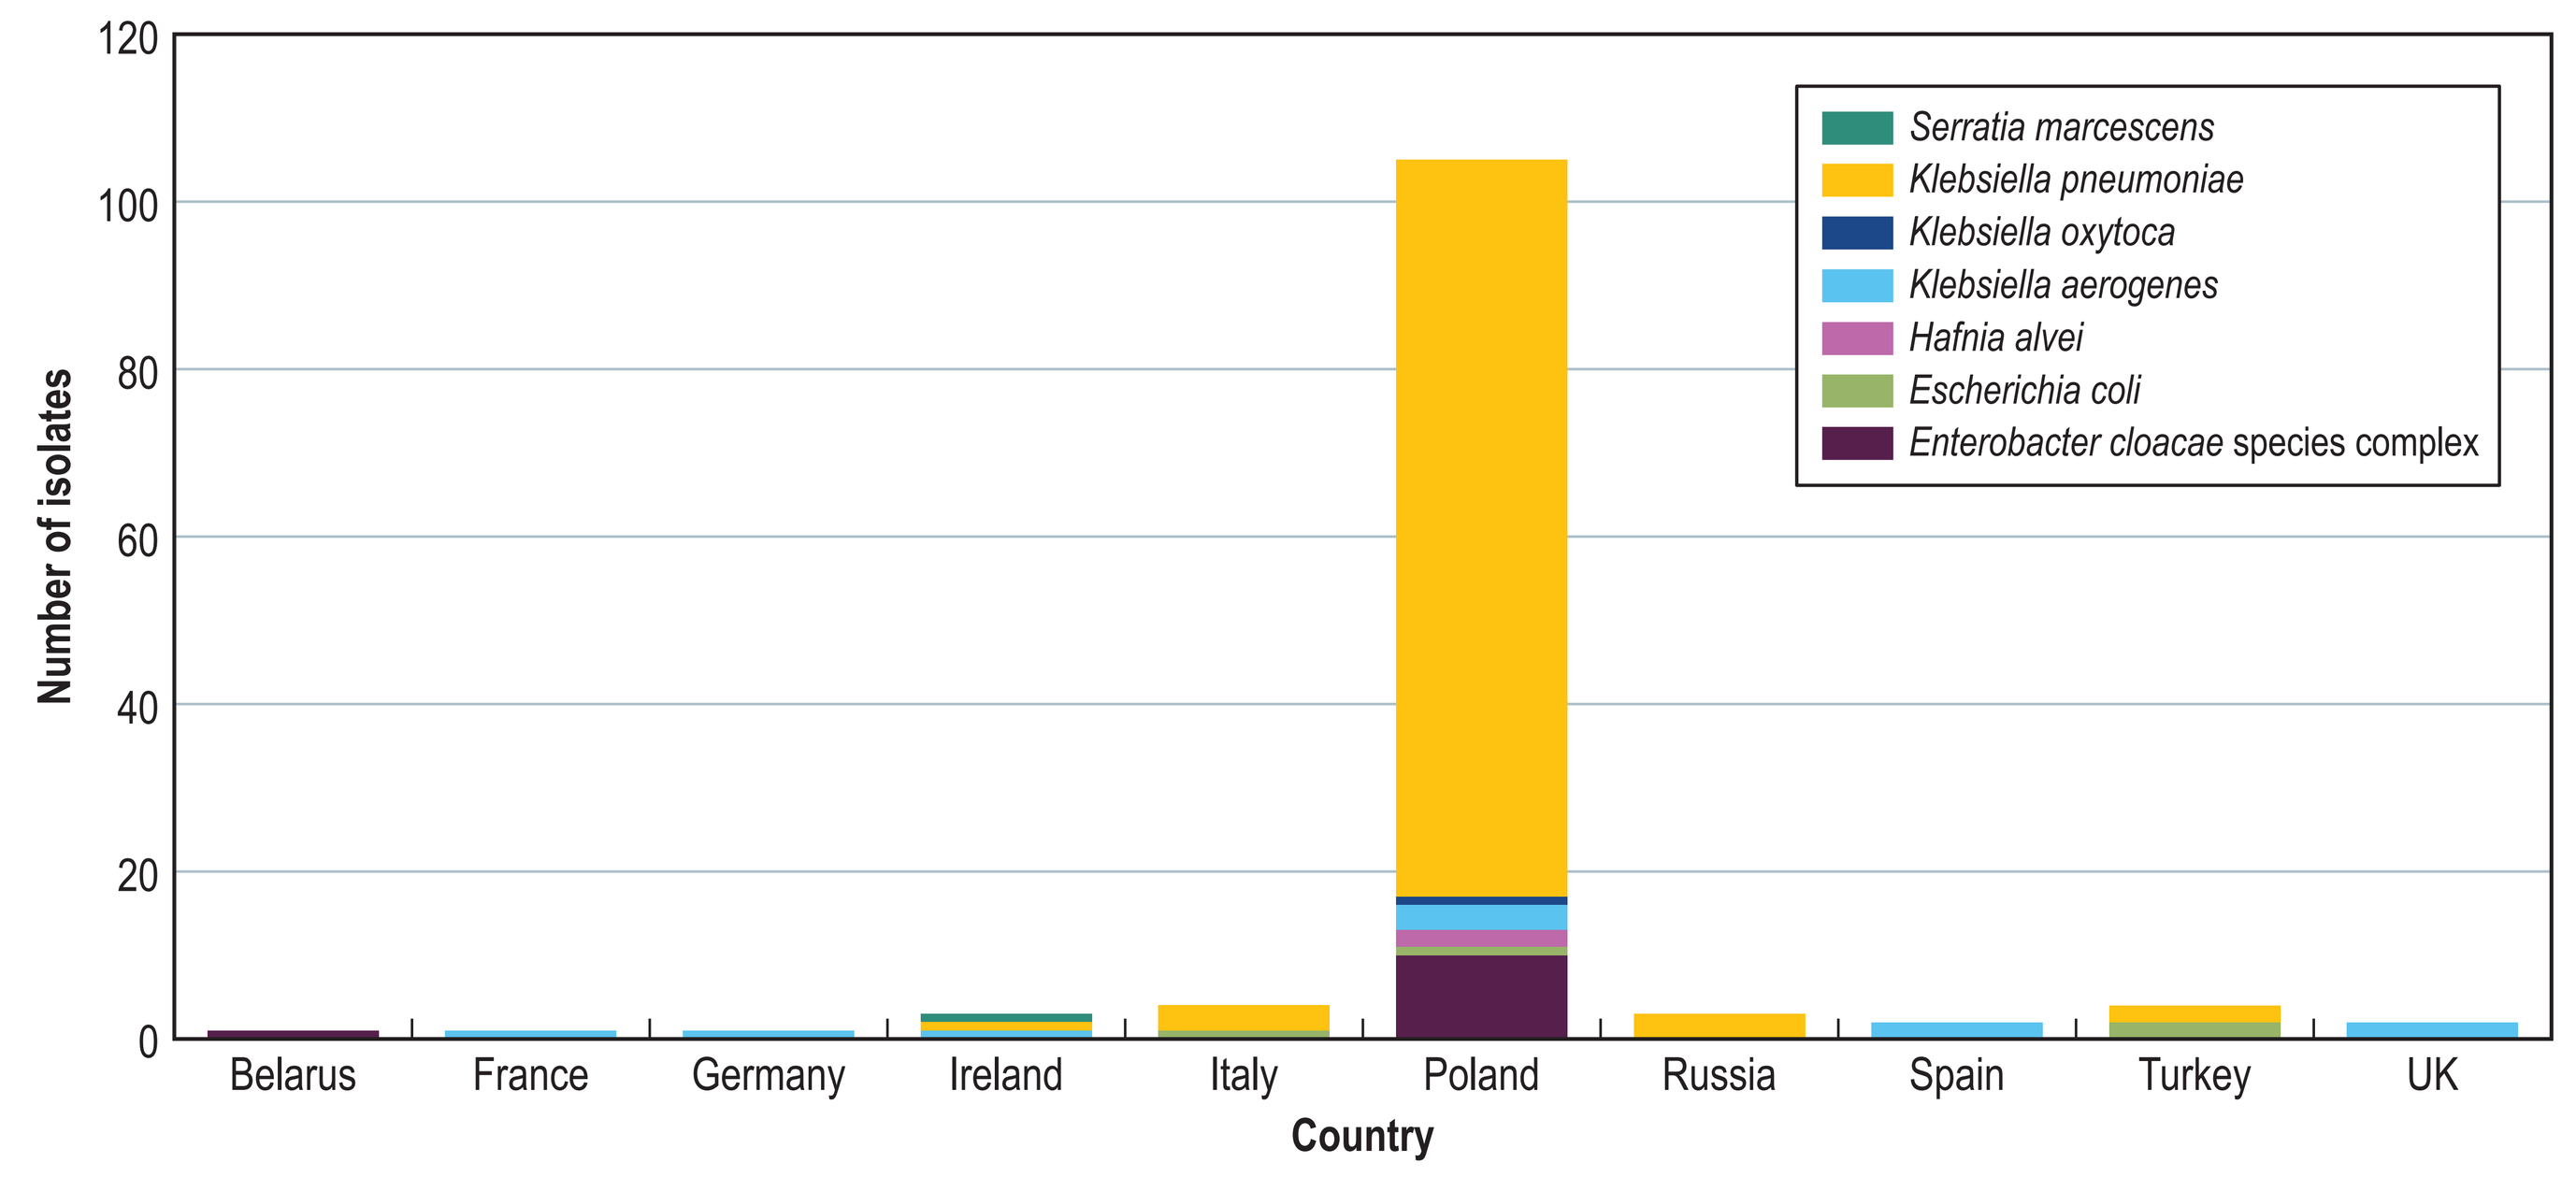
120
100
80
60
40
20
0
Number of isolates
Serratia marcescens
Klebsiella pneumoniae
Klebsiella oxytoca
Klebsiella aerogenes
Hafnia alvei
Escherichia coli
Enterobacter cloacae species complex
Belarus
France
Germany
Ireland
Italy
Poland
Russia
Spain
Turkey
UK
Country

Figure 2. *K. pneumoniae* sequence type (ST) distribution of nonCP CRE isolates by country

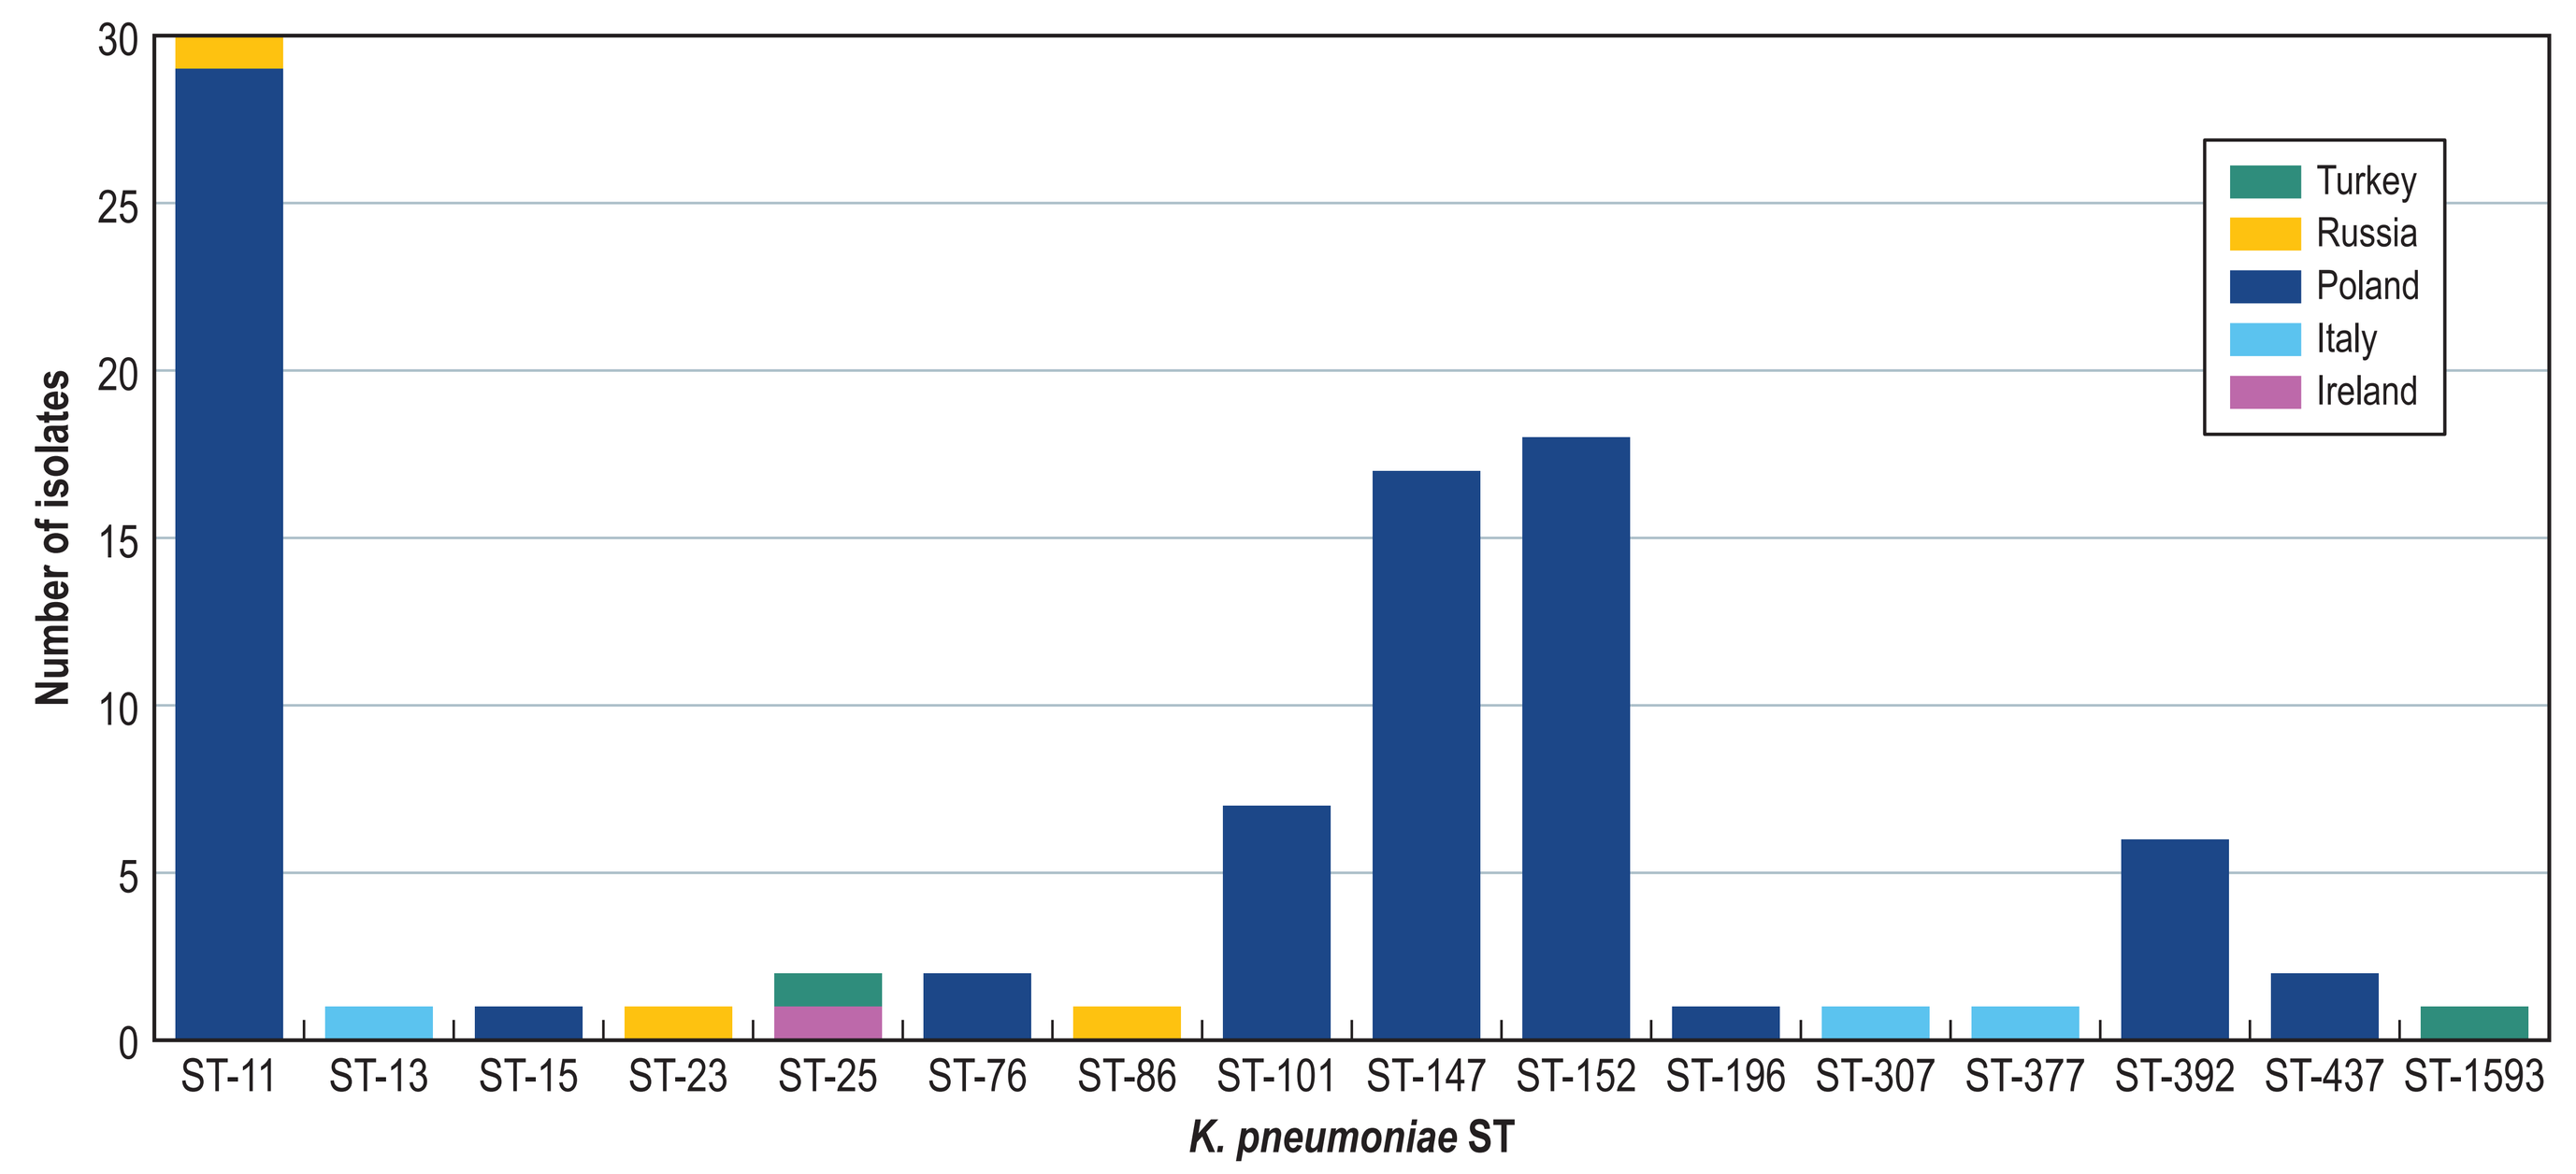
30
25
20
15
10
5
0
Number of isolates
Turkey
Russia
Poland
Italy
Ireland
ST-11
ST-13
ST-15
ST-23
ST-25
ST-76
ST-86
ST-101
ST-147
ST-152
ST-196
ST-307
ST-377
ST-392
ST-437
ST-1593
K. pneumoniae ST

Figure 3. MIC distribution of meropenem-vaborbactam (MVB) and meropenem (MER) tested against all nonCP CRE isolates and against *K. pneumoniae* isolates with OmpK35 or OmpK36 disruptions

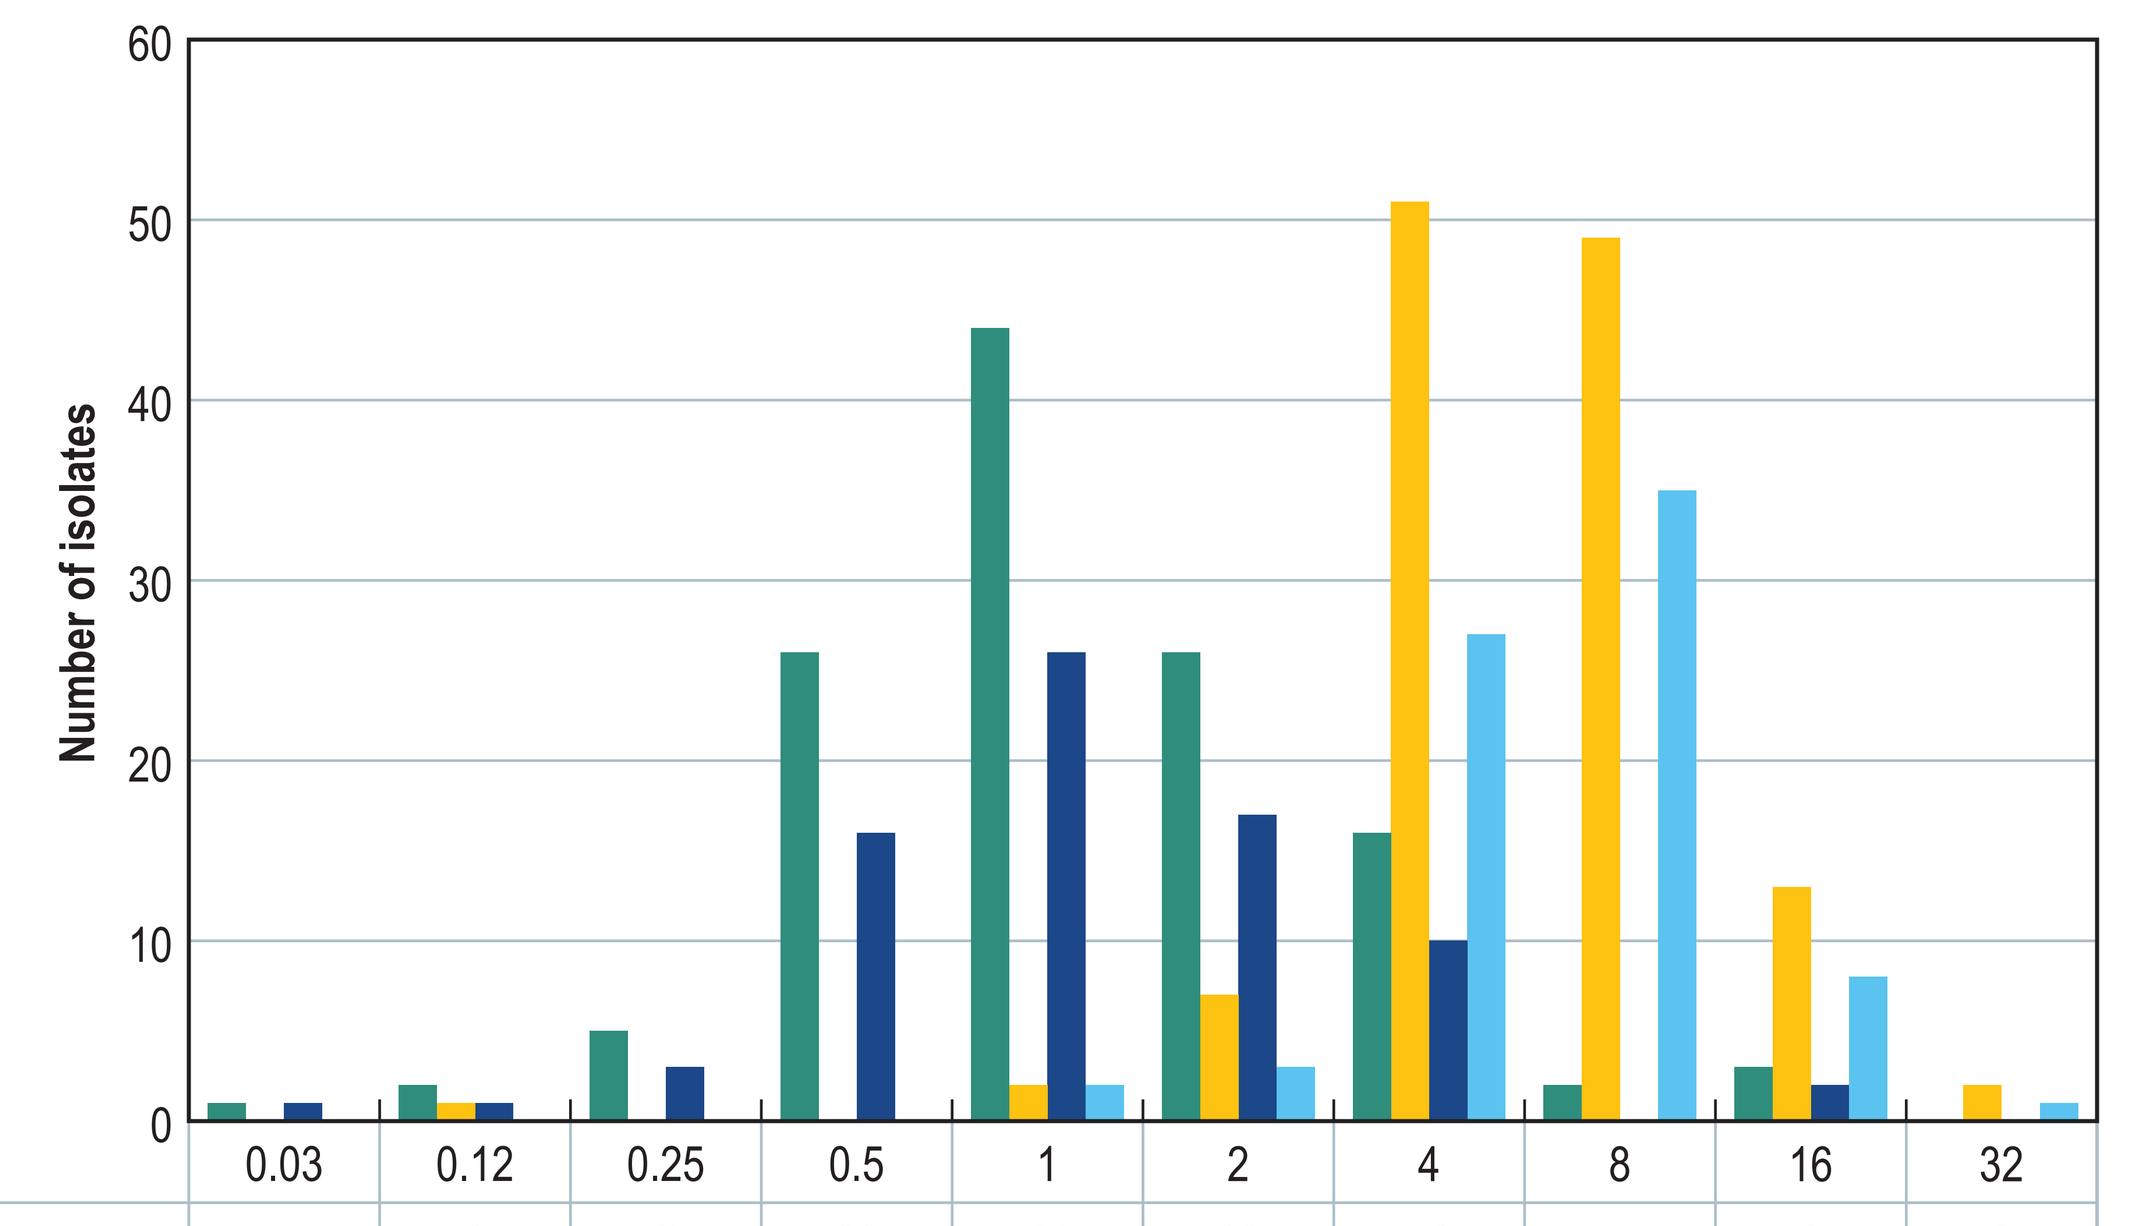
60
50
40
30
20
10
0
Number of isolates

	0.03	0.12	0.25	0.5	1	2	4	8	16	32
MVB All isolates	1	2	5	26	44	26	16	2	3	0
MER All isolates	0	1	0	0	2	7	51	49	13	2
MVB *K. pneumoniae* OMP mutations	1	1	3	16	26	17	10		2	
MER *K. pneumoniae* OMP mutations	0	0	0	0	2	3	27	35	8	1

MIC (mg/L)

Conclusions

- These results demonstrate meropenem-vaborbactam was the most active drug tested against CRE isolates that lack known carbapenemases, with 97.6% of these isolates considered susceptible.
 - 8.0% were susceptible to meropenem.
 - The higher EUCAST breakpoint of meropenem-vaborbactam reflect the higher dose of the meropenem component and a maximal inhibitory effect of the vaborbactam component.
- *K. pneumoniae* was the most common nonCP CRE accounting for 77.6%.
 - 84% of the *K. pneumoniae* isolates were from Poland and represented 9 different ST types.
- Resistance mechanisms observed among the nonCP CRE isolates in this study included multiple acquired β-lactamases (CTX-M-15, OXA, SHV, and TEM) and/or disruption of OmpC (*K. aerogenes* and *E. cloacae*) or OmpK35 or K36 in *K. pneumoniae*.
- The increased activity of meropenem-vaborbactam against these isolates shows that inhbition of the non-carbapenemase β-lactamases by vaborbactam increases the activity of meropenem alone.
- These results suggest that meropenem-vaborbactam may be a useful treatment for infections caused by CREs that lack a carbapenemase.

Acknowledgements

This poster has been funded by A. Menarini Industrie Farmaceutiche Riunite SRL.

References

CLSI. *M07 Eleventh edition. Methods for Dilution Antimicrobial Susceptibility Testing for Bacteria That Grow Aerobically.* Clinical and Laboratory Standards Institute, Wayne, PA, 2018.

EUCAST. The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters, v 11.0, January 1, 2021. http://www.eucast.org (Access Date Jan 31, 2021).

Castanheira M, Rhomberg PR, Flamm RK and Jones RN. Effect of the β-lactamase inhibitor vaborbactam combined with meropenem against serine carbapenemase-producing *Enterobacteriaceae*. Antimicrob Agents Chemother 60:545-8. 2016.

Contact

Dee Shortridge
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2021_MeroVaborVEuroCRE.pdf

Charges may apply. No personal information is stored.

ECCMID 2021, July 9–12, 2021