ECCMID 2024 | Poster #P2452

Proposed zosurabalpin broth microdilution quality control ranges using Clinical and Laboratory Standards Institute M23 Tier 2 criteria

Michael D. Huband[1], Joshua M. Maher[1], Yahse Edah[1], Mariana Castanheira[1], Séverine Louvel[2], Claudia Zampaloni[2]

[1] Element Iowa City (JMI Laboratories), IA, USA, [2] Roche Pharma Research and Early Development, Cardiovascular & Metabolism, Immunology, Infectious Diseases and Ophthalmology, Roche Innovation Center Basel, F. Hoffmann La Roche Ltd, Grenzacherstrasse 124, 4070 Basel, Switzerland

Introduction

- New antibacterials with activity against Gram-negative pathogens including *Acinetobacter baumannii* are urgently needed. Zosurabalpin (RG6006) is a first-in-class novel tethered macrocyclic peptide antibiotic in Phase 1 clinical development (NCT05614895).
- Zosurabalpin is active against *Acinetobacter* spp., including carbapenem-resistant *A. baumannii-calcoaceticus* complex organisms, exerting its antimicrobial activity through inhibition of the lipopolysaccharide transporter ($LptB_2FGC$).
- Broth microdilution susceptibility testing of zosurabalpin in cation-adjusted Mueller-Hinton broth (CAMHB) supplemented with 20% heat inactivated horse serum was approved at the January 2024 Clinical and Laboratory Standards Institute (CLSI) meeting in Tempe, Arizona.
- In this study, we performed CLSI M23 Tier 2 broth microdilution quality control (QC) testing to generate proposed zosurabalpin QC ranges.

Materials and Methods

- CLSI M23 Tier 2 broth microdilution QC studies were conducted with zosurabalpin and meropenem against *A. baumannii* NCTC 13304, *A. baumannii* ROB00867, *A. baumannii* ROB07643, and *Pseudomonas aeruginosa* ATCC 27853.
- A trailing effect and/or skipped wells are observed with some *A. baumannii* isolates when zosurabalpin is tested in CAMHB, thus preventing a clear MIC determination.
- Zosurabalpin MIC testing was conducted in CAMHB and CAMHB supplemented with 10% and 20% heat-inactivated horse serum.
- Zosurabalpin MIC values were read at substantial reduction of growth and complete (100%) growth inhibition when tested in CAMHB.
- Zosurabalpin MIC values were read at complete (100%) growth inhibition when tested in CAMHB supplemented with 10% and 20% heat inactivated horse serum.
- The Tier 2 broth microdilution QC study utilized 8 laboratories, 3 media lots from 3 different manufacturers, and 10 MIC replicates per QC strain tested over ≥3 days.

Results

- Proposed zosurabalpin MIC QC ranges against the 3 *A. baumannii* strains using CAMHB containing 10% or 20% horse serum (100% growth inhibition MIC reading endpoint) were the same or within ±1 dilution of those obtained in CAMHB where the MIC reading endpoint was interpreted using substantial reduction of growth (Table 1). The addition of horse serum made the MIC endpoints easier to read and did not significantly alter the proposed QC ranges.
- A 3-dilution broth microdilution QC range (0.016–0.06 µg/mL) containing 100% of all zosurabalpin MIC values is proposed when testing zosurabalpin in CAMHB against *A. baumannii* NCTC 13304 and the MIC endpoint is read at substantial growth reduction (Table 1 and Figure 1).
- A 4-dilution broth microdilution QC range (0.016–0.12 µg/mL) containing 99.2%–100% of all zosurabalpin MIC values is proposed when testing zosurabalpin in CAMHB or CAMHB containing 10% or 20% horse serum against *A. baumannii* NCTC 13304 and the MIC is read at 100% growth inhibition (Table 1 and Figures 2–4).
- A 3-dilution broth microdilution QC range (0.06–0.25 µg/mL) containing 97.9%–98.3% of all zosurabalpin MIC values is proposed when testing zosurabalpin in CAMHB containing 10% or 20% horse serum against *A. baumannii* ROB00867 (Table 1).
- A 3-dilution broth microdilution QC range (0.016–0.06 µg/mL) containing 95.8%–97.9% of all zosurabalpin MIC values is proposed when testing zosurabalpin in CAMHB containing 10% or 20% horse serum against *A. baumannii* ROB07643 (Table 1).
- No QC ranges are proposed for *A. baumannii* ROB00867 or ROB07643 when testing in CAMHB and the MIC is read at complete (100%) inhibition due to strong MIC trailing (zosurabalpin MIC values, >8 µg/mL) (Table 1).
- All (100%; 240/240) of the meropenem MIC values against *A. baumannii* NCTC 13304 and 97.9% (235/240) of the meropenem MIC values against *P. aeruginosa* ATCC 27853 were within current CLSI QC ranges published in the M100 (2023) document (data not shown).

Figure 1. Zosurabalpin MIC distributions by medium lot for *Acinetobacter baumannii* NCTC 13304 when tested in CAMHB and the MIC endpoint is read as substantial growth reduction

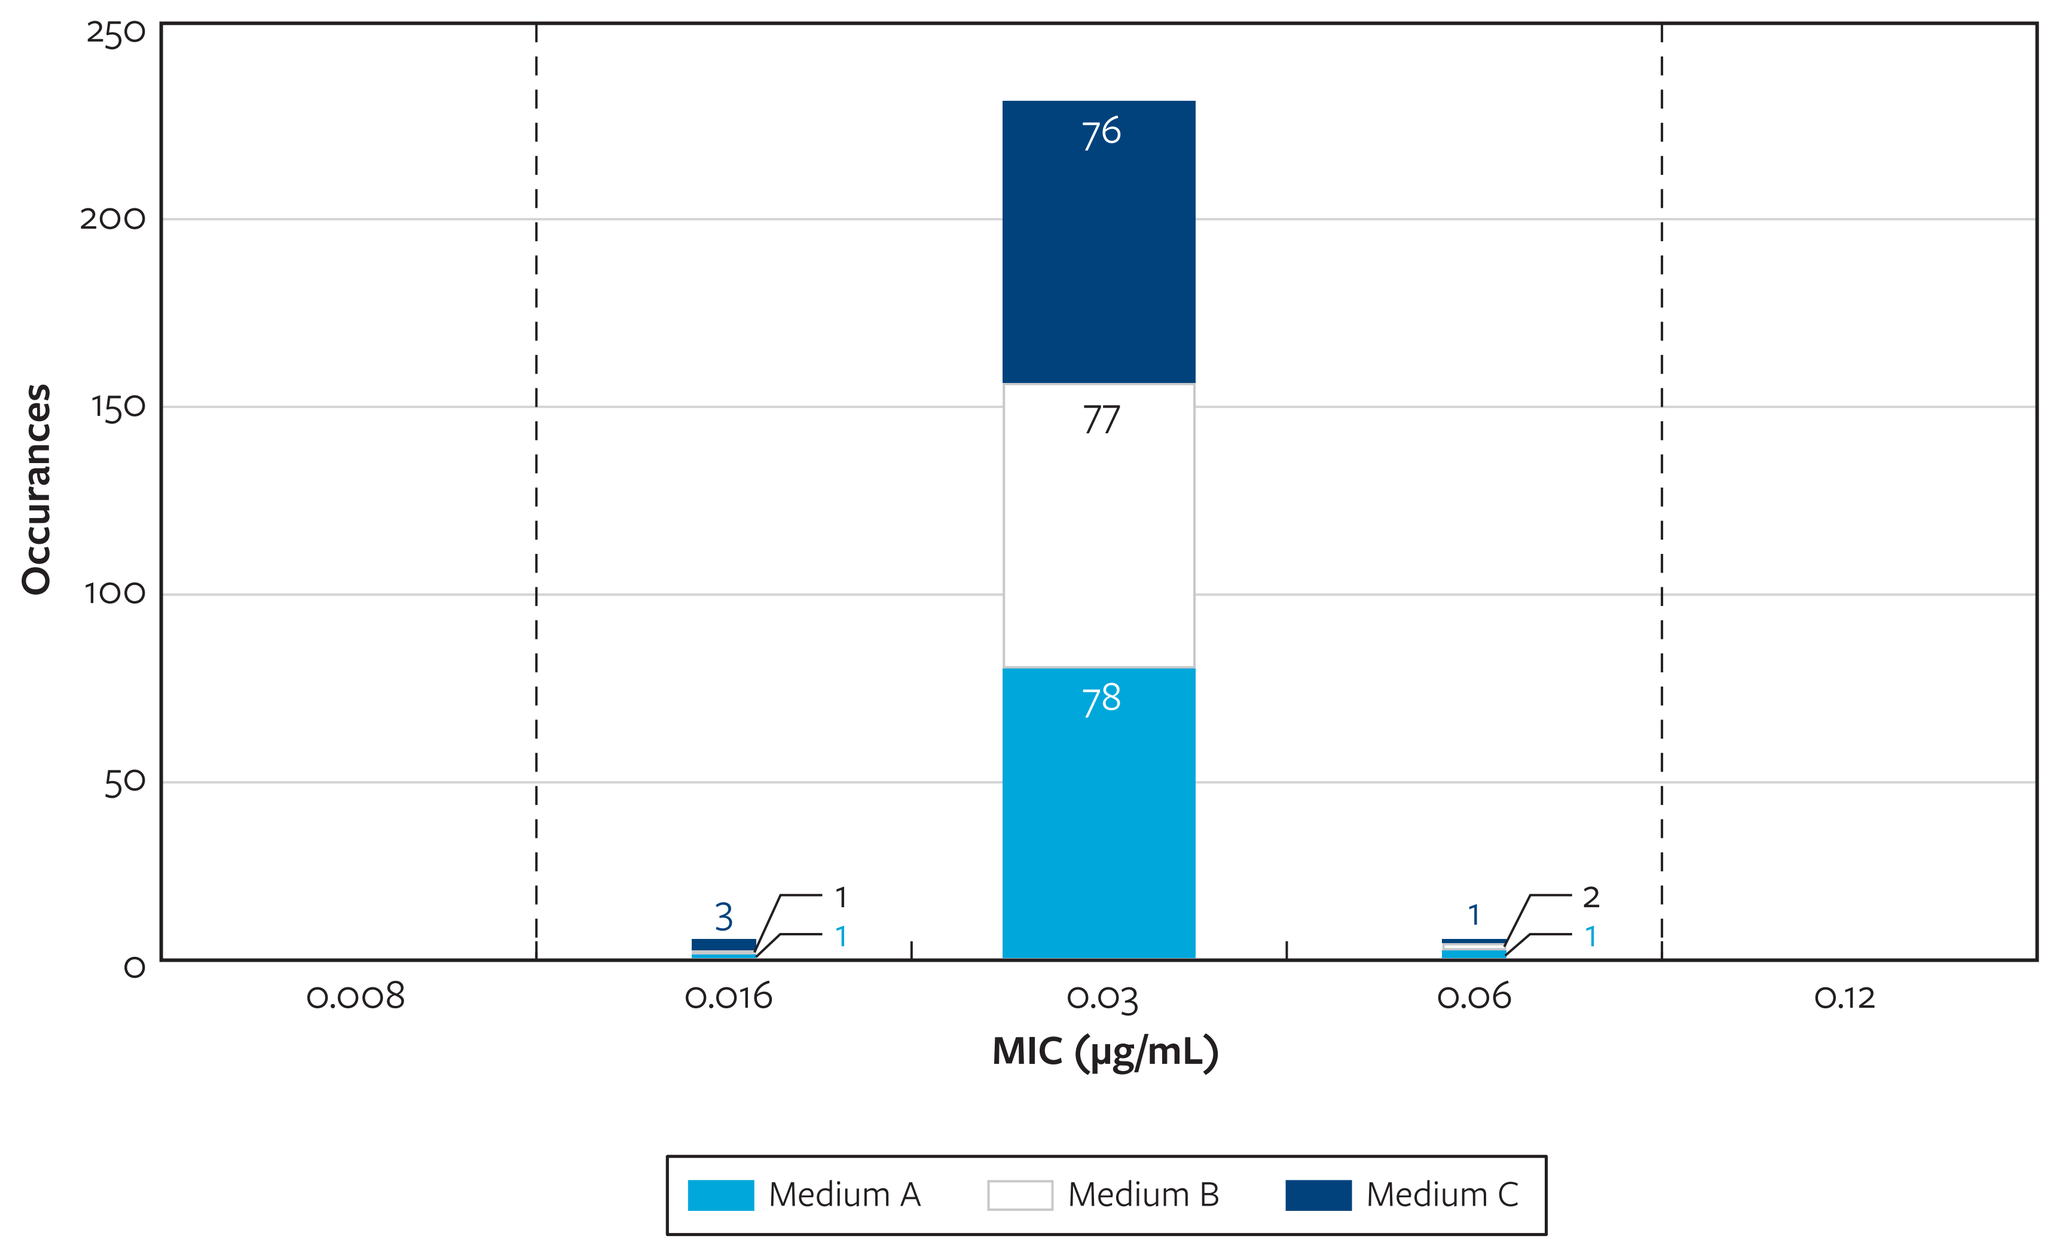

Occurances
250
200
150
100
50
0
0.008
0.016
0.03
0.06
0.12
MIC (µg/mL)
3
1
1
76
77
78
1
2
1
Medium A
Medium B
Medium C

A, Difco, Lot #3150800; B, BBL (BD), Lot #2343309; C, Oxoid, Lot #3566128.

Figure 2. Zosurabalpin MIC distributions by medium lot for *Acinetobacter baumannii* NCTC 13304 when tested in CAMHB and the MIC is read at 100% growth inhibition

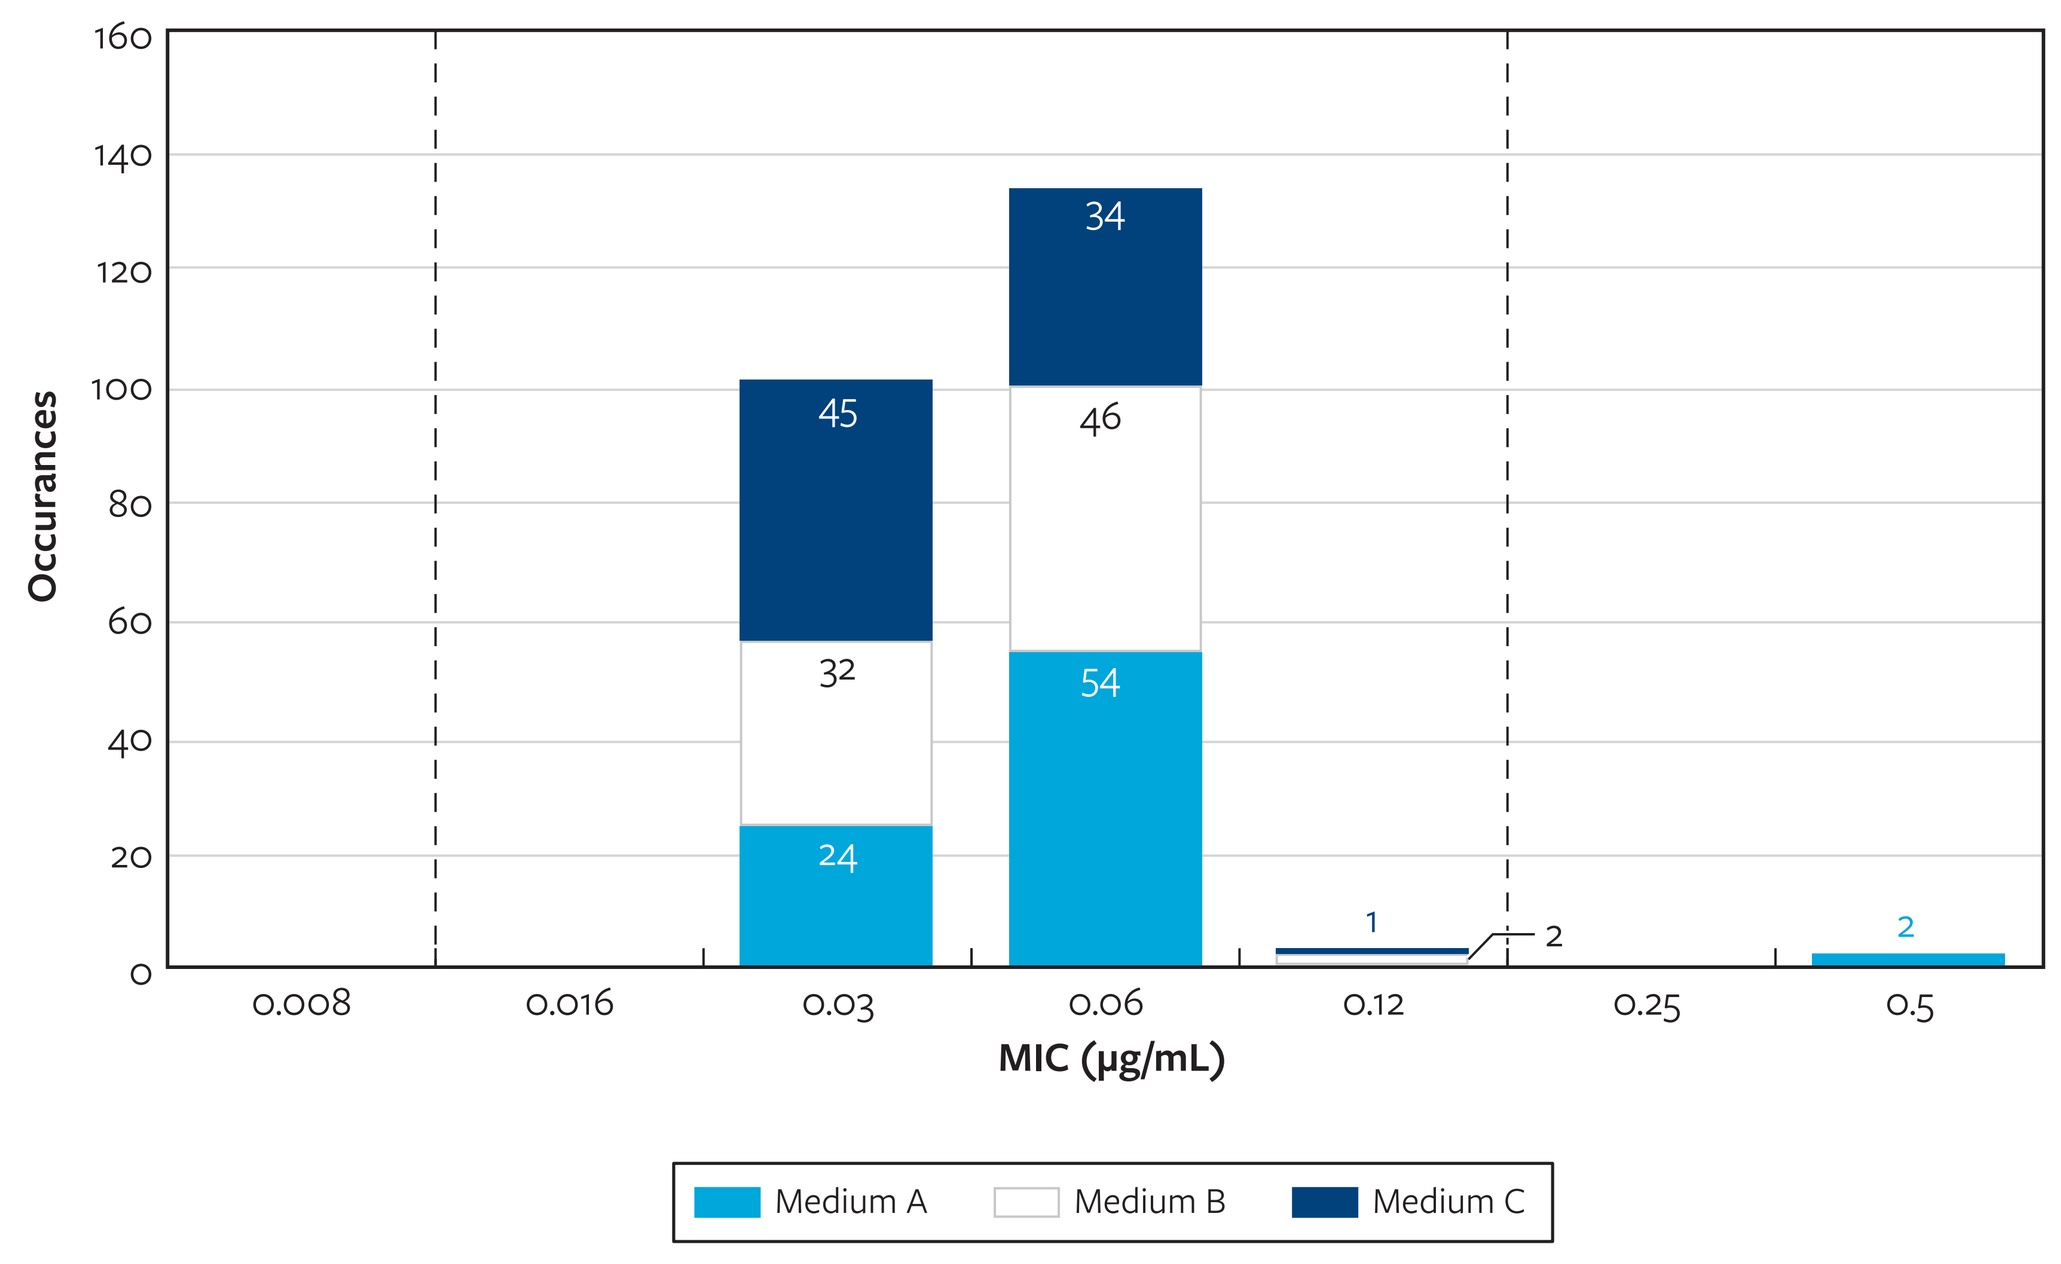

Occurances
160
140
120
100
80
60
40
20
0
0.008
0.016
0.03
0.06
0.12
0.25
0.5
MIC (µg/mL)
45
32
24
34
46
54
1
2
2
Medium A
Medium B
Medium C

A, Difco, Lot #3150800; B, BBL (BD), Lot #2343309; C, Oxoid, Lot #3566128.
[a] An 74.8% zosurabalpin MIC shoulder was observed at 0.03 µg/mL.

Figure 3. Zosurabalpin MIC distributions by medium lot for *Acinetobacter baumannii* NCTC 13304 when tested in CAMHB + 10% horse serum

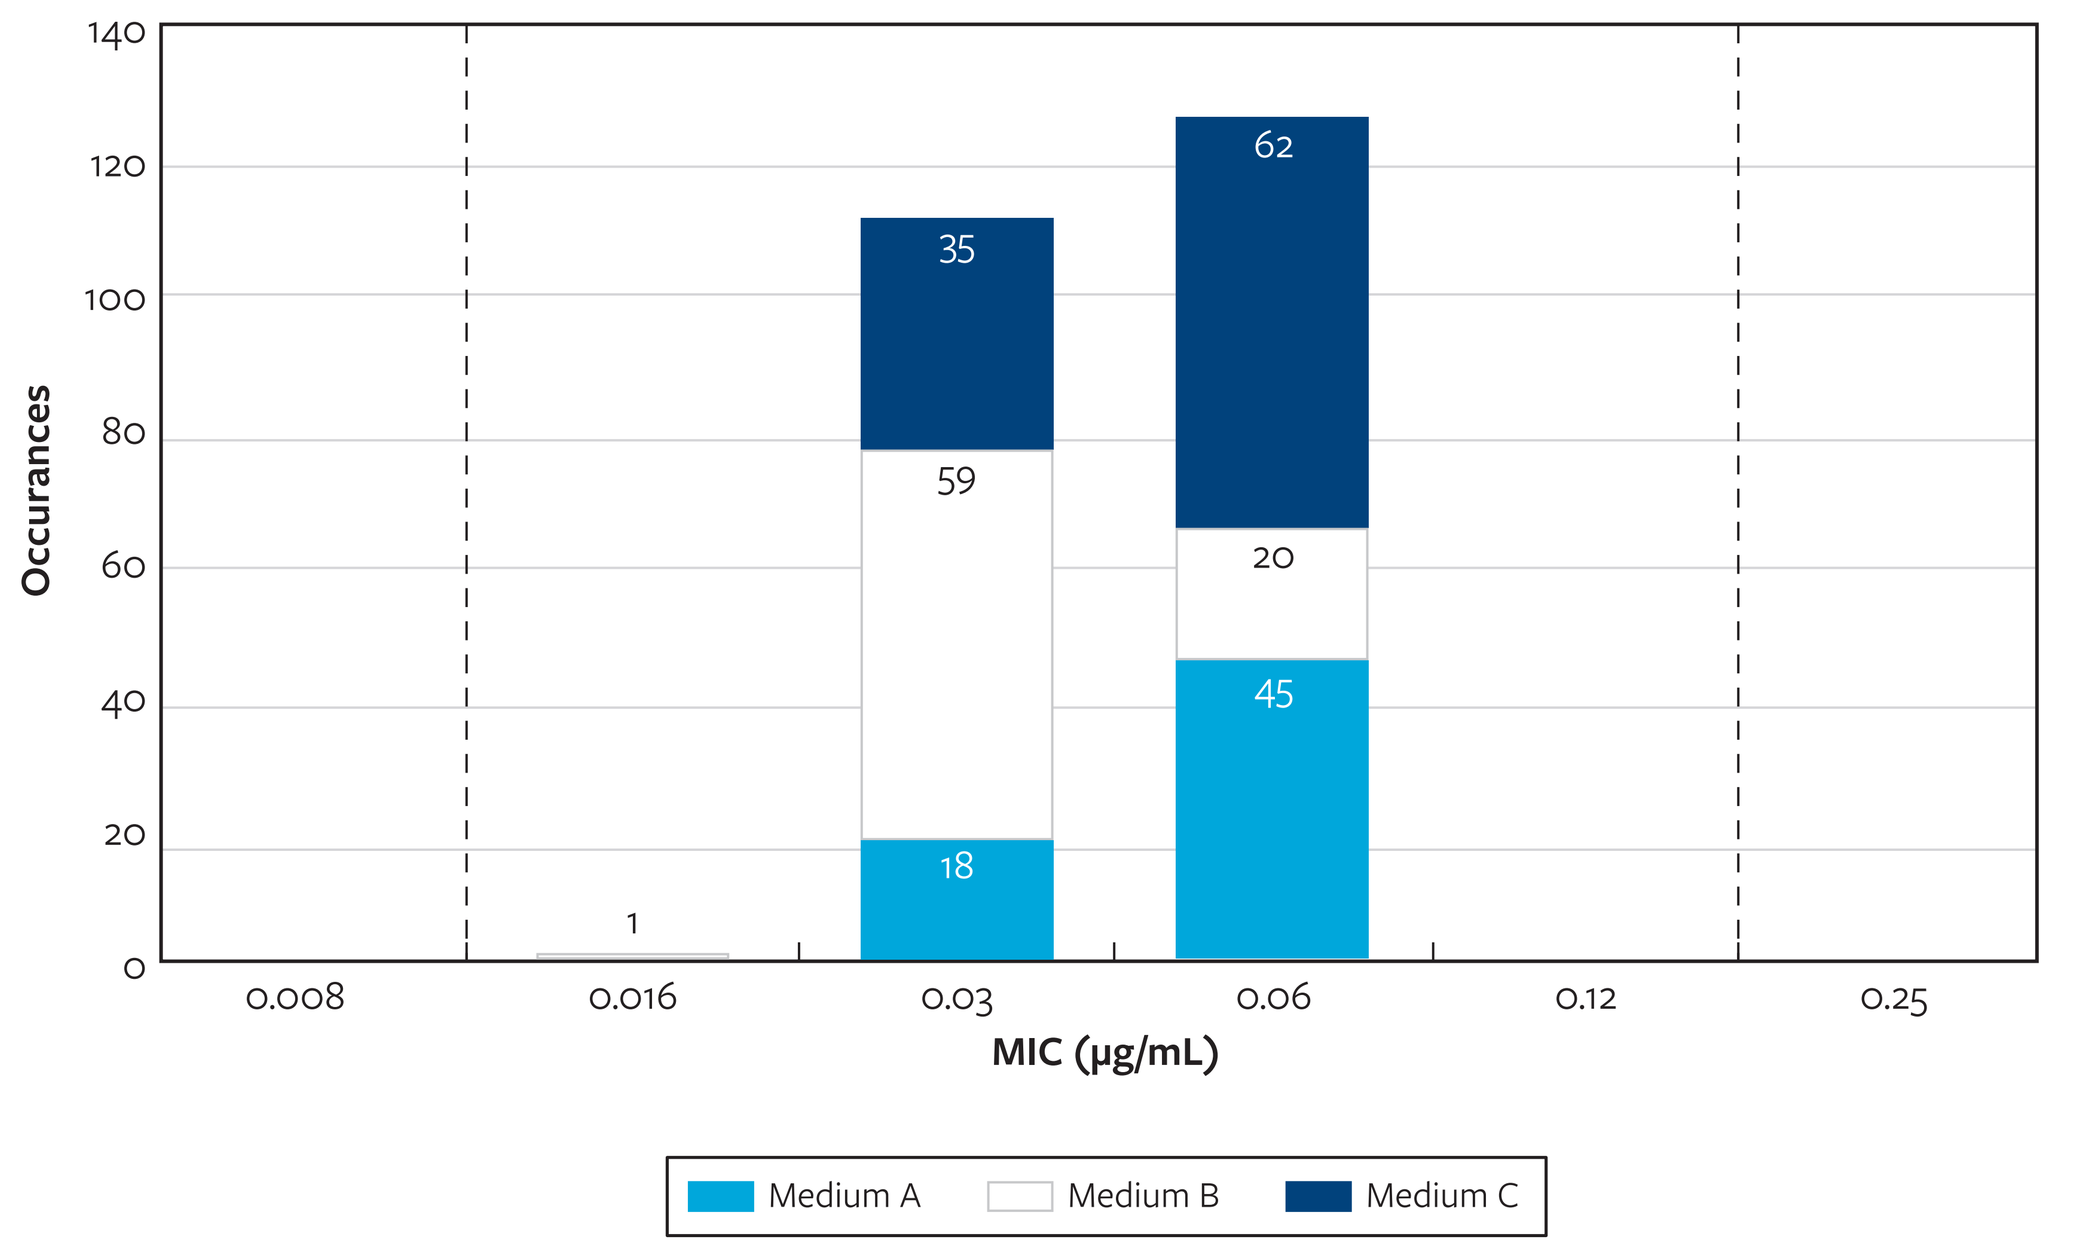

Occurances
140
120
100
80
60
40
20
0
0.008
0.016
0.03
0.06
0.12
0.25
MIC (µg/mL)
1
35
59
18
62
20
45
Medium A
Medium B
Medium C

A, Difco, Lot #3150800; B, BBL (BD), Lot #2343309; C, Oxoid, Lot #3566128.
[a] An 88.2% zosurabalpin MIC shoulder was observed at 0.03 µg/mL.

Figure 4. Zosurabalpin MIC distributions by medium lot for *Acinetobacter baumannii* NCTC 13304 when tested in CAMHB + 20% horse serum

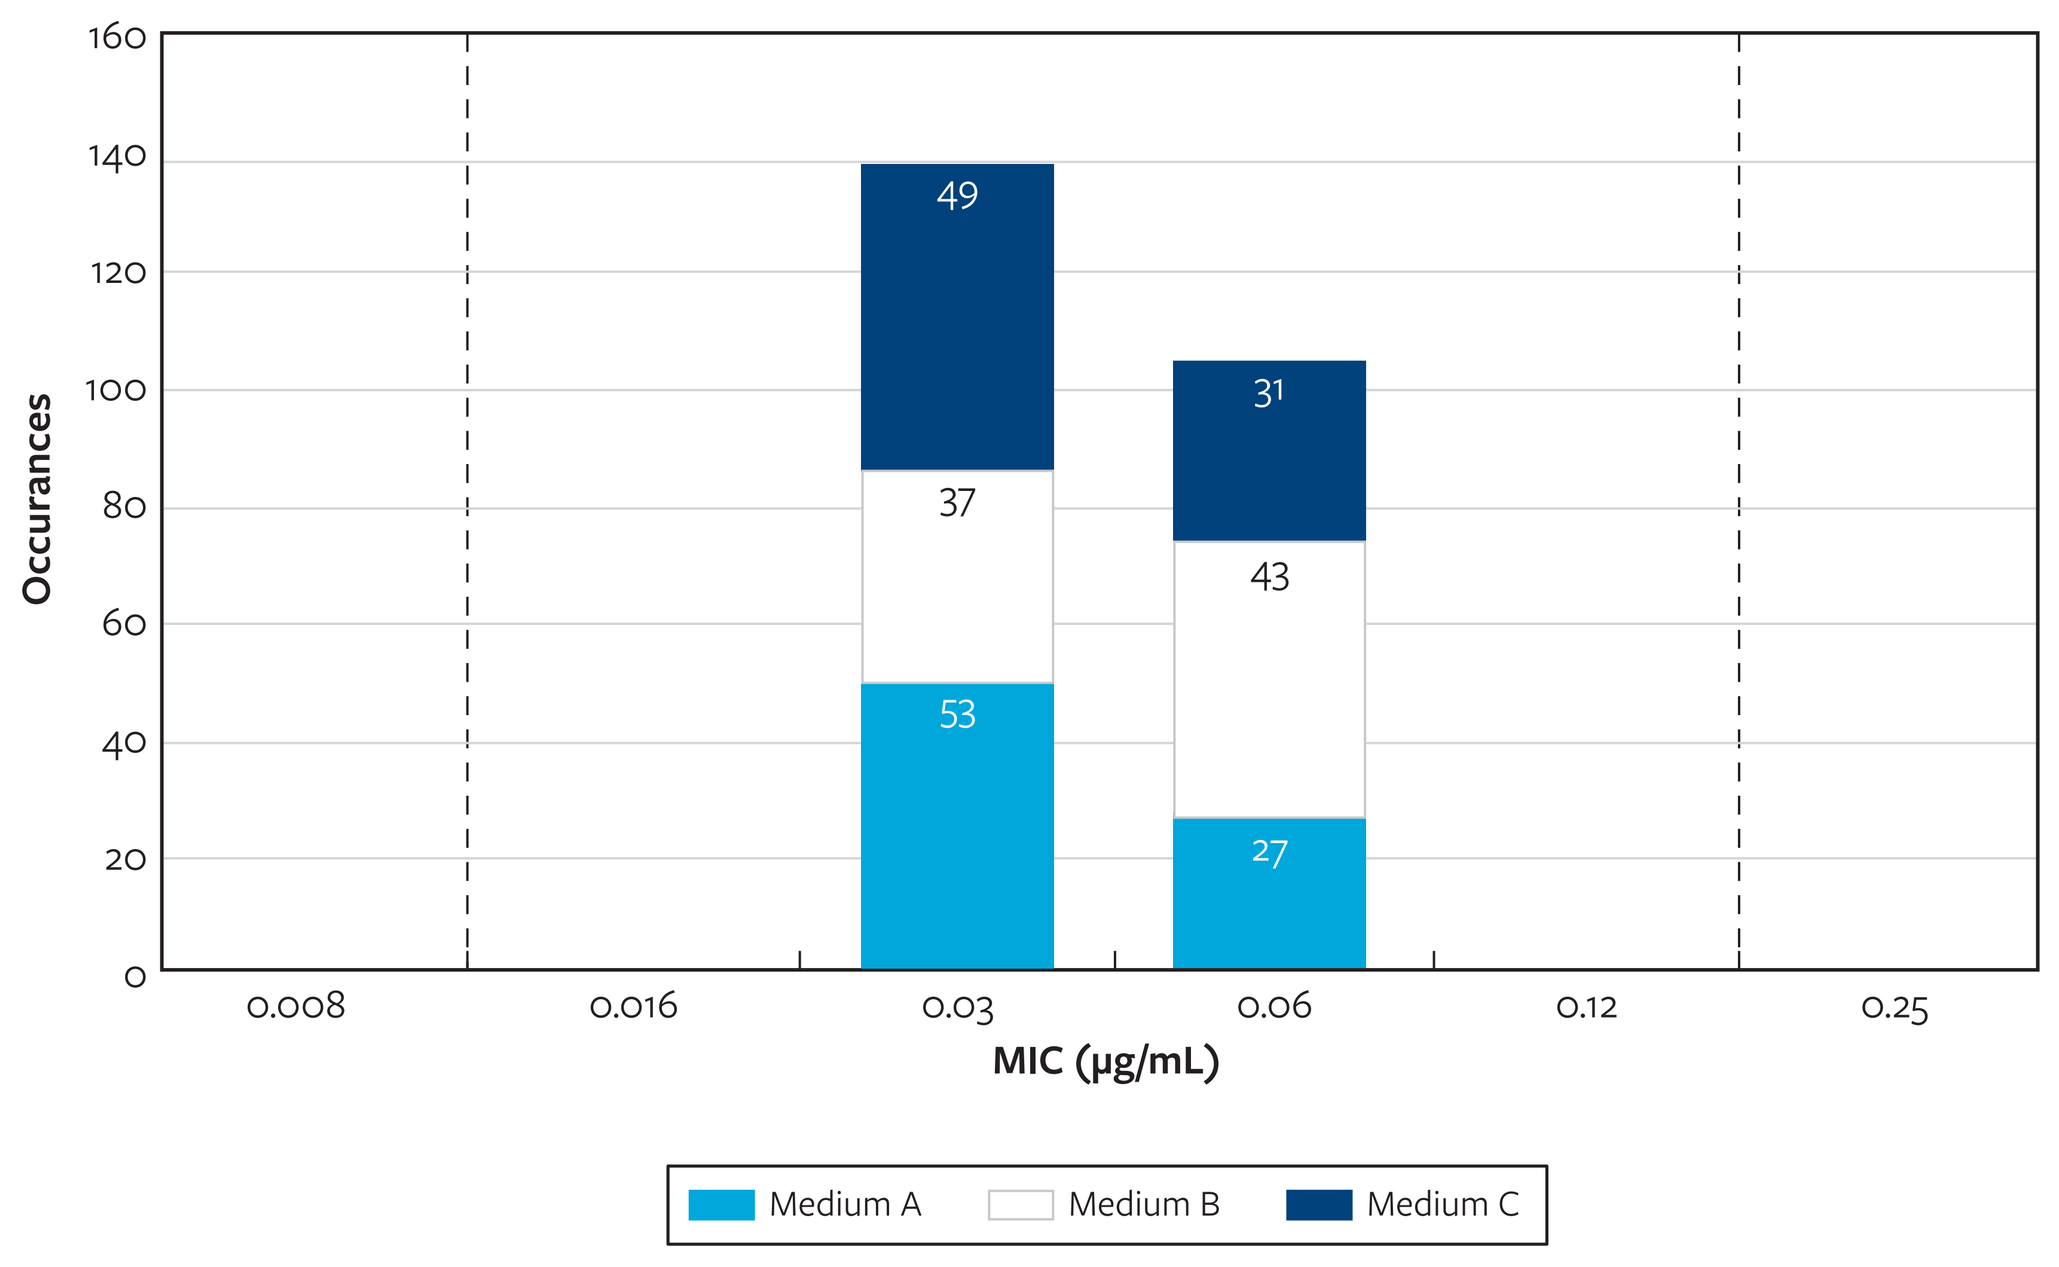

Occurances
160
140
120
100
80
60
40
20
0
0.008
0.016
0.03
0.06
0.12
0.25
MIC (µg/mL)
49
37
53
31
43
27
Medium A
Medium B
Medium C

A, Difco, Lot #3150800; B, BBL (BD), Lot #2343309; C, Oxoid, Lot #3566128.
[a] A 72.7% zosurabalpin MIC shoulder was observed at 0.06 µg/mL.

Table 1. Proposed zosurabalpin quality control ranges

	Proposed Zosurabalpin MIC QC Ranges (µg/mL)			
	CAMHB (Substantial Growth Reduction)	CAMHB (100% Growth Inhibition)	CAMHB +10% Horse Serum (100% Growth Inhibition)	CAMHB +20% horse serum (100% Growth Inhibition)
Strain Tested	(% in range; no. of dilutions)			
A. baumannii NCTC 13304	0.016 – 0.06[a] (100.0%; 3)	0.016 – 0.12[b] (100.0%; 4)	0.016 – 0.12[a] (100.0%; 4)	0.016 – 0.12[a] (100.0%; 4)
A. baumannii ROB00867	0.03 – 0.12[a] (97.9%; 3)	No proposed QC range	0.06 – 0.25[a] (98.3%; 3)	0.06 – 0.25[a] (97.9%; 3)
A. baumannii ROB07643	0.016 – 0.06[b] (99.0%; 3)	No proposed QC range	0.016 – 0.06[b] (97.9%; 3)	0.016 – 0.06[b] (95.8%; 3)
P. aeruginosa ATCC 27853	No proposed QC range	No proposed QC range	No proposed QC range	No proposed QC range

[a] CLSI criteria.
[b] CLSI criteria and the RangeFinder statistical program.

Conclusions

- Establishing a CLSI broth microdilution QC range for zosurabalpin against *A. baumannii* NCTC 13304 (0.016–0.12 mg/L) in CAMHB containing 20% heat inactivated horse serum will assist clinical laboratories and reference laboratories participating in clinical trials.

Acknowledgements

This project has been supported in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Biomedical Advanced Research and Development Authority, under contract number HHSO100201600038C.

References

CLSI. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.

CLSI. M23E6. Development of in vitro susceptibility test methods, breakpoints, and quality control parameters. 6^{th} ed. CLSI guideline M23. Clinical and Laboratory Standards Institute; 2023.

CLSI. M100Ed33. Performance standards for antimicrobial susceptibility testing. Wayne, PA, Clinical and Laboratory Standards Institute, 2023.

Contact

Michael Huband
Element Iowa City
(JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317

Phone: (319) 665-3370 x203
Fax: (319) 665-3371
Email: michael.huband@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ECCMID2024_23-ROC-08_P1_M23_MIC.pdf

Charges may apply. No personal information is stored.

ECCMID 2024, April 27–30, 2024, Barcelona, Spain