ESCMID 2026 | Poster #P3308

Activity of manogepix against *Candida auris*

Marisa L. Winkler, J. Hank Kimbrough, Samuel Edeker, Abby Klauer, Paul R. Rhomberg, Mariana Castanheira
Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- *Candida auris* is a rising threat worldwide due to high antifungal resistance rates and tolerance to cleaning solutions
- Fosmanogepix (prodrug of manogepix) is a novel antifungal agent in Phase 3 clinical trials for the treatment of invasive yeast and mould infections
- Manogepix targets the fungal Gwt1 enzyme, which is a different mechanism of action relative to currently approved antifungal agents: azoles, echinocandins, and amphotericin B
- There is no cross-resistance among manogepix and other antifungal agents
- The *in vitro* activity of manogepix and comparator antifungal agents was evaluated against *C. auris* isolates

Methods

- A total of 169 *C. auris* isolates were collected from 20 different hospitals in 9 different countries over 7 years
- Only 1 isolate per patient episode was included
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing
- Isolates were tested by CLSI reference broth microdilution method (M27) to obtain minimal inhibitory concentration (MIC) measurements
- CDC breakpoints were applied for comparator agents; no breakpoints or epidemiological cutoff values are available for manogepix
- Whole genome sequencing was performed on isolates resistant to one or more echinocandins with analysis of FKS hotspot (HS) regions

Table 1. MIC range, $MIC_{50/90}$, and % organisms non-resistant for manogepix and comparator agents for *C. auris*

Antifungal agent	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	% not resistant[1]
Manogepix	≤0.002 to 0.25	0.015/0.03	NA
Micafungin	0.06 to >4	0.25/0.5	97.0
Anidulafungin	0.06 to >4	0.25/0.5	97.6
Caspofungin	0.015 to >4	0.12/0.5	92.3
Fluconazole	2 to >128	128/>128	7.7
Itraconazole	0.06 to >8	0.5/1	NA
Posaconazole	0.03 to >8	0.25/0.5	NA
Voriconazole	0.015 to >8	1/2	NA
Amphotericin B	0.5 to 4	1/2	56.8

MIC, minimal inhibitory concentration; NA, not applicable, no breakpoints defined
[1] As defined in https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html

Figure 1. Number of *C. auris* isolates collected per year

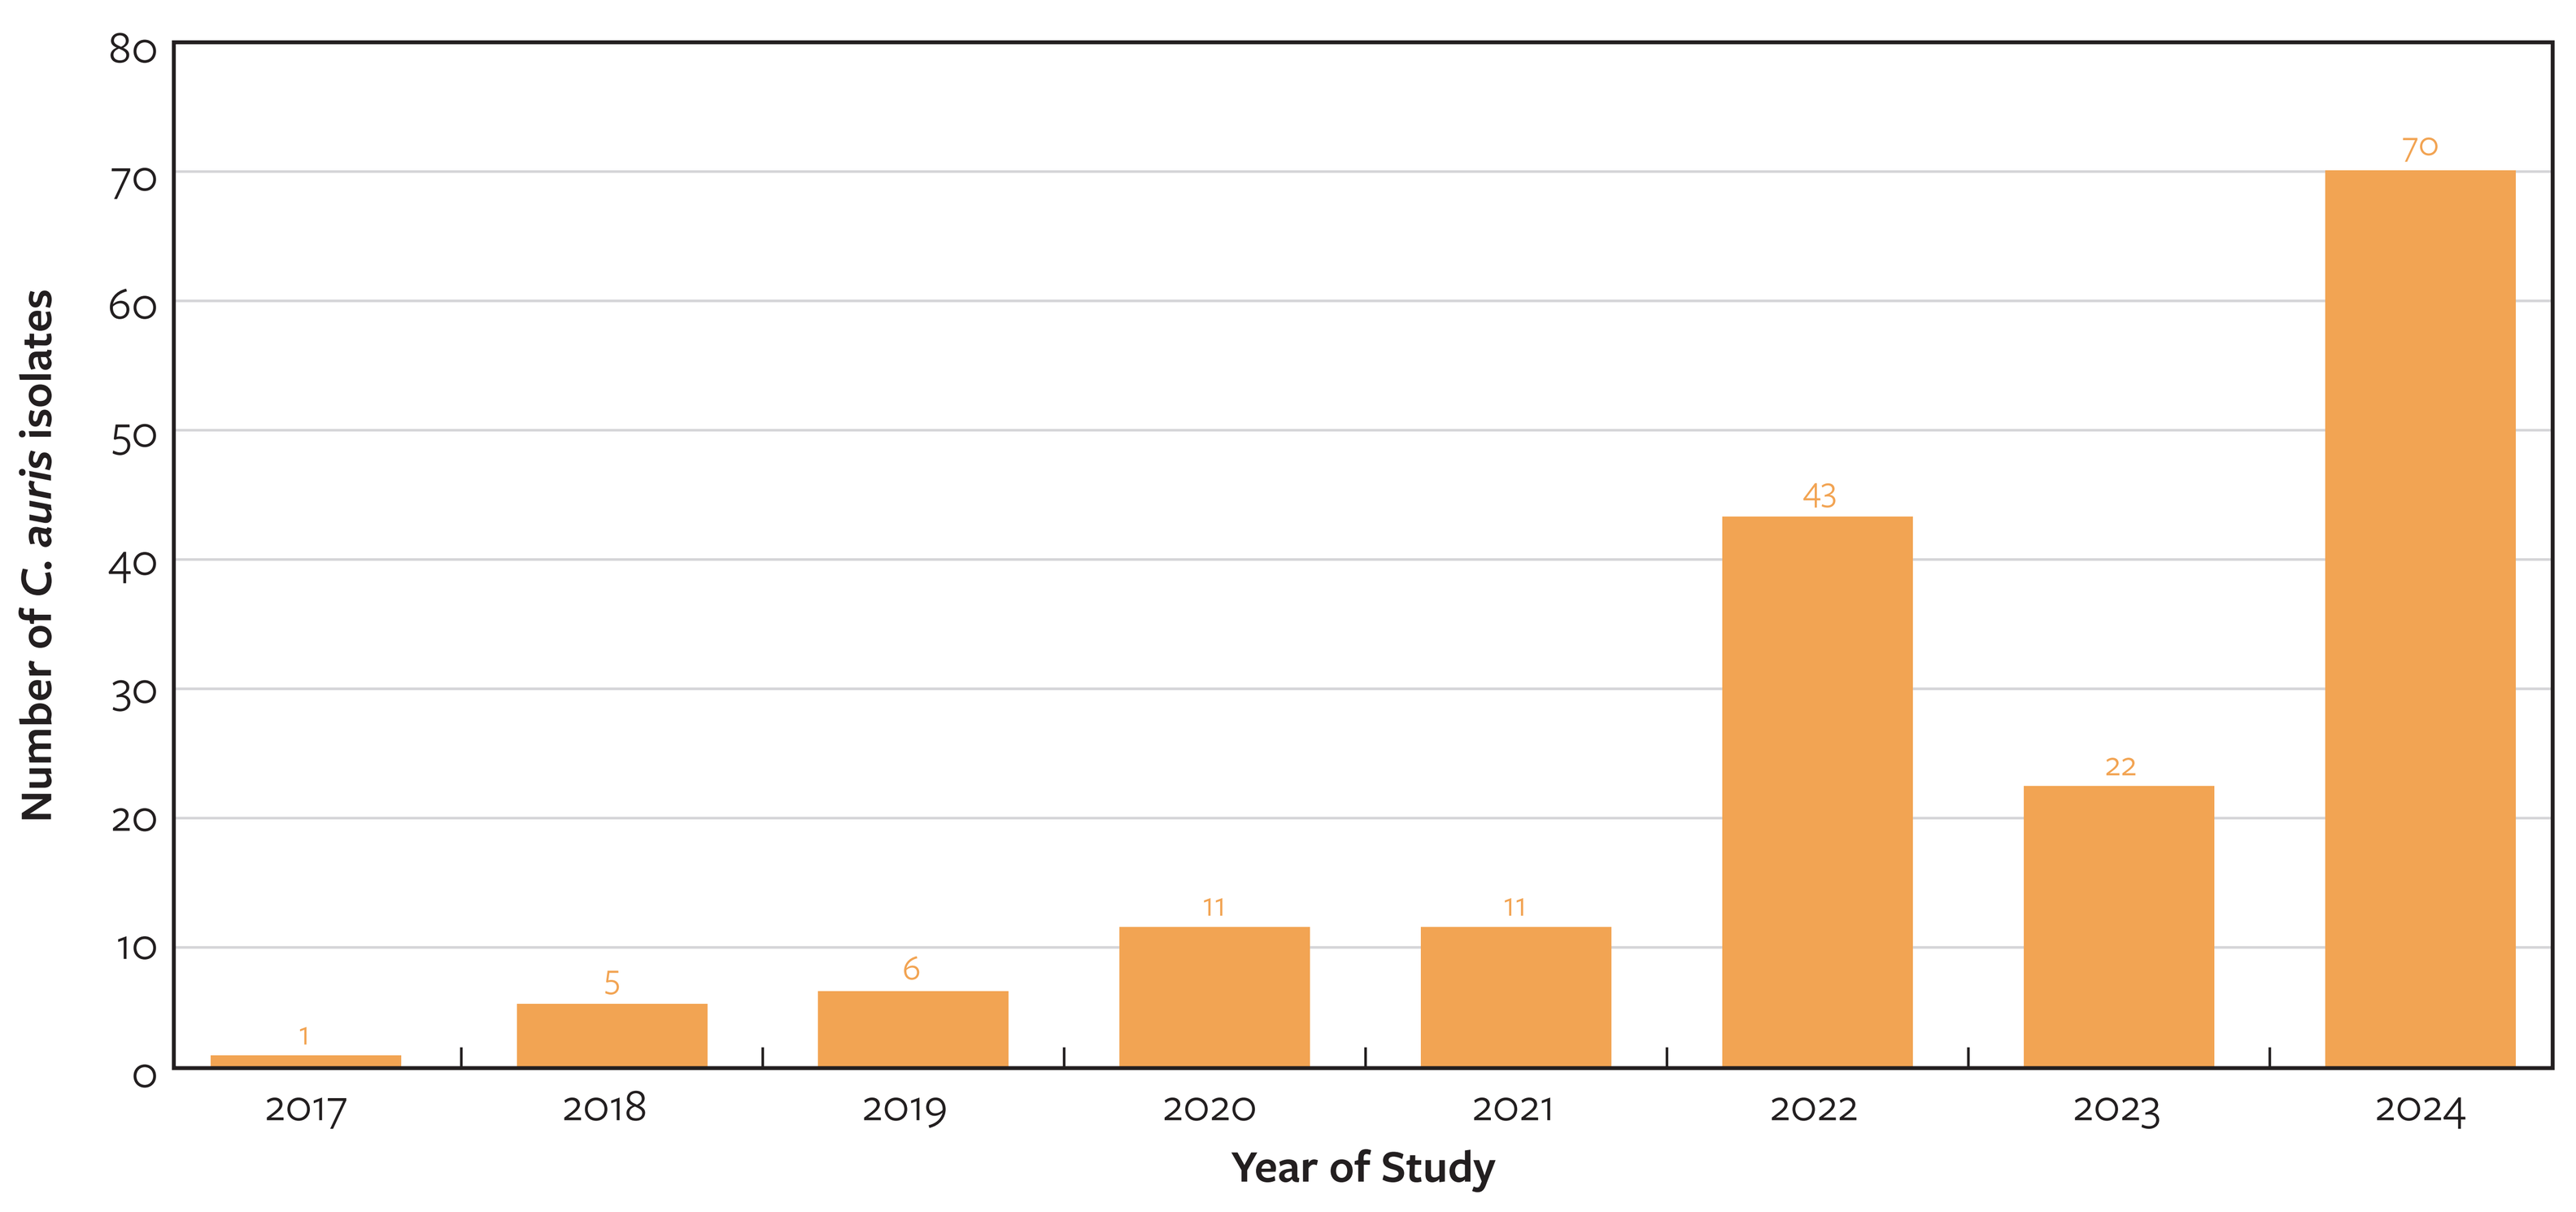
Number of C. auris isolates
Year of Study
0
10
20
30
40
50
60
70
80
2017
2018
2019
2020
2021
2022
2023
2024
1
5
6
11
11
43
22
70

Results

- The number of isolates increased from 1 in 2017 to 70 in 2024 (Figure 1)
- Isolates were from various infection sources (Figure 2)
- Manogepix $MIC_{50/90}$ values against *Candida auris* were 0.015/0.03 mg/L (Table 1)
 - The highest observed manogepix MIC was 0.25 mg/L
 - Manogepix $MIC_{50/90}$ values were within 2 doubling dilutions across the years of study
 - In 2020, $MIC_{50/90}$ values were 0.004/0.015 mg/L; in 2024 $MIC_{50/90}$ values were 0.015/0.03 mg/L
- There were 156 isolates resistant to fluconazole and 73 isolates resistant to amphotericin B
 - Low manogepix MICs were maintained against all resistant subgroups (Table 2)
- Echinocandin resistance was first observed in 2022
 - 13 isolates were resistant to ≥1 echinocandin
 - From Panama, USA, Israel, Turkey, Greece
 - 1 isolate (2.3%) in 2022, 5 isolates (22.7%) in 2023, and 7 isolates (10.0%) in 2024
 - 9 isolates had FKS1 HS alterations (Table 3)
 - The 4 isolates without FKS1 alterations were only resistant to caspofungin

Conclusions

- Manogepix has potent *in vitro* activity among 169 *C. auris* isolates collected since 2017
- More echinocandin-resistant isolates have been observed since 2022
 - No trend towards increasing manogepix MICs were observed over this time period
- Manogepix also maintained low MICs against isolates with FKS1 alterations
- Manogepix is a promising novel antifungal agent for the treatment of infections due to yeast including organisms of critical concern like *C. auris*

Figure 2. Infection source of collected *C. auris* isolates

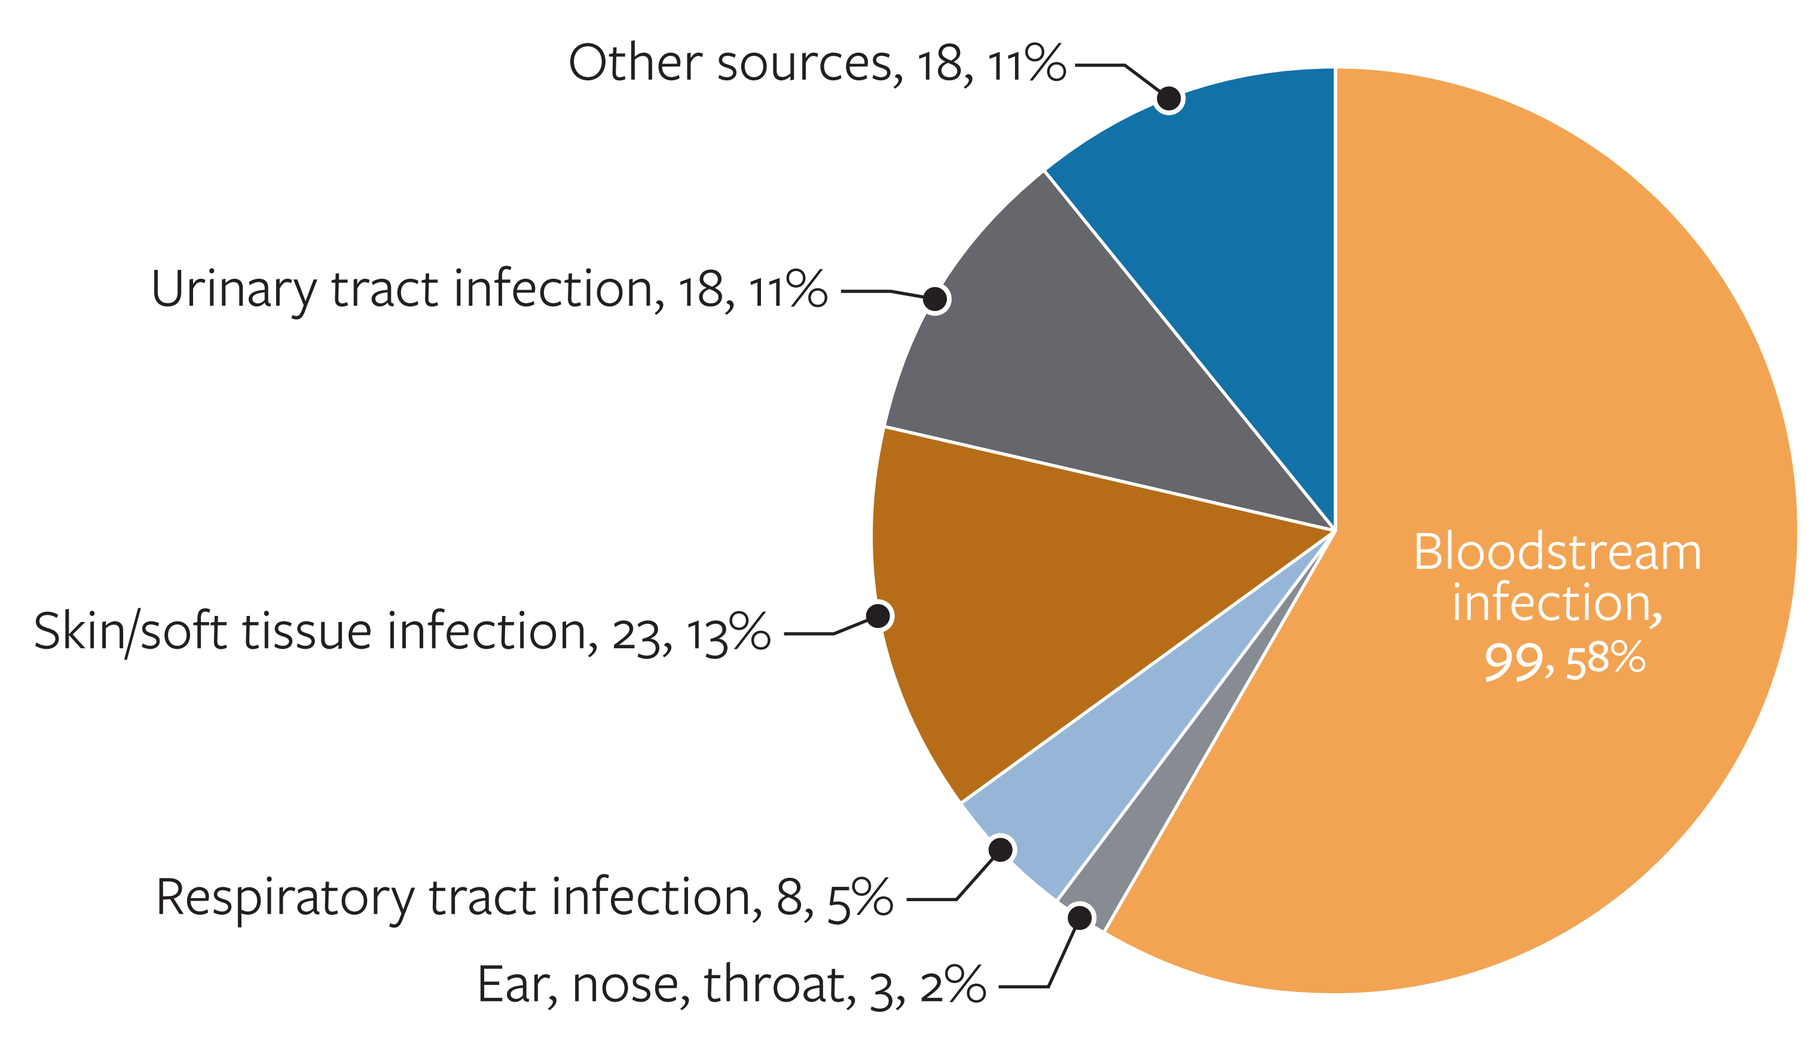
Other sources, 18, 11%
Urinary tract infection, 18, 11%
Skin/soft tissue infection, 23, 13%
Respiratory tract infection, 8, 5%
Ear, nose, throat, 3, 2%
Bloodstream infection, 99, 58%

Table 2. MICs for manogepix and comparators against fluconazole-resistant and amphotericin B-resistant *C. auris*

Susceptibility pattern (n)	Manogepix		Anidulafungin		Amphotericin B		Fluconazole	
	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)
FLC-resistant (156)	0.002 – 0.25	0.015/0.03	0.06 – >4	0.25/0.5	0.5 – 4	1/2	32 – >128	>128/>128
AmB-resistant (73)	0.004 – 0.25	0.015/0.06	0.06 – 2	0.25/0.5	2 – 4	2/2	16 – >128	>128/>128
FLC and AmB-resistant (72)	0.004 – 0.25	0.015/0.06	0.06 – 2	0.25/0.5	2 – 4	2/2	32 – >128	>128/>128
Pan-resistant (8)[2]	0.008 – 0.25	0.03/[1]	0.5 – 2	1/[1]	2	2/[1]	32 – >128	>128/[1]

[1] Unable to calculate MIC_{90} if < 10 isolates
[2] Pan-resistant defined as resistant to amphotericin B, fluconazole, and ≥1 echinocandin

Table 3. Results of FKS sequencing for echinocandin-resistant *C. auris* isolates

Year of collection	Collection site	Manogepix MIC (mg/L)	Anidulafungin MIC (mg/L)	Caspofungin MIC (mg/L)	Micafungin MIC (mg/L)	FKS1 HS1	FKS1 HS2
2022	Panama	0.004	4	>4	4	WT	R1354G
2023	LA, USA	0.008	>4	>4	>4	S639F	WT
2023	LA, USA	0.008	>4	>4	>4	S639F	WT
2023	Turkey	0.008	1	>4	1	WT	R1354H
2023	VA, USA	0.015	1	>4	0.5	D642Y	WT
2023	VA, USA	0.015	2	>4	0.5	D642Y	WT
2024	LA, USA	0.03	>4	>4	>4	S639F	WT
2024	Israel	0.03	1	>4	0.5	WT	WT
2024	VA, USA	0.25	2	>4	1	D642Y	WT
2024	TX, USA	0.015	0.5	>4	1	WT	WT
2024	Greece	0.03	2	>4	>4	F635Y	WT
2024	Greece	0.06	0.5	>4	0.25	WT	WT
2024	Greece	0.12	0.5	>4	0.25	WT	WT

MIC, minimal inhibitory concentration; HS, hotspot; LA, Louisiana; USA, United States of America; VA, Virginia; TX, Texas; WT, wildtype

Funding

This study was supported by Basilea and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration for Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under OT number 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, JH Kimbrough, S Edeker, A Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

References

Pappas, P.G., Kauffman, C.A., Andes, D.R., Clancy, C.J., Marr, K.A., Ostrosky-Zeichner, L., et al., *Clinical Practice Guideline for the Management of Candidiasis: 2016 Update by the Infectious Diseases Society of America.* Clin Infect Dis, 2016. 62(4): e1-50.

Centers for Disease Control. *Antifungal susceptibility testing for C. auris*. February 3, 2026. https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html

CLSI, *M27 M44S Ed3. Performance standards for antifungal susceptibility testing of yeasts*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M27 Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Contact

element

Scan Me

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ESCMID2026_24-BAS-01_A5_MANOGEPIX_C_AURIS.pdf

Charges may apply. No personal information is stored.

ESCMID Global 2026, April 17–21, 2026 Munich, Germany