ESCMID 2026 | Poster #P3320

Activity of Manogepix and Comparator Antifungals Against Mould Isolates Collected from Invasive Infections in Intensive Care Units (ICU) and Non-ICU Settings (2017–2024)

MD Huband, ML Winkler, PR Rhomberg, S Edeker, AL Klauer, M Castanheira
Element Iowa City (JMI Laboratories), North Liberty, Iowa USA

Introduction

- Manogepix is a novel antifungal agent targeting the fungal Gwt1 enzyme with potent *in vitro* activity against many species of yeast and mould.
- The manogepix prodrug, fosmanogepix, is being evaluated in a Phase 3 clinical trial (NCT06925321) for the treatment of adults with invasive mould infections.
- Few studies describe the distribution and susceptibility for invasive mould isolates from the intensive care unit (ICU) compared to non-ICU settings.
- In this study, we evaluated the species distribution and susceptibility of manogepix against 2,659 mould isolates according to ICU versus non-ICU in a global collection of contemporary isolates (2017–2024).

Materials and Methods

- 2,659 non-duplicate mould isolates were collected during 2017–2024 from patients in 67 medical centers in North America (27 sites; 38.2%), Europe (27 sites; 40.8%), Asia-Pacific (11 sites; 19.3%), and Latin America (2 sites; 1.7%).
- Isolates were collected from the following indications: pneumonia in hospitalized patients (PIHP; 63.7%), skin and skin structure infections (SSSI; 9.9%), bloodstream infections (BSI; 1.5%), urinary tract infections (UTI; 0.05%), intra-abdominal infections (IAI; 0.15%), and other infection types (24.7%).
- All isolates were tested using reference broth microdilution methodology following Clinical and Laboratory Standards Institute (CLSI) M38 (2017) and M38M51S (2022) criteria.
- Fungal isolate identifications were confirmed using matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Billerica, MA) and/or sequencing based methods.
- Manogepix MEC endpoints were determined for mould isolates using the same reading criteria as echinocandins.

Results

- Based on MEC_{90} values, manogepix was the most potent antifungal tested against 2,659 mould isolates from ICU and non-ICU patients with MEC_{90} values of 0.06 mg/L and 0.03 mg/L, respectively (Figures 1–2).
- The top 5 organism groups from ICU patients were *Aspergillus fumigatus* (63.3%, $MEC_{50/90}$, 0.016/0.03 mg/L), *A. flavus* species complex (9.3%, $MEC_{50/90}$, 0.016/0.03 mg/L), *A. niger* species complex (8.2%, $MEC_{50/90}$, ≤0.008/0.03 mg/L), *A. terreus* species complex (3.5%, $MEC_{50/90}$, 0.008/0.03 mg/L), and *Rhizopus* spp. (3.5%, $MEC_{50/90}$, 4/8 mg/L) (Table 1).
- The top 5 organism groups from non-ICU patients were *A. fumigatus* (50.9%, $MEC_{50/90}$, 0.016/0.03 mg/L), *A. niger* species complex (11.4%, $MEC_{50/90}$, ≤0.008/0.016 mg/L), *A. flavus* species complex (8.3%, $MEC_{50/90}$, 0.016/0.03 mg/L), *Fusarium* spp. (4.6%, $MEC_{50/90}$, 0.016/0.03 mg/L), and *A. terreus* species complex (3.3%, $MEC_{50/90}$, 0.016/0.03 mg/L) (Table 1).
- Notable activity was observed for manogepix against *Aspergillus* spp. isolates from ICU and non-ICU patients including *A. fumigatus*, *A. nidulans* species complex, *A. flavus* species complex, *A. niger* species complex, *A. terreus* species complex, *A. tubingensis*, *A. ustus* species complex, *A. versicolor* species complex, and other *Aspergillus* spp. ($MEC_{50/90}$ values, ≤0.008–0.016/0.016–0.06 mg/L) (Table 1).
- Manogepix was active against other mould isolates from ICU and non-ICU patients including *Fusarium* spp. ($MEC_{50/90}$, 0.016/0.03–0.12 mg/L), *Lomentospora prolificans* ($MEC_{50/90}$, 0.016/0.06 mg/L), *Paecilomyces variotii* ($MEC_{50/90}$, ≤0.008/≤0.008 mg/L), *Purpureocillium lilacinum* ($MEC_{50/90}$, ≤0.008/0.016 mg/L), *Rasamsonia argillacea* species complex ($MEC_{50/90}$, ≤0.008/0.016 mg/L), and *Scedosporium* spp. ($MEC_{50/90}$, 0.03/0.06 mg/L) (Table 1) including isolates demonstrating elevated MIC values to other drug classes.
- Pneumonia in hospitalized patients, skin and skin structure infections, and other infection types accounted for 97% of ICU and 99% of non-ICU infections.

Conclusions

- Mould species distributions were slightly different between ICU and non-ICU settings.
- Manogepix showed potent *in vitro* activity against mould isolates from ICU and non-ICU patients caused by *Aspergillus* spp., *Exophiala* spp., *Fusarium* spp., *L. prolificans*, *P. variotii*, *P. lilacinum*, *R. argillacea* species complex, and *Scedosporium* spp. including isolates demonstrating elevated MIC values to other drug classes.
- The data supports the continued Phase 3 development of fosmanogepix in the treatment of invasive mould infections.

Acknowledgments

This project has been funded in whole or in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration for Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA) under OT number 75A50124C00033; and Basilea Pharmaceutica Ltd. The contract and federal funding are not an endorsement of the study results, product, or company.

References

CLSI. *Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 3rd ed. CLSI standard M38. Wayne, PA: Clinical and Laboratory Standards Institute: 2017.

CLSI. *Performance standards for antifungal susceptibility testing of filamentous fungi*. 3rd ed. CLSI supplement M38M51S. Clinical and Laboratory Standards Institute: 2022.

Contact

Marisa L. Winkler, MD, Associate Director
Element Iowa City (JMI Laboratories)
335 Beaver Kreek Centre
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/ESCMID2026_24-BAS-01_A6_MOLD_ICU.pdf

Charges may apply.
No personal information is stored.

Table 1. Activity of manogepix against 2,659 mould isolates from ICU and non-ICU patients (2017–2024)

	Manogepix $MEC_{50/90}$ (mg/L) (no. tested)			MIC or $MEC_{50/90}$ (mg/L) (% susceptible)		
Organism	All	ICU	Non-ICU	AMB	MFG	VCZ
Aspergillus fumigatus	0.016/0.03 (1,425)	**0.016/0.03 (415)**	**0.016/0.03 (793)**	1/2 (—)	≤0.008/0.016 (—)	0.5/0.5 (90.5%) [a]
A. nidulans species complex	0.016/0.03 (52)	**0.016/— (9)**	**0.016/0.03 (29)**	2/2 (—)	≤0.008/0.03 (—)	0.12/0.25 (—)
A. flavus species complex	0.016/0.06 (236)	**0.016/0.03 (61)**	**0.016/0.03 (129)**	2/2 (—)	≤0.008/0.016 (—)	0.5/1 (—)
A. niger species complex	≤0.008/0.016 (283)	**≤0.008/0.03 (54)**	**≤0.008/0.016 (177)**	0.25/1 (—)	≤0.008/0.016 (—)	1/2 (—)
A. terreus species complex	0.016/0.03 (94)	**0.008/0.03 (23)**	**0.016/0.03 (52)**	2/4 (—)	≤0.008/0.016 (—)	0.5/0.5 (—)
A. tubingensis	≤0.008/0.03 (23)	**0.002/— (2)**	**≤0.008/0.03 (10)**	0.12/0.5 (—)	≤0.008/0.03 (—)	2/4 (—)
A. ustus species complex	≤0.008/0.016 (15)	**≤0.008/— (3)**	**≤0.008/0.016 (10)**	1/4 (—)	0.016/0.03 (—)	4/8 (—)
A. versicolor species complex	0.016/0.03 (11)	**—/— (0)**	**0.016/— (9)**	2/2 (—)	0.016/0.03 (—)	0.5/1 (—)
Other *Aspergillus* spp. [b]	0.016/0.03 (45)	**0.016/— (6)**	**0.016/0.03 (30)**	2/4 (—)	0.008/0.03 (—)	0.5/4 (—)
Exophiala spp. [c]	≤0.008/≤0.008 (16)	**—/— (1)**	**≤0.008/≤0.008 (15)**	0.5/2 (—)	>4/>4 (—)	0.12/0.25 (—)
Fusarium fujikuroi species complex	0.016/0.12 (24)	**0.016/— (3)**	**0.016/0.03 (17)**	2/>2 (—)	>4/>4 (—)	4/>8 (—)
F. oxysporum species complex	0.016/0.06 (22)	**0.06/— (4)**	**0.016/0.06 (18)**	2/4 (—)	>4/>4 (—)	4/8 (—)
F. solani species complex	0.016/0.03 (46)	**0.016/0.03 (10)**	**0.016/0.016 (26)**	2/2 (—)	>4/>4 (—)	8/>8 (—)
Other *Fusarium* spp. [d]	0.016/2 (15)	**0.004/— (2)**	**0.016/2 (11)**	1/2 (—)	>4/>4 (—)	4/>8 (—)
Lomentospora prolificans	0.016/0.06 (25)	**0.016/— (2)**	**0.016/0.06 (20)**	>2/>2 (—)	>4/>4 (—)	>8/>8 (—)
Mucor spp. [e]	1/>4 (26)	**1/— (6)**	**2/>4 (12)**	0.25/0.5 (—)	>4/>4 (—)	>8/>8 (—)
Paecilomyces variotii	≤0.008/≤0.008 (14)	**0.004/— (3)**	**≤0.008/— (9)**	0.12/0.5 (—)	≤0.008/≤0.008 (—)	4/8 (—)
Purpureocillium lilacinum	≤0.008/0.016 (21)	**0.008/— (5)**	**≤0.008/0.016 (14)**	>2/>2 (—)	0.03/0.12 (—)	0.25/0.5 (—)
Rasamsonia argillacea species complex	≤0.008/0.016 (12)	**—/— (1)**	**≤0.008/0.016 (11)**	1/2 (—)	≤0.008/≤0.008 (—)	>8/>8 (—)
Rhizopus spp. [f]	4/>4 (65)	**4/8 (23)**	**4/>4 (34)**	0.5/1 (—)	>4/>4 (—)	8/>8 (—)
Scedosporium spp. [g]	0.03/0.06 (94)	**0.016/— (5)**	**0.03/0.06 (75)**	>2/>2 (—)	0.5/>4 (—)	1/2 (—)

AMB = Amphotericin B; MFG = Micafungin; VCZ = Voriconazole.
[a] CLSI breakpoints applied.
[b] Organisms include: *Aspergillus alabamensis* (1), *A. brasiliensis* (2), *A. caelatus* (1), *A. clavatus* (2), *A. fumisynnematus* (1), *A. hiratsukae* (1), *A. hortai* (1), *A. lentulus* (9), *A. melleus* (1), *A. nomius* (1), *A. ochraceus* species complex (1), *A. parasiticus* (3), *A. sclerotiorum* (5), *A. sydowii* (5), *A. tamarii* (3), *A. thermomutatus* (2), *A. udagawae* (2), *A. unguis* (3), and *A. welwitschiae* (1).
[c] Organisms include: *Exophiala attenuata* (2), *E. dermatitidis* (13), and unspeciated *Exophiala* (1).
[d] Organisms include: *Fusarium annulatum* (2), *F. dimerum* (1), *F. dimerum* species complex (1), *F. falciforme* (1), *F. incarnatum-equiseti* species complex (6), *F. musae* (1), *F. petroliphilum* (1), and unspeciated *Fusarium* (2).
[e] Organisms include: *Mucor circinelloides* (15), *M. circinelloides/M. ramosissimus* (2), *M. indicus* (1), and unspeciated *Mucor* (8).
[f] Organisms include: *Rhizopus microsporus* (32), *R. oryzae* species complex (27), and unspeciated *Rhizopus* (6).
[g] Organisms include: *Scedosporium apiospermum* (10), *S. apiospermum / Scedosporium boydii* (53), *S. aurantiacum* (13), *S. boydii* (12), *S. dehoogii* (3), *S. minutisporum* (2), and unspeciated *Scedosporium* (1).

Figure 1. Activity of manogepix and comparators tested against 656 mould isolates from ICU patients

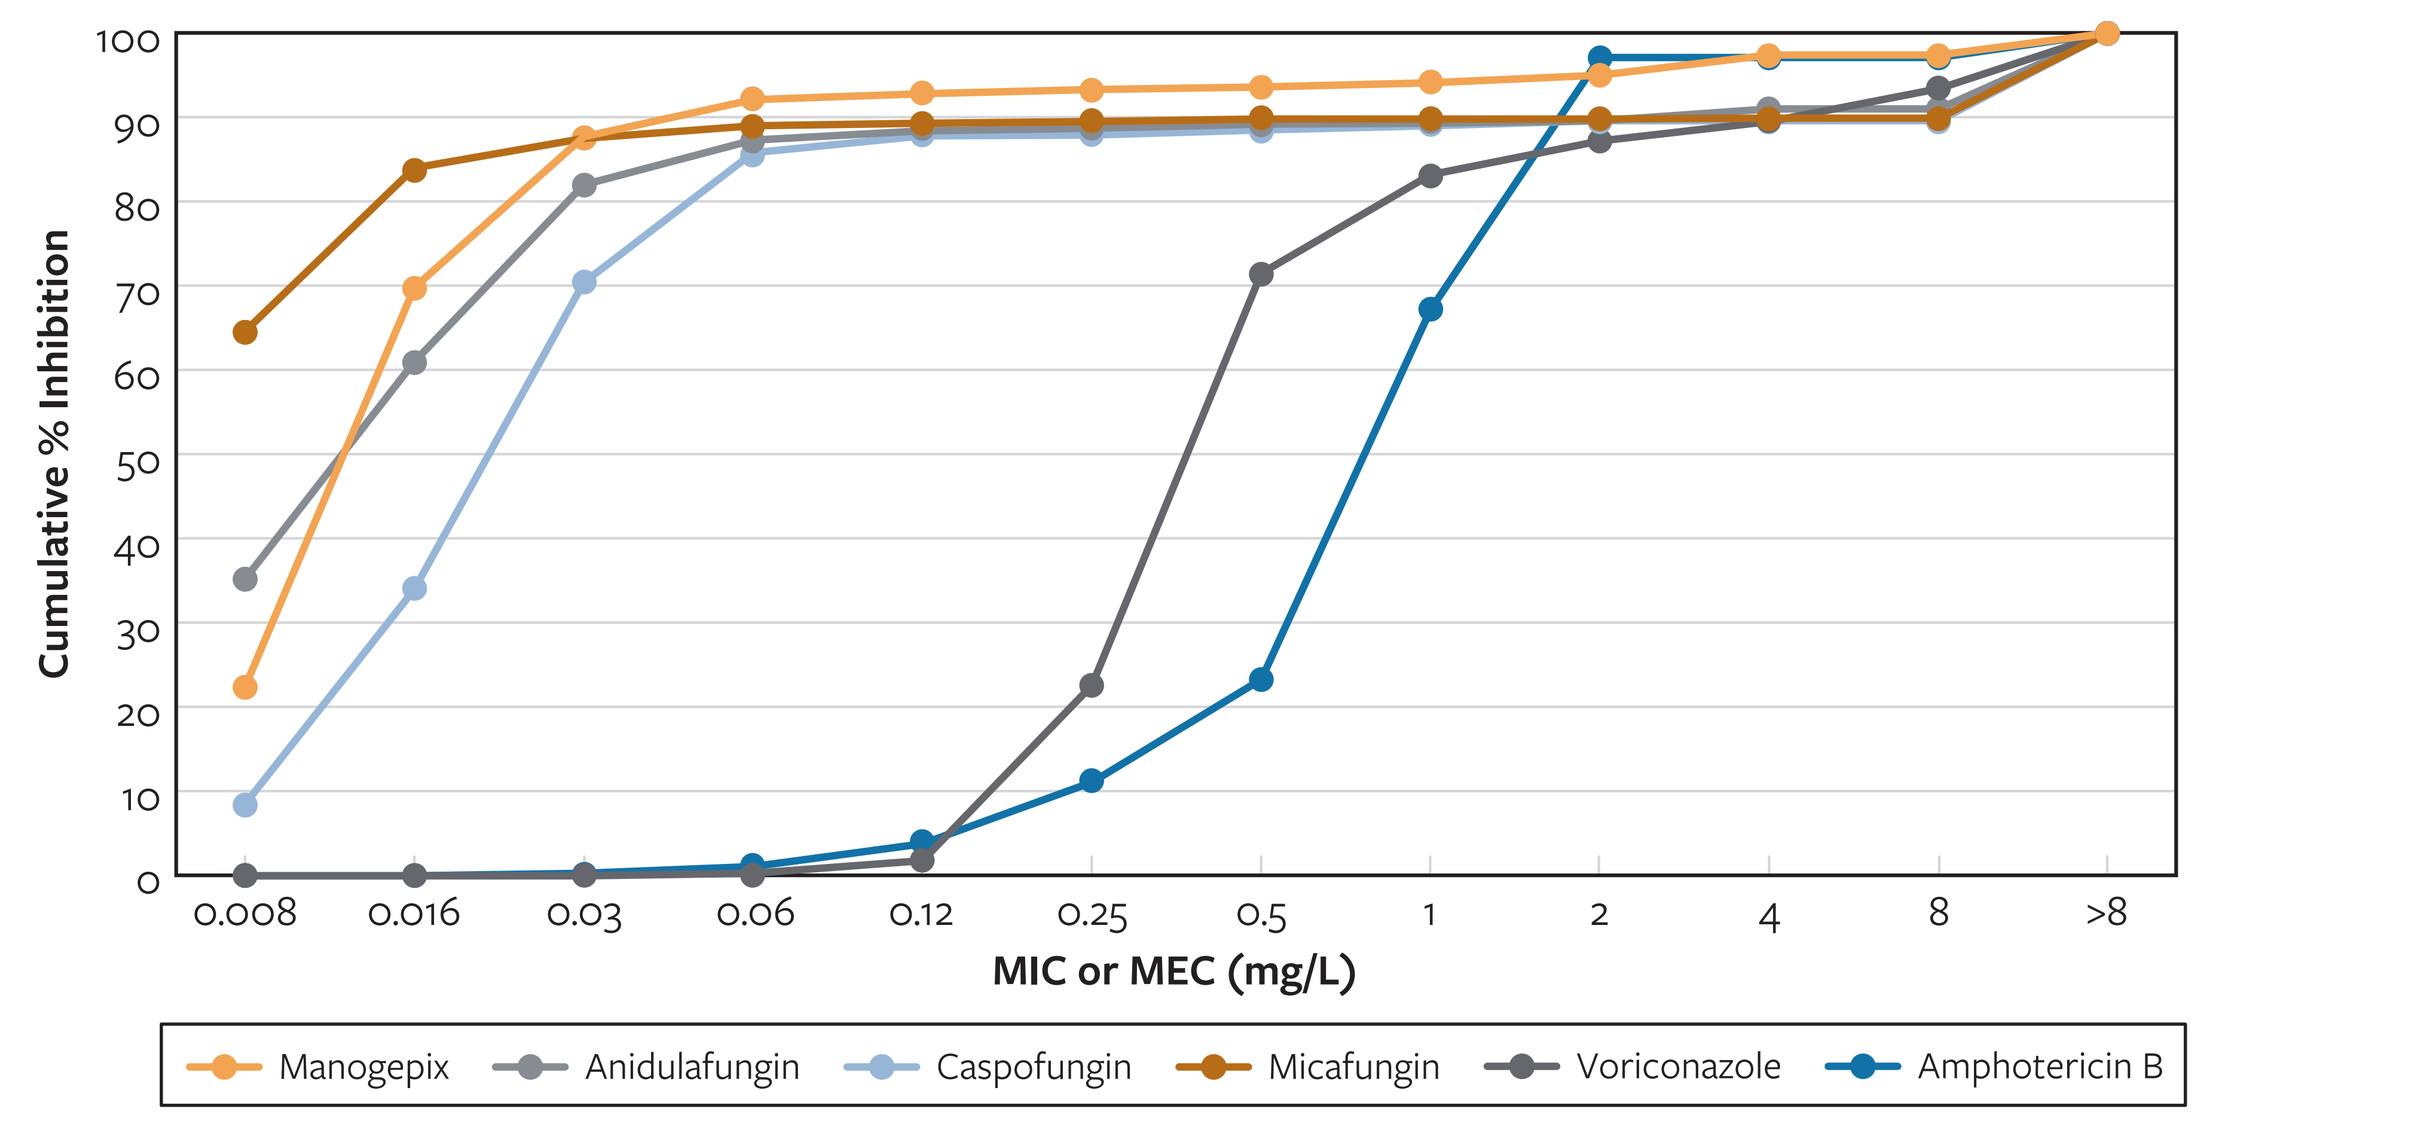
Cumulative % Inhibition
0
10
20
30
40
50
60
70
80
90
100
0.008
0.016
0.03
0.06
0.12
0.25
0.5
1
2
4
8
>8
MIC or MEC (mg/L)
Manogepix
Anidulafungin
Caspofungin
Micafungin
Voriconazole
Amphotericin B

Organisms include *Aspergillus* spp. (573), *Exophiala dermatitidis* (1), *Fusarium* spp. (19), *Gibberella fujikuroi* species complex (3), *Lichtheimia* spp. (8), *Lomentospora prolificans* (2), *Mucor* spp. (6), *Paecilomyces variotii* (3), *Purpureocillium lilacinum* (5), *Rasamsonia argillacea* species complex (1), *Rhizomucor pusillus* (5), *Rhizopus* spp. (23), *Sarocladium kiliense* (1), *Scedosporium* spp. (5), unspeciated *Cunninghamella* (2), unspeciated *Syncephalastrum* (1), and *Verruconis gallopava* (1).

Figure 2. Activity of manogepix and comparators tested against 1,557 mould isolates from non-ICU patients

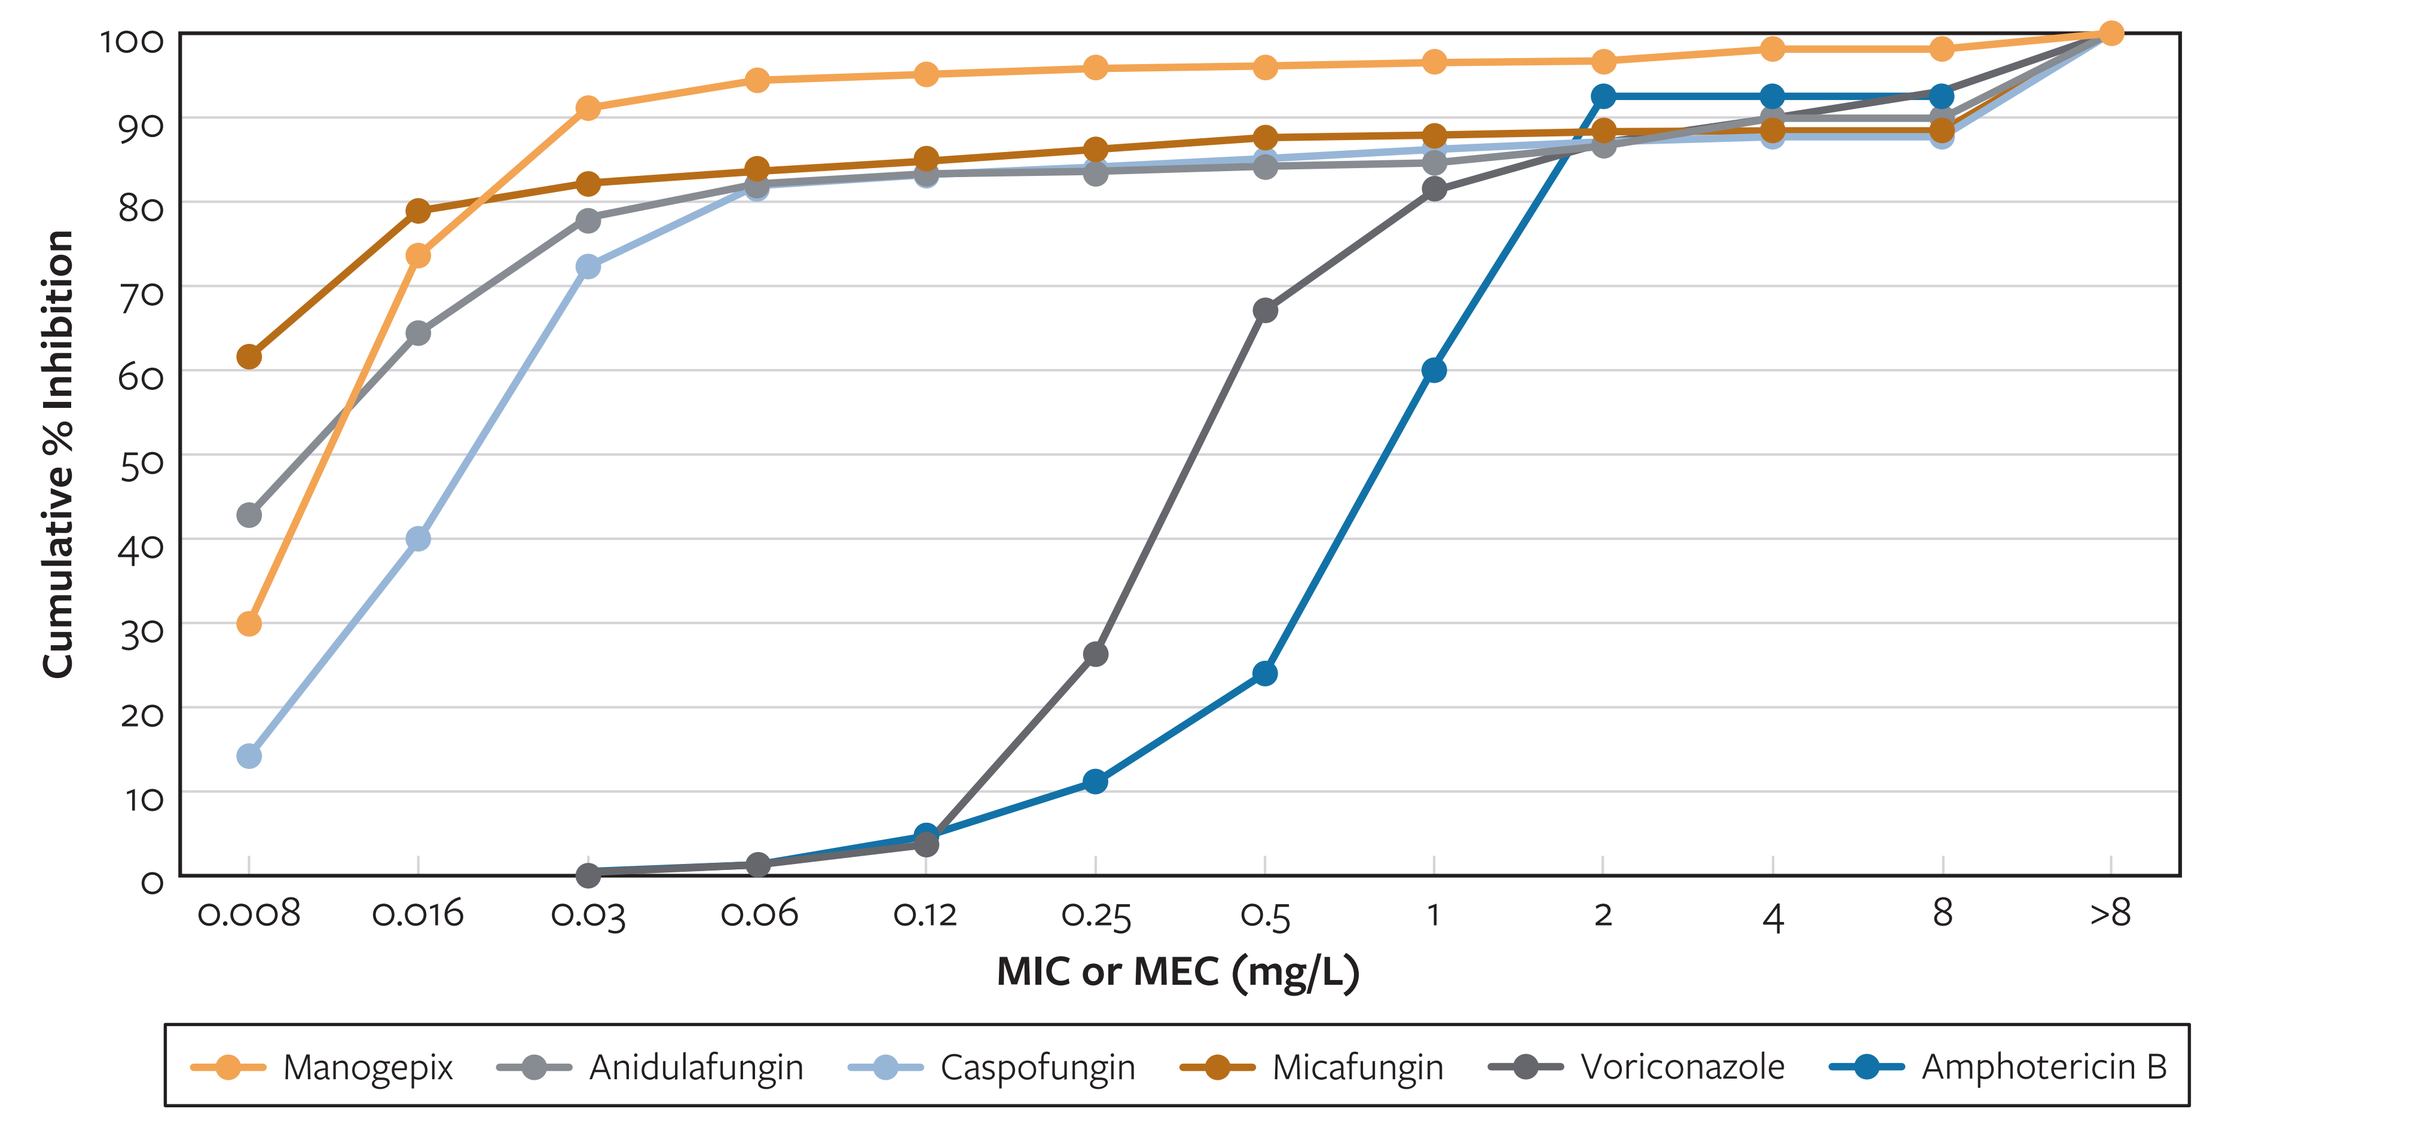
Cumulative % Inhibition
0
10
20
30
40
50
60
70
80
90
100
0.008
0.016
0.03
0.06
0.12
0.25
0.5
1
2
4
8
>8
MIC or MEC (mg/L)
Manogepix
Anidulafungin
Caspofungin
Micafungin
Voriconazole
Amphotericin B

Organisms include *Alternaria* spp. (2), *Aspergillus* spp. (1,240), *Aureobasidium pullulans* (1), *Coprinopsis cinerea* (1), *Exophiala* spp. (14), *Fusarium* spp. (72), *Gibberella fujikuroi* species complex (17), *Lichtheimia* spp. (8), *Lomentospora prolificans* (20), *Medicopsis romeroi* (1), *Microascus cirrosus* (1), *Monascus ruber* (1), *Mucor* spp. (12), *Paecilomyces* spp. (16), *Penicillium* spp. (6), *Pleurostomophora richardsiae* (1), *Purpureocillium lilacinum* (14), *Rasamsonia* spp. (11), *Rhizomucor pusillus* (1), *Rhizopus* spp. (34), *Scedosporium* spp. (75), *Scopulariopsis* spp. (3), *Trichoderma* spp. (4), unspeciated *Acremonium* (1), unspeciated *Chaetomium* (1), unspeciated *Coprinellus* (1), unspeciated *Cunninghamella* (2), unspeciated *Curvularia* (7), unspeciated *Phialemoniopsis* (1), unspeciated *Sarocladium* (1), unspeciated *Syncephalastrum* (2), and *Verruconis gallopava* (1).

ESCMID Global 2026, April 17–21, 2026 Munich, Germany