C1-109a

Dissemination and Genetic Context Analysis of *bla*$_{VIM-6}$ Among *Pseudomonas aeruginosa* in Asia-Pacific Nations

M CASTANHEIRA, JM BELL, JD TURNIDGE, RE MENDES, HS SADER, RN JONES

JMI Laboratories, North Liberty, IA; Women's and Children's Hospital, Adelaide, Australia

ICAAC 2007

JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370
fax 319.665.3371
ronald-jones@jmilabs.com

AMENDED ABSTRACT

Background: *bla*$_{VIM-6}$ was reported from two *P. putida* clinical isolates recovered in Singapore (SG; 2004), but the context of this gene has not been described. We evaluated the dissemination and the genetic environment of *bla*$_{VIM-6}$, now in *P. aeruginosa* (PSA) across the Asia-Pacific (APAC) region.

Methods: Detection of MßL genes was performed with degenerated primers by real-time PCR. Positive samples were tested with primers targeting class 1 integron conserved structures achoring in the MßL gene. Amplicons generated were sequenced on both strands. Integron PCR products were digested with DraI and XbaI and the band patterns used to determine the similarity among integrons carrying this MßL. Each integron type was fully sequenced. VIM-6-producing isolates were also ribotyped to evaluate possible clonality.

Results: Among 196 carbapenem-resistant PSA collected in 2006 from 10 nations, 66 (33.6%) were MßL-producers and *bla*$_{VIM-6}$ was detected in 16 strains (8.1%). VIM-6 producing PSA were isolated from India (12 strains), Indonesia (2) and 1 strain each from Korea and the Philippines. 13 ribotype patterns were observed and 3 clones of 2 strains each were detected in 3 hospitals in India. Isolates from various countries showed distinct ribotypes. Class 1 integrons of 3.5 (India only) and 5 Kb were detected among *bla*$_{VIM-6}$-carrying strains. 2 of 5 Indian hospitals had both integrons. Integrons of the same size had the same RFLP patterns. *bla*$_{VIM-6}$ was located in the 1st position followed by *bla*$_{OXA-10}$ and *aacA4*. 5 Kb integrons showed the same genetic arrangement as 3.5 Kb integrons, but with the addition of an unknown gene and an *aadA1* preceding *qacE∆1/sul1*.

Conclusions: *bla*$_{VIM-6}$ was observed in 5 APAC nations (4 new countries + SG). The genetic diversity of these isolates and the presence of related, yet distinct, integrons indicates the mobility of genetic structures carrying *bla*$_{VIM-6}$ and/or the presence of a common bacterial reservoir for this element in the region.

INTRODUCTION

Production of metallo-ß-lactamases (MßLs) is not the most prevalent resistance mechanism against carbapenems found in *Pseudomonas* species. However, MßLs constitute one of the most important clinically and epidemiologically, since these enzymes can hydrolyze the vast majority of available ß-lactam agents. MßLs are not inhibited by the ß-lactamase inhibitors currently marketed or in development. Additionally, the acquired genes encoding these enzymes are carried in mobile structures, which facilitate its dissemination and usually harbor other resistance elements.

Among the five types of reported MßLs (IMP, VIM, SPM-1, GIM-1 and SIM-1), IMP- and VIM-variants are the most prevalent and they have been described in numerous countries. Among the VIM-types, VIM-2 appears to be the most dominant genotype and has been described in 23 countries. A new VIM-type enzyme was identified in two *Pseudomonas putida* isolates recovered in a tertiary hospital located in Singapore in 2000. This enzyme, named VIM-6, shows only two amino acid differences compared to VIM-2, which is probably its genetic ancestor. The gene encoding VIM-6 was carried in a class 1 integron, but the structure of this integron has not been described.

In this study we report 16 *bla*$_{VIM-6}$-carrying *P. aeruginosa* isolated from 4 countries from the Asia-Pacific (APAC) region and their genetic context.

MATERIALS AND METHODS

Bacterial isolates. During 2006, 42 medical centers located in 10 countries in the APAC region were recruited to participate to the SENTRY Antimicrobial Surveillance Program. Isolates were consecutively collected from bloodstream, respiratory tract, skin and soft tissue, and urinary tract infections according to defined protocols. Only clinically significant isolates were included in the study; one per patient episode. Species identification was confirmed by standard biochemical tests and use of Vitek Systems (bioMérieux; Hazelwood, Missouri, USA), when necessary.

Susceptibility testing. All isolates were susceptibility tested against more than 25 antimicrobials by the broth microdilution procedure described by the Clinical and Laboratory Standards Institute (CLSI; 2006) using validated panels manufactured by TREK Diagnostics (Cleveland, Ohio, USA). Interpretations of susceptibility testing results were by CLSI (2007) breakpoint criteria. *Escherichia coli* ATCC 25922 and *P. aeruginosa* ATCC 27853 were concurrently tested for quality assurance.

MßL detection. Isolates non-susceptible to imipenem and meropenem (MIC, ≥8 µg/ml) were tested with multiplex PCR using MßL generic primers in a Real-time platform, recently described. Amplicons obtained were sequenced on both strands. The nucleotide sequences and deduced amino acid sequences were analyzed using Lasergene software package (DNASTAR, Madison, WI) and compared with the sequences available through the internet using BLAST (http://www.ncbi.nlm.nih.gov/blast/).

Molecular Typing. MßL-producing isolates were ribotyped using the RiboPrinter Microbial Characterization System® (Qualicon, Wilmington, Delaware). Overnight cultures were treated with lysis buffer and placed into the automated system. In brief, this automated process includes bacterial cell lysis, cleavage of DNA using the restriction enzime PvuII, size separation using gel electrophoresis and modified Southern blotting. Results were analyzed by the RiboPrinter and isolates were considered to have the same ribotype if the similarity coefficient was ≥0.93.

Class 1 Integron Analysis. Additional primers designed in the 5' conserved sequence (CS) and 3'CS regions of class 1 integron were used in combination with the MßL primers to determine the size of the integron structure. All amplicons generated with *bla*$_{VIM}$ primers anchoring in the 3'CS were digested with DraI and XbaI in separated reactions. The band pattern obtained was visually compared in 2% agarose electrophoresis. One integron of each type was fully sequenced and analyzed as described above.

RESULTS

- Among 719 *P. aeruginosa* strains collected in the APAC region in 2006, 196 (27.3%) were non-susceptible (MIC, ≥8 µg/ml) to imipenem or meropenem, and among those, 66 (33.6%; 9.1% overall) isolates carried a MßL gene.
- *bla*$_{VIM-6}$ was detected in 16 (8.1%) isolates recovered in India (12), Indonesia (2), Korea (1) and the Philippines (1; Table 1).
- Ribotyping showed 13 different patterns among *bla*$_{VIM-6}$-carrying isolates (Figure 1). Four clones of two isolates each were observed in India (3) and in Indonesia. The other nine strains showed unique ribotypes.
- PCR and sequencing reactions of the class 1 integron CS primers combined with the primer annealing in *bla*$_{VIM-6}$ showed that this resistance gene was carried in the first position of the integron in all strains and that these structures were 3.5 and 5 Kb in size (Table 1).
- 3.5 Kb integrons carrying *bla*$_{VIM-6}$ were found in three distinct medical centers, but only in India.
- Integrons of the same size generated identical RFLP patterns with both restriction enzymes, indicating a similar content and arrangement.
- Sequencing showed that 3.5 Kb integrons carried downstream of *bla*$_{VIM-6}$, the gene for *bla*$_{OXA-10}$, *aacA4* encoding resistance to aminoglycosides, and the *qacE∆1/sul1* structure (Figure 2).
- The 5 Kb integron possessed the same gene content as the 3.5 Kb integrons with two additional gene cassettes. A 331 bp *orf* encoding a protein showing 78% similarity with QACE∆1 was found following *aacA4*. This unknown gene was located upstream of an *aadA1* that preceded the 3'CS of the class 1 integron (Figure 2).

Table 1. Characteristics of 16 *bla*$_{VIM-6}$-carrying *P. aeruginosa* isolated in 2006 in the APAC region.

Isolate	Country	Ribotype	Integron size
225-22-D	Korea	105-1034-2	ND[a]
228-23-D	Philippines	258-35-7	ND[a]
241-09-D	Indonesia	105-566-6	5 Kb
241-73.2-D	Indonesia	105-566-6	5 Kb
244-07-D	India	105-566-2	3.5 Kb
244-10-D	India	258-347-6	5 Kb
244-18-C	India	258-347-8	5 Kb
244-24-D	India	258-352-2	5 Kb
244-29-D	India	258-352-2	5 Kb
247-03-D	India	258-348-6	5 Kb
247-06-D	India	258-348-6	5 Kb
248-11-A	India	252-45-6	3.5 Kb
248-26-C	India	252-45-6	3.5 Kb
248-77-C	India	127-114-4	5 Kb
252-05-C	India	258-147-6	3.5 Kb
252-09-C	India	222-134-4	3.5 Kb

a. Not determined.

Figure 1. Ribotyping of 16 *bla*$_{VIM-6}$-carrying *P. aeruginosa* isolated in 2006 in the APAC region. Three clones with two isolates each were detected in India and one clone was observed in Indonesia (indicated with different colors).

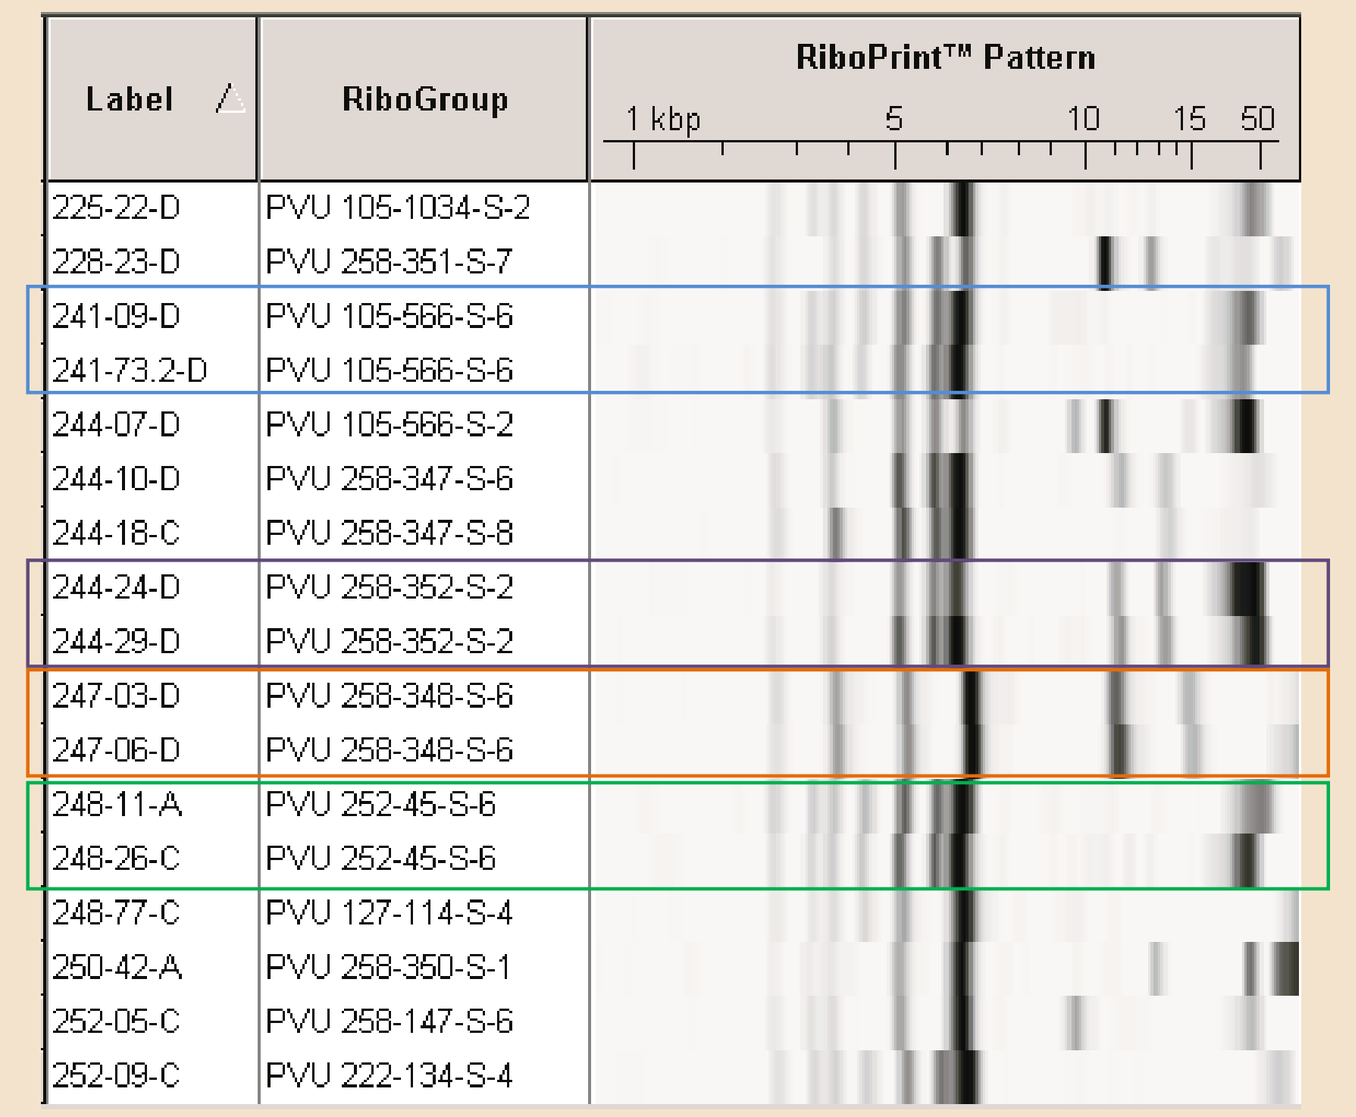

Label
RiboGroup
RiboPrint™ Pattern
1 kbp
5
10
15
50
225-22-D PVU 105-1034-S-2
228-23-D PVU 258-351-S-7
241-09-D PVU 105-566-S-6
241-73.2-D PVU 105-566-S-6
244-07-D PVU 105-566-S-2
244-10-D PVU 258-347-S-6
244-18-C PVU 258-347-S-8
244-24-D PVU 258-352-S-2
244-29-D PVU 258-352-S-2
247-03-D PVU 258-348-S-6
247-06-D PVU 258-348-S-6
248-11-A PVU 252-45-S-6
248-26-C PVU 252-45-S-6
248-77-C PVU 127-114-S-4
250-42-A PVU 258-350-S-1
252-05-C PVU 258-147-S-6
252-09-C PVU 222-134-S-4

Figure 2. Schematic representation of *bla*$_{VIM-6}$-carrying integrons detected in carbapenem-resistant *P. aeruginosa* isolates from the APAC region. The 5 Kb integrons possessed an *orf* and an *aadA1* that were not present in 3.5 Kb structures. Filed circles represent the 59-be recombination sites.

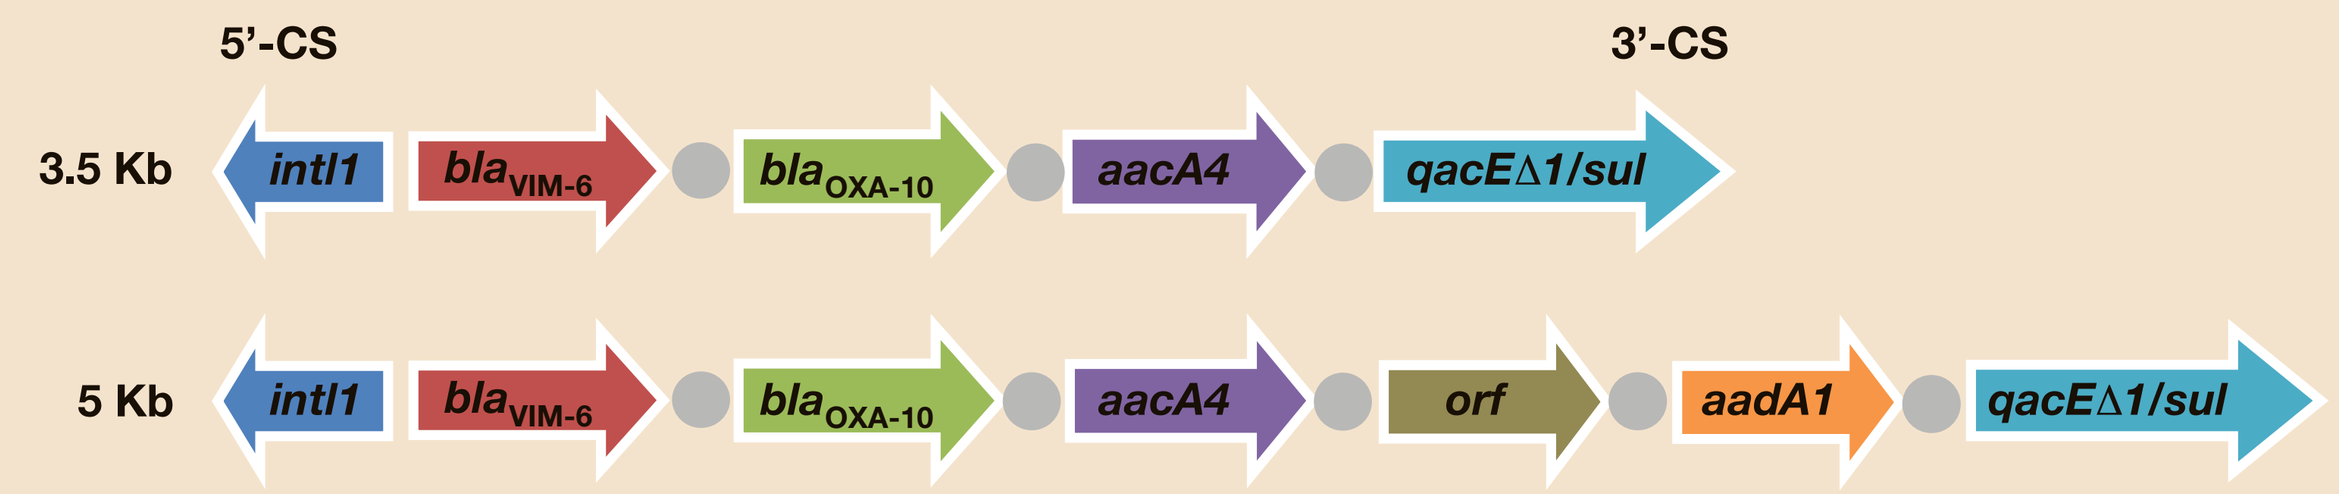

5'-CS
3'-CS
3.5 Kb
intI1
blaVIM-6
blaOXA-10
aacA4
qacE∆1/sul
5 Kb
intI1
blaVIM-6
blaOXA-10
aacA4
orf
aadA1
qacE∆1/sul

CONCLUSIONS

- The 5 Kb integrons found in this study seems to be identical to the *bla*$_{VIM-6}$-integron carried by the isolate found in Singapore which was deposited in the GenBank (# EF522838), but yet not described.
- Further studies are necessary to characterize the unknown *orf* found in this study, since this gene can encode resistance to other clinically important antimicrobial agents and play a role in the selection of strains harboring these mobile elements.
- The spread and high genetic diversity observed among *bla*$_{VIM-6}$-harboring isolates can be attributed to a promiscuous mobile structure that carries this gene, probably associated to an environmental source for this MßL gene in the region.
- Class 1 integrons play an important role in the dissemination and increasing prevalence of MßL-producing *P. aeruginosa* isolates. Hospital infection control measures may have limited effects because the dissemination of MßL-encoding genes seems to be related to the spread of genetic elements carrying these resistance elements.

SELECTED REFERENCES

1. Clinical and Laboratory Standards Institute. (2006). *M7-A7, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - seventh edition.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute. (2007). *M100-S17, Performance standards for antimicrobial susceptibility testing, 17th informational supplement*. Wayne, PA: CLSI.
3. Koh TH, Wang GC, Sng LH (2004). IMP-1 and a novel metallo-ß-lactamase, VIM-6, in fluorescent pseudomonads isolated in Singapore. *Antimicrob Agents Chemother* 48: 2334-2336.
4. Mendes RE, Kiyota KA, Monteiro J, Castanheira M, Andrade SS, Gales AC, Pignatari AC, Tufik S (2007). Rapid detection and identification of metallo-ß-lactamase-encoding genes by multiplex real-time PCR assay and melt curve analysis. *J Clin Microbiol* 45: 544-547.
5. Queenan AM, Bush K (2007). Carbapenemases: The versatile ß-lactamases. *Clin Microbiol Rev* 20: 440-458.
6. Toleman MA, Vinodh H, Sekar U, Kamat V, Walsh TR (2007). *bla*$_{VIM-2}$-harboring integrons isolated in India, Russia, and the United States arise from an ancestral class 1 integron predating the formation of the 3' conserved sequence. *Antimicrob Agents Chemother* 51: 2636-2638.