E-280

Antimicrobial Activity of ME1036 and Ceftaroline Tested against Clinical Strains of Community-Acquired Methicillin-Resistant *Staphylococcus aureus* (CA-MRSA)

HS SADER, TR FRITSCHE, RN JONES

JMI Laboratories, North Liberty, IA, USA

ICAAC 2007
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370
fax 319.665.3371
helio-sader@jmilabs.com

ABSTRACT

Background: ME1036 (ME), formerly CP5609, is a new parenteral carbapenem with a 7-acylated imidazo[5,1-beta]thiazole-2-yl group at the C-2 position. Ceftaroline (CTR) is formulated as a novel *N*-phosphono prodrug of an anti-MRSA cephalosporin that has high affinity for penicillin-binding protein (PBP) 2a. We evaluated the potency of both investigational drugs, ME and CTR against CA-MRSA.

Methods: 152 well characterized CA-MRSA isolated from human infections in 2000-2005 from geographically diverse USA locations (47 medical centers) were susceptibility tested by CLSI broth microdilution method against CTR, ME and 12 comparators. All organisms were tested for Panton-Valentine leukocidin (PVL) and SCC*mec* genes by PCR and typed by PFGE. *agr* type was evaluated on selected strains.

Results: ME was two- to four-fold more potent than CTR and 256-fold more active than ceftriaxone (CRO; see Table). High rates of resistance to CRO, amoxicillin/clavulanate and erythromycin were observed; while imipenem, levofloxacin and tetracycline showed good activity against tested strains. All isolates exhibited positive PCR results for PVL gene and SCC*mec* type IV. 95.6% of strains showed *agr* I and 1 strain was *agr* III. 103 strains (68%) had the USA300 PFGE clonal type, while 40 (26%) strains showed PFGE patterns with only a 1-2 band difference from USA300. Strains of the USA400 clone comprised 5%.

	Cumulative % inhibited at MIC (µg/ml) of:								
Antimicrobials	0.06	0.12	0.25	0.5	1	2	4	8	16
ME1036	11	80	97	100	100	100	100	100	100
Ceftaroline	0	0	1	98	100	100	100	100	100
Ceftriaxone	0	0	0	0	0	0	0	0	1
Imipenem	-[a]	-	-	91	97	100	100	100	100
Meropenem	-	-	-	6	20	90	98	100	100
Amoxicillin/ clavulanate	-	-	0	0	0	1	1	81	-

a. - = dilution concentration not tested.

Conclusion: USA300, which represents the major CA-MRSA epidemic/endemic clone in the USA, predominated among the collection of strains evaluated. CTR and ME were very active against a well characterized and geographically diverse collection of CA-MRSA.

INTRODUCTION

Ceftaroline fosamil (formerly PPI-0903, TAK-599) is a *N*-phosphono-type prodrug cephalosporin. Its active form, ceftaroline, is released in vivo upon hydrolysis of the phosphonate group. This cephem has documented high affinity for PBP2a and potent in vitro activity against oxacillin (methicillin)-resistant *S. aureus* (MRSA) and many other gram-positive organisms, while retaining activity against gram-negative bacilli. Ceftaroline has also exhibited excellent bactericidal activity versus drug resistant *Streptococcus pneumoniae* strains.

ME1036 (formerly CP5609) is a novel parenteral carbapenem with a 7-acylated imidazo[5,1-*b*]thiazole-2-yl group directly attached to the carbapenem moiety of the C-2 position. ME1036 has demonstrated high affinity to altered staphylococcal PBP2' and potent in vitro activity against MRSA. This compound has also shown potent in vitro activities against penicillin-resistant *S. pneumoniae*, *Enterococcus faecalis*, *Haemophilus influenzae*, and many members of the family Enterobacteriaceae.

The objective of the present study was to evaluate the in vitro activities of these two novel ß-lactam compounds against community-acquired MRSA (CA-MRSA) strains isolated from patients throughout the United States (USA).

MATERIALS AND METHODS

Bacterial isolates: A collection of 152 strains from geographically diverse locations (47 medical centers) were selected from surveillance programs performed in North America to include well characterized CA-MRSA isolated from human infections. The isolates were collected in the 2000-2005 period.

Susceptibility testing: Reagent grade ceftaroline and ME1036 compounds were provided by Cerexa, Inc. (Alameda, CA). Comparator agents were purchased from Sigma Chemical Co. (St. Louis, MO) or obtained from their respective manufacturers in the USA. Minimum inhibitory concentrations (MICs) were determined and interpreted by reference broth microdilution methods according to procedures published by the Clinical and Laboratory Standards Institute (CLSI, formerly the National Committee for Clinical Laboratory Standards).

Molecular characterization: PCR amplification of Panton-Valentine leukocidin (PVL) genes (*lukF-PV* and *lukS-PV*) was performed with primers listed below and procedures described by Lina et al. luk-PV-F: ATC ATT AGG TAA AAT GTC TGG ACA TGA TCC A, luk-PV-R: GCA TCA AST GTA TTG GAT AGC AAA AGC. All PVL-positive isolates were characterized for the type of SCC*mec* gene cassette using a multiplex PCR strategy. The primers amplified various DNA segments within SCC*mec* characteristic to each of the types I, II, III, and IV. The *mecA* gene was amplified as part of the multiplex PCR to serve as an internal control. PCR products were separated on 2% agarose gel in TAE buffer on Criterion Sub-cell GT system (Bio-Rad, Hercules, CA) and stained with ethidium bromide. SCC*mec* types were assigned based on the number and sizes of the amplicons obtained.

Epidemiologic typing: PVL-positive CA-MRSA isolates were subjected to pulsed-field gel electrophoresis (PFGE) according to the procedure described by Tenover et al. PFGE patterns were compared to CA-MRSA clones prevalent in the USA. The PFGE patterns were designated by a capital letter (eg. C, F, G and K). Strains were assigned with the same PFGE pattern only when all bands matched. When there was one or two bands difference, the strains were assigned as a sub-type or variant of the major type, which was designated with the same capital letter followed by an Arabic number (Example: C1, C2, C3, etc.).

RESULTS

- Both ceftaroline and ME1036 were highly active against the collection of CA-MRSA evaluated. Ceftaroline showed an MIC_{50} and MIC_{90} of 0.5 µg/ml, and the highest MIC value was 1 µg/ml; ME1036 showed an MIC_{50} of 0.12 µg/ml, MIC_{90} of 0.25 µg/ml and highest MIC value of only 0.5 µg/ml (Table 1).

- All isolates appeared susceptible to imipenem (highest MIC was 2 µg/ml), clindamycin (all MIC values at ≤0.25 µg/ml), trimethoprim/sulfamethoxazole (all MIC values at ≤0.25 µg/ml), linezolid (highest MIC was 2 µg/ml), daptomycin (highest MIC was 1 µg/ml) and vancomycin (highest MIC was 2 µg/ml). Furthermore, the vast majority (>90%) of strains were test-susceptible to meropenem (98.0% susceptible), levofloxacin (92.8%) and tetracycline (94.7%). In contrast, the vast majority of strains were resistant to ceftriaxone, amoxicillin/clavulanate and erythromycin (Table 1).

- Ceftaroline MIC range was very narrow (0.25 – 1 µg/ml), and 148 strains (97.4%) had an MIC result of 0.5 µg/ml. ME1036 MIC values ranged from 0.06 to only 0.5 µg/ml, with 69.7% of strains with a MIC value of 0.12 µg/ml (Table 2).

- Ceftaroline and ME1036 were the most active ß-lactams tested against this collection of CA-MRSA (Table 2).

- Ceftriaxone and amoxicillin/clavulanate showed limited activity against these CA-MRSA with 98.7% of strains showing an MIC value greater than or equal to the respective resistant breakpoint. In contrast, the carbapenems imipenem (MIC_{90}, ≤0.5 µg/ml) and meropenem (MIC_{90}, 2 µg/ml), were very active in vitro against this collection of strains, even though MRSA isolates are considered resistant to all marketed ß-lactams according to CLSI criteria (Table 2).

Table 1. In vitro activity of ceftaroline and ME1036 tested against a selected collection (152 strains) of well-characterized community-acquired methicillin-resistant *S. aureus*.

	MIC (µg/ml):			%	%
Antimicrobial agent	50%	90%	Range	susceptible	resistant
Ceftaroline	0.5	0.5	0.25-1	-[a]	-
ME1036	0.12	0.25	0.06-0.5	-[a]	-
Ceftriaxone	32	>32	16->32	0.0 (0.0)[b]	98.7
Imipenem	≤0.5	≤0.5	≤0.5-2	100.0 (0.0)[b]	0.0
Meropenem	2	2	≤0.5-8	98.0 (0.0)[b]	0.0
Amoxicillin/clavulanate	8	>8	2->8	1.3 (0.0)[b]	98.7
Erythromycin	>8	>8	2->8	1.3	98.0
Clindamycin	≤0.25	≤0.25	≤0.25	100.0	0.0
Levofloxacin	0.25	0.5	≤0.12->4	92.8	7.2
Tetracycline	≤0.5	≤0.5	≤0.5->16	94.7	4.6
Trim/sulfa[c]	≤0.25	≤0.25	≤0.25	100.0	0.0
Linezolid	2	2	1-2	100.0	-
Daptomycin	0.25	0.5	0.25-1	100.0	0.0
Vancomycin	1	1	0.5-2	100.0	0.0

a. = no breakpoint has been established by the CLSI for this category.
b. Number in parenthesis indicates population of strains considered susceptible by CLSI M100-S17 criteria.
c. Trimethoprim/sulfamethoxazole

Table 2. MIC distributions for ceftaroline, ME1036 and four other ß-lactams tested against a collection of community-acquired methicillin-resistant *S. aureus* strains isolated in the USA (152 strains).

	Number of strains (cumulative %) inhibited at MIC (µg/ml) of: Antimicrobial agent								
	≤0.06	0.12	0.25	0.5	1	2	4	8	16
Ceftaroline	0 (0.0)	0 (0.0)	1 (0.7)	148 (98.0)	3 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)
ME1036	16 (10.5)	106 (80.3)	26 (97.4)	4 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)
Ceftriaxone	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	2 (1.3)
Imipenem	-[a]	-	-	139 (91.4)	9 (97.4)	4 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)
Meropenem	-	-	-	9 (5.9)	22 (20.4)	106 (90.1)	12 (98.0)	3 (100.0)	0 (100.0)
Amoxicillin/clavulanate	-	-	0 (0.0)	0 (0.0)	0 (0.0)	1 (0.7)	2 (1.3)	121 (80.9)	-

a. - = untested dilution concentration.

- All isolates showed positive PCR results for PVL genes and SCC*mec* type IV.

- PCR screening for *agr* type was performed in a randomly selected group of 23 strains, and 22 of those showed *agr* type I while the remaining strain showed *agr* type II.

- The vast majority of strains (94.1%) showed a major PFGE type C, which is equivalent to that presented by clone USA300 and variants. One hundred and three strains (67.8%) showed an identical PFGE pattern (C = USA300-0114) while 40 other strains showed a recognized variation (one or two bands different) of the major PFGE pattern (C1 to C9 and C11 to C15). Furthermore, seven strains (4.6%) showed major pattern F (F, F1 and F2), which is equivalent to that presented by clone USA400 (Table 3).

Table 3. Frequency of PFGE patterns observed among the CA-MRSA collection evaluated in this study.

PFGE pattern	Frequency (%)
C[a]	103 (67.8)
C1	1 (0.7)
C2	2 (1.3)
C3	19 (12.5)
C4	5 (3.3)
C5	1 (0.7)
C6	1 (0.7)
C7	1 (0.7)
C8	4 (2.6)
C9	1 (0.7)
C11	1 (0.7)
C12	1 (0.7)
C13	1 (0.7)
C14	1 (0.7)
C15	1 (0.7)
All C variants (USA300)	143 (94.1)
F[a]	1 (0.7)
F1	5 (3.3)
F2	1 (0.7)
All F variants (USA400)	7 (4.6)
G	1 (0.7)
K	1 (0.7)
Other patterns	2 (1.4)

a. PFGE pattern C is equivalent to USA300 while pattern F is equivalent to USA400.

CONCLUSIONS

- The results of this study showed that ceftaroline and ME1036 were very active against a well-characterized collection of CA-MRSA isolates obtained from patients throughout the USA. The highest MIC observed for ceftaroline was 1 µg/ml and for ME1036 was 0.5 µg/ml.

- The collection of CA-MRSA strains evaluated was dominated by strains with PFGE patterns identical or similar to that of USA300 strain (94.1% of strains) which represents the predominant CA-MRSA epidemic/endemic clone on this continent.

- These compounds may represent an important addition to the antimicrobial armamentarium for treatment of both community- and hospital-acquired infections.

SELECTED REFERENCES

1. Clinical and Laboratory Standards Institute. (2006). *M7-A7, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically;approved standard - seventh edition.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute. (2007). *M100-S17, Performance standards for antimicrobial susceptibility testing, 17th informational supplement.* Wayne, PA: CLSI.
3. Deurenberg RH, Vink C, Kalenic S, Friedrich AW, Bruggeman CA, Stobberingh EE (2007). The molecular evolution of methicillin-resistant *Staphylococcus aureus*. *Clin Microbiol Infect* 13: 222-235.
4. Kurazono M, Hirai Y, Takahata S, Takayama Y, Yoshida T, Shitara E, Yonezawa M (2004). Efficacy of ME1036 against Enterobacteriaceae harboring a variety of ß-lactamases including ESBLs, Abstr. F-331. 44th ICAAC, Washington, D.C., 197.
5. Kurazono M, Ida T, Yamada K, Hirai Y, Maruyama T, Shitara E, Yonezawa M (2004). In vitro activities of ME1036 (CP5609), a novel parenteral carbapenem, against methicillin-resistant staphylococci. *Antimicrob Agents Chemother* 48: 2831-2837.
6. Page MG (2006). Anti-MRSA ß-lactams in development. *Curr Opin Pharmacol* 6: 480-485.
7. Sader HS, Fritsche TR, Kaniga K, Ge Y, Jones RN (2005). Antimicrobial activity and spectrum of PPI-0903M (T-91825), a novel cephalosporin, tested against a worldwide collection of clinical strains. *Antimicrob Agents Chemother* 49: 3501-3512.
8. Tenover FC, McDougal LK, Goering RV, Killgore G, Projan SJ, Patel JB, Dunman PM (2006). Characterization of a strain of community-associated methicillin-resistant *Staphylococcus aureus* widely disseminated in the United States. *J Clin Microbiol* 44: 108-118.

ACKNOWLEDGEMENTS

These studies were supported by Cerexa, Inc., a wholly-owned subsidiary of Forest Laboratories, Inc.