C2-138

Ceftaroline Activity Against Staphylococci and Characterization of *S. aureus* with Elevated Ceftaroline MIC Values: Results from the Ceftaroline Surveillance Program

H.S. SADER[1], R.E. MENDES[1], D. BIEK[2], I. CRITCHLEY[2], R.N. JONES[1]

[1]JMI Laboratories, North Liberty, IA; [2]Cerexa Inc., Oakland, CA (a wholly-owned subsidiary of Forest Laboratories, Inc., New York, NY)

ICAAC 2009
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, 319.665.3371
helio-sader@jmilabs.com

Amended Abstract

Introduction: Ceftaroline (CPT) is a novel parenteral cephalosporin with broad-spectrum activity including MRSA. In order to monitor the in vitro staphylococcal activity of CPT, clinical isolates of *S. aureus* (SA) and coagulase-negative staphylococci (CoNS) were collected from 55 medical centers in the USA, 12 European countries, and Israel.

Methods: Unique clinical SA strains (6665) and CoNS (1072) were consecutively collected from >50 hospitals in 2008 and tested for susceptibility (S) by CLSI broth microdilution methods against CPT and numerous comparators. SA with CPT MIC at >2 μg/mL were further tested as follows: *mecA*, *mecR1* and *mecI* were PCR amplified and sequenced; PVL genes and SCC*mec* types by PCR; clonality by PFGE, *spa* and MLST by reference methods.

Results: 56.8 and 27.2% of SA were MRSA in the USA and EU, respectively. CPT was very active against oxacillin-S SA (MSSA), MRSA and CoNS with MIC_{90}s of 0.25-0.5, 1-2 and 0.5-1 μg/mL, respectively. CPT was 16-fold more potent than ceftriaxone against MSSA. All MRSA were inhibited at ≤2 μg/mL of CPT except 4 strains from a Greek hospital with a CPT MIC of 4 μg/mL. These 4 clonal strains had no change in *mecR1, mecA* promoter and ribosomal binding site, but showed 3 amino acid (AA) mutations ($N \rightarrow K_{146/204}$; $E \rightarrow K_{150}$) in the non-penicillin binding domain (nPBD) of PBP2a. *mecI* could not be detected on these 4 strains, all of which were PVL negative, SCC*mec* III and related to the ST-239 / *spa* t037 clone.

Organism (no. [USA/EU])	Cumulative % (USA/EU) inhibited at ceftaroline MIC (μg/mL) of: ≤0.06	0.12	0.25	0.5	1	2	4
MSSA (1,711/1,966)	1.0/1.0	4.6/5.1	90.3/88.4	100/>99.9	100/100	-/-	-/-
MRSA (2,254/734)	0.0/0.0	0.1/0.0	1.1/1.9	35.1/28.5	94.8/82.6	100/99.5	100/100
CoNS (638/434)	19.4/15.7	32.3/24.4	59.4/50.0	90.3/80.2	98.1/91.2	100/98.2	100/100

Conclusions: Staphylococci, including MRSA, recently isolated in USA and EU were generally very S to CPT. AA mutations in the nPBD and lack of repressor (*mecI*) might explain elevation of CPT MIC observed with 4 clonally-related isolates.

Introduction

Staphylococcus aureus is a major cause of infections in hospitalized patients and possesses or can acquire a remarkable number of mechanisms for producing antimicrobial resistance. *S. aureus* infections are usually associated with complicated skin and skin structure infections (cSSSI) and bacteremia, but are also related to pulmonary infections. *Staphylococcus epidermidis* and other coagulase-negative staphylococci (CoNS), previously dismissed as culture contaminants, have assumed great importance as true pathogens. Infections caused by these organisms usually involve indwelling foreign bodies or devices, and resistance to multiple antimicrobial agents may further complicate therapy.

Ceftaroline is a novel, parenteral, broad-spectrum cephalosporin exhibiting bactericidal activity against gram-positive organisms, including methicillin (oxacillin)-resistant *S. aureus* (MRSA) and multidrug-resistant *Streptococcus pneumoniae* (MDRSP), as well as common gram-negative pathogens. Ceftaroline is currently in phase III clinical development. Favorable results have been reported from phase II and III trials on the efficacy and safety profile of ceftaroline for treatment of cSSSI and from phase III community-acquired bacterial pneumonia (CABP) trials.

The objective of this study was to evaluate the antimicrobial activity and spectrum of ceftaroline and comparator agents tested against clinical bacterial isolates of *S. aureus* and CoNS recently collected in medical institutions geographically dispersed throughout the United States (USA) and Europe.

Materials and Methods

Bacterial Isolates

Unique *S. aureus* clinical strains (6665) and CoNS (1072) were consecutively collected in 2008 from 55 medical centers in the USA, 12 European countries, and Israel.

Antimicrobial Susceptibility

All strains were tested for antimicrobial susceptibility by the broth microdilution method. Dry-form, validated microdilution panels and broth reagents were manufactured by TREK Diagnostics (Cleveland, OH, USA). Mueller-Hinton Broth adjusted to contain physiological levels of calcium (50 mg/L) was used when testing daptomycin. Comparator agents included those representing the most common classes and examples of drugs used for the empiric or directed treatment of the indicated pathogen. Concurrent testing of quality control (QC) strains determined that proper test conditions and procedures were used. The following strains were included: *S. aureus* American Type Culture Collection (ATCC) 29213, *Enterococcus faecalis* ATCC 29212, and *S. pneumoniae* ATCC 49619. Susceptibility percentages and validation of QC results were based on the Clinical and Laboratory Standards Institute (CLSI) guidelines or breakpoints. No criteria for ceftaroline susceptibility have been established.

Characterization of *S. aureus* Strains with Ceftaroline MIC of 4 μg/mL

Isolates with elevated ceftaroline MIC values (4 μg/mL) were epidemiologically typed by pulsed-field gel electrophoresis (PFGE) and the patterns obtained were compared to those of major USA and international MRSA clones (NARSA, www.narsa.net). Single (*spa*) and multilocus sequence typing (MLST) were performed on 1 representative of each of 2 PFGE clusters observed among the 4 isolates.

PVL (*lukF-PV* and *lukS-PV*) screening was performed by Real-Time (RT) PCR method and primers targeting the *tuf* gene from *Staphylococcus* spp. were added into the reaction for internal control purposes. SCC*mec* types (I through VI) were also determined using a multiplex PCR approach. Custom primers were designed to PCR amplify the entire *mecA* gene. Amplicons obtained were sequenced on both strands. The nucleotide and deduced amino acid sequences were analyzed using Lasergene software package (DNASTAR, Madison, WI, USA). Penicillin-binding protein (PBP) 2a-deduced amino acid sequences were compared with wild-type PBP 2a sequences available through GenBank (http://www.ncbi.nlm.nih.gov/blast/). *mecA* upstream regions of the evaluated isolates were also compared with wild-type sequences available through GenBank.

Results

- The highest ceftaroline MIC value among methicillin-susceptible *S. aureus* (MSSA) strains was only 1 μg/mL (1 isolate [0.03%]), and 88.4 to 90.3% of strains were inhibited at a ceftaroline MIC of ≤0.25 μg/mL (Table 1). Ceftaroline ($MIC_{50/90}$, 0.25 μg/mL) was 16-fold more active than ceftriaxone ($MIC_{50/90}$, 4 μg/mL), 8- to 16-fold more active than cefepime (MIC_{50}, 2 μg/mL and MIC_{90}, 4 μg/mL), and 8-fold more active than linezolid ($MIC_{50/90}$, 2 μg/mL) when tested against MSSA (Tables 2 and 3).

- Against MRSA, all isolates were inhibited at a ceftaroline MIC of ≤2 μg/mL, except for 4 isolates (0.06% of *S. aureus* and 0.13% of MRSA tested) that had ceftaroline MICs of 4 μg/mL (Table 1). These four MRSA isolates were from a single medical center located in Greece and showed a dominant PFGE pattern (83.8% similarity; Figure 1).

- Although ceftaroline MICs were slightly higher (approximately 4-fold) among MRSA (MIC_{50}, 1 μg/mL) compared with MSSA (MIC_{50}, 0.25 μg/mL), the activity of ceftaroline was significantly greater than that of other cephalosporins tested against MRSA .

- Ceftaroline potency against MRSA was similar to or 2-fold greater than that of linezolid (MIC_{50} and MIC_{90}, 2 μg/mL) and vancomycin (MIC_{50}, 1 μg/mL and MIC_{90}, 2 μg/mL; Tables 2 and 3).

- Isolates from the USA exhibited markedly higher resistance rates to oxacillin (56.8%) when compared with isolates from Europe (27.2%). MRSA strains showed high rates of resistance to erythromycin (92.9% in the USA and 66.6% in Europe), clindamycin (34.9% to 38.3%), and levofloxacin (70.3 to 84.1%; Tables 2 and 3).

- Ceftaroline was slightly more active against CoNS than against *S. aureus*. Although all oxacillin-resistant CoNS should be considered resistant to all β-lactams by the CLSI interpretive standard, ceftaroline was very active against oxacillin-resistant CoNS collected in the USA and European hospitals.

- The 4 rare MRSA isolates with ceftaroline MIC of 4 μg/mL clustered within a single epidemic group (coefficient similarity of 83.8%) and their PFGE patterns were most similar to that of the Hungarian/Brazilian clone. Furthermore, the 4 isolates could be further clustered within 2 groups of 2 isolates each (Figure 1). Representative isolates of each PFGE group were studied by *spa* and MLST typing. Both isolates were *spa* t037 and ST-239, which is also consistent with the Hungarian/Brazilian clone.

- All 4 MRSA isolates with ceftaroline MIC of 4 μg/mL were PVL-negative and harbored SCC*mec* type IIIA. Sequencing analysis of *mecA* gene revealed that all 4 evaluated isolates had 3 amino acid substitutions ($N \rightarrow K_{146/204}$; $E \rightarrow K_{150}$) in the non-penicillin-binding domain (nPBD) of PBP2a when compared with the consensus sequence from *S. aureus* COL, except for *S. aureus* isolate 062-13101A, which showed an additional $H \rightarrow N_{351}$ mutation. No nucleotide mutation was observed in *mecR1, mecA* promoter, or ribosomal binding site in any of the strains.

Table 1. Summary of Ceftaroline Activity Against Staphylococci from the USA and Europe.

Organism/region (no. of strains)	No. of organisms (cumulative %) inhibited at ceftaroline MIC (μg/mL) of: ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4
USA										
S. aureus (3965)	1 (0.0)	2 (0.1)	1 (0.1)	13 (0.4)	64 (2.0)	1489 (39.6)	933 (63.1)	1344 (97.0)	118 (100.0)	-
Oxacillin-susceptible (1711)	1 (0.1)	2 (0.2)	1 (0.2)	13 (1.0)	61 (4.6)	1467 (90.3)	166 (100.0)	-	-	-
Oxacillin-resistant (2254)	-	-	-	-	3 (0.1)	22 (1.1)	767 (35.1)	1344 (94.8)	118 (100.0)	-
CoNS (638)	2 (0.3)	2 (0.6)	17 (3.3)	103 (19.4)	82 (32.3)	173 (59.4)	197 (90.3)	50 (98.1)	12 (100.0)	-
Europe										
S. aureus (2700)	-	-	4 (0.1)	16 (0.7)	80 (3.7)	1651 (64.9)	423 (80.5)	398 (95.3)	124 (99.9)	4 (100.0)[a]
Oxacillin-susceptible (1966)	-	-	4 (0.2)	16 (1.0)	80 (5.1)	1637 (88.4)	228 (99.9)	1 (100.0)	-	-
Oxacillin-resistant (734)	-	-	-	-	-	14 (1.9)	195 (28.5)	397 (82.6)	124 (99.5)	4 (100.0)
CoNS (434)	0 (0.0)	1 (0.2)	11 (2.8)	56 (15.7)	38 (24.4)	111 (50.0)	131 (80.2)	48 (91.2)	30 (98.2)	8 (100.0)

a. These 4 isolates were collected from a single medical center in Greece and clustered in 1 epidemic group based on PFGE patterns (Figure 1).
CoNS = coagulase-negative staphylococci; PFGE = pulsed-field gel electrophoresis.

Table 2. In Vitro Activity of Ceftaroline and Selected Antimicrobial Agents Tested Against Staphylococci from the USA .

Antimicrobial agent (no. of strains)	MIC_{50}	MIC_{90}	Range	% Susceptible[a]	% Resistant[a]
S. aureus (3965)					
Ceftaroline	0.5	1	≤0.008 – 2	-	-
Oxacillin	>2	>2	≤0.25 – >2	43.2	56.8
Ceftriaxone	32	>32	≤0.25 – >32	43.2	56.8
Cefepime	8	>16	0.25 – >16	43.2	56.8
Erythromycin	>4	>4	≤0.25 – >4	31.3	68.0
Clindamycin	≤0.25	>2	≤0.25 – >2	76.8	22.9
Levofloxacin	≤0.5	>4	≤0.5 – >4	54.7	44.7
Trim/sulfa[b]	≤0.5	≤0.5	≤0.5 – >2	98.5	1.5
Linezolid	2	2	0.25 – >8	99.9	-
Vancomycin	1	1	≤0.12 – 2	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 4	99.8	-
MSSA[c] (1711)					
Ceftaroline	0.25	0.25	≤0.008 – 0.5	-	-
Oxacillin	0.5	0.5	≤0.25 – 2	100.0	0.0
Ceftriaxone	4	4	0.5 – 16	99.6	0.0
Cefepime	2	4	0.25 – 8	100.0	0.0
Erythromycin	≤0.25	>4	≤0.25 – >4	64.1	35.2
Clindamycin	≤0.25	≤0.25	≤0.25 – >2	92.7	7.1
Levofloxacin	≤0.5	4	≤0.5 – >4	88.4	11.1
Trim/sulfa	≤0.5	≤0.5	≤0.5 – >2	98.4	1.6
Linezolid	2	2	0.25 – 2	100.0	-
Vancomycin	1	1	≤0.12 – 2	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 1	100.0	-
MRSA[d] (2254)					
Ceftaroline	1	1	0.12 – 2	-	-
Oxacillin	>2	>2	>2	0.0	100.0
Ceftriaxone	>32	>32	≤0.25 – >32	0.0	100.0
Cefepime	16	>16	1 – >16	0.0	100.0
Erythromycin	>4	>4	≤0.25 – >4	6.5	92.9
Clindamycin	≤0.25	>2	≤0.25 – >2	64.7	34.9
Levofloxacin	>4	>4	≤0.5 – >4	29.1	70.3
Trim/sulfa	≤0.5	≤0.5	≤0.5 – >2	98.5	1.5
Linezolid	2	2	0.25 – >8	99.9	-
Vancomycin	1	1	0.25 – 2	100.0	0.0
Daptomycin	0.25	0.5	0.12 – 4	99.7	-
CoNS[e] (638)					
Ceftaroline	0.25	0.5	≤0.008 – 2	-	-
Oxacillin	>2	>2	≤0.25 – >2	28.8	71.2
Ceftriaxone	8	>32	≤0.25 – >32	28.8	71.2
Cefepime	2	>16	≤0.12 – >16	28.8	71.2
Erythromycin	>4	>4	≤0.25 – >4	32.6	66.8
Clindamycin	≤0.25	>2	≤0.25 – >2	64.9	33.1
Levofloxacin	4	>4	≤0.5 – >4	43.6	54.2
Trim/sulfa	≤0.5	>2	≤0.5 – >2	59.4	40.6
Linezolid	1	1	0.25 – >8	98.4	-
Vancomycin	1	2	0.25 – 4	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 4	99.5	-

a. Criteria as published by the Clinical and Laboratory Standards Institute (CLSI, 2009). - = No breakpoints have been established by the CLSI.
b. Trim/sulfa = Trimethoprim/sulfamethoxazole.
c. MSSA = methicillin-susceptible *S. aureus*.
d. MRSA = methicillin-resistant *S. aureus*.
e. Includes: *Staphylococcus auricularis* (8 strains), *Staphylococcus capitis* (27 strains), *Staphylococcus cohnii* (1 strain), *Staphylococcus epidermidis* (276 strains), *Staphylococcus haemolyticus* (26 strains), *Staphylococcus hominis* (52 strains), *Staphylococcus intermedius* (1 strain), *Staphylococcus lugdunensis* (24 strains), *Staphylococcus saprophyticus* (5 strains), *Staphylococcus sciuri* (1 strain), *Staphylococcus simulans* (3 strains), *Staphylococcus warnerii* (13 strains), *Staphylococcus xylosus* (1 strain), and unspeciated coagulase-negative staphylococci (200 strains).

Table 3. In Vitro Activity of Ceftaroline and Selected Antimicrobial Agents Tested Against Staphylococci from Europe .

Antimicrobial agent (no. of strains)	MIC_{50}	MIC_{90}	Range	% Susceptible[a]	% Resistant[a]
S. aureus (2700)					
Ceftaroline	0.25	1	0.03 – 4	-	-
Oxacillin	0.5	>2	≤0.25 – >2	72.8	27.2
Ceftriaxone	4	>32	1 – >32	72.8	27.2
Cefepime	2	>16	≤0.12 – >16	72.8	27.2
Erythromycin	≤0.25	>4	≤0.25 – >4	70.0	28.6
Clindamycin	≤0.25	>2	≤0.25 – >2	88.0	11.8
Levofloxacin	≤0.5	>4	≤0.5 – >4	72.5	26.8
Trim/sulfa[b]	≤0.5	≤0.5	≤0.5 – >2	99.3	0.7
Linezolid	2	2	0.25 – 4	100.0	-
Vancomycin	1	1	0.25 – 2	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 1	100.0	-
MSSA[c] (1966)					
Ceftaroline	0.25	0.5	0.03 – 1	-	-
Oxacillin	0.5	0.5	≤0.25 – 2	100.0	0.0
Ceftriaxone	4	4	1 – 32	99.8	0.0
Cefepime	2	4	0.25 – 8	100.0	0.0
Erythromycin	≤0.25	>4	≤0.25 – >4	84.2	14.4
Clindamycin	≤0.25	≤0.25	≤0.25 – >2	98.0	1.9
Levofloxacin	≤0.5	≤0.5	≤0.5 – >4	94.0	5.4
Trim/sulfa	≤0.5	≤0.5	≤0.5 – >2	99.5	0.5
Linezolid	2	2	0.5 – 2	100.0	-
Vancomycin	1	1	0.25 – 2	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 1	100.0	-
MRSA[d] (734)					
Ceftaroline	1	2	0.25 – 4	-	-
Oxacillin	>2	>2	>2	0.0	100.0
Ceftriaxone	>32	>32	4 – >32	0.0	100.0
Cefepime	>16	>16	≤0.12 – >16	0.0	100.0
Erythromycin	>4	>4	≤0.25 – >4	32.0	66.6
Clindamycin	≤0.25	>2	≤0.25 – >2	61.2	38.3
Levofloxacin	>4	>4	≤0.5 – >4	15.0	84.1
Trim/sulfa	≤0.5	1	≤0.5 – >2	98.9	1.1
Linezolid	2	2	0.25 – 4	100.0	-
Vancomycin	1	1	0.25 – 2	100.0	0.0
Daptomycin	0.25	0.5	0.12 – 1	100.0	-
CoNS[e] (434)					
Ceftaroline	0.25	1	0.015 – 4	-	-
Oxacillin	>2	>2	≤0.25 – >2	24.0	76.0
Ceftriaxone	16	>32	≤0.25 – >32	24.0	76.0
Cefepime	4	>16	0.25 – >16	24.0	76.0
Erythromycin	>4	>4	≤0.25 – >4	35.5	64.1
Clindamycin	≤0.25	>2	≤0.25 – >2	67.1	30.0
Levofloxacin	4	>4	≤0.5 – >4	45.4	52.3
Trim/sulfa	≤0.5	>2	≤0.5 – >2	63.6	36.4
Linezolid	1	1	0.12 – >8	99.5	-
Vancomycin	1	2	≤0.12 – 4	100.0	0.0
Daptomycin	0.25	0.5	≤0.06 – 2	99.8	-

a. Criteria as published by the Clinical and Laboratory Standards Institute (CLSI, 2009). - = No breakpoint has been established by the CLSI.
b. Trim/sulfa = Trimethoprim/sulfamethoxazole.
c. MSSA = methicillin-susceptible *S. aureus*.
d. MRSA = methicillin-resistant *S. aureus*.
e. Includes: *Staphylococcus auricularis* (1 strain), *Staphylococcus capitis* (17 strains), *Staphylococcus caprae* (1 strain), *Staphylococcus carnosus* (1 strain), *Staphylococcus chromogenes* (2 strains), *Staphylococcus cohnii* (1 strain), *Staphylococcus epidermidis* (211 strains), *Staphylococcus haemolyticus* (28 strains), *Staphylococcus hominis* (36 strains), *Staphylococcus lentus* (3 strains), *Staphylococcus lugdunensis* (7 strains), *Staphylococcus saprophyticus* (12 strains), *Staphylococcus sciuri* (2 strains), *Staphylococcus simulans* (1 strain), *Staphylococcus warnerii* (9 strains), *Staphylococcus xylosus* (2 strains), and unspeciated coagulase-negative staphylococci (100 strains).

Figure 1. PFGE Pattern Analysis of 4 *S. aureus* Isolates with Ceftaroline MIC Values of 4 μg/mL and *S. aureus* Control Isolate (062-4981X).

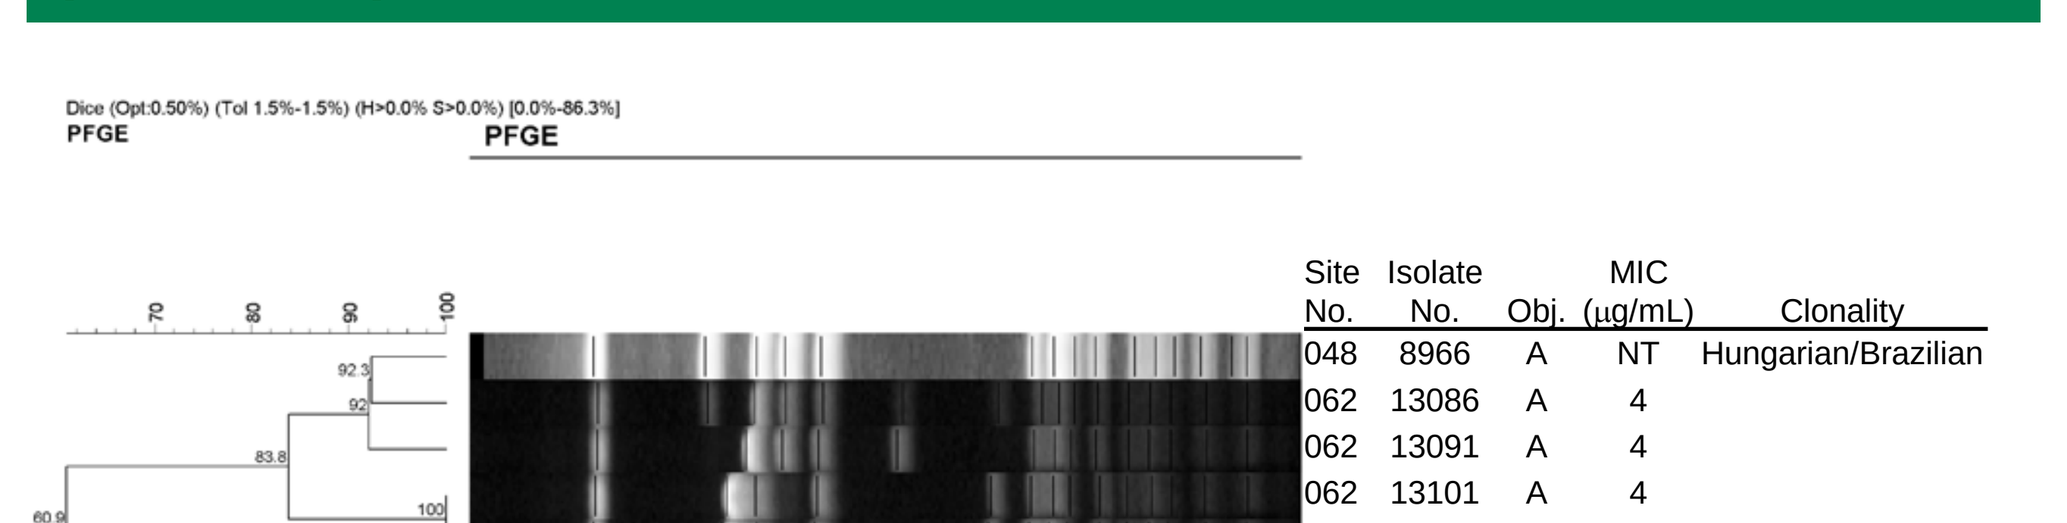

Dice (Opt:0.50%) (Tol 1.5%-1.5%) (H>0.0% S>0.0%) [0.0%-86.3%]
PFGE
PFGE
Site No.
Isolate No.
Obj.
MIC (μg/mL)
Clonality
048 8966 A NT Hungarian/Brazilian
062 13086 A 4
062 13091 A 4
062 13101 A 4
062 4993 X 4
062 4981 X 1

A representative strain (048-8966A) belonging to the Hungarian/Brazilian clone is also shown for comparison purposes. Percent similarities were identified on a dendrogram derived from the unweighted pair group method using arithmetic averages and based on Dice coefficients. Band position tolerance and optimization were set at 1.5% and 0.5%, respectively. Isolates showing similarity coefficient ≥80% were considered as genetically related. NT reads not tested.

Conclusions

- Ceftaroline demonstrated excellent activity against MRSA with 94.8 and 82.6% of strains in the USA and Europe, respectively, being inhibited at ≤1 µg/mL, and more than 99.9% inhibited by ≤2 µg/mL.

- Ceftaroline provides a significant potency advantage (≥ 16-fold) compared to ceftriaxone or cefepime against MSSA.

- Only four *S. aureus* isolates (0.06%) displayed ceftaroline MIC values >2 μg/mL (all at 4 μg/mL). These isolates were collected from a single medical center located in Athens, Greece, and appear to be derived from the same ancestor according to the molecular typing results.

- This favorable antimicrobial profile demonstrates that ceftaroline is a very promising anti-staphylococcal therapeutic option widely applicable in North America and Europe.

References

1. Clinical and Laboratory Standards Institute (2009). *M07-A8, Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - eighth edition.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2009). *M100-S19, Performance standards for antimicrobial susceptibility testing, 19th informational supplement.* Wayne, PA: CLSI.
3. Corey R, Wilcox M, Talbot GH (2008). CANVAS-1: randomized, double-blinded phase 3 study (P903-06) of the efficacy and safety of ceftaroline vs vancomycin plus aztreonam in complicated skin and skin structure infections (cSSSI). *48th ICAAC, October 25-28, 2008.*
4. Enright MC, Day NP, Davies CE, Peacock SJ, Spratt BG (2000). Multilocus sequence typing for characterization of methicillin-resistant and methicillin-susceptible clones of *Staphylococcus aureus*. *J Clin Microbiol*, 38: 1008-1015.
5. Kanafani ZA and Corey GR (2009). Ceftaroline: a cephalosporin with expanded Gram-positive activity. *Future Microbiol.* 4:25-33.
6. Lina G, Piemont Y, Godail-Gamot F, Bes M, Peter MO, Gauduchon V, Vandenesch F, Etienne J (1999). Involvement of Panton-Valentine leukocidin-producing *Staphylococcus aureus* in primary skin infections and pneumonia. *Clin Infect Dis,* 29: 1128-1132.
7. Milheirico C, Oliveira DC, de Lencastre H (2007). Update to the multiplex PCR strategy for assignment of *mec* element types in *Staphylococcus aureus*. *Antimicrob Agents Chemother* 51: 3374-3377.
8. Shopsin B, Gomez M, Montgomery SO, Smith DH, Waddington M, Dodge DE, Bost DA, Riehman M, Naidich S, Kreiswirth BN (1999). Evaluation of protein A gene polymorphic region DNA sequencing for typing of *Staphylococcus aureus* strains. *J Clin Microbiol*, 37: 3556-3563.
9. Talbot GH, Thye D, Das A, Ge Y (2007). Phase 2 study of ceftaroline versus standard therapy in the treatment of complicated skin and skin structure infections. *Antimicrob. Agents Chemother.* 51:3612-3616.

Acknowledgement

Supported by Forest Laboratories, Inc.