C2-621

Decreasing Susceptibility Rates Among *Acinetobacter* spp. Clinical Isolates Collected Worldwide (2005-2008)

RE MENDES, RN JONES, HS SADER

JMI Laboratories, North Liberty, IA, USA

ICAAC 2009
JMI Laboratories
North Liberty, IA, USA
www.jmilabs.com
319.665.3370, fax 319.665.3371
rodrigo-mendes@jmilabs.com

ABSTRACT

Background: Antimicrobial susceptibility of *Acinetobacter* spp. varies widely by region and is continuously being recognized as a serious therapeutic problem.

Methods: 4,204 *Acinetobacter* spp. were collected in USA (17%), European (EU; 22%), Latin American (LA; 25%) and Asian (36%) hospitals during 2005-2008 and tested for susceptibility by CLSI broth microdilution methods. 133 medical centers in 30 countries were surveyed. Tigecycline breakpoints established by the USA-FDA for Enterobacteriaceae (≤2 µg/ml for susceptible) were applied for comparison purposes. Annual susceptibility trends were determined by χ^2 test.

Results: Isolates were mainly from bacteremia (46.6%) and pneumonia (38.6%). Tigecycline ($MIC_{50/90}$, 0.5/2 µg/ml; 97.1% susceptible overall) and polymyxin B ($MIC_{50/90}$, ≤0.5/≤0.5 µg/ml; >99% susceptible) were the most active agents in all regions. Susceptibility rates to other agents were usually low and showed significant decline during the study period in all regions (**Table**). A slight reduction in tigecycline susceptibility rates occurred in Europe and Latin America in 2007-2008. Tigecycline MIC distribution was generally bimodal with highest MIC occurrences at 0.12 and 1 µg/ml and yearly variations. In 2008, imipenem and amikacin susceptibility rates were 73 and 73, 60 and 42, 59 and 28, and 62 and 52% in the USA, Europe, Latin America and Asia, respectively. Among countries with >100 samples tested, imipenem and amikacin susceptibility rates were greatest in Indonesia and Japan (93-97%). Imipenem susceptibility rates were as low as 24% in Argentina and 46-48% in Turkey and India; while amikacin susceptibility rates were <25% in Argentina, India, Mexico and Poland.

Conclusions: Continuously emerging resistance was observed among *Acinetobacter* spp. worldwide. Only tigecycline and polymyxin B showed good (>95%) in vitro activity against *Acinetobacter* spp. Although tigecycline showed an overall trend toward decreased susceptibility at ≤2 µg/ml, it was restricted to specific regions and years. Tigecycline should be further evaluated for treatment of *Acine*tobacter spp. infections.

Antimicrobial agents	2005 (609)	2006 (1,145)	2007 (1,436)	2008 (1,014)	Overall	P
	Worldwide % susceptibility rate by year (No. of strains)					
Tigecycline[a]	97.5	98.8	96.0	96.5 (-1.0%)[b]	97.1	0.006
Imipenem	77.0	65.9	63.9	50.2 (-26.8%)	63.0	<0.0001
Ampicillin/sulbactam	52.1	46.2	46.5	32.1 (-20.0%)	43.8	<0.0001
Cefepime	44.0	40.1	42.1	30.9 (-13.1%)	39.1	<0.0001
Amikacin	49.3	46.4	53.2	39.0 (-10.3%)	47.3	<0.0001
Polymyxin B	99.7	99.2	99.3	99.2 (-0.5%)	99.3	0.647

a. USA-FDA breakpoints for Enterobacteriaceae were applied for comparison purposes only.
b. Percentage difference in susceptibility rates from the years 2005 through 2008.

INTRODUCTION

Acinetobacter spp. is widely distributed in the environment and is part of the endogenous human bacterial flora, particularly skin, oral cavity and respiratory tract. These organisms are also found in many health care environments, are a very effective human colonizer among hospitalized patients and extremely capable of acquiring resistance determinants. These characteristics make *Acinetobacter* spp. a successful nosocomial pathogen.

Usually recognized as an opportunistic pathogen, *Acinetobacter* spp. represents the third leading cause of pneumonias in intensive care units (ICUs), and is responsible for up to 10% of hospital-acquired infections (HAIs). Furthermore, nearly 30% of *A. baumannii* recovered from ICU patients are resistant to at least three classes of antimicrobial agents, often including fluoroquinolones and carbapenems. The emergence of multidrug-resistant (MDR) *A. baumannii* and the dearth of new therapeutic options has increased the use of polymyxins B and E (colistin).

Tigecycline, a semisynthetic tetracycline-like agent, is active against several bacterial species and is stable against tetracycline resistance mechanisms. This agent inhibits protein translation by binding to the 30S rRNA, blocking the entry of amino-acyl tRNA molecules into the A site of the ribosome. Tigecycline was approved by the European Medicines Agency (EMEA) and the United States (USA) Food and Drug Administration (FDA) for the treatment of complicated skin and skin structure infections and intra-abdominal infections. This study evaluated the antimicrobial activity of tigecycline and comparator agents tested against a worldwide collection of *Acinetobacter* spp. isolated during the years of 2005 through 2008.

MATERIALS AND METHODS

Bacterial Strain Collection. A total of 4,204 *Acinetobacter* spp. were collected from 133 hospitals in 30 countries located in North America (USA; 17%), Europe (22%), Latin America (25%) and the Asia-Pacific region (APAC; 36%). The isolates were recovered from 2005 through 2008 as part of various global antimicrobial surveillance programs. Bacterial identification was confirmed by the central monitoring site (JMI Laboratories, Iowa, USA) using standard algorithms and an automated system, when needed (Vitek® 2; bioMerieux, Missouri, USA).

Susceptibility Test Methods. The isolates were tested for susceptibility by the reference Clinical and Laboratory Standards Institute (CLSI, formerly NCCLS) broth microdilution method using commercially prepared and validated panels (TREK Diagnostic Systems, Ohio, USA) in cation-adjusted Mueller-Hinton broth.

Susceptibility percentages were obtained based upon the CLSI breakpoints to determine susceptibility rates over the four-year interval. Tigecycline breakpoints established by the USA-FDA for Enterobacteriaceae (≤2 µg/ml for susceptible) were applied for comparison purposes. Annual susceptibility trends were determined by χ^2 test using the Epi Info™ Version 3.4.1 software package (Centers for Disease Control and Prevention, Georgia, USA). *P* values < 0.05 were considered to be significant.

Concurrent testing of quality control (QC) strains assured proper test conditions, procedures and included: *Staphylococcus aureus* ATCC 29213, *Enterococcus faecalis* ATCC 29212, *Escherichia coli* ATCC 25922 and *Pseudomonas aeruginosa* ATCC 27853. Validation of QC results was based upon the CLSI guidelines (M100-S19, 2009); all QC MIC results were within CLSI listed ranges.

ACKNOWLEDGEMENT

The study was supported by a grant from Wyeth Pharmaceuticals.

SELECTED REFERENCES

1. Adams MD, Goglin K, Molyneaux N, Hujer KM, Lavender H, Jamison JJ, MacDonald IJ, Martin KM, Russo T, Campagnari AA, Hujer AM, Bonomo RA Gill SR (2008). Comparative genome sequence analysis of multidrug-resistant *Acinetobacter baumannii*. *J Bacteriol* 190: 8053-8064.
2. Clinical and Laboratory Standards Institute (2009). *M07-A8. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - eighth edition*. Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2009). *M100-S19. Performance standards for antimicrobial susceptibility testing, 19th informational supplement.* Wayne, PA: CLSI.
4. Jones RN, Ferraro MJ, Reller LB, Schreckenberger PC, Swenson JM Sader HS (2007). Multicenter studies of tigecycline disk diffusion susceptibility results for *Acinetobacter* spp. *J Clin Microbiol* 45: 227-230.
5. Livermore DM (2005). Tigecycline: what is it, and where should it be used? *J Antimicrob Chemother* 56: 611-614.
6. Perez F, Endimiani A Bonomo RA (2008). Why are we afraid of *Acinetobacter baumannii*? *Expert Rev Anti Infect Ther* 6: 269-271.
7. Perez F, Hujer AM, Hujer KM, Decker BK, Rather PN Bonomo RA (2007). Global challenge of multidrug-resistant *Acinetobacter baumannii*. *Antimicrob Agents Chemother* 51: 3471-3484.

RESULTS

- The isolates were recovered from blood (46.6%) and respiratory tract specimens (38.6%), followed by skin and skin structure (12.7%), urinary tract (0.4%), catheter (0.1%) and other less prevalent clinical specimen types (1.5%).
- Tigecycline exhibited a bimodal MIC distribution (highest occurrences at MIC values of 0.25 and 1 µg/ml) during the 2005 and 2007 years sampling (**Figure 1A**). Whereas the tigecycline MIC distribution during 2008 year was more unimodal (modal MIC, 1 µg/ml).
- Tigecycline MIC distribution was bimodal when tested against *Acinetobacter* spp. isolates from the USA and the APAC region (**Figure 1B and E**), while unimodal against strains from Latin America with modal MIC value at 0.5 µg/ml (**Figure 1D**).
- A shift in the tigecycline MIC distribution was observed among *Acinetobacter* spp. from Europe over the study period (**Figure 1C**). Tigecycline showed a bimodal MIC distribution during 2005 and 2006, becoming more unimodal in 2007 and 2008, which was caused by isolates from Poland, Spain, Greece, Israel and Turkey (data not shown).

Table 1. Antimicrobial activity of tigecycline against *Acinetobacter* spp. isolates clustered by country (≥50 strains), continent, specimen type and year of sampling (2005-2008).

Region	Country/Specimen/Year	0.06	0.12	0.25	0.5	1	2	4	>4	MIC (µg/ml) 50%	90%	Susceptibility[a]
		No. of organisms (cumulative %) inhibited by tigecycline at MIC (µg/ml) of:										
Overall (4,204)		208 (4.9)	657 (20.6)	774 (39.0)	922 (60.9)	1082 (86.7)	440 (97.1)	108 (99.7)	13 (100.0)	0.5	2	97.1
North America	USA (719)	49 (6.8)	136 (25.7)	146 (46.0)	86 (58.0)	155 (79.6)	106 (94.3)	38 (99.6)	3 (100.0)	0.5	2	94.3
Europe	France (78)	5 (6.4)	12 (21.8)	24 (52.6)	9 (64.1)	18 (87.2)	4 (92.3)	4 (97.4)	2 (100.0)	0.25	2	92.3
	Germany (65)	7 (10.8)	27 (52.3)	21 (84.6)	8 (96.9)	2 (100.0)	–	–	–	0.12	0.5	100.0
	Poland (117)	1 (0.9)	2 (2.6)	13 (13.7)	20 (30.8)	38 (63.2)	36 (94.0)	7 (100.0)	–	1	2	94.0
	Greece (62)	2 (3.2)	3 (8.1)	6 (17.7)	9 (32.3)	24 (71.0)	17 (98.4)	1 (100.0)	–	1	2	98.4
	Italy (74)	1 (1.4)	2 (4.1)	9 (16.2)	22 (45.9)	24 (78.4)	11 (93.2)	5 (100.0)	–	1	2	93.2
	Israel (83)	2 (2.4)	4 (7.2)	12 (21.7)	16 (41.0)	31 (78.3)	18 (100.0)	–	–	1	2	100.0
	Turkey (306)	5 (1.6)	29 (11.1)	32 (21.6)	60 (41.2)	132 (84.3)	43 (98.4)	4 (99.7)	1 (100.0)	1	2	98.4
Latin America	Argentina (211)	–	5 (2.4)	23 (13.3)	81 (51.7)	60 (80.1)	32 (95.3)	9 (99.5)	1 (100.0)	0.5	2	95.3
	Brazil (506)	13 (2.6)	70 (6.4)	87 (33.6)	122 (57.7)	135 (84.4)	57 (95.7)	22 (100.0)	–	0.5	2	95.7
	Chile (125)	3 (2.4)	6 (7.2)	17 (20.8)	60 (68.8)	26 (89.6)	11 (98.4)	2 (100.0)	–	0.5	2	98.4
	Mexico (213)	7 (3.3)	24 (14.6)	58 (41.8)	82 (80.3)	25 (92.0)	12 (97.7)	1 (98.1)	4 (100.0)	0.5	1	97.7
Asia-Pacific	India (256)	9 (3.5)	31 (15.6)	41 (31.6)	89 (66.4)	69 (93.4)	14 (98.8)	2 (99.6)	1 (100.0)	0.5	1	98.8
	Indonesia (137)	10 (7.3)	40 (36.5)	48 (71.5)	17 (83.9)	15 (94.9)	6 (99.3)	–	1 (100.0)	0.25	1	99.3
	Japan (193)	16 (8.3)	76 (47.7)	67 (82.4)	14 (89.6)	17 (98.4)	1 (99.9)	2 (100.0)	–	0.25	1	99.0
	Korea (81)	2 (2.5)	5 (8.6)	5 (14.8)	23 (43.2)	39 (91.4)	7 (100.0)	–	–	1	1	100.0
	China (577)	43 (7.5)	105 (25.6)	96 (42.3)	139 (66.4)	157 (93.6)	34 (99.5)	3(100.0)	–	0.5	1	99.5
	Taiwan (93)	1 (1.1)	4 (5.4)	8 (14.0)	13 (28.0)	43 (74.2)	21 (96.8)	3 (100.0)	–	1	2	96.8
	Thailand (88)	6 (6.8)	15 (23.9)	15 (40.9)	18 (61.4)	27 (92.0)	5 (97.7)	2 (100.0)	–	0.5	1	100.0
Continent	North America (USA; 719)	49 (6.8)	136 (25.7)	146 (46.0)	86 (58.0)	155 (79.6)	106 (94.3)	38 (99.6)	3 (100.0)	0.5	2	94.3
	Europe (909)	39 (4.3)	110 (16.4)	147 (32.6)	161 (50.3)	292 (82.4)	133 (97.0)	24 (99.7)	3 (100.0)	0.5	2	97.0
	Latin America (1,055)	23 (2.2)	105 (12.1)	185 (29.7)	345 (62.4)	246 (85.7)	112 (96.3)	34 (99.5)	5 (100.0)	0.5	2	96.3
	Asia-Pacific (1,521)	97 (6.4)	306 (26.5)	296 (46.0)	330 (67.7)	389 (93.2)	89 (99.1)	12 (99.9)	2 (100.0)	0.5	1	99.1
Clinical specimen[b]	Bloodstream (1,960)	101 (5.2)	307 (20.8)	383 (40.4)	419 (61.7)	481 (86.3)	210 (97.0)	56 (99.8)	3 (100.0)	0.5	2	97.0
	RTI (1,623)	81 (5.0)	255 (20.7)	259 (36.7)	377 (59.9)	427 (86.2)	175 (97.0)	45 (99.8)	4 (100.0)	0.5	2	97.0
	SSSI (535)	23 (4.3)	70 (17.4)	109 (37.8)	114 (59.1)	154 (87.9)	52 (97.6)	7 (98.9)	6 (100.0)	0.5	2	97.6
Year	2005 (609)	29 (4.8)	92 (19.9)	145 (43.7)	127 (64.5)	142 (87.8)	59 (97.5)	14 (99.8)	1 (100.0)	0.5	2	97.5
	2006 (1,145)	65 (5.7)	197 (22.9)	178 (38.4)	314 (65.9)	285 (90.7)	92 (98.8)	12 (99.8)	2 (100.0)	0.5	1	98.8
	2007 (1,436)	66 (4.6)	240 (21.3)	298 (42.1)	269 (60.8)	332 (83.9)	174 (96.0)	52 (99.7)	5 (100.0)	0.5	2	96.0
	2008 (1,014)	48 (4.7)	128 (17.4)	153 (32.4)	212 (53.4)	323 (85.2)	115 (96.5)	30 (99.5)	5 (100.0)	0.5	2	96.5

a. Tigecycline breakpoints established by the USA-FDA for Enterobacteriaceae (≤2 µg/ml for susceptible) were applied for comparison purposes.
b. RTI, respiratory tract infections; SSSI, skin and skin structure infections.

Table 2. Spectrum of activity of tigecycline and comparator agents when tested against *Acinetobacter* spp. clinical isolates.

Antimicrobial agents	2005 MIC (µg/ml) 50%	2005 90%	2005 Susceptibility[a]	2006 MIC (µg/ml) 50%	2006 90%	2006 Susceptibility	2007 MIC (µg/ml) 50%	2007 90%	2007 Susceptibility	2008 MIC (µg/ml) 50%	2008 90%	2008 Susceptibility	Overall Susceptibility
Tigecycline	0.5	2	97.5	0.5	1	98.8	0.5	2	96.0	0.5	2	96.5	97.1
Imipenem	1	>8	77.0	2	>8	65.9	2	>8	63.9	4	>8	50.2	63.0
Meropenem	2	>8	70.8	2	>8	63.8	2	>8	63.0	8	>8	48.9	60.9
Ceftazidime	>16	>16	34.5	>16	>16	35.8	>16	>16	39.3	>16	>16	26.9	34.7
Cefepime	16	>16	44.0	16	>16	40.1	16	>16	42.1	>16	>16	30.9	39.1
A/S[b]	8	>16	52.1	16	>16	46.2	16	>16	46.5	>16	>16	32.1	43.8
P/T[b]	>64	>64	35.5	>64	>64	36.3	>64	>64	35.3	>64	>64	24.1	32.9
Amikacin	32	>32	49.3	>32	>32	46.4	8	>32	53.2	>32	>32	39.0	47.3
Gentamicin	>8	>8	42.2	>8	>8	38.9	>8	>8	44.9	>8	>8	37.2	41.0
Levofloxacin	4	>4	36.6	>4	>4	37.6	>4	>4	41.7	>4	>4	29.0	36.8
Minocycline	NT[c]	NT	NT	≤1	8	88.4	0.5	8	86.5	0.5	8	80.7	85.5
Tetracycline	4	>8	54.5	8	>8	45.0	8	>8	47.8	8	>8	37.1	45.4
Tobramycin	2	>16	58.6	2	>16	52.5	2	>16	58.0	16	>16	45.9	53.7
Polymyxin B	≤0.5	≤0.5	99.7	≤0.5	1	99.2	≤0.5	≤0.5	99.3	≤0.5	1	99.2	99.3

a. Tigecycline breakpoints established by the USA-FDA for Enterobacteriaceae (≤2 µg/ml for susceptible) were applied for comparison purposes.
b. A/S, ampicillin/sulbactam; P/T, piperacillin/tazobactam.
c. NT, not tested.

Figure 1: Tigecycline MIC distribution when tested against *Acinetobacter* spp. clinical isolates listed by region (A. Overall; B. North America [USA]; C. Europe; D. Latin America; E. APAC [Asia-Pacific]) and year within the sampling interval.

A. Overall

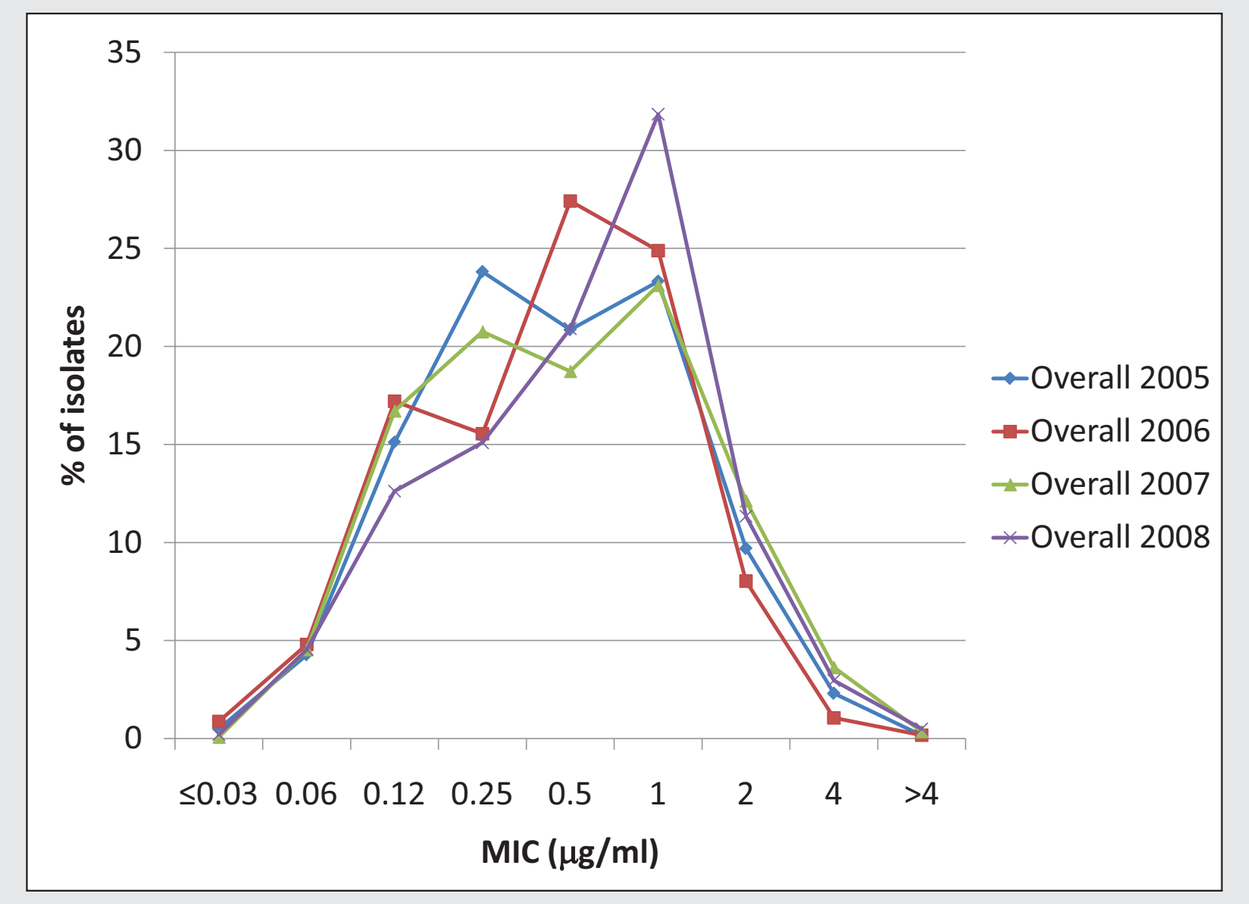

% of isolates
MIC (µg/ml)
≤0.03 0.06 0.12 0.25 0.5 1 2 4 >4
Overall 2005
Overall 2006
Overall 2007
Overall 2008

B. North America (USA)

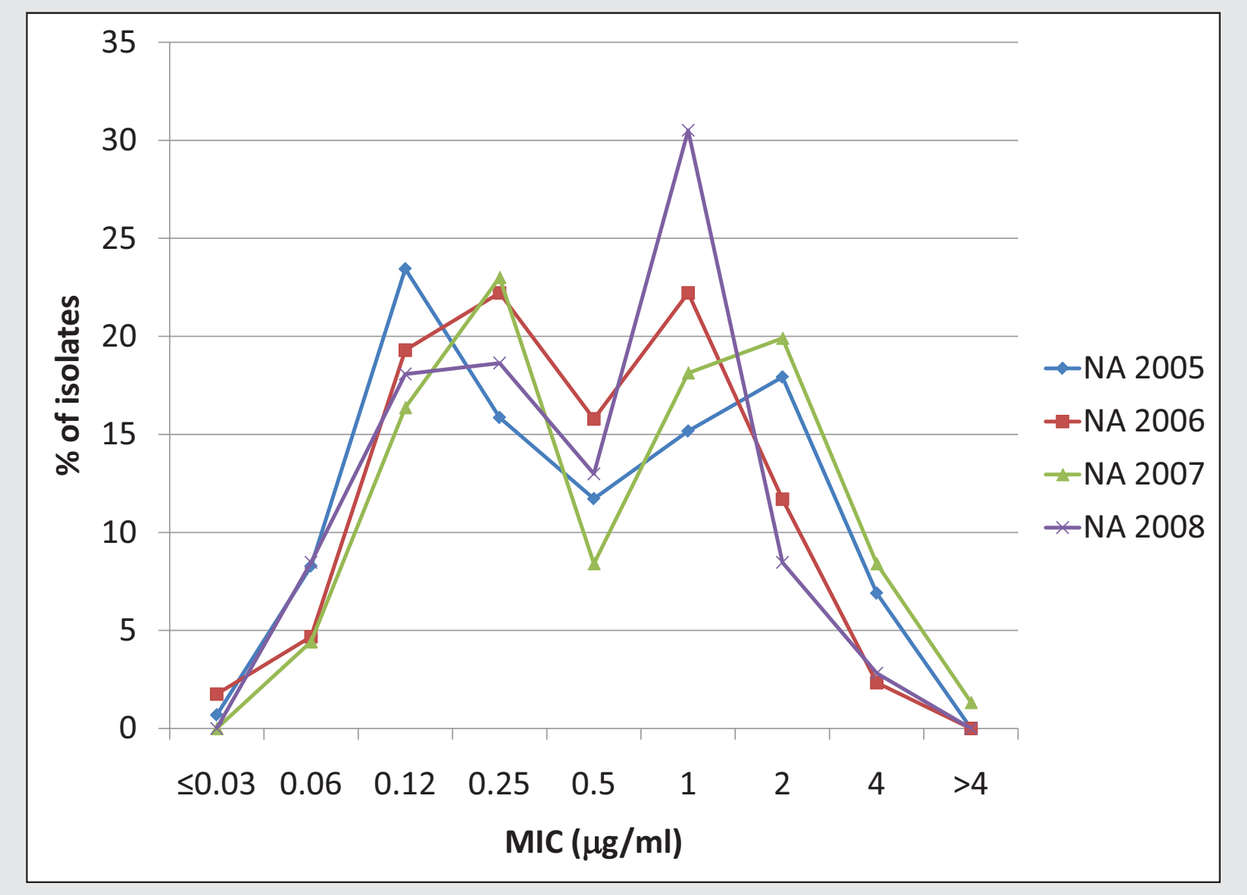

% of isolates
MIC (µg/ml)
≤0.03 0.06 0.12 0.25 0.5 1 2 4 >4
NA 2005
NA 2006
NA 2007
NA 2008

C. Europe

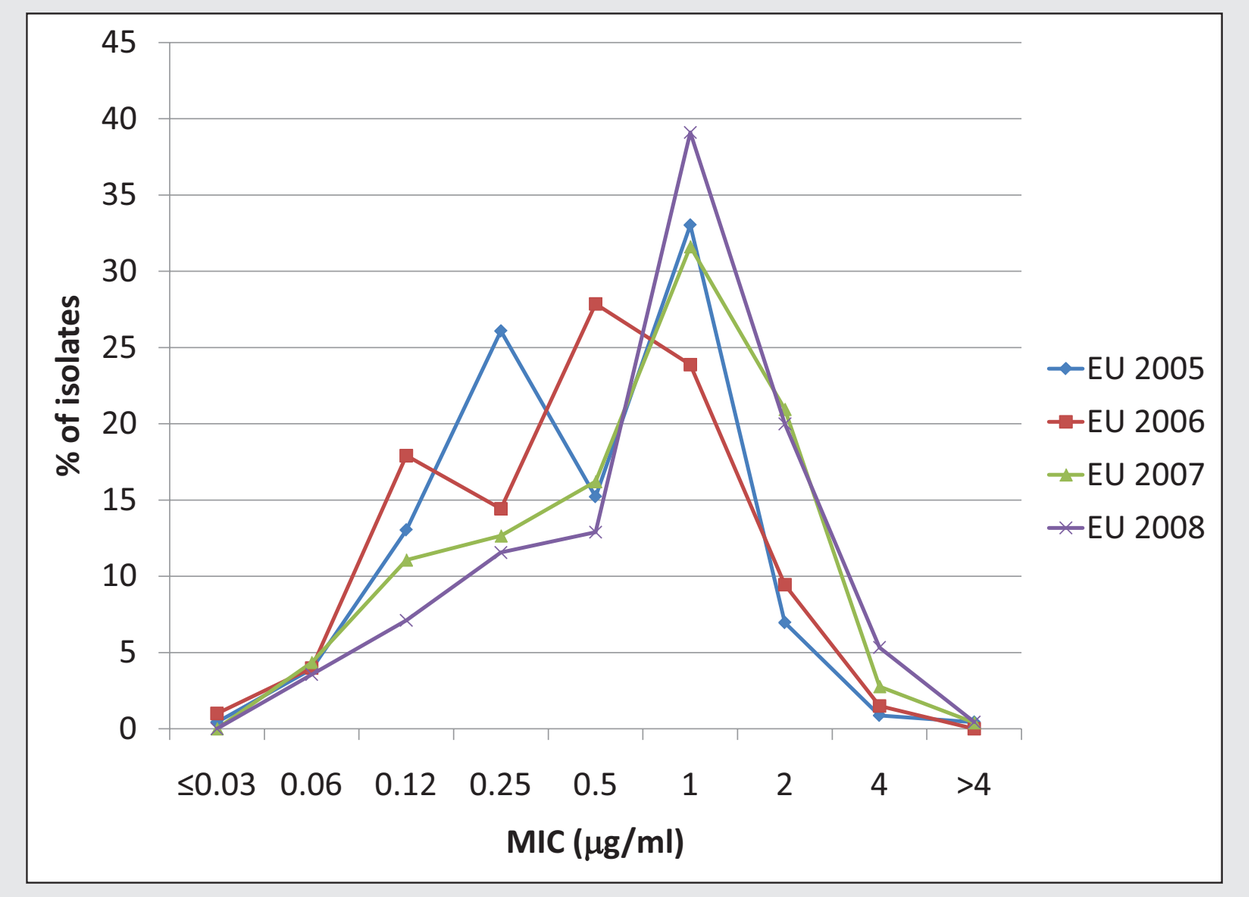

% of isolates
MIC (µg/ml)
≤0.03 0.06 0.12 0.25 0.5 1 2 4 >4
EU 2005
EU 2006
EU 2007
EU 2008

D. Latin America

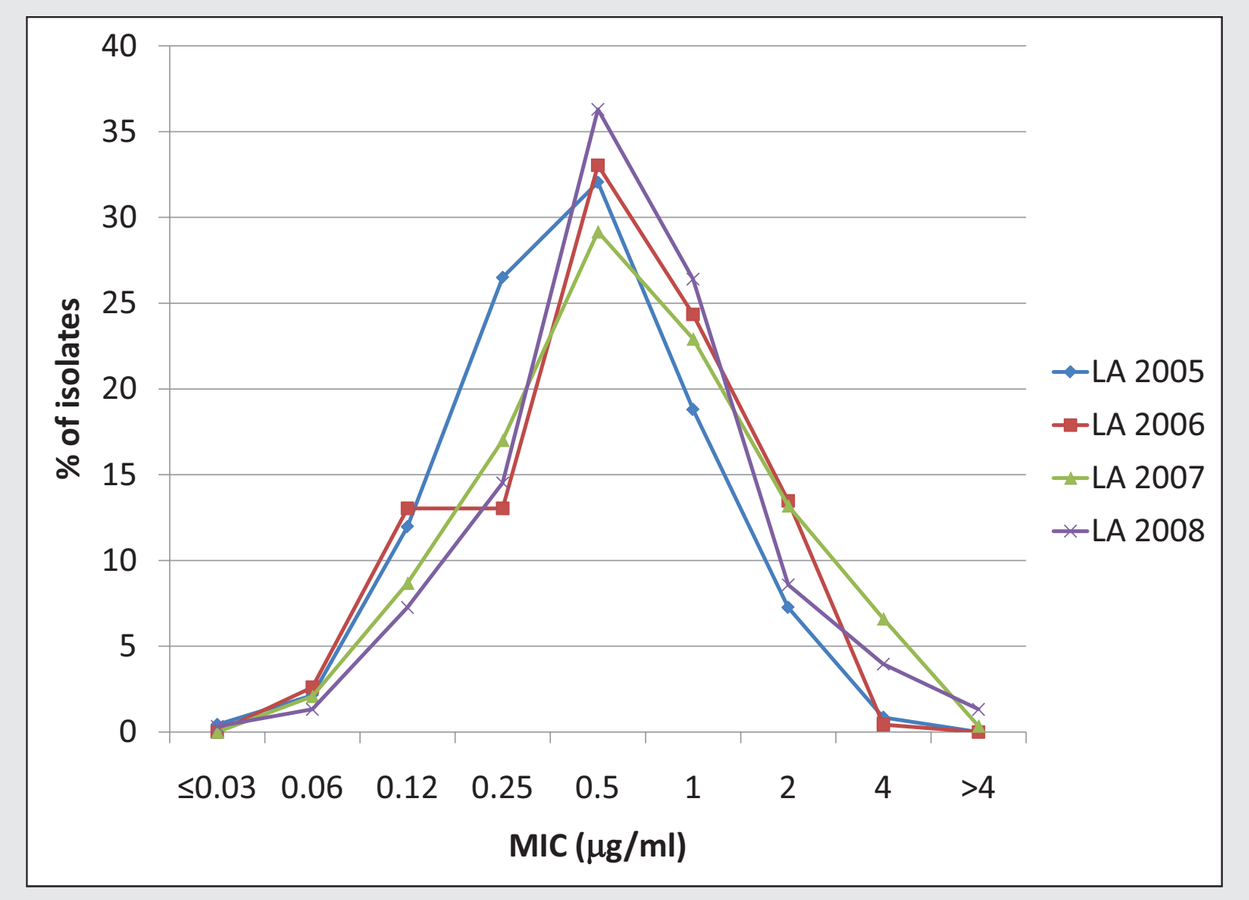

% of isolates
MIC (µg/ml)
≤0.03 0.06 0.12 0.25 0.5 1 2 4 >4
LA 2005
LA 2006
LA 2007
LA 2008

E. APAC

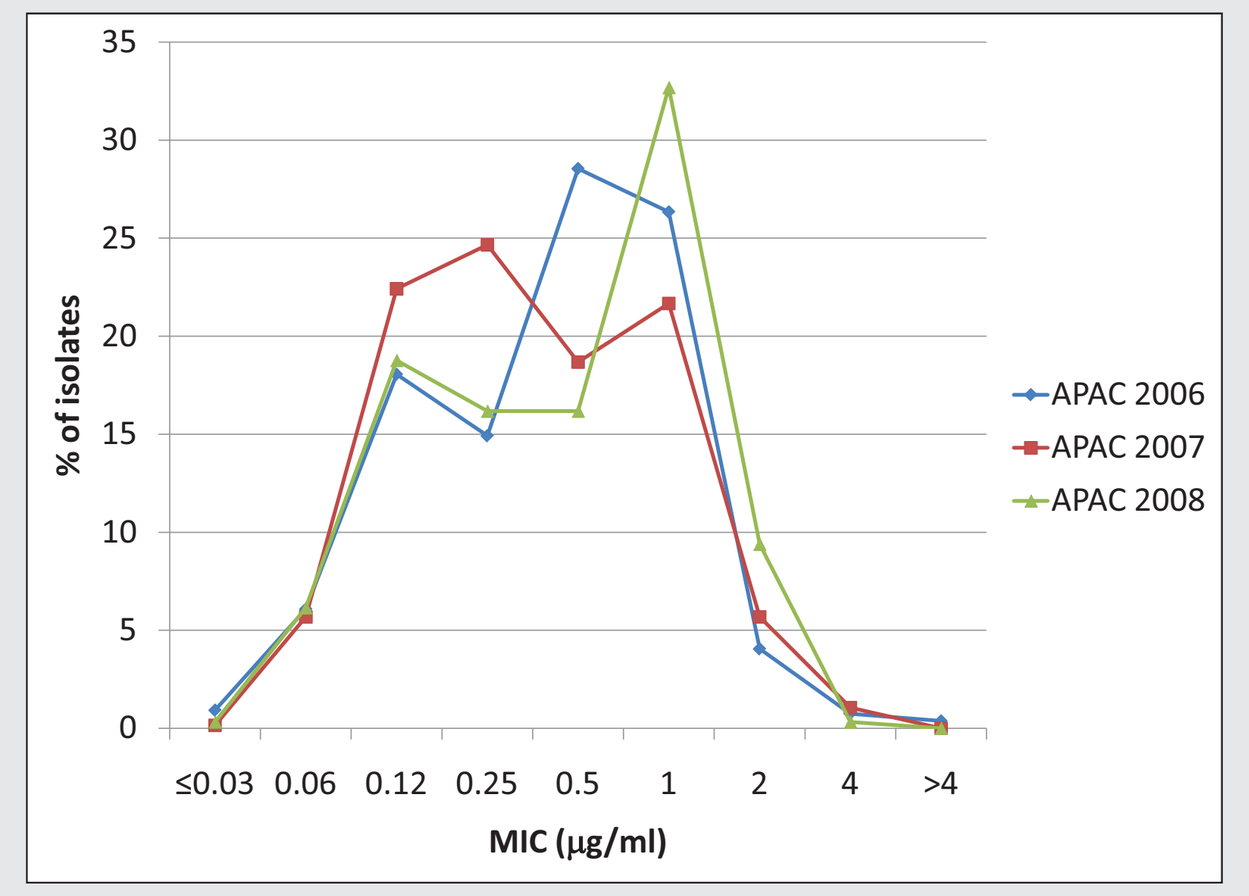

% of isolates
MIC (µg/ml)
≤0.03 0.06 0.12 0.25 0.5 1 2 4 >4
APAC 2006
APAC 2007
APAC 2008

- Overall, tigecycline demonstrated slightly higher potency against *Acinetobacter* spp. originating from countries in the APAC region (MIC_{90}, 1 µg/ml; **Table 1**) compared to isolates recovered from countries in the other geographic areas (MIC_{90}, 2 µg/ml).
- Tigecycline MIC_{90} results did not vary significantly by year of sampling (1 – 2 µg/ml) or clinical specimen type (2 µg/ml; **Table 1**).
- Tigecycline non-susceptibility rates (MIC, ≥4 µg/ml) were highest in Switzerland (11.1%), France (7.7%), Italy (6.8%), Poland (6.0%) and the USA (5.7%).
- Based on overall MIC_{90} values, tigecycline (MIC_{90}, 1-2 µg/ml; ≥96.0% susceptible) and polymyxin B (MIC_{90}, ≤0.5-1 µg/ml; ≥99.2% susceptible) were the most potent compounds, followed by minocycline (MIC_{90}, 8 µg/ml; ≥80.7% susceptible; **Table 2**). The remaining comparator agents showed limited activity.
- Except for polymyxin B (99.7-99.2%; *P*=0.647), all antimicrobial agents tested showed a trend toward decreasing susceptibility rates over the study period (**Table 2**).
- A slight reduction in tigecycline susceptibility rates occurred in Europe (from 98.7-98.5% in 2005-2006, to 96.8-94.2% in 2007-2008) and Latin America (from 99.1-99.6% in 2005-2006, to 93.1-94.7% in 2007-2008) during the study period (data not shown).

CONCLUSIONS

- Overall, a shift in the tigecycline MIC distribution (from bimodal to unimodal) was observed over the study period. However, this alteration was likely driven by the MIC distribution shift noted among 2007-2008 isolates in specific European countries.
- Only tigecycline and polymyxin B demonstrated acceptable (>95%) in vitro activity against this worldwide collection of *Acinetobacter* spp. clinical isolates.
- Tigecycline was very active against *Acinetobacter* spp. ($MIC_{50/90}$, 0.5/2 µg/ml) and 97.1% of 4,204 strains were inhibited at ≤2 µg/ml. Although tigecycline showed a trend toward decreased susceptibility at ≤2 µg/ml, this trend was restricted to *Acinetobacter* spp. isolated from Europe and Latin America (2007 and 2008) and was only a 1.0% decrease in the susceptibility overall (**Table 2**).
- *Acinetobacter* spp. is a constantly evolving pathogen and decreasing susceptibility rates were observed during a recent four-year study interval. These results emphasize the need for new therapeutic options, and tigecycline may be an alternative to polymyxins and should be further evaluated for treatment of *Acinetobacter* spp. infections via structured clinical trials.