POSTER # F1-1626

ANTIMICROBIAL ACTIVITY OF A NOVEL POLYMYXIN ANALOG (CB-182,804) TESTED AGAINST CLINICAL STRAINS OF GRAM-NEGATIVE BACILLI, INCLUDING COLISTIN-RESISTANT ORGANISMS

H.S. SADER, P.R. RHOMBERG, R.N. JONES

Cubist Pharmaceuticals, Inc., Lexington, MA.

H.S. SADER
JMI LABORATORIES
345 BEAVER KREEK CENTRE, SUITE A
NORTH LIBERTY, IA 52246 USA
HELIO-SADER@JMILABS.COM

ABSTRACT

Background: CB-182,804 is structurally related to polymyxins and has shown rapid bactericidal activity against many Gram-negative (GN) pathogens, including multidrug-resistant (MDR) organisms.

Methods: 455 strains selected from various surveillance programs were susceptibility (S) tested by CLSI broth microdilution methods. Strains with acquired resistance (R) to colistin (COL) and/or selected broad-spectrum antimicrobials were included in all organism groups. Organisms tested were: *P. aeruginosa* (PSA; 100), *Acinetobacter* spp. (ASP; 81), *B. cepacia* (BC; 18) *S. maltophilia* (XM; 25), *E. coli* (EC; 80), *K. pneumoniae* (KPN; 81), *Citrobacter* spp. (CSP; 20), *Enterobacter* spp. (ESP; 20), indole-positive Proteae (IPP; 10), *P. mirabilis* (PM; 10) and *S. marcescens* (SM; 10).

Results: CB-182,804 was very active against PSA ($MIC_{50/90}$, 0.5/2 µg/ml) and ASP ($MIC_{50/90}$, 1/4 µg/ml), including MDR strains. CB-182,804 was also active against *E. coli* ($MIC_{50/90}$, 1/2 µg/ml) and KPN ($MIC_{50/90}$, 1/4 µg/ml), including ESBL and serine carbapenemase (KPC-2 or 3) producers. When compared to COL, CB-182,804 was more active against PSA (MIC_{50} comparisons) and slightly less active against ASP, EC and KPN. CSP had lower MICs for CB-182,804 and COL, while both compounds exhibited variable activity against XM and ESP, and limited activity against BC, IPP, PM and SM.

Conclusions: CB-182,804 exhibited in vitro activity and spectrum similar to COL. CB-182,804 was very active against COL-S strains that were R to all other currently available antimicrobials for clinical use and may represent and an important therapeutic option for infections caused by pan-R GN bacilli. Cross-R with COL was observed.

INTRODUCTION

- The continued worldwide emergence of Gram-negative bacteria resistant to most available antimicrobial agents (so called "pan-resistant" organisms) has severely compromised the treatment of bacterial infections and led to increasing use of colistin and polymyxin B as "salvage" therapy.
- Colistin and polymyxin B possess antibacterial activity against a wide variety of Gram-negative pathogens. The great majority of *Pseudomonas aeruginosa*, *Acinetobacter* spp., *Escherichia coli*, *Klebsiella* spp. and *Citrobacter* spp., all common and important nosocomial pathogens with increasing antimicrobial resistance, are usually susceptible to the polymyxin class agents.
- CB-182,804 (Figure 1) is structurally related to polymyxins and has shown rapid bactericidal activity against many Gram-negative pathogens, including multidrug-resistant (MDR) organisms. In the present study, we evaluated the antimicrobial activity of CB-182,804 tested by reference methods against a collection of clinical Gram-negative bacilli enriched with MDR organisms.

Figure 1. Structure of CB-182,804

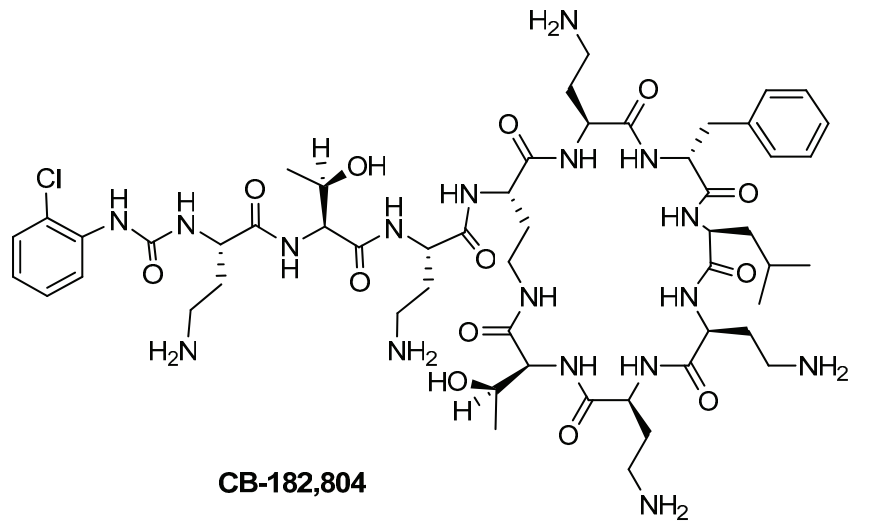

CB-182,804

METHODS

Bacterial isolates.

- A total of 455 Gram-negative bacilli causing clinical infections submitted to various surveillance programs (JMI Laboratories, North Liberty, IA) were utilized. Tested strains included *Enterobacteriaceae* (231 strains; 13 species) and non-fermentative Gram-negative bacilli (224 strains; 9 species). These challenge strains were selected to include resistance to colistin, carbapenems and/or broad-spectrum cephalosporins. Strains were primarily collected in 2006 except for a few with very uncommon resistance mechanisms or patterns, which were collected in previous years. Species identification was confirmed by standard biochemical tests and or the Vitek Systems (bioMerieux, Hazelwood, MO), when necessary.

Antimicrobial susceptibility testing.

- All isolates were tested for susceptibility using broth microdilution methods as described by the Clinical and Laboratory Standards Institute (CLSI; M07-A8, 2009) with cation-adjusted Mueller-Hinton broth. CB-182,804 and colistin were tested as well as 12 additional comparator agents. Susceptibility breakpoint interpretations were those found in CLSI documents (M100-S20-U), and quality control (QC) was assured with concurrent testing of E. coli ATCC 25922 and *Pseudomonas aeruginosa* ATCC 27853. All QC results were within CLSI specified ranges.

Genotypic detection of resistance.

- Multiplex PCR approaches were used to detect genes encoding plasmidic AmpCs, metallo-β-lactamases (IMP, VIM, SPM, GIM, SIM) and serine-carbapenemases (KPC, IMI/NMC-A, SME, OXA). Amplicons were sequenced and the nucleotide sequences and deduced amino acid sequences were analyzed using the Lasergene software package (DNASTAR, Madison, WI). Sequences were compared to others available via internet sources (http://www.ncbi.nlm.nih.gov/blast/).

RESULTS

- CB-182,804 (MIC_{50}, 0.5 µg/ml and MIC_{90}, 2 µg/ml) was very active and slightly more potent than colistin (MIC_{50}, 1 µg/ml and MIC_{90}, 2 µg/ml) when tested against *P. aeruginosa*, including MDR strains (resistant to carbapenems and/or aminoglycosides and/or fluoroquinolones; Tables 1 and 2).
- Acinetobacter spp. strains were very susceptible to CB-182,804 (MIC_{50}, 1 µg/ml and MIC_{90}, 4 µg/ml). CB-182,804 activity against *Acinetobacter* spp. was comparable to that of colistin (MIC_{50}, 0.5 µg/ml and MIC_{90}, 4 µg/ml; Tables 1 and 2).
- CB-182,804 was active against *E. coli* (MIC50, 1 µg/ml and MIC_{90}, 2 µg/ml) and *K. pneumoniae* (MIC_{50}, 1 µg/ml and MIC_{90}, 4 µg/ml), including strains producing extended-spectrum β-lactamases (ESBLs) and serine carbapenemases, such as KPC-2 or 3 (Table 3). CB-182,804 was less active than colistin ($MIC_{50/90}$, 0.25/0.5 µg/ml for *E. coli* and 0.5/2 µg/ml for *K. pneumoniae* respectively) against these organisms (Tables 1 and 2).
- *Citrobacter* spp. strains showed the lowest MIC values for CB-182,804 (MIC_{90}, 0.5 µg/ml) and colistin (MIC_{90}, 0.25 µg/ml; Tables 1 and 2).
- CB-182,804 and colistin possesed variable activity against *Enterobacter* spp. and *S. maltophilia*; while all strains of *B. cepacia*, indole-positive *Proteae*, *P. mirabilis* and *S. marcescens* showed elevated MIC values for CB-182,804 and colistin (Table 1).
- Table 2 summarizes the *in vitro* activities of CB-182,804 and all comparator agents evaluated. Resistance to comparator agents were generally high since a great proportion of strains with acquired resistance to colistin and/or carbapenems and/or broad-spectrum cephalosporins were pre-selected for study, and included in all organism groups evaluated.

Table 1. Frequency of occurrence and cumulative percentage inhibited at MIC values for CB-182,804 and colistin against a large collection of Gram-negative bacilli.

	No. of isolates (cumulative %) inhibited at MIC (µg/ml) of:										
Organism (no. tested)	≤0.12	0.25	0.5	1	2	4	8	16	32	64	>[a]
P. aeruginosa (100)											
CB-182,804	0 (0.0)	5 (5.0)	70 (75.0)	11 (86.0)	4 (90.0)	1 (91.0)	0 (91.0)	0 (91.0)	-	-	9 (100.0)
Colistin	0 (0.0)	3 (3.0)	19 (22.0)	62 (84.0)	6 (90.0)	2 (92.0)	2 (94.0)	1 (95.0)	1 (96.0)	0 (96.0)	4 (100.0)
Acinetobacter spp. (81)											
CB-182,804	1 (1.2)	5 (7.4)	18 (29.6)	39 (77.8)	9 (88.9)	3 (92.6)	2 (95.1)	2 (97.5)	-	-	2 (100.0)
Colistin	4 (4.9)	23 (33.3)	37 (79.0)	5 (85.2)	1 (86.4)	3 (90.1)	2 (92.6)	1 (93.8)	1 (95.1)	0 (95.1)	4 (100.0)
B. cepacia (18)											
CB-182,804	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	-	-	18 (100.0)
Colistin	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (5.0)	0 (5.0)	17 (100.0)
S. maltophilia (25)											
CB-182,804	0 (0.0)	1 (4.0)	3 (16.0)	0 (16.0)	3 (28.0)	2 (36.0)	5 (56.0)	4 (72.0)	-	-	7 (100.0)
Colistin	2 (8.0)	3 (20.0)	6 (44.0)	4 (60.0)	4 (76.0)	0 (76.0)	3 (88.0)	1 (92.0)	2 (100.0)	0 (100.0)	0 (100.0)
E. coli (80)											
CB-182,804	0 (0.0)	1 (1.3)	9 (12.5)	42 (65.0)	23 (93.8)	0 (93.8)	0 (93.8)	1 (95.0)	-	-	4 (100.0)
Colistin	3 (3.8)	40 (53.8)	32 (93.8)	0 (93.8)	0 (93.8)	1 (95.0)	3 (98.8)	0 (98.8)	0 (98.8)	0 (98.8)	1 (100.0)
K. pneumoniae (81)											
CB-182,804	0 (0.0)	1 (1.2)	14 (18.5)	50 (80.3)	7 (88.9)	1 (90.1)	0 (90.1)	3 (93.8)	-	-	5 (100.0)
Colistin	0 (0.0)	37 (46.7)	30 (82.7)	5 (88.9)	1 (90.2)	0 (90.2)	3 (93.8)	3 (97.5)	2 (100.0)	0 (100.0)	0 (100.0)
Citrobacter spp. (20)											
CB-182,804	0 (0.0)	1 (5.0)	18 (95.0)	1 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	-	-	0 (100.0)
Colistin	0 (0.0)	18 (90.0)	2 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)	0 (100.0)
Enterobacter spp. (20)											
CB-182,804	0 (0.0)	0 (0.0)	5 (25.0)	9 (70.0)	1 (75.0)	0 (75.0)	0 (75.0)	0 (75.0)	-	-	5 (100.0)
Colistin	0 (0.0)	13 (65.0)	2 (75.0)	0 (75.0)	0 (75.0)	1 (80.0)	0 (80.0)	0 (80.0)	1 (90.0)	1 (95.0)	2 (100.0)
Indole-positive *Proteae* (10)											
CB-182,804	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	-	-	10 (100.0)
Colistin	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	10 (100.0)
P. mirabilis (10)											
CB-182,804	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	-	-	10 (100.0)
Colistin	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	10 (100.0)
S. marcescens (10)											
CB-182,804	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	-	-	10 (100.0)
Colistin	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	10 (100.0)

a. Indicates >16 µg/ml for CB-182,804 and >64 µg/ml for colistin.
b. - = dilution not tested.

Table 2. Activity of CB-182,804 and comparator antimicrobial agents when tested against Gram-negative bacilli.

Organism/Antimicrobial agent	MIC_{50}	MIC_{90}	Range	%Susceptible / %Resistant[a]
Pseudomonas aeruginosa (100)				
CB-182,804	0.5	2	0.25->16	- / -
Colistin	1	2	0.25->64	90.0 / 8.0
Meropenem	2	>32	0.06->32	57.0 / 28.0
Ceftazidime	8	>16	≤1->16	51.0 / 44.0
Piperacillin/tazobactam	16	>64	≤0.5->64	67.0 / 33.0
Amikacin	8	>32	≤4->32	66.0 / 32.0
Ciprofloxacin	1	>4	≤0.03->4	51.0 / 42.0
Acinetobacter spp. (81)				
CB-182,804	1	4	0.12->16	- / -
Colistin	0.5	4	0.12->64	86.4 / 13.6
Meropenem	1	32	0.06->32	69.1 / 28.4
Ceftazidime	16	>16	≤1->16	46.9 / 48.1
Cefepime	8	>16	≤0.12->16	51.9 / 30.9
Piperacillin/tazobactam	16	>64	≤0.5->64	51.9 / 30.9
Amikacin	≤4	>32	≤4->32	69.1 / 24.7
Ciprofloxacin	1	>4	≤0.03->4	50.6 / 49.4
Tigecycline	0.5	2	≤0.03-4	- / -
Burkholderia cepacia (18)				
CB-182,804	>16	>16	>16	- / -
Colistin	>64	>64	32->64	0.0 / 100.0[c]
Meropenem	2	16	0.06-16	83.3 / 11.1
Ceftazidime	2	>16	≤1->16	88.9 / 11.1
Cefepime	4	>16	1->16	- / -
Piperacillin/tazobactam	2	>64	≤0.5->64	- / -
Amikacin	>32	>32	≤4->32	- / -
Ciprofloxacin	2	>4	1->4	- / -
Tigecycline	2	>4	0.25->4	- / -
Stenotrophomonas maltophilia (25)				
CB-182,804	8	>16	0.25->16	- / -
Colistin	1	16	0.12-32	76.0 / 24.0[c]
Ceftazidime	16	>16	≤1->16	40.0 / 44.0
Amikacin	>32	>32	≤4->32	- / -
Ciprofloxacin	2	>4	0.5->4	- / -
Tigecycline	0.5	2	0.12-8	- / -
Escherichia coli (80)				
CB-182,804	1	2	0.25->16	- / -
Colistin	0.25	0.5	0.12->64	- / -
Meropenem	0.03	0.06	≤0.015-8	96.3 / 1.2
Ceftazidime	≤1	>16	≤1->16	75.0 / 22.5
Piperacillin/tazobactam	2	64	≤0.5->64	82.3 / 8.9
Amikacin	≤4	8	≤4->32	97.5 / 1.3
Ciprofloxacin	≤0.03	>4	≤0.03->4	57.5 / 41.3
Tigecycline[b]	0.12	0.25	0.06-1	100.0 / 0.0
Klebsiella pneumoniae (81)				
CB-182,804	1	4	0.25->16	- / -
Colistin	0.5	2	0.25-32	60.1 / 9.9[c]
Meropenem	0.03	8	≤0.015->32	87.7 / 11.1
Ceftazidime	≤1	>16	≤1->16	67.9 / 29.6
Piperacillin/tazobactam	4	>64	≤0.5->64	72.8 / 22.2
Amikacin	≤4	32	≤4->32	84.0 / 4.9
Ciprofloxacin	≤0.03	>4	≤0.03->4	75.3 / 24.7
Tigecycline[b]	0.25	1	0.06-2	100.0 / 0.0
Citrobacter spp. (20)				
CB-182,804	0.5	0.5	0.25-1	- / -
Colistin	0.25	0.25	0.25-0.5	100.0 / 0.0[c]
Meropenem	0.03	0.06	≤0.015-0.12	100.0 / 0.0
Ceftazidime	≤1	>16	≤1->16	50.0 / 50.0
Piperacillin/tazobactam	4	32	1->64	75.0 / 5.0
Amikacin	≤4	≤4	≤4-8	100.0 / 0.0
Ciprofloxacin	≤0.03	0.12	≤0.03-0.25	100.0 / 0.0
Tigecycline[b]	0.25	0.25	0.12-0.5	100.0 / 0.0
Enterobacter spp. (20)				
CB-182,804	1	>16	0.5->16	- / -
Colistin	0.25	64	0.25->64	75.0 / 20.0
Meropenem	0.03	0.25	≤0.015-16	95.0 / 5.0
Ceftazidime	≤1	>16	≤1->16	75.0 / 25.0
Piperacillin/tazobactam	2	>64	1->64	75.0 / 15.0
Amikacin	≤4	≤4	≤4	100.0 / 0.0
Ciprofloxacin	≤0.03	>4	≤0.03->4	85.0 / 15.0
Tigecycline[b]	0.5	1	0.25-2	100.0 / 0.0
Indole-positive *Proteae* (10)				
CB-182,804	>16	>16	>16	- / -
Colistin	>64	>64	>64	0.0 / 100.0[c]
Meropenem	0.06	0.12	0.06-0.12	100.0 / 0.0
Ceftazidime	≤1	8	≤1->16	80.0 / 10.0
Piperacillin/tazobactam	2	4	≤0.5-4	100.0 / 0.0
Amikacin	≤4	8	≤4-8	100.0 / 0.0
Ciprofloxacin	>4	>4	≤0.03->4	40.0 / 60.0
Tigecycline	1	2	0.5-2	100.0 / 0.0
Proteus mirabilis (10)				
CB-182,804	>16	>16	>16	- / -
Colistin	>64	>64	>64	0.0 / 100.0[c]
Meropenem	0.06	0.06	0.03-0.06	100.0 / 0.0
Ceftazidime	≤1	4	≤1-4	100.0 / 0.0
Piperacillin/tazobactam	≤0.5	1	≤0.5-2	100.0 / 0.0
Amikacin	≤4	8	≤4-32	90.0 / 0.0
Ciprofloxacin	≤0.03	>4	≤0.03->4	80.0 / 20.0
Tigecycline	2	4	0.5-4	80.0 / 20.0
Serratia marcescens (10)				
CB-182,804	>16	>16	>16	- / -
Colistin	>64	>64	>64	0.0 / 100.0[c]
Meropenem	0.06	0.25	0.03-0.25	100.0 / 0.0
Ceftazidime	≤1	>16	≤1->16	50.0 / 50.0
Piperacillin/tazobactam	2	32	1->64	80.0 / 10.0
Amikacin	≤4	32	≤4-32	80.0 / 0.0
Ciprofloxacin	0.12	1	0.06->4	90.0 / 10.0
Tigecycline	1	2	0.5-2	100.0 / 0.0

a. Criteria as published by the CLSI [2010]. - = no interpretation criteria available.
b. US-FDA breakpoints were applied [Tygacil Product Insert, 2009].
c. Breakpoints established by the CLSI for *P. aeruginosa* were applied for comparison purpose only [CLSI 2010].

Table 3. Activity of CB-182,804 and selected comparator agents tested against β-lactamase producing Gram-negative organisms.

Organism (no. tested)/ Antimicrobial agent	MIC_{50}	MIC_{90}	MIC range	%S / %R
P. aeruginosa (19)[*]				
CB-182,804	0.5	1	0.5->16	- / -
Colistin	1	2	0.5-8	94.7 / 5.3
Meropenem	>32	>32	16->32	0.0 / 100.0
Ceftazidime	>16	>16	>16	0.0 / 100.0
A. baumanni (10)[†]				
CB-182,804	1	1	0.5-2	- / -
Colistin	0.5	0.5	0.25-0.5	100.0 / 0.0
Meropenem	16	>32	16->32	0.0 / 100.0
Ceftazidime	>16	>16	>16	0.0 / 100.0
E. coli (12)[‡]				
CB-182,804	1	2	0.25-2	- / -
Colisitin	0.5	0.5	0.12-0.5	100.0 / 0.0
Meropenem	2	4	0.03-8	66.7 / 25.0
Ceftazidime	16	>16	2->16	16.7 / 83.3
K. pneumoniae (10)[§]				
CB-182,804	1	1	0.5-1	- / -
Colistin	0.5	0.5	0.25-0.5	100.0 / 0.0
Meropenem	16	>32	1->32	10.0 / 90.0
Ceftazidime	>16	>16	>16	0.0/ 100.0

[*] Includes strains producing IMP (6), VIM (7) and SPM (6) metallo-β-lactamases.
[†] Includes strains producing OXA-23 (4), -24 (2) and -58 (4).
[‡] Includes strains producing KPC-2/3 (7), plasmidic AmpC enzymes (4) and SHV-5 (1).
[§] Includes strains producing KPC (6) and VIM (4) carbapenemases.

CONCLUSIONS

- CB-182,804 exhibited in vitro potency and a spectrum of activity most similar to that of colistin. CB-182,804 was active against colistin susceptible (MIC, ≤2 µg/ml) *P. aeruginosa* and *Acinetobacter* spp., including MDR strains.
- CB-182,804 was also active against *E. coli*, *K. pneumoniae*, *Citrobacter* spp., and *Enterobacter* spp. isolates including MDR strains. However, isolates with elevated colistin MIC results also exhibited elevated MIC values for CB-182,804.
- CB-182,804 showed limited activity against organisms intrinsically resistant to colistin, such as indole-positive *Proteae*, *P. mirabilis* and *S. marcescens*.
- CB-182,804 was active against colistin-susceptible strains that were resistant to all other currently available antimicrobials for clinical use, and this new agent may represent an important therapeutic option for infections caused by pan-resistant Gram-negative bacilli.

Selected References

1. Clinical and Laboratory Standards Institute (2009). *M07-A8. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eighth edition* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2010). M100-S20-U. *Performance standards for antimicrobial susceptibility testing: 20th informational supplement.* Wayne, PA: CLSI.
3. Gales AC, Jones RN, Sader HS (2006). Global assessment of the antimicrobial activity of polymyxin B against 54 731 clinical isolates of Gram-negative bacilli: report from the SENTRY antimicrobial surveillance programme (2001-2004). *Clin Microbiol Infect* 12: 315-321.
4. Giamarellou H, Poulakou G (2009). Multidrug-resistant Gram-negative infections: What are the treatment options? *Drugs* 69: 1879-1901.
5. Li J, Nation RL, Turnidge JD, Milne RW, Coulthard K, Rayner CR, Paterson DL (2006). Colistin: The re-emerging antibiotic for multidrug-resistant Gram-negative bacterial infections. *Lancet Infect Dis* 6: 589-601. 6. Livermore DM (2009). Has the era of untreatable infections arrived? *J Antimicrob Chemother* 64 Suppl 1: i29-i36.
7. Michalopoulos A, Falagas ME (2008). Colistin and polymyxin B in critical care. *Crit Care Clin* 24: 377-391.
8. Sader HS, Ferraro MJ, Reller B, Schreckenbrger P, Swenson JM, Jones RN (2006). Assessment of colistin and polymyxin B antimicrobial susceptibility testing methods against non-fermentative Gram-negative bacilli (NFGNB). Abstr. C173. *106TH ASM*, Orlando, Florida, USA.
9. Tygacil Package Insert (2009). Available at www.wyeth.com. Accessed August 2009.
10. Zavascki AP, Goldani LZ, Li J, Nation RL (2007). Polymyxin B for the treatment of multidrug-resistant pathogens: A critical review. *J Antimicrob Chemother* 60: 1206-1215.

ACKNOWLEDGMENT

This study was supported by a research grant from Cubist Pharmaceuticals.

This study was funded by Cubist Pharmaceuticals.

50th Interscience Conference on Antimicrobial Agents and Chemotherapy

September 12 –15, 2010, Boston, MA, USA