C-553

ICAAC/ICC 2015
San Diego, CA
September 17-21, 2015

In Vitro Activity of Tedizolid Against Streptococci and Enterococci Isolated From the United States and Europe in 2014

David Farrell[1], Mekki Bensaci[2*]

[1]JMI Laboratories, North Liberty, IA, USA; [2]Merck & Co., Inc., Kenilworth, NJ, USA

*Presenting author

Contact Information:
Mekki Bensaci, PhD
Merck & Co., Inc.
Kenilworth, NJ
908-740-4000
mekki.bensaci@merck.com

Introduction

- Streptococci and enterococci are common causative pathogens of acute bacterial skin and skin structure infections (ABSSSI)[1,2]
- Although β-hemolytic streptococci (BHS) remain susceptible to commonly used antibacterials, enterococci (predominantly *Enterococcus faecium*) are increasingly resistant to vancomycin, an antibacterial of last resort[1,3]
- Tedizolid phosphate, the prodrug of the active moiety tedizolid, is an oxazolidinone antibacterial currently approved for the treatment of ABSSSI in a number of countries, including the United States, Canada, and those of the European Union[4-6]
- Tedizolid is generally at least 4-fold more potent *in vitro* than linezolid against susceptible strains of streptococci and enterococci (including vancomycin-nonsusceptible enterococci) based on the minimum inhibitory concentration necessary to inhibit growth of 90% of the isolates (MIC_{90})[7-9]
- Two Phase 3 trials, ESTABLISH-1 (NCT01170221) and ESTABLISH-2 (NCT01421511), demonstrated the noninferior efficacy of tedizolid (200 mg once daily for 6 days) to linezolid (600 mg twice daily for 10 days) in patients with ABSSSI[10-12]
- The Surveillance of Tedizolid Activity and Resistance (STAR) program is an ongoing effort to monitor the activity of tedizolid against clinically relevant Gram-positive pathogens
- We report the *in vitro* activity of tedizolid, linezolid, and comparator agents against streptococcal and enterococcal isolates collected from the United States and Europe as part of the STAR program in 2014. MIC values of tedizolid and linezolid obtained at 80% and 100% reads were compared

Methods

Surveillance and Susceptibility Testing

- Nonduplicate, nonconsecutive, single-patient isolates of BHS, viridans group streptococci (including *Streptococcus anginosus* group: *S. anginosus, S. constellatus,* and *S. intermedius*), and enterococci were collected in 2014 from 29 medical centers in all 9 US census regions and from 34 medical centers in 15 European countries (considered among them for this analysis: Turkey, Ukraine, and Israel)
 - BHS: Total, 802; United States, 563; Europe, 239
 - Viridans group streptococci: Total, 370; United States, 260; Europe, 110
 - Enterococci: Total, 1290; United States, 524; Europe, 766
- Susceptibility testing was performed by broth microdilution in accordance with Clinical and Laboratory Standards Institute (CLSI) M07-A10 guidelines and CLSI M100-S25 guidelines at JMI Laboratories[13-15]
 - MIC values for tedizolid and linezolid were read in 2 ways:
 - MIC at the first well in which trailing begins (80% read; designated tedizolid 80 and linezolid 80)
 - MIC at endpoint (100% growth inhibition; designated tedizolid 100 and linezolid 100)
- Susceptibility results for tedizolid were interpreted using both European Committee on Antimicrobial Susceptibility Testing (EUCAST)-approved and U.S. Food and Drug Administration (FDA)-approved breakpoints[4,5]:
 - BHS
 - EUCAST: ≤0.5 μg/mL (susceptible) and >0.5 μg/mL (resistant) for groups A, B, C, G[5]
 - FDA: ≤0.5 μg/mL (susceptible) (*Streptococcus pyogenes* and *Streptococcus agalactiae* only)[4]
 - *S. anginosus* group (including *S. anginosus, S. constellatus*, and *S. intermedius*)
 - EUCAST: ≤0.25 μg/mL (susceptible) and >0.25 μg/mL (resistant)[5]
 - FDA: ≤0.25 μg/mL (susceptible)[4]
 - *Enterococcus faecalis*
 - FDA: ≤0.5 μg/mL (susceptible)[4]
- For comparator antibacterial agents, susceptibility results were interpreted according to CLSI and EUCAST criteria[14,16]

Results

Activity of Tedizolid and Linezolid Against BHS

- Tedizolid had potent activity against BHS, including *S. pyogenes* and *S. agalactiae* in 2014, with MIC_{90} of 0.12 μg/mL for tedizolid 80 and 0.25 μg/mL for tedizolid 100 (**Table 1**)
- The MIC_{90} of tedizolid was 8-fold lower than that of linezolid when read at 80% inhibition (0.12 μg/mL vs 1 μg/mL, respectively) and 4-fold lower when read at 100% inhibition (0.25 μg/mL vs 1 μg/mL, respectively)
- All *S. pyogenes* and *S. agalactiae* isolates were susceptible to tedizolid and linezolid
- Tedizolid was active against isolates that were resistant to clindamycin, erythromycin, levofloxacin, and tetracycline

Table 1. Activity of Tedizolid and Comparators Against BHS Isolated in 2014 From the United States and Europe

Pathogen (N)	Drug	Range	MIC_{50} μg/mL	MIC_{90} μg/mL	%S/I/R (EUCAST)	%S/I/R (CLSI)
All BHS[a] (802)	Tedizolid 80	0.03–0.25	0.12	0.12	–/–/–	–/–/–
	Tedizolid 100	0.06–0.5	0.12	0.25	–/–/–	–/–/–
	Linezolid 80	0.25–1	1	1	100.0/0.0/0.0	100.0/–/–
	Linezolid 100	0.5–4	1	1	–/–/–	–/–/–
	Ceftaroline	≤0.015–0.03	≤ 0.015	≤ 0.015	–/–/–	100.0 /–/–
	Ceftriaxone	≤0.06–0.25	≤0.06	0.12	100.0/0.0/0.0	100.0/–/–
	Clindamycin	≤0.25–>2	≤0.25	>2	83.9/0.0/16.1	82.9/1.0/16.1
	Daptomycin	≤0.06–0.5	0.12	0.25	100.0/0.0/0.0	100.0/–/–
	Erythromycin	≤0.12–>16	≤0.12	>16	68.9/1.5/29.6	68.9/1.5/29.6
	Levofloxacin	≤0.12–>4	0.5	1	97.8/1.7/0.5	99.5/0.0/0.5
	Penicillin	≤0.06–0.12	≤0.06	≤0.06	100.0/0.0/0.0	100.0/–/–
	Tetracycline	≤0.5–>8	1	>8	50.4/0.8/48.8	51.2/2.0 /46.8
	Vancomycin	0.25–0.5	0.25	0.5	100.0/0.0/0.0	100.0/–/–
S. pyogenes (334)	Tedizolid 80	0.03–0.25	0.12	0.12	–/–/–	–/–/–
	Tedizolid 100	0.06–0.5	0.12	0.25	–/–/–	100.0/0.0/0.0[b]
	Linezolid 80	0.5–1	0.5	1	100.0/0.0/0.0	100.0/–/–
	Linezolid 100	0.5–4	1	2	–/–/–	–/–/–
	Ceftaroline	≤0.015-0.03	≤0.015	≤0.015	–/–/–	100.0/–/–
	Ceftriaxone	≤0.06–0.25	≤0.06	≤0.06	–/–/–	100.0/–/–
	Clindamycin	≤0.25–>2	≤0.25	≤0.25	93.7/0.0/6.3	93.4/0.3/6.3
	Daptomycin	≤0.06–0.25	≤0.06	0.12	100.0/0.0/0.0	100.0/–/–
	Erythromycin	≤0.12–>16	≤0.12	4	86.5/0.9/12.6	86.5/0.9/12.6
	Levofloxacin	≤0.12–>4	0.5	1	96.7/3.0/0.3	99.7/0.0/0.3
	Penicillin	≤0.06–0.12	≤0.06	≤0.06	100.0/0.0/0.0	100.0/–/–
	Tetracycline	≤0.5->8	≤0.5	>8	80.5/0.9/18.6	81.4/0.9 /17.7
	Vancomycin	0.25–0.5	0.25	0.5	100.0/0.0/0.0	100.0/–/–
S. agalactiae (347)	Tedizolid 80	0.06–0.25	0.12	0.12	–/–/–	–/–/–
	Tedizolid 100	0.06–0.5	0.25	0.25	–/–/–	100.0/0.0/0.0[b]
	Linezolid 80	0.25–1	1	1	100.0/0.0/0.0	100.0/–/–
	Linezolid 100	0.5–4	1	1	–/–/–	–/–/–
	Ceftaroline	≤0.015–0.03	≤0.015	0.03	–/–/–	100.0/–/–
	Ceftriaxone	≤0.06–0.25	≤0.06	0.12	–/–/–	100.0/–/–
	Clindamycin	≤0.25–>2	≤0.25	>2	71.4/0.0/28.6	69.9/1.5/28.6
	Daptomycin	≤0.06–0.5	0.25	0.25	100.0/0.0/0.0	100.0/–/–
	Erythromycin	≤0.12–>16	0.25	>16	50.9/2.0/47.1	50.9/2.0/47.1
	Levofloxacin	0.25–>4	0.5	1	98.6/0.8/0.6	99.4/0.0/0.6
	Penicillin	≤0.06–0.12	≤0.06	≤0.06	100.0/0.0/0.0	100.0/–/–
	Tetracycline	≤0.5–>8	>8	>8	21.9/0.6/77.5	22.5/0.6/76.9
	Vancomycin	0.25–0.5	0.5	0.5	100.0/0.0/0.0	100.0/–/–

BHS, β-hemolytic streptococci; CLSI, Clinical and Laboratory Standards Institute; EUCAST, European Committee on Antimicrobial Susceptibility Testing; FDA, US Food and Drug Administration; I, intermediate; MIC_{50}, minimum inhibitory concentration required to inhibit growth of 50% of isolates; MIC_{90}, minimum inhibitory concentration required to inhibit growth of 90% of isolates; R, resistant; S, susceptible; –, no breakpoint available for interpretation.
[a]BHS spp include *S. pyogenes* (334), *S. agalactiae* (347), Group G Streptococcus (1) and *S. dysgalactiae* (120).
[b]FDA-approved breakpoint applied.

Activity of Tedizolid and Linezolid Against Viridans Group Streptococci (N=126)

- Tedizolid had potent activity against viridans group streptococci in 2014, with MIC_{90} of 0.12 μg/mL for tedizolid 80 and 0.25 μg/mL for tedizolid 100 (**Table 2**)
- The MIC_{90} of tedizolid was 8-fold lower than that of linezolid when read at 80% inhibition (0.12 μg/mL vs 1 μg/mL, respectively) and 4-fold lower when read at 100% inhibition (0.25 μg/mL vs 1 μg/mL, respectively)
- All *S. anginosus* group isolates were susceptible to tedizolid and linezolid

Table 2. Activity of Tedizolid and Comparators Against Viridans Group Streptococci Isolated in 2014 From the United States and Europe (N=370)

Pathogen (N)	Drug	Range	MIC_{50} μg/mL	MIC_{90} μg/mL	%S/I/R (EUCAST)	%S/I/R (CLSI)
Viridans Group Streptococci[a] (370)	Tedizolid 80	≤0.008–0.25	0.12	0.12	–/–/–	–/–/–
	Tedizolid 100	≤0.008–0.25	0.12	0.25	–/–/–	–/–/–
	Linezolid 80	0.12–1	0.5	1	–/–/–	–/–/–
	Linezolid 100	≤0.12–2	1	1	–/–/–	–/–/–
	Ceftaroline	≤0.015–4	0.03	0.06	–/–/–	–/–/–
	Ceftriaxone	≤0.06–>8	0.25	1	88.9/0.0/11.1	93.5/2.4/4.1
	Clindamycin	≤0.25–>2	≤0.25	>2	85.7/0.0/14.3	84.6/1.1/14.3
	Daptomycin	≤0.06–1	0.25	0.5	–/–/–	100/–/–
	Erythromycin	≤0.12–>16	≤0.12	>16	–/–/–	52.2/3.2/44.6
	Levofloxacin	≤0.12–>4	1	2	–/–/–	94.1/1.0/4.9
	Penicillin	≤0.06–>0.12	≤0.06	>0.12	83.2/12.7/4.1	77.0/18.9/4.1
	Tetracycline	≤0.5–>8	≤0.5	>8	–/–/–	63.8/3.0/33.2
	Vancomycin	≤0.12–1	0.5	0.5	100.0/0.0/0.0	100.0/–/–
S.anginosus (66)	Tedizolid 80	≤0.06-0.12	0.06	0.12	–/–/–	100.0/–/–[b]
	Tedizolid 100	≤ 0.06-0.25	0.12	0.25	–/–/–	100.0/–/–[b]
	Linezolid 80	0.5-1	1	1	–/–/–	100.0/–/–
	Linezolid 100	0.5-1	1	1	–/–/–	–/–/–
	Ceftaroline	≤0.015-0.06	0.03	0.03	–/–/–	–/–/–
	Ceftriaxone	≤0.06-0.5	0.25	0.5	100.0/0.0/0.0	100.0/0.0/0.0
	Clindamycin	≤0.25->2	≤0.25	≤0.25	92.3/0.0/7.7	92.3/0.0/7.7
	Daptomycin	0.12-0.5	0.25	0.5	–/–/–	100.0/–/–
	Erythromycin	≤0.12->16	≤0.12	4	–/–/–	76.9/0.0/23.1
	Levofloxacin	≤0.12-1	0.5	1	–/–/–	100.0/0.0/0.0
	Penicillin	≤0.06	≤0.06	≤0.06	100.0/0.0/0.0	100.0/0.0/0.0
	Tetracycline	≤0.5->8	4	>8	–/–/–	46.2/7.6/46.2
	Vancomycin	0.5	0.5	0.5	100.0/0.0/0.0	100.0/–/–
S. constellatus (37)	Tedizolid 80	0.03–0.12	0.06	0.12	–/–/–	100.0/–/–[b]
	Tedizolid 100	0.03–0.25	0.12	0.12	–/–/–	100.0/–/–[b]
	Linezolid 80	0.25–1	0.5	1	–/–/–	100.0/–/–
	Linezolid 100	0.25–2	1	1	–/–/–	–/–/–
	Ceftaroline	≤0.015–0.06	0.03	0.06	–/–/–	–/–/–
	Ceftriaxone	≤0.06-0.5	0.5	0.5	100.0/0.0/0.0	100.0/0.0/0.0
	Clindamycin	≤0.25–>2	≤0.25	>2	83.8/0.0/16.2	83.8/0.0/16.2
	Daptomycin	≤0.06–0.5	0.25	0.5	–/–/–	100.0/–/–
	Erythromycin	≤0.12–>16	≤0.12	>16	–/–/–	81.1/0.0/18.9
	Levofloxacin	≤0.12–0.5	0.5	0.5	–/–/–	100.0/0.0/0.0
	Penicillin	≤0.06–>0.12	≤0.06	0.12	100.0/0.0/0.0	97.3/2.7/0.0
	Tetracycline	≤0.5–>8	≤0.5	>8	–/–/–	73.0/0.0/27.0
	Vancomycin	0.25–1	0.5	1	100.0/0.0/0.0	100.0/–/–

CLSI, Clinical and Laboratory Standards Institute; EUCAST, European Committee on Antimicrobial Susceptibility Testing; FDA, US Food and Drug Administration; I, intermediate; MIC_{50}, minimum inhibitory concentration required to inhibit growth of 50% of isolates; MIC_{90}, minimum inhibitory concentration required to inhibit growth of 90% of isolates; R, resistant; S, susceptible; –, no breakpoint available for interpretation.
[a]Viridans Group Streptococci spp include *S. australis* (3), *S. canis* (5), *S. constellatus* (37), *S. cristatus* (6), *S. gallolyticus* (11), *S. gordonii* (14), *S. lutetiensis* (2), *S. anginosus* group (13), *S. mitis* group (24), *S. mitis/oralis* (53), *S. mutans* (3), *S. parasanguinis* (13), *S. salivarius* group (11), *S. salivarius* (14), *S. sobrinus* (1), *S. suis* (1), *S. anginosus* (66), *S. bovis* group (3), *S. infantis* (1), *S. intermedius* (10), *S. mitis* (6), *S. oralis* (57), *S. sanguinis* (14), *S. vestibularis* (2).
[b]FDA-approved breakpoint applied.

Activity of Tedizolid and Linezolid Against Enterococci

- Tedizolid had potent activity against all enterococci, including *E. faecalis* and *E. faecium*, with MIC_{90} of 0.25 μg/mL for tedizolid 80 and 0.5 μg/mL for tedizolid 100 (**Table 3**)
- Based on MIC_{90} values, the potency of tedizolid was 4-fold higher than that of linezolid when read at both 80% and 100%
- Tedizolid was equally active against vancomycin-nonsusceptible enterococci and vancomycin-susceptible enterococci with MIC_{90} of 0.25 μg/mL (tedizolid 80) and 0.5 μg/mL (tedizolid 100) for each (data not shown)
- Of 1290 enterococcal isolates, 1 (0.08%) and 23 (1.78%) isolates had a tedizolid MIC ≥1 μg/mL at 80% and 100% reads, respectively. A linezolid MIC ≥8 μg/mL was reported for 1 (0.08%) and 11 (0.85%) isolates at 80% and 100% reads, respectively
 - Of the 23 isolates with a tedizolid MIC ≥1 μg/mL, 19 were *E. faecium* and 4 were *E. faecalis*
 - Of the 11 isolates with a linezolid MIC ≥8 μg/mL, 7 were *E. faecium* and 4 were *E. faecalis*

Table 3. Activity of Tedizolid and Comparators Against Enterococci Isolated in 2014 From the United States and Europe

Pathogen (N)	Drug	Range	MIC_{50} μg/mL	MIC_{90} μg/mL	%S/I/R (EUCAST)	%S/I/R (CLSI)
Enterococci spp[a] (1290)	Tedizolid 80	0.03 –1	0.12	0.25	–/–/–	–/–/–
	Tedizolid 100	0.03–>1	0.25	0.5	–/–/–	–/–/–
	Linezolid 80	≤0.12 –8	1	1	99.9/0.0/0.1	99.6/0.3/0.1
	Linezolid 100	0.25–>8	2	2	–/–/–	–/–/–
	Ampicillin	≤0.25–>8	1	>8	67.3/0.3/32.4	67.6/0.0/32.4
	Daptomycin	≤0.06 –4	1	2	–/–/–	100.0/–/–
	Levofloxacin	≤0.12–>4	>4	>4	49.6/0.0/50.4	48.4/1.2/50.4
	Teicoplanin	≤2–>16	≤2	>16	85.3/0.0/14.7	85.7/1.6/12.7
	Tetracycline	≤0.5->8	>8	>8	–/–/–	27.6/1.2/71.2
	Vancomycin	≤0.12–>16	1	>16	83.4/0.0/16.6	83.4/0.6/16.0
E. faecalis (815)	Tedizolid 80	0.06–1	0.12	0.25	–/–/–	–/–/–
	Tedizolid 100	0.06–1	0.25	0.25	–/–/–	99.5/–/–[b]
	Linezolid 80	≤0.12–8	1	1	99.9/0.0/0.1	99.6/0.3/0.1
	Linezolid 100	0.5– >8	2	2	–/–/–	–/–/–
	Ampicillin	≤0.25–8	1	2	99.9/0.1/0.0	100.0/0.0/0.0
	Daptomycin	≤0.06–4	1	2	–/–/–	100.0/–/–
	Levofloxacin	0.25–>4	1	>4	70.3/0.0/29.7	69.8/0.5/29.7
	Teicoplanin	≤ 2–> 16	≤2	≤2	98.7/0.0/1.3	98.8/0.0/1.2
	Tetracycline	≤0.5–>8	>8	>8	–/–/–	23.0/0.3/76.7
	Vancomycin	0.25–>16	1	2	98.7/0.0/1.3	98.7/0.2/1.1
E. faecium (446)	Tedizolid 80	0.03–0.5	0.12	0.25	–/–/–	–/–/–
	Tedizolid 100	0.06–>1	0.25	0.5	–/–/–	–/–/–
	Linezolid 80	0.25–4	1	1	100.0/0.0/0.0	99.6/0.4/0.0
	Linezolid 100	0.25–>8	2	2	–/–/–	–/–/–
	Ampicillin	≤0.25–>8	>8	>8	6.1/0.6/93.3	6.7/0.0/93.3
	Daptomycin	0.12–4	2	4	–/–/–	100.0/–/–
	Levofloxacin	0.5–>4	>4	>4	9.6/0.0/90.4	7.4/2.2/90.4
	Teicoplanin	≤2–>16	≤2	>16	59.9/0.0/40.1	60.8/4.7/34.5
	Tetracycline	≤0.5–>8	>8	>8	–/–/–	33.6/2.5/63.9
	Vancomycin	0.25–>16	1	>16	55.2/0.0/44.8	55.2/0.4/44.4
VNS *E. faecalis* (11)	Tedizolid 80	0.12 – 0.25	0.12	0.25	–/–/–	100.0/–/–[b]
	Tedizolid 100	0.12 – 0.5	0.25	0.25	–/–/–	–/–/–
	Linezolid 80	0.5–1	1	1	100.0/0.0/0.0	100.0/0.0/0.0
	Linezolid 100	1-2	1	2	–/–/–	–/–/–
	Ampicillin	1-4	1	2	100.0/0.0/0.0	100.0/0.0/0.0
	Daptomycin	0.5–2	1	2	–/–/–	100.0/0.0/0.0
	Levofloxacin	2–>4	>4	>4	9.1/0.0/90.9	9.1/0.0/90.9
	Teicoplanin	≤2->16	>16	>16	18.2/0.0/81.8	18.2/0.0/81.8
	Tetracycline	>8	>8	>8	–/–/–	0.0/0.0/100.0
	Vancomycin	8–>16	>16	>16	0.0/0.0/100.0	0.0/18.2/81.8
VNS *E. faecium* (200)	Tedizolid 80	0.06 – 0.5	0.12	0.25	–/–/–	–/–/–
	Tedizolid 100	0.12 –>1	0.25	0.5	–/–/–	–/–/–
	Linezolid 80	0.5–4	1	1	100.0/0.0/0.0	99.0/1.0/0.0
	Linezolid 100	0.5–>8	1	2	–/–/–	–/–/–
	Ampicillin	>8	>8	>8	0.0/0.0/100.0	0.0/0.0/100.0
	Daptomycin	0.12–4	2	2	–/–/–	100.0/–/–
	Levofloxacin	>4	>4	>4	0.0/0.0/100.0	0.0/0.0/100.0
	Teicoplanin	≤2–>16	>16	>16	10.5/0.0/89.5	12.5/10.5/77.0
	Tetracycline	≤0.5–>8	>8	>8	–/–/–	21.0/4.0/75.0
	Vancomycin	16–>16	>16	>16	0.0/0.0/100.0	0.0/1.0/99.0

CLSI, Clinical and Laboratory Standards Institute; EUCAST, European Committee on Antimicrobial Susceptibility Testing; FDA, US Food and Drug Administration; I, intermediate; MIC_{50}, minimum inhibitory concentration required to inhibit growth of 50% of isolates; MIC_{90}, minimum inhibitory concentration required to inhibit growth of 90% of isolates; R, resistant; S, susceptible; VNS, vancomycin-nonsusceptible; –, no breakpoint available for interpretation.
[a]Enterococci spp include *E. avium* (2), *E. casseliflavus* (6), *E. cecorum* (1), *E. durans* (1), *E. faecalis* (815), *E. faecium* (446), *E. gallinarum* (14), *E. hirae* (3), *E. raffinosus* (2).
[b]FDA-approved breakpoint applied.

Comparison of Tedizolid and Linezolid Activity Against BHS, Viridans Group Streptococci, and Enterococci

- The activity of tedizolid was similar against isolates of BHS, viridans group streptococci, and enterococci
 - At 80% read, >95% of BHS, viridans group streptococci, and enterococci were inhibited at an MIC of 0.25 μg/mL
 - At 100% read, >95% of BHS and viridans group streptococci were inhibited at an MIC of 0.25 μg/mL and >98% of enterococci were inhibited at an MIC of 0.5 μg/mL
- For all 3 organism groups, tedizolid was at least 4-fold more potent than linezolid with MIC_{90} of 0.12 to 0.25 μg/mL versus 1 μg/mL, respectively, at 80% read and MIC_{90} of 0.5 μg/mL versus 2 μg/mL, respectively, at 100% read

Figure 1. Tedizolid and linezolid MIC distributions (at 80% and 100% reads) for all BHS, viridans group streptococci, and enterococci isolated from the United States and Europe, 2014.

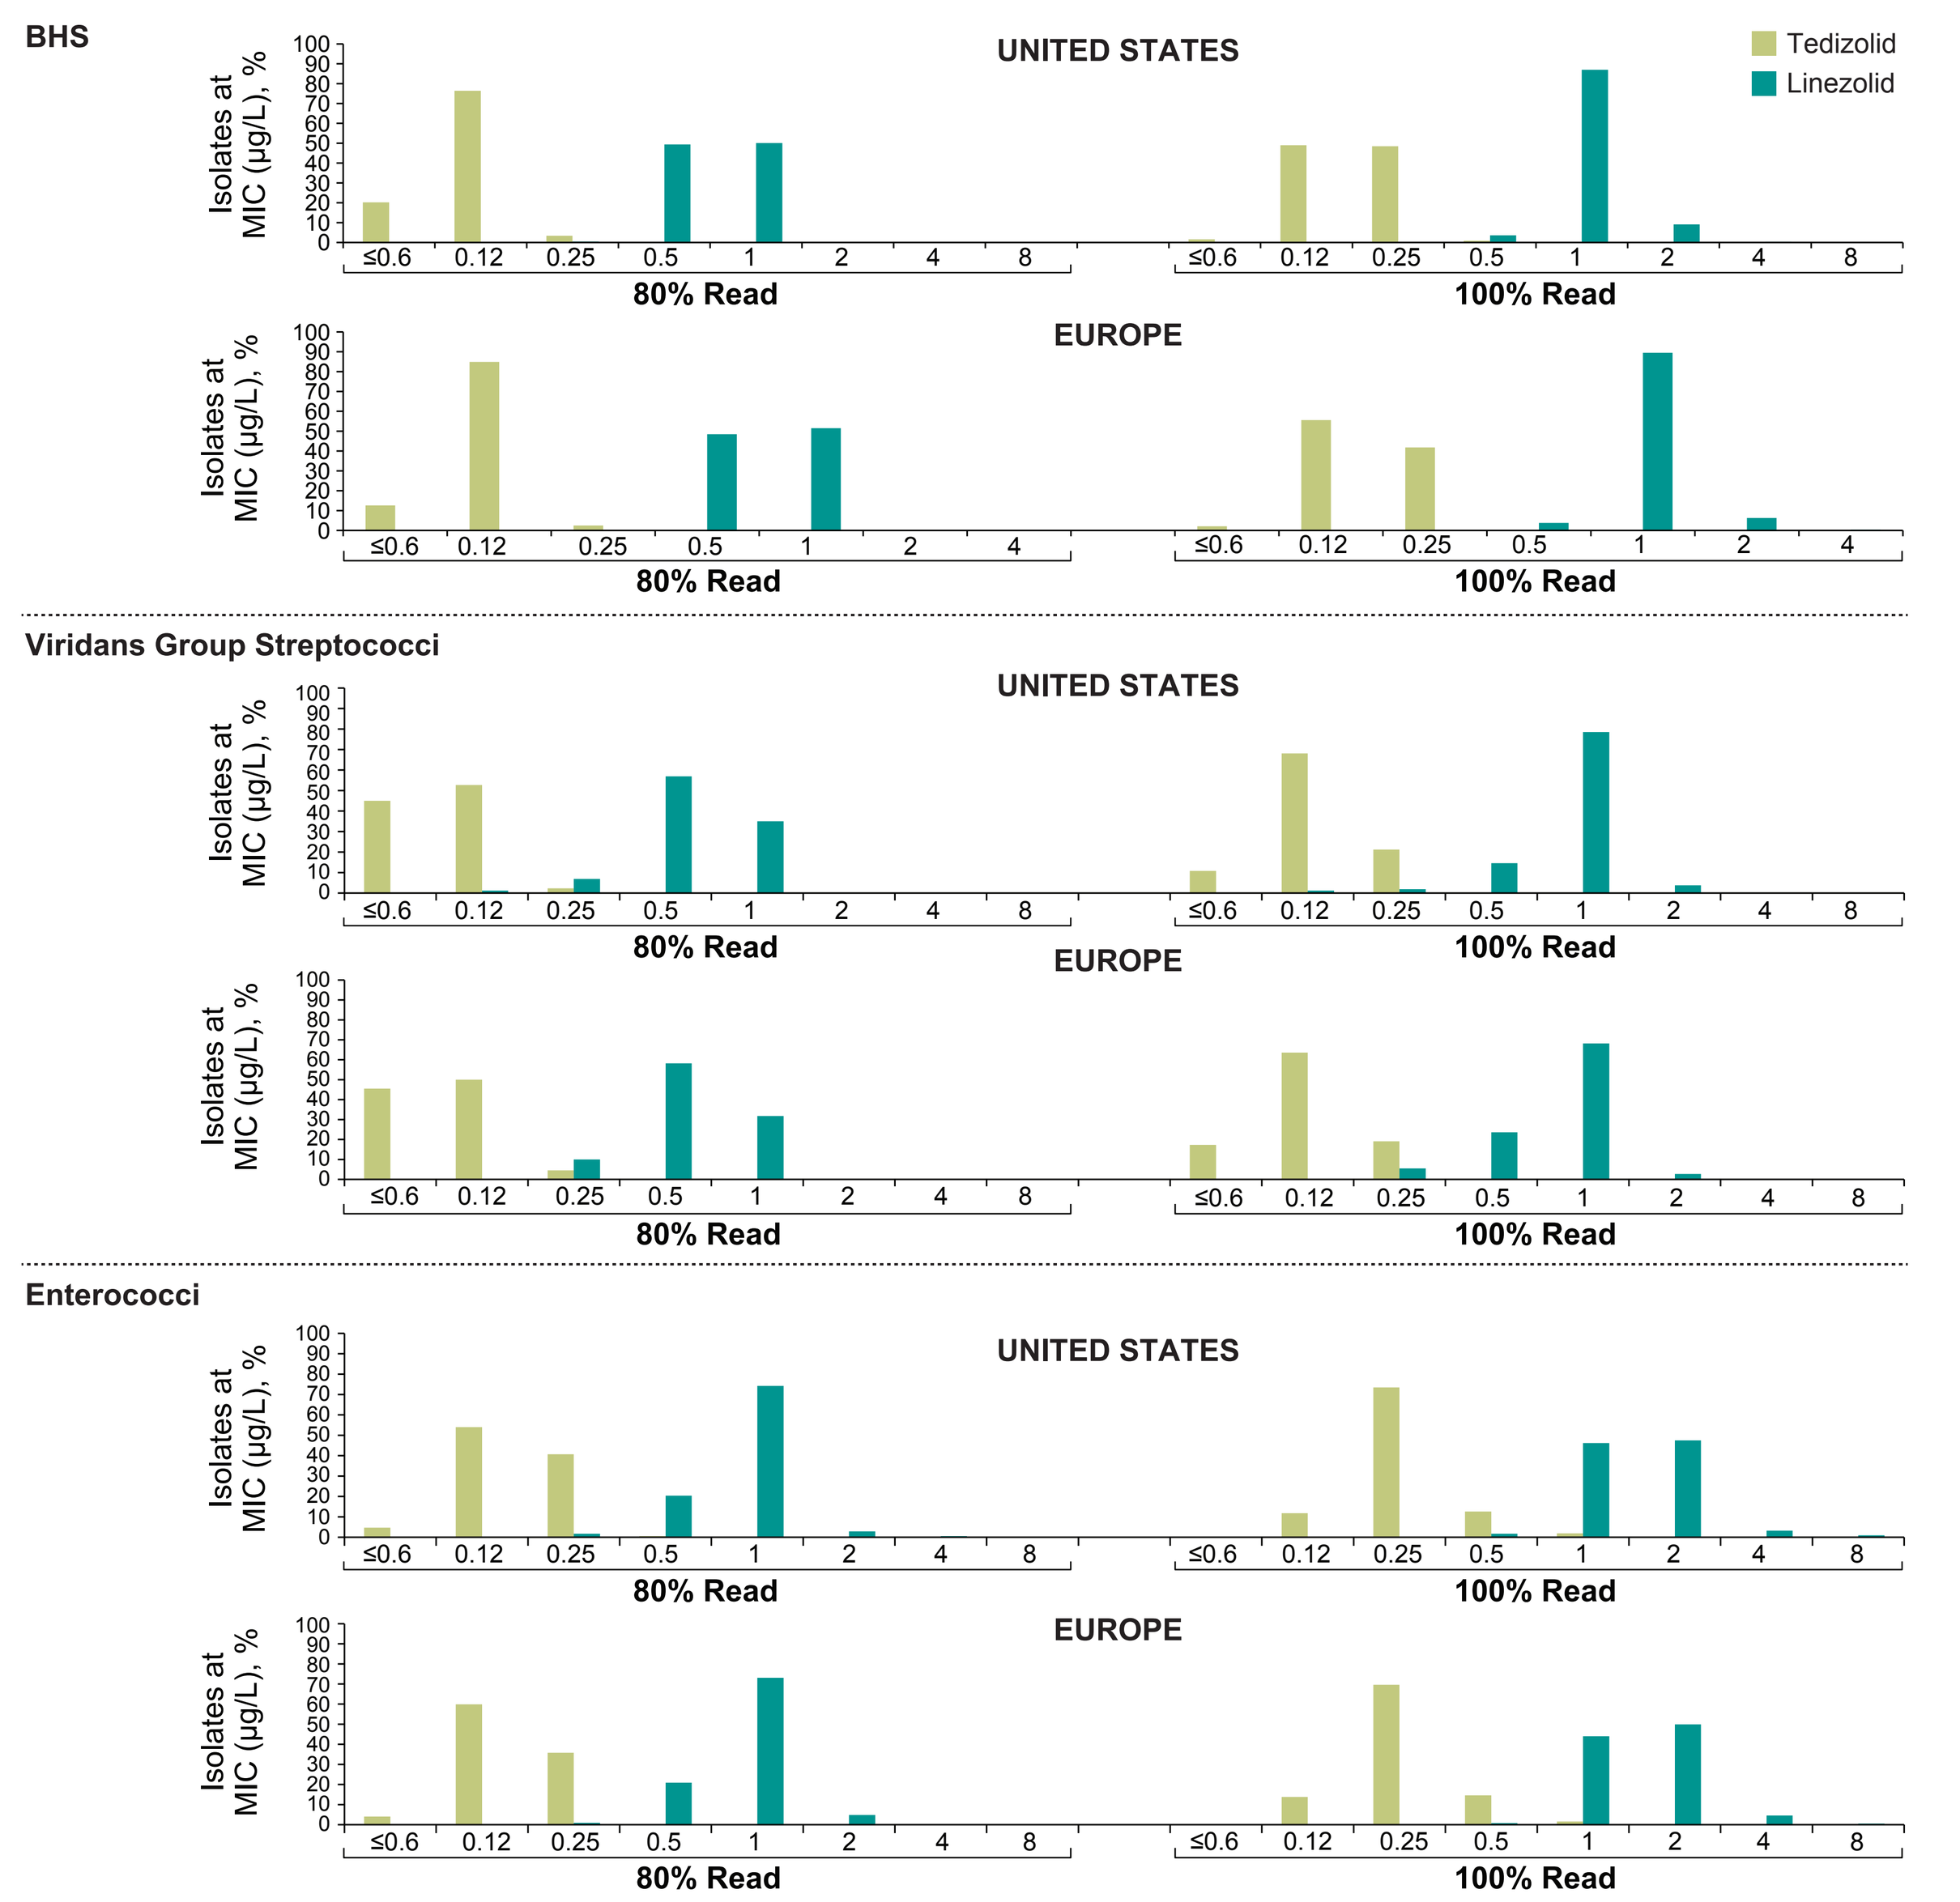
BHS
Viridans Group Streptococci
Enterococci
UNITED STATES
EUROPE
Tedizolid
Linezolid
Isolates at MIC (μg/L), %
≤0.6
0.12
0.25
0.5
1
2
4
8
80% Read
100% Read

BHS, β-hemolytic streptococci.

Conclusions

- Tedizolid demonstrated potent activity against recent clinical isolates of BHS, viridans group streptococci, and enterococci, including vancomycin-nonsusceptible enterococci, isolated from the United States and Europe
- Tedizolid demonstrated 4- to 8-fold more potent *in vitro* activity than linezolid against streptococcal and enterococcal isolates, regardless of whether the MIC was read at 80% or 100%
- All BHS and viridans group streptococci isolates had tedizolid MIC values of ≤0.5 and 0.25 μg/mL, respectively
- All *S. pyogenes, S. agalactiae*, and *S.anginosus* were susceptible to tedizolid
- Four of 815 *E. faecalis* isolates were resistant to tedizolid when the MICs were read at 100%

Acknowledgments

Funding for this research was provided by Merck & Co., Inc., Kenilworth, NJ, USA. Medical writing and editorial assistance were provided by Norma Padilla, PhD, of Apothecom, Yardley, PA, USA. This assistance was funded by Merck & Co., Inc., Kenilworth, NJ, USA.

References

1. Moet GJ et al. *Diag Microbiol Infect Dis.* 2007;57:7-13.
2. Fritsche TR et al. *Diag Microbiol Infect Dis.* 2007;58:19-26.
3. Werner G et al. *Euro Surveill.* 2008;13:1-11.
4. SIVEXTRO® (tedizolid phosphate) [prescribing information]. Kenilworth, NJ: Merck & Co., Inc.; 2015.
5. SIVEXTRO® (tedizolid phosphate) [summary of product characteristics]. Weybridge, Surrey, UK: Cubist (UK) Ltd.; 2015.
6. SIVEXTRO (tedizolid phosphate). Health Canada website www.hc-sc.gc.ca/dhp-mps/prodpharma/sbd-smd/drug-med/sbd_smd_2015_sivextro_173603-eng.php. Accessed August 26, 2015.
7. Brown SD et al. *Antimicrob Agents Chemother.* 2010;54:2063-2069.
8. Schaadt RD et al. *Antimicrob Agents Chemother.* 2009;53:3236-3239.
9. Thomson KS et al. *Antimicrob Agents Chemother.* 2013;57:2892-2895.
10. Prokocimer P et al. *JAMA.* 2013;309:559-569.
11. Moran GJ et al. *Lancet Infect Dis.* 2014;14:696-705.
12. Shorr AF et al. *Antimicrob Agents Chemother.* 2015;59:864-871.
13. Clinical and Laboratory Standards Institute. M07-A10. *Methods for Dilution Antimicrobial Susceptibility Tests for Bacteria that Grow Aerobically*; Approved standard. Wayne, PA: CLSI; 2015.
14. Clinical and Laboratory Standards Institute. M100-S25. Performance standards for antimicrobial susceptibility testing: 25th informational supplement. Wayne, PA: CLSI; 2015.
15. Clinical and Laboratory Standards Institute. 2012. Summary minutes of the subcommittee on antimicrobial susceptibility testing, 22-24 January 2012.
16. The European Committee on Antimicrobial Susceptibility Testing. Breakpoint tables for interpretation of MICs and zone diameters. Version 5.0, 2015. www.eucast.org/clinical_breakpoints. Accessed March 30, 2015.

http://bit.ly/1iqAX72

Copyright © 2015 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.