Poster # 247

Antimicrobial Activity of Ceftolozane/Tazobactam Tested Against Gram-negative Bacterial Isolates From Hospitalized Patients With Pneumonia in United States Hospitals (2013)

D.J. Farrell, H.S. Sader, R.K. Flamm, R.N. Jones

JMI Laboratories, North Liberty, IA, USA

David J. Farrell, Ph.D., D(ABMM)
JMI Laboratories
345 Beaver Kreek Ctr, Ste A
North Liberty, IA, 52317
Tel: 319-665-3370
E-mail: david-farrell@jmilabs.com

ABSTRACT

BACKGROUND: Ceftolozane/tazobactam (TOL/TAZ) is a novel antibacterial with activity against *Pseudomonas aeruginosa* (PSA) and other common Gram-negative (GN) pathogens. TOL/TAZ is currently under clinical development for the treatment of nosocomial pneumonia, complicated intra-abdominal infections, and complicated urinary tract infections (cUTIs). The in vitro activity of TOL/TAZ was tested against GN pathogens in patients hospitalized with pneumonia in USA hospitals.

METHODS: 1438 isolates were consecutively collected in 29 USA hospitals from patients with pneumonia in 2013. Susceptibility (S) testing was performed by CLSI broth microdilution methods (TOL/TAZ at a fixed 4 µg/mL of TAZ).

RESULTS: PSA was the most common pathogen (40.4%) and TOL/TAZ was the most active β-lactam tested against PSA (97.6% inhibited at ≤8 µg/mL). PSA exhibited moderate S to meropenem (MEM, 78.1%), ceftazidime (CAZ; 83.0%), cefepime (FEP, 81.2%), piperacillin/TAZ (PIP/TAZ; 75.7%), levofloxacin (LVX; 72.6%), and gentamicin (GEN; 86.0%). TOL/TAZ exhibited activity against CAZ-non-S, MEM-non-S PSA, and MDR PSA isolates (Table). TOL/TAZ was active against *Klebsiella pneumoniae* (KPN; $MIC_{50/90}$, 0.5/>32 µg/mL) but activity was lower ($MIC_{50/90}$, 32/>32 µg/mL) against ESBL-phenotype KPN (31.2%); similar to all β-lactams (including MER [32.2% S]) and LEV (18.6% S) and GEN (57.6% S). TOL/TAZ inhibited 84.2% of MEM-S-ESBL-KPN at ≤8 µg/mL. TOL/TAZ was active against *Escherichia coli* (MIC_{90}, 0.5 µg/mL), including ESBL-phenotype isolates (MIC_{90}, 1 µg/mL). TOL/TAZ inhibited 93.4 and 96.2% *Enterobacter* spp. (ESP) and *Serratia* spp., respectively, at ≤8 µg/mL, and demonstrated activity against CAZ-non-S ESP (70.3% inhibited at ≤8 µg/mL). TOL/TAZ was active against *Proteus mirabilis* (MIC_{90}, 0.5 µg/mL), *Citrobacter* spp. (MIC_{90}, 4 µg/mL), and indole (+) *Proteae* (MIC_{90}, 1 µg/mL). All β-lactams had limited activity against *Acinetobacter* spp.

Organism (No. Tested)	No. of Isolates (Cumulative %) Inhibited at TOL/TAZ MIC (µg/mL)								
	≤0.5	1	2	4	8	16	32	>32	$MIC_{50/90}$
P. aeruginosa (581)	300 (51.6)	170 (80.9)	56 (90.5)	26 (95.0)	15 (97.6)	5 (98.5)	1 (98.6)	8 (100.0)	0.5/2
CAZ-non-S (99)	4 (4.0)	12 (16.2)	34 (50.5)	21 (71.7)	14 (85.9)	5 (90.9)	1 (91.9)	8 (100.0)	2/16
MEM-non-S (127)	32 (25.2)	46 (61.4)	14 (72.4)	16 (85.0)	8 (91.3)	4 (94.5)	0 (94.5)	7 (100.0)	1/8
MDR (94)	8 (8.5)	23 (33.0)	26 (60.6)	16 (77.7)	8 (86.2)	4 (90.4)	1 (91.5)	8 (100.0)	2/16
K. pneumoniae (189)	128 (67.7)	13 (74.6)	4 (76.7)	1 (77.3)	2 (78.3)	7 (82.0)	11 (87.8)	23 (100.0)	0.5/>32
ESBL-phenotype (59)	8 (13.6)	4 (20.3)	3 (25.4)	1 (27.1)	2 (30.5)	7 (42.4)	11 (61.0)	23 (100.0)	32/>32
MEM-S-ESBL (19)	8 (42.1)	4 (63.2)	2 (73.7)	1 (79.0)	1 (84.2)	1 (89.5)	0 (89.5)	2 (100.0)	1/>32
Enterobacter spp. (167)	124 (74.3)	6 (77.8)	12 (85.0)	8 (89.8)	6 (93.4)	5 (96.4)	3 (98.2)	3 (100.0)	0.25/8
Serratia spp. (156)	98 (62.8)	37 (86.5)	9 (92.3)	5 (95.5)	1 (96.2)	0 (96.2)	1 (96.8)	5 (100.0)	0.5/2
E. coli (134)	129 (96.3)	3 (98.5)	1 (99.3)	0 (99.3)	1 (100.0)	-	-	-	0.25/0.5
ESBL-phenotype (20)	17 (85.0)	1 (90.0)	1 (95.0)	0 (95.0)	1 (100.0)	-	-	-	0.25/1

CONCLUSIONS: In GN isolates from hospitalized patients with pneumoniae in USA hospitals, TOL/TAZ demonstrated greater in vitro activity than currently available cephalosporins, carbapenems, and PIP/TAZ when tested against PSA, including MDR strains. Additionally, TOL/TAZ demonstrated greater activity than currently available cephalosporins and PIP/TAZ against Enterobacteriaceae from pneumonia specimens.

INTRODUCTION

- Ceftolozane/tazobactam is an antibacterial consisting of ceftolozane, a novel antipseudomonal cephalosporin, with tazobactam, a well-established β-lactamase inhibitor.
- Ceftolozane exerts its bactericidal activity by inhibiting essential penicillin-binding proteins, resulting in inhibition of cell-wall synthesis and subsequent cell death. Ceftolozane has demonstrated greater activity against *Pseudomonas aeruginosa* when directly compared with ceftazidime and cefepime.
- Tazobactam is a potent inhibitor of most common Class A and some Class C β-lactamases that protects ceftolozane from hydrolysis, by binding to the active site of these enzymes, and broadens coverage to include most extended-spectrum β-lactamase (ESBL)–producing Enterobacteriaceae and some AmpC-derepressed Enterobacteriaceae.
- In clinical trials, ceftolozane/tazobactam demonstrated superior clinical efficacy to high-dose levofloxacin for the treatment of patients with complicated lower urinary tract infection/pyelonephritis. Ceftolozane/tazobactam plus metronidazole was as efficacious as meropenem in patients with complicated intra-abdominal infection (cIAI).

INTRODUCTION (cont'd)

- Gram-negative bacilli are the major cause of pneumonia in hospitalized patients and, with increasing antimicrobial resistance in these pathogens and empirical therapy for these infections becoming increasingly difficult, development of new therapeutic options is highly imperative.
- Phase 3 trials to assess the efficacy and safety of ceftolozane/tazobactam versus meropenem in the treatment of ventilated nosocomial pneumonia are ongoing. In the present study, we evaluated the in vitro activity of ceftolozane/tazobactam against Gram-negative pathogens isolated from patients hospitalized with pneumonia in 29 USA hospitals in 2013.

MATERIALS AND METHODS

Organism collection: The organism collection included only Gram-negative bacilli collected from hospitalized patients with a diagnosis of pneumonia. In 2013, a total of 1438 unique patient organisms were consecutively collected from 29 USA medical centers. Species identification was performed at the participant medical center and confirmed at the monitoring laboratory (JMI Laboratories, North Liberty, IA, USA) using the VITEK 2 System (bioMerieux, Hazelwood, MO, USA) or MALDI-TOF (Bruker Daltonics Inc., Billerica, MA, USA), when necessary. Only 1 strain per patient infection episode was included in this surveillance study.

Susceptibility testing: Isolates were tested for susceptibility to multiple antimicrobial agents at a reference laboratory (JMI Laboratories) by standardized broth microdilution methods as described by the Clinical and Laboratory Standards Institute (CLSI) M07-A9 document. Minimum inhibitory concentration (MIC) results were interpreted according to CLSI criteria in M100-S24 (2014), as well as European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoint tables (version 4.0, January 2014). *Escherichia coli* and *Klebsiella* spp. isolates with MIC of ≥2 µg/mL for ceftazidime or ceftriaxone or aztreonam were categorized as ESBL-phenotype. To better evaluate the activities of ceftolozane/tazobactam against *P. aeruginosa*, strains were stratified by susceptibility pattern to ceftazidime and meropenem. Multidrug-resistant (MDR), extensively drug-resistant (XDR), and pandrug-resistant (PDR) bacteria were classified as such as per recently recommended guidelines by Magiorakos et al. (2012), using antimicrobial class representative agents and CLSI susceptibility MIC breakpoints. Classifications were based on the following recommended parameters: MDR = non-susceptible to ≥3 antimicrobial classes; XDR = susceptible to ≤2 antimicrobial classes; PDR = non-susceptible to all antimicrobial classes. Quality control (QC) strains included: *E. coli* ATCC 25922 and 35218 and *P. aeruginosa* ATCC 27853. QC ranges and interpretive criteria for comparator compounds used the CLSI M100-S24 guidelines and all QC results were within published ranges.

RESULTS

- *P. aeruginosa* was the most common pathogen (40.4%) and ceftolozane/tazobactam was the most active β-lactam tested against *P. aeruginosa* (MIC required to inhibit the growth of 50%/90% of organisms [$MIC_{50/90}$], 0.5/2 µg/mL; 97.6% inhibited at ≤8 µg/mL; **Tables 1** to **3**). *P. aeruginosa* was moderately susceptible to ceftazidime (83.0%), cefepime (81.2%), meropenem (78.1%), piperacillin/tazobactam (75.7%), levofloxacin (72.6%), and gentamicin (86.0%). Most isolates were susceptible to amikacin (95.2%) and colistin (99.8%; **Table 3**).
- Ceftolozane/tazobactam showed activity against ceftazidime-non-susceptible (85.9% inhibited at ≤8 µg/mL), cefepime-non-susceptible (88.1% inhibited at ≤8 µg/mL), meropenem-non-susceptible *P. aeruginosa* (91.3% inhibited at ≤8 µg/mL), and isolates non-susceptible to meropenem + ceftazidime + piperacillin/tazobactam (78.3% inhibited at ≤8 µg/mL), and other antimicrobial agents (**Tables 1, 2; Figure 1**). Ceftolozane/tazobactam inhibited 86.2% of MDR (16.2% of all *P. aeruginosa* isolates) and 77.1% of XDR (8.3% of all *P. aeruginosa* isolates) *P. aeruginosa* isolates at MICs of ≤8 µg/mL (**Table 1**). No PDR strains of *P. aeruginosa* were found.
- Ceftolozane/tazobactam was very active ($MIC_{50/90}$, 0.5/4 µg/mL; 90.6/92.3% inhibited at ≤4/≤8 µg/mL) against 776 Enterobacteriaceae and retained activity against many MDR (13.3% of all Enterobacteriaceae isolates) and XDR (3.6% of all Enterobacteriaceae isolates), inhibiting 49.5% of MDR isolates and 17.9% of XDR isolates at MIC values of ≤8 µg/mL (**Table 1**).
- Ceftolozane/tazobactam was active against *E. coli* ($MIC_{50/90}$, 0.25/0.5 µg/mL; 99.3/100.0% inhibited at ≤4/≤8 µg/mL), including ESBL-phenotype isolates ($MIC_{50/90}$, 0.25/1 µg/mL; 95.0/100.0% inhibited at ≤4/≤8 µg/mL; **Table 1**). All *E. coli* isolates were susceptible to meropenem, tigecycline, and colistin (EUCAST interpretive criteria only available for colistin; **Table 3**). Susceptibility rates (CLSI criteria) were high to modest for other agents: ceftazidime (92.5%), piperacillin/tazobactam (92.4%), cefepime (88.8%), ceftriaxone (85.1%), gentamicin (85.7%), and levofloxacin (58.2%; **Table 3**).
- Ceftolozane/tazobactam ($MIC_{50/90}$, 0.25/32 µg/mL) inhibited 82.7% of *Klebsiella* spp., including ESBL producers, at MICs ≤8 µg/mL. Among ESBL-phenotype *Klebsiella* spp., whereas only 36.4/% were inhibited at ≤8 µg/mL, 88.0% of meropenem-susceptible, ESBL-phenotype *Klebsiella* spp. strains were inhibited at ≤8 µg/mL of ceftolozane/tazobactam (**Tables 1** and **2**).
- Against *K. pneumoniae*, including ESBL, 78.3% of isolates were inhibited at ceftolozane/tazobactam MICs ≤8 µg/mL. 96.7% of isolates were susceptible (EUCAST criteria) to colistin and 100.0% to tigecycline (CLSI criteria); resistance rates by CLSI criteria were as follows: for ceftazidime (27.0%), cefepime (23.8%), ceftriaxone (29.1%), piperacillin/tazobactam (24.3%), meropenem (20.1%), levofloxacin (24.3%), and gentamicin (10.6%; **Table 3**).
- When tested against *Enterobacter* spp., ceftolozane/tazobactam ($MIC_{50/90}$, 0.25/8 µg/mL; 93.4% inhibited at ≤8 µg/mL) showed greater activity than ceftazidime ($MIC_{50/90}$, 0.25/>32 µg/mL; 77.8% susceptible [CLSI]) and piperacillin/tazobactam ($MIC_{50/90}$, 4/64 µg/mL; 80.6% susceptible [CLSI]; **Table 3**), and demonstrated activity against ceftazidime-non-susceptible strains (70.3% inhibited at ≤8 µg/mL; **Table 2**).
- Ceftolozane/tazobactam was active against *Serratia marcescens* ($MIC_{50/90}$, 0.5/2 µg/mL; 96.2% inhibited at ≤8 µg/mL), *Proteus mirabilis* ($MIC_{50/90}$, 0.5/0.5 µg/mL; 100.0% inhibited at ≤8 µg/mL), *Citrobacter* spp. ($MIC_{50/90}$, 0.25/4 µg/mL; 95.5% inhibited at ≤8 µg/mL), and indole-positive *Proteae* ($MIC_{50/90}$, 0.5/1 µg/mL; 100.0% inhibited at ≤8 µg/mL; **Table 2**).
- All β-lactams had limited activity against *Acinetobacter* spp. (**Table 3**).

RESULTS (cont'd)

Table 1. Summary of Ceftolozane/Tazobactam Activity Tested Against Gram-negative Bacilli by Species and Resistance Phenotype Isolated From Patients Hospitalized With Pneumonia (USA, 2013)

Organism (No. Tested)/ Resistance Phenotype	Number of Isolates (Cumulative %) Inhibited at Ceftolozane/Tazobactam MIC (µg/mL)											MIC (µg/mL)	
	≤0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32	MIC_{50}	MIC_{90}
P. aeruginosa (581)	3 (0.5)	5 (1.4)	22 (5.2)	270 (51.6)	170 (80.9)	56 (90.5)	26 (95.0)	15 (97.6)	5 (98.5)	1 (98.6)	8 (100.0)	0.5	2
CAZ-non-S (99)	0 (0.0)	0 (0.0)	0 (0.0)	4 (4.0)	12 (16.2)	34 (50.5)	21 (71.7)	14 (85.9)	5 (90.9)	1 (91.9)	8 (100.0)	2	16
FEP-non-S (109)	0 (0.0)	0 (0.0)	0 (0.0)	3 (2.8)	19 (20.2)	37 (54.1)	24 (76.2)	13 (88.1)	5 (92.7)	0 (92.7)	8 (100.0)	2	16
MEM-non-S (127)	0 (0.0)	0 (0.0)	0 (0.0)	32 (25.2)	46 (61.4)	14 (72.4)	16 (85.0)	8 (91.3)	4 (94.5)	0 (94.5)	7 (100.0)	1	8
P/T-non-S (141)	0 (0.0)	0 (0.0)	0 (0.0)	11 (7.8)	42 (37.6)	40 (66.0)	22 (81.6)	14 (91.5)	5 (95.0)	0 (95.0)	7 (100.0)	2	8
CAZ & MEM & P/T-non-S (46)	0 (0.0)	0 (0.0)	0 (0.0)	1 (2.2)	3 (8.7)	10 (30.4)	14 (60.9)	8 (78.3)	4 (87.0)	0 (87.0)	6 (100.0)	4	>32
LVX-non-S (159)	0 (0.0)	1 (0.6)	4 (3.1)	35 (25.2)	59 (62.3)	31 (81.8)	12 (89.3)	6 (93.1)	3 (95.0)	1 (95.6)	7 (100.0)	1	8
GEN-non-S (81)	0 (0.0)	0 (0.0)	2 (2.5)	17 (23.5)	20 (48.2)	21 (74.1)	7 (82.7)	5 (88.9)	1 (90.1)	1 (91.4)	7 (100.0)	2	16
MDR (94)	0 (0.0)	0 (0.0)	0 (0.0)	8 (8.5)	23 (33.0)	26 (60.6)	16 (77.7)	8 (86.2)	4 (90.4)	1 (91.5)	8 (100.0)	2	16
XDR (48)	0 (0.0)	0 (0.0)	0 (0.0)	2 (4.2)	4 (12.5)	14 (41.7)	9 (60.4)	8 (77.1)	4 (85.4)	0 (85.4)	7 (100.0)	4	>32
Acinetobacter spp.[a] (81)	5 (6.2)	0 (6.2)	1 (7.4)	4 (12.4)	5 (18.5)	7 (27.2)	4 (32.1)	6 (39.5)	7 (48.2)	9 (59.3)	33 (100.0)	32	>32
All Enterobacteriaceae (776)	2 (0.3)	105 (13.8)	262 (47.6)	220 (75.9)	65 (84.3)	31 (88.3)	18 (90.6)	13 (92.3)	13 (93.9)	16 (96.0)	31 (100.0)	0.5	4
MDR Enterobacteriaceae (103)	0 (0.0)	0 (0.0)	4 (3.9)	14 (17.5)	9 (26.2)	11 (36.9)	10 (46.6)	3 (49.5)	7 (56.3)	14 (69.9)	31 (100.0)	16	>32
XDR Enterobacteriaceae (28)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (3.6)	0 (3.6)	3 (14.3)	1 (17.9)	1 (21.4)	6 (42.9)	16 (100.0)	>32	>32
E. coli (134)	1 (0.8)	46 (35.1)	63 (82.1)	19 (96.3)	3 (98.5)	1 (99.3)	0 (99.3)	1 (100.0)	-	-	-	0.25	0.5
SBL phenotype (20)	0 (0.0)	0 (0.0)	10 (50.0)	7 (85.0)	1 (90.0)	1 (95.0)	0 (95.0)	1 (100.0)	-	-	-	0.25	1
Klebsiella spp.[b] (243)	1 (0.4)	41 (17.3)	88 (53.5)	45 (72.0)	15 (78.2)	8 (81.5)	1 (81.9)	2 (82.7)	7 (85.6)	12 (90.5)	23 (100.0)	0.25	32
ESBL phenotype (66)	0 (0.0)	0 (0.0)	3 (4.6)	7 (15.2)	4 (21.2)	7 (31.8)	1 (33.3)	2 (36.4)	7 (47.0)	12 (65.2)	23 (100.0)	32	>32
ESBL phenotype & MEM-S (25)	0 (0.0)	0 (0.0)	3 (12.0)	7 (40.0)	4 (56.0)	6 (80.0)	1 (84.0)	1 (88.0)	1 (92.0)	0 (92.0)	2 (100.0)	1	16
Enterobacter spp.[c] (167)	0 (0.0)	13 (7.9)	85 (58.7)	26 (74.3)	6 (77.8)	12 (85.0)	8 (89.8)	6 (93.4)	5 (96.4)	3 (98.2)	3 (100.0)	0.25	8
CAZ-non-S (37)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (2.7)	11 (32.4)	8 (54.1)	6 (70.3)	5 (83.8)	3 (91.9)	3 (100.0)	4	32
S. marcescens (156)	0 (0.0)	1 (0.6)	3 (2.6)	94 (62.8)	37 (86.5)	9 (92.3)	5 (95.5)	1 (96.2)	0 (96.2)	1 (96.8)	5 (100.0)	0.5	2
Proteus mirabilis (31)	0 (0.0)	0 (0.0)	5 (16.1)	24 (93.6)	1 (96.8)	1 (100.0)	-	-	-	-	-	0.5	0.5
Citrobacter spp.[d] (22)	0 (0.0)	10 (9.1)	4 (54.6)	0 (54.6)	0 (54.6)	0 (54.6)	4 (90.9)	8 (95.5)	1 (100.0)	-	-	0.25	4
Indole-positive *Proteus* spp.[e] (23)	0 (0.0)	2 (8.7)	8 (43.5)	8 (78.3)	3 (91.3)	0 (91.3)	0 (91.3)	2 (100.0)	-	-	-	0.5	1

CAZ = ceftazidime; non-S = non-susceptible; FEP = cefepime; MEM = meropenem; P/T = piperacillin/tazobactam; LVX = levofloxacin; GEN = gentamicin.
[a]Includes *A. baumannii* (75 strains), *A. nosocomialis* (1 strain), *A. berezinae* (1 strain), *A. junii* (1 strain), *A. ursingii* (1 strain), *A. pittii* (1 strain), *A. soli* (1 strain); [b]Includes *K. pneumoniae* (189 strains), *K. oxytoca* (53), *K. pneumoniae* subsp. *ozaenae* (1); [c]Includes *E. cloacae* (111 strains), *E. aerogenes* (48), *E. asburiae* (5), *E. cancerogenus* (1), *E. gergoviae* (1), *E. amnigenus* (1); [d]Includes *C. freundii* (16), *C. koseri* (5), *C. braakii* (1); [e]Includes *Morganella morganii* (13 strains), *Providencia stuartii* (7), *Providencia rettgeri* (3).

Table 2. Comparative Activity of Ceftolozane/Tazobactam, Ceftazidime, Piperacillin/Tazobactam and Meropenem When Tested Against Bacterial Isolates From Patients Hospitalized With Pneumonia (% Susceptible) (USA, 2013).

Organism	MIC_{50}/MIC_{90} (µg/mL)/%Susceptible (S)[a,b]			
	Ceftolozane/ Tazobactam	Ceftazidime	Piperacillin/ Tazobactam	Meropenem
P. aeruginosa (581)	97.6	83.0	75.7	78.1
E. coli (134)	100	92.5	92.4	100.0
Klebsiella spp. (243)	82.7	77.0	75.3	83.1
Enterobacter spp. (167)	93.4	77.8	80.6	98.8
S. marcescens (156)	96.2	93.6	88.5	97.4
P. mirabilis (31)	100.0	100.0	100.0	100.0
Indole-positive *Proteus* spp. (23)	100.0	95.7	95.7	100.0
Citrobacter spp. (22)	95.5	68.2	77.3	100.0
Acinetobacter spp. (81)	39.5	32.1	27.2	32.1

[a]CLSI (2014) interpretative criteria for ceftazidime (CAZ), piperacillin/tazobactam and meropenem (MEM).
[b]For comparison purposes, % inhibited at ≤8 µg/mL of ceftolozane/tazobactam.

Figure 1. Comparative Activity of Ceftolozane/Tazobactam, Ceftazidime, Piperacillin/Tazobactam, and Meropenem When Tested Against Bacterial Isolates From Patients Hospitalized With Pneumonia (% Non-susceptible) (USA, 2013)

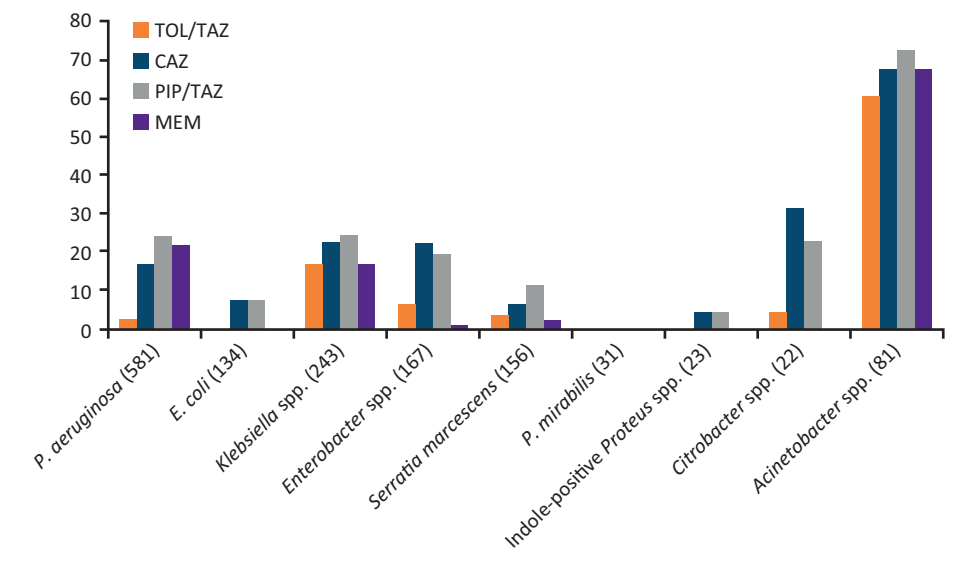
80
70
60
50
40
30
20
10
0
TOL/TAZ
CAZ
PIP/TAZ
MEM
P. aeruginosa (581)
E. coli (134)
Klebsiella spp. (243)
Enterobacter spp. (167)
Serratia marcescens (156)
P. mirabilis (31)
Indole-positive Proteus spp. (23)
Citrobacter spp. (22)
Acinetobacter spp. (81)

For comparison purposes, % resistant at >8 µg/mL of ceftolozane/tazobactam.

Table 3. Activity of Ceftolozane/Tazobactam and comparator Antimicrobial Agents When Tested Against Bacterial Isolates From Patients Hospitalized With Pneumonia (USA, 2013).

Organisms (No. Tested)/Antimicrobial Agent	MIC (µg/mL)		%S/%I/%R	
	MIC_{50}	MIC_{90}	CLSI[a]	EUCAST[a]
P. aeruginosa (581)				
Ceftolozane/tazobactam	0.5	2	(97.6)[b]	
Ceftazidime	2	32	83.0/4.1/12.9	83.0/0.0/17.0
Cefepime	4	16	81.2/9.8/9.0	81.2/0.0/18.8
Meropenem	0.5	8	78.1/7.6/14.3	78.1/14.3/7.6
Piperacillin/tazobactam	8	>64	75.7/11.2/13.1	75.7/0.0/24.3
Levofloxacin	0.5	>4	72.6/8.6/18.8	60.8/11.8/27.4
Gentamicin	2	>8	86.0/3.1/10.9	86.0/0.0/14.0
Amikacin	4	8	95.2/1.4/3.4	91.2/4.0/4.8
Colistin	1	2	99.8/0.0/0.2	99.8/0.0/0.2
E. coli (131)				
Ceftolozane/tazobactam	0.25	0.5	(100.0)[b]	
Ceftazidime	0.25	2	92.5/0.0/7.5	87.3/5.2/7.5
Cefepime	≤0.5	4	88.8/4.5/6.7	86.6/4.4/9.0
Ceftriaxone	≤0.06	>8	85.1/0.0/14.9	85.1/0.0/14.9
Piperacillin/tazobactam	2	16	92.4/4.5/3.1	88.5/3.9/7.6
Meropenem	≤0.06	≤0.06	100.0/0.0/0.0	100.0/0.0/0.0
Levofloxacin	≤0.12	>4	58.2/0.8/41.0	58.2/0.0/41.8
Gentamicin	≤1	>8	85.7/0.0/14.3	85.7/0.0/14.3
Tigecycline[c]	0.12	0.25	100.0/0.0/0.0	100.0/0.0/0.0
Colistin	0.5	0.5	-/-/-	100.0/0.0/0.0
K. pneumoniae (189)				
Ceftolozane/tazobactam	0.5	>32	(78.3)[b]	
Ceftazidime	0.25	>32	70.9/2.1/27.0	69.3/1.6/29.1
Cefepime	≤0.5	>16	72.5/3.7/23.8	71.4/2.1/26.5
Ceftriaxone	≤0.06	>8	69.8/1.1/29.1	69.8/1.1/29.1
Piperacillin/tazobactam	8	>64	72.5/3.2/24.3	65.1/7.4/27.5
Meropenem	≤0.06	>8	78.8/1.1/20.1	79.9/5.8/14.3
Levofloxacin	≤0.12	>4	74.6/1.1/24.3	74.1/0.5/25.4
Gentamicin	≤1	>8	86.8/2.6/10.6	86.2/0.6/13.2
Tigecycline[c]	0.25	1	100.0/0.0/0.0	91.5/8.5/0.0
Colistin	0.5	1	-/-/-	96.7/0.0/3.3
Enterobacter spp. (167)				
Ceftolozane/tazobactam	0.25	8	(93.4)[b]	
Ceftazidime	0.25	>32	77.8/1.8/20.4	74.5/4.3/21.2
Cefepime	≤0.5	2	91.6/4.8/3.6	88.0/8.4/3.6
Ceftriaxone	0.25	>8	72.2/4.3/23.5	72.2/4.3/23.5
Piperacillin/tazobactam	4	64	80.6/11.5/7.9	78.2/2.4/19.4
Meropenem	≤0.06	≤0.06	98.8/1.2/0.0	100.0/0.0/0.0
Levofloxacin	≤0.12	0.25	97.0/1.2/1.8	97.0/0.0/3.0
Gentamicin	≤1	≤1	97.0/0.6/2.4	97.0/0.0/3.0
Tigecycline[b]	0.25	0.5	100.0/0.0/0.0	95.8/4.2/0.0
Colistin	0.5	>8	-/-/-	80.7/0.0/19.3
S. marcescens (156)				
Ceftolozane/tazobactam	0.5	2	(96.2)[b]	
Ceftazidime	0.25	0.5	93.6/1.3/5.1	92.3/1.3/6.4
Cefepime	≤0.5	≤0.5	94.2/3.9/1.9	93.6/1.9/4.5
Ceftriaxone	0.25	8	81.0/2.7/16.3	81.0/2.7/16.3
Piperacillin/tazobactam	2	32	88.5/7.0/4.5	86.5/2.0/11.5
Meropenem	≤0.06	≤0.06	97.4/0.0/2.6	97.4/0.7/1.9
Levofloxacin	≤0.12	1	95.5/1.9/2.6	90.4/5.1/4.5
Gentamicin	≤1	≤1	96.1/2.0/1.9	95.5/0.6/3.9
Tigecycline[b]	0.5	1	99.4/0.6/0.0	98.1/1.3/0.6
Colistin	>8	>8	-/-/-	5.1/0.0/94.9
P. mirabilis (31)				
Ceftolozane/tazobactam	0.5	0.5	(100.0)[b]	
Ceftazidime	0.06	0.06	100.0/0.0/0.0	96.8/3.2/0.0
Cefepime	≤0.5	≤0.5	96.8/0.0/3.2	96.8/0.0/3.2
Ceftriaxone	≤0.06	≤0.06	96.8/0.0/3.2	96.8/0.0/3.2
Piperacillin/tazobactam	≤0.5	1	100.0/0.0/0.0	100.0/0.0/0.0
Meropenem	≤0.06	≤0.06	100.0/0.0/0.0	100.0/0.0/0.0
Levofloxacin	≤0.12	>4	64.5/12.9/22.6	64.5/0.0/35.5
Gentamicin	≤1	8	87.1/9.7/3.2	80.6/6.5/12.9
Tigecycline[b]	1	4	83.9/16.1/0.0	51.6/32.3/16.1
Colistin	>8	>8	-/-/-	0.0/0.0/100.0
Citrobacter spp. (22)				
Ceftolozane/tazobactam	0.25	4	(95.5)[b]	
Ceftazidime	0.5	>32	68.2/0.0/31.8	68.2/0.0/31.8
Cefepime	≤0.5	1	90.9/4.6/4.5	90.9/4.6/4.5
Ceftriaxone	0.25	>8	68.2/0.0/31.8	68.2/0.0/31.8
Piperacillin/tazobactam	4	64	77.3/18.2/4.5	77.3/0.0/22.7
Meropenem	≤0.06	≤0.06	100.0/0.0/0.0	100.0/0.0/0.0
Levofloxacin	≤0.12	4	86.4/9.1/4.5	86.4/0/13.6
Gentamicin	≤1	≤1	95.5/0.0/4.5	95.5/0.0/4.5
Tigecycline[b]	0.25	0.5	100.0/0.0/0.0	100.0/0.0/0.0
Colistin	0.5	1	-/-/-	95.5/0.0/4.5
Indole-positive *Proteae* (23)				
Ceftolozane/tazobactam	0.5	1	(100.0)[b]	
Ceftazidime	0.12	4	95.7/4.3/0.0	87.0/8.7/4.3
Cefepime	≤0.5	≤0.5	95.7/4.3/0.0	95.7/4.3/0.0
Ceftriaxone	≤0.06	4	95.7/4.3/0.0	95.7/4.3/0.0
Piperacillin/tazobactam	≤0.5	8	95.7/4.3/0.0	91.3/4.4/4.3
Meropenem	≤0.06	0.12	100.0/0.0/0.0	100.0/0.0/0.0
Levofloxacin	≤0.12	>4	82.6/4.4/13.0	69.6/13.0/17.4
Gentamicin	≤1	>8	78.3/8.7/13.0	73.9/4.4/21.7
Tigecycline[b]	0.5	1	95.7/4.3/0.0	91.3/4.4/4.3
Colistin	>8	>8	-/-/-	0.0/0.0/100.0
Acinetobacter spp. (81)				
Ceftolozane/tazobactam	32	>32	(39.5)[b]	
Ceftazidime	>32	>32	32.1/6.2/61.7	-/-/-
Cefepime	>16	>16	28.7/7.6/63.7	-/-/-
Meropenem	>8	>8	32.1/4.9/63.0	29.6/7.4/63.0
Ampicillin/sulbactam	16	>32	37.0/14.9/48.1	-/-/-
Piperacillin/tazobactam	>64	>64	27.2/6.1/66.7	-/-/-
Levofloxacin	>4	>4	27.2/0.0/72.8	27.2/0.0/72.8
Gentamicin	>8	>8	37.0/2.5/60.5	37.0/0.0/63.0
Tigecycline	1	2	-/-/-	-/-/-
Colistin	1	2	95.0/0.0/5.0	95.0/0.0/5.0

[a]Criteria as published by the CLSI [2014] and EUCAST [2014].
[b]Percentage inhibited at ceftolozane/tazobactam MICs of ≤8 µg/mL; for comparison purpose only.
[c]In the absence of CLSI breakpoints, USA-FDA breakpoints were applied when available [Tygacil Product Insert, 2012].
I = intermediate; R = resistant; S = susceptible.

CONCLUSIONS

- Ceftolozane/tazobactam demonstrated greater in vitro activity, and susceptibility at ≤8 µg/mL, than currently available cephalosporins, carbapenems, and piperacillin/tazobactam when tested against *P. aeruginosa*.
- Ceftolozane/tazobactam exhibited activity against many MDR and XDR *P. aeruginosa*.
- Against Enterobacteriaceae (including MDR and XDR strains), ceftolozane/tazobactam activity was greater than those of the other cephalosporins tested and piperacillin/tazobactam.
- Ceftolozane/tazobactam may represent a valuable treatment option for Gram-negative infections, including pneumonia caused by MDR and XDR *P. aeruginosa*.

REFERENCES

Clinical and Laboratory Standards Institute. 2012. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: ninth edition. CLSI document M07-A9. CLSI, Wayne, PA.

Clinical and Laboratory Standards Institute. 2014. Performance standards for antimicrobial susceptibility testing: 24th informational supplement. CLSI document M100-S24. CLSI, Wayne, PA.

Eckmann C, et al. Presented at: European Congress of Clinical Microbiology and Infectious Diseases (ECCMID); Barcelona, Spain; May 10-13, 2014. Poster P0266a.

European Committee on Antimicrobial Susceptibility Testing. 2014. Breakpoint tables for interpretation of MICs and zone diameters. Version 4.0, January 2014. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 1, 2014.

Farrell DJ, et al. *Antimicrob Agents Chemother*. 2013;57:6305-6310.

Farrell DJ, et al. *Int J Antimicrob Agents*. 2014;43:533-539.

Magiorakos AP, et al. *Clin Microbiol Infect*. 2012;18:268-281.

Wagenlehner F, et al. Presented at: European Congress of Clinical Microbiology and Infectious Diseases (ECCMID); Barcelona, Spain; May 10-13, 2014. Poster eP449.

ACKNOWLEDGMENTS

This study was funded in part by educational/research grants from Cubist Pharmaceuticals (Lexington, MA, USA). Editorial and layout support for this poster was provided by PAREXEL and funded by Cubist Pharmaceuticals.

This study was funded by Cubist Pharmaceuticals.

Presented at IDWeek: A joint meeting of IDSA • SHEA • HIVMA • PIDS

October 8-12, 2014, Philadelphia, PA