Antimicrobial Activity of Ceftazidime-Avibactam and Comparator Agents Tested against Gram-Negative Organisms Isolated from Complicated Urinary Tract Infections: Results from the International Network for Optimal Resistance Monitoring (INFORM) Program

IDWeek 2018
Poster #1523

Helio S. Sader, Mariana Castanheira, Rodrigo E. Mendes, Robert K. Flamm,

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- Urinary tract infections (UTIs) are among the most frequent healthcare-associated infections and represent a major source of gram-negative bacteremia
- Gram-negative bacteria (GNB), specifically *Enterobacteriaceae*, are common causes of community-acquired and healthcare-associated UTIs
- UTIs caused by antibiotic-resistant GNB are a growing concern due to limited therapeutic options
- Ceftazidime-avibactam is approved by the United States Food and Drug Administration (US FDA) and by the European Medicines Agency (EMA) to treat complicated urinary tract infections (cUTIs), including pyelonephritis, as well as hospital-acquired bacterial pneumonia, including ventilator-associated bacterial pneumonia (HABP/VABP) and complicated intra-abdominal infections (cIAIs) in combination with metronidazole
- We evaluated the antimicrobial susceptibility of GNB isolated from patients with cUTIs in US medical centers

MATERIALS AND METHODS

Bacterial isolates

- Unique patient isolates were consecutively collected from patients with cUTIs in 83 medical centers from 2015 to 2017, and the GNB (n=9,403) were susceptibility tested against ceftazidime-avibactam and comparator agents through the International Network for Optimal Resistance Monitoring (INFORM) Program
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program
- Carbapenem-resistant *Enterobacteriaceae* (CRE) isolates were defined as displaying imipenem, meropenem, and/or doripenem MIC values at ≥4 mg/L (CLSI, 2018)
 - Imipenem was not applied to *Proteus mirabilis* and indole-positive Proteeae due to their intrinsically elevated MIC values

Table 1. Antimicrobial activity of ceftazidime-avibactam tested against the main gram-negative organisms isolated from patients with urinary tract infections (USA, 2015–2017)

Organism/organism group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of: ≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	MIC_{50}	MIC_{90}
Enterobacteriaceae (8,803)	457 5.2	988 16.4	2,832 48.6	3,169 84.6	1,035 96.3	231 99.0	68 99.7	13 99.9	8 >99.9	1 >99.9	1 100.0		0.12	0.25
Escherichia coli (5,771)	401 6.9	602 17.4	1,882 50.0	2,169 87.6	629 98.5	75 99.8	11 >99.9	1 >99.9	1 100.0				0.12	0.25
Klebsiella pneumoniae (1,355)	40 3.0	66 7.8	410 38.1	556 79.1	187 92.9	58 97.2	28 99.3	6 99.7	3 99.9	0 99.9	1 100.0		0.12	0.25
Proteus mirabilis (572)	6 1.0	256 45.8	277 94.2	28 99.1	5 100.0								0.06	0.06
Enterobacter cloacae species complex (254)	2 0.8	1 1.2	19 8.7	87 42.9	77 73.2	52 93.7	12 98.4	3 99.6	1 100.0				0.25	0.5
Enterobacter aerogenes (168)	1 0.6	8 5.4	34 25.6	88 78.0	28 94.6	4 97.0	3 98.8	1 99.4	1 100.0				0.12	0.25
Citrobacter freundii species complex (161)	4 2.5	3 4.3	30 23.0	67 64.6	41 90.1	12 97.5	3 99.4	0 99.4	0 99.4	1 100.0			0.12	0.25
Klebsiella oxytoca (160)	0 0.0	4 2.5	68 45.0	64 85.0	18 96.2	5 99.4	0 99.4	1 100.0					0.12	0.25
Indole-positive Proteeae (142)	0 0.0	24 16.9	65 62.7	27 81.7	11 89.4	10 96.5	4 99.3	0 99.3	1 100.0				0.06	0.5
Pseudomonas aeruginosa (531)					5 0.9	11 3.0	173 35.6	225 78.0	83 93.6	29 99.1	4 99.8	1 100.0	2	4

Table 2. Antimicrobial activity of ceftazidime-avibactam and comparator agents tested against *Enterobacteriaceae* and *P. aeruginosa* isolated from patients with complicated urinary tract infections (INFORM Program; 2015–2017)

Organism/organism group (no. of isolates) Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R
Enterobacteriaceae (8,803)				
Ceftazidime-avibactam	0.12	0.25	>99.9	<0.1
Ceftolozane-tazobactam	0.25	0.5	97.7	1.6
Ceftriaxone	≤0.06	>8	87.2	12.5
Ceftazidime	0.12	4	90.2	8.2
Cefepime	≤0.12	1	91.0	6.8
Ampicillin-sulbactam	8	>32	57.7	26.0
Piperacillin-tazobactam	2	8	96.1	1.9
Meropenem	≤0.015	0.06	99.3	0.6
Ciprofloxacin	≤0.03	>4	78.7	20.4
Levofloxacin	0.06	>4	79.7	18.6
Gentamicin	0.5	4	90.4	8.7
Amikacin	2	4	99.5	0.2
TMP-SMX[b]	≤0.5	>4	73.7	26.3
Tigecycline	0.25	1	97.3[c]	0.3[c]
Colistin	0.12	8	89.7[d]	
ESBL-producing *Enterobacteriaceae* (744)[e]				
Ceftazidime-avibactam	0.12	0.25	100.0	0.0
Ceftolozane-tazobactam	0.5	1	96.7	1.4
Ceftriaxone	>8	>8	0.4	99.2
Ceftazidime	16	>32	25.8	61.4
Cefepime	>16	>16	9.8	71.3
Ampicillin-sulbactam	32	>32	17.1	65.6
Piperacillin-tazobactam	4	32	88.6	3.6
Meropenem	0.03	0.06	99.5	0.0
Aztreonam	>16	>16	13.8	74.7
Ciprofloxacin	>4	>4	16.7	80.1
Levofloxacin	>4	>4	21.9	74.6
Gentamicin	1	>8	62.8	35.6
Amikacin	4	8	97.7	0.7
TMP-SMX[b]	>4	>4	32.1	67.9
Tigecycline	0.25	0.5	99.2[c]	0.1[c]
Colistin	0.12	0.25	98.8[d]	
Carbapenemase-producing *Enterobacteriaceae* (56)[f]				
Ceftazidime-avibactam	0.5	2	98.2	1.8
Ceftolozane-tazobactam	>16	>16	5.3	89.5
Ceftriaxone	>8	>8	3.6	96.4
Ceftazidime	>32	>32	3.6	92.9
Cefepime	>16	>16	8.9	75.0
Ampicillin-sulbactam	>32	>32	0.0	100.0
Piperacillin-tazobactam	>64	>64	5.4	87.5
Meropenem	>16	>16	7.1	87.5
Aztreonam	>4	>4	0.0	96.4
Ciprofloxacin	>4	>4	10.7	83.9
Levofloxacin	4	>8	14.3	78.6
Gentamicin	16	>32	46.4	25.0
Amikacin	>4	>4	58.9	14.3
TMP-SMX[b]	0.5	1	14.3	85.7
Tigecycline	0.12	>8	100.0	0.0[c]
Colistin			83.9[d]	
Pseudomonas aeruginosa (531)				
Ceftazidime-avibactam	2	4	99.1	0.9
Ceftolozane-tazobactam	0.5	1	99.5	0.5
Ceftazidime	2	16	88.5	7.0
Cefepime	2	16	89.5	2.4
Piperacillin-tazobactam	4	32	84.6	5.5
Meropenem	0.5	8	83.6	10.2
Aztreonam	8	>16	71.2	16.5
Ciprofloxacin	0.25	>4	74.0	21.1
Levofloxacin	0.5	>4	68.5	24.1
Gentamicin	2	8	87.6	6.8
Amikacin	4	8	97.9	0.9
Tobramycin	0.5	2	94.4	4.6
Colistin	≤0.5	1	100.0	0.0

[a] Criteria as published by CLSI 2018.
[b] TMP-SMX, trimethoprim-sulfamethoxazole.
[c] FDA breakpoints published 2017-DEC-13.
[d] Percentage of wild type based on ECV. CLSI M100 (2018).
[e] Extended-spectrum β-lactamase (ESBL)-producing *Enterobacteriaceae* excluded carbapenemase-producing isolates and included isolates of *Enterobacter cloacae* species complex (12), *Escherichia coli* (622), *Klebsiella oxytoca* (5), *K. pneumoniae* (102), and *Proteus mirabilis* (3).
[f] *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (3), *Escherichia coli* (2), *Klebsiella oxytoca* (3), *K. pneumoniae* (45), *Serratia marcescens* (2).

Susceptibility testing

- Broth microdilution test method was conducted according to the Clinical and Laboratory Standards Institute (CLSI) standards
- Avibactam was provided by Allergan (Irvine, California, USA) and combined with ceftazidime (avibactam at fixed concentration of 4 mg/L) for susceptibility testing
- Ceftolozane stock solution was obtained from ThermoFisher Scientific (Cleveland, Ohio, USA) and combined with tazobactam (acquired from United States Pharmacopeia [USP]) at fixed concentration of 4 mg/L for susceptibility testing
- All other compounds were obtained from USP or Sigma-Aldrich (St. Louis, Missouri, USA)

Screening for β-lactamase-encoding genes

- *Enterobacteriaceae* isolates displaying MIC values ≥2 mg/L for at least 2 β-lactams (ie, ceftazidime, ceftriaxone, aztreonam, or cefepime) and all CRE isolates underwent next-generation sequencing to test for β-lactamase-encoding genes
- Libraries were normalized using the bead-based normalization procedure (Illumina) and sequenced on MiSeq
- FASTQ files were assembled using SPAdes Assembler and subjected to a proprietary software (JMI Laboratories) to screen β-lactamase genes

Figure 1. Frequency of organisms isolated from patients with bacteremia from US medical centers (INFORM Program, 2017)

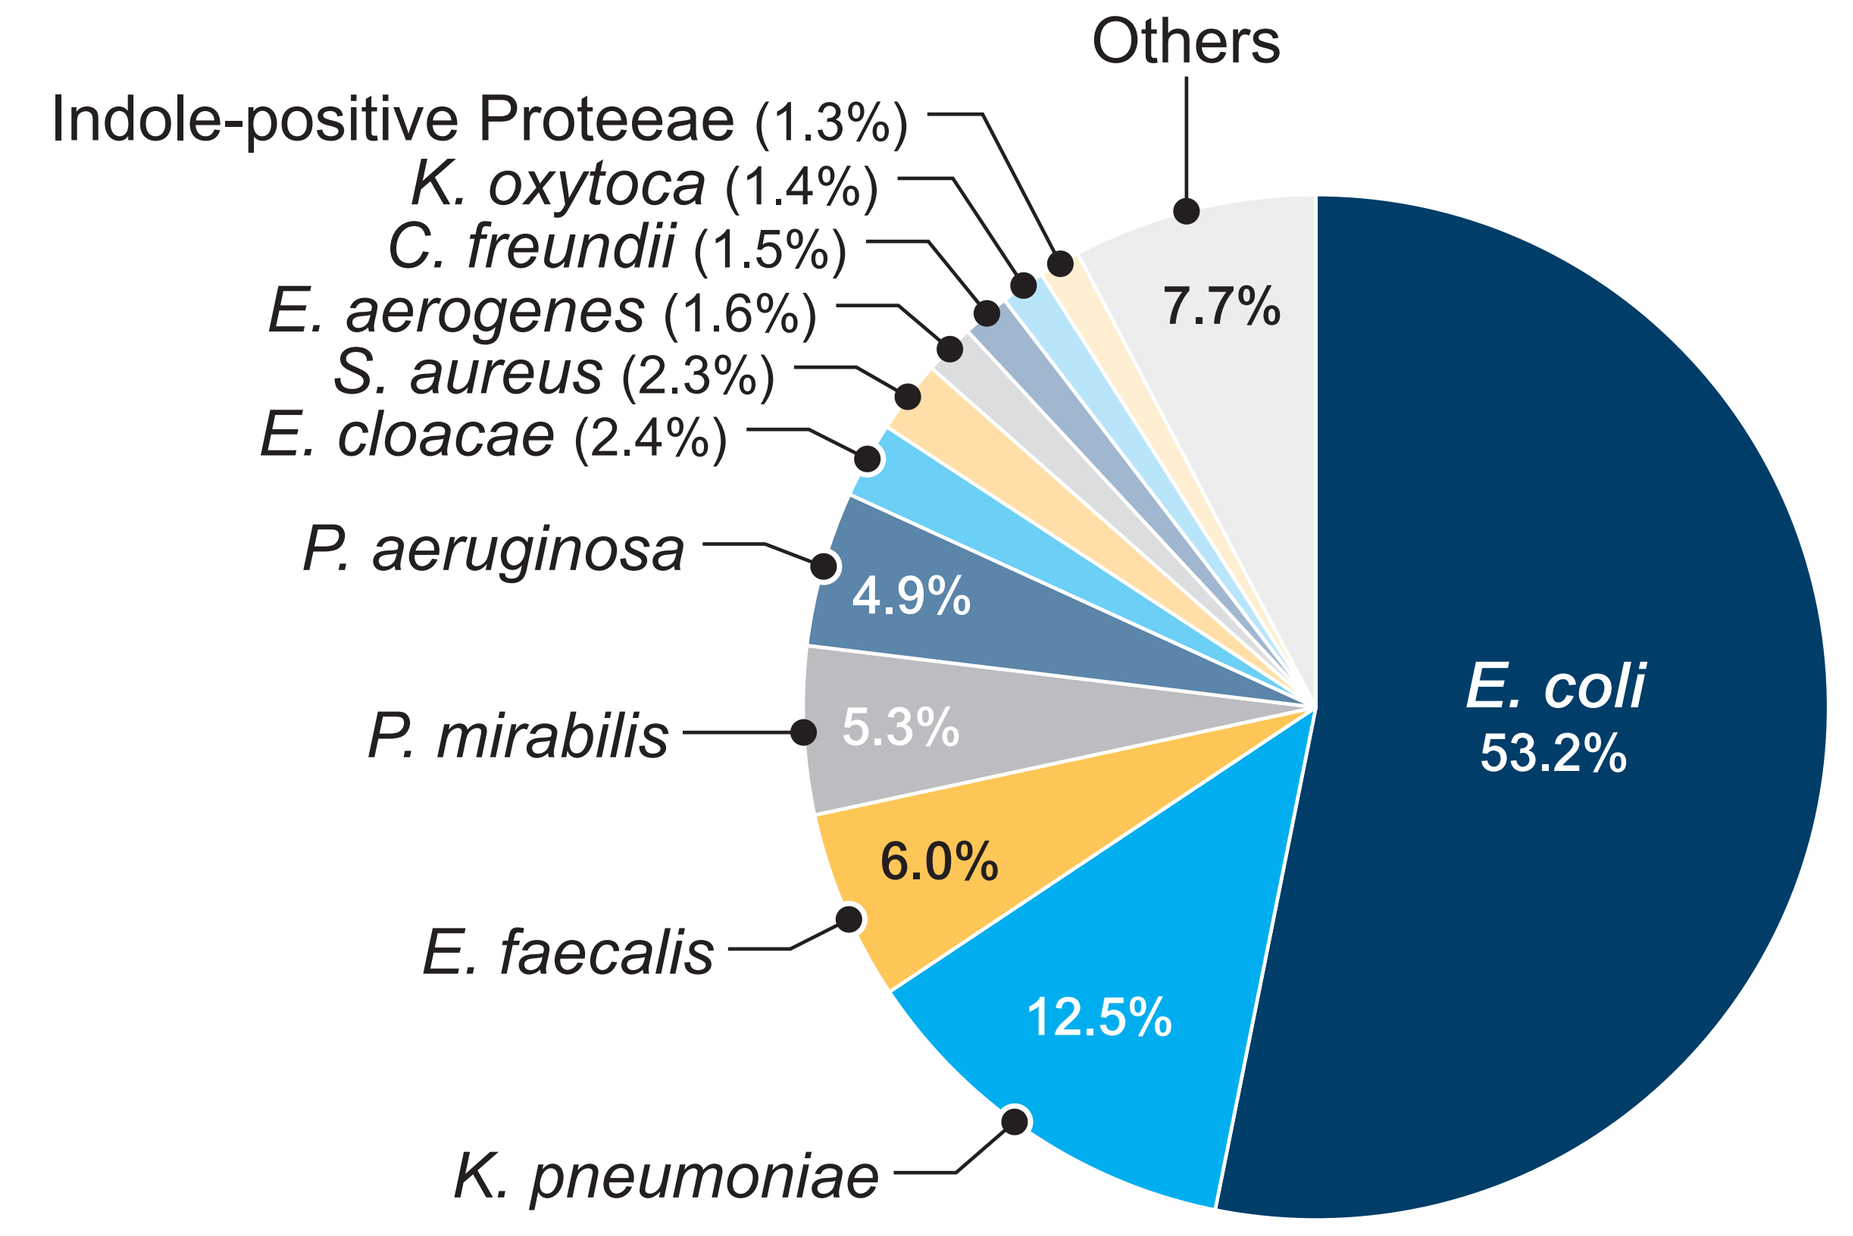
Others
7.7%
Indole-positive Proteeae (1.3%)
K. oxytoca (1.4%)
C. freundii (1.5%)
E. aerogenes (1.6%)
S. aureus (2.3%)
E. cloacae (2.4%)
P. aeruginosa
4.9%
P. mirabilis
5.3%
6.0%
E. faecalis
12.5%
K. pneumoniae
E. coli
53.2%

RESULTS

- The most common organisms were *Escherichia coli* (53.2%), *Klebsiella pneumoniae* (12.5%), *Enterococcus faecalis* (6.0%), *P. mirabilis* (5.3%), *Pseudomonas aeruginosa* (4.9%), and *Enterobacter cloacae* (2.4%; Figure 1)
- Ceftazidime-avibactam inhibited >99.9% of all *Enterobacteriaceae* isolates, including all *E. coli*, *P. mirabilis*, and *E. cloacae* isolates, at ≤8 mg/L, the susceptible (S) breakpoint (Tables 1 and 2)
- Ceftazidime-avibactam was also highly active against extended-spectrum β-lactamase (ESBL)-producing (n=744; $MIC_{50/90}$, 0.12/0.25 mg/L; 100.0%S) and carbapenemase-producing (n=56; $MIC_{50/90}$, 0.5/2 mg/L; 98.2%S) *Enterobacteriaceae* (Table 2 and Figure 2)
- Only 1 *Enterobacteriaceae* isolate was ceftazidime-avibactam-resistant, a *K. pneumoniae* with a ceftazidime-avibactam MIC of 16 mg/L that produced a KPC-2 and a SHV-12 and exhibited decreased expression of OmpK36
- Meropenem and ceftolozane-tazobactam were active against 99.5% and 96.7% of ESBL-producing *Enterobacteriaceae*, respectively, but exhibited very limited activity against carbapenemase-producing *Enterobacteriaceae* (Table 2)
- Among *E. cloacae* (27.2% ceftazidime-nonsusceptible), susceptibility rates to ceftazidime-avibactam, ceftolozane-tazobactam, and meropenem were 100.0%, 80.0%, and 98.8%, respectively (data not shown)
- Among 56 carbapenemase-producing strains, 54 (96.4%) produced a KPC-like and 2 (3.6%) produced an SME-4; *K. pneumoniae* represented 80.3% (45/56) of the collection (Table 3)
- Ceftazidime-avibactam was also highly active against *P. aeruginosa* ($MIC_{50/90}$, 2/4 mg/L; 99.1%S) and retained good activity against isolates nonsusceptible to meropenem, ceftazidime, and piperacillin-tazobactam (n=29; $MIC_{50/90}$, 4/16 mg/L; 86.2%S; data not shown)
- *Acinetobacter* spp. (n=69; 0.7% of GNB) exhibited high rates of resistance to most antimicrobial agents tested (data not shown)

Figure 2. Ceftazidime-avibactam MIC distributions when testing extended-spectrum β-lactamase (ESBL)-producing (excluding carbapenemases) and carbapenemase-producing *Enterobacteriaceae* isolates (USA, 2015–2017)

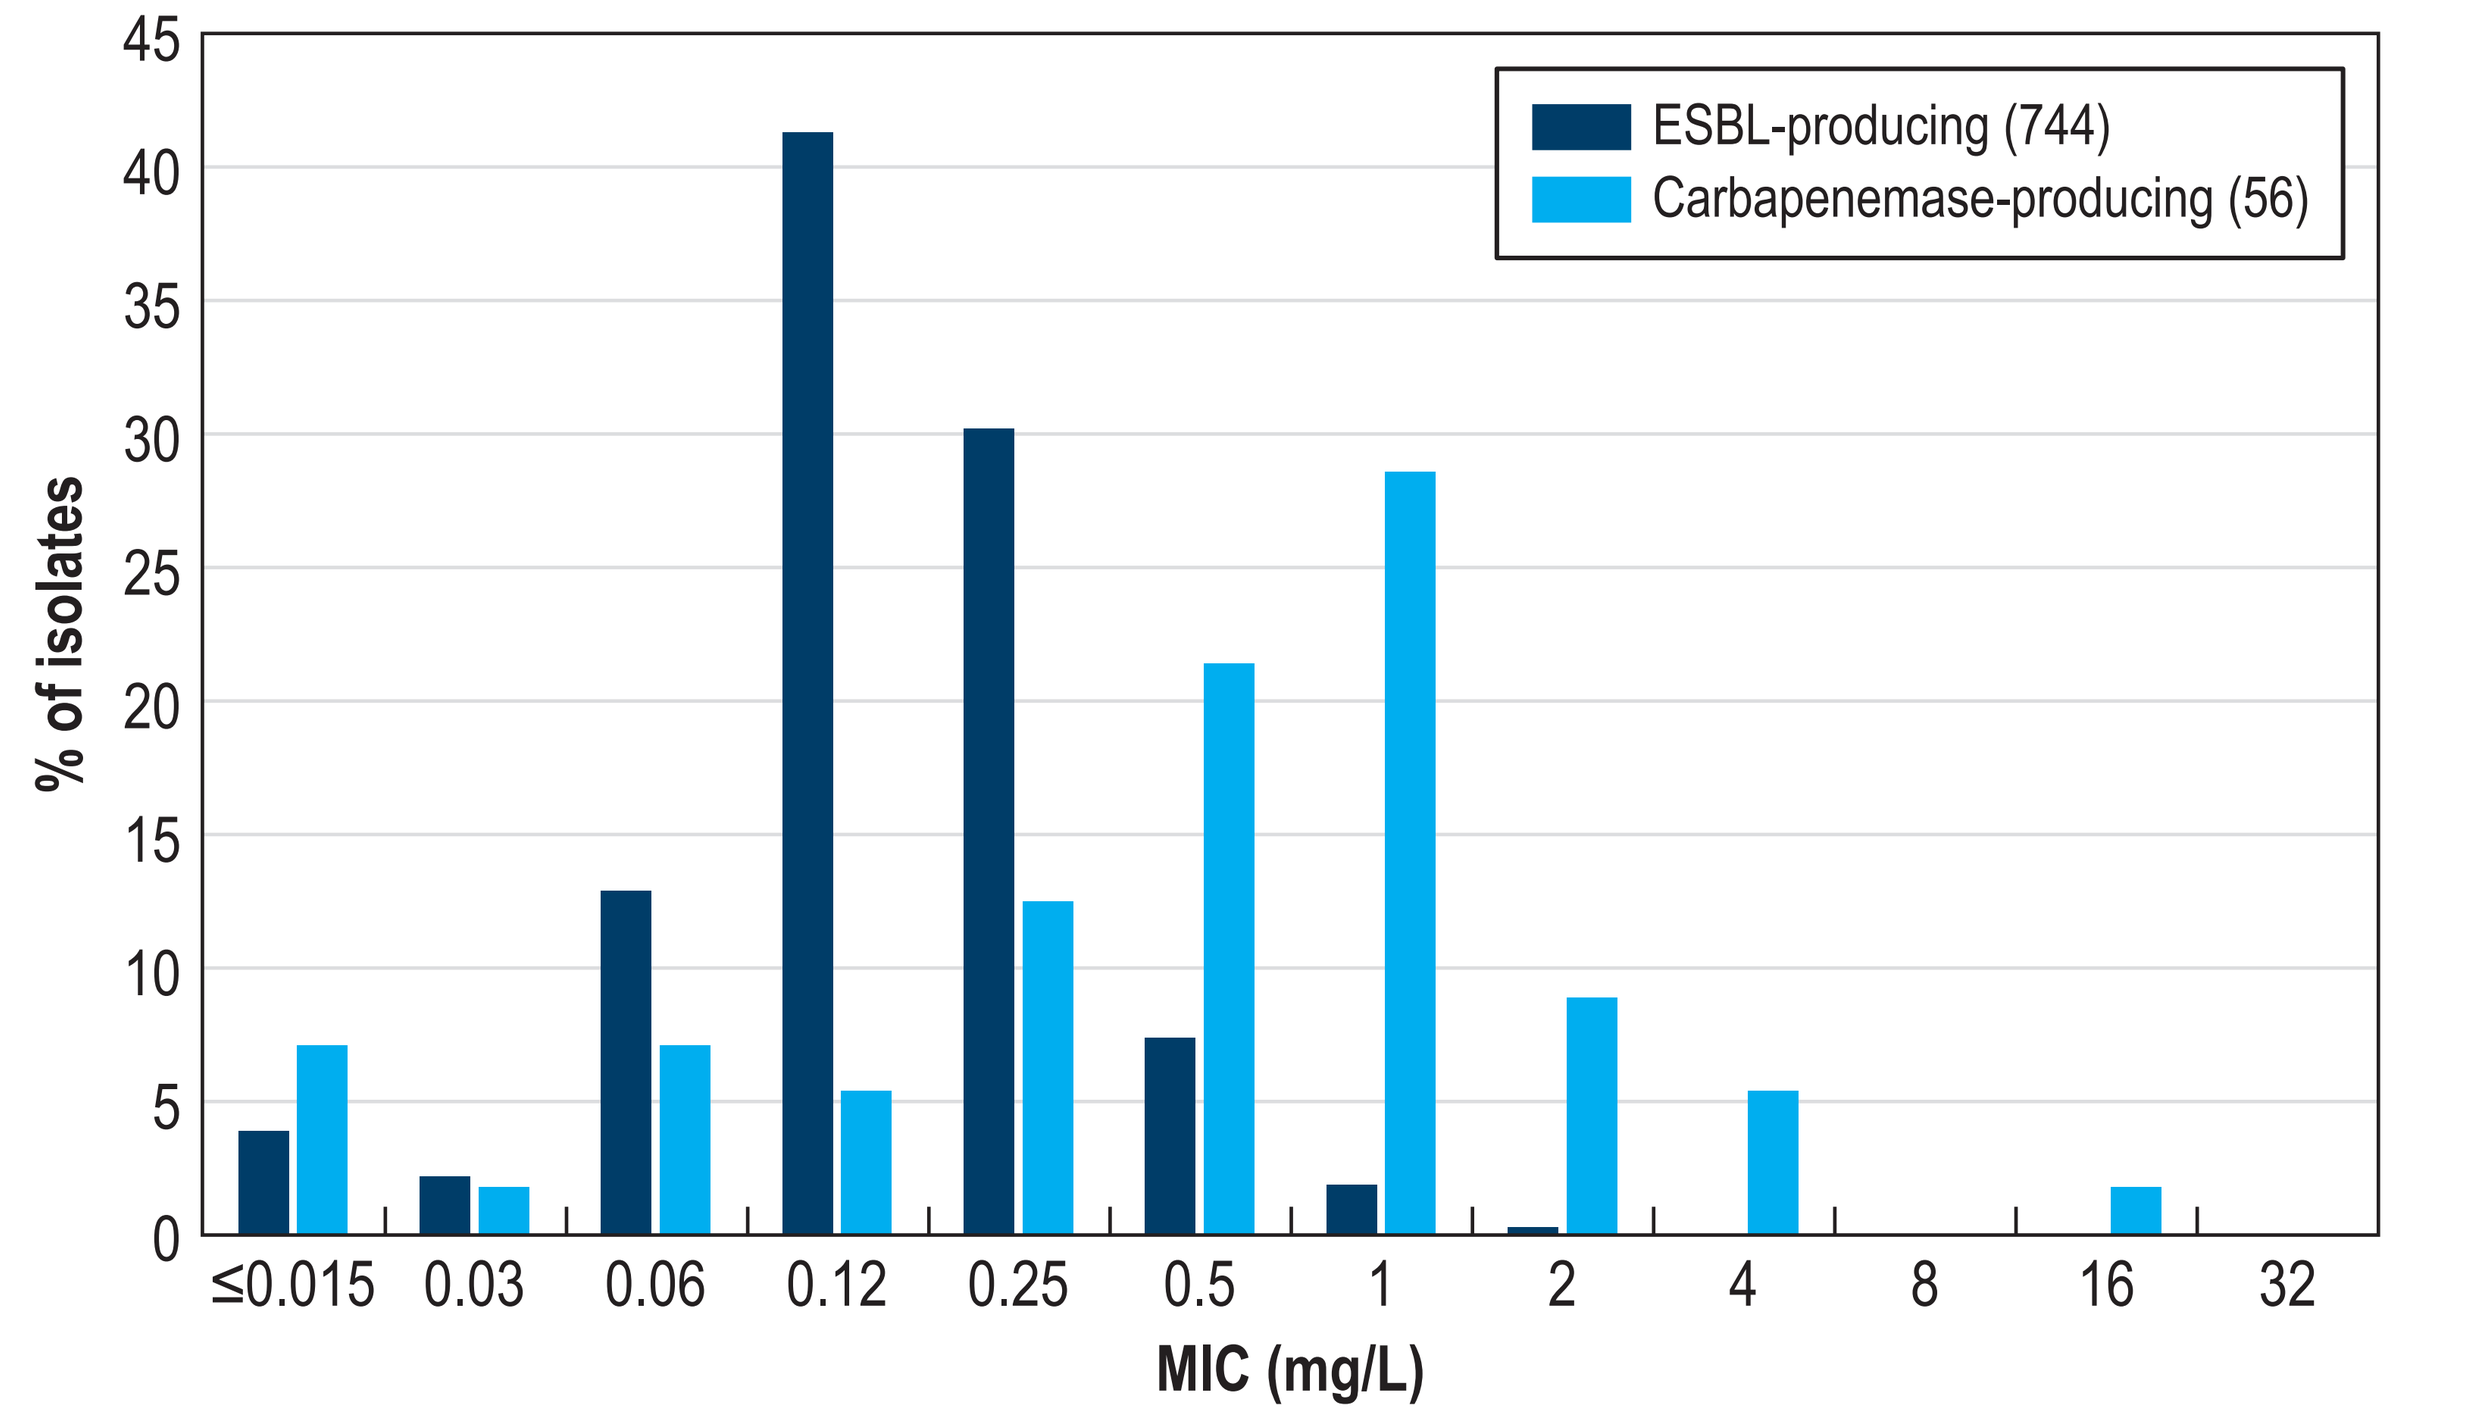
ESBL-producing (744)
Carbapenemase-producing (56)
% of isolates
0
5
10
15
20
25
30
35
40
45
≤0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
MIC (mg/L)

Table 3. Carbapenemases produced by *Enterobacteriaceae* isolated from patients with UTI in US medical centers (INFORM Program, 2015–2017)

Organism Carbapenemase	No. of isolates
Klebsiella pneumoniae	**45**
KPC-2	8
KPC-3	17
KPC-like	20
Enterobacter cloacae	**3**
KPC-2	1
KPC-3	2
Klebsiella oxytoca	**3**
KPC-3	2
KPC-like	1
Escherichia coli	**2**
KPC-3	2
Serratia marcescens	**2**
SME-4	2
Citrobacter freundii	**1**
KPC-3	1
Total	**56**

CONCLUSIONS

- Ceftazidime-avibactam demonstrated potent activity against a large collection of contemporary (2015-2017) GNB isolated from patients with cUTIs in US medical centers, including ESBL-producing and carbapenemase-producing *Enterobacteriaceae* isolates
- Ceftazidime-avibactam and ceftolozane-tazobactam showed similar coverage (%S) against *P. aeruginosa* (99.1%S vs. 99.5%S)

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the INFORM Program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi. Accessed January 2017.
2. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition.* Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
4. Flores-Mireles AL, Walker JN, Caparon M, Hultgren SJ (2015). Urinary tract infections: epidemiology, mechanisms of infection and treatment options. *Nat Rev Microbiol* 13:269-84
5. Sader HS, Castanheira M, Duncan LR, Flamm RK (2018). Antimicrobial susceptibility of *Enterobacteriaceae* and *Pseudomonas aeruginosa* isolates from United States medical centers stratified by infection type: Results from the International Network for Optimal Resistance Monitoring (INFORM) surveillance program, 2015–2016. *Diagn Microbiol Infect Dis* 92: 69-74.

Visit www.allergancongressposters.com/370797

Allergan

IDWeek 2018, October 3–7, 2018 in San Francisco, California