Comparison of Ceftazidime-Avibactam and Ceftolozane-Tazobactam *In Vitro* Activities when Tested against Gram-Negative Bacteria Isolated from Patients Hospitalized with Pneumonia in US Medical Centers (2017)

IDWeek 2018
Poster #2427

Helio S. Sader, Robert K. Flamm, Mariana Castanheira

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- The initial antimicrobial management of patients with pneumonia is determined mostly by the understanding of causative pathogens, and there is very little current information regarding the frequency and antimicrobial susceptibility of organisms causing healthcare-associated pneumonia
- Although *Staphylococcus aureus* is a significant cause of pneumonia in hospitalized patients, the importance of gram-negative organisms such as *Pseudomonas aeruginosa* and *Enterobacteriaceae* species, mainly *Klebsiella pneumoniae*, *Enterobacter* spp., and *Escherichia coli*, has increased substantially in recent years
- Ceftazidime-avibactam is approved by the United States Food and Drug Administration (US FDA) and by the European Medicine Agency (EMA) to treat hospital-acquired bacterial pneumonia, including ventilator-associated bacterial pneumonia (HABP/VABP)
- Ceftazidime-avibactam is also approved to treat complicated intra-abdominal infections (cIAIs) in combination with metronidazole, as well as complicated urinary tract infections, including pyelonephritis
- We evaluated the frequency and antimicrobial susceptibility of gram-negative bacteria isolated from patients hospitalized with pneumonia in US medical centers, and assessed the activity and spectrum of 2 recently approved β-lactamase inhibitor combinations, ceftazidime-avibactam and ceftolozane-tazobactam, and many other antimicrobial agents currently used to treat pneumonia

MATERIALS AND METHODS

Bacterial isolates

- A total of 5,432 bacterial isolates were consecutively collected (1/infection episode) from 70 US medical centers in 2017
- Among those, 1,865 *Enterobacteriaceae* and 1,337 *Pseudomonas aeruginosa* isolates were tested for susceptibility as part of the International Network for Optimal Resistance Monitoring (INFORM) Program
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program

Resistant subsets

- Carbapenem-resistant *Enterobacteriaceae* (CRE) isolates were defined as displaying imipenem, meropenem, and/or doripenem MIC values at ≥4 mg/L (CLSI, 2018)
 - Imipenem was not applied to *Proteus mirabilis* and indole-positive Proteeae isolates due to the intrinsically elevated MIC values

Table 1 Antimicrobial activity of ceftazidime-avibactam tested against *Enterobacteriaceae* and *P. aeruginosa* from pneumonia in hospitalized patients

Organism/ organism group (no. of isolates)	No. of isolates and cumulative % at MIC (mg/L) of: ≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32	MIC_{50}	MIC_{90}
Enterobacteriaceae (1,865)	24 1.3	64 4.7	389 25.6	730 64.7	410 86.7	168 95.7	46 98.2	24 99.5	8 99.9	1 99.9	0 99.9	1[a] 100.0		0.12	0.5
ESBL-producing (176)	4 2.3	3 4.0	6 7.4	63 43.2	66 80.7	25 94.9	8 99.4	1 100.0						0.25	0.5
MDR (224)	8 3.6	8 7.1	27 19.2	37 35.7	50 58.0	35 73.7	29 86.6	22 96.4	6 99.1	1 99.6	0 99.6	1[a] 100.0		0.25	2
XDR (41)			4 9.8	1 12.2	7 29.3	7 46.3	9 68.3	10 92.7	2 97.6	0 97.6	0 97.6	1[a] 100.0		1	2
CRE (50)	1 2.0	0 2.0	0 2.0	1 4.0	6 16.0	9 34.0	15 64.0	14 92.0	3 98.0	0 98.0	0 98.0	1[a] 100.0		1	2
P. aeruginosa (1,337)		1 0.1	4 0.4	12 1.3	12 2.2	61 6.7	297 28.9	593 73.3	201 88.3	105 96.2	30 98.4	8 99.0	13 100.0	2	8
Meropenem-nonsusceptible (>2 mg/L; 386)						4 1.0	37 10.6	106 38.1	115 67.9	78 88.1	27 95.1	7 96.9	12 100.0	4	16
MDR (331)					1 0.3	4 1.5	20 7.6	81 32.0	93 60.1	82 84.9	29 93.7	8 96.1	13 100.0	4	16
XDR (204)							6 2.9	46 25.5	60 54.9	50 79.4	22 90.2	8 94.1	12 100.0	4	16

[a] A *Serratia marcescens* isolate from New York, NY, with KPC-3, OXA-10, OXA-9, SRT-like, TEM-1, and alterations in *ompC* and *ompF*.

- Multidrug-resistant (MDR) and extensively drug-resistant (XDR) *Enterobacteriaceae* and *P. aeruginosa* strains were classified according to recommended guidelines (Magiorakos et al., 2012) as follows:
 - MDR = nonsusceptible (NS; CLSI breakpoints) to at least 3 antimicrobial classes
 - XDR = susceptible (S) to 2 or fewer antimicrobial classes

Susceptibility testing

- Broth microdilution test method was conducted according to CLSI
- Avibactam was provided by Allergan (Irvine, California, USA) and combined with ceftazidime (avibactam at fixed concentration of 4 mg/L) for susceptibility testing
- Ceftolozane stock solution was obtained from ThermoFisher Scientific (Cleveland, Ohio, USA) and combined with tazobactam (acquired from United States Pharmacopeia [USP]) at fixed concentration of 4 mg/L for susceptibility testing
- All other compounds were obtained from USP or Sigma-Aldrich (St. Louis, Missouri, USA)

Screening for β-lactamase-encoding genes

- *Enterobacteriaceae* isolates displaying MIC values ≥2 mg/L for at least 2 β-lactams (ie, ceftazidime, ceftriaxone, aztreonam, or cefepime) and all CRE isolates were tested for β-lactamase-encoding genes using next-generation sequencing
- Libraries were normalized using the bead-based normalization procedure (Illumina) and sequenced on MiSeq
- FASTQ files were assembled using SPAdes Assembler and subjected to a proprietary software (JMI Laboratories) for screening of β-lactamase genes

RESULTS

- The most common organisms isolated from patients hospitalized with pneumonia were *S. aureus* (27.5%), *P. aeruginosa* (24.7%), *Klebsiella pneumoniae* (7.9%), *E. coli* (6.5%), *Stenotrophomonas maltophilia* (4.5%), and *Enterobacter cloacae* (4.2%); overall, 68.7% of the organisms were gram-negative and 31.3% were gram-positive organisms (Figure 1)
- Ceftazidime-avibactam exhibited potent activity against *Enterobacteriaceae* ($MIC_{50/90}$, 0.12/0.5 mg/L; 99.9% susceptible [S]), including extended-spectrum β-lactamase (ESBL)-producing strains ($MIC_{50/90}$, 0.25/0.5 mg/L; 100.0%S), MDR ($MIC_{50/90}$, 0.25/2 mg/L; 99.6%S), and XDR isolates ($MIC_{50/90}$, 1/2 mg/L; 97.6%S; Tables 1 and 2 and Figure 2)
- Only 1 *Enterobacteriaceae* isolate (0.05%) was ceftazidime-avibactam resistant, a *Serratia marcescens* isolate from New York, NY, with KPC-3, OXA-10, OXA-9, SRT-like, TEM-1, and alterations in *ompC* and *ompF*

Figure 1. Frequency of organisms isolated from patients hospitalized with pneumonia in US medical centers (INFORM Program, 2017)

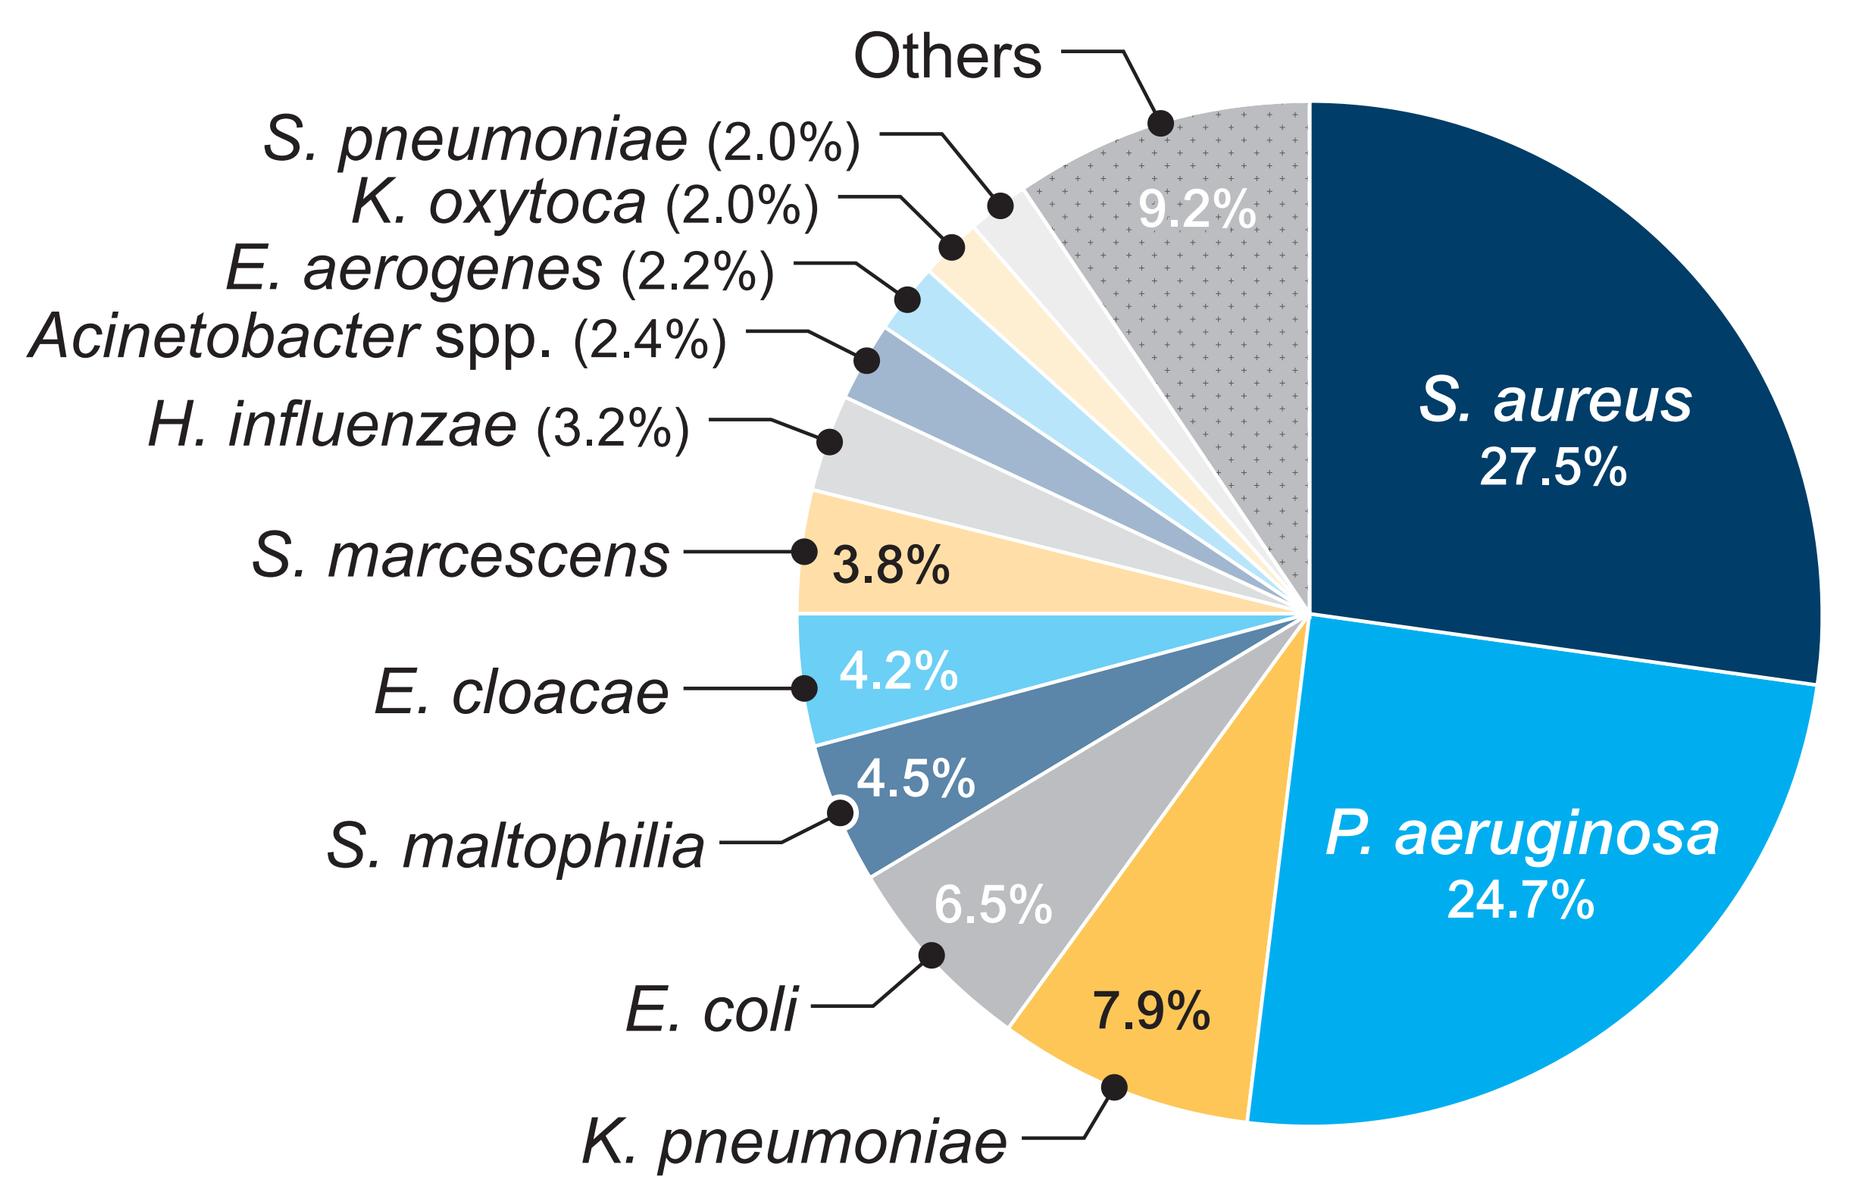
Others
9.2%
S. pneumoniae (2.0%)
K. oxytoca (2.0%)
E. aerogenes (2.2%)
Acinetobacter spp. (2.4%)
H. influenzae (3.2%)
S. marcescens
3.8%
E. cloacae
4.2%
S. maltophilia
4.5%
E. coli
6.5%
K. pneumoniae
7.9%
S. aureus
27.5%
P. aeruginosa
24.7%

Figure 2. Antimicrobial susceptibility of *Enterobacteriaceae* isolated from patients hospitalized with pneumonia in US medical centers (INFORM Program, 2017)

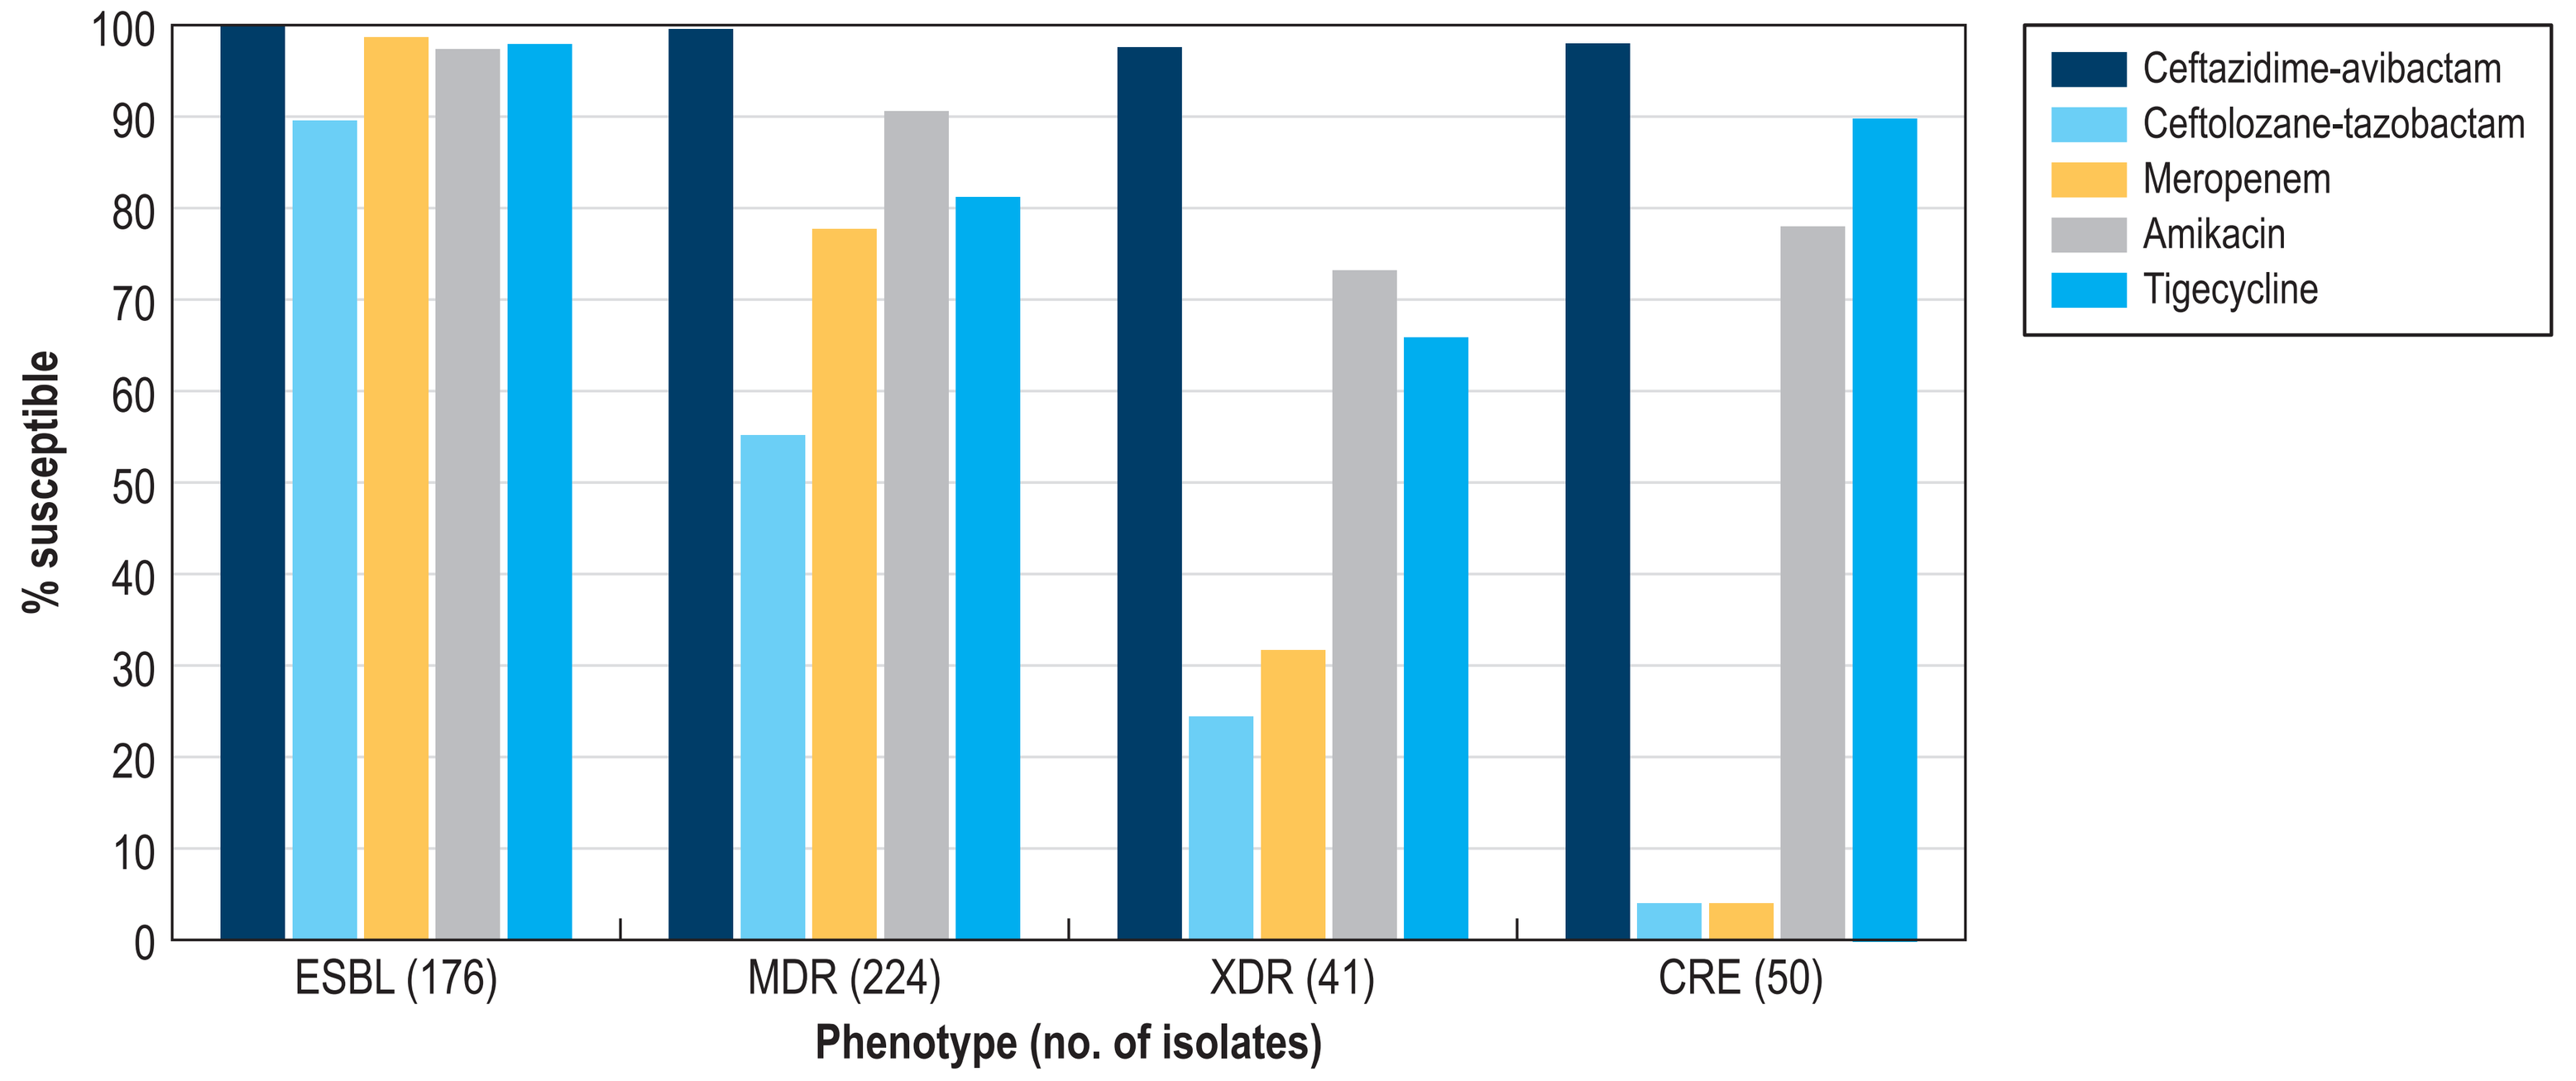
% susceptible
100
90
80
70
60
50
40
30
20
10
0
ESBL (176)
MDR (224)
XDR (41)
CRE (50)
Phenotype (no. of isolates)
Ceftazidime-avibactam
Ceftolozane-tazobactam
Meropenem
Amikacin
Tigecycline

Abbreviations: ESBL, extended-spectrum β-lactamases (excluding carbapenemase-producing strains); MDR, multidrug-resistant; XDR, extensively drug-resistant, CRE, carbapenem-resistant *Enterobacteriaceae.*

Figure 3. Antimicrobial susceptibility of *P. aeruginosa* isolated from patients hospitalized with pneumonia in US medical centers (INFORM Program, 2017)

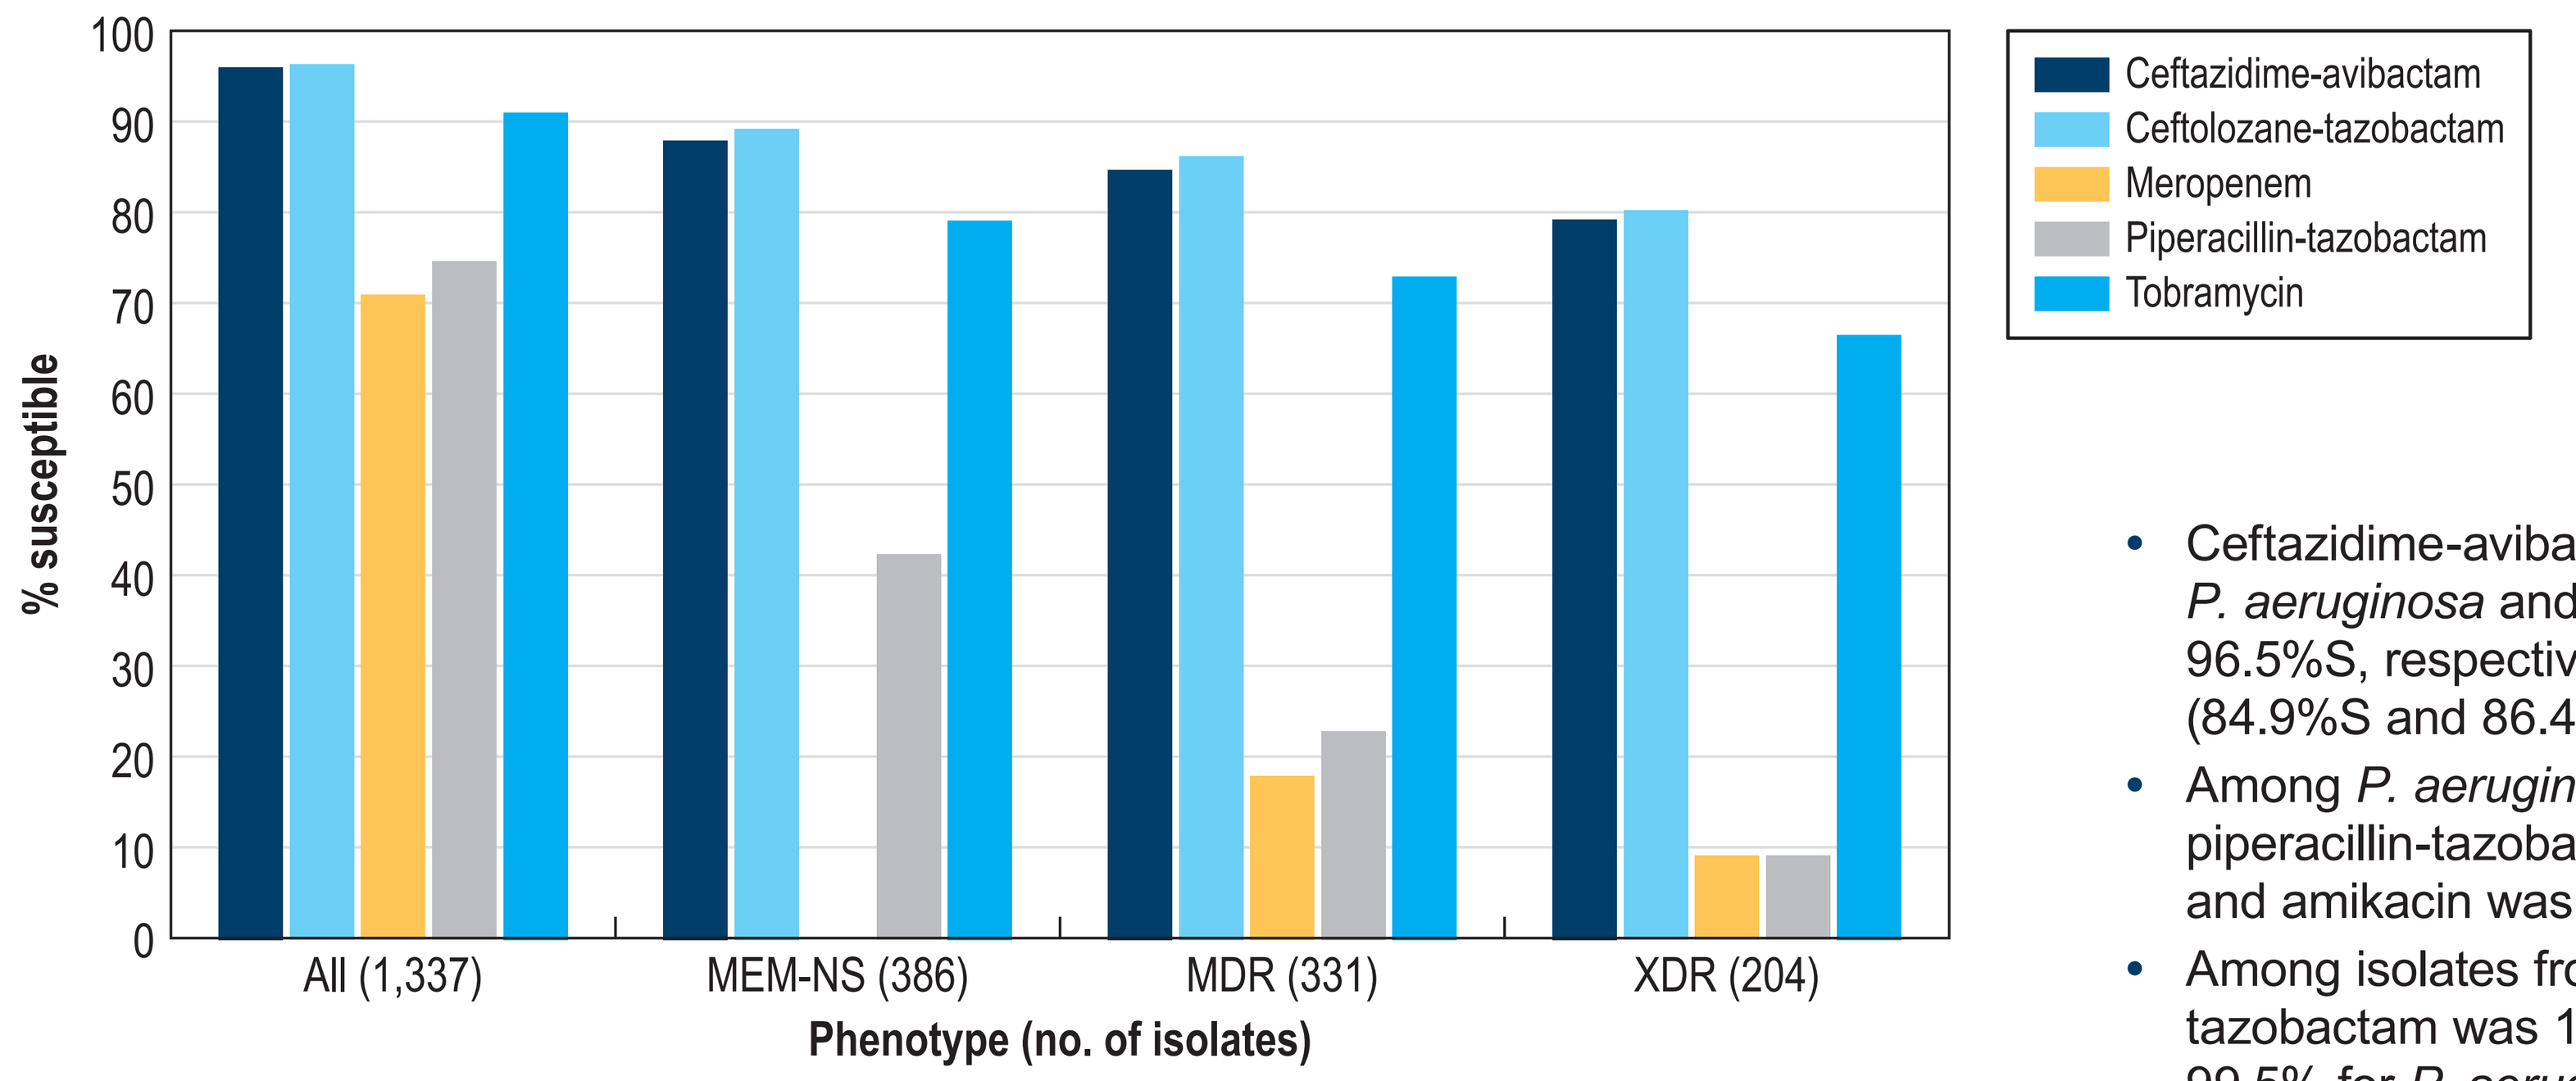
% susceptible
100
90
80
70
60
50
40
30
20
10
0
All (1,337)
MEM-NS (386)
MDR (331)
XDR (204)
Phenotype (no. of isolates)
Ceftazidime-avibactam
Ceftolozane-tazobactam
Meropenem
Piperacillin-tazobactam
Tobramycin

Abbreviations: MEM-NS, meropenem-nonsusceptible; MDR, multidrug-resistant; XDR, extensively drug-resistant.

- The most active agents against *Enterobacteriaceae* were ceftazidime-avibactam (>99.9%S), amikacin (98.7%S), the carbapenems meropenem and doripenem (97.3%S), and tigecycline (94.1%S), but only ceftazidime-avibactam and tigecycline retained good activity (≥90%S) against CRE (98.0% and 90.0%S, respectively; Table 2 and Figure 2)
- The most active agents against MDR *Enterobacteriaceae* were ceftazidime-avibactam (99.6%S) and amikacin (90.6%S), whereas ceftolozane-tazobactam and meropenem were active against only 55.2% and 77.7% of these organisms, respectively (Figure 2)
- Ceftazidime-avibactam was the most active agent tested against XDR *Enterobacteriaceae* (97.6%S) followed by amikacin (73.2%S) and tigecycline (65.9%S; Figure 2)
- Among ESBL-producing *Enterobacteriaceae* (excluding carbapenemase-producing) isolates, susceptibility to ceftazidime-avibactam, ceftolozane-tazobactam, and meropenem were 100.0%, 84.0%, and 99.4%, respectively (Table 2)

Table 2 Antimicrobial activity of ceftazidime-avibactam and comparator agents tested against *Enterobacteriaceae* and *P. aeruginosa* isolated from patients with pneumonia (INFORM Program; 2015–2017)

Organism/organism group (no. of isolates) Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	CLSI[a] %S	CLSI[a] %R
Enterobacteriaceae (1,865)				
Ceftazidime-avibactam	0.12	0.5	99.9	0.1
Ceftolozane-tazobactam	0.25	2	90.3	7.2
Piperacillin-tazobactam	2	64	86.5	7.7
Ceftazidime	0.25	>32	81.2	17.1
Ceftriaxone	0.12	>8	76.1	21.9
Cefepime	≤0.12	8	86.7 [b]	9.2
Meropenem	0.03	0.06	97.3	2.4
Doripenem	≤0.06	0.25	97.3	2.5
Levofloxacin	0.06	16	83.7	14.8
Gentamicin	0.5	2	90.8	7.9
Amikacin	2	4	98.7	0.2
Tigecycline [c]	0.5	2	94.1	0.8
Colistin[d]	0.12	>8	77.7[d]	
ESBL-producing isolates (176) [e]				
Ceftazidime-avibactam	0.25	0.5	100.0	0.0
Ceftolozane-tazobactam	0.5	8	84.0	12.6
Piperacillin-tazobactam	4	64	81.2	9.7
Ceftriaxone	>8	>8	0.6	96.0
Ceftazidime	32	>32	17.6	72.7
Cefepime	>16	>16	12.5 [b]	66.5
Meropenem	0.03	0.06	99.4	0.6
Doripenem	≤0.06	0.12	98.9	1.1
Levofloxacin	8	>16	38.1	59.7
Gentamicin	1	>16	55.7	39.2
Amikacin	2	8	96.0	0.0
Tigecycline [c]	0.25	1	97.7	0.0
Colistin[d]	0.12	0.25	98.8 [d]	

Organism/organism group (no. of isolates) Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	CLSI[a] %S	CLSI[a] %R
Pseudomonas aeruginosa (1,337)				
Ceftazidime-avibactam	2	8	96.2	3.8
Ceftolozane-tazobactam	0.5	2	96.5	1.8
Piperacillin-tazobactam	8	128	74.8	13.7
Ceftazidime	2	32	79.9	15.0
Cefepime	4	16	80.0	8.7
Meropenem	0.5	16	71.1	20.4
Doripenem	0.5	>8	72.9	19.7
Levofloxacin	1	16	69.3	20.7
Gentamicin	2	16	78.4	10.9
Amikacin	4	16	93.2	3.6
Tobramycin	1	4	91.2	6.9
Colistin	0.5	1	100.0	0.0
MDR *P. aeruginosa* isolates (331)				
Ceftazidime-avibactam	4	16	84.9	15.1
Ceftolozane-tazobactam	2	8	86.4	7.3
Piperacillin-tazobactam	64	>128	23.0	45.9
Ceftazidime	16	>32	34.7	48.9
Cefepime	16	>16	31.1	32.0
Meropenem	8	32	18.1	68.6
Doripenem	8	>8	18.5	69.7
Levofloxacin	8	>16	20.0	61.2
Gentamicin	8	>16	41.7	31.7
Amikacin	8	>32	81.6	10.3
Tobramycin	2	>16	73.1	22.4
Colistin	0.5	1	100.0	0.0

[a] Criteria as published by CLSI 2018.
[b] Intermediate interpreted as susceptible-dose dependent.
[c] FDA breakpoints published 2017-DEC-13.
[d] Percentage of wild type based on ECV. CLSI M100 (2018).
[e] Organisms include: *Citrobacter freundii* species complex (1), *C. koseri* (2), *Enterobacter cloacae* species complex (12), *Escherichia coli* (87), *Klebsiella oxytoca* (4), *K. pneumoniae* (69), *Proteus mirabilis* (1).

- Ceftazidime-avibactam and ceftolozane-tazobactam were very active against *P. aeruginosa* and exhibited similar coverage against these organisms (96.2%S and 96.5%S, respectively), including meropenem-nonsusceptible (88.1%S and 89.4%S), MDR (84.9%S and 86.4%S), and XDR (79.4%S and 80.4%S) isolates (Table 2 and Figure 3)
- Among *P. aeruginosa* isolates nonsusceptible to ceftazidime, meropenem, and piperacillin-tazobactam, susceptibility to ceftazidime-avibactam, ceftolozane-tazobactam, and amikacin was 73.7%, 76.6%, and 82.6%, respectively (data not shown)
- Among isolates from VABP, susceptibility to ceftazidime-avibactam and ceftolozane-tazobactam was 100.0% and 90.2% for *Enterobacteriaceae* (n=266) and 97.8% and 99.5% for *P. aeruginosa* (n=183), respectively (data not shown)

CONCLUSIONS

- Ceftazidime-avibactam demonstrated potent activity against a large US collection (n=3,202) of contemporary *Enterobacteriaceae* and *P. aeruginosa* isolates from patients with pneumonia, including organisms resistant to most currently available agents, such as CRE and meropenem-nonsusceptible *P. aeruginosa*
- Ceftazidime-avibactam and ceftolozane-tazobactam showed similar coverage (%S) against *P. aeruginosa* (96.2%S vs. 96.5%S), including against MDR (84.9%S vs. 86.4%S) and XDR (79.4%S vs. 80.4%S) isolates
- Ceftolozane-tazobactam was less active than ceftazidime-avibactam against *Enterobacteriaceae* in general and exhibited limited activity against resistant subsets
- Ceftazidime-avibactam represents a valuable option for treating patients hospitalized with pneumonia in US medical centers

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the INFORM Program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi. Accessed January 2017.
2. Castanheira M, Mendes RE, Jones RN, Sader HS (2016). Changes in the frequencies of beta-lactamase genes among *Enterobacteriaceae* isolates in U.S. Hospitals, 2012 to 2014: Activity of ceftazidime-avibactam tested against beta-Lactamase-producing isolates. *Antimicrob Agents Chemother* 60: 4770-4777.
3. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition.* Wayne, PA: CLSI.
5. Magiorakos AP, Srinivasan A, Carey RB, Carmeli Y, Falagas ME, Giske CG, Harbarth S, Hindler JF, Kahlmeter G, Olsson-Liljequist B, Paterson DL, Rice LB, Stelling J, Struelens MJ, Vatopoulos A, Weber JT, Monnet DL (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268-281.
6. Sader HS, Castanheira M, Mendes RE, Flamm RK (2018). Frequency and antimicrobial susceptibility of gram-negative bacteria isolated from patients with pneumonia hospitalized in ICUs of US medical centres (2015–17). *J Antimicrob Chemother* [in press].

Visit www.allergancongressposters.com/814333

IDWeek 2018, October 3–7, 2018 in San Francisco, California