IDWeek 2018
Poster #1000

Antimicrobial Susceptibility of Gram-Positive Bacteria Isolated from Patients Hospitalized with Bacteremia in United States and European Medical Centers: Results from the International Dalbavancin Evaluation of Activity (IDEA) Program

Helio S. Sader[1], Robert K. Flamm[1], Urania Rappo[2], Dmitri Debabov[2], Mariana Castanheira[1], Rodrigo E. Mendes[1]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Allergan, Irvine, California, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit www.allergancongressposters.com/683917

Charges may apply.
No personal information is stored.

INTRODUCTION

- Bloodstream infections (BSIs) are a major cause of morbidity and mortality worldwide
- The etiology of BSIs may vary significantly according to the type of patient and source of infection
- Dalbavancin belongs to the lipoglycopeptide class of antimicrobial agents that act by interrupting bacterial cell wall synthesis resulting in bacterial death, and it was approved in the United States (USA; 2014) and Europe (EUR; 2015) to treat adults with acute bacterial skin and skin structure infections (ABSSSIs) caused by susceptible isolates of *Staphylococcus aureus*, including methicillin-resistant (MRSA) and -susceptible *S. aureus* (MSSA), *Streptococcus pyogenes*, *Streptococcus agalactiae*, *Streptococcus dysgalactiae*, *Streptococcus anginosus* group, and vancomycin-susceptible *Enterococcus faecalis*
- Dalbavancin allows for convenient parenteral administration, which can be a single dose of 1500 mg or a dose of 1000 mg followed by 500 mg a week later for treating ABSSSI
- Dalbavancin is not licensed for treating BSI, but is potentially important in treating infections due to highly resistant gram-positive cocci (GPC)
- We evaluated dalbavancin *in vitro* activity and potency when tested against a large collection of GPC isolates collected from patients with BSIs

MATERIALS AND METHODS

Bacterial isolates

- A total of 8,296 unique GPC were consecutively collected from 33 United States (n=4,409) and 39 European (n=3,887) medical centers in 2015–2017 by the International Dalbavancin Evaluation of Activity (IDEA) Program
- Isolates were determined to be clinically significant based on local guidelines and were submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA)
- Participating laboratories initially identified isolates and JMI confirmed bacterial identifications by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany)

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility (S) by broth microdilution following guidelines in the Clinical and Laboratory Standards Institute (CLSI) M07 document (2018)
- CLSI (2018)-approved dalbavancin breakpoints (≤0.25 mg/L) and breakpoint criteria for comparator agents for indicated species were applied
- Quality assurance was performed by concurrently testing CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213, *E. faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619)

RESULTS

- The most common organisms were *S. aureus* (48.3% in USA, 44.3% in EUR), coagulase-negative staphylococci (CoNS; 14.3% in USA, 15.6% in EUR), and *E. faecalis* (11.5% in USA, 13.1% in EUR; Figure 1)
- All *S. aureus* isolates were susceptible to dalbavancin ($MIC_{50/90}$, 0.03/0.03 mg/L), linezolid ($MIC_{50/90}$, 1/2 mg/L), vancomycin ($MIC_{50/90}$, 1/1 mg/L), and teicoplanin ($MIC_{50/90}$, ≤0.5/≤0.5 mg/L); >99.9% were susceptible to daptomycin ($MIC_{50/90}$, 0.25/0.5 mg/L; Tables 1 and 2)
- Based on MIC_{50}, dalbavancin ($MIC_{50/90}$, 0.03/0.03 mg/L) was 8-fold more active than daptomycin ($MIC_{50/90}$, 0.25/0.5 mg/L) and 32-fold more active than vancomycin ($MIC_{50/90}$, 1/1 mg/L) against *S. aureus*, and dalbavancin activity was not adversely affected by oxacillin resistance (Table 2)
- Among CoNS isolates, 99.9% were inhibited at a dalbavancin MIC of ≤0.25 mg/L ($MIC_{50/90}$, 0.03/0.06 mg/L; Table 1); susceptibility to daptomycin ($MIC_{50/90}$, 0.5/0.5 mg/L), linezolid ($MIC_{50/90}$, 0.5/1 mg/L), vancomycin ($MIC_{50/90}$, 1/2 mg/L), and teicoplanin ($MIC_{50/90}$, 2/4 mg/L) were 100.0%, 96.7%, 100.0%, and 98.3%, respectively (Table 2)
- Among *E. faecalis* isolates, 97.7% were dalbavancin-susceptible (96.4% in USA, 99.0% in EUR; $MIC_{50/90}$, 0.03/0.06 mg/L), 97.5% were vancomycin-susceptible (96.1% in USA, 99.0% in EUR; $MIC_{50/90}$, 1/2 mg/L), and all isolates were susceptible to ampicillin ($MIC_{50/90}$, 1/1 mg/L), daptomycin ($MIC_{50/90}$, 0.5/1 mg/L), and linezolid ($MIC_{50/90}$,1/2 mg/L; Table 2)
- Among *E. faecium* isolates (n=656; 7.9% overall), 63.9% were inhibited at ≤0.25 mg/L of dalbavancin (33.4% in USA, 87.5% in EUR) and 61.6% were vancomycin-susceptible (32.8% in USA, 84.0% in EUR; Tables 1 and 2)
- Dalbavancin was highly active against β-hemolytic streptococci (BHS; n=686 [8.3% overall]; $MIC_{50/90}$, 0.015/0.03 mg/L) and viridans group streptococci (VGS; n=432 [5.2% overall]; $MIC_{50/90}$, 0.015/0.03 mg/L; Tables 1 and 2)

Figure 1 Frequency of gram-positive bacteria isolated from patients with BSIs (SENTRY Program, 2015–2017)

A. United States **B.** Europe

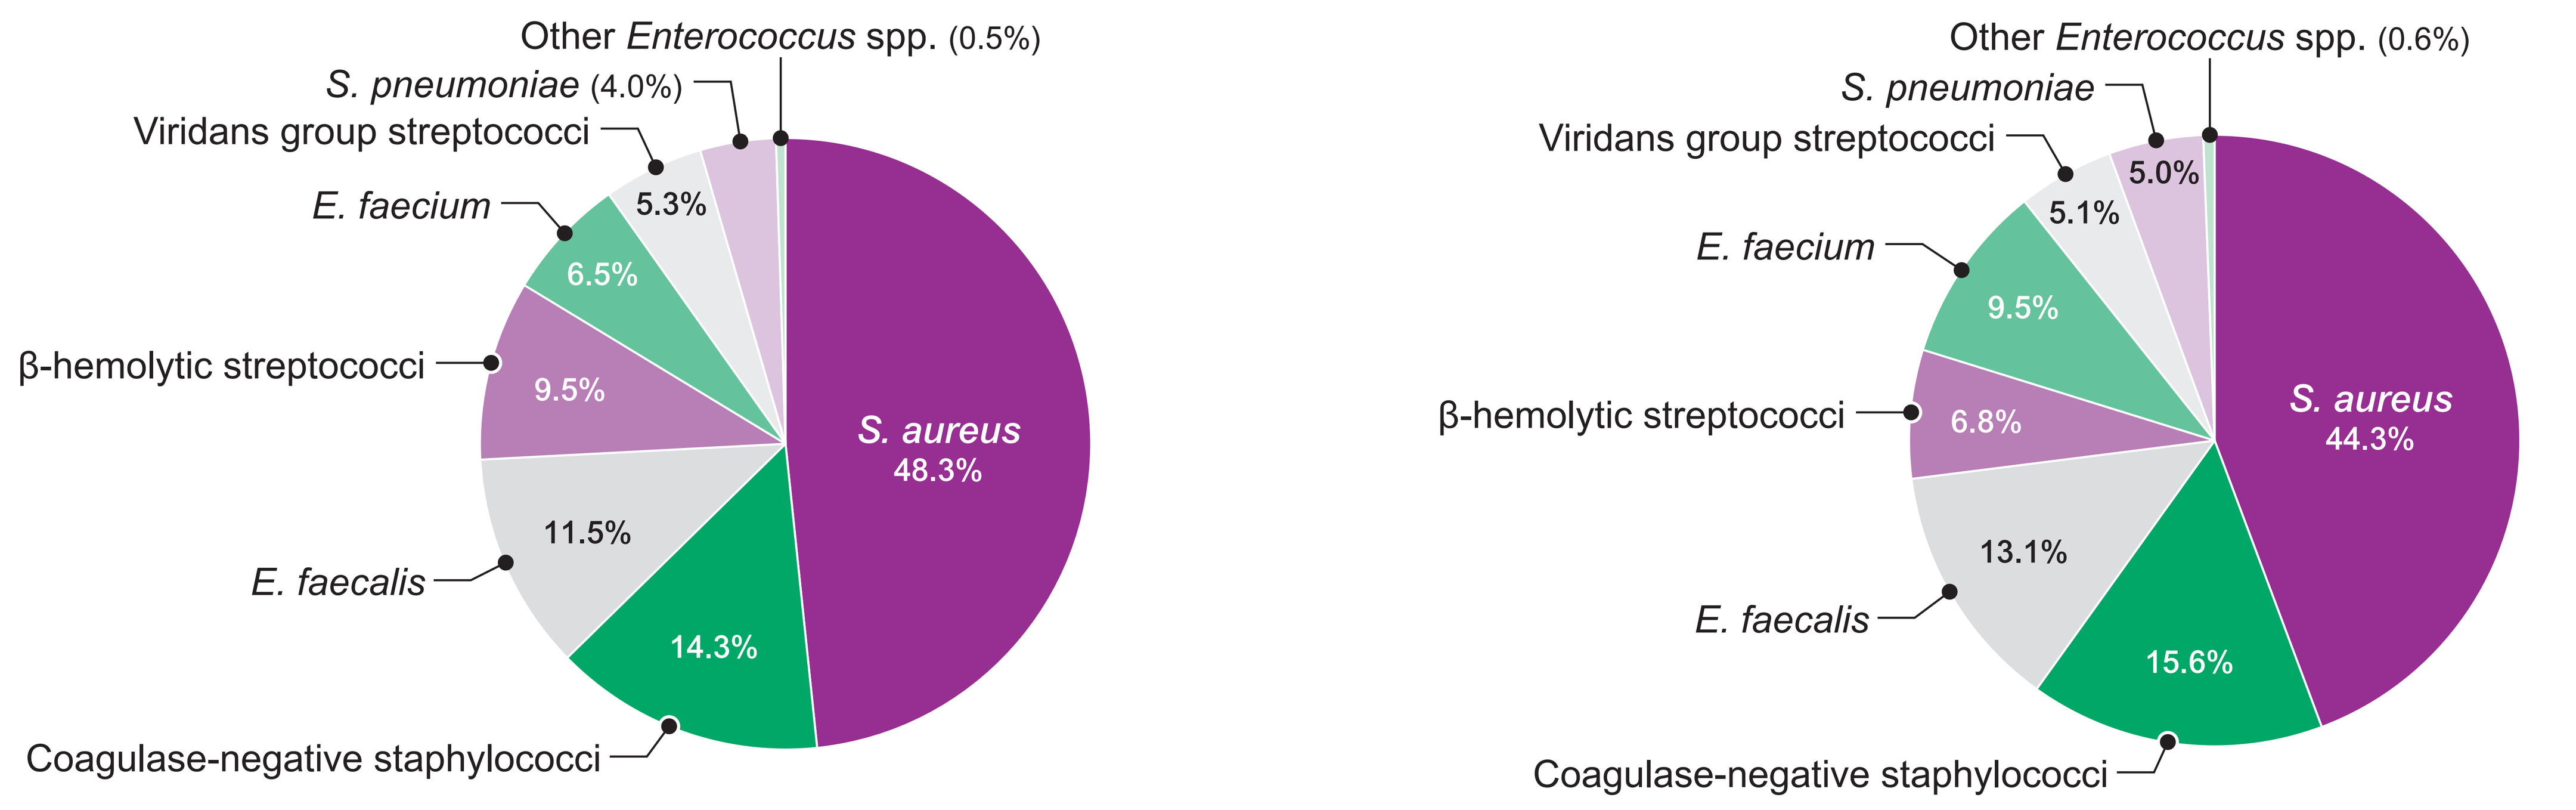
Other *Enterococcus* spp. (0.5%)
S. pneumoniae (4.0%)
Viridans group streptococci
5.3%
E. faecium
6.5%
β-hemolytic streptococci
9.5%
S. aureus
48.3%
11.5%
E. faecalis
14.3%
Coagulase-negative staphylococci
Other *Enterococcus* spp. (0.6%)
S. pneumoniae
Viridans group streptococci
5.0%
5.1%
E. faecium
9.5%
β-hemolytic streptococci
6.8%
S. aureus
44.3%
13.1%
E. faecalis
15.6%
Coagulase-negative staphylococci

CONCLUSIONS

- Dalbavancin was very active against *S. aureus*, CoNS, vancomycin-susceptible enterococci, BHS, and VGS isolated from patients with BSIs
- Based on MIC_{50} values, dalbavancin was generally 8- to 32-fold more active than daptomycin and vancomycin against these organisms
- These results support further investigations to determine the role of dalbavancin in the treatment of BSI

Table 1 Antimicrobial activity of dalbavancin tested against the main organisms and organism groups from United States and Europe combined

Organism/organism group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of:												MIC_{50}	MIC_{90}
	≤0.002	0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	>[a]		
Staphylococcus aureus (3,852)	0 0.0	4 0.1	24 0.7	418 11.6	3,060 91.0	332 99.6	12 99.9	2 100.0					0.03	0.03
Coagulase-negative staphylococci (1,235)	1 0.1	8 0.7	34 3.5	280 26.2	655 79.2	200 95.4	53 99.7	3 99.9				1 100.0	0.03	0.06
Staphylococcus epidermidis (755)	0 0.0	2 0.3	8 1.3	153 21.6	473 84.2	110 98.8	8 99.9	0 99.9				1 100.0	0.03	0.06
Staphylococcus haemolyticus (110)		0 0.0	1 0.9	2 2.7	18 19.1	46 60.9	40 97.3	3 100.0					0.06	0.12
Staphylococcus hominis (182)		0 0.0	1 0.5	66 36.8	98 90.7	16 99.5	1 100.0						0.03	0.03
Enterococcus spp. (1,720)				166 9.7	790 55.6	374 77.3	109 83.7	21 84.9	1 84.9	3 85.1	12 85.8	244 100.0	0.03	>2
Enterococcus faecalis (1,018)				118 11.6	667 77.1	197 96.5	11 97.5	2 97.7	0 97.7	0 97.7	1 97.8	22 100.0	0.03	0.06
Enterococcus faecium (656)				42 6.4	109 23.0	166 48.3	85 61.3	17 63.9	1 64.0	3 64.5	11 66.2	222 100.0	0.12	>2
Other *Enterococcus* spp. (46)				6 13.0	14 43.5	11 67.4	13 95.7	2 100.0					0.06	0.12
β-hemolytic streptococci (686)		121 17.6	207 47.8	257 85.3	75 96.2	24 99.7	2 100.0						0.015	0.03
Streptococcus agalactiae (298)		5 1.7	59 21.5	180 81.9	42 96.0	12 100.0							0.015	0.03
Streptococcus dysgalactiae (153)		15 9.8	59 48.4	43 76.5	24 92.2	10 98.7	2 100.0						0.015	0.03
Streptococcus pyogenes (235)		101 43.0	89 80.9	34 95.3	9 99.1	2 100.0							0.008	0.015
Viridans group streptococci (432)		82 19.0	91 40.0	111 65.7	116 92.6	24 98.1	8 100.0						0.015	0.03
Streptococcus anginosus group (64)		59 92.2	5 100.0										≤0.004	≤0.004
Streptococcus bovis group (62)		6 9.7	10 25.8	19 56.5	27 100.0								0.015	0.03
Streptococcus mitis group (245)		14 5.7	60 30.2	75 60.8	75 91.4	15 97.6	6 100.0						0.015	0.03
Streptococcus mutans group (7)			0 0.0	1 14.3	5 85.7	1 100.0							0.03	
Streptococcus salivarius group (54)		3 5.6	16 35.2	16 64.8	9 81.5	8 96.3	2 100.0						0.015	0.06
Streptococcus pneumoniae (371)		5 1.3	103 29.1	240 93.8	23 100.0								0.015	0.015

[a] Greater than the highest concentration tested.

Table 2 Antimicrobial activity of dalbavancin and comparator agents tested against 8,296 gram-positive cocci isolated from patients with BSIs in the United States (USA, n=4,409) and Europe (EUR; n=3,887)

Organism/ antimicrobial agent (no. tested in USA/ EUR)	MIC_{50}[a]	MIC_{90}[a]	USA		EUR	
			%S[a]	%R[a]	%S[a]	%R[a]
S. aureus (2,130/1,722)						
Dalbavancin	0.03	0.03	100.0		100.0	
Teicoplanin	≤0.5	≤0.5	100.0	0.0	100.0	0.0
Vancomycin	1	1	100.0	0.0	100.0	0.0
Daptomycin	0.25	0.5	>99.9		100.0	
Linezolid	1	2	100.0	0.0	100.0	0.0
Oxacillin	0.5	>2	58.4	41.6	74.8	25.2
Clindamycin	≤0.25	>2	85.1	14.6	93.6	6.4
Levofloxacin	0.25	>4	62.2	37.0	76.2	23.4
Tetracycline	≤0.5	≤0.5	96.2	3.1	94.3	5.0
TMP-SMX[b]	≤0.5	≤0.5	97.6	2.4	99.2	0.8
CoNS (629/606)						
Dalbavancin	0.03	0.06	(100.0)[c]		(100.0)[c]	
Teicoplanin	2	4	98.3	0.0	98.8	0.0
Vancomycin	1	2	100.0	0.0	100.0	0.0
Daptomycin	0.5	0.5	100.0		99.8	
Linezolid	0.5	1	96.7	3.3	98.5	1.5
Oxacillin	>2	>2	34.3	65.7	28.9	71.1
Clindamycin	≤0.25	>2	65.2	33.4	71.1	26.6
Levofloxacin	2	>4	50.9	47.4	42.4	52.1
Tetracycline	≤0.5	>8	83.3	15.9	88.1	9.7
TMP-SMX	≤0.5	>4	65.3	34.7	63.7	36.3
E. faecalis (507/511)						
Dalbavancin	0.03	0.06	96.4[d]		99.0[d]	
Teicoplanin	≤2	≤2	96.4	3.6	99.0	1.0
Vancomycin	1	2	96.1	3.9	99.0	1.0
Daptomycin	0.5	1	100.0		100.0	
Linezolid	1	2	100.0	0.0	100.0	0.0
Ampicillin	1	1	100.0	0.0	100.0	0.0
Levofloxacin	1	>4	75.3	24.7	68.9	29.9
E. faecium (287/369)						
Dalbavancin	>2	>2	(33.4)[d]		(87.5)[d]	
Teicoplanin	≤2	>16	34.5	58.9	87.5	10.3
Vancomycin	1	>16	32.8	67.2	84.0	14.9
Daptomycin	1	2	98.6		100.0	
Linezolid	1	2	99.0	0.7	98.9	0.5
Ampicillin	>8	>8	18.8	81.2	9.2	90.8
Levofloxacin	>4	>4	11.1	82.2	4.9	91.9

Organism/ antimicrobial agent (no. tested in USA/ EUR)	MIC_{50}[a]	MIC_{90}[a]	USA		EUR	
			%S[a]	%R[a]	%S[a]	%R[a]
β-hemolytic streptococci (420/266)						
Dalbavancin	0.015	0.03	100.0[e]		100.0[e]	
Vancomycin	0.5	0.5	100.0		100.0	
Daptomycin	0.12	0.25	100.0		100.0	
Linezolid	1	2	100.0		100.0	
Ceftriaxone	≤0.03	0.06	100.0		100.0	
Penicillin	≤0.03	0.06	100.0		100.0	
Clindamycin	≤0.25	>2	81.7	17.6	90.6	9.0
Levofloxacin	0.5	1	99.3	0.2	98.5	1.5
Tetracycline	1	>4	48.8	49.3	56.0	43.2
Viridans group streptococci (235/197)						
Dalbavancin	0.015	0.03	100.0[e]		100.0[e]	
Vancomycin	0.5	0.5	100.0		100.0	
Daptomycin	0.25	0.5	100.0		100.0	
Linezolid	1	1	100.0		100.0	
Ceftriaxone	0.12	1	93.2	4.7	96.4	1.5
Penicillin	0.06	1	67.7	5.1	78.2	1.5
Clindamycin	≤0.25	>2	86.4	12.8	91.9	8.1
Levofloxacin	1	>4	87.7	11.5	97.0	1.5
Tetracycline	1	>4	61.3	35.3	70.1	27.9
S. pneumoniae (177/194)						
Dalbavancin	0.015	0.015	100.0[e]		100.0[e]	
Vancomycin	0.25	0.5	100.0		100.0	
Linezolid	1	2	100.0		100.0	
Ceftriaxone	0.03	0.5	92.7[f] 98.9[g]	1.1[f] 0.0[g]	91.2[f] 95.9[g]	4.1[f] 1.5[g]
Penicillin	≤0.06	1	75.1[h] 98.3[i]	6.2[h] 0.0[i]	83.5[h] 94.8[i]	8.2[h] 0.0[i]
Clindamycin	≤0.25	≤0.25	90.3	9.1	87.1	12.9
Erythromycin	0.03	>2	64.4	33.9	82.5	17.0
Levofloxacin	1	1	100.0	0.0	99.0	1.0
Tetracycline	≤0.25	>4	85.9	14.1	83.0	17.0
TMP-SMX	≤0.5	2	82.5	6.8	77.3	14.9

[a] MIC_{50} and MIC_{90} values for the US and EUR collection combined; criteria as published by CLSI 2018.
[b] TMP-SMX, trimethoprim-sulfamethoxazole.
[c] *S. aureus* breakpoint was applied for comparison purpose only.
[d] Breakpoint applied to all *E. faecalis* and *E. faecium* isolates, but approved for vancomycin-susceptible *E. faecalis* isolates only.
[e] Breakpoint applied to all *Streptococcus* spp., but approved for *S. pyogenes*, *S. agalactiae*, *S. dysgalactiae*, and *S. anginosus* group only.
[f] Using meningitis breakpoints.
[g] Using non-meningitis breakpoints.
[h] Using oral breakpoints.
[i] Using parenteral, non-meningitis breakpoints

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the IDEA and the SENTRY programs for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.
3. Dalvance® (2018). Available at https://www.allergan.com/assets/pdf/dalvance_pi. Accessed 22 August 2018.
4. Pfaller MA, Mendes RE, Duncan LR, et al. (2018). Activity of dalbavancin and comparator agents against gram-positive cocci from clinical infections in the USA and Europe 2015-16. *J Antimicrob Chemother in press*.
5. Timsit JF, Soubirou JF, Voiriot G, et al. (2014). Treatment of bloodstream infections in ICUs. *BMC Infect Dis* 14: 489.
6. Tobudic S, Forstner C, Burgmann H, et al. (2018). Dalbavancin as primary and sequential treatment for gram-positive infective endocarditis: 2-year experience at the General Hospital of Vienna. *Clin Infect Dis* 67: 795-798.
7. Vincent LL, Otto CM (2018). Infective endocarditis: Update on epidemiology, outcomes, and management. *Curr Cardiol Rep* 20: 86.

IDWeek 2018, October 3–7, 2018, San Francisco, California