IDWeek 2018
Poster #1081

Antimicrobial Activity of Dalbavancin Tested against Gram-Positive Organisms Isolated from Patients with Infective Endocarditis in United States and European Medical Centers

Helio S. Sader, Rodrigo E. Mendes, Michael A. Pfaller, Robert K. Flamm

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit www.allergancongressposters.com/294795

Charges may apply.
No personal information is stored.

INTRODUCTION

- Despite improvements in its management, infective endocarditis (IE) remains associated with high mortality and severe complications
- Managing IE requires an aggressive and prolonged treatment approach with specific antimicrobial treatment or a combination of effective antibiotics and surgery to control the infectious source
- Dalbavancin belongs to the lipoglycopeptide class of antimicrobial agents that act by interrupting bacterial cell wall synthesis resulting in bacterial death, and it was approved in the United States (2014) and Europe (2015) to treat adults with acute bacterial skin and skin structure infections (ABSSSIs) caused by susceptible isolates of *Staphylococcus aureus*, including methicillin-resistant (MRSA) and -susceptible *S. aureus* (MSSA), *Streptococcus pyogenes*, *Streptococcus agalactiae*, *Streptococcus dysgalactiae*, *Streptococcus anginosus* group, and vancomycin-susceptible *Enterococcus faecalis*
- Dalbavancin allows for very convenient parenteral administration, which can be a single dose of 1500 mg or a dose of 1000 mg followed by 500 mg a week later for treating ABSSSI
- Dalbavancin is not licensed for treating IE, but is potentially important in treating infections due to highly resistant gram-positive cocci (GPC)
- We evaluated dalbavancin *in vitro* activity and potency when tested against a large collection of GPC isolates responsible for IE

MATERIALS AND METHODS

Bacterial isolates

- A total of 626 organisms recovered from patients with a diagnosis of bacterial endocarditis in the United States (n=222) and Europe (n=404) by the SENTRY Antimicrobial Surveillance Program (2007–2017) were included in this investigation
- Isolates were determined to be clinically significant based on local guidelines and were submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA)
- Participating laboratories initially identified isolates and JMI confirmed bacterial identifications by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany)

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following guidelines in the CLSI M07 document (2018)
- CLSI (2018)-approved dalbavancin breakpoints (≤0.25 mg/L) and breakpoint criteria for comparator agents for indicated species were applied
- Quality assurance was performed by concurrently testing CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213, *E. faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619)

RESULTS

- *S. aureus* (48.4%) was the most common pathogen associated with IE, followed by *E. faecalis* (19.6%), and viridans group streptococci (VGS; 12.5%), coagulase-negative staphylococci (CoNS; 7.3%), and *E. faecium* (5.6%; Figure 1)
- Dalbavancin (MIC_{50} and MIC_{90}, 0.06 mg/L) and daptomycin ($MIC_{50/90}$, 0.25/0.5 mg/L) showed complete activity (100.0%S) against *S. aureus*, but dalbavancin MIC values were 4- to 8-fold lower than those of daptomycin (Tables 1 and 2)
- Linezolid ($MIC_{50/90}$, 1/2 mg/L), vancomycin (MIC_{50} and MIC_{90}, 1 mg/L), and teicoplanin (MIC_{50} and MIC_{90}, ≤2 mg/L) were also active against all *S. aureus* (Table 2)

Table 1. Dalbavancin MIC distribution when tested against 626 gram-positive organisms causing infective endocarditis (SENTRY Program, 2007–2017)

Organism (no. tested)	No. of isolates (cumulative %) at MIC (mg/L) of:					MIC_{50} (mg/L)	MIC_{90} (mg/L)
	≤0.03	0.06	0.12	0.25	>0.25		
S. aureus (303)	140	142	20	1		0.06	0.06
	(46.2)	(93.1)	(99.7)	(100.0)			
MSSA (202)	88	102	12			0.06	0.06
	(43.6)	(94.1)	(100.0)				
MRSA (101)	52	40	8	1		≤0.03	0.06
	(51.5)	(91.1)	(99.0)	(100.0)			
E. faecalis (123)	61	50	8	1	2	0.06	0.06
	(49.6)	(90.2)	(96.7)	(97.6)	(100.0)		
VGS (78)	70	6	2				
	(89.7)	(97.4)	(100.0)			≤0.03	0.06
CoNS (46)	24	14	8				
	52.20)	82.60)	100.00)			≤0.03	0.12
E. faecium (35)	6	6	9	2	12		
	(17.1)	(34.3)	(60.0)	(65.7)	(100.0)	0.12	>0.25
BHS (24)	20	3	1				
	(83.3)	(95.8)	(100.0)			≤0.03	0.06
S. pneumoniae (15)	15						
	(100)					≤0.03	≤0.03
All organisms (626)	338	221	48	4	15		
	(54.0)	(89.3)	(97.0)	(97.6)	(100.0)	≤0.03	0.12

Abbreviations: MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; VGS, viridans group streptococci; CoNS, coagulase-negative staphylococci; BHS, β-hemolytic streptococci.

- Ceftaroline exhibited potent activity against MSSA (MIC_{50} and MIC_{90}, 0.25 mg/L; 100.0%S) and inhibited 78.4% of MRSA isolates ($MIC_{50/90}$, 1/2 mg/L) at ≤1 mg/L (Table 2)
- All *E. faecalis* isolates were susceptible to ampicillin ($MIC_{50/90}$, ≤1/2 mg/L), daptomycin ($MIC_{50/90}$, 1/2 mg/L), and linezolid ($MIC_{50/90}$, 1/2 mg/L), whereas 97.6% (120/123) of isolates were susceptible to dalbavancin (MIC_{50} and MIC_{90}, 0.06 mg/L) and vancomycin ($MIC_{50/90}$, 1/2 mg/L; Table 2)
- Against *E. faecalis*, dalbavancin MIC values (MIC_{50} and MIC_{90}, 0.06 mg/L) were 16- to 32-fold lower than daptomycin and vancomycin ($MIC_{50/90}$, 1/2 mg/L for both compounds; Table 2)
- All VGS and CoNS isolates were susceptible to dalbavancin, daptomycin, vancomycin, and linezolid (Table 2), and the highest ceftaroline MICs were 0.5 mg/L for VGS and 4 mg/L for CoNS (93.5% inhibited at ≤1 mg/L; data not shown)
- Against *E. faecium*, 65.7% of isolates were inhibited at ≤0.25 mg/L of dalbavancin and 62.9% were vancomycin-susceptible (Tables 1 and 2)
- All *E. faecium* were susceptible to daptomycin (MIC_{50} and MIC_{90}, 2 mg/L) and linezolid ($MIC_{50/90}$, 1/2 mg/L; Table 2)
- β-hemolytic streptococci (BHS) were susceptible to most antimicrobial agents
- Only 66.7% of *S. pneumoniae* isolates were susceptible to penicillin at ≤0.06 mg/L (MIC_{50} and MIC_{90}, ≤0.03 mg/L)

CONCLUSIONS

- Dalbavancin exhibited potent *in vitro* activity against a large collection of gram-positive isolates recovered from patients with endocarditis in US and European medical centers
- Dalbavancin MIC values were 4- to 8-fold lower than those of daptomycin and 16-fold lower than those of vancomycin when tested against *S. aureus*
- These results support further investigations to determine the role of dalbavancin in the treatment of infective endocarditis

Table 2. Antimicrobial activity of dalbavancin and comparator agents tested against gram-positive bacteria isolated from patients with infective endocarditis (SENTRY Program, 2007–2017)

Organism/organism group (no. of isolates) Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	CLSI[a] %S	CLSI[a] %R
S. aureus (303)				
Dalbavancin	0.06	0.06	100.0	
Daptomycin	0.25	0.5	100.0	
Teicoplanin	≤2	≤2	100.0	0.0
Vancomycin	1	1	100.0	0.0
Oxacillin	0.5	>2	66.7	33.3
Ceftaroline	0.25	1	92.9	0.0
Levofloxacin	≤0.5	>4	72.9	26.7
Linezolid	1	2	100.0	0.0
Methicillin-resistant *S. aureus* (101)				
Dalbavancin	≤0.03	0.06	100.0	
Daptomycin	0.25	0.5	100.0	
Teicoplanin	≤2	≤2	100.0	0.0
Vancomycin	1	1	100.0	0.0
Oxacillin	>2	>2	0.0	100.0
Ceftaroline	1	2	78.4	0.0
Levofloxacin	>4	>4	28.7	71.3
Linezolid	1	2	100.0	0.0
E. faecalis (123)				
Dalbavancin	0.06	0.06	97.6 [b]	
Daptomycin	1	2	100.0	
Teicoplanin	≤2	≤2	98.4	1.6
Vancomycin	1	2	96.7	3.3
Levofloxacin	2	>4	68.3	29.3
Linezolid	1	2	100.0	0.0
Ampicillin	≤1	2	100.0	0.0
Viridans group streptococci (78)[c]				
Dalbavancin	≤0.03	0.06	100.0 [d]	
Daptomycin	0.25	1	100.0	
Vancomycin	0.5	0.5	100.0	
Levofloxacin	1	2	98.7	0.0
Linezolid	0.5	1	100.0	
Ceftriaxone	≤0.25	0.5	96.2	2.6
Ceftaroline	≤0.015	0.06		
Penicillin	≤0.06	0.5	83.3	1.3
Coagulase-negative staphylococci (46)[e]				
Dalbavancin	≤0.03	0.12		
Daptomycin	0.25	0.5	100.0	
Teicoplanin	≤2	8	97.8	0.0
Vancomycin	1	2	100.0	0.0
Oxacillin	2	>2	32.6	67.4
Ceftaroline	0.25	0.5		
Levofloxacin	0.25	>4	54.3	39.1
Linezolid	0.5	1	100.0	0.0
E. faecium (35)				
Dalbavancin	0.12	>0.25		
Daptomycin	2	2	100.0	
Teicoplanin	≤2	>16	65.7	34.3
Vancomycin	1	>16	62.9	37.1
Levofloxacin	>4	>4	5.7	88.6
Linezolid	1	2	100.0	0.0
Ampicillin	>8	>8	11.4	88.6

[a] Criteria as published by CLSI 2018.
[b] Breakpoint applied to all *E. faecalis*, but approved for vancomycin-susceptible isolates only.
[c] Organisms include: *Streptococcus anginosus* (5), *S. anginosus* group (1), *S. bovis* group (4), *S. cristatus* (1), *S. gallolyticus* (8), *S. gordonii* (5), *S. mitis* group (8), *S. mitis/oralis* (3), *S. mutans* (2), *S. oralis* (8), *S. parasanguinis* (3), *S. salivarius* (4), *S. salivarius* group (1), *S. sanguinis* (13), *S. sinensis* (1), *S. vestibularis* (1), unspeciated viridans group streptococci (10).
[d] Breakpoint applied to all *Streptococcus* spp., but approved for *S. pyogenes*, *S. agalactiae*, *S. dysgalactiae*, and *S. anginosus* group only.
[e] Organisms include: *Staphylococcus capitis* (1), *S. caprae* (1), *S. cohnii* (1), *S. epidermidis* (27), *S. haemolyticus* (6), *S. hominis* (4), *S. lugdunensis* (4), *S. pasteuri* (1), *S. pettenkoferi* (1).

Figure 1. Frequency of gram-positive bacteria isolated from patients with infective endocarditis (SENTRY Program, 2007–2017).

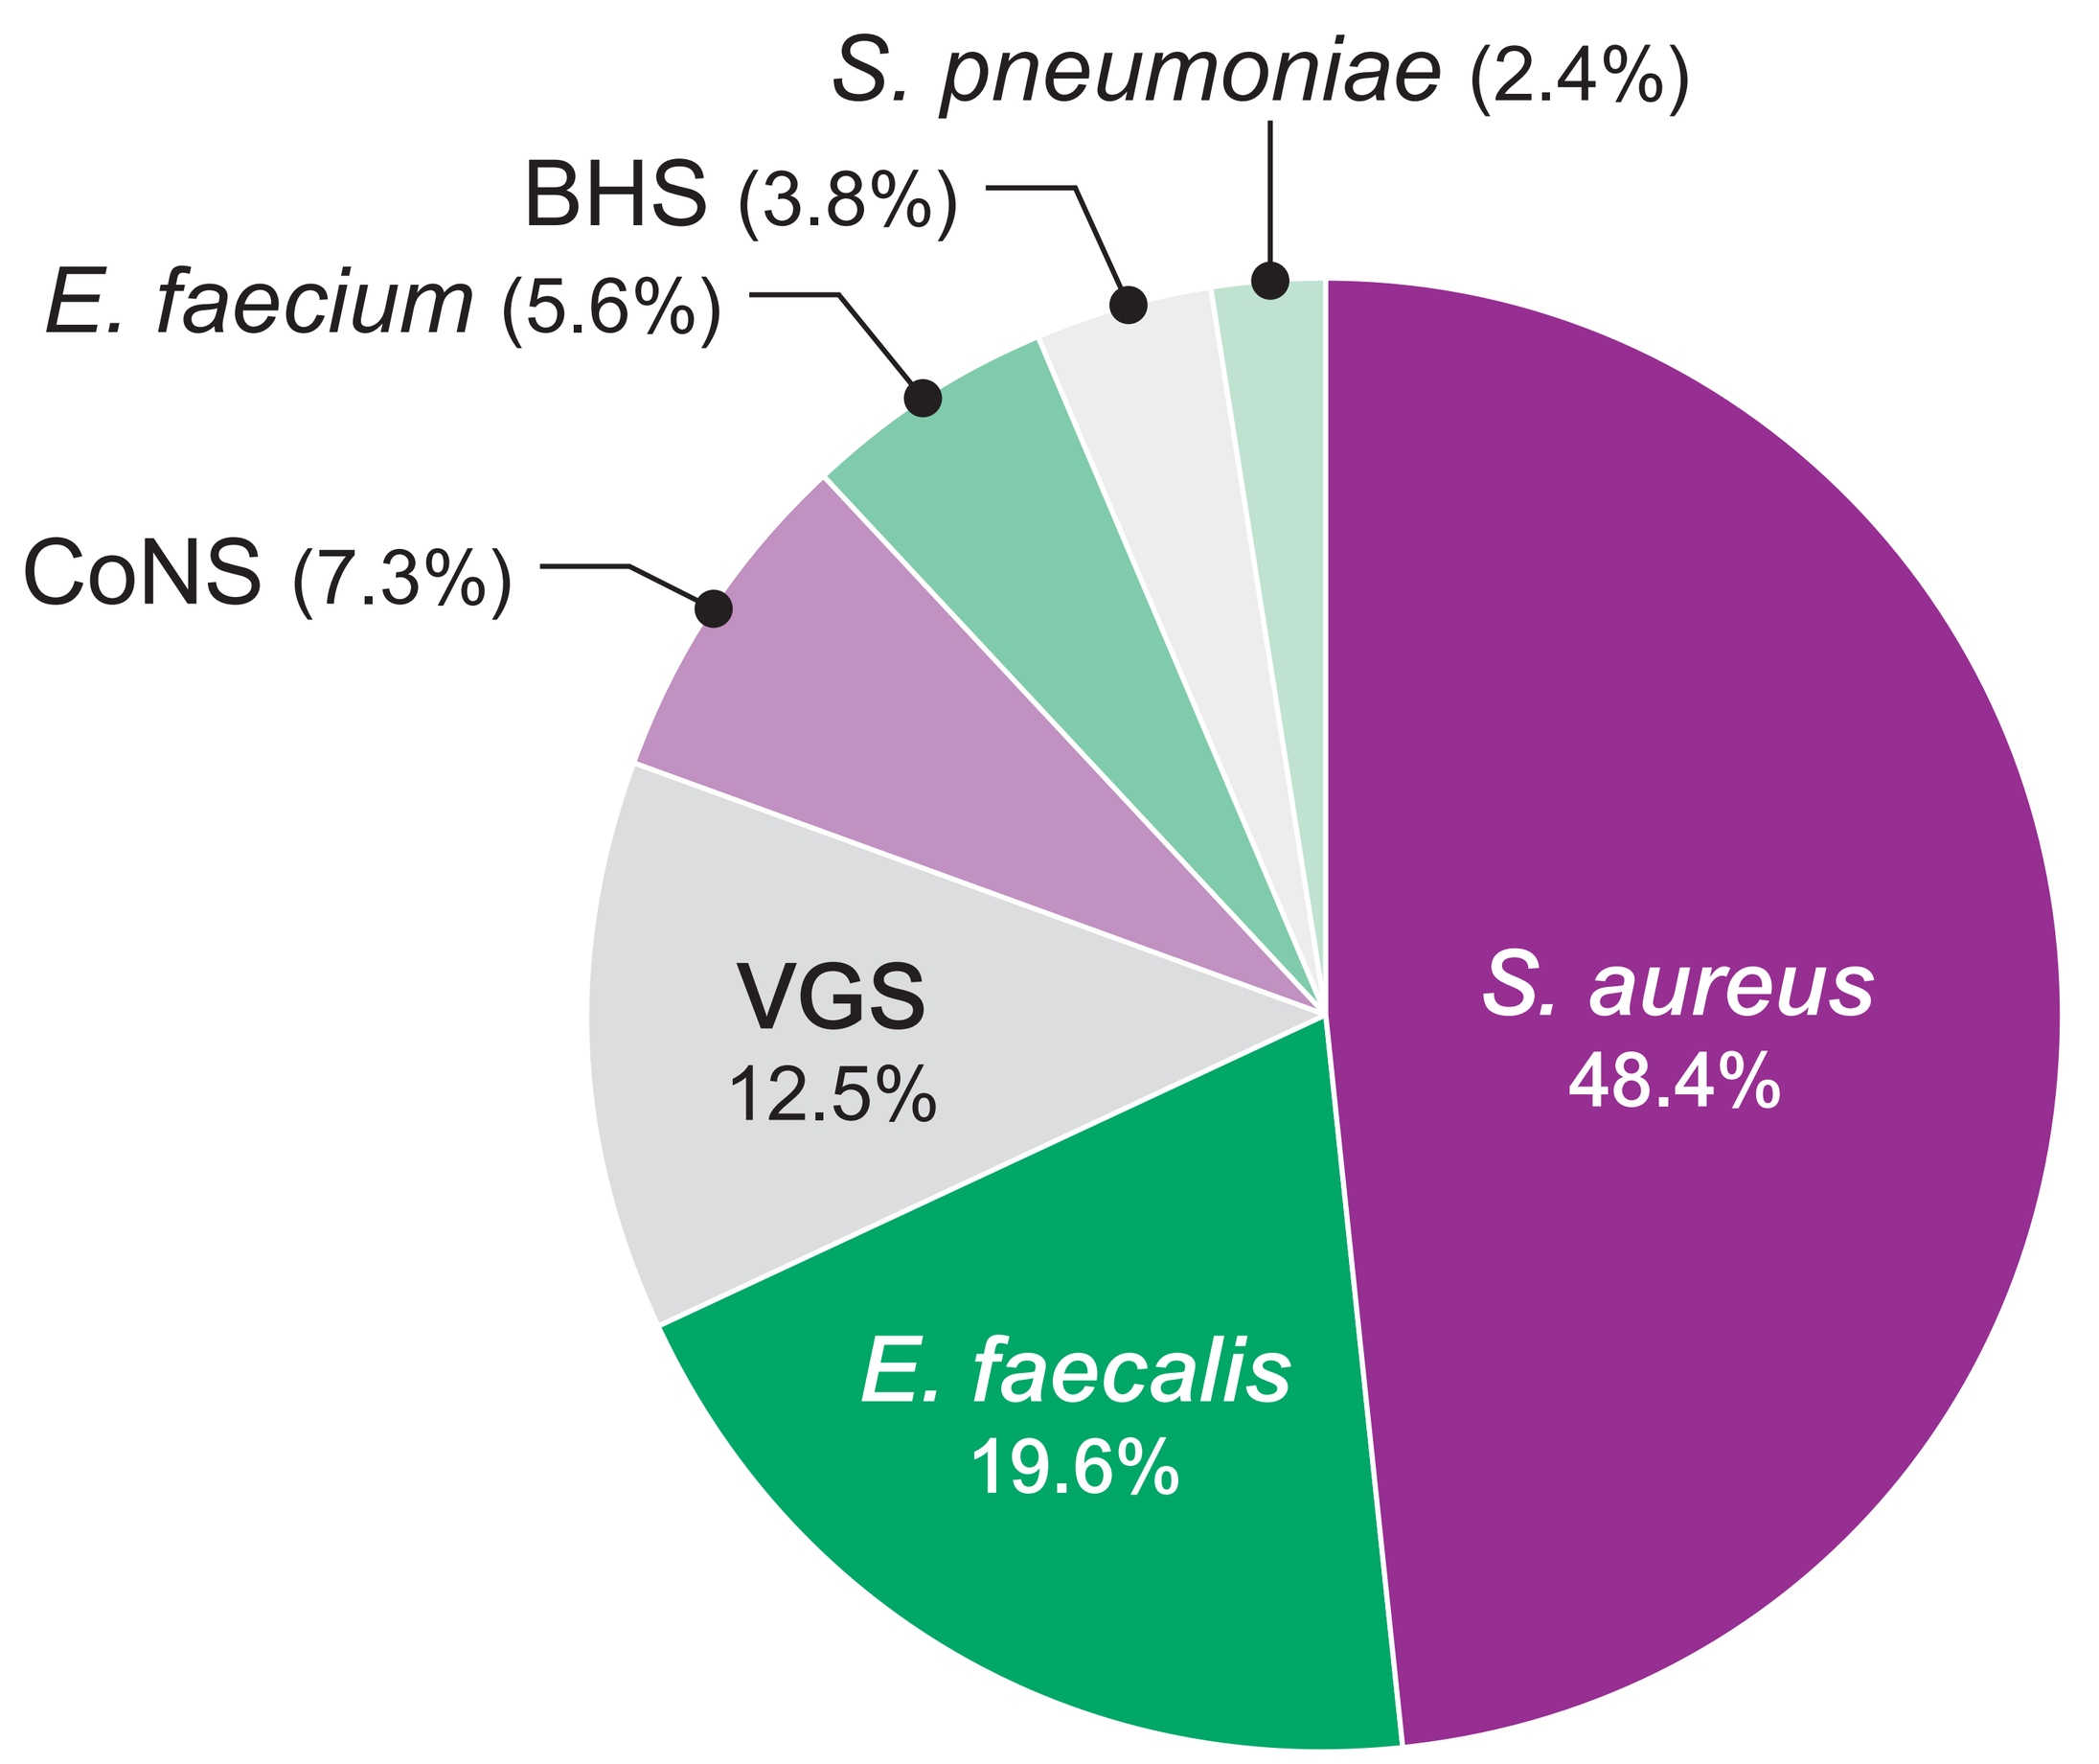
S. pneumoniae (2.4%)
BHS (3.8%)
E. faecium (5.6%)
CoNS (7.3%)
VGS 12.5%
S. aureus 48.4%
E. faecalis 19.6%

Abbreviations: VGS, viridans group streptococci; CoNS, coagulase-negative staphylococci; BHS, β-hemolytic streptococci.

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the International Dalbavancin Evaluation of Activity (IDEA) and the SENTRY programs for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. Clinical and Laboratory Standards Institute (2018). *M100Ed28E. Performance standards for antimicrobial susceptibility testing: 28th informational supplement.* Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.
3. Dalvance® (2018). Available at https://www.allergan.com/assets/pdf/dalvance_pi. Accessed 22 August 2018.
4. Pfaller MA, Mendes RE, Duncan LR, et al. (2018). Activity of dalbavancin and comparator agents against gram-positive cocci from clinical infections in the USA and Europe 2015-16. *J Antimicrob Chemother in press*.
5. Tobudic S, Forstner C, Burgmann H, et al. (2018). Dalbavancin as primary and sequential treatment for gram-positive infective endocarditis: 2-year experience at the General Hospital of Vienna. *Clin Infect Dis* 67: 795-798.
6. Vincent LL, Otto CM (2018). Infective endocarditis: Update on epidemiology, outcomes, and management. *Curr Cardiol Rep* 20: 86.

IDWeek 2018, October 3–7, 2018, San Francisco, California