ID Week 2018

Poster #1364

Efficacy of Omadacycline against Molecularly Characterized Gram-Positive and Gram-Negative Pathogens Causing Infections in the Phase 3 CABP and ABSSSI Clinical Trials

RE MENDES[1], M CASTANHEIRA[2], ES ARMSTRONG[2], JN STEENBERGEN[2], RK FLAMM[1]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2] Paratek Pharmaceuticals, King of Prussia, Pennsylvania, USA

Contact Information:
Rodrigo E. Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://paratekpharma.com/media/1583/idweek-2018-1364-phase3-molec-char-pathogens.pdf

Charges may apply.
No personal information is stored.

Introduction

- Omadacycline represents a new class of tetracycline-related compounds, the aminomethylcyclines, that possesses antibacterial activity from its ability to bind to the 30S subunit of the bacterial ribosome and inhibit bacterial protein synthesis
- Structural modifications at the C-7 and C-9 positions allow omadacycline to overcome the 2 main mechanisms of resistance to tetracyclines: efflux pumps and ribosomal protection (see poster #1377)
- One phase 3 trial for community-acquired bacterial pneumonia (CABP; OPTIC) and 2 phase 3 trials for acute bacterial skin and skin structure infections (ABSSSIs; OASIS-1 and OASIS-2) were completed
- This study provided the molecular characterization of clinical trial isolates and evaluated the efficacy results of omadacycline against subsets of macrolide or doxycycline-nonsusceptible isolates

Materials and Methods

Bacterial organisms

- Gram-positive (n=24) and -negative (n=17) clinical isolates recovered from subjects enrolled in the OPTIC (n=26), OASIS-1 (n=10), or OASIS-2 (n=5) trials were selected for molecular characterization and included in this investigation
- Gram-positive isolates were selected based on the presence of tetracycline and/or macrolide resistance phenotypes; Gram-negative isolates were selected based on nonsusceptibility to tetracycline

Antimicrobial susceptibility testing

- Selected isolates were tested for antimicrobial susceptibility using 96-well frozen-form broth microdilution panels containing cation-adjusted Mueller-Hinton broth and panels were manufactured per the Clinical and Laboratory Standards Institute (CLSI) specifications described in the M07-A10 (2015) document
- MIC values were validated by concurrently testing the appropriate gram-positive and -negative strains recommended by the CLSI M100 documents
 - MIC results obtained against ATCC quality-control strains were interpreted according to published criteria per CLSI M100 guidelines
- MIC values obtained against clinical isolates were interpreted using published CLSI (M100, 2017) breakpoints, when available

Characterizing tetracycline resistance mechanisms

- Whole genome extraction and sequencing
 - Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, Ohio, USA), which was used as input material for library construction
 - DNA libraries were prepared using the Nextera™ library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on a MiSeq Sequencer (JMI Laboratories, North Liberty, Iowa, USA)
- Sequence analysis
 - FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.9.0, and an in-house designed software pipeline was applied to align the assembled sequences against numerous determinants associated with MLS_B phenotype (including rRNA methylases, efflux pumps, and inactivating enzymes) and tetracycline resistance (such as ribosomal protection, enzymatic, and efflux pumps) (Table 1)
 - Multilocus sequence typing (MLST) was performed by extracting previously defined sets of 7 housekeeping gene fragments (~500 bp) for each species included in the study and each fragment was compared to known allele variants for each locus (housekeeping gene) on the MLST website (http://www.mlst.net)

Efficacy analysis

- The efficacy endpoint was investigator's assessment of clinical response at post therapy evaluation (PTE)

Table 1 Genes associated with the MLS_B and tetracycline resistance phenotype investigated

Resistance genes encoding for[a]:					
MLS_B		Inactivating enzymes			
rRNA methylase	Efflux	Esterase	Lyases	Transferases	Phosphorylases
erm(A), (B), (C), (D), (E), (F), (G), (H), (I), (N), (O), (Q), (R), (S), (T), (U), (V), (W), (X), (Y), (Z), (30), (31), (33), (32), (33), (34), (35), (36), (37), (38), (39), (40), (41), (42), (43), (44), (45), (46)	*mef*(A), (B), (C), (En2) *msr*(A), (C), (D), (E) *car*(A) *lmr*(A), (B) *ole*(B), (C) *srm*(B) *tlr*(C) *lsa*(A), (B), (C), (E) *vga*(A), (B), (C), (D), (E) *vgb*, *vgb*(A), (B) *eat*(A) *sal*(A) *mac*(A), (B) *mre*(A)	*ere*(A), (A2), (B), (D)	*vgb*(A), (A)LC, (B)	*lnu*(A), (A'), (AN2), (B), (C), (D), (E), (F), (P) *vat*(A), (B), (C) *vat*(D), (E), (F), (H) *mgt* *ole*(B), (C), (D), (I) *lin*	*mph*(A), (B), (C), (D), (E), (F), (G)
cfr, *cfr*(B), *cfr*(C), *myr*(A), *tsn*(R),	*optrA*				
Tetracycline	**Efflux**	**Ribosomal protection**	**Enzymatic**	**Unknown**	
	tet(A), *tet*(B), *tet*(C), *tet*(D), *tet*(E), *tet*(59), *tet*(G), *tet*(H), *tet*(J), *tet*(V), *tet*(Y), *tet*(Z), *tet*(30), *tet*(31), *tet*(33), *tet*(57), *tet*(35), *tet*(39), *tet*(41), *tet*(K), *tet*(L), *tet*(38), *tet*(45), *tet*(58), *tetA*(P), *tet*(40), *otr*(B), *otr*(C), *tcr*, *tet*(42), *tet*(43), *tetAB*(46), *tetAB*(60)	*tet*(M), *tet*(O), *tet*(S), *tet*(W), *tet*(32), *tet*(Q), *tet*(T), *tet*(36), *otr*(A), *tetB*(P), *tet*, *tet*(44)	*tet*(X), *tet*(37), *tet*(34), *tet*(47), *tet*(48), *tet*(49), *tet*(50), *tet*(51), *tet*(52), *tet*(53), *tet*(54), *tet*(55), *tet*(56)	*tet*(U)	

[a] Adapted from http://faculty.washington.edu/marilynr/ermwebA.pdf.

Table 2 Summary of results obtained from *S. pneumoniae* clinical trial isolates

Isolate	Study	Source isolate no.	Macrolide resistance determinant(s)[a]	MLST		MIC (µg/mL)						Treatment arm	Clinical outcome[b]
				ST	Profile	OMC	AZI	ERY	CLI	DOX	TET		
6	CABP	D604669	*erm*(B), *tet*(M)	ST62	2-5-29-12-16-3-14	0.06	>1	>0.5	>0.5	>8	>4	OMC	Success
7	CABP	E101733	*erm*(B), *tet*(M)	ST62	2-5-29-12-16-3-14	0.03	>1	>0.5	>0.5	8	>4	MXF	Success
8	CABP	E101737	*erm*(B), *tet*(M)	ST179	7-14-40-12-1-1-14	0.03	>1	>0.5	>0.5	8	>4	OMC	Success
9	CABP	D604807	*mef*(A), *msr*(D)	ST13568	1-5-4-5-587-1-8	0.03	>1	>0.5	0.06	0.12	0.25	OMC	Success
10	CABP	D606264	*mef*(A), *msr*(D)	ST9	1-5-4-5-5-1-8	0.03	>1	>0.5	0.12	0.12	0.25	OMC	Success
11	CABP	D759414	*erm*(B), *tet*(M)	ST180	7-15-2-10-6-1-22	0.06	>1	>0.5	0.12	8	>4	MXF	Success
13	CABP	E052867	*erm*(B), *mef*(A), *msr*(D), *tet*(M)	ST320	4-16-19-15-6-20-1	0.03	>1	>0.5	>0.5	2	>4	MXF	Success
14	CABP	D780411	*mef*(A), *msr*(D)	ST100	5-12-29-12-9-39-18	0.03	>1	>0.5	0.06	0.06	0.12	OMC	Success
15	CABP	D791162	*erm*(B), *tet*(M)	ST2467	7-25-4-1-15-20-28	0.03	>1	>0.5	>0.5	2	>4	OMC	Success
16	CABP	D826829	*erm*(B), *tet*(M)	ST2744	7-6-9-2-6-1-68	0.06	>1	>0.5	>0.5	8	>4	OMC	Success
17	CABP	E216193	*erm*(B), *tet*(M)	ST13570	1-597-1-8-11-5-11	0.06	>1	>0.5	>0.5	>8	>4	OMC	Success
18	CABP	D818811	*erm*(B), *tet*(M)	ST4310	32-28-1-1-15-1-14	0.03	>1	>0.5	>0.5	8	>4	OMC	Success
19	CABP	D735404	*tet*(M)	ST2031	7-8-53-6-10-6-14	0.06	0.5	0.06	0.12	4	>4	OMC	Success
20	CABP	E141031	*mef*(A), *msr*(D), *tet*(M)	ST719	15-29-2-21-30-1-14	0.03	>1	>0.5	0.06	8	>4	MXF	Success
21	CABP	E140987	*mef*(A), *msr*(D), *tet*(M)	ST1611	2-19-2-17-15-22-14	0.03	>1	>0.5	0.12	4	>4	MXF	Success
27	ABSSSI	TR2011200870	*erm*(B), *tet*(M)	ST180	7-15-2-10-6-1-22	0.03	>1	>0.5	>0.5	0.06	0.12	OMC	Success

CABP, community-acquired bacterial pneumonia; MLST, multilocus sequence typing; OMC, omadacycline; AZI, azithromycin; ERY, erythromycin; CLI, clindamycin; DOX, doxycycline; TET, tetracycline; MXF, moxifloxacin; ABSSSI, acute bacterial skin and skin structure infection.
[a] Listed gene or a variant with >99% identity detected in each case. *tet*(M) detected in isolate TR2011200870 had a premature stop codon.
[b] The efficacy endpoint was investigator's assessment of clinical response at post therapy evaluation.

Table 3 Summary of results obtained from *S. aureus* clinical trial isolates

Isolate	Study	Source isolate no.	Macrolide resistance determinant(s)[a]	MLST		MIC (µg/mL)					Treatment arm	Clinical outcome[b]
				ST	Profile	OMC	AZI	ERY	CLI	DOX		
1	ABSSSI	R4091141	*tet*(38), *tet*(K)	ST8	3-3-1-1-4-4-3	0.25	2	0.5	0.12	8	OMC	Success
2	ABSSSI	R6041266	*tet*(38), *tet*(M), *erm*(C)	ST8	3-3-1-1-4-4-3	0.5	>4	>4	>4	8	LZD	Success
4	ABSSSI	R7212058	*tet*(38), *tet*(M), *mph*(C), *msr*(A)	ST8	3-3-1-1-4-4-3	0.25	8	>4	0.12	8	LZD	Success
5	CABP	D665005	*tet*(38), *tet*(M), *tet*(L)	ST8	3-3-1-1-4-4-3	0.25	1	0.25	0.12	16	MXF	Success
22	ABSSSI	D661166	*tet*(38), *tet*(M)	ST3081[c]	3-4-1-4-12-1-10	0.25	2	0.5	0.12	8	LZD	Success
24	ABSSSI	EJ14045	*tet*(38), *tet*(M), *erm*(C), *mph*(C), *msr*(A)	ST8	3-3-1-1-4-4-3	0.25	>4	>4	>4	8	LZD	Success
25	ABSSSI	EJ29451	*tet*(38), *tet*(M)	ST4302	1-648-1-8-11-5-11	0.25	2	0.5	0.12	8	LZD	Indeterminate
26	ABSSSI	EJ03946	*tet*(38), *tet*(M)	ST4302	1-648-1-8-11-5-11	0.25	2	0.5	0.12	8	LZD	Indeterminate

ABSSSI, acute bacterial skin and skin structure infection; MLST, multilocus sequence typing; OMC, omadacycline; AZI, azithromycin; ERY, erythromycin; CLI, clindamycin; DOX, doxycycline; MXF, moxifloxacin; LZD, linezolid; CABP, community-acquired bacterial pneumonia.
[a] Listed gene or a variant with >99% identity was detected in each case; *tet*(38) is intrinsic in *S. aureus*.
[b] The efficacy endpoint was investigator's assessment of clinical response at post therapy evaluation.
[c] ST3081 is single locus variant (SLV) of ST5, while ST37 is a SLV of ST36.

Table 4 Summary of results obtained from gram-negative clinical trial isolates

Isolate	Study	Source isolate no.	Organism	*tet* gene[a]	MLST		MIC (µg/mL)					Treatment arm	Clinical outcome[c]
					ST	Profile	OMC	TIG	DOX	MIN	TET		
2	ABSSSI	D661170	*E. coli*	*tet*(A)	372	17-88-103-19-36-23-44-26	2	0.25	8	1	>16	LZD	Success
10	ABSSSI	EJ03755	*E. coli*	*tet*(A)	131	53-40-47-13-36-28-29	1	0.25	8	2	>16	OMC	Success
16	CABP	D670591	*E. coli*	*tet*(A)	131	53-40-47-13-36-28-29	2	0.25	8	2	>16	OMC	Indeterminate
17	CABP	D826823	*E. coli*	*tet*(A)	131	53-40-47-13-36-28-29	1	0.25	8	2	>16	MXF	Success
22	CABP	D604732	*E. coli*	*tet*(B)	73	36-24-9-13-17-11-25	1	0.25	>8	32	>16	OMC	Success
30	ABSSSI	D873248	*E. coli*	*tet*(B)	744	10-11-135-8-8-8-2	0.5	0.12	>8	8	>16	LZD	Success
31	CABP	EK21636	*E. coli*	*tet*(A), *tet*(B)	744	10-11-135-8-8-8-2	2	0.25	>8	8	>16	MXF	Success
32	CABP	D658102	*E. coli*	*tet*(A)	NA[b]	6-4-12-1-20-18-7	2	0.25	>8	2	>16	MXF	Success
4	ABSSSI	D648088	*E. cloacae*	*tet*(D)	NA	59-40-6-9-62-6-6	2	0.5	>8	32	>16	OMC	Success
6	ABSSSI	D687070	*E. cloacae*	*tet*(D)	419	59-9-86-9-67-37-6	2	0.5	>8	32	>16	OMC	Success
8	ABSSSI	D873239	*K. pneumoniae*	*tet*(A)	17	2-1-1-1-4-4-4	2	0.5	>8	4	>16	LZD	Success
13	CABP	D665009	*K. pneumoniae*	*tet*(A)	307	4-1-2-52-1-1-7	8	1	>8	8	>16	OMC	Failure
19	CABP	D944286	*K. pneumoniae*	*tet*(A)	23	2-1-1-1-9-4-12	16	2	>8	>32	>16	OMC	Success
20	CABP	D736002	*K. pneumoniae*	*tet*(A)	NA	2-3-2-1-10-1-43	4	1	>8	8	>16	OMC	Success
21	CABP	D736023	*K. pneumoniae*	*tet*(A)	NA	2-3-2-1-10-1-43	8	1	>8	4	>16	MXF	Success
25	CABP	D969351	*K. pneumoniae*	*tet*(A)	1	4-4-1-1-7-4-10	16	2	>8	32	>16	OMC	Success
28	ABSSSI	R7081262	*K. pneumoniae*	ND	14	1-6-1-1-1-1-1	8	4	>8	16	>16	LZD	Success

ABSSSI, acute bacterial skin and skin structure infection; MLST, multilocus sequence typing; OMC, omadacycline; TIG, tigecycline; DOX, doxycycline; MIN, minocycline; TET, tetracycline; MXF, moxifloxacin; LZD, linezolid; CABP, community-acquired bacterial pneumonia.
[a] ND, *tet* genes investigated were not detected. Wild-type *rpsJ* (S10 rRNA) sequences were observed in all isolates.
[b] NA, ST not assigned yet for this profile.
[c] The efficacy endpoint was investigator's assessment of clinical response at post therapy evaluation.

Results

- All but 1 *S. pneumoniae* carried MLS_B-associated genes, while tetracycline- and doxycycline-nonsusceptible isolates (n=12) had *tet*(M) (Table 2)
- *S. aureus* (8 isolates) exhibited a doxycycline-nonsusceptible phenotype (MIC, 8–16 µg/mL) and omadacycline MIC values of 0.25–0.5 µg/mL (Table 3)
 - All *S. aureus* carried *tet*(M), except for 1 isolate with a *tet*(K) gene and 1 isolate with *tet*(M) and *tet*(L) genes (Table 3)
- All *Escherichia coli* (8 isolates; omadacycline MIC, 0.5–2 µg/mL), *Enterobacter cloacae* (2 isolates; omadacycline MIC, 2 µg/mL), and *Klebsiella pneumoniae* (6 isolates; omadacycline MIC, 2–16 µg/mL) carried tetracycline efflux-pump genes, *tet*(A) and/or *tet*(B), *tet*(D), and *tet*(A) (omadacycline MIC, 0.03–0.06 µg/mL), respectively (Table 4)
 - *tet* genes were not detected in 1 *K. pneumoniae* (omadacycline MIC, 8 µg/mL) isolate
 - A total of 3 *E. coli* isolates belonged to the epidemic ST131 lineage (Table 4)
- Clinical success was noted in 14/16 (87.5%) omadacycline-treated subjects in OPTIC, and 5/5 (100%) omadacycline-treated subjects in the combined OASIS studies
 - All omadacycline-treated subjects with *S. pneumoniae* at baseline had outcomes of clinical success at PTE
 - Two linezolid-treated subjects with *Staphylococcus aureus* (omadacycline MIC, 0.25 µg/mL) from OASIS-1 and 1 omadacycline-treated subject from OPTIC with *E. coli* (omadacycline MIC, 2 µg/mL) had indeterminate clinical responses at PTE
 - One omadacycline-treated subject from OPTIC with *K. pneumoniae* (omadacycline MIC, 8 µg/mL) was a clinical failure at PTE

Conclusions

- Omadacycline demonstrated clinical success regardless of MIC result or presence of tetracycline and/or MLS_B resistance mechanisms
- This study expands on the analysis of omadacycline efficacy data among subjects infected with nonsusceptible pathogens

Acknowledgements

This study was sponsored by Paratek Pharmaceuticals, Inc., King of Prussia, PA.

References

Cho JC, Childs-Kean LM, Zmarlicka MT, Crotty MP (2018). Return of the tetracyclines: omadacycline, a novel aminomethylcycline antimicrobial. *Drugs Today* 54: 209-217.

Clinical and Laboratory Standards Institute (2017). *M100Ed27E. Performance standards for antimicrobial susceptibility testing: 27th Informational Supplement.* Wayne, PA, USA.

Clinical and Laboratory Standards Institute (2015). *M07-A10*. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard—10th Edition. Wayne, USA.

Tygacil® Package Insert, Wyeth Pharmaceuticals (2015). Available at https://www.accessdata.fda.gov/drugsatfda_docs/label/2018/021821s045lbl.pdf

Pfaller MA, Huband MD, Shortridge D, Flamm RK (2018). Surveillance of Omadacycline Activity Tested against Clinical Isolates from the United States and Europe as Part of the 2016 SENTRY Antimicrobial Surveillance Program. *Antimicrob Agents Chemother.* 62: e02327-17.

Roberts MC (2003). Tetracycline therapy: Update. *Clin Infect Dis* 36: 462–467.