303

Prevalence of Main Gram-positive Pathogens Causing Bloodstream Infections in US Medical Centers (2010–2015) and Analysis of Oritavancin *In Vitro* Activity

RE Mendes, HS Sader, MD Huband, RK Flamm JMI Laboratories, North Liberty, Iowa, USA

Rodrigo E. Mendes, PhD
JMI Laboratories
335 Beaver Kreek Centre
North Liberty, IA 52317 USA
rodrigo-mendes@jmilabs.com

Abstract

Background: US healthcare agencies have introduced measures to address the burden of nosocomial infections. This study assessed the prevalence of the most common Gram-positive organisms causing bacteremia in US hospitals from 2010–2015 and the *in vitro* activity of oritavancin and comparator agents against these pathogens.

Methods: A total of 10,592 Gram-positive organisms recovered from blood during the SENTRY Antimicrobial Surveillance Program for US were included. Isolates were collected from 33 US sites and were identified by standard biochemical algorithms and MALDI-TOF MS. Susceptibility testing was performed by CLSI methods.

Results: *S. aureus* represented 47.1% of isolates recovered from blood during the six-year period, followed by *Enterococcus* spp. (21.9%; 12.1% and 8.6% of *E. faecalis* and *E. faecium*, respectively), coagulase-negative staphylococci (CoNS; 10.5%), beta-hemolytic streptococci (BHS; 8.0%), and viridans group streptococci (VGS; 4.3%). *S. aureus* rates increased from 46.2% to 52.3% from 2010 to 2012, and decreased to 44.3% in 2015. MRSA rates declined from 45.7% in 2010 to 39.4% in 2014, and increased to 45.4% in 2015. *Enterococcus* spp. rates decreased from 25.5% to 18.0% mostly because *E. faecium* rates declined from 10.1% to 6.7%. CoNS prevalence increased from 8.2% to 15.5% over time. Oritavancin had MIC$_{50/90}$ values of 0.03/0.06 µg/mL against staphylococci, while MIC$_{50/90}$ results for daptomycin and vancomycin were 0.25/0.5 µg/mL and 1/1-2 µg/mL, respectively. Only oritavancin (MIC$_{50/90}$, 0.015/0.06 µg/mL), daptomycin (MIC$_{50/90}$, 1/2 µg/mL), and linezolid (MIC$_{50/90}$, 1/1 µg/mL) were active against enterococci (32.8% VRE). Oritavancin (MIC$_{50/90}$, 0.03/0.12 µg/mL; 99.4% susceptible), penicillin (MIC$_{50/90}$, ≤0.06/≤0.06 µg/mL; 100.0% susceptible), and daptomycin (MIC$_{50/90}$, 0.12/0.25 µg/mL; 100.0% susceptible) were the most active agents against BHS, while oritavancin (MIC$_{50/90}$, 0.015/0.06 µg/mL; 100.0% susceptible) was the most active agent against VGS.

Conclusions: In this set of bacteremia isolates, *S. aureus* and MRSA rates remained elevated, while CoNS consolidated as an emergent pathogen. In contrast, rates of enterococcal infections, mainly those due to *E. faecium*, appeared to be declining. Oritavancin had potent *in vitro* activity against these Gram-positive isolates that caused invasive infections in US hospitals.

Background

Healthcare-associated infections (HAI) are associated with higher healthcare costs and may be considered as a surrogate for hospital quality of care. HAI also are associated with considerable morbidity and mortality among infected patients and continue to increase despite laborious and costly infection prevention efforts. In the last decade, US healthcare agencies have introduced measures to address the burden of nosocomial infections. Methicillin-resistant *Staphylococcus aureus* (MRSA) and vancomycin-resistant enterococci (VRE) are among the top five most prevalent Gram-positive organisms causing HAI. The Centers for Disease Control and Prevention and the Society for Healthcare Epidemiology of America recommend contact (gloves and gown) precautions for healthcare workers caring for hospitalized patients colonized or infected with MRSA or VRE.

Oritavancin was approved by the US Food and Drug Administration (FDA, August 2014) and European Medicines Agency (EMA, March 2015) for the treatment of adults with acute bacterial skin and skin structure infections (ABSSSI). Oritavancin has documented potent *in vitro* activity against Gram-positive clinical isolates. This study assessed the prevalence of the most common Gram-positive organisms causing bacteremia in US hospitals during 2010-2015 and prevalence trends over time. In addition, *in vitro* activity of oritavancin and comparator agents was evaluated against these pathogens.

Methods

Bacterial isolate collection. The prevalence analysis included a total of 10,592 Gram-positive organisms recovered from blood of patients hospitalized in 33 US sites from 2010 to 2015. Only one pathogen per patient/infection episode was included, and the isolates were subsequently submitted to the monitoring laboratory (JMI Laboratories, North Liberty, Iowa, US) as part of the SENTRY Antimicrobial Surveillance Program. The participating laboratory primarily identified the isolates and the reference monitoring laboratory (JMI Laboratories) confirmed bacterial identification by standard algorithms and supported by Matrix Assisted Laser Desorption Ionization Time-of-Flight Mass Spectrometry (MALDI-TOF MS) (Bruker Daltonics, Bremen, Germany).

Antimicrobial susceptibility test methods. The antimicrobial activity analysis used a subset of 7,158 isolates (2010 – 2015) that were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07-A10 document. Bacterial inoculum density was monitored by colony counts to assure an adequate number of cells for each testing event. Validation of the MIC values was performed by concurrent testing of CLSI-recommended quality control (QC) reference strains (*S. aureus* ATCC 29213, *Enterococcus faecalis* ATCC 29212 and *Streptococcus pneumoniae* ATCC 49619). All QC results were within published acceptable ranges (M100-S26). MIC interpretations were based on the CLSI (M100-S26) breakpoint criteria, as available.

Results

- *S. aureus* represented 47.1% of Gram-positive isolates recovered from blood during the six-year period, followed by *Enterococcus* spp. (21.9%; 12.1% and 8.6% of *E. faecalis* and *E. faecium*, respectively), coagulase-negative staphylococci (CoNS; 10.5%), beta-hemolytic streptococci (BHS; 8.0%), *S. pneumoniae* (5.1%) and viridans group streptococci (VGS; 4.3%; Figure 1).
- *S. aureus* rates increased from 46.1% to 52.2% from 2010 to 2012, and decreased to 43.9% in 2015. MRSA rates declined from 21.1% in 2010 to 18.7% in 2014, and increased to 19.9% in 2015 (Figure 2). CoNS prevalence increased from 8.2% to 15.5% over time.
- Overall, *E. faecalis* (55.5%) represented the majority of enterococcal isolates collected during the study interval, followed closely by *E. faecium* (39.5%). *Enterococcus* spp. rates decreased during the period (from 25.5% to 18.0%), and prevalence rates for both *E. faecalis* (from 13.9% to 10.4%) and *E. faecium* (from 10.1% to 6.7%) decreased during the six-year period (Figure 3).
- While the vancomycin resistance (VRE) rates remained low among *E. faecalis* (4.0% overall), 76.4% of *E. faecium* were vancomycin-resistant (Table 2). However, the rate of vancomycin resistance in *E. faecium* decreased from 80.7% in 2010 to 68.0% in 2015 (Figure 3).
- *Streptococcus agalactiae* and *Streptococcus mitis* group were the most prevalent species/group within the BHS and VGS populations, respectively (Figure 1). Overall, the prevalence rates for both BHS (from 7.7% to 9.1%) and VGS (from 3.7% to 5.5%) increased during the study period (Figure 4).
- Oritavancin (MIC$_{50/90}$, 0.03/0.06 µg/mL) inhibited 99.7% of *S. aureus* at the breakpoint for susceptibility (≤0.12 µg/mL), and had a MIC$_{90}$ value eight- to 16-fold lower than daptomycin (MIC$_{50/90}$, 0.25/0.5 µg/mL), ceftaroline (MIC$_{50/90}$, 0.25/1 µg/mL) and vancomycin (MIC$_{50/90}$, 1/1 µg/mL; Tables 1 and 2).
- Oritavancin inhibited 97.4% of all enterococcal isolates at the breakpoint for susceptibility for *E. faecalis* (i.e., ≤0.12 µg/mL for vancomycin-susceptible only), and 97.9% and 96.8% of *E. faecium* and *E. faecalis* at ≤0.12 µg/mL, respectively (Table 1).
- Only oritavancin (MIC$_{50/90}$, 0.03/0.12 µg/mL), daptomycin (MIC$_{50/90}$, 2/2 µg/mL), and linezolid (MIC$_{50/90}$, 1/2 µg/mL) showed *in vitro* activity against *E. faecium* (76.1% VRE). Oritavancin (MIC$_{50/90}$, 0.015/0.06 µg/mL), daptomycin (MIC$_{50/90}$, 1/1 µg/mL), linezolid (MIC$_{50/90}$, 1/1 µg/mL) and ampicillin (MIC$_{50/90}$, ≤1/2 µg/mL) were active against *E. faecalis* (Table 2).
- CoNS isolates demonstrated a multidrug-resistant (MDR) phenotype with susceptibility rates for most agents between 32.5 – 83.2%, except for daptomycin (100.0% susceptible), linezolid (98.7% susceptible), and vancomycin (100.0% susceptible; Table 2).
- Oritavancin MIC$_{50/90}$ results (MIC$_{50/90}$, 0.03/0.06 µg/mL) against CoNS were eight-fold lower than those of ceftaroline (MIC$_{50/90}$, 0.25/0.5 µg/mL) and daptomycin (MIC$_{50/90}$, 0.25/0.5 µg/mL), and 16- to 32-fold lower than those obtained for linezolid (MIC$_{50/90}$, 0.5/1 µg/mL) and vancomycin (MIC$_{50/90}$, 1/2 µg/mL; Table 2).
- Oritavancin (MIC$_{50/90}$, 0.03/0.12 µg/mL; 99.4% susceptible), penicillin (MIC$_{50/90}$, ≤0.06/≤0.06 µg/mL; 100.0% susceptible), and daptomycin (MIC$_{50/90}$, 0.12/0.25 µg/mL; 100.0% susceptible) were the most active agents against BHS, while oritavancin (MIC$_{50/90}$, 0.015/0.06 µg/mL; 100.0% susceptible) was the most active agent against VGS.

Figure 1. Prevalence all Gram-positive organisms causing bacteremia in US hospitals from 2010 to 2015.

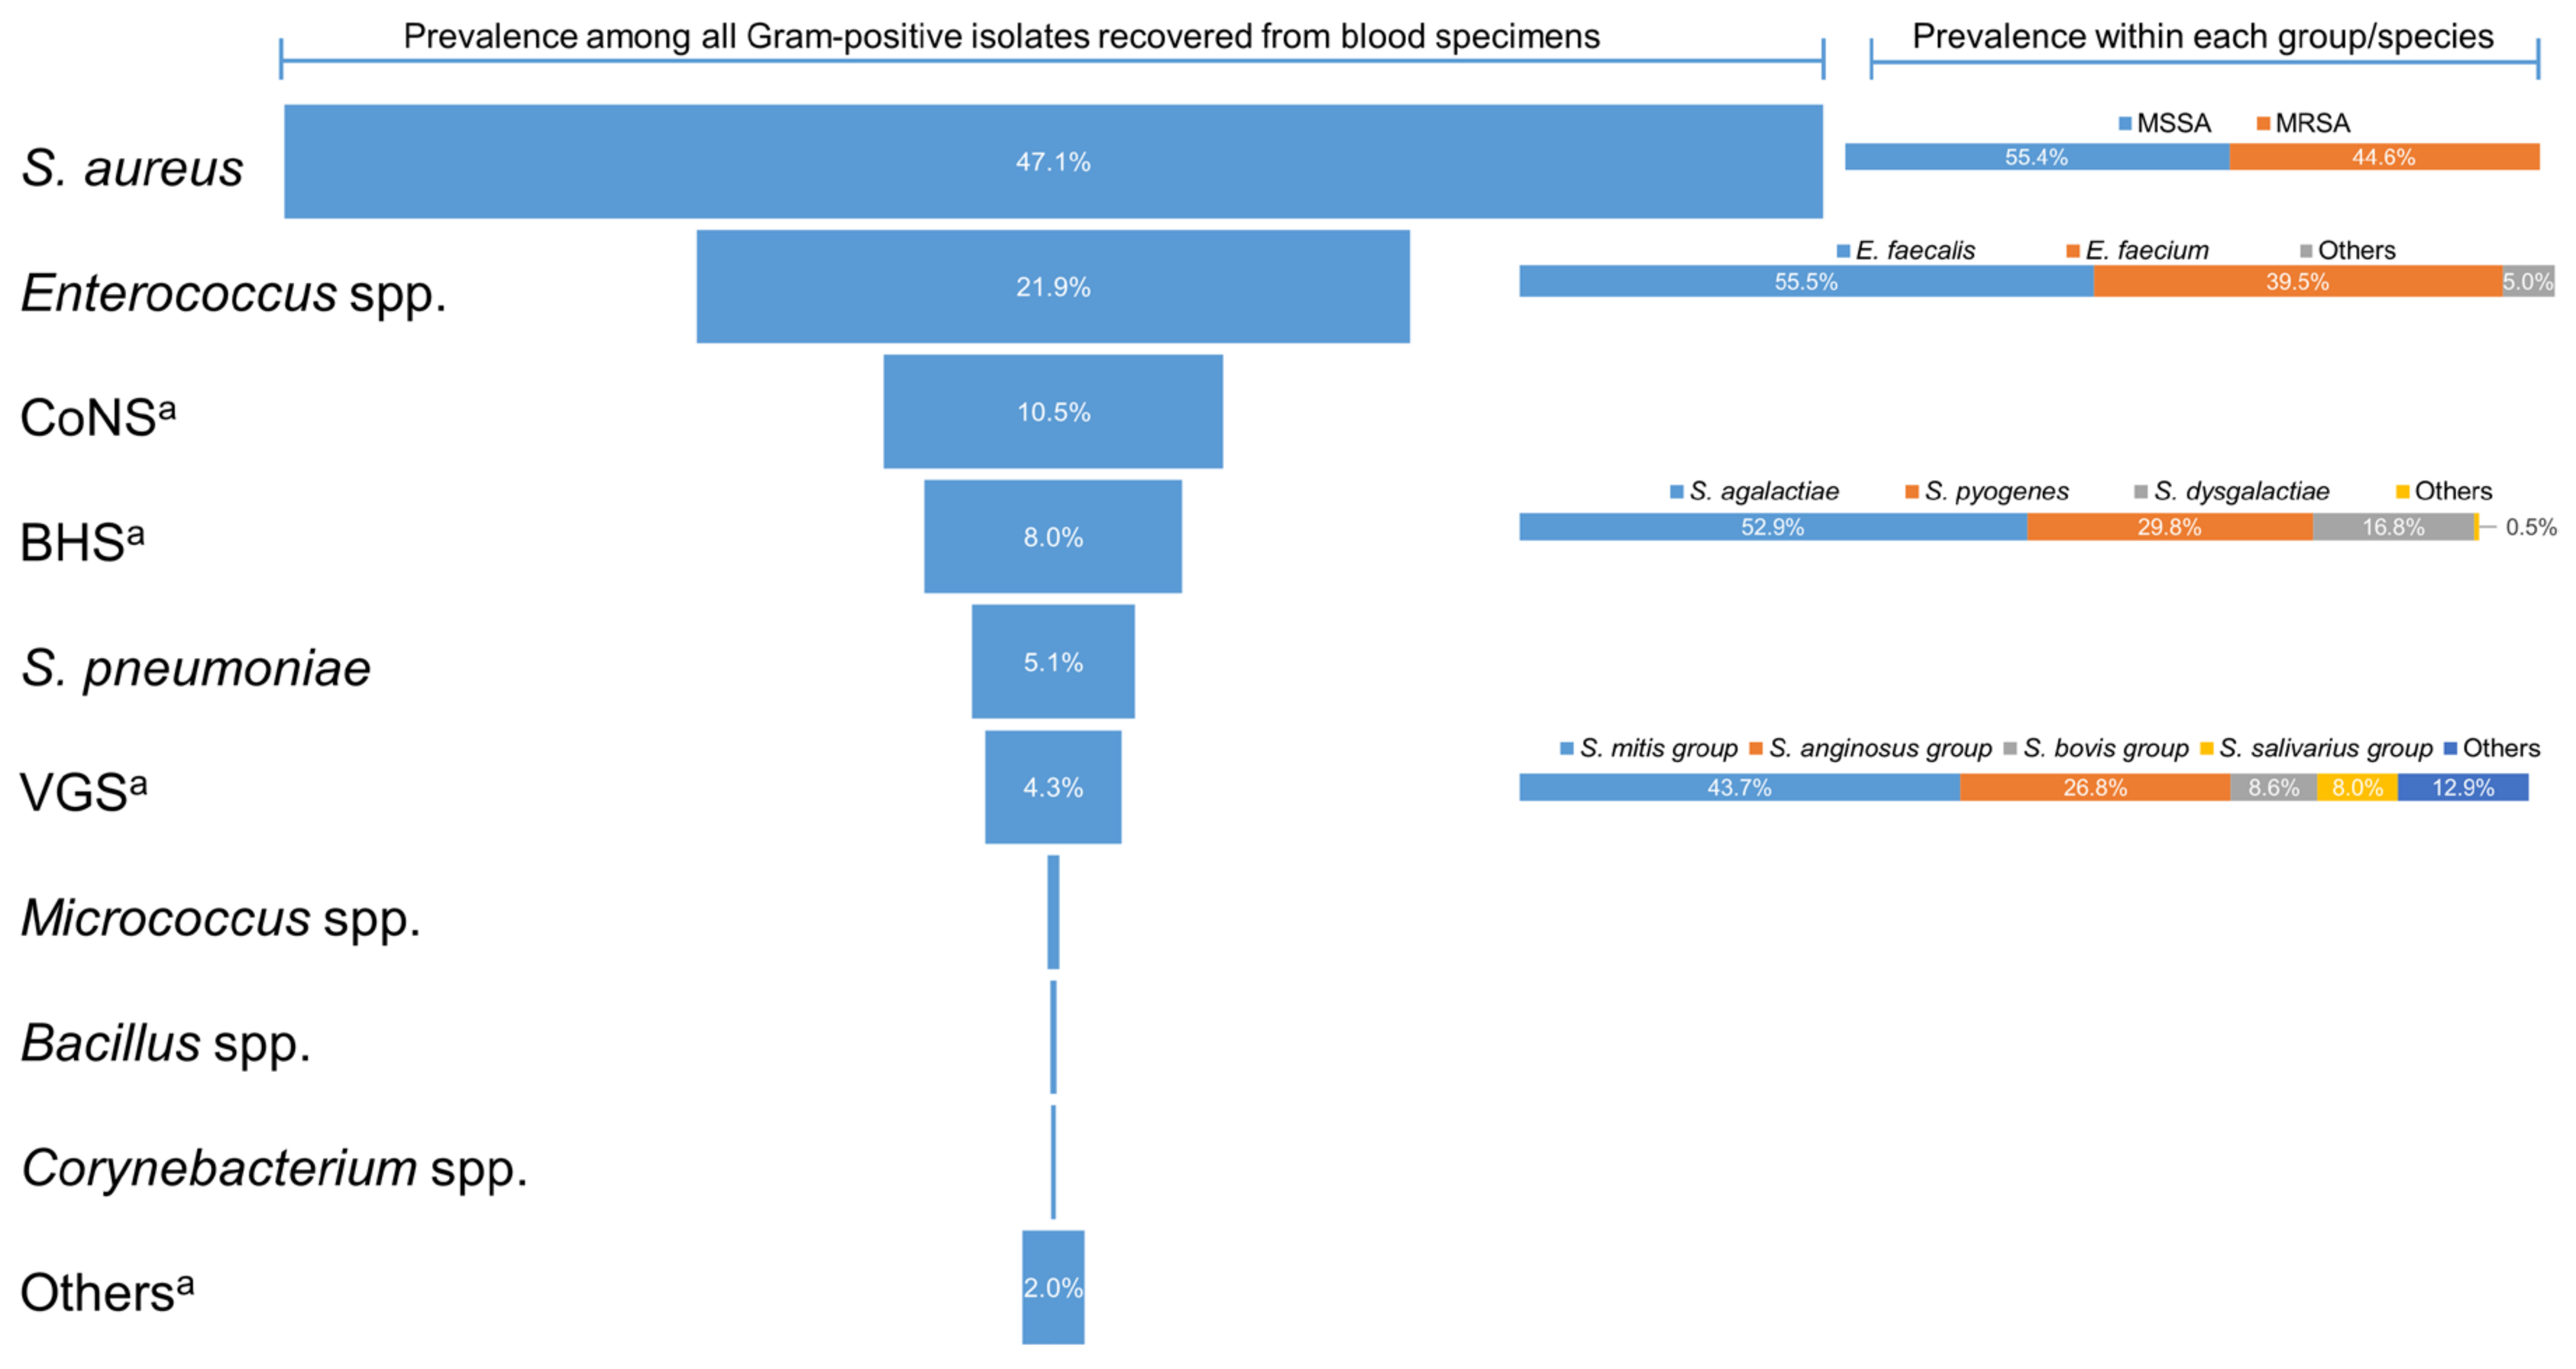

Prevalence among all Gram-positive isolates recovered from blood specimens
Prevalence within each group/species
S. aureus 47.1%
MSSA 55.4% MRSA 44.6%
Enterococcus spp. 21.9%
E. faecalis 55.5% E. faecium 39.5% Others 5.0%
CoNSa 10.5%
BHSa 8.0%
S. agalactiae 52.9% S. pyogenes 29.8% S. dysgalactiae 16.8% Others 0.5%
S. pneumoniae 5.1%
VGSa 4.3%
S. mitis group 43.7% S. anginosus group 26.8% S. bovis group 8.6% S. salivarius group 8.0% Others 12.9%
Micrococcus spp.
Bacillus spp.
Corynebacterium spp.
Othersa 2.0%

[a] CoNS = coagulase-negative staphylococci; BHS = β-hemolytic streptococci; VGS = Viridans group streptococci. Others are represented by 40 different genera.

Table 1. Antimicrobial activity of oritavancin against the main organisms and organism groups included in this study.

Organism (number tested)	Number of isolates at each MIC (µg/mL; cumulative %)							MIC (µg/mL)	
	≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	50%	90%
***S. aureus* (3,875)**	161 (4.2%)	1216 (35.5%)	1408 (71.9%)	857 (94.0%)	223 (99.7%)	10 (100.0%)		0.03	0.06
***Enterococcus* spp.[a] (1,705)**	488 (28.6%)	486 (57.2%)	360 (78.3%)	227 (91.6%)	98 (97.4%)	38 (99.6%)	7 (100.0%)	0.015	0.06
***Enterococcus faecium* (679)**	202 (29.7%)	103 (44.9%)	144 (66.1%)	145 (87.5%)	71 (97.9%)	14 (100.0%)		0.03	0.12
***Enterococcus faecalis* (965)**	239 (24.8%)	374 (63.6%)	212 (85.6%)	81 (94.0%)	27 (96.8%)	24 (99.3%)	7 (100.0%)	0.015	0.06
CoNS[b] (758)	142 (18.7%)	119 (34.4%)	254 (67.9%)	197 (93.9%)	44 (99.7%)	2 (100.0%)		0.03	0.06
BHS[c] (530)	44 (8.3%)	140 (34.7%)	145 (62.1%)	97 (80.4%)	68 (93.2%)	33 (99.4%)	3 (100.0%)	0.03	0.12
VGS[d] (290)	140 (48.3%)	50 (65.5%)	41 (79.7%)	32 (90.7%)	23 (98.6%)	4 (100.0%)		0.015	0.06

[a] Includes: *Enterococcus avium* (8), *E. casseliflavus* (14), *E. durans* (8), *E. faecalis* (965), *E. faecium* (679), *E. gallinarum* (17), *E. hirae* (5), *E. raffinosus* (5), other *Enterococcus* spp. (4).
[b] CoNS = coagulase-negative staphylococci and includes: *Staphylococcus auricularis* (9), *S. capitis* (37), *S. caprae* (5), *S. cohnii* (1), *S. epidermidis* (427), *S. haemolyticus* (33), *S. hominis* (75), *S. intermedius* (1), *S. lugdunensis* (19), *S. pasteuri* (1), *S. pettenkoferi* (2), *S. saprophyticus* (4), *S. simulans* (4), *S. warneri* (20), *Staphylococcus* spp. (120).
[c] BHS = β-hemolytic streptococci and includes: *Streptococcus agalactiae* (309), *S. dysgalactiae* (41), *S. pyogenes* (180).
[d] VGS = Viridans group streptococci and includes: *Streptococcus alactolyticus* (2), *S. anginosus* (31), *S. anginosus* group (5), *S. australis* (3), *S. bovis* group (12), *S. constellatus* (3), *S. gallolyticus* (16), *S. gordonii* (7), *S. infantarius* (1), *S. intermedius* (5), *S. lutetiensis* (1), *S. mitis* (3), *S. mitis* group (46), *S. mitis/oralis* (27), *S. mutans* (4), *S. oralis* (24), *S. parasanguinis* (14), *S. salivarius* (19), *S. salivarius* group (5), *S. sanguinis* (24), *S. sinensis* (1), *S. vestibularis* (2), *Streptococcus* spp. (35).

Table 2. Antimicrobial activity of oritavancin and comparator agents tested against the main organisms and organism groups included in this study.

Organism[a] (no. tested) Antimicrobial Agent[b]	MIC (µg/mL)		MIC Range (µg/mL)	CLSI[c]			EUCAST[c]		
	50%	90%		%S	%I	%R	%S	%I	%R
***S. aureus* (3,875)**									
Oritavancin	0.03	0.06	≤0.008 — 0.25	99.7	-	-	99.7	-	0.3
Ceftaroline	0.25	1	≤0.06 — 2	96.9	3.1	0.0	96.9	-	3.1
Clindamycin	≤0.25	>2	≤0.25 — >2	82.6	0.2	17.2	82.3	0.3	17.4
Daptomycin	0.25	0.5	≤0.12 — 4	99.8	-	-	99.8	-	0.2
Erythromycin	>4	>4	≤0.25 — >4	43.4	3.4	53.1	43.8	1.0	55.2
Levofloxacin	≤0.5	>4	≤0.5 — >4	61.3	1.1	37.6	61.3	1.1	37.6
Linezolid	1	1	≤0.12 — 4	100.0	-	0.0	100.0	-	0.0
Minocycline	≤0.06	0.12	≤0.06 — 8	99.3	0.7	0.0	97.7	0.3	2.0
Oxacillin	0.5	>2	≤0.25 — >2	55.5	-	44.5	55.5	-	44.5
Tetracycline	≤0.5	≤0.5	≤0.5 — >8	95.7	0.6	3.7	93.4	1.6	5.0
TMP-SMX	≤0.5	≤0.5	≤0.5 — >4	98.3	-	1.7	98.3	0.2	1.5
Vancomycin	1	1	≤0.12 — 2	100.0	0.0	0.0	100.0	-	0.0
***E. faecium* (679)**									
Oritavancin	0.03	0.12	≤0.008 — 0.25	-	-	-	-	-	-
Ampicillin	>8	>8	≤1 — >8	9.3	-	90.7	8.7	0.6	90.7
Daptomycin	2	2	≤0.25 — >8	99.4	-	-	-	-	-
Erythromycin	>4	>4	≤0.25 — >4	3.2	17.4	79.4	-	-	-
Levofloxacin	>4	>4	≤0.5 — >4	7.7	2.4	90.0	10.0	-	90.0[d]
Linezolid	1	2	≤0.25 — >8	98.8	0.7	0.4	99.6	-	0.4
Tetracycline	>8	>8	≤1 — >8	23.4	1.6	75.0	-	-	-
Vancomycin	>16	>16	≤0.5 — >16	23.6	0.3	76.1	23.6	-	76.4
***E. faecalis* (965)**									
Oritavancin	0.015	0.06	≤0.008 — 0.5	99.5	-	-	-	-	-
Ampicillin	≤1	2	≤1 — 4	100.0	-	0.0	100.0	0.0	0.0
Daptomycin	1	1	≤0.25 — 4	100.0	-	-	-	-	-
Erythromycin	>4	>4	≤0.25 — >4	8.3	34.8	57.0	-	-	-
Levofloxacin	1	>4	≤0.5 — >4	69.0	0.5	30.5	69.5	-	30.5[d]
Linezolid	1	1	≤0.25 — >8	99.9	0.0	0.1	99.9	-	0.1
Tetracycline	>8	>8	≤1 — >8	24.8	0.3	74.9	-	-	-
Vancomycin	1	2	≤0.5 — >16	95.8	0.2	4.0	95.8	-	4.2
CoNS (758)									
Oritavancin	0.03	0.06	≤0.008 — 0.25	-	-	-	-	-	-
Ceftaroline	0.25	0.5	≤0.06 — 2	-	-	-	-	-	-
Clindamycin	≤0.25	>2	≤0.25 — >2	64.9	2.4	32.7	64.2	0.7	35.1
Daptomycin	0.25	0.5	≤0.12 — 1	100.0	-	-	100.0	-	0.0
Erythromycin	>4	>4	≤0.25 — >4	32.7	2.5	64.8	33.4	0.9	65.7
Levofloxacin	4	>4	≤0.5 — >4	45.0	2.0	53.0	45.0	2.0	53.0
Linezolid	0.5	1	≤0.12 — >8	98.7	-	1.3	98.7	-	1.3
Oxacillin	>2	>2	≤0.25 — >2	32.5	-	67.5	32.5	-	67.5
Tetracycline	≤0.5	>8	≤0.5 — >8	83.2	1.5	15.3	76.9	6.2	16.9
TMP-SMX	≤0.5	>4	≤0.5 — >4	65.7	-	34.3	65.7	17.9	16.4
Vancomycin	1	2	≤0.12 — 4	100.0	0.0	0.0	100.0	-	0.0
BHS (530)									
Oritavancin	0.03	0.12	≤0.008 — 0.5	99.4	-	-	99.4	-	0.6
Ceftaroline	≤0.008	0.015	≤0.008 — 0.015	100.0	-	-	100.0	-	0.0
Clindamycin	≤0.25	>2	≤0.25 — >2	79.4	0.6	20.0	80.0	-	20.0
Daptomycin	0.12	0.25	≤0.06 — 0.5	100.0	-	-	100.0	-	0.0
Erythromycin	≤0.25	>4	≤0.25 — >4	61.3	1.1	37.5	61.3	1.1	37.5
Levofloxacin	≤0.5	1	≤0.5 — >4	99.1	0.2	0.8	95.5	3.6	0.9
Linezolid	1	1	≤0.12 — 1	100.0	-	-	100.0	0.0	0.0
Penicillin	≤0.06	≤0.06	≤0.06 — 0.12	100.0	-	-	100.0	-	0.0
Tetracycline	>8	>8	≤0.5 — >8	41.6	1.5	56.9	41.6	0.0	58.4
TMP-SMX	≤0.5	≤0.5	≤0.5 — >4	-	-	-	-	-	-
Vancomycin	0.5	0.5	0.12 — 1	100.0	-	-	100.0	-	0.0
VGS (290)									
Oritavancin	0.015	0.06	≤0.008 — 0.25	100.0	-	-	100.0	-	0.0
Ceftaroline	0.015	0.12	≤0.008 — 0.5	-	-	-	-	-	-
Clindamycin	≤0.25	>2	≤0.25 — >2	86.9	2.1	11.1	88.9	-	11.1
Daptomycin	0.25	0.5	≤0.06 — 1	100.0	-	-	-	-	-
Erythromycin	1	>4	≤0.25 — >4	41.4	3.4	55.2	-	-	-
Levofloxacin	1	2	≤0.5 — >4	91.0	1.0	7.9	-	-	-
Linezolid	0.5	1	≤0.12 — 4	99.7	-	-	-	-	-
Penicillin	≤0.06	1	≤0.06 — >4	71.7	23.1	5.2	79.0	15.9	5.2
Tetracycline	≤0.5	>8	≤0.5 — >8	65.5	3.4	31.0	-	-	-
TMP-SMX	≤0.5	4	≤0.5 — >4	-	-	-	-	-	-
Vancomycin	0.5	0.5	≤0.12 — 1	100.0	-	-	100.0	-	0.0

[a] CoNS = coagulase-negative staphylococci; BHS = β-hemolytic streptococci; VGS = Viridans group streptococci. Refer to footnotes of Table 1 for species included in each group of organisms.
[b] TMP-SMX = Trimethoprim-sulfamethoxazole.
[c] Breakpoint criteria were those from CLSI (2016) or EUCAST (2016). "-" breakpoint not available.
[d] Uncomplicated UTI only.

Figure 2. Prevalence of *S. aureus* (MRSA and MSSA) and CoNS causing bacteremia in US hospitals from 2010 to 2015.

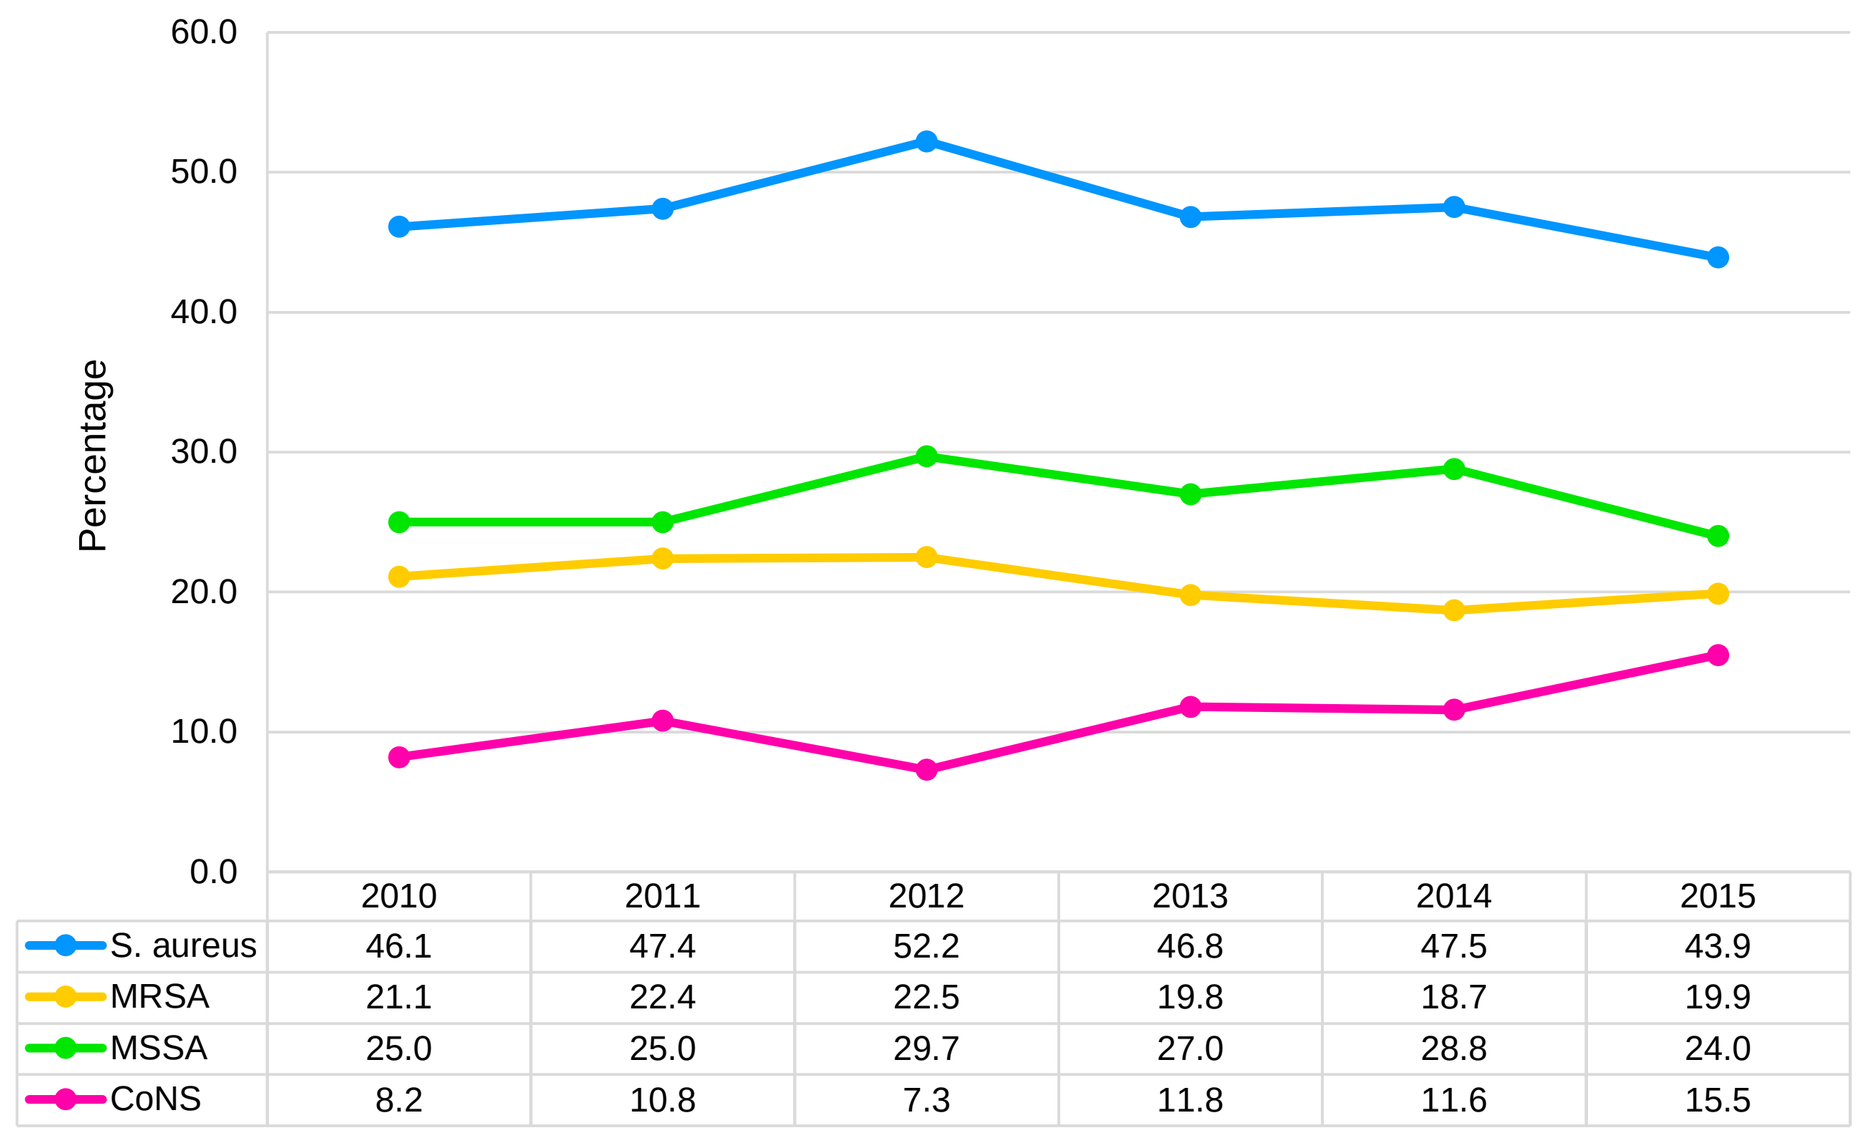

Percentage
60.0 50.0 40.0 30.0 20.0 10.0 0.0
| | 2010 | 2011 | 2012 | 2013 | 2014 | 2015 |
S. aureus 46.1 47.4 52.2 46.8 47.5 43.9
MRSA 21.1 22.4 22.5 19.8 18.7 19.9
MSSA 25.0 25.0 29.7 27.0 28.8 24.0
CoNS 8.2 10.8 7.3 11.8 11.6 15.5

Figure 3. Prevalence of *Enterococcus* spp. causing bacteremia in US hospitals from 2010 to 2015 (VRE rates within *E. faecium*).

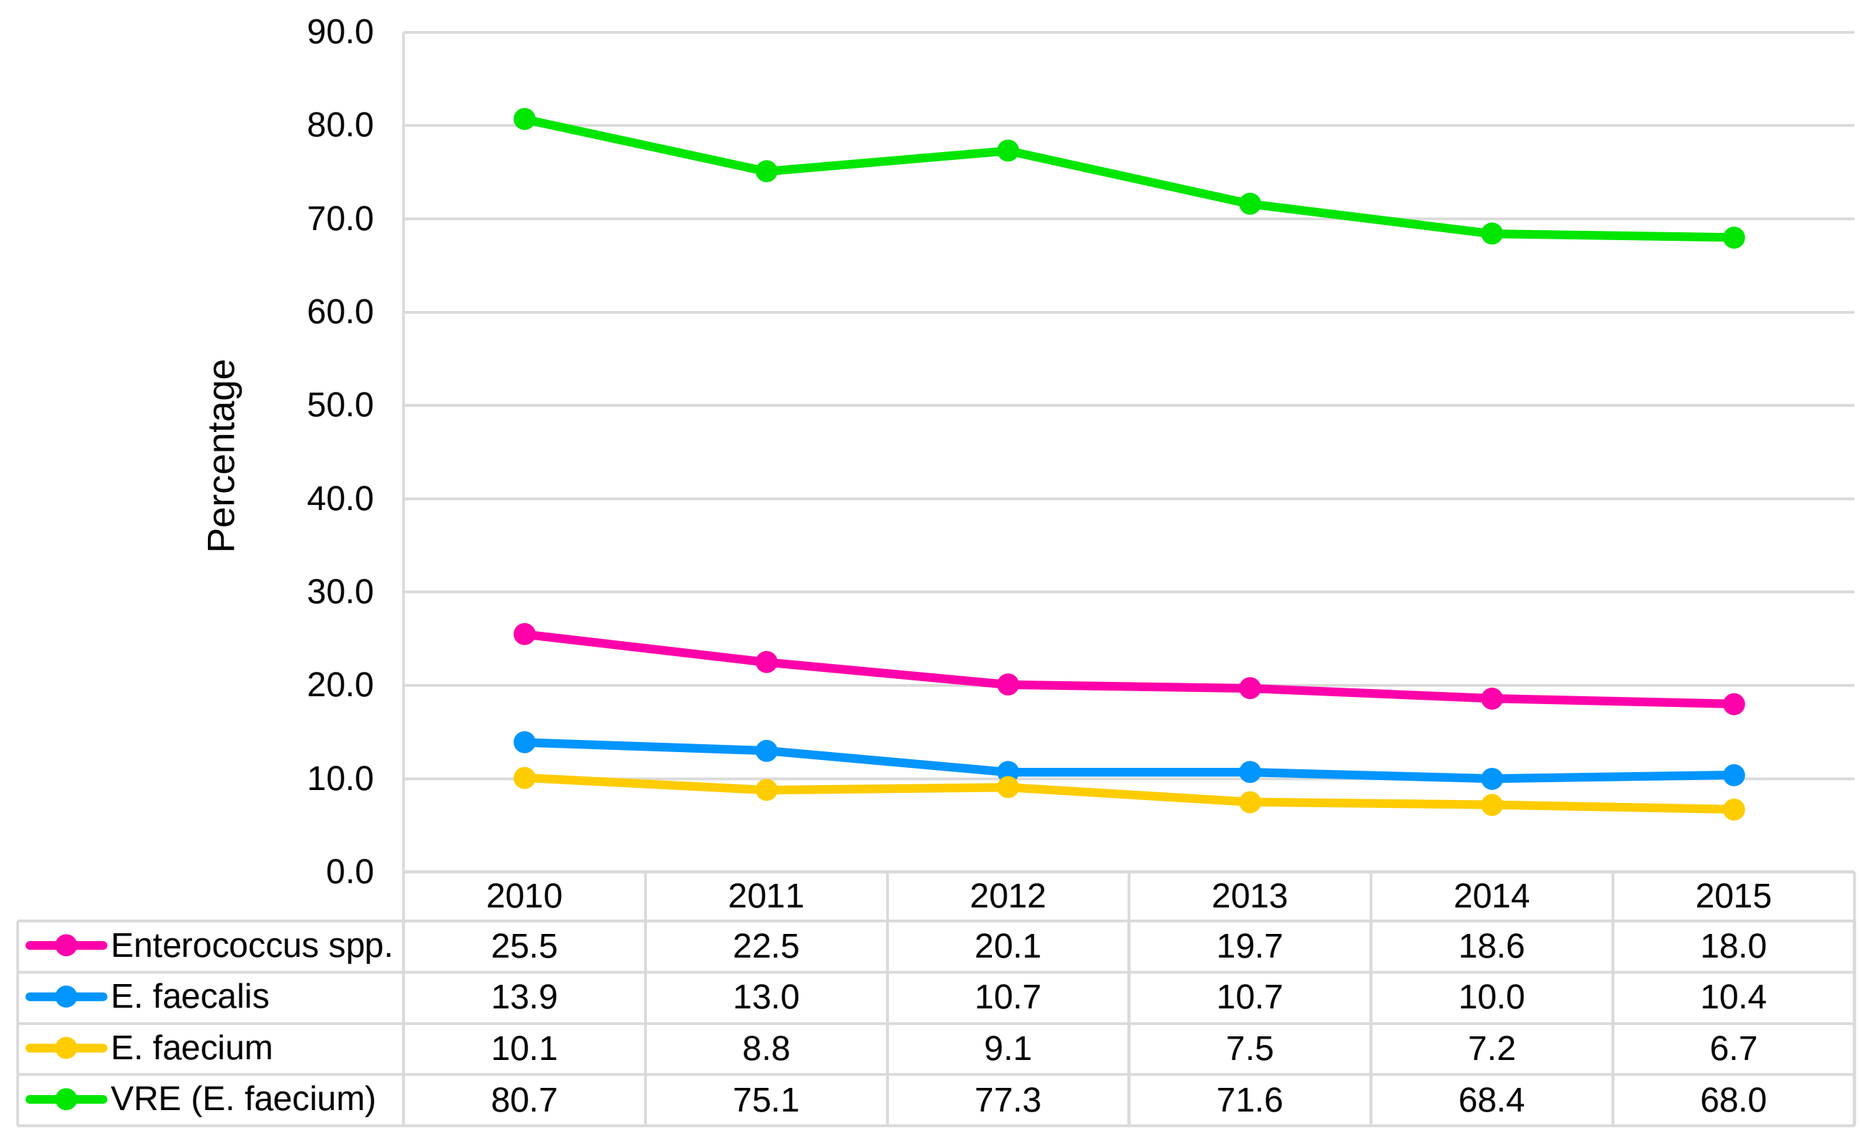

Percentage
90.0 80.0 70.0 60.0 50.0 40.0 30.0 20.0 10.0 0.0
2010 2011 2012 2013 2014 2015
Enterococcus spp. 25.5 22.5 20.1 19.7 18.6 18.0
E. faecalis 13.9 13.0 10.7 10.7 10.0 10.4
E. faecium 10.1 8.8 9.1 7.5 7.2 6.7
VRE (E. faecium) 80.7 75.1 77.3 71.6 68.4 68.0

Figure 4. Prevalence of β-hemolytic streptococci (BHS) and viridans group streptococci (VGS) causing bacteremia in US hospitals from 2010 to 2015.

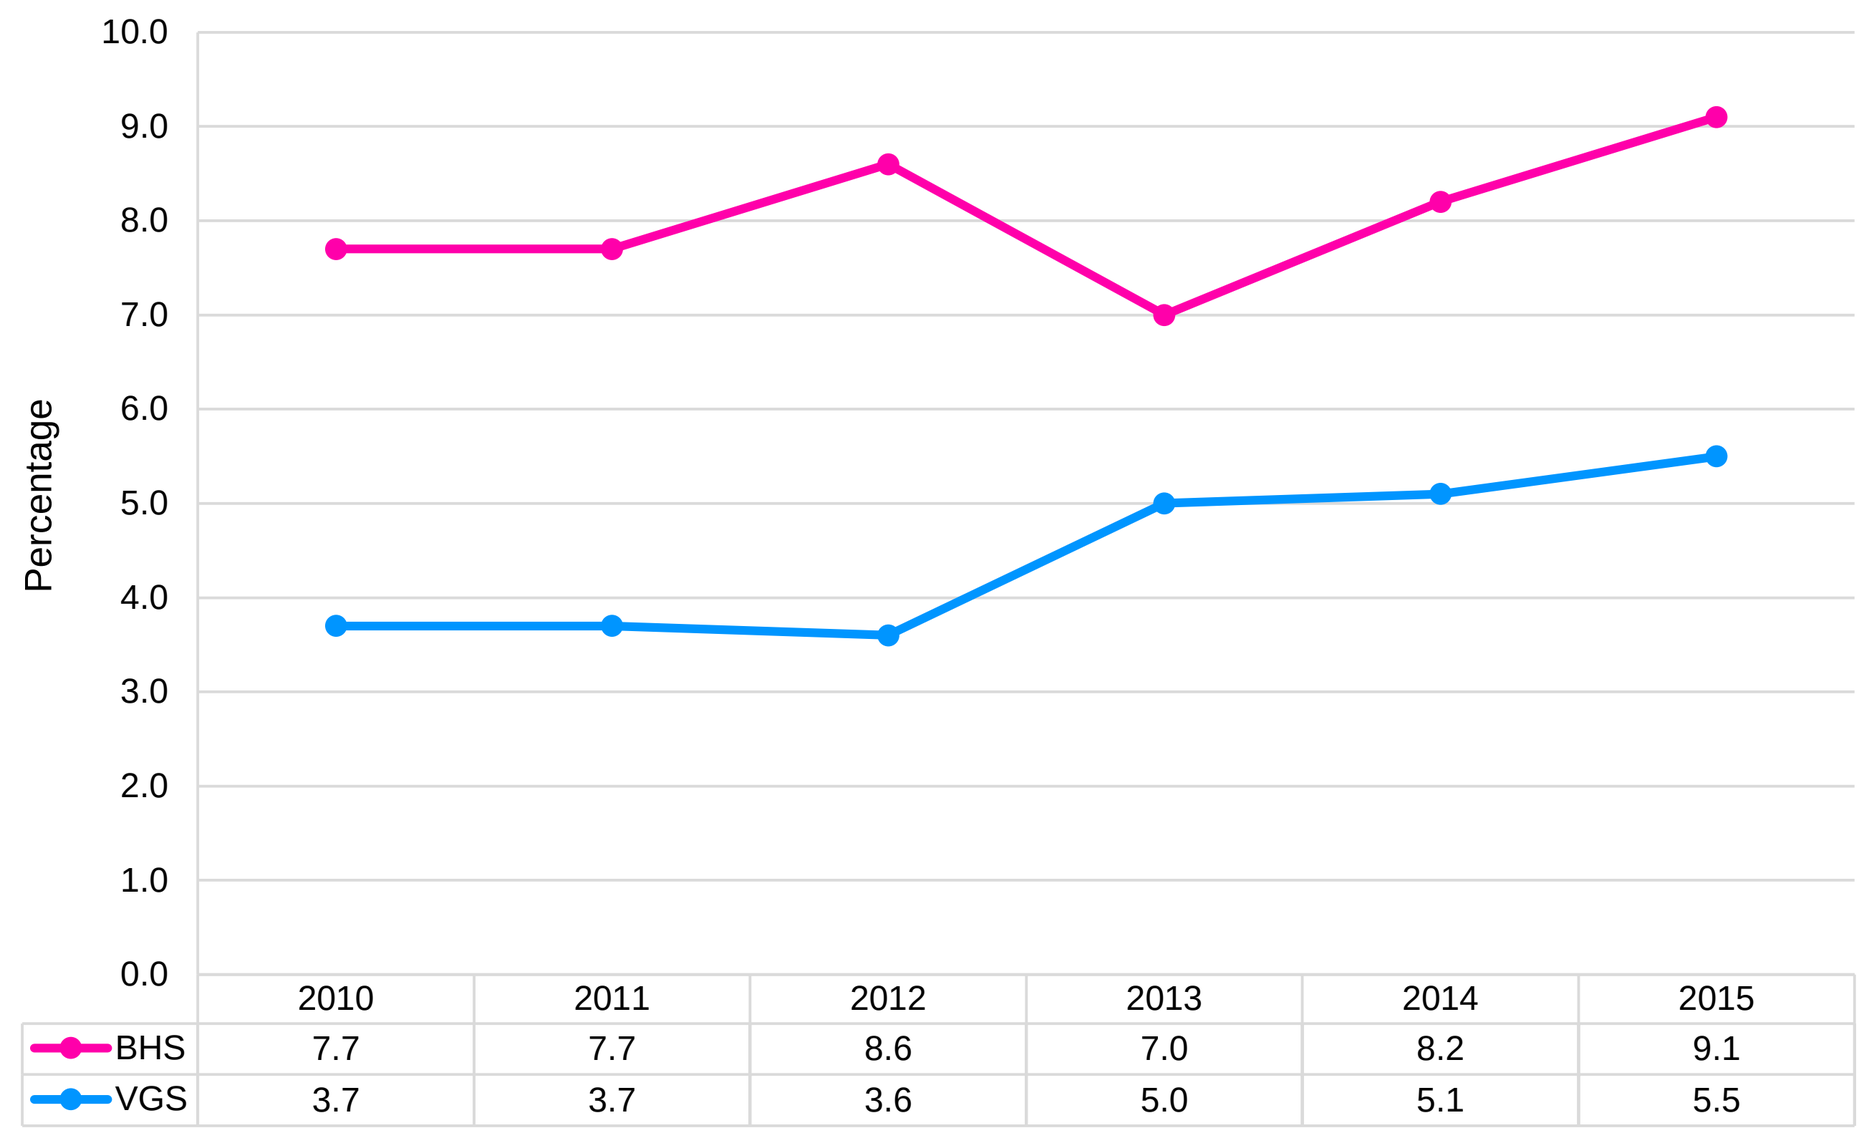

Percentage
10.0 9.0 8.0 7.0 6.0 5.0 4.0 3.0 2.0 1.0 0.0
2010 2011 2012 2013 2014 2015
BHS 7.7 7.7 8.6 7.0 8.2 9.1
VGS 3.7 3.7 3.6 5.0 5.1 5.5

Conclusions

- Prevalence of *S. aureus* and MRSA causing bacteremia in US hospitals remains elevated, although these rates showed signs of decline during the study period.
- Likewise, prevalence rates for both *E. faecalis* and *E. faecium* declined during the six-year interval. However, the rate of *E. faecium* (9.1%) in 2012 was similar to the rate of *E. faecalis* (10.7%), which is strikingly different from decades ago when *E. faecalis* predominated.
- In contrast, rates of CoNS isolates increased consistently, as did VGS rates. Although representing approximately only 15% of the bacteremia cases in the last study year, CoNS isolates have consolidated as an emergent pathogen. This becomes a concern as these isolates usually exhibit an MDR phenotype.
- Oritavancin demonstrated potent *in vitro* activity against the Gram-positive isolates in this study causing bloodstream infections in US hospitals. Against organisms/groups for which oritavancin breakpoints are available, oritavancin demonstrated a high level of activity (99.4% – 100.0% susceptible).

Disclosures

This study was sponsored by an educational/research grant from The Medicines Company (Parsippany, New Jersey, United States) via the SENTRY Antimicrobial Surveillance Program platform.

References

1. Ajao AO, Harris AD, Roghmann MC, Johnson JK, Zhan M, McGregor JC, Furuno JP (2011). Systematic review of measurement and adjustment for colonization pressure in studies of methicillin-resistant *Staphylococcus aureus*, vancomycin-resistant enterococci, and *Clostridium difficile* acquisition. *Infect Control Hosp Epidemiol* 32: 481-489.
2. Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: Tenth edition*. Wayne, PA, USA.
3. Clinical and Laboratory Standards Institute (2016). *M100-S26. Performance standards for antimicrobial susceptibility testing: 26th Informational supplement.* Wayne, PA, USA.
4. EUCAST (2016). Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, January 2016. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2016.
5. Mendes RE, Sader HS, Flamm RK, Farrell DJ, Jones RN (2014). Oritavancin Activity against *Staphylococcus aureus* Causing Invasive Infections in U.S. and European Hospitals: a 5-Year International Surveillance Program. *Antimicrob Agents Chemother* 58: 2921–2924.
6. Snyder GM, Thom KA, Furuno JP, Perencevich EN, Roghmann MC, Strauss SM, Netzer G, Harris AD (2008). Detection of methicillin-resistant *Staphylococcus aureus* and vancomycin-resistant enterococci on the gowns and gloves of healthcare workers. *Infect Control Hosp Epidemiol* 29: 583-589.

Presented at IDWeek 2016, October 26-30, 2016, New Orleans, LA