Antimicrobial Activity of Aztreonam-Avibactam and Comparator Agents Tested against Contemporary (2016) Clinical *Enterobacteriaceae* Isolates

IDWeek 2017
Poster #1233

HS Sader, RE Mendes, D Shortridge, RK Flamm, M Castanheira

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- Avibactam effectively inactivates the vast majority of clinically relevant serine β-lactamases, including class A (such as KPC), class C (AmpC), and some class D (OXA) β-lactamases; however, avibactam and all other β-lactamase inhibitors currently available for clinical use do not inhibit metallo-β-lactamases (MBLs)
- Aztreonam has a unique feature compared to other β-lactams in that it is stable to hydrolysis by MBLs
- Aztreonam was approved by the United States (US) Food and Drug Administration (FDA) in 1986, and it is still the only clinically available member of the monobactam class
- Because *Enterobacteriaceae* isolates that produce an MBL usually coproduce a serine β-lactamase, aztreonam was combined with avibactam, and this novel β-lactamase-inhibitor combination is under clinical development (NCT01689207; available at https://clinicaltrials.gov/)
- We assessed the *in vitro* activity of aztreonam-avibactam when tested against a large collection of contemporary (2016) clinical *Enterobacteriaceae* isolates recovered from patients hospitalized in U.S. medical centers, and carbapenem-resistant *Enterobacteriaceae* (CRE) isolates collected worldwide, including NDM, KPC, OXA, VIM, and SME producers

MATERIALS AND METHODS

Bacterial isolates

- A total of 10,451 *Enterobacteriaceae* isolates were consecutively collected from 84 medical centers in 37 states from all 9 U.S. Census divisions in 2016
- Isolates were collected from patients with urinary tract infections (n = 4,222; 40.4%), pneumonia (n = 2,051; 19.6%), skin and skin structure infections (n = 1,806; 17.3%), bloodstream infections (n = 1,641; 15.7%), intra-abdominal infections (n = 398; 3.8%), and other infection types (n = 333; 3.2%) according to defined protocols
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program
- Species identification was confirmed by standard biochemical tests and using the MALDI Biotyper (Bruker Daltonics, Billerica, Massachusetts, USA) according to the manufacturer instructions, where necessary

Resistant subsets

- Isolates were categorized as multidrug-resistant (MDR), extensively drug-resistant (XDR), or pan-drug resistant (PDR) according to criteria published by Magiorakos *et al.* (2012)
- CRE was defined as resistant (MIC, ≥4 µg/mL [CLSI]) to imipenem (imipenem was **not** applied to *Proteus mirabilis* or to indole-positive *Proteeae*), meropenem, or doripenem

Ex-US isolates

- With the purpose of assessing the *in vitro* activity of aztreonam-avibactam against *Enterobacteriaceae*-producing carbapenemases that are uncommon in the US, we included a collection of contemporary CRE clinical isolates from outside the US
- The collection comprised 250 CRE isolates from 38 centers in 25 other countries (ex-US) in 2016

Whole genome sequencing

- Total genomic DNA was extracted using the fully-automated ThermoScientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, Ohio, USA)
- DNA extracts were quantified using the Qubit™ High Sensitivity DS-DNA assay (Invitrogen, ThermoFisher Inc.) and normalized to 0.2 ng/µL
- A total of 1 ng high-quality genomic DNA was used as input material for library construction using the Nextera XT™ DNA library preparation kit (Illumina, San Diego, California, USA)
 – Libraries were normalized using the bead-based normalization procedure (Illumina) and sequenced on MiSeq
 – Fastq files generated were assembled using SPAdes Assembler and subjected to a proprietary software (JMI Laboratories) for screening β-lactamase genes

Figure 1 Antimicrobial activity of aztreonam-avibactam tested against carbapenem-resistant *Enterobacteriaceae* (CRE) isolates

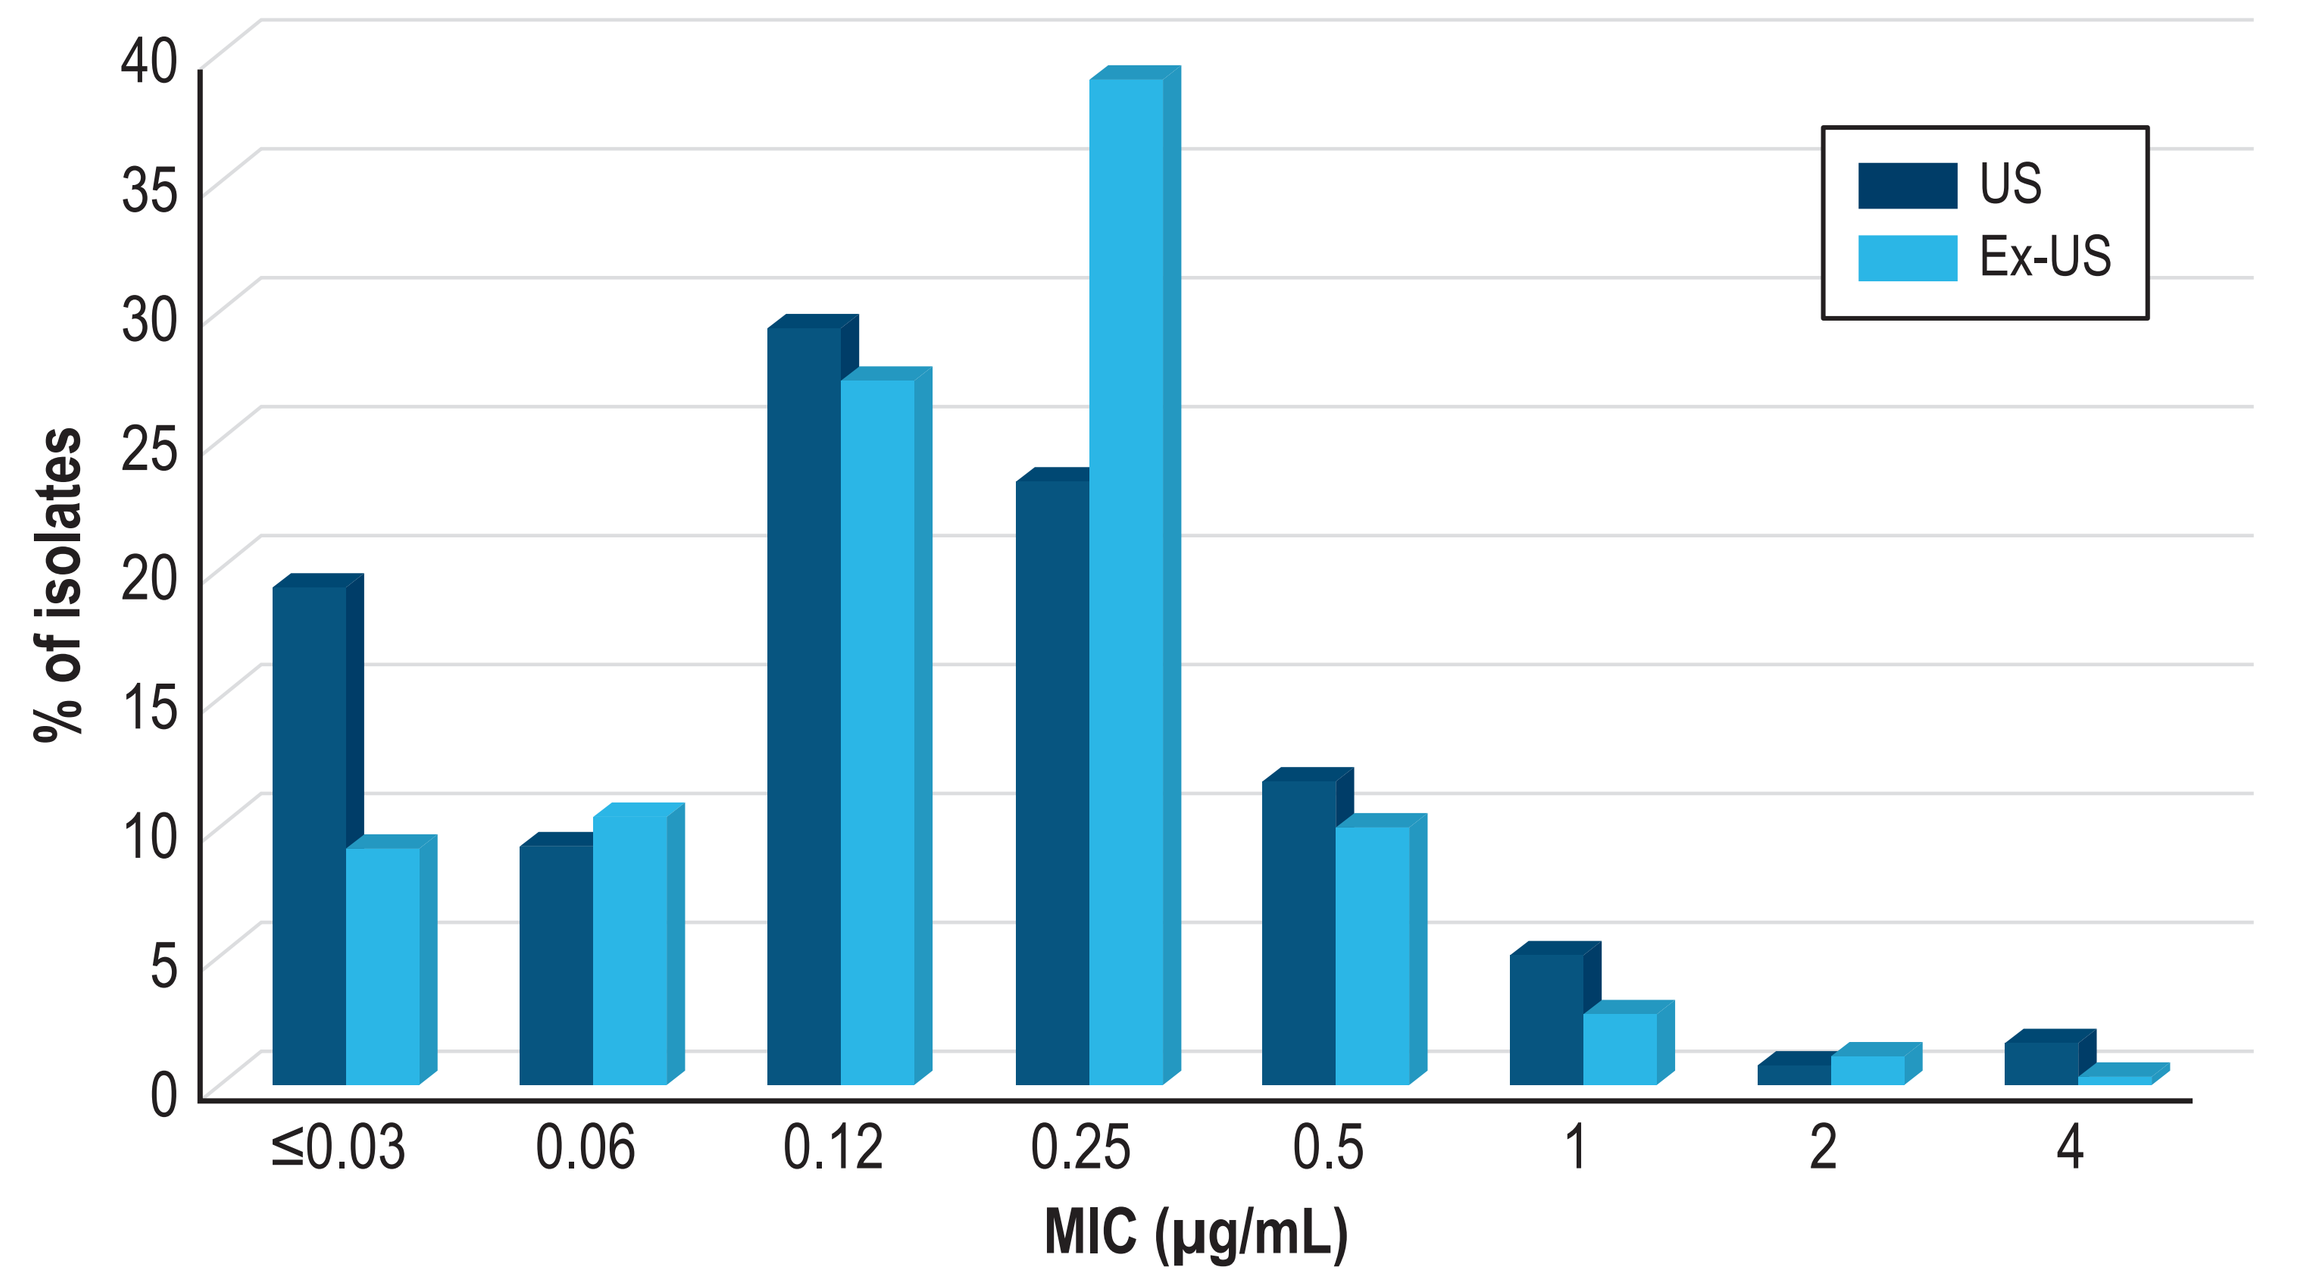

% of isolates
40
35
30
25
20
15
10
5
0
≤0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/mL)
US
Ex-US

Table 1 Antimicrobial activity of aztreonam-avibactam tested against *Enterobacteriaceae* from US hospitals (2016)

Organism / organism group (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %) ≤0.03	0.06	0.12	0.25	0.5	1	2	4	8	MIC_{50}	MIC_{90}
Enterobacteriaceae (10,451)	6,486 (62.1)	2,378 (84.8)	1,023 (94.6)	333 (97.8)	152 (99.2)	64 (99.9)	10 (>99.9)	4 (>99.9)	1 (100.0)	≤0.03	0.12
MDR (876)	408 (46.6)	157 (64.5)	133 (79.7)	92 (90.2)	48 (95.7)	26 (98.6)	7 (99.4)	4 (99.9)	1 (100.0)	0.06	0.25
XDR (111)	28 (25.2)	8 (32.4)	27 (56.8)	27 (81.1)	14 (93.7)	5 (98.2)	2 (100.0)			0.12	0.5
PDR (2)	1 (50.0)	0 (50.0)	1 (100.0)							≤0.03	

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; PDR, pan drug-resistant.

Table 2 Activity of aztreonam-avibactam and comparator antimicrobial agents when tested against isolates from United States hospitals (2016)

Organisms / antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R
All *Enterobacteriaceae* (US; 10,451)				
Aztreonam-avibactam[b]	≤0.03	0.12	NA[b]	NA
Aztreonam	0.06	16	87.8	10.9
Ceftriaxone	≤0.06	>8	84.4	14.6
Piperacillin-tazobactam	2	16	92.5	3.9
Meropenem	0.03	0.06	98.8	1.0
Levofloxacin	0.06	>4	83.1	14.8
Gentamicin	0.5	2	91.5	7.4
Amikacin	2	4	99.4	0.2
Tigecycline[c]	0.25	1	96.8	0.1
Colistin[d]	0.25	>8	79.1	20.9
CRE (US; 120)[e]				
Aztreonam-avibactam[b]	0.12	0.5	NA	NA
Aztreonam	>16	>16	2.5	97.5
Ceftriaxone	>8	>8	1.7	97.5
Piperacillin-tazobactam	>64	>64	3.3	89.2
Meropenem	8	>32	4.2	87.5
Levofloxacin	>4	>4	26.7	66.7
Gentamicin	8	>8	43.3	34.2
Amikacin	8	32	74.2	6.7
Tigecycline[c]	0.5	2	96.7	0.0
Colistin[d]	0.25	>8	81.7	18.3
MDR (US; 876)[f]				
Aztreonam-avibactam[b]	0.06	0.25	NA	NA
Aztreonam	>16	>16	34.1	62.6
Ceftriaxone	>8	>8	23.4	74.2
Piperacillin-tazobactam	16	>64	56.8	25.6
Meropenem	0.06	4	86.2	11.8
Levofloxacin	>4	>4	21.8	68.4
Gentamicin	8	>8	40.0	49.4
Amikacin	4	16	93.4	1.7
Tigecycline[c]	0.5	4	82.4	0.3
Colistin[d]	0.25	>8	58.3	41.7
XDR (US; 111)[g]				
Aztreonam-avibactam[b]	0.12	0.5	NA	NA
Aztreonam	>16	>16	16.2	82.9
Ceftriaxone	>8	>8	4.5	93.7
Piperacillin-tazobactam	>64	>64	13.5	78.4
Meropenem	8	>32	30.6	64.0
Levofloxacin	>4	>4	9.9	80.2
Gentamicin	>8	>8	18.9	51.4
Amikacin	8	>32	65.8	11.7
Tigecycline[c]	1	4	79.3	0.0
Colistin[d]	0.25	>8	57.7	42.3
CRE ex-US (250)[h]				
Aztreonam-avibactam[b]	0.25	0.5	NA	NA
Aztreonam	>16	>16	6.0	93.6
Ceftriaxone	>8	>8	3.2	96.0
Piperacillin-tazobactam	>64	>64	0.4	97.2
Meropenem	32	>32	2.4	94.0
Levofloxacin	>4	>4	14.4	83.2
Gentamicin	2	>8	51.6	47.2
Amikacin	8	>32	58.0	23.2
Tigecycline[c]	0.5	2	98.0	0.4
Colistin[d]	0.25	>8	79.2	20.8

[a] Criteria as published by CLSI
[b] NA, not applicable. Breakpoints for aztreonam-avibactam have not been defined.
[c] Breakpoints from US FDA Package Insert
[d] EUCAST breakpoints of ≤2 µg/mL for susceptible and ≥4 for resistant were applied
[e] Organisms include: *Citrobacter freundii* species complex (4), *Enterobacter aerogenes* (5), *E. cloacae* species complex (15), *Escherichia coli* (5), *Klebsiella oxytoca* (7), *K. pneumoniae* (74), *Proteus mirabilis* (2), *Providencia stuartii* (1), *Raoultella ornithinolytica* (1), *Serratia marcescens* (4), unspeciated *Raoultella* (2)
[f] Organisms include: *Citrobacter freundii* species complex (20), *C. koseri* (1), *Enterobacter aerogenes* (11), *E. cloacae* species complex (75), *Escherichia coli* (242), *Hafnia alvei* (3), *Klebsiella oxytoca* (22), *K. pneumoniae* (194), *Morganella morganii* (51), *Proteus mirabilis* (158), *P. vulgaris* group (1), *Providencia rettgeri* (5), *P. stuartii* (58), *Raoultella ornithinolytica* (1), *Serratia liquefaciens* (1), *S. marcescens* (30), unspeciated *Raoultella* (2), unspeciated *Serratia* (1)
[g] Organisms include: *Citrobacter freundii* species complex (4), *Enterobacter cloacae* species complex (14), *Escherichia coli* (1), *Klebsiella oxytoca* (3), *K. pneumoniae* (60), *Morganella morganii* (6), *Proteus mirabilis* (9), *Providencia stuartii* (8), *Raoultella ornithinolytica* (1), *Serratia marcescens* (4), unspeciated *Raoultella* (1)
[h] Organisms include: *Citrobacter freundii* (3), *Enterobacter aerogenes* (2), *E. cloacae* (14), *Escherichia coli* (10), *Klebsiella oxytoca* (4), *K. pneumoniae* (210), *Pluralibacter gergoviae* (1), *Proteus mirabilis* (2), *Providencia stuartii* (1), *Raoultella ornithinolytica* (1), *Serratia marcescens* (2)
Abbreviations: CRE, carbapenem-resistant *Enterobacteriaceae*; MDR, multidrug-resistant; XDR, extensively drug-resistant

Table 3 Carbapenemase results by organism for 120 carbapenem-resistant *Enterobacteriaceae* (CRE) isolates collected during 2016 in the United States

Organism (no. tested)	No. of positive results IMP-27	NDM-1	KPC-2	KPC-3	KPC-4	SME-4	No. tested negative
Overall (120)	**1**	**1**	**35**	**66**	**1**	**2**	**14**
Citrobacter freundii (4)			2	2			
Enterobacter aerogenes (5)			1				4
Escherichia coli (5)			2				3
Enterobacter cloacae (15)		1	1	9	1		3
Klebsiella oxytoca (7)			1	6			
Klebsiella pneumoniae (74)			27	44			3
Proteus mirabilis (2)	1						1
Providencia stuartii (1)			1				
Unspeciated *Raoultella* (2)				2			
Raoultella ornithinolytica (1)				1			
Serratia marcescens (4)				2		2	

RESULTS

- All US *Enterobacteriaceae* isolates ($MIC_{50/90}$, ≤0.03/0.12 µg/mL), except for 1 *Escherichia coli* strain with an aztreonam-avibactam MIC of 8 µg/mL (Table 1), and all ex-US CRE isolates ($MIC_{50/90}$, 0.25/0.5 µg/mL) were inhibited at aztreonam-avibactam MIC of ≤4 µg/mL (Figure 1)
- Among US isolates, aztreonam-avibactam was also very active against CRE (n=120; $MIC_{50/90}$, 0.12/0.5 µg/mL; highest MIC, 4 µg/mL), MDR (n=876; $MIC_{50/90}$, 0.06/0.25 µg/mL), XDR (n=111; $MIC_{50/90}$, 0.12/0.5 µg/mL), PDR (n=2; MICs ≤0.03 and 0.12 µg/mL), and ceftazidime-nonsusceptible *Enterobacter cloacae* ($MIC_{50/90}$, 0.25/1 µg/mL) isolates (Tables 1 and 2)
- Meropenem was very active against US *Enterobacteriaceae* isolates overall ($MIC_{50/90}$, 0.03/0.06 µg/mL; 98.8%S per CLSI), but showed limited activity against MDR (86.2%S) and XDR (30.6%S) isolates (Table 2)
- Amikacin and colistin were active against 74.2% and 81.7% of US CRE isolates, 93.4% and 58.3% US MDR, 65.8% and 57.7% of US XDR, and 58.0% and 79.2% of ex-US CRE isolates, respectively (Table 2)
- A total of 106 carbapenemase-encoding genes were detected in 106 US CRE isolates, including 102 KPC-like, 2 SME-4, 1 NDM-1, and 1 IMP-27 (Table 3)
- 248 carbapenemase-encoding genes were identified on 241 ex-US CRE isolates, including 124 KPC-like, 64 OXA-like, 50 NDM-like, 7 VIM-1, 2 IMP-4, and 1 SME-4 (Table 4)
- All CRE isolates, including all carbapenemase-producing *Enterobacteriaceae* (US and ex-US), were inhibited at aztreonam-avibactam MIC of ≤4 µg/mL (Figure 1 and Table 5)
- Whole genome sequencing results of the isolate with the aztreonam-avibactam MIC of 8 µg/mL showed 3 serine β-lactamase genes (bla_{CMY-42}, $bla_{OXA-1/30}$, and bla_{TEM-1}) and a 4-amino acid insertion (RIKY) at position 335 of PBP3. This isolate also displayed reduced expression of OmpF, and alterations in OmpC, whereas expression of AcrAB-TolC multidrug efflux pump was similar to baseline

Table 4 Carbapenemase results by organism for 250 carbapenem-resistant *Enterobacteriaceae* (CRE) isolates collected during 2016 worldwide (ex-US)

Organism (no. tested)	No. of positive results[a] IMP-4	NDM-1	NDM-5	NDM-6	NDM-7	VIM-1	OXA-23	OXA-48	OXA-232	OXA-244	OXA-370	KPC-2	KPC-3	KPC-12	SME-4	No. tested negative
Overall (250)	**2**	**40**	**2**	**1**	**7**	**7**	**1**	**56**	**5**	**1**	**1**	**47**	**76**	**1**	**1**	**9**
Citrobacter freundii (3)						2						1				
Enterobacter aerogenes (2)																2
Escherichia coli (10)		2	2		1			1	2			2	2			
Enterobacter cloacae (14)		5			2	3		2				1	1			
Pluralibacter gergoviae (1)												1				
Klebsiella oxytoca (4)		2										2				
Klebsiella pneumoniae (210)	2	30		1	4	1		50	3	1	1	40	73	1	1	7
Proteus mirabilis (2)		1					1									
Providencia stuartii (1)						1										
Raoultella ornithinolytica (1)								1								
Serratia marcescens (2)								2								

[a] Some isolates had more than 1 carbapenemase.

Table 5 Antimicrobial activity of aztreonam-avibactam tested against carbapenem-resistant *Enterobacteriaceae* (CRE) isolates stratified by carbapenemase type

Organism / organism group (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %) ≤0.03	0.06	0.12	0.25	0.5	1	2	4	MIC_{50}	MIC_{90}
United States										
All CRE isolates (120)	23 (19.2)	11 (28.3)	35 (57.5)	28 (80.8)	14 (92.5)	6 (97.5)	1 (98.3)	2 (100.0)	0.12	0.5
KPC-producers (102)	20 (19.6)	10 (29.4)	32 (60.8)	25 (85.3)	11 (96.1)	4 (100.0)			0.25	0.5
SME-4-producers (2)			1 (50.0)	1 (100.0)					0.12	
MBL-producers (2)	1 (50.0)	0 (50.0)	0 (50.0)	0 (50.0)	0 (50.0)	1 (100.0)			≤0.03	
Carbapenemase negative (14)	2 (14.3)	1 (21.4)	2 (35.7)	2 (50.0)	3 (71.4)	1 (78.6)	1 (85.7)	2 (100.0)	0.25	4
Worldwide (ex-US)										
All CRE isolates (250)	23 (9.2)	26 (19.6)	68 (46.8)	97 (85.6)	25 (95.6)	7 (98.4)	3 (99.6)	1 (100.0)	0.25	0.5
KPC-producers (excluding MBL-producers; 124)	13 (10.5)	16 (23.4)	30 (47.6)	43 (82.3)	17 (96.0)	5 (100.0)			0.25	0.5
OXA-48-like-producers (excluding MBL-producers; 58)[a]	2 (3.4)	2 (6.9)	17 (36.2)	33 (93.1)	4 (100.0)				0.25	0.25
MBL-producers (59)[b]	7 (11.9)	7 (23.7)	21 (59.3)	17 (88.1)	4 (94.9)	0 (94.9)	2 (98.3)	1 (100.0)	0.12	0.5
Carbapenemase negative (9)		1 (11.1)	0 (11.1)	5 (66.7)	0 (66.7)	2 (8.9)	1 (100.0)		0.25	

[a] Includes 56 OXA-48-, 1 OXA-244-, and 1 OXA-370-producing strains
[b] Includes 36 NDM-1-, 7 NDM-7-, 7 VIM-1-, 3 NDM-1 plus OXA-232-, 2 NDM-5 plus OXA-232-, 2 IMP-4-, 1 NDM-1 plus OXA-48-, and 1 NDM-6-producing strains
Abbreviations: CRE, carbapenem-resistant *Enterobacteriaceae*; MBL, metallo-β-lactamase

CONCLUSIONS

- Aztreonam-avibactam demonstrated potent *in vitro* activity against a large collection of contemporary (2016) *Enterobacteriaceae* isolates from patients in US hospitals and CRE isolates collected worldwide, including NDM, KPC, OXA, VIM, and SME producers
- Results of the *in vitro* activity of aztreonam combined with avibactam presented here, coupled with clinical data currently available for aztreonam used alone and for avibactam used in combination with ceftazidime, indicate that aztreonam-avibactam may represent a valuable option for treating infections caused by CRE, MDR, and XDR *Enterobacteriaceae*, including MBL-producing strains

ACKNOWLEDGEMENTS

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing this poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. Alm RA, Johnstone MR, Lahiri SD (2015). Characterization of *Escherichia coli* NDM isolates with decreased susceptibility to aztreonam/avibactam: role of a novel insertion in PBP3. *J Antimicrob Chemother* 70: 1420-1428.
2. Brogden RN, Heel RC (1986). Aztreonam. A review of its antibacterial activity, pharmacokinetic properties and therapeutic use. *Drugs* 31: 96-130.
3. Castanheira M, Mendes RE, Jones RN, et al. (2016). Changes in the frequencies of beta-lactamase genes among *Enterobacteriaceae* isolates in U.S. hospitals, 2012 to 2014: activity of ceftazidime-avibactam tested against beta-lactamase-producing isolates. *Antimicrob Agents Chemother* 60: 4770-4777.
4. Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.
5. EUCAST (2017). Breakpoint tables for interpretation of MIC's and zone diameters. Version 7.1, March 2017. Available at http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_7.1_Breakpoint_Tables.pdf. Accessed March 2017.
6. Falcone M, Paterson D (2016). Spotlight on ceftazidime/avibactam: a new option for MDR Gram-negative infections. *J Antimicrob Chemother* 71: 2713-2722.
7. Magiorakos AP, Srinivasan A, Carey RB, et al. (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268-281.
8. Ramsey C, MacGowan AP (2016). A review of the pharmacokinetics and pharmacodynamics of aztreonam. *J Antimicrob Chemother* 71: 2704-2712.
9. Tygacil Package Insert (2016). Available at www.tygacil.com. Accessed February 23, 2017.
10. Vasoo S, Cunningham SA, Cole NC, et al. (2015). *In vitro* activities of ceftazidime-avibactam, aztreonam-avibactam, and a panel of older and contemporary antimicrobial agents against carbapenemase-producing gram-negative bacilli. *Antimicrob Agents Chemother* 59: 7842-7846.

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit www.allergancongressposters.com/194928

Charges may apply.
No personal information is stored.

Allergan

IDWeek 2017, October 4–8, San Diego, California